EE638

ISPOR Annual European Congress, 12th-15th November 2023, Copenhagen, Denmark

Cost-effectiveness of Vasopressin alone or in Combination with SOC in the Treatment of Sepsis and Septic Shock patients

Evelyn Walter[a], Gerald Eichhober[a], Marco Voit[a]

[a] IPF Institute for Pharmaeconomic Research, Vienna, Austria

Objectives

Sepsis is a burdensome condition worldwide in terms of morbidity, mortality and financial cost to health systems. The recent Surviving Sepsis Campaign (SSC) has published new guidelines for septic patients. However, deviation from guidelines is frequent in intensive care units. A retrospective cohort study using a least absolute shrinkage and selection operator penalized machine learning model, carried out by Kalimouttou and colleagues, has identified and analyzed 22 recommendations (77,2% compliance with guidelines) extracted from 2 US databases. Among the 22 items, 6 are associated with a reduction in 28-day all-cause mortality; one is Vasopressin (OR: 0.74, 95%CI 0.65;0.86). The aim of this analysis is to analyze the cost-effectiveness of adding vasopressin to standard-of-care (SOC) versus SOC to assess outcomes (costs, mortality, ICU stay, requirement of RRT etc.)

Methods

A decision tree structure plus Markov model is used to describe the time-span of the sepsis related hospitalization over 28 days. A Markov structure is then applied to represent long-term outcomes including survival, adverse effects (ESRD and atrial fibrillation [AF]) and quality-adjusted life-years (QALYs). Resource-utilization and direct costs (2023 €) were derived from published sources from the German payer's perspective. A willingness-to-pay threshold set at 25,000 €/QALY gained was incorporated. One-way and probabilistic sensitivity analysis were performed.

Figure 1: Model design

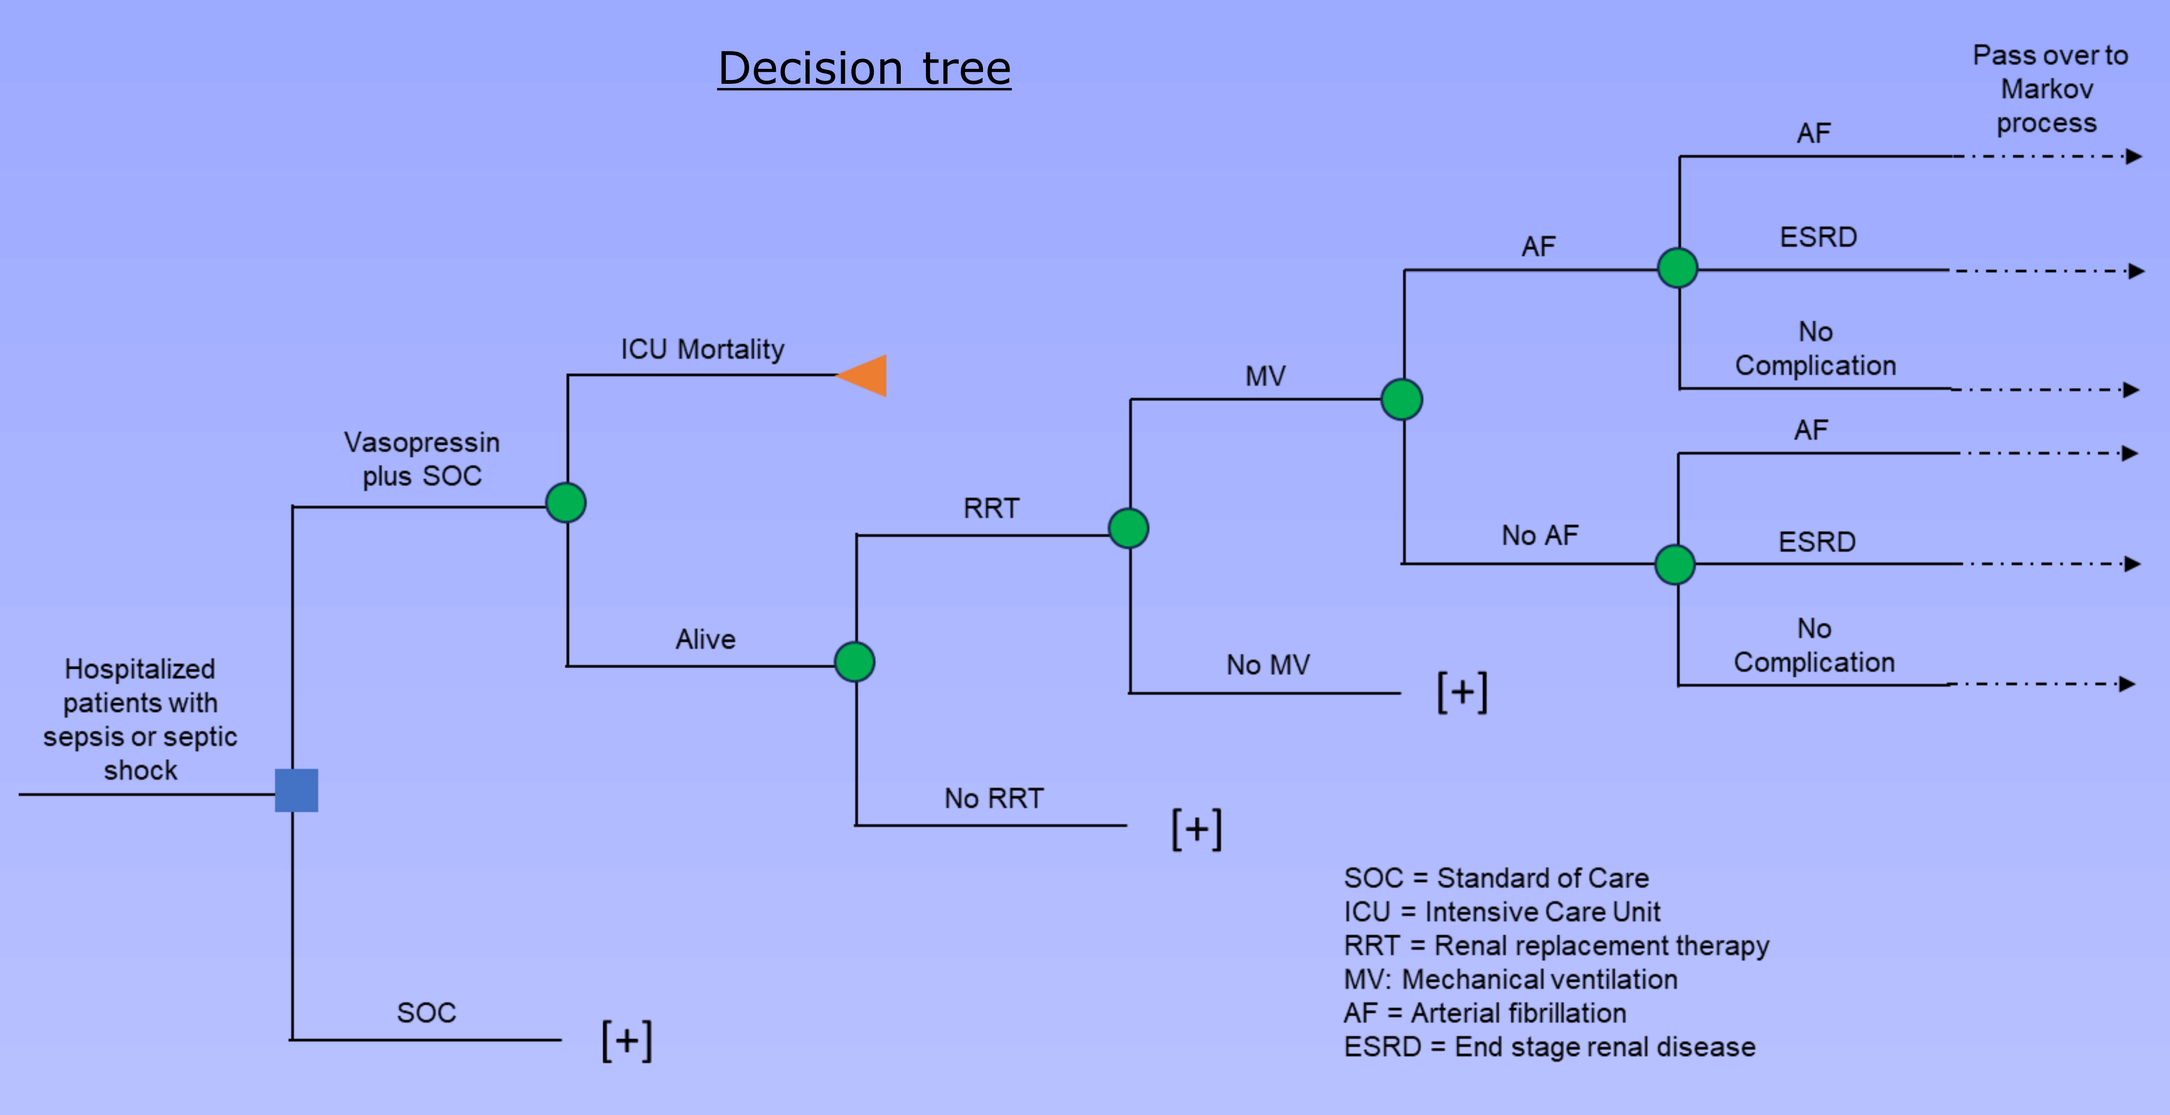
Decision tree
Hospitalized patients with sepsis or septic shock
Vasopressin plus SOC
SOC
ICU Mortality
Alive
RRT
No RRT
MV
No MV
AF
No AF
ESRD
No Complication
Pass over to Markov process
[+]
SOC = Standard of Care
ICU = Intensive Care Unit
RRT = Renal replacement therapy
MV: Mechanical ventilation
AF = Arterial fibrillation
ESRD = End stage renal disease

