Comprehensive and standardized value assessment of innovative cancer treatments

HPR181

A multi-criteria decision analysis tool for Switzerland

Kuhlmey, F.[1], Bill, M.[1], Fischer, B.[1], Oniangue-Ndza, C.[2], Telser, H.[1]

[1] Polynomics SA, Olten, Switzerland; [2] Bristol Myers Squibb, Steinhausen, Switzerland

Objectives

The existing procedures for reimbursing new drugs and drug combinations encounter challenges in making effective reimbursement decisions, particularly in systems with public health insurance such as in Switzerland.

The aim of this project is to develop a tool that better reflects the diverse value components of innovative medications and enables a standardized assessment of new treatment methods in a general context. This evaluation process should be quicker and more transparent than current procedures. It is applicable to the Swiss reimbursement context.

The proposed model was tested using the treatment of advanced renal cell carcinoma as a case study.

Methods

A MCDA framework

Multi-criteria decision analysis (MCDA) is a method to guide complex decisions. It evaluates multiple factors simultaneously, helping decision-makers prioritize options transparently. We utilized the framework proposed in the ISPOR task force report for MCDA (Marsh et al., 2016) and applied it to evaluate drugs used in advanced cancer treatments.

Figure 1. MCDA steps according to ISPOR-guidelines

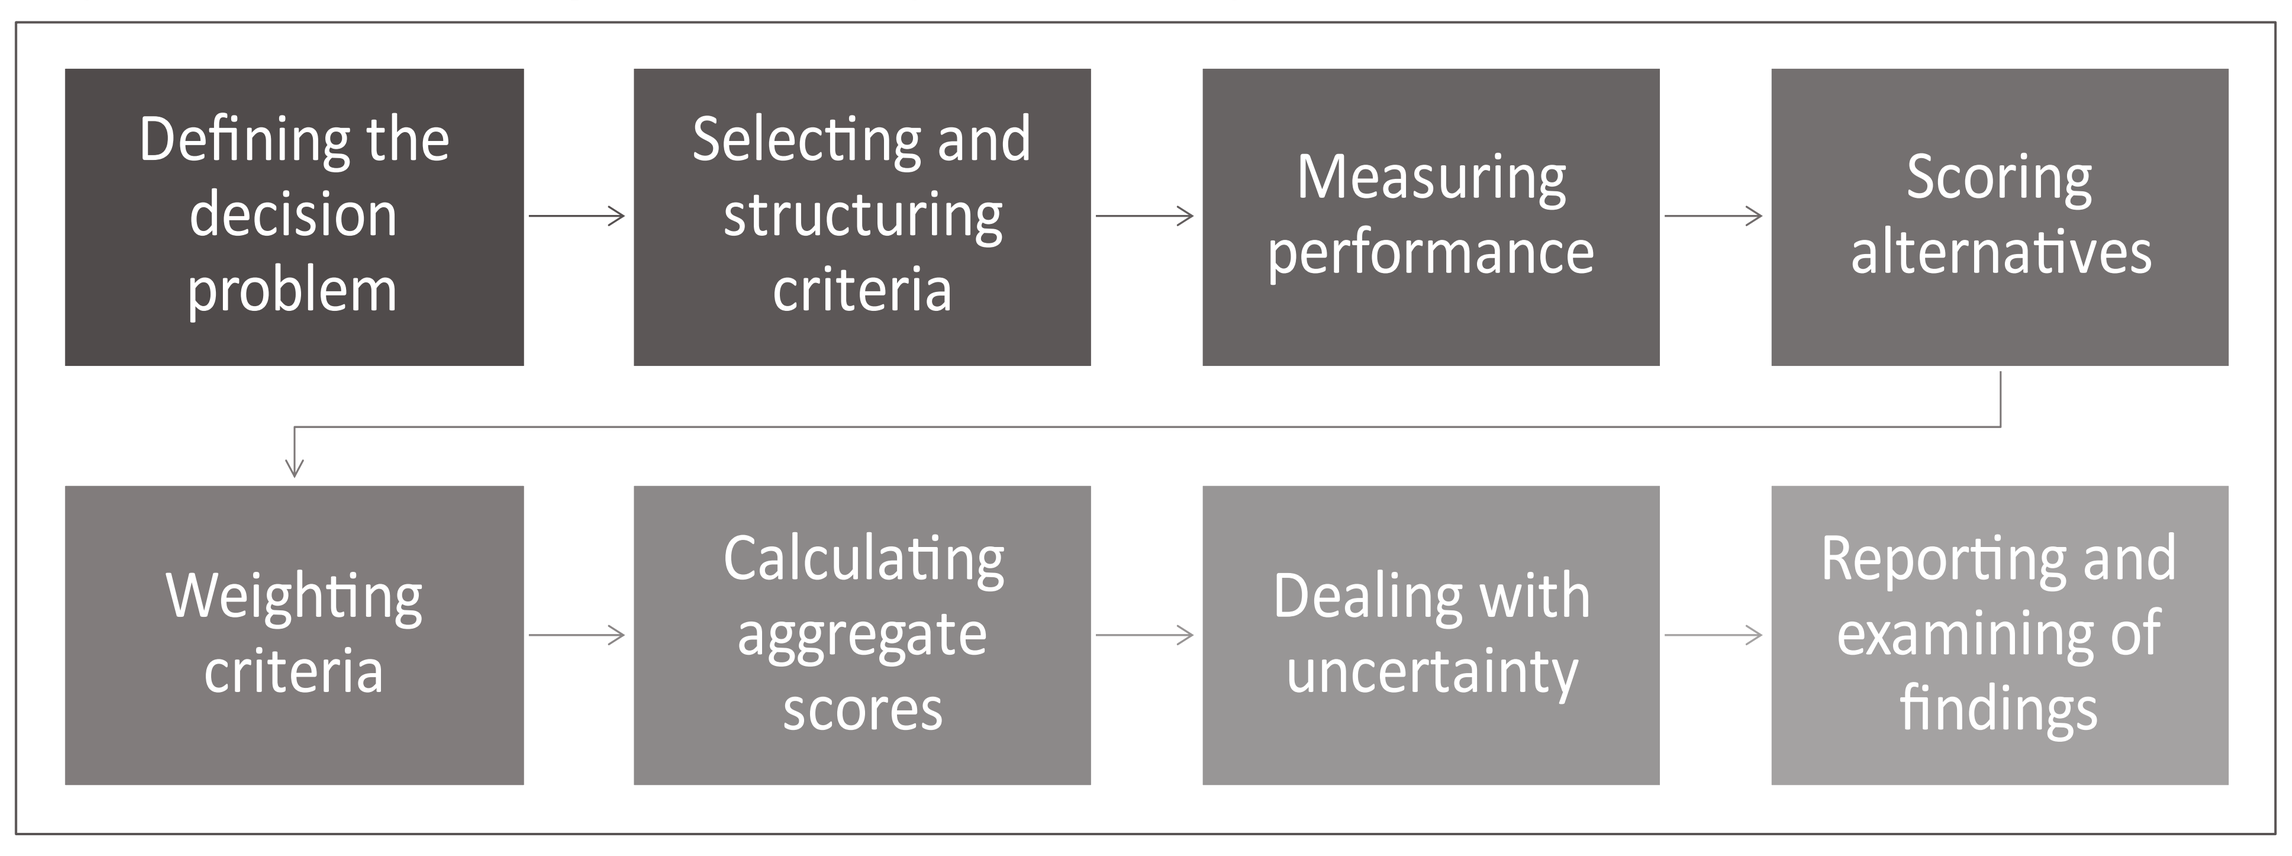
Defining the decision problem
Selecting and structuring criteria
Measuring performance
Scoring alternatives
Weighting criteria
Calculating aggregate scores
Dealing with uncertainty
Reporting and examining of findings

