Internal

ECONOMIC BURDEN ON DIABETES AND CARDIOVASCULAR DISEASE: REAL WORLD DATA AND EMPIRICAL ANALYSIS IN ALGERIA

Tebaibia A[1-2], A. Mammeri [1-2], F. Hamrour [1-2], O. Hocine[1-2], M. Ammi[1-2], N. Slimani[1-2], S. Zekri[1-2], Aissaoui A[3], Hachelaf Z[3]
Internal Medicine Department, EPH El Biar (Ex. Birtraria), Algiers, Algeria1,
Algiers University 1, Algeria2
Sanofi Algeria3

Poster # EE475
ISPOR 2023
Nov 12–15, 2023
Copenhagen Denmark

BACKGROUND

- In Algeria, as in many other developing countries, the number of people with type 2 diabetes is increasing reaching 14.4%[2] of prevalence, this is mainly due to urbanization, an aging population, a sedentary lifestyle and an unbalanced diet[1].
- Cardiovascular disease is a leading cause of death in Algeria, a direct consequence of a growing prevalence of the various cardiovascular metabolic risk factors[3].
- All these increases in diabetes prevalence and its complications has led to significant economic burden for healthcare system
- In our Knowledge, no specific study has evaluated this economic burden in Algeria.

OBJECTIVE

To assess direct cost of diabetes and cardiovascular diseases management from hospital perspective

To compare these costs based on real world data.

METHODS

- This was an observational, monocentric, retrospective study conducted during 2 years (2019 & 2020) from patients medical records.
- **Study population**:

Number of patients included are 2300

- **Patient inclusion criteria**: Patient diagnosed in diabetes and cardiovascular disease at service level during study period.

Availability of the patient's medical record containing medical data and resources used by the patient during his or her disease management

- Participating Center:

Study was set up in internal medicine department in EPH* Birtraria Algiers, Algeria.