Decision tree. The square is a decision node from which the competing strategies originate. The circles are chance nodes from which a study result leads to a particular management of the patient. The triangles are the end-points of each pathway and represent the final outcome.

- The decision tree describes a period of 28 days.
- The Odds Ratio (OR) of mortality over 28 days (= hospital or ICU mortality) is 0.74 when Vasopressin is administered according to the recommendation (Kalimoutou et al. 2023)
- In addition the need for Mechanical Ventilation (MV) decreases (OR: 0.94).
- To be able to show the long-term consequences of arrhythmia, these data were supplemented with the literature (Lam et al. 2020)

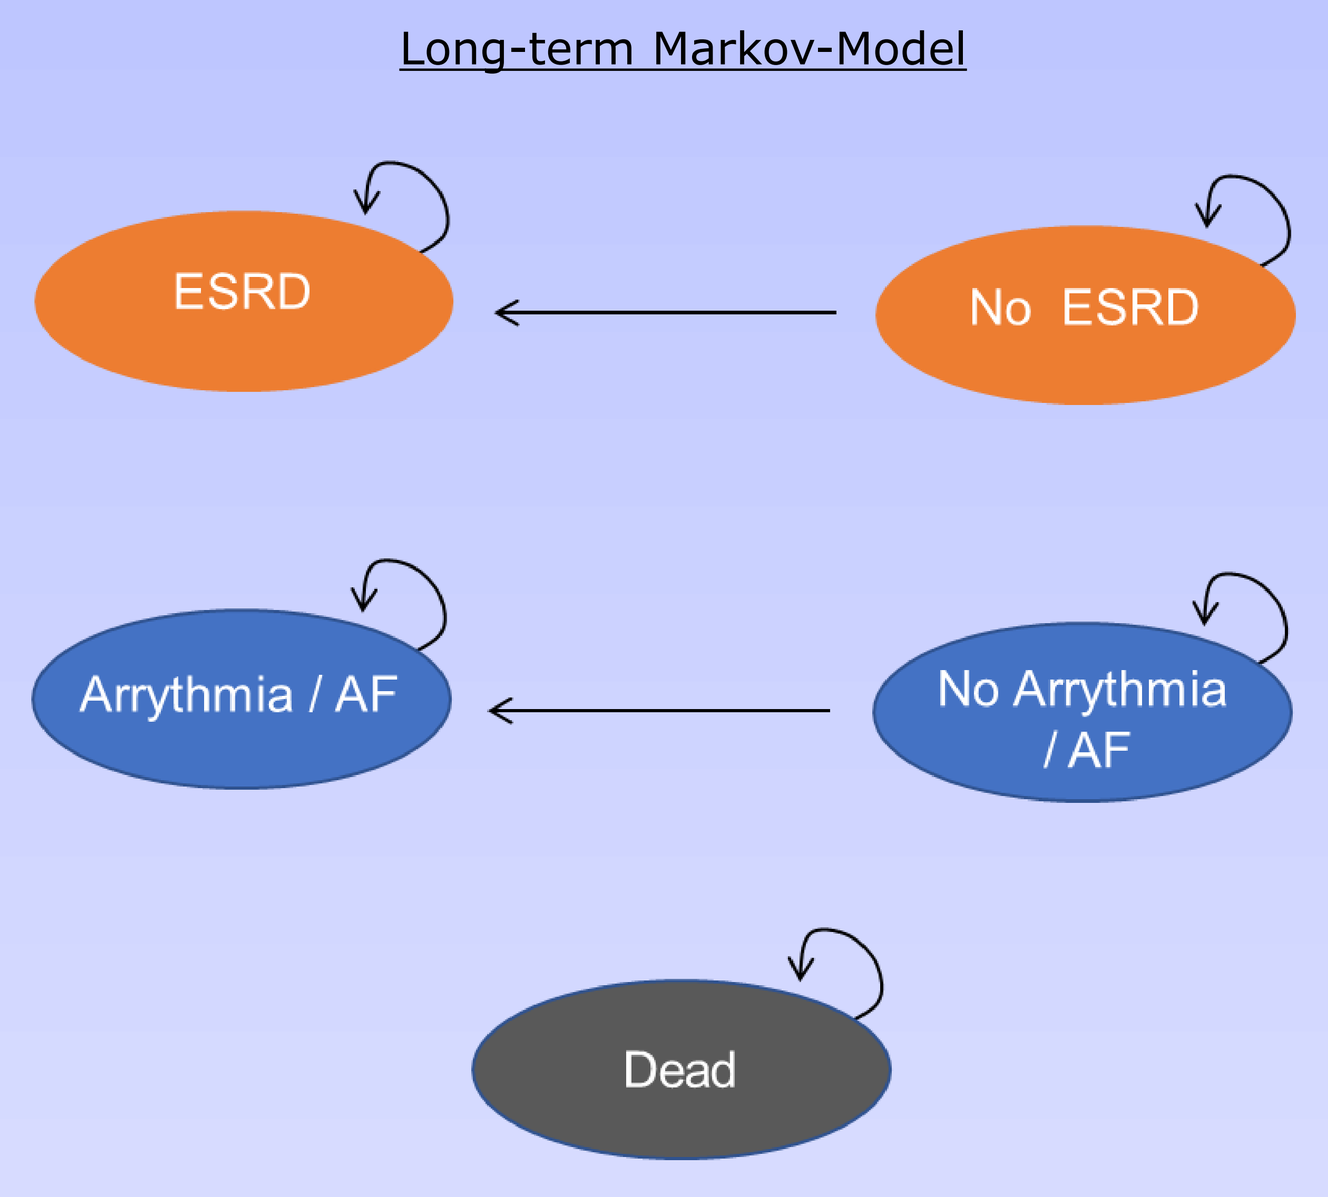
Long-term Markov-Model
ESRD
No ESRD
Arrythmia / AF
No Arrythmia / AF
Dead

- All patients who are alive after 28 days run into the Markov model.
- Patients who have not discontinued RRT after 28 days and require renal replacement therapy will be considered for dialysis in the long-term model. 41% quit therapy after 28 days (Chen et al. 2022).
- AF occurrence following sepsis hospitalization was more common among patients with new-onset AF during sepsis. Data over 5 years from the literature were used to map the long-term consequences (Walkey et al. 2014).
- The death state is an absorbing state and can be entered from any state.

Source: own developed

Tab. 1: Overview of methods applied

Methods	
Type of study	**Cost-utility analysis (CUA) and Cost-effectiveness analysis (CEA)**
Type of the model	**Deterministic hybrid model** using • A decision tree and • A Markov-cohort-state-transition model
Perspective	**German health care systems perspective** (payers' perspective; direct costs)
Time horizon Cycle length	**Lifetime** First cycle 28 days, second cycle 337 days than yearly cycles.
Discount rate	**3% for costs and 3% for outcomes**
Population	**Patients with sepsis or septic shock**. Eligible participants were adults (mean age 66 years).
Intervention	**Vasopressin** plus standard-of-care (SOC) versus SOC
Comparator	**BSC** (Bundel of recommendations without Vasopressin)
Direct costs	**Direct costs:** Costs hospital stay (normal ward), costs ICU stay, costs RRT, cost of re-hospitalization, dialysis costs, AF costs.
Outcomes	• **Quality-adjusted life-years (QALYs)** based on EQ-5D from the literature • **Disutilities** for AF and ESRD
Results	**Incremental cost utility ratio (ICUR) & incremental cost effectiveness ratio (ICER)**
Timing	**2023**

Source: own developed

Mortality

Sepsis and septic shock are subsequently associated with increased mortality. Patients with additional complications are at increased risk. This is taken into account in the model as follows (Lam et al. 2020):

- In case of renal replacement therapy the OR amounts to 2.17 (Range, 2.15-2.20).
- In case of AF the OR amounts to 2.47 (Range, 1.89-3.24).
- The long-term mortality following a sepsis were analyzed by Wang et al. (2014). Based on a multivariable analysis, the association of sepsis with increased all-cause mortality persisted for up to 5 years, after adjustment for confounders. HR were published which compare the mortality after sepsis with the overall population. These HR were uses for patients without AF. HR <1 year 13.07 to 5+ years 1.41.

Direct costs

The model includes costs of hospital stay (normal ward), costs of ICU stay, costs of RRT, cost of re-hospitalization and dialysis costs. Costs were presented from the payer perspective.

- Costs for Vasopressin are included in the diagnosis-related group (G-DRG) costs and do not represent a single cost component.
- Costs of hospital stay were derived from the G-DGR catalogue. All relevant G-DGR codes were selected to calculate an average value for cost per hospital stay. Points per G-DRG code were multiplied with the 2023 base-rate of 4,030.32 Euro per point. This results in an average cost per day of 731.47 € for an average stay of 16 days.

Tab. 2: Unit cost data

Cost components	Unit	Costs	Source
Hospital stay (normal ward)	day	731.47 €	G-DRG 2023
ICU without MV	day	1,618.00 €	Martin et al. 2008
ICU with MV	day	2,015.08 €	Martin et al. 2008
RRT; hospital setting	day	476.25 €	G-DRG; ZE01.01
Readmission	stay	25,092.12 €	calculated
ESRD / Dialysis	year	65,017.11 €	Häckl et al. 2021
AF cost	year	4,394.15 €	Reinhold et al. 2012

- Costs for an ICU day were retrieved from the literature. Martin et al. (2008) have published costs per day and distinguish between patients require mechanical ventilation (MV) and patients without MV.
- Costs taken from the literature were inflated to 2023.