The model yields an aggregate value score for each therapy that is calculated as the sum of the weighted attribute values and is normalized between 0 and 100. The value gain from an innovative treatment option follows as the difference between the value score from the innovation and the standard-of-care (SOC).

B Stakeholder selection for attributes definition

Input from stakeholders, including Swiss oncologists, patient representatives, and health economists, was gathered and employed to identify the model's attributes, i.e., the value components.

The specific form of the value functions (linear or non-linear) as well as the minima and maxima of the attributes were further refined with the assistance of a focus group. To maintain the model's versatility, in some cases, multiple variants were permitted for measuring an attribute.

C DCE for weighting and scoring of attributes

To determine attribute weighting and assessment, we elicited preferences of the Swiss population through a Discrete Choice Experiment (DCE), a decompositional method to elicit preferences.

The main survey took place in February 2023. It covered the Swiss population aged 18 and above from the German- or French-speaking regions of Switzerland. The sample was stratified by age, gender, and language region. In total, 1,034 interviews were completed. A total of 16 decision scenarios were created, divided into two blocks. Each respondent had to make eight decisions.

The statistical analysis is based on a decision-theoretical model (McFadden, 2001). A panel-data mixed logit model was estimated with a decision dummy as the dependent variable and the attributes as independent categorical variables. There were no other covariates related to respondents' characteristics, as the sample is representative of the Swiss population, from which we aim to elicit preferences.

This estimation allows the calculation of attribute weights and also defines the non-linear value function.

D Case study for model validation and measuring of performance

We consider the treatment of patients with intermediate or poor risk renal cell carcinoma. The case study is based on the phase III CheckMate 214 trial comparing snivolumab plus ipilimumab versus sunitinib. The following table describes the sources we used to extract the required information to feed into our model.

Table 1. References for the case study

Reference	Quality assessment
Motzer RJ et al. Nivolumab plus ipilimumab versus sunitinib in advanced renal-cell carcinoma. *New England Journal of Medicine*. 2018.	▪ Peer reviewed ▪ Phase III study
Albiges L et al. Nivolumab plus ipilimumab versus sunitinib for first-line treatment of advanced renal cell carcinoma: extended 4-year follow-up of the phase III CheckMate 214 trial. *ESMO Open*. 2020.	▪ Peer reviewed ▪ Phase III study
Cella D et al. Patient-reported outcomes of patients with advanced renal cell carcinoma treated with nivolumab plus ipilimumab versus sunitinib (CheckMate 214): A randomized, phase 3 trial. *The Lancet Oncology*. 2019.	▪ Peer reviewed ▪ Phase III study
Oniangue-Ndza C et al. Cost effectiveness analysis of nivolumab in combination with ipilimumab for the first-line treatment of advanced/metastatic renal cell carcinoma in Switzerland. ISPOR Europe Conference 2019.	▪ Conference-review
Expert opinion	▪ Not peer-reviewed

Results

A MCDA model

In collaboration with experts in oncology, health economics, and the pharmaceutical industry, the following value tree depicted for advanced cancer treatments was developed.

