Cost Effectiveness Analysis of Levonorgestrel Vs Etonorgestrel implants in Contraception from the Mexican Context

EE402

Monroy-Cruz, Bárbara[1]; Ramirez-Gámez, Jocelyn[1]; Quintana, Sandra[1]

[1.] Bayer de México, México

INTRODUCTION

Teen pregnancy in Mexico is a health and socioeconomic problem, which widens gender and social gaps. [1] It has a negative impact on the education, income and health of the teenagers, placing them in a vulnerable postion.[2]

According to recent data, Mexico is one of the countries with the highest rate of teen pregnancy in the world, with 66 pregnancies per 1,000 teens every year (360,135 teen pregnancies expected for 2023 in Mexico).[3]

Subdermal implants can help in reducing teen pregnancy rates due to their duration coverage, price and effectiveness, which is not user dependent.

OBJECTIVES

Currently, two subdermal implants are available in the Mexican public healthcare institutions: levonorgestrel (LNG) and etonorgestrel (ENG), which differ in duration of coverage, discontinuation rates and price.

Therefore, to know which one represent the most efficient option, a cost effectiveness analysis was conducted from the Mexican public healthcare system perspective.

METHODS

A cost effectiveness analysis was conducted to estimate the Incremental Cost Effectiveness Ratio per year of coverage with LNG or ENG from the public payer´s perspective.[4]

A decision tree model was developed in MicrosoftExcel®2013 to simulate 1,000 teenagers. Only direct medical costs were considered. Prices were gathered from Mexican official sources as of April 2023.[5]