Figure 1: Study design

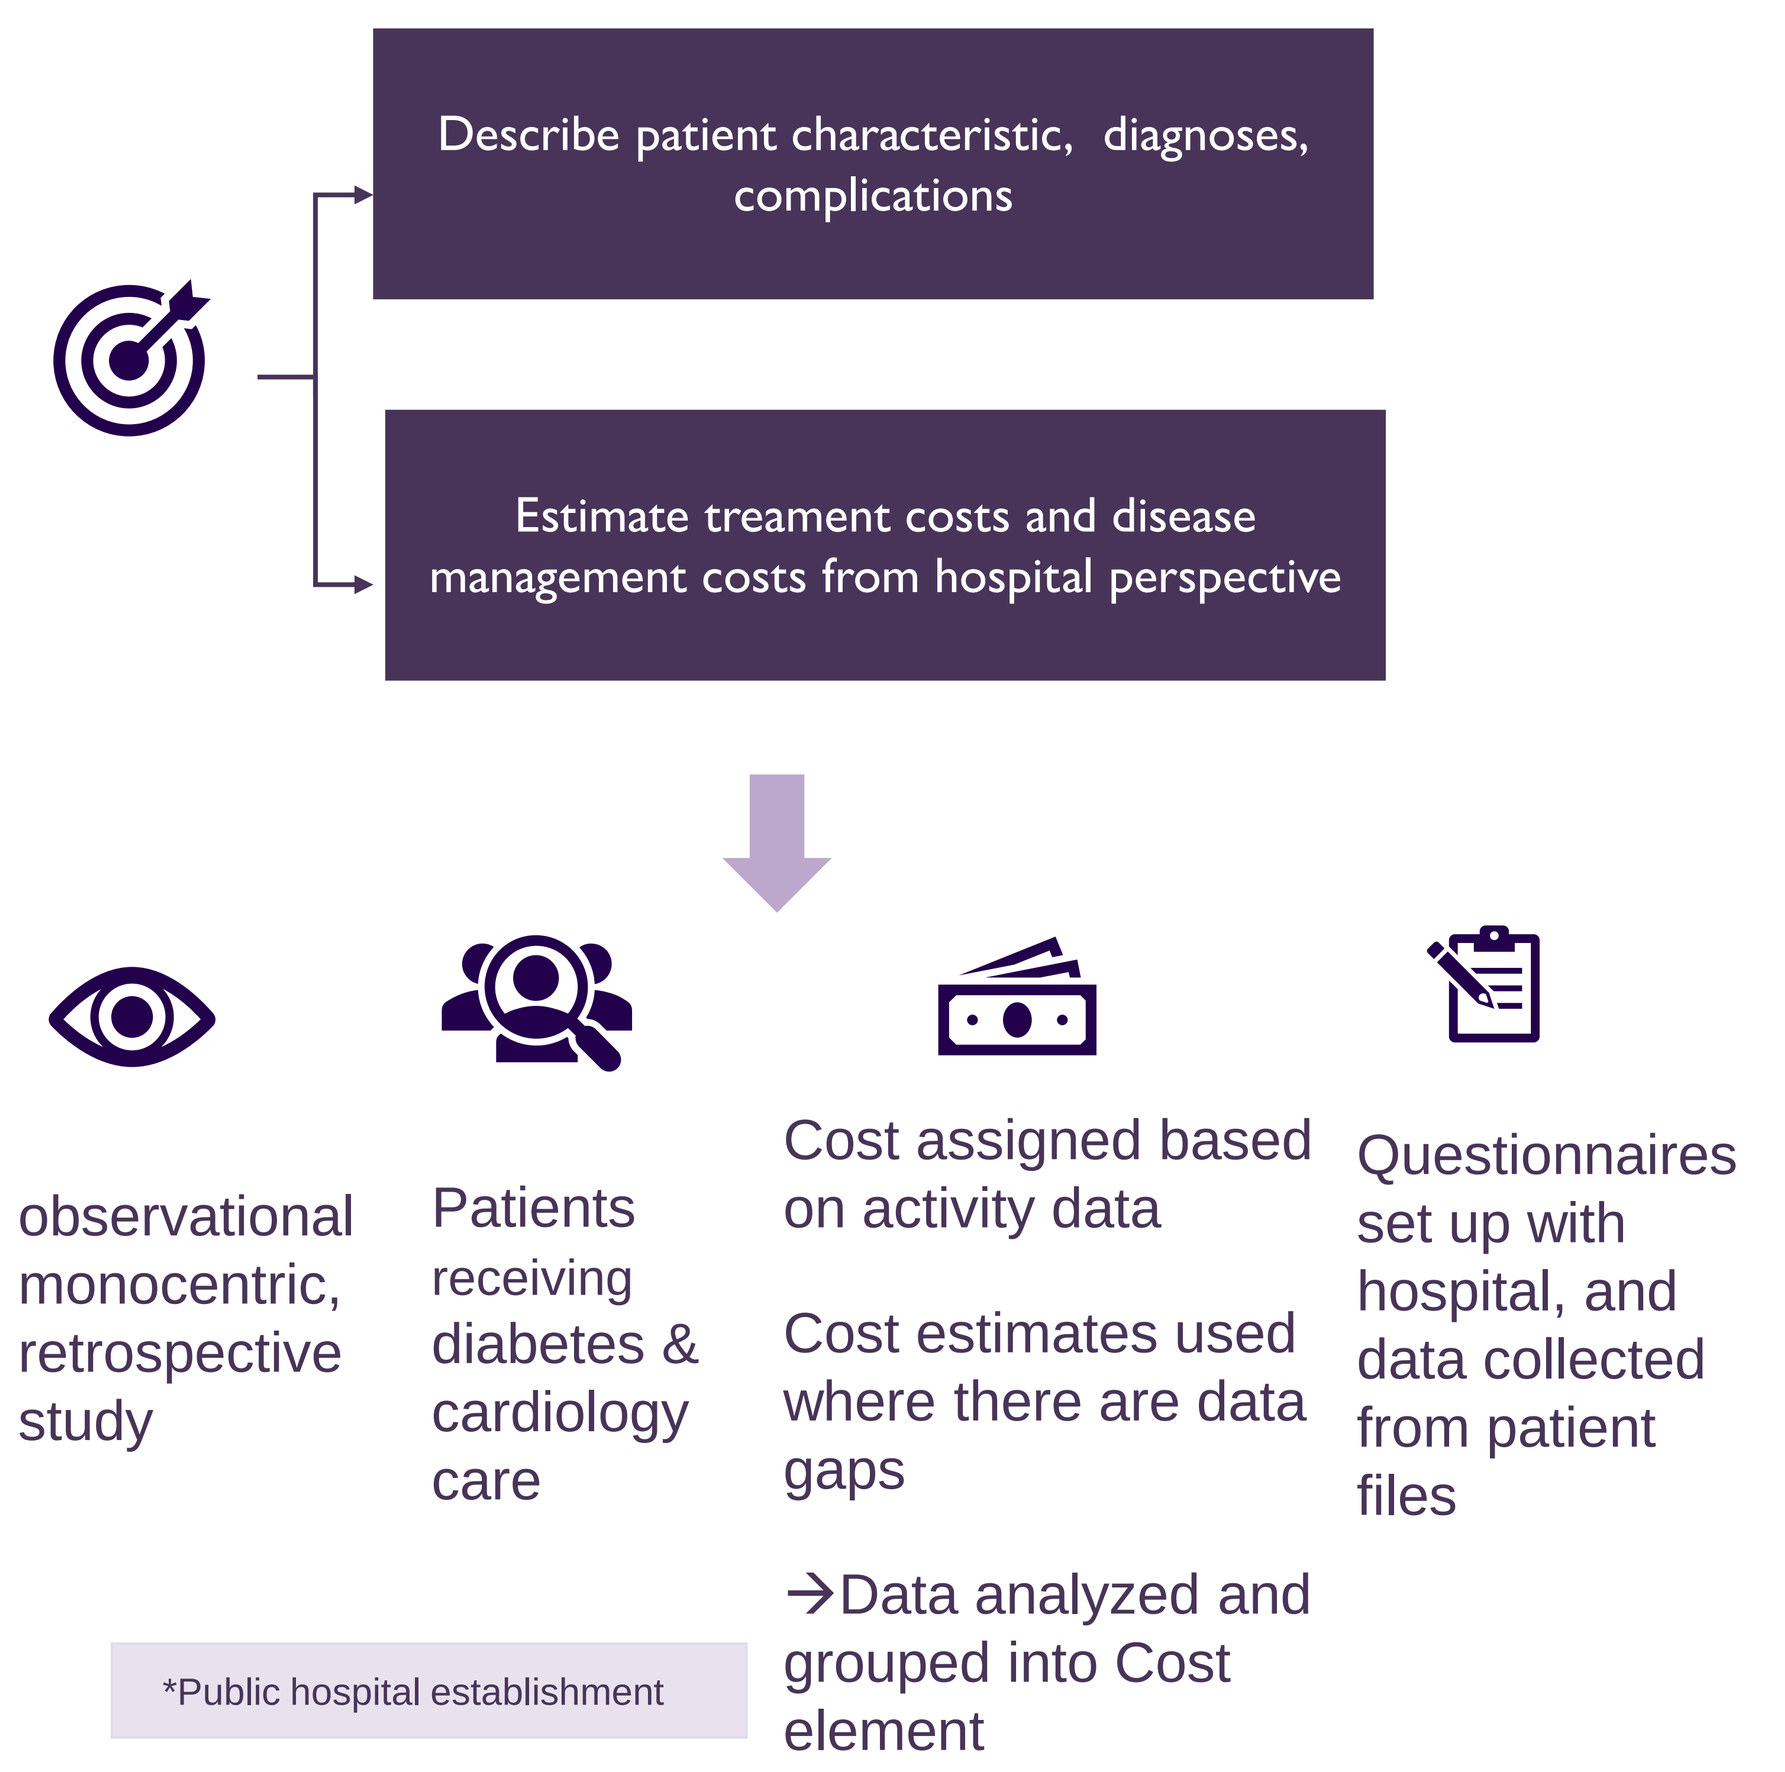
Describe patient characteristic, diagnoses, complications
Estimate treament costs and disease management costs from hospital perspective
observational monocentric, retrospective study
Patients receiving diabetes & cardiology care
Cost assigned based on activity data
Cost estimates used where there are data gaps
→Data analyzed and grouped into Cost element
Questionnaires set up with hospital, and data collected from patient files

*Public hospital establishment

RESULTS

This study has captured the patients characteristic and cost elements for 2300 patients at the internal medicine department, which represents 46% of total patients, repartition per therapeutic area is 54% for cardiology and 37% for diabetology, remaining 6 % were suffering from other pathologies managed in the internal medicine service. Average age of patient groups is 64 years for cardiovascular disease and 60 years for diabetes. 85% of diabetes patients had at least one