Table 2. Overview of attributes

	Criteria	Attribute	Measurement
Patient benefit	Treatment success	Survival time (ST)	▪ Median observed survival (OS) or Median progression-free survival (PFS)
		Health-related quality of life (QoL)	▪ SF-36, SF-12, EQ-5D, QLQ-C30 (prioritized) ▪ Consider alternatives suggested by experts ▪ Use utilities that are normalized between 0 (death) and 100 (perfect health)
	Safety profile	Adverse events	▪ Percentage of patients with at least one adverse event of Grade 3, 4, or 5
		Patient convenience	▪ Frequency of administration ▪ Administration location (ambulatory, at home)
Societal benefit	Disease burden	Disability-adjusted life years (DALY)	▪ DALY or (Disability Weights * Prevalence of indication) ▪ Indicator: Proportion of total DALY for cancer and Switzerland
	Costs	Indirect costs	▪ Number of lost work months (patient and caregivers) for the entire treatment period ▪ Calculation or expert estimation
		Additional treatment costs	▪ Costs of typical accompanying services for the considered drug therapy (per patient, entire treatment period) ▪ Calculation or expert estimation

B Discrete choice experiment (DCE)

We define the value on the utility scale for all possible variations of the metrics in our value tree for advanced cancer therapies.

The following table contains the minimum and maximum values for each attribute as well as the information for which attributes a non-linear value function is assumed and therefore three values are used.

Table 3. Specifications of all attributes in the DCE

Attribute	Unit	Level 1	Level 2	Level 3	Value function
1 Survival time (here: Median OS)	Months	3	24	60	Non-linear
2 Long-term quality of life (health-related QoL)	PROM utility score	10	45	80	Non-linear
3 Severe adverse events	Percent of patients	5	80		Linear
4 Treatment frequency	Every … days	1	30		Linear
5 Treatment location: ambulatory (0), at home (1)	Dummy variable	0	1		Linear
6 Disease burden in society	DALY per 100k pop. per year	46	460	920	Non-linear
7 Indirect costs to society	Months	1	6		Linear
8 Additional treatment costs	1'000 CHF	10	80		Linear

Each surveyed individual encountered eight distinct decision scenarios in which they had to determine which option they personally preferred. The PROM utility scores of 10, 45, and 80 were referred to as "severe", "significant", and "mild" limitations, respectively and the disease burden was described as "low", "moderate", and "high".

The following figure displays one example of the decision scenarios.