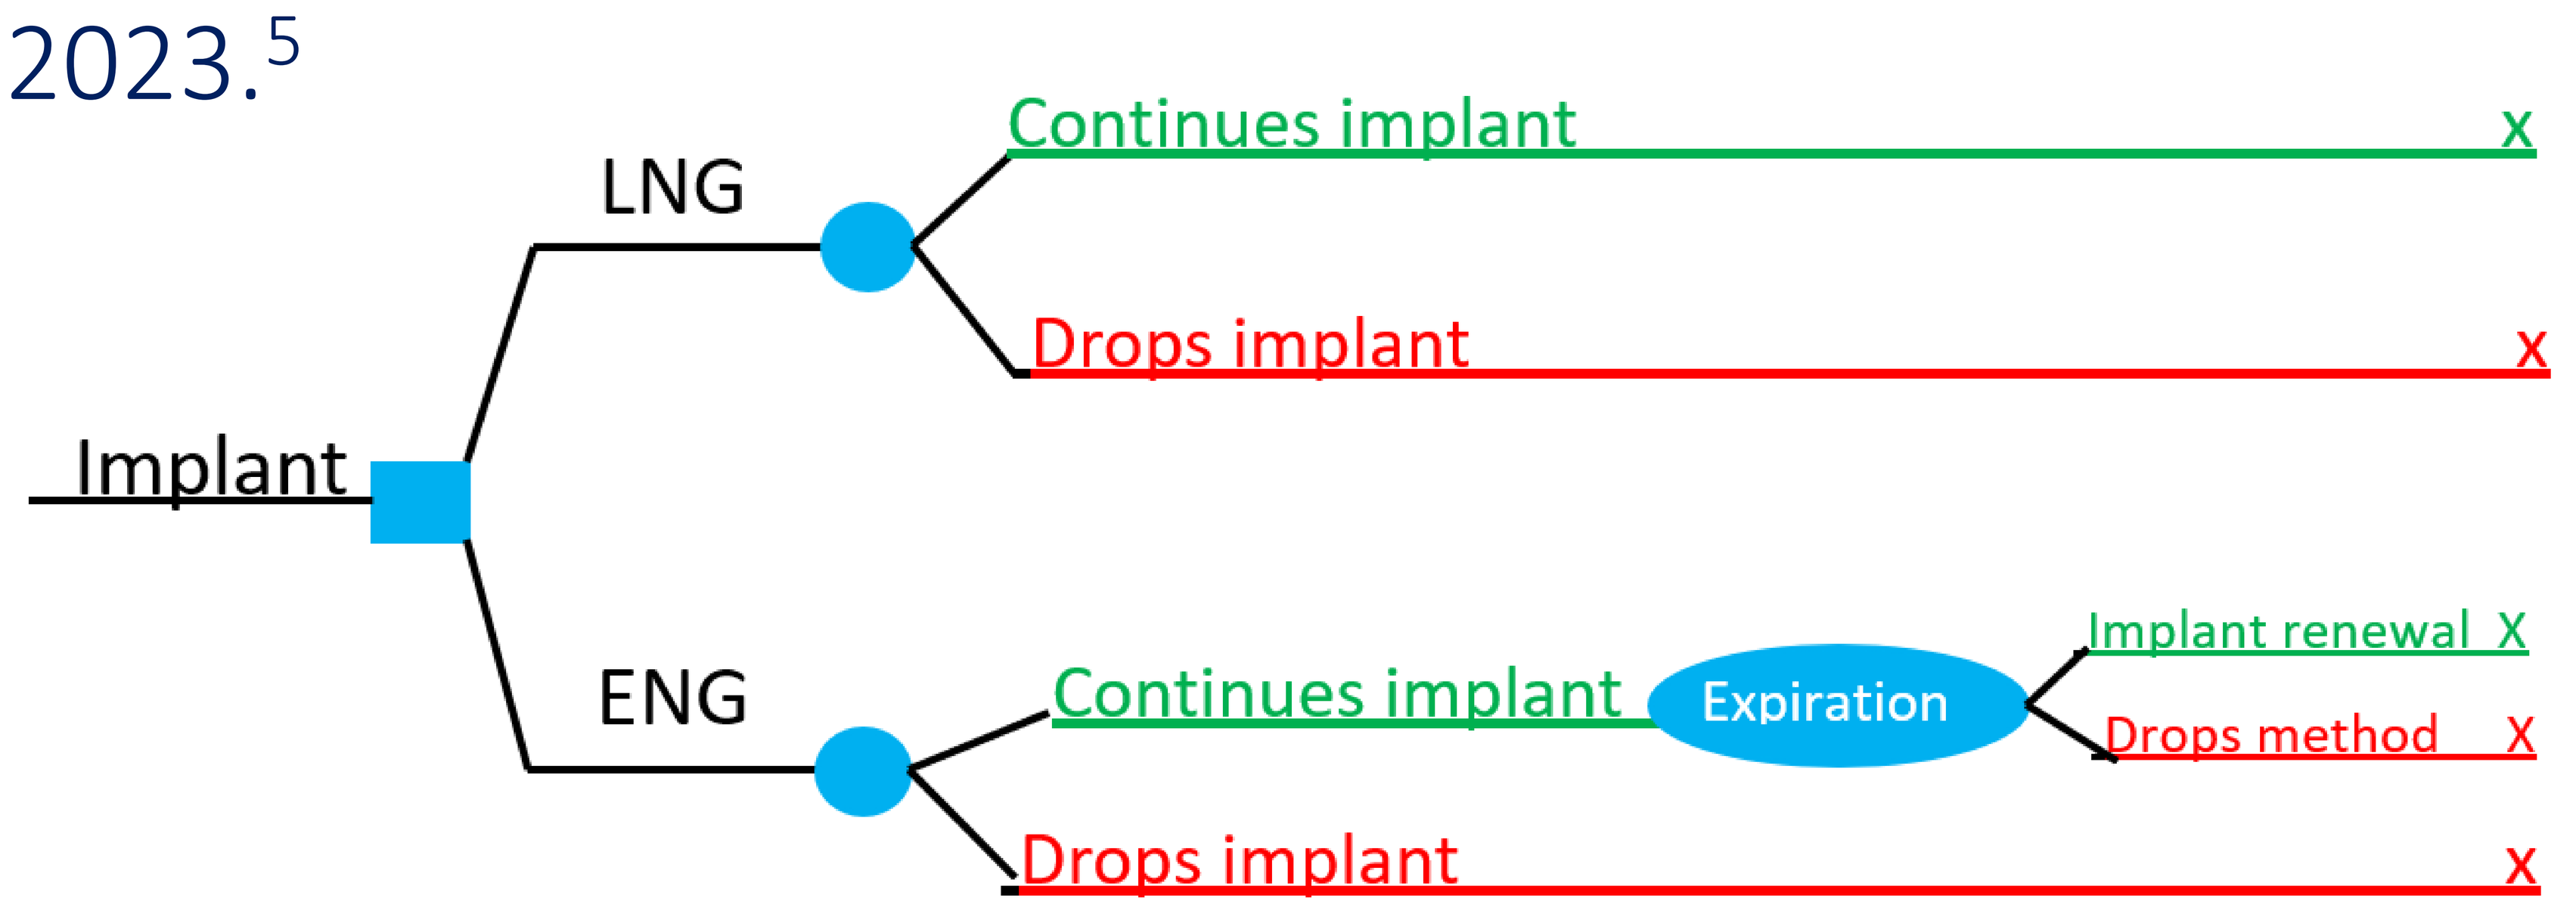
Implant
LNG
Continues implant x
Drops implant x
ENG
Continues implant
Expiration
Implant renewal X
Drops method X
Drops implant x