POSTER HIGHLIGHTS

Figure 2: Patient characteristic

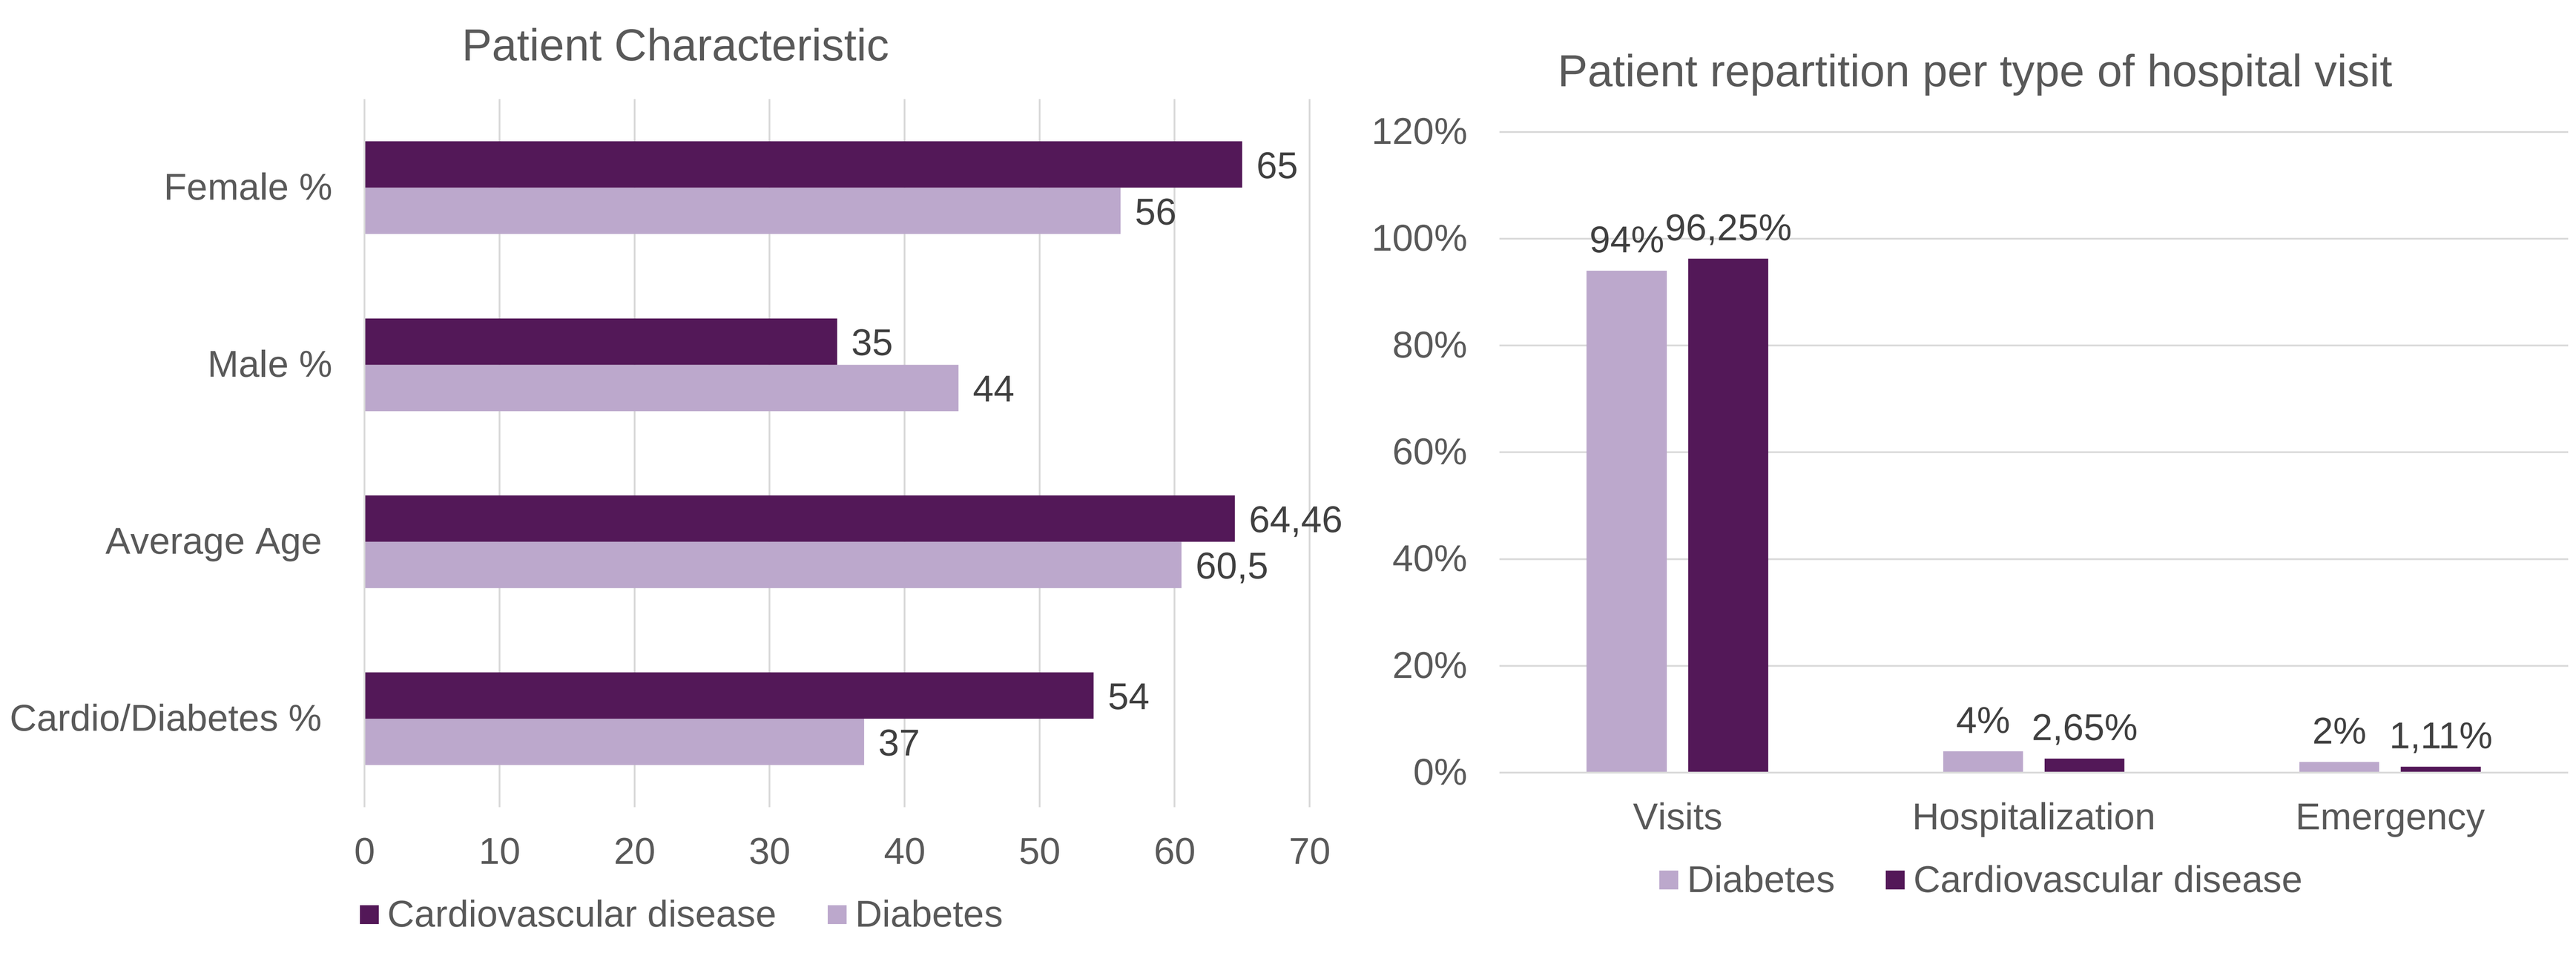
Patient Characteristic
Female %
Male %
Average Age
Cardio/Diabetes %
65
56
35
44
64,46
60,5
54
37
0
10
20
30
40
50
60
70
Cardiovascular disease
Diabetes
Patient repartition per type of hospital visit
120%
100%
80%
60%
40%
20%
0%
94%
96,25%
4%
2,65%
2%
1,11%
Visits
Hospitalization
Emergency
Diabetes
Cardiovascular disease