Figure 2. Example of a decision scenario

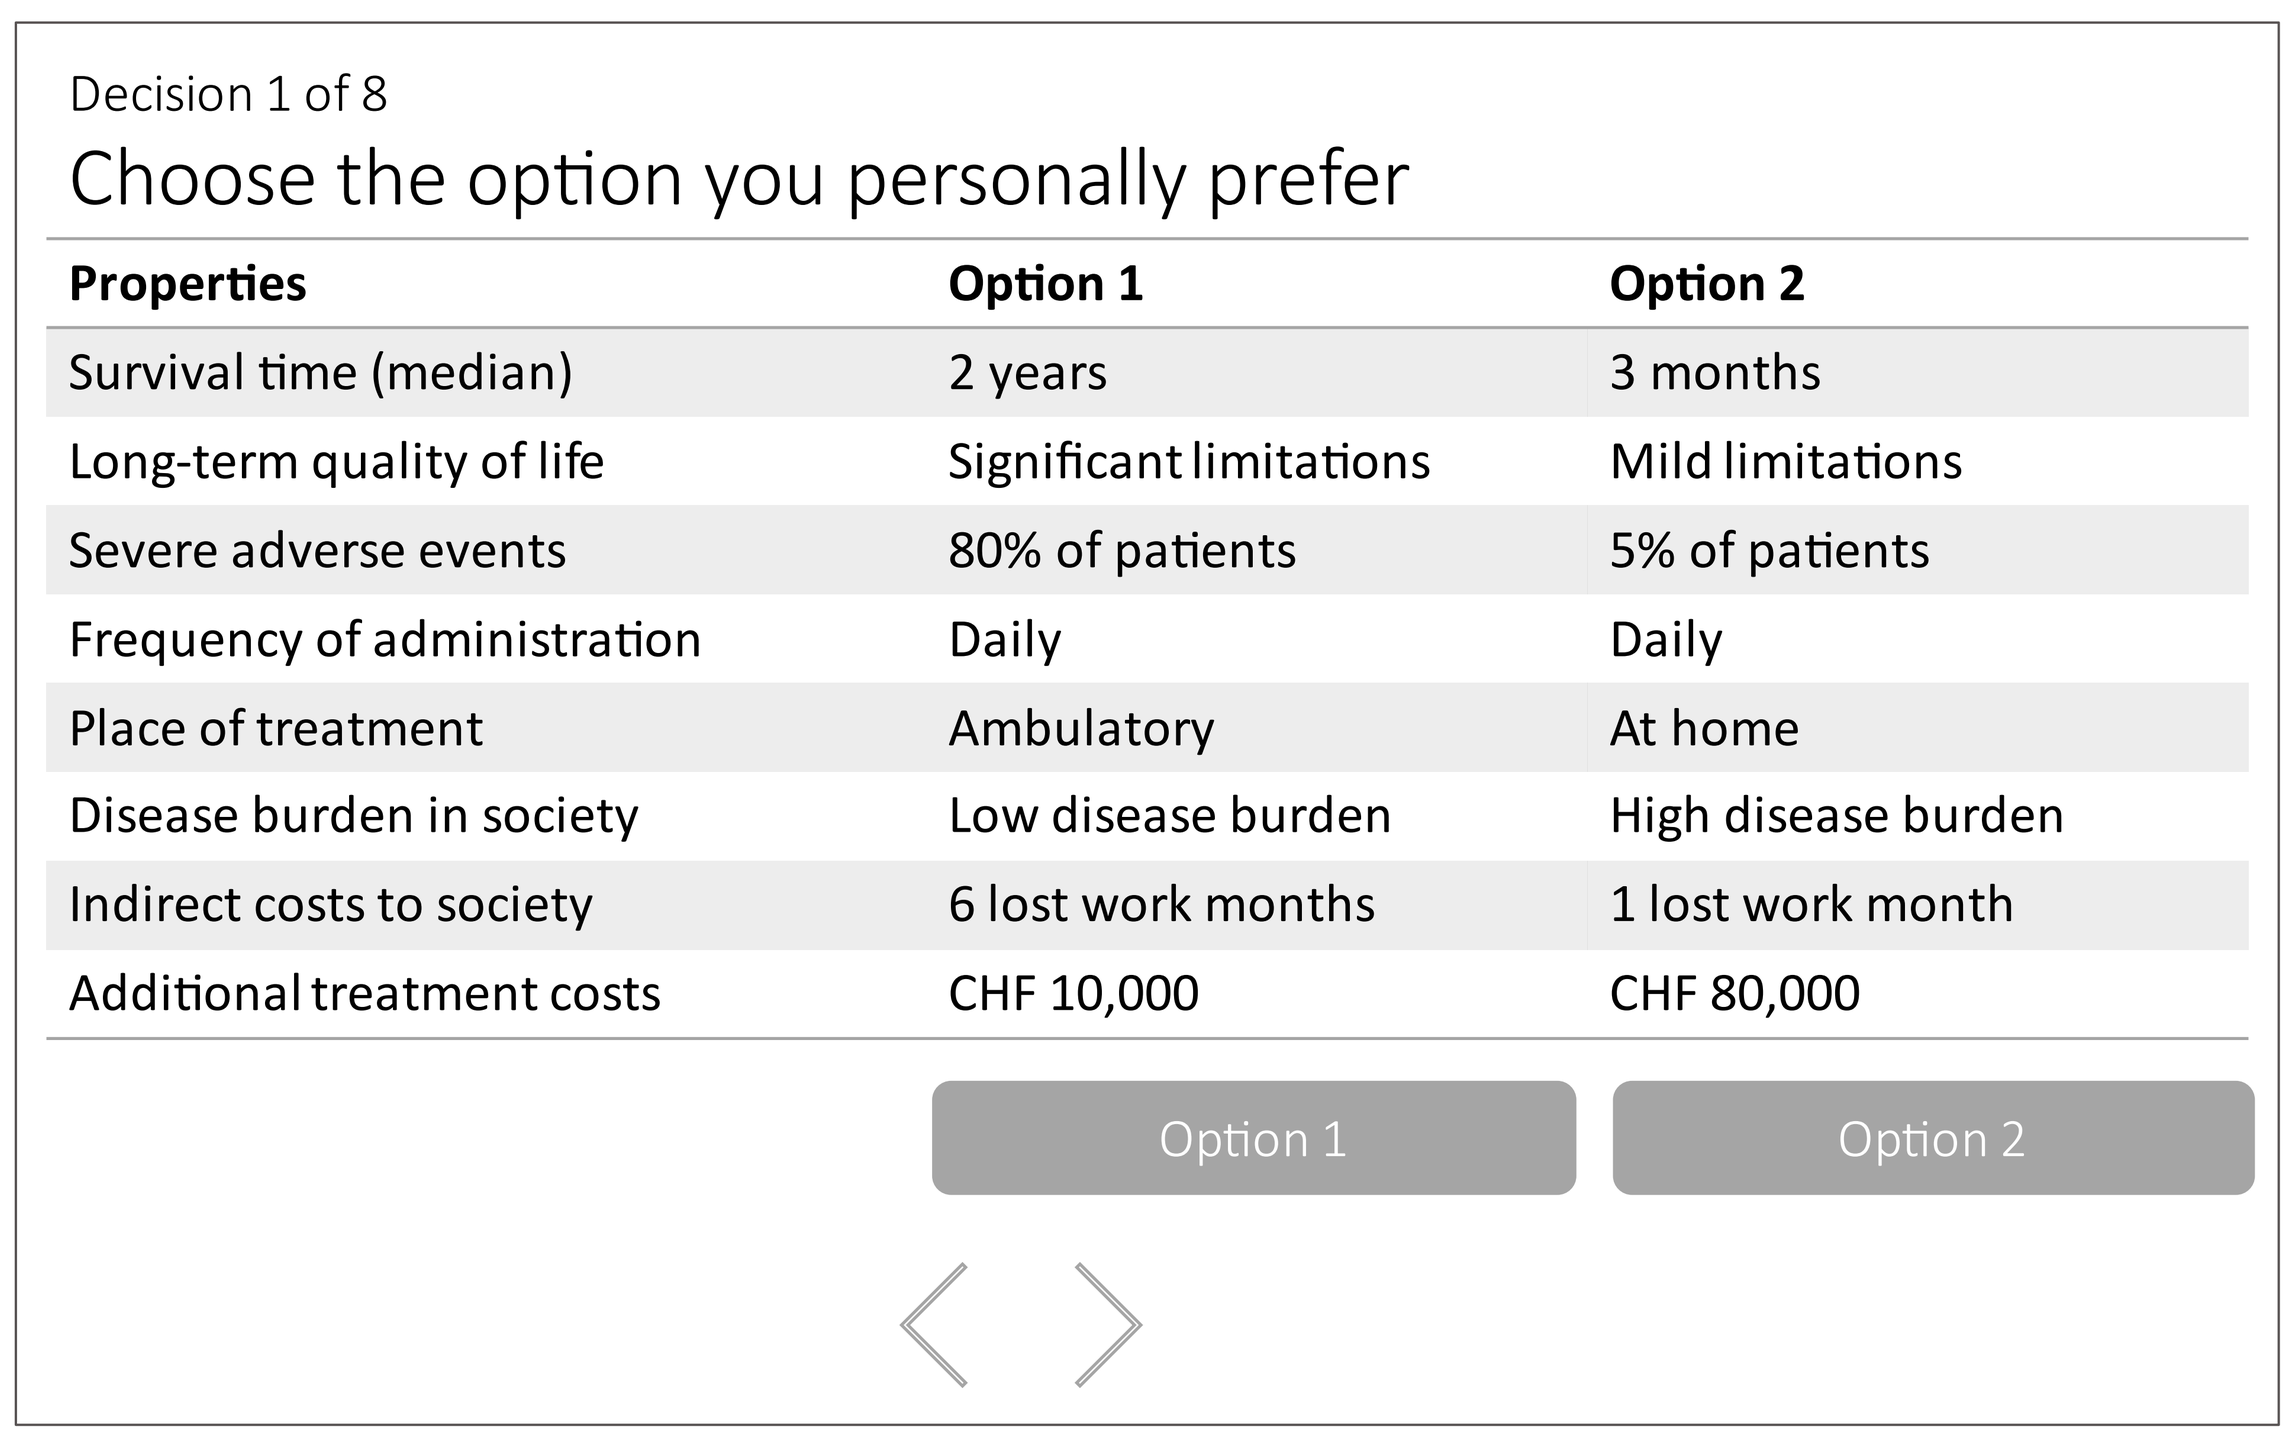

Decision 1 of 8

Choose the option you personally prefer

Properties	Option 1	Option 2
Survival time (median)	2 years	3 months
Long-term quality of life	Significant limitations	Mild limitations
Severe adverse events	80% of patients	5% of patients
Frequency of administration	Daily	Daily
Place of treatment	Ambulatory	At home
Disease burden in society	Low disease burden	High disease burden
Indirect costs to society	6 lost work months	1 lost work month
Additional treatment costs	CHF 10,000	CHF 80,000

Option 1 Option 2

All attribute levels, except for moderate disease burden (compared to low disease burden) and indirect costs to society, are statistically significant and different from zero. The signs and magnitudes of the coefficients are plausible.

Table 4. Coefficients from the DCE

Attribute	Value	Coefficient	Standard error	p-Value
Survival time after therapy initiation (Median)	3 months	0	.	.
	2 years	1.625***	0.114	0.000
	5 years	2.537***	0.160	0.000
Long-term quality of life	Severe limitations	0	.	.
	Moderate limitations	1.013***	0.055	0.000
	Mild limitations	1.496***	0.075	0.000
Severe adverse events	5%	0	.	.
	80%	-0.792***	0.049	0.000
Treatment frequency	Daily	0	.	.
	Monthly	0.194***	0.032	0.000
Treatment location	Ambulatory	0	.	.
	At home	0.305***	0.035	0.000
Disease burden in society	Low disease burden	0	.	.
	Moderate disease burden	-0.023	0.031	0.473
	High disease burden	-0.138***	0.029	0.000
Indirect costs to society	1 month of lost work	0	.	.
	6 months of lost work	-0.029	0.023	0.193
Additional treatment costs	10'000 CHF	0	.	.
	80'000 CHF	-0.242***	0.027	0.000

***: significant at the 0.1% level, **: 1% level, *: 5% level.

Based on the coefficients from the DCE, the weights for each attribute were calculated. Patient benefit accounts for a total weight of 93 percent, while attributes measuring societal benefit receive a total weight of 7 percent.