The analysis was conducted under a 5-year time horizon to capture the difference in implant durations. A 5% annual discount rate was applied according to Mexican guidelines.[6]

Variable	LNG	ENG
Price (MXN)	$447.6	$1,102.9
Coverage duration (years)	5	3
Continuation rates, years 1, 2 and 3 (%)	88%, 77%, 52%	89%, 75%, 12%

RESULTS

From the perspective of the Mexican public payer, the results suggests that LNG is a dominant alternative vs ENG, as it offers more years of contraceptive coverage (598 extra years) at a lower cost (- $870,301 MXN).

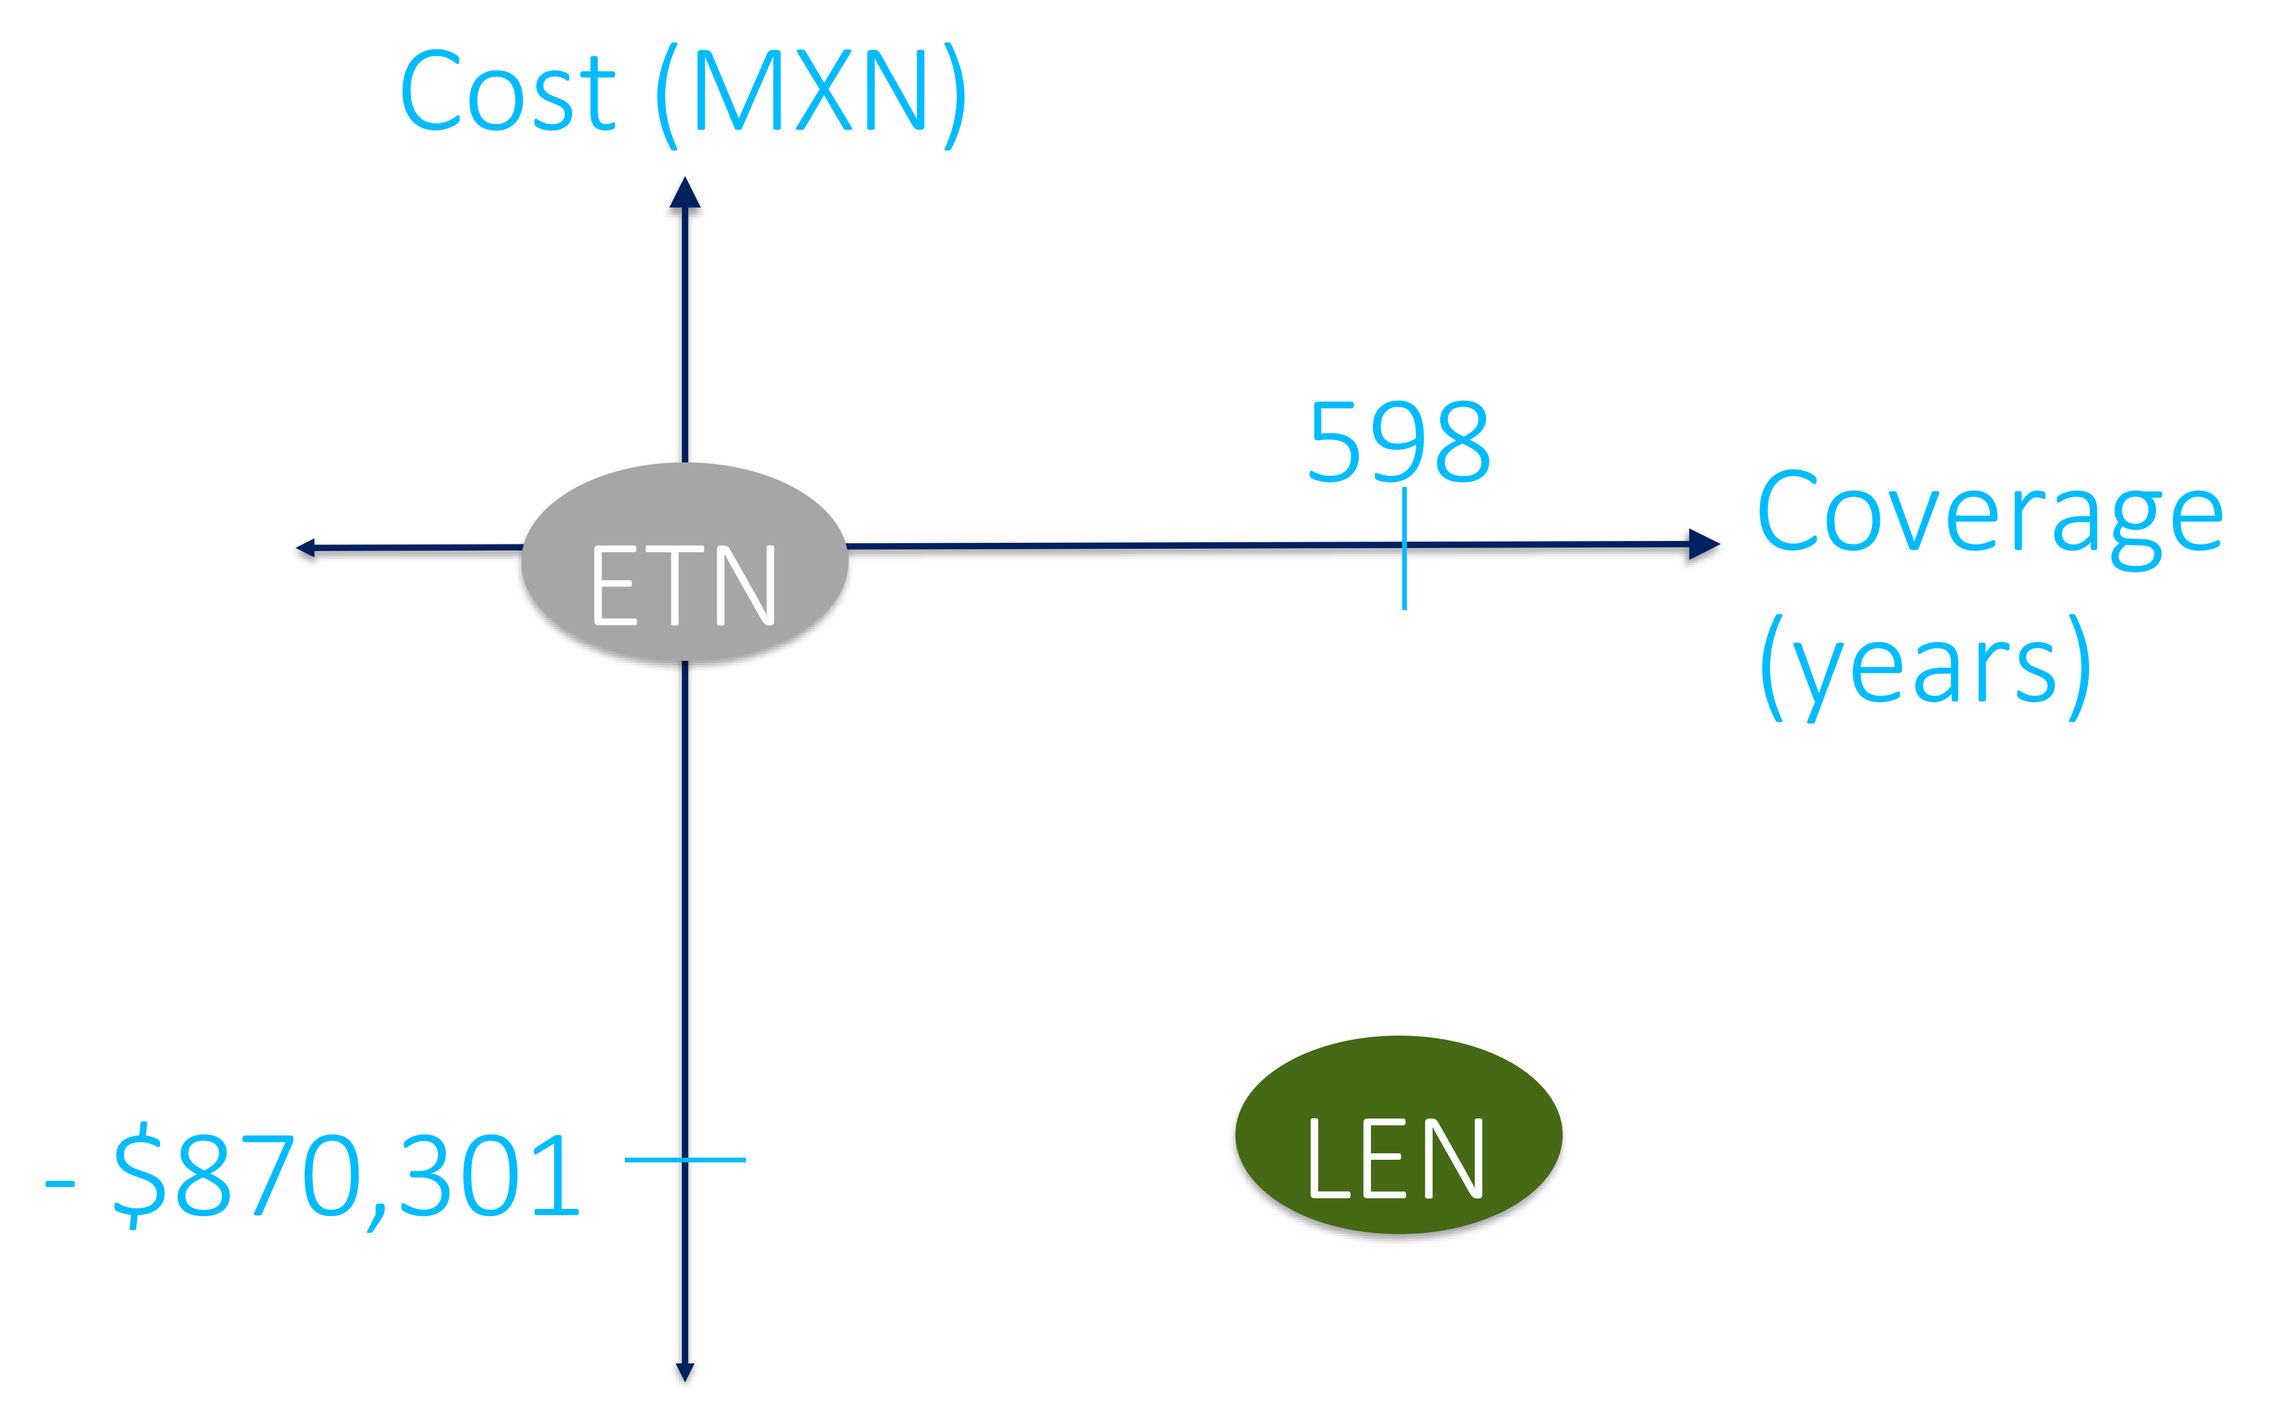
Cost (MXN)
598
Coverage (years)
ETN
- $870,301
LEN

CONCLUSIONS

LNG offers greater adherence vs ENG due to the higher duration (5 vs 3 years) and higher continuation rates, protecting teenagers against unplanned pregnancies.

Therefore, investing on LNG can bring great benefits to the society, the healthcare system, but ultimately to the teenagers, the most vulnerable group.

REFERENCES

LNG: Levonorgestrel; **ENG:** Etonogestrel;

1. Estrategia Nacional para la Prevención del Embarazo en Adolescentes, 2022, Secretaria de Gobernación, Mexico; **2**. UNFPA, 2018, Consecuencias socioeconómicas del embarazo en adolescentes en México; **3.** CONAPO, Proyección poblacional 2016-2050; **4.** Bahamondes L, et al., 2015, A 3-year multicentre randomized controlled trial of etonogestrel- and levonorgestrel-releasing contraceptive implants, with non-randomized matched copper-intrauterine device controls. Hum Reprod., ;30(11):2527-38; **5.** Programa Anual de Adquisiciones, Arrendamientos y Servicios (website) ISSSTE; **6.** Consejo de Salubridad General, 2023, Guía para la conducción de estudios de evaluación económica para la actualización del Compendio Nacional de Insumos para la Salud, Mexico.