Figure 3: Complications description

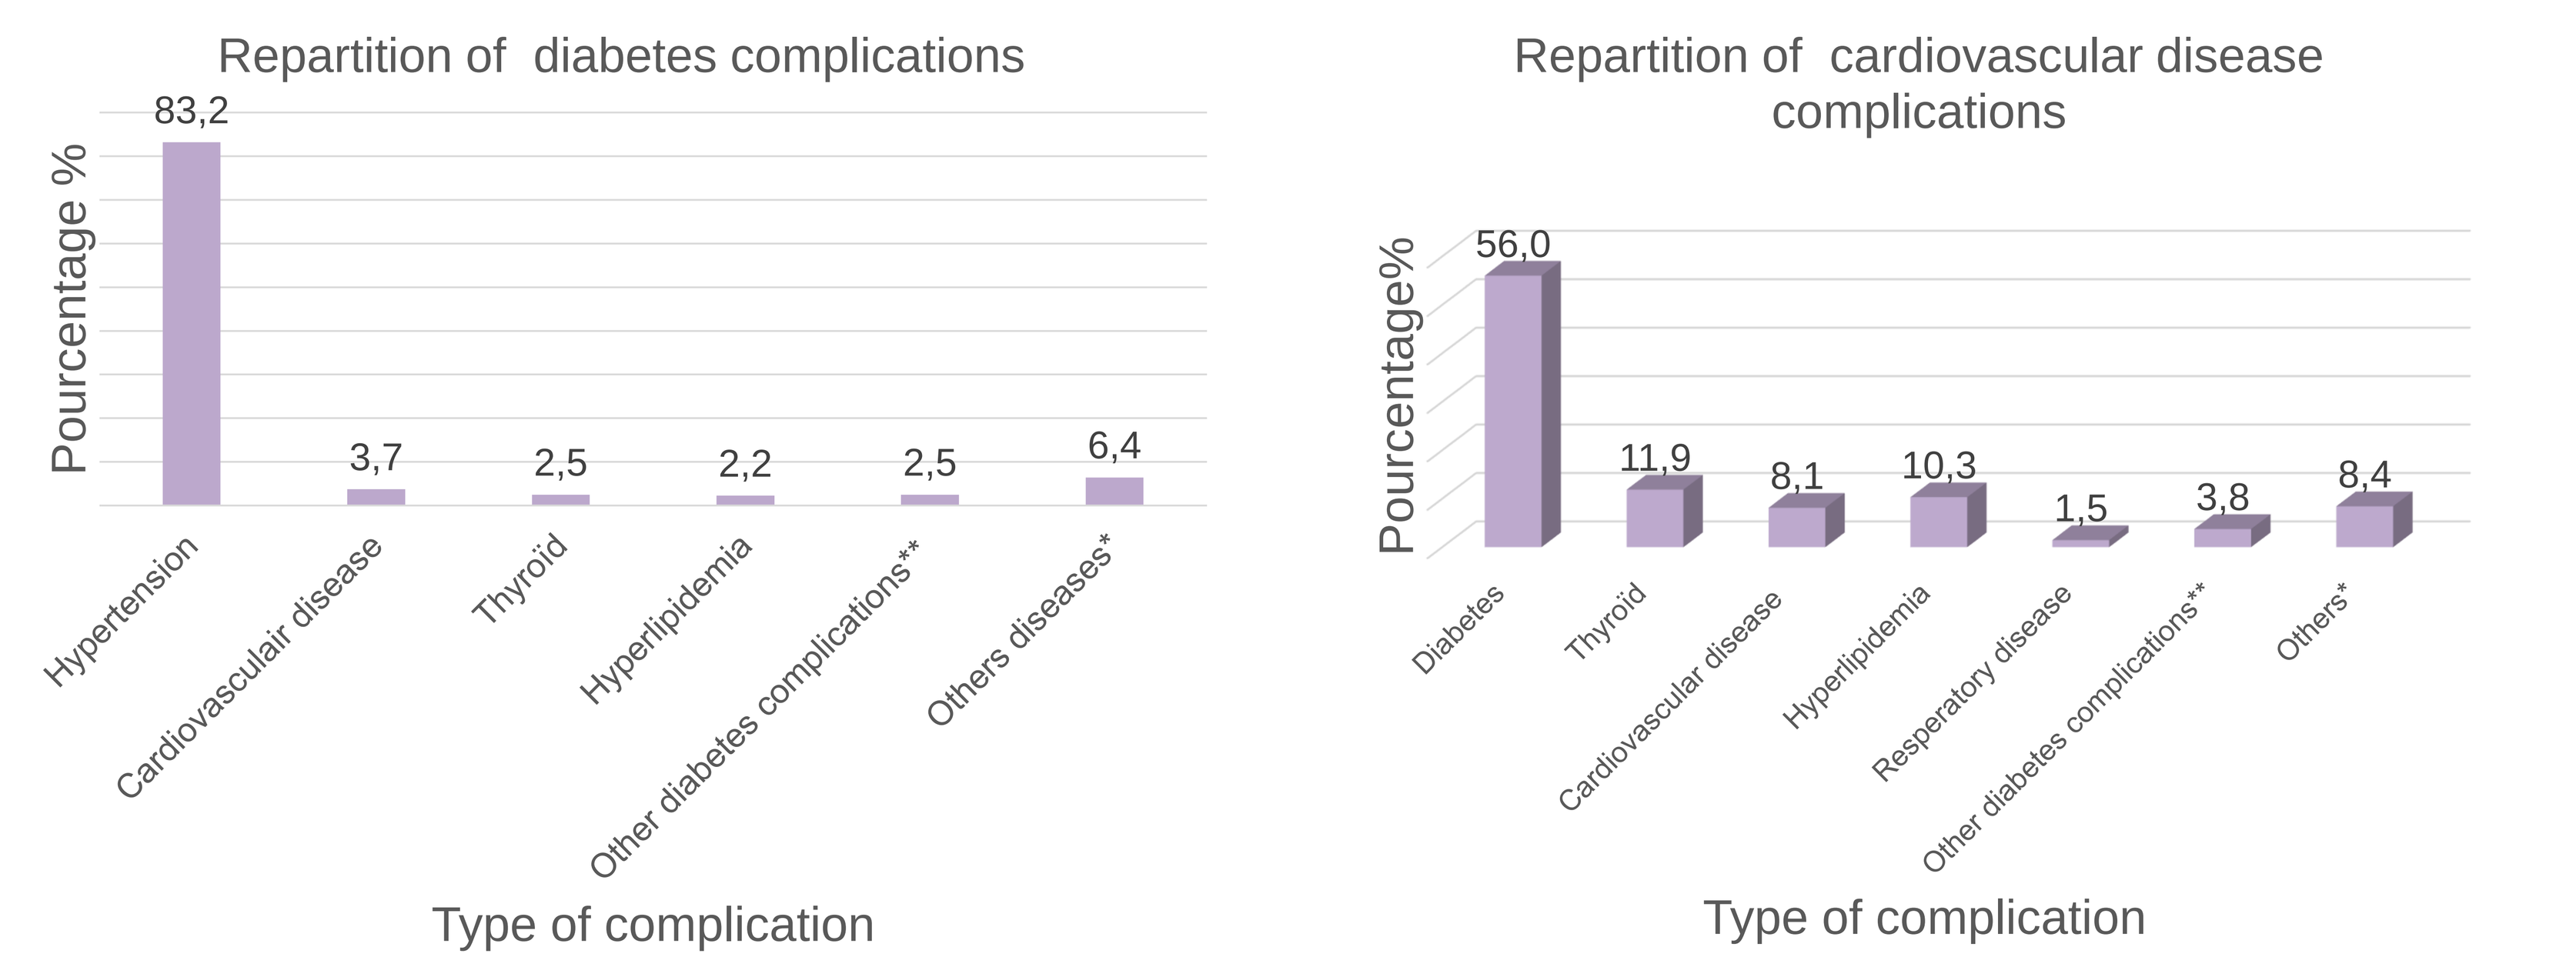
Repartition of diabetes complications
Pourcentage %
83,2
3,7
2,5
2,2
2,5
6,4
Hypertension
Cardiovasculair disease
Thyroïd
Hyperlipidemia
Other diabetes complications**
Others diseases*
Type of complication
Repartition of cardiovascular disease complications
Pourcentage%
56,0
11,9
8,1
10,3
1,5
3,8
8,4
Diabetes
Thyroïd
Cardiovascular disease
Hyperlipidemia
Resperatory disease
Other diabetes complications**
Others*
Type of complication