Results

Adding Vasopressin to the SOC to manage sepsis/septic shock is associated with direct costs of 72,135 € (SD, 3,218 €). SOC is associated with total costs of 84,104 € [SD, 4,414 €]. Adding Vasopressin yields cost savings of about 11,969 € per patient and a QALY gain of 0.53 or 6.3 months in perfect health. Patients in the Vasopressin-group exhibit a shorter ICU-stay (-1 day) and a lower RRT requirement (-11.5%). Vasopressin plus SOC is the dominant strategy.

Tab. 3: Cost effectiveness Results

Costs	Vasopressin plus SOC	SOC	Incremental
TOTAL COSTS	**72,135.72 €**	**84,104.35 €**	**-11.968.63 €**
Outcomes			
QALYs	5.645	5.117	0.527
ICUR	**DOMINANT**		
Net monetary Benefit	**25,154.11 €**		
LYs	8.771	8.026	0.745
ICER	**DOMINANT**		

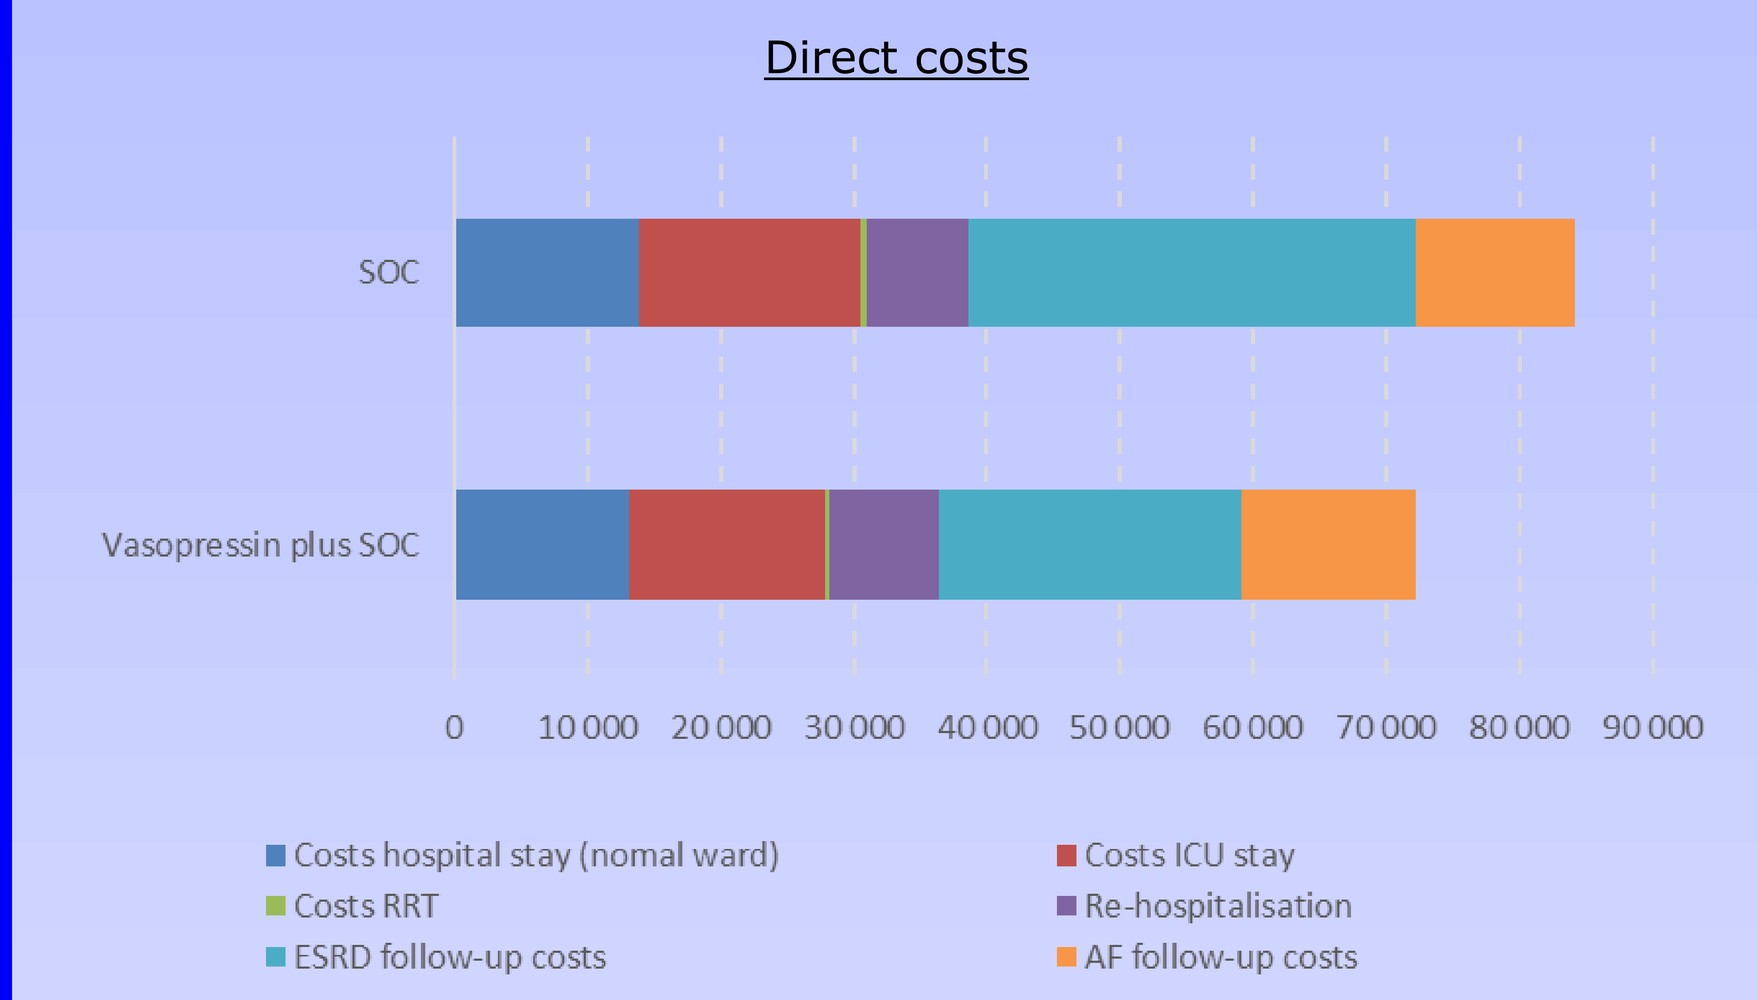
Direct costs
SOC
Vasopressin plus SOC
0
10 000
20 000
30 000
40 000
50 000
60 000
70 000
80 000
90 000
Costs hospital stay (nomal ward)
Costs ICU stay
Costs RRT
Re-hospitalisation
ESRD follow-up costs
AF follow-up costs