The resulting weighting was determined for the advanced cancer therapy setting and should not be simply transferred to another context. However, in the present context, the weighting can be considered robust.

There are no significant differences observed between different population groups, and a previous expert survey also yielded a similar result.

Figure 3. Weighting of attributes in the setting of advanced cancer therapies

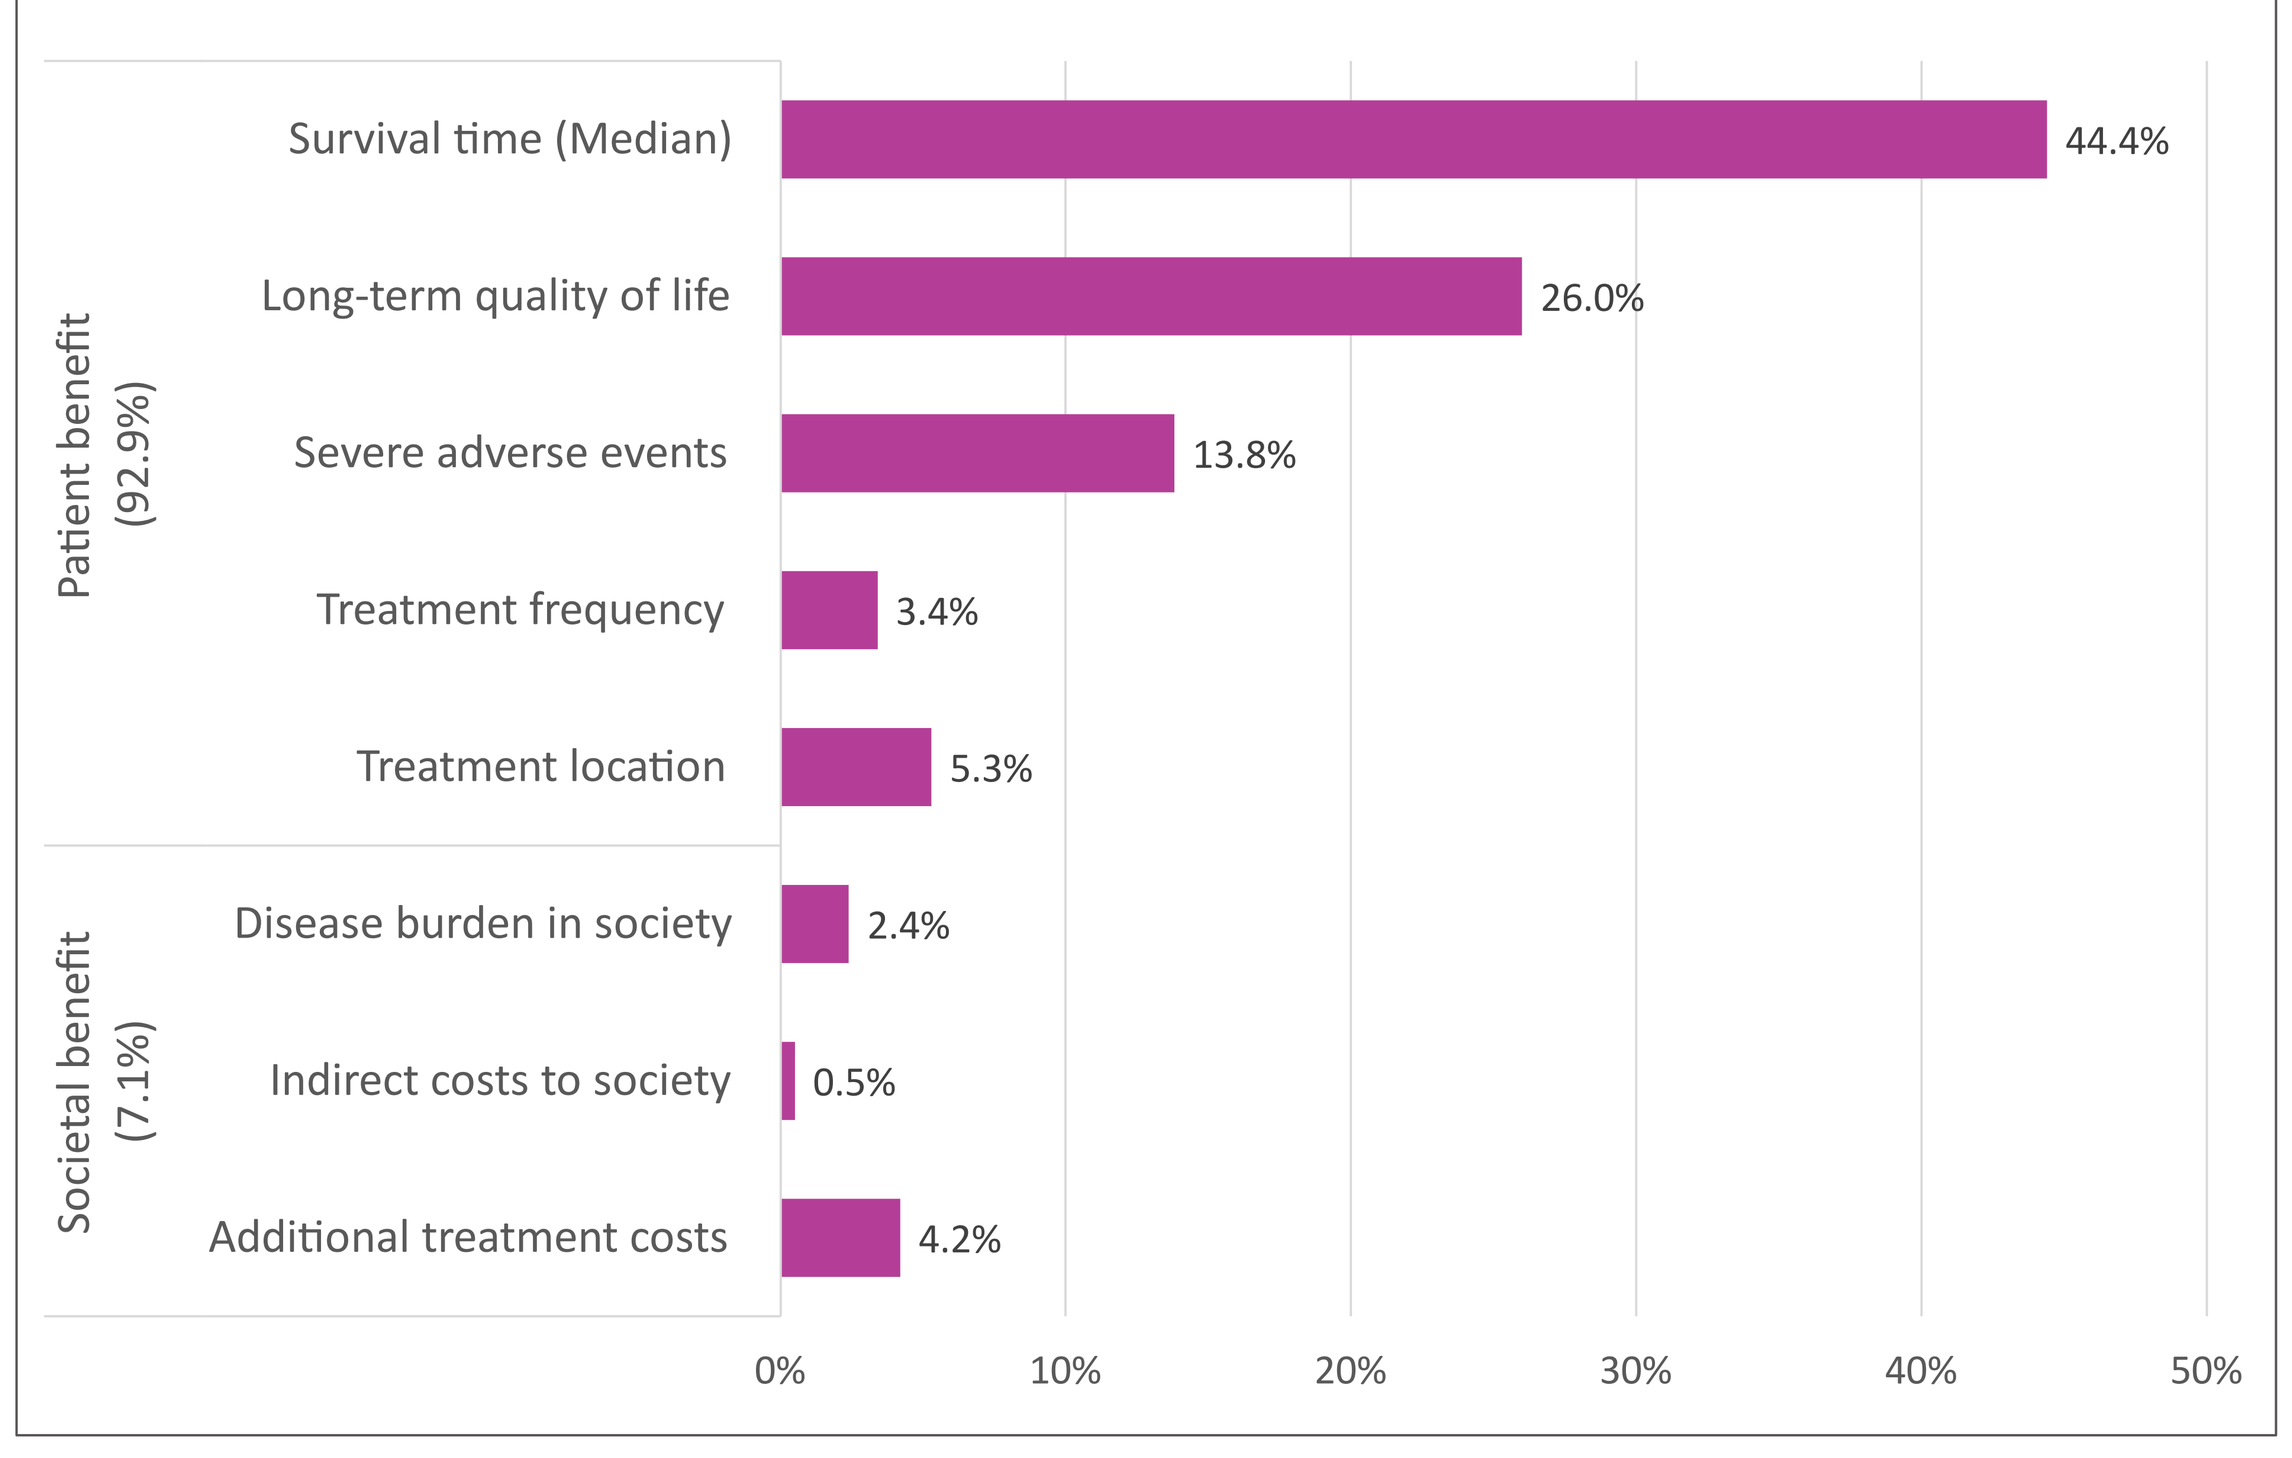
Patient benefit (92.9%)
Survival time (Median) 44.4%
Long-term quality of life 26.0%
Severe adverse events 13.8%
Treatment frequency 3.4%
Treatment location 5.3%
Societal benefit (7.1%)
Disease burden in society 2.4%
Indirect costs to society 0.5%
Additional treatment costs 4.2%
0% 10% 20% 30% 40% 50%