**Other diabetes complications : renal impairment, acidecetosis, diabetic foot, retinopathy
*Other diseases :Alzheimer, cancer, epilepsy, fatigue, skin disease, bones and resperatory disease

**Other diabetes complications : renal impairment, retinopathy
*Others diseases :Anemia, Alzheimer, cancer, epilepsy, fatigue, skin disease , arthrosis, crohn disease, Liver disease, parkinson..

Figure 4: Costs results

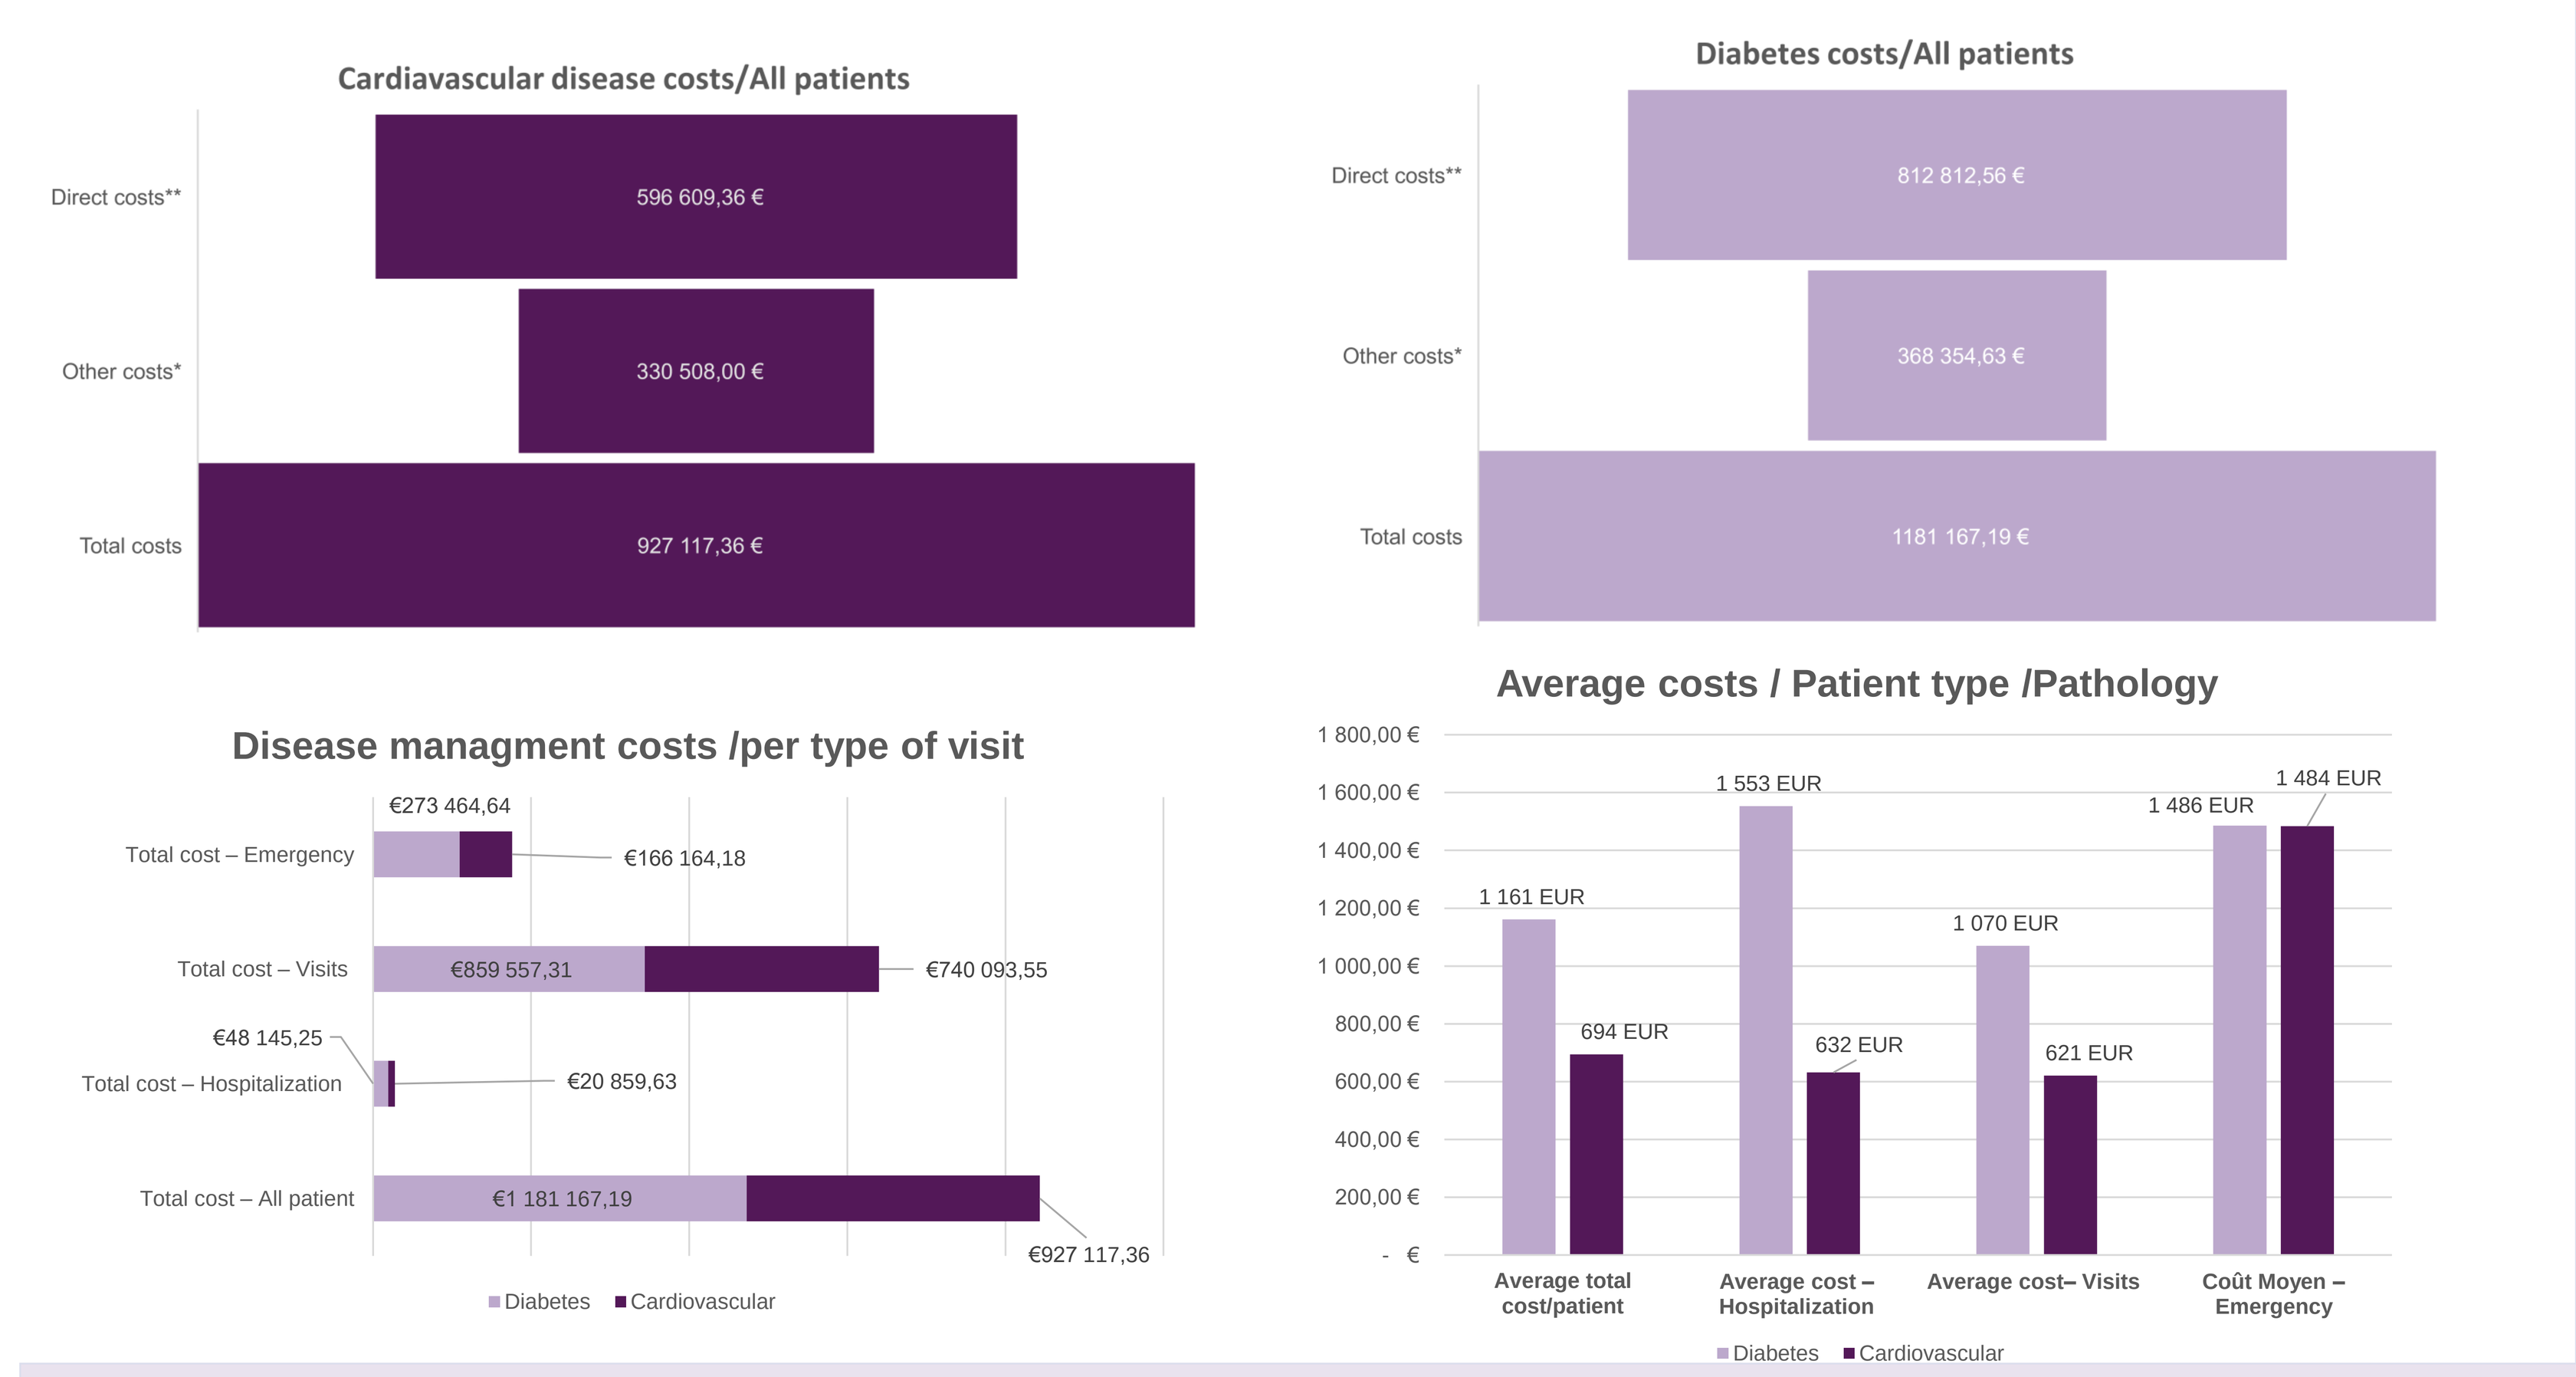
Cardiavascular disease costs/All patients
Direct costs**
596 609,36 €
Other costs*
330 508,00 €
Total costs
927 117,36 €
Diabetes costs/All patients
Direct costs**
812 812,56 €
Other costs*
368 354,63 €
Total costs
1181 167,19 €
Disease managment costs /per type of visit
Total cost – Emergency
€273 464,64
€166 164,18
Total cost – Visits
€859 557,31
€740 093,55
Total cost – Hospitalization
€48 145,25
€20 859,63
Total cost – All patient
€1 181 167,19
€927 117,36
Diabetes
Cardiovascular
Average costs / Patient type /Pathology
1 800,00 €
1 600,00 €
1 400,00 €
1 200,00 €
1 000,00 €
800,00 €
600,00 €
400,00 €
200,00 €
- €
1 161 EUR
694 EUR
1 553 EUR
632 EUR
1 070 EUR
621 EUR
1 486 EUR
1 484 EUR
Average total cost/patient
Average cost – Hospitalization
Average cost– Visits
Coût Moyen – Emergency
Diabetes
Cardiovascular