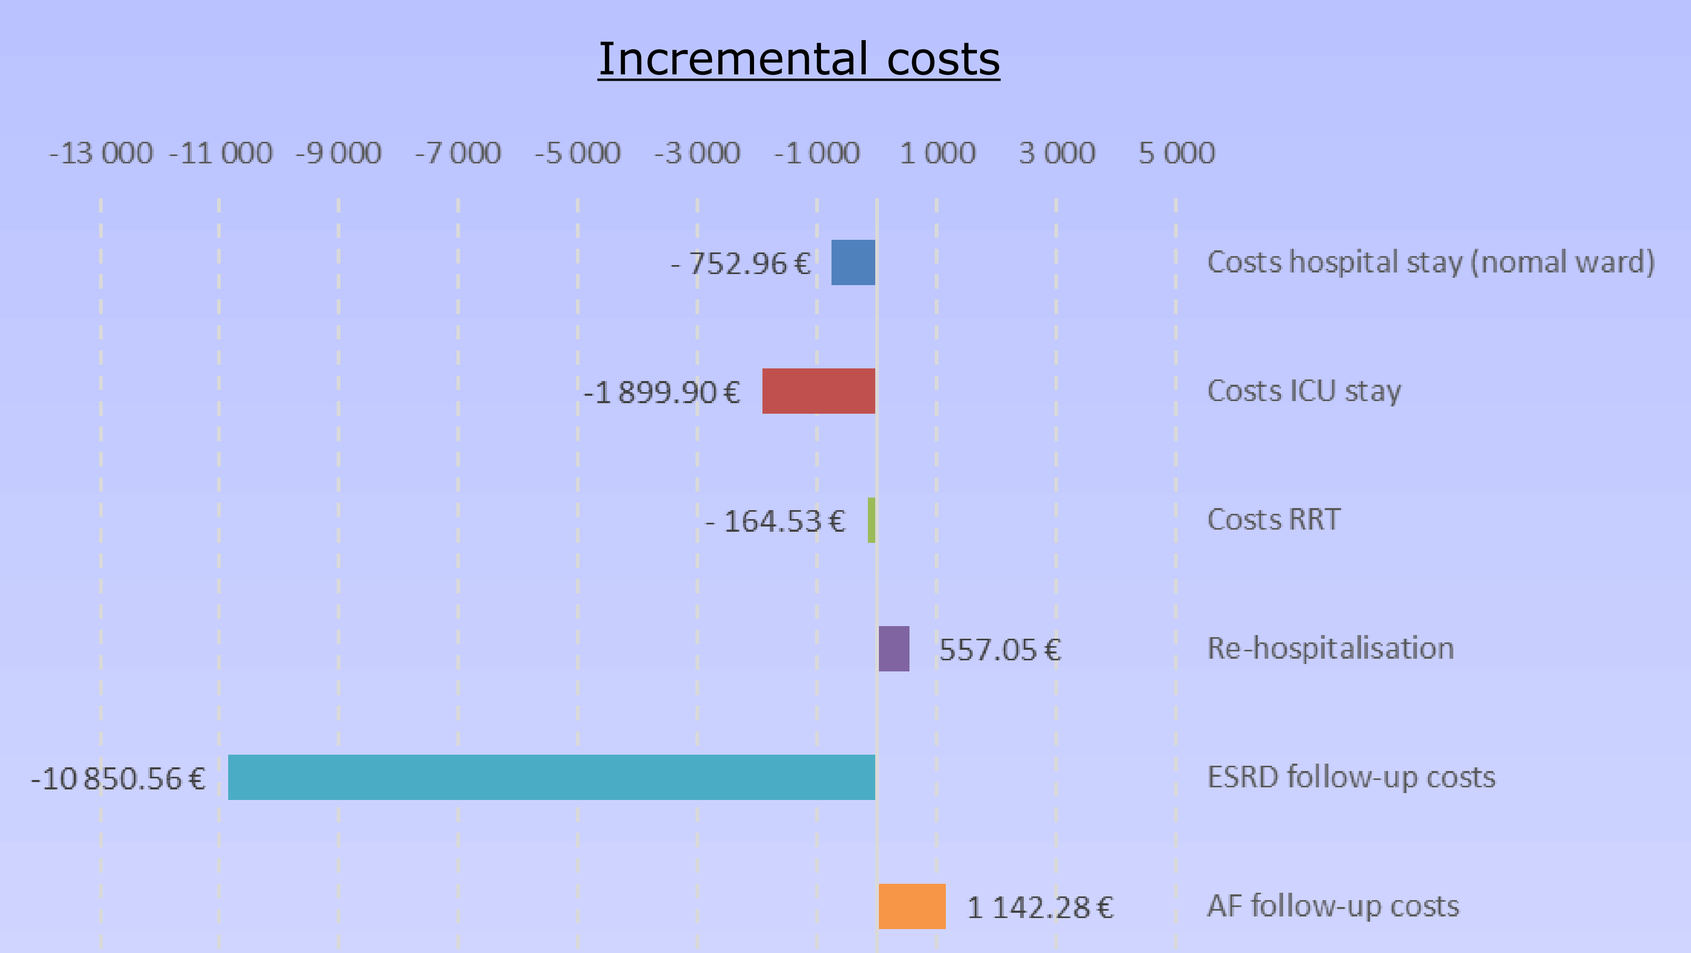
Incremental costs
-13 000 -11 000 -9 000 -7 000 -5 000 -3 000 -1 000 1 000 3 000 5 000
- 752.96 €
-1 899.90 €
- 164.53 €
557.05 €
-10 850.56 €
1 142.28 €
Costs hospital stay (nomal ward)
Costs ICU stay
Costs RRT
Re-hospitalisation
ESRD follow-up costs
AF follow-up costs

- The **additional administration of vasopressin** leads to savings of hospitalization costs; That is a total of 2,817.39 € which could be saved within the first 28 days.
- **Additional costs** are incurred for the **rehospitalizations**, as there is a lower hospital mortality after 28 days (16.2% vs. 21.9%)
- Furthermore, there are **higher follow-up costs for dialysis and AF treatment**, also due to the higher survival rate.

Source: own calculations

Sensitivity Analysis

Probabilistic sensitivity analysis (PSA) and deterministic one-way sensitivity analysis (OWSA) were carried out to examine the robustness of the model.

Fig. 2: Sensitivity Analysis

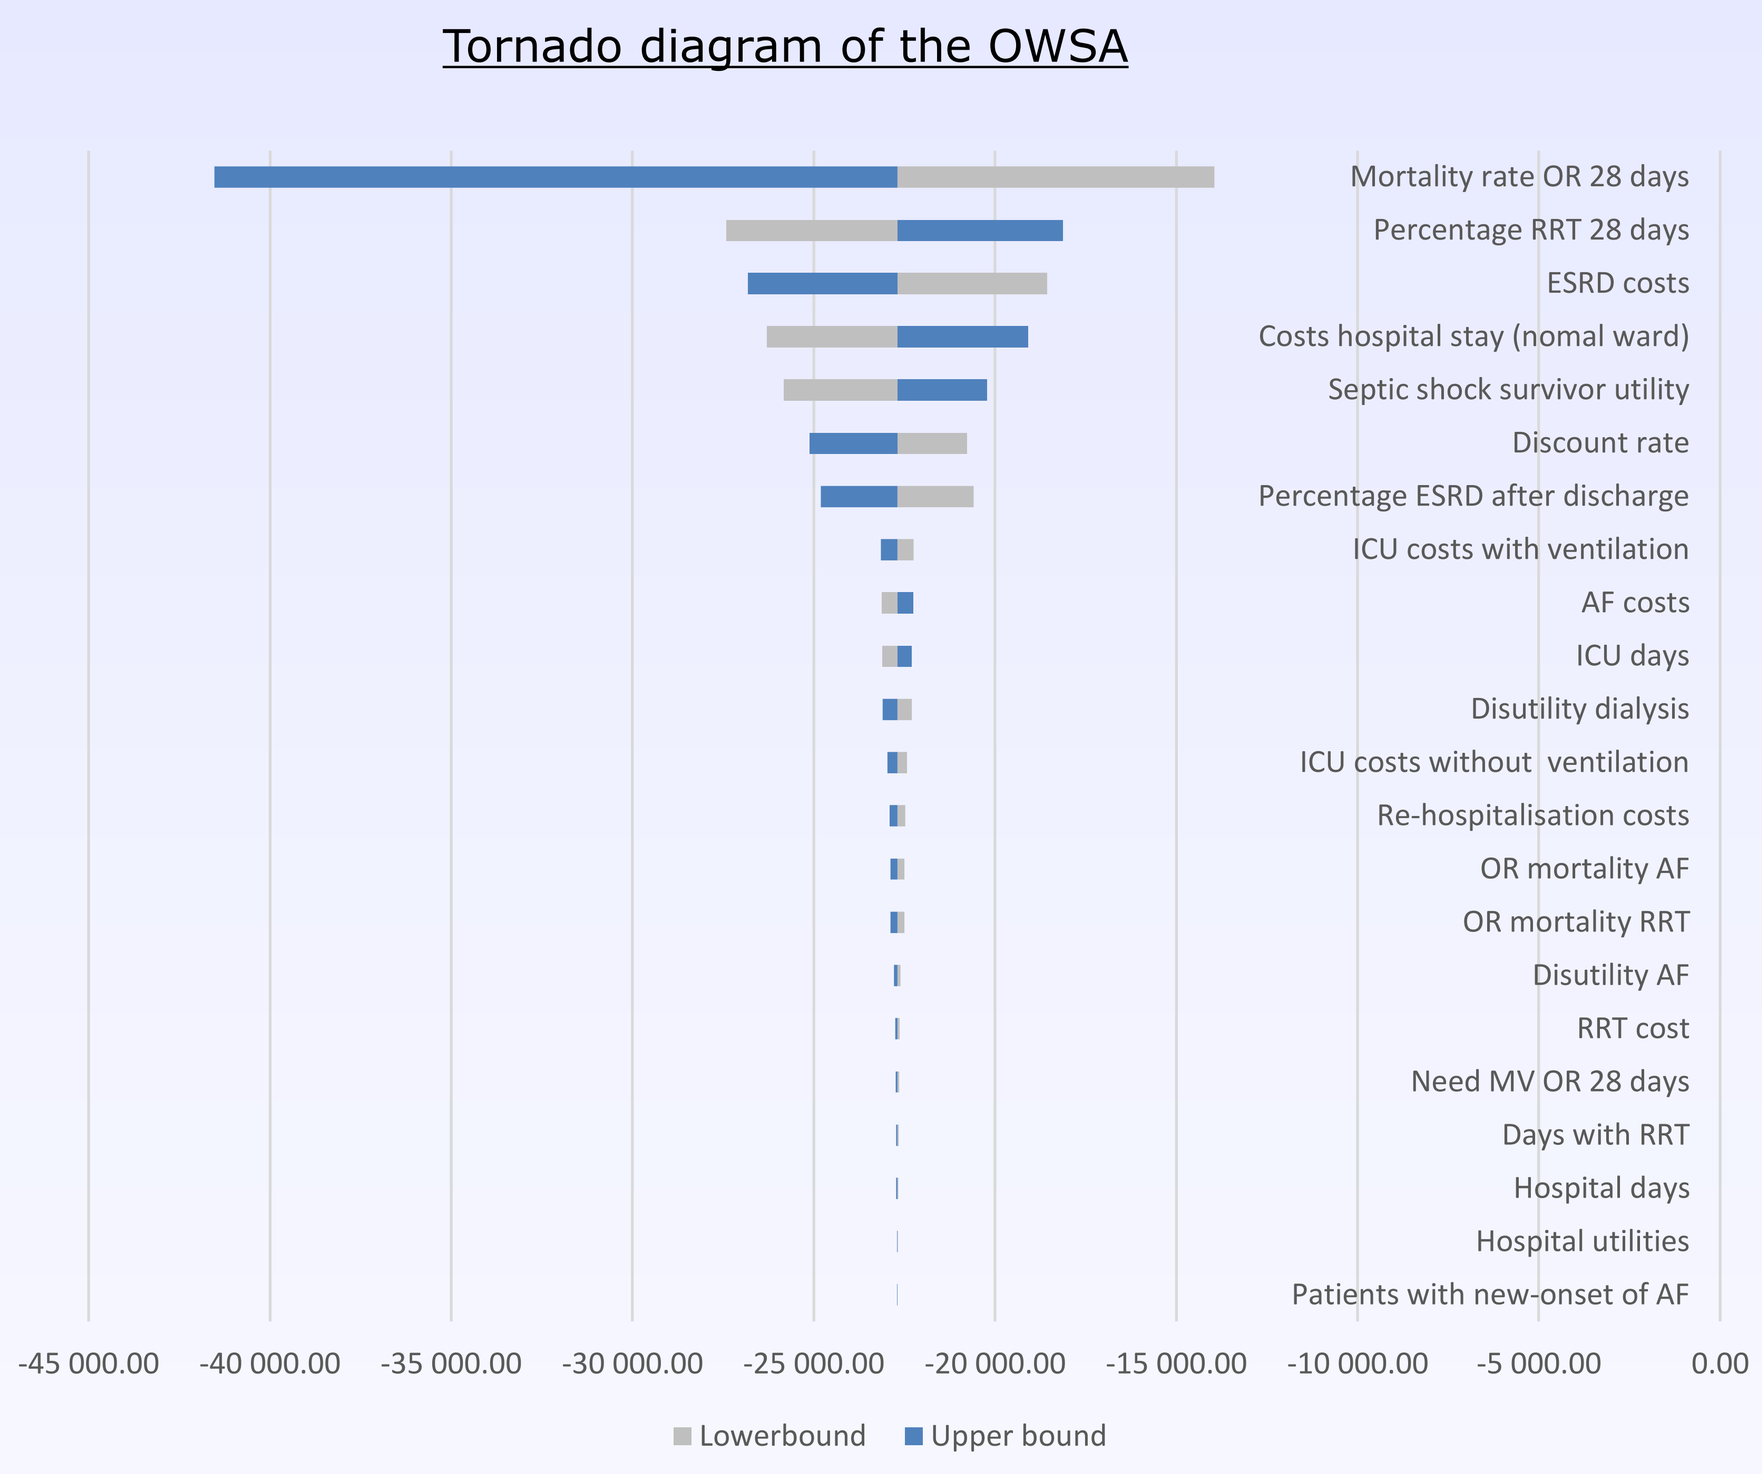
Tornado diagram of the OWSA
Mortality rate OR 28 days
Percentage RRT 28 days
ESRD costs
Costs hospital stay (nomal ward)
Septic shock survivor utility
Discount rate
Percentage ESRD after discharge
ICU costs with ventilation
AF costs
ICU days
Disutility dialysis
ICU costs without ventilation
Re-hospitalisation costs
OR mortality AF
OR mortality RRT
Disutility AF
RRT cost
Need MV OR 28 days
Days with RRT
Hospital days
Hospital utilities
Patients with new-onset of AF
-45 000.00 -40 000.00 -35 000.00 -30 000.00 -25 000.00 -20 000.00 -15 000.00 -10 000.00 -5 000.00 0.00
Lowerbound
Upper bound