*Other costs : All costs not involved in t direct management of the disease : ,: administrative, financial, IT costs
**Direct costs : physician costs, nurse costs, drug costs & Tests and diagnosis costs

Figure 5 : Costs per patient

	Average cost/patient/Diabetes
Tests and diagnosis costs	147,49 EUR
Drug costs	560,50 EUR
Nurse costs	25,22 EUR
Physician costs	325,19 EUR
Total costs	1 058,40 EUR

	Average cost/ patient/Cardiovascular
Tests and diagnosis costs	152,82 EUR
Drug cots	146,02 EUR
Nurse costs	23,05 EUR
Physician costs	263,10 EUR
Total costs	584,99 EUR

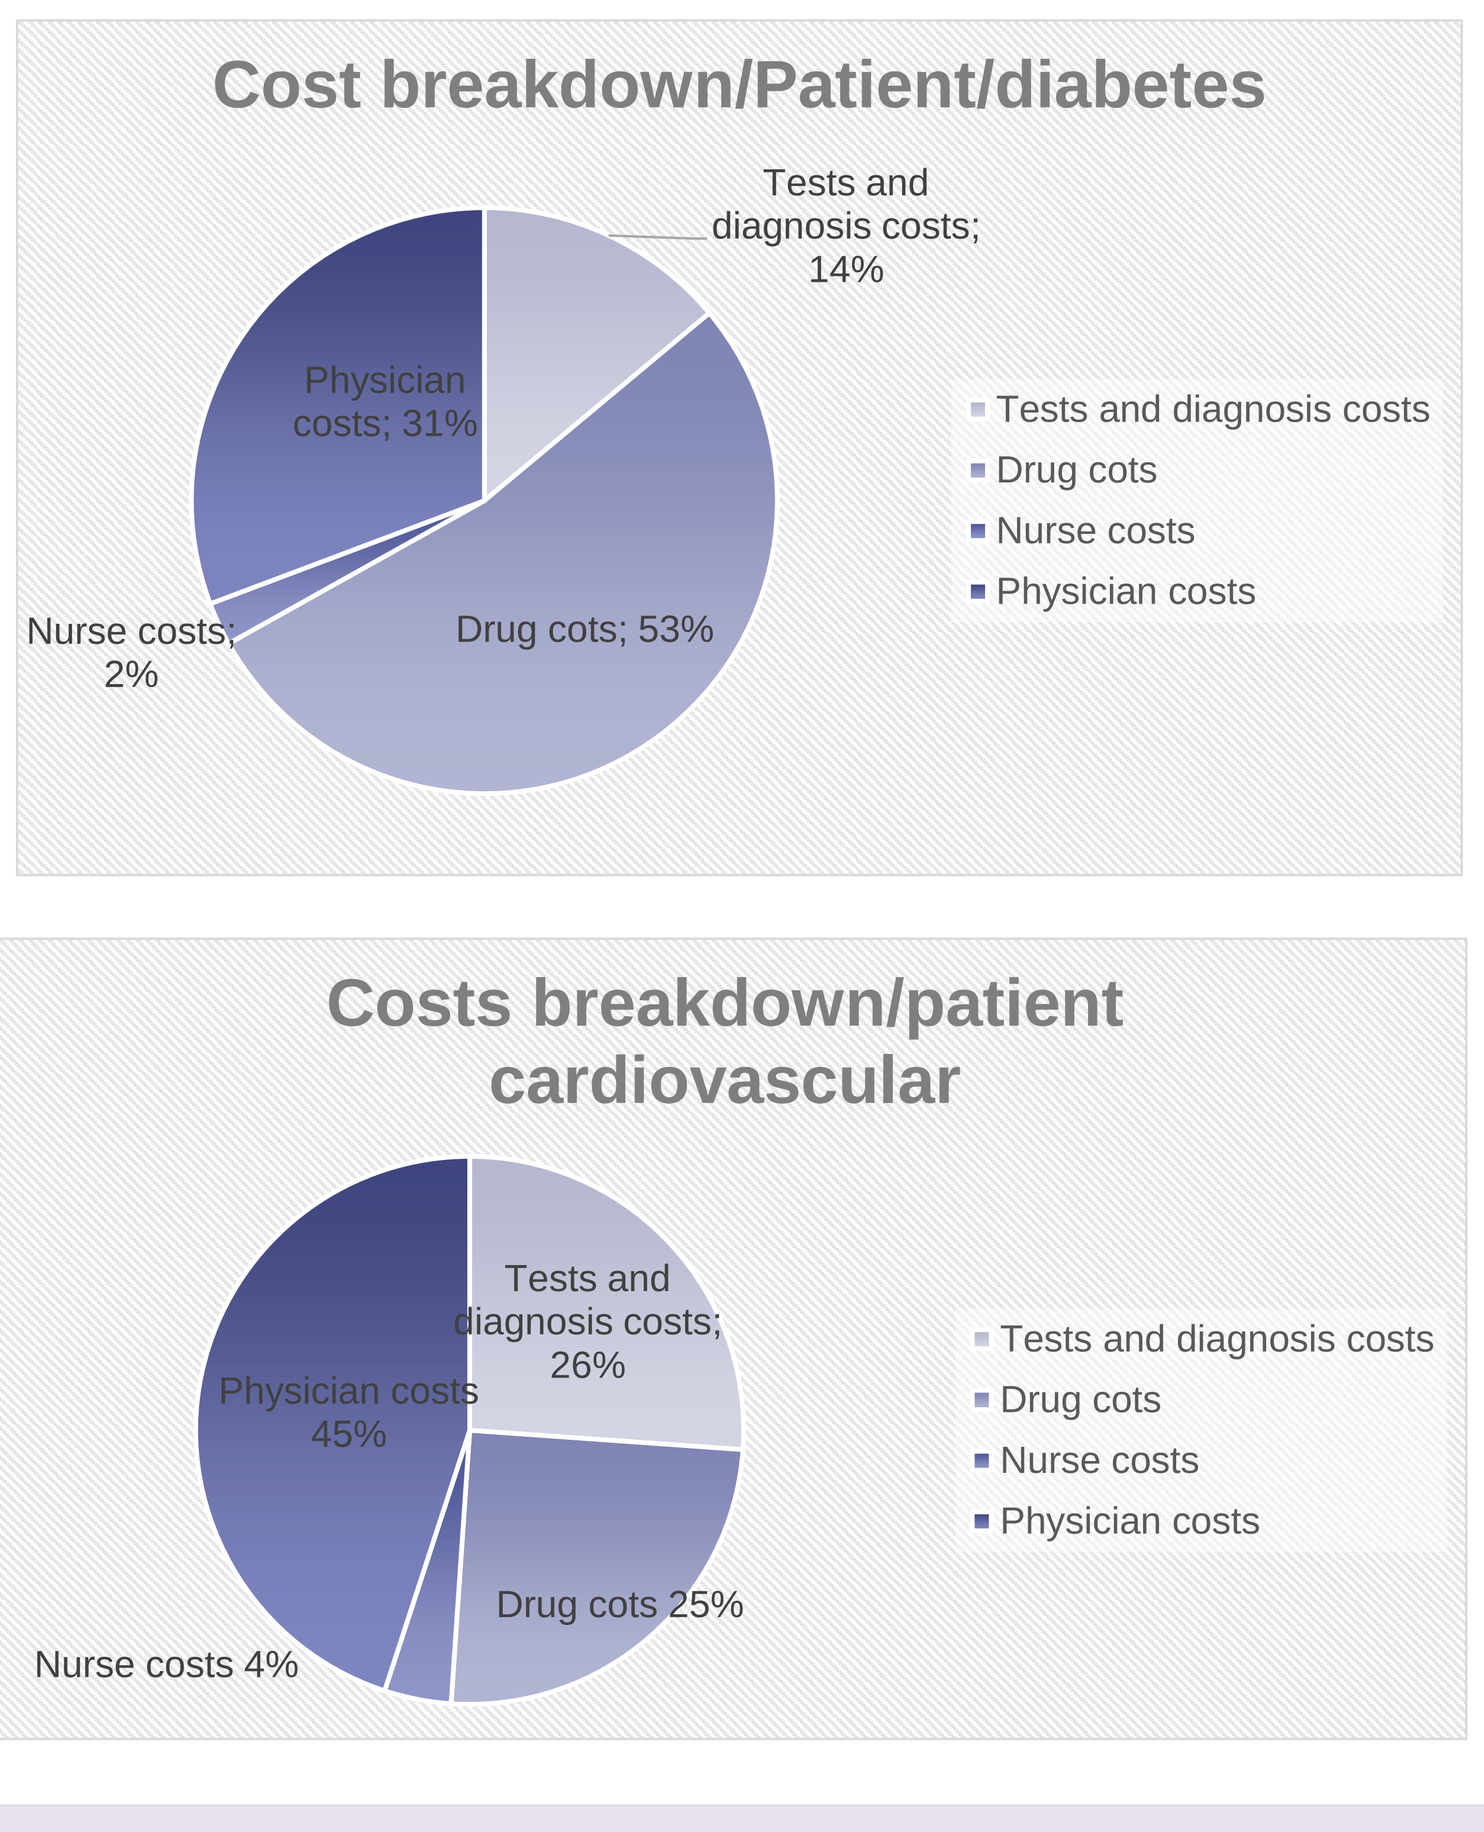
Cost breakdown/Patient/diabetes
Tests and diagnosis costs; 14%
Physician costs; 31%
Nurse costs; 2%
Drug cots; 53%
Tests and diagnosis costs
Drug cots
Nurse costs
Physician costs
Costs breakdown/patient cardiovascular
Tests and diagnosis costs; 26%
Physician costs 45%
Drug cots 25%
Nurse costs 4%
Tests and diagnosis costs
Drug cots
Nurse costs
Physician costs