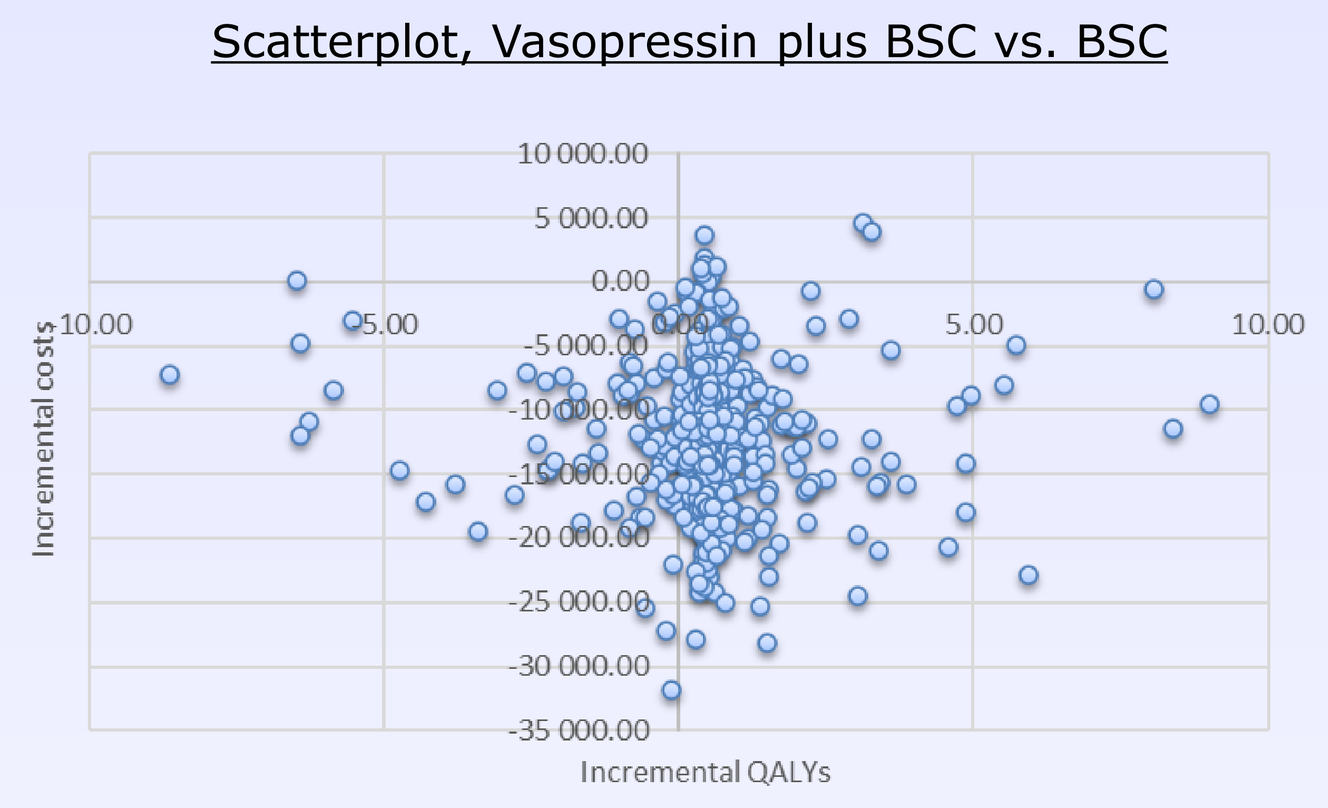
Scatterplot, Vasopressin plus BSC vs. BSC
Incremental costs
Incremental QALYs
10 000.00
5 000.00
0.00
-5 000.00
-10 000.00
-15 000.00
-20 000.00
-25 000.00
-30 000.00
-35 000.00
-10.00
-5.00
5.00
10.00

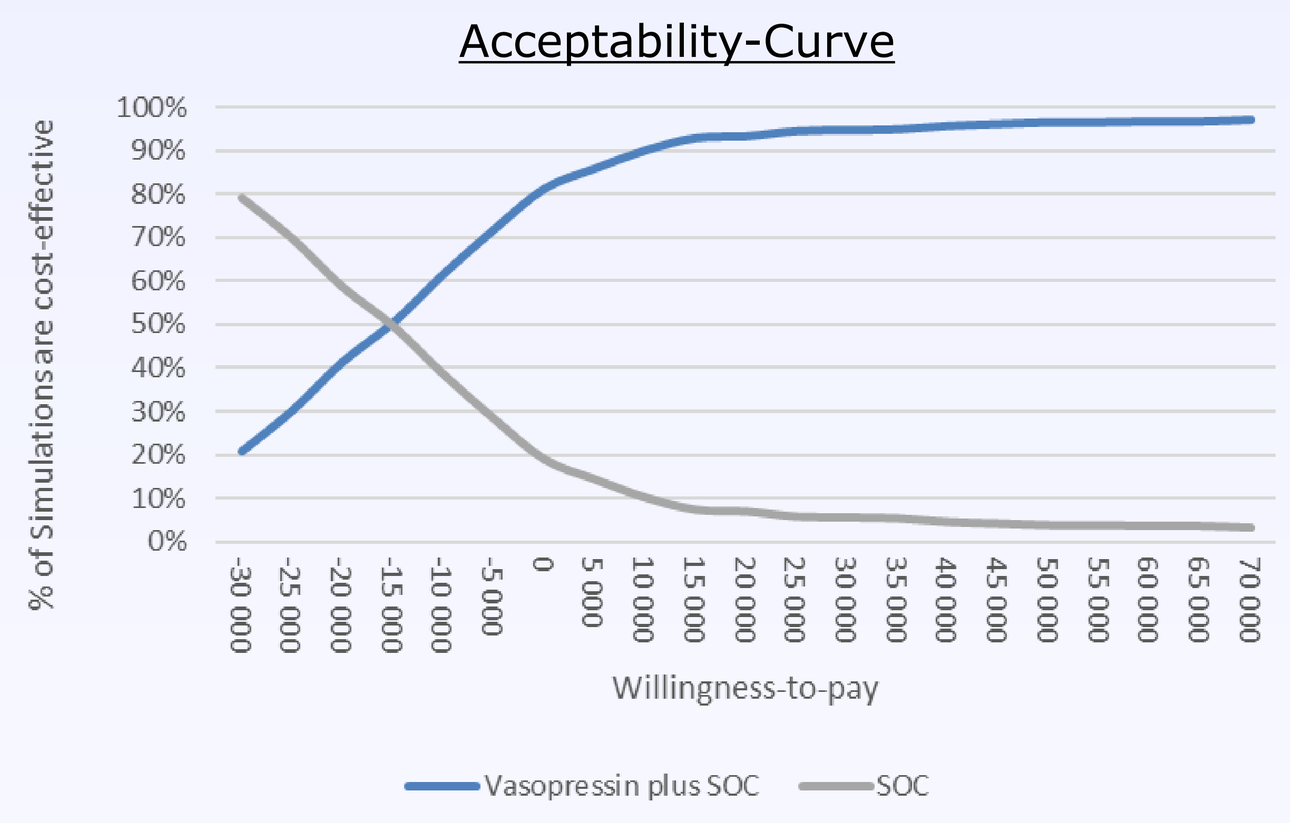
Acceptability-Curve
% of Simulations are cost-effective
Willingness-to-pay
Vasopressin plus SOC
SOC

Source: own calculations

The Monte-Carlo PSA results of 500 second-order simulations plotting incremental costs versus incremental effects. At a willingness-to-pay threshold of 25,000 €, 95.2% of simulation are in the cost-effective range.
OWSA confirms probabilistic results. Variations in the inputs like OR for mortality within first 28 days, the percentage of RRT in the first 28 days and ESRD costs have biggest influence on the result.

Conclusion

In patients with sepsis/septic shock, the use of Vasopressin plus SOC is highly cost-effective and associated with savings for the German healthcare system.

References

Kalimouttou A, Lerner I, Cheurfa C, Jannot AS, Pirracchio R. Machine-learning-derived sepsis bundle of care. Intensive Care Med. 2023;49(1):26-36. doi:10.1007/s00134-022-06928-2
Lam SW, Barreto EF, Scott R, Kashani KB, Khanna AK, Bauer SR. Cost-effectiveness of second-line vasopressors for the treatment of septic shock. J Crit Care. 2020;55:48-55. doi:10.1016/j.jcrc.2019.10.005
Chen JCY, Hu B, Frank RD, Kashani KB. Inpatient Kidney Function Recovery among Septic Shock Patients Who Initiated Kidney Replacement Therapy in the Hospital. Nephron. 2020;144(8):363-371. doi:10.1159/000507999
Wang HE, Szychowski JM, Griffin R, Safford MM, Shapiro NI, Howard G. Long-term mortality after community-acquired sepsis: a longitudinal population-based cohort study. BMJ Open. 2014;4(1):e004283. Published 2014 Jan 17. doi:10.1136/bmjopen-2013-004283

Additional literature with the author

CONTACT:
Dr. Evelyn Walter
IPF Institute for Pharmaeconomic Research
Wolfengasse 4/7
1010 Vienna, Austria
Phone: +43-1-5132007-13
Fax: +43-1-5132007-15
Email: e.walter@ipf-ac.at
Web: www.ipf-ac.at

Funding: This study was funded by a grant from AOP Orphan Pharmaceuticals GmbH