RESULTS (continued)

comorbidity or complication related to diabetes which was hypertension and 56% for cardiovascular disease groupe which was diabetes. Total direct cost was 1.4millions euros (almost 600K € for cardiovascular disease and 812 K € for diabetes).

The average cost of care is significantly higher for a diabetic patient versus cardiology patient, a gap of 67%.

This gap widens further when it comes to the average cost of a hospitalized patient, since between a diabetes and cardiovascular disease patient, the difference is estimated at ≈146%.

It is estimated that there is a difference of 72% between the average cost of a diabetes patient vs cardiovascular disease patient when seen in visits.

For a patient admitted to the emergency room, there is no difference in average cost between a diabetes and cardiovascular disease patient.

Average medical resources consumed per diabetes patient is 1 058,40 € (560,50 € for drug costs, 147,49€, for Tests and diagnosis costs, 325,19 € physician costs and 25,22 € for nurses costs) and for cardiovascular disease patients 584,99 € (146,02 € for drug costs, 152,82€, for Tests and diagnosis costs, 263,10 € physician costs and 23,05 € for nurses costs)

DISCUSSION

- This study provides the first and one of the most detailed descriptions of inpatient resource utilization from hospital perspective, patient cohort was large which allowed to obtain a statistically significant results.
- Diabetes care appears to be costly in comparison to cardiovascular diseases costs, this is mainly explained by higher treatment cost of this pathology, this study suggests further that this cost item is the highest overall hospital expenditure,
- Other resources spent during the study period of 2 years for both patient sub groups remains equivalent
- As there is no local study that assessed this type of cost, it is not possible to perform comparison

The limits of this study :

- Only hospital perspective disease management and costs was considered, other costs were note included.
- Detailed cost of complications and other disease costs were not assessed.

CONCLUSIONS

Diabetes and cardiovascular disease healthcare utilization are significantly high, the direct costs generated by these diseases in Algeria represent a real economic and societal burden for patients and health authorities.

These results will help raise awareness and stimulate further scientific research and investment for these disease management, resource optimization, and improved quality of life for patients.

FUNDING : This study was sponsored by Sanofi.

REFERENCES :
1-Brondex A. et all , 2012/ 2-WHO stepwise, 2017/ 3-Cardiometabolic risk in Algeria: past and present, A. Mammer, A. Tebaibia, October 2019