C Case study

From the data sources we extract the attribute specifications summarized in the following table.

Table 5. Attribute levels

Attribute	Measurement	nivolumab + ipilimumab	sunitinib	Source
Survival	Median OS	48.1 months	26.6 months	Albiges et al. (2020)
Long-term quality of life	EQ-5D 3L UK	82	75	Cella et al. (2019)
Severe adverse events	Percentage of adverse events	48%	64%	Albiges et al. (2020)
Patient convenience	Treatment frequency	14.5 days	1.5 days	Motzer et al. (2018)
	Treatment location	Ambulatory (clinic or hospital)	At home	Motzer et al. (2018)
Disease burden	DALY (Rate per 100k pop. per year)	31, used: minimum level of 46	As in innovation	Global burden of disease database, Cella et al. (2019)
Costs	Indirect costs (months of work loss)	1 month	1 month	-
	Additional treatment costs	72,100 CHF	73,335 CHF	Oniangue-Ndza et al. (2019)

For both treatment options, the respective input values are provided, along with their corresponding individual derived values score. (In the next table, for example, an input of 48.1 months of median survival time translates to a weighted value score of 43.4.)

The bottom row of the following table shows the sum of the individual utilities. Accordingly, the innovation scores 80 points, while the comparator scores 68 points. The theoretical maximum is 100 points.

Table 6. Assessment

Attributes	Unit	nivolumab + ipilimumab Input	nivolumab + ipilimumab Value score	sunitinib Input	sunitinib Value score
1 Survival time (Median OS)	Months	48.1	43.4	26.6	30.8
2 Long-term quality of life	Utilities according to list	80.0	26.0	75.0	25.4
3 Severe adverse events	Share of patients	48.0	5.9	64.0	2.9
4 Treatment frequency	in days	14.5	1.6	1.5	0.1
5 Treatment at home	1: yes, 0: no	0.0	0.0	1.0	5.3
6 Disease burden	DALY / 100k capita/year	46.0	2.4	46.0	2.4
7 Indirect costs (months of work loss)	Months	1.0	0.5	1.0	0.5
8 Additional treatment costs	1,000 CHF	72.1	0.5	73.3	0.4
Total value score			80		68

Despite having collected sufficient and representative data through the population survey, allowing for precise estimation of weights, the calculation of individual benefits and the overall assessment provide an arithmetic precision that is not justified. Direct interpretation of the benefit difference is not recommended. We propose a categorization of differences in the value scores that allows for an interpretation of the improvement in benefits.

D Dealing with uncertainty

The resulting value increase (12 units) in our case study turns out to be fairly robust, as we show in our sensitivity analyses. We address four uncertainties, for which we indicate how much the original difference in the value scores can change.

1. Long-term quality of life (QoL) is not observed long enough. In our baseline we are pessimistic about the innovation. If we are optimistic, the value score difference increases by 1 unit.
2. The total value score is obtained from a QoL-measure of 80 and above, which indicates a ceiling for the innovation (with value 82). This censoring can be avoided by rescaling the value function to a higher limit. If we set the limit to 90, the value score difference increases by 1 unit.
3. Structural uncertainty exists with regard to the shape of the three nonlinear value functions (Table 3). If we use the square root function instead of the quadratic function, the value score difference decreases by 1 unit.
4. Missing or incomplete data: We have no information on indirect costs. The utility difference potentially changes by the weight of the attribute (Figure 3: plus/minus 0.5).

Figure 4. Sensitivity of the value score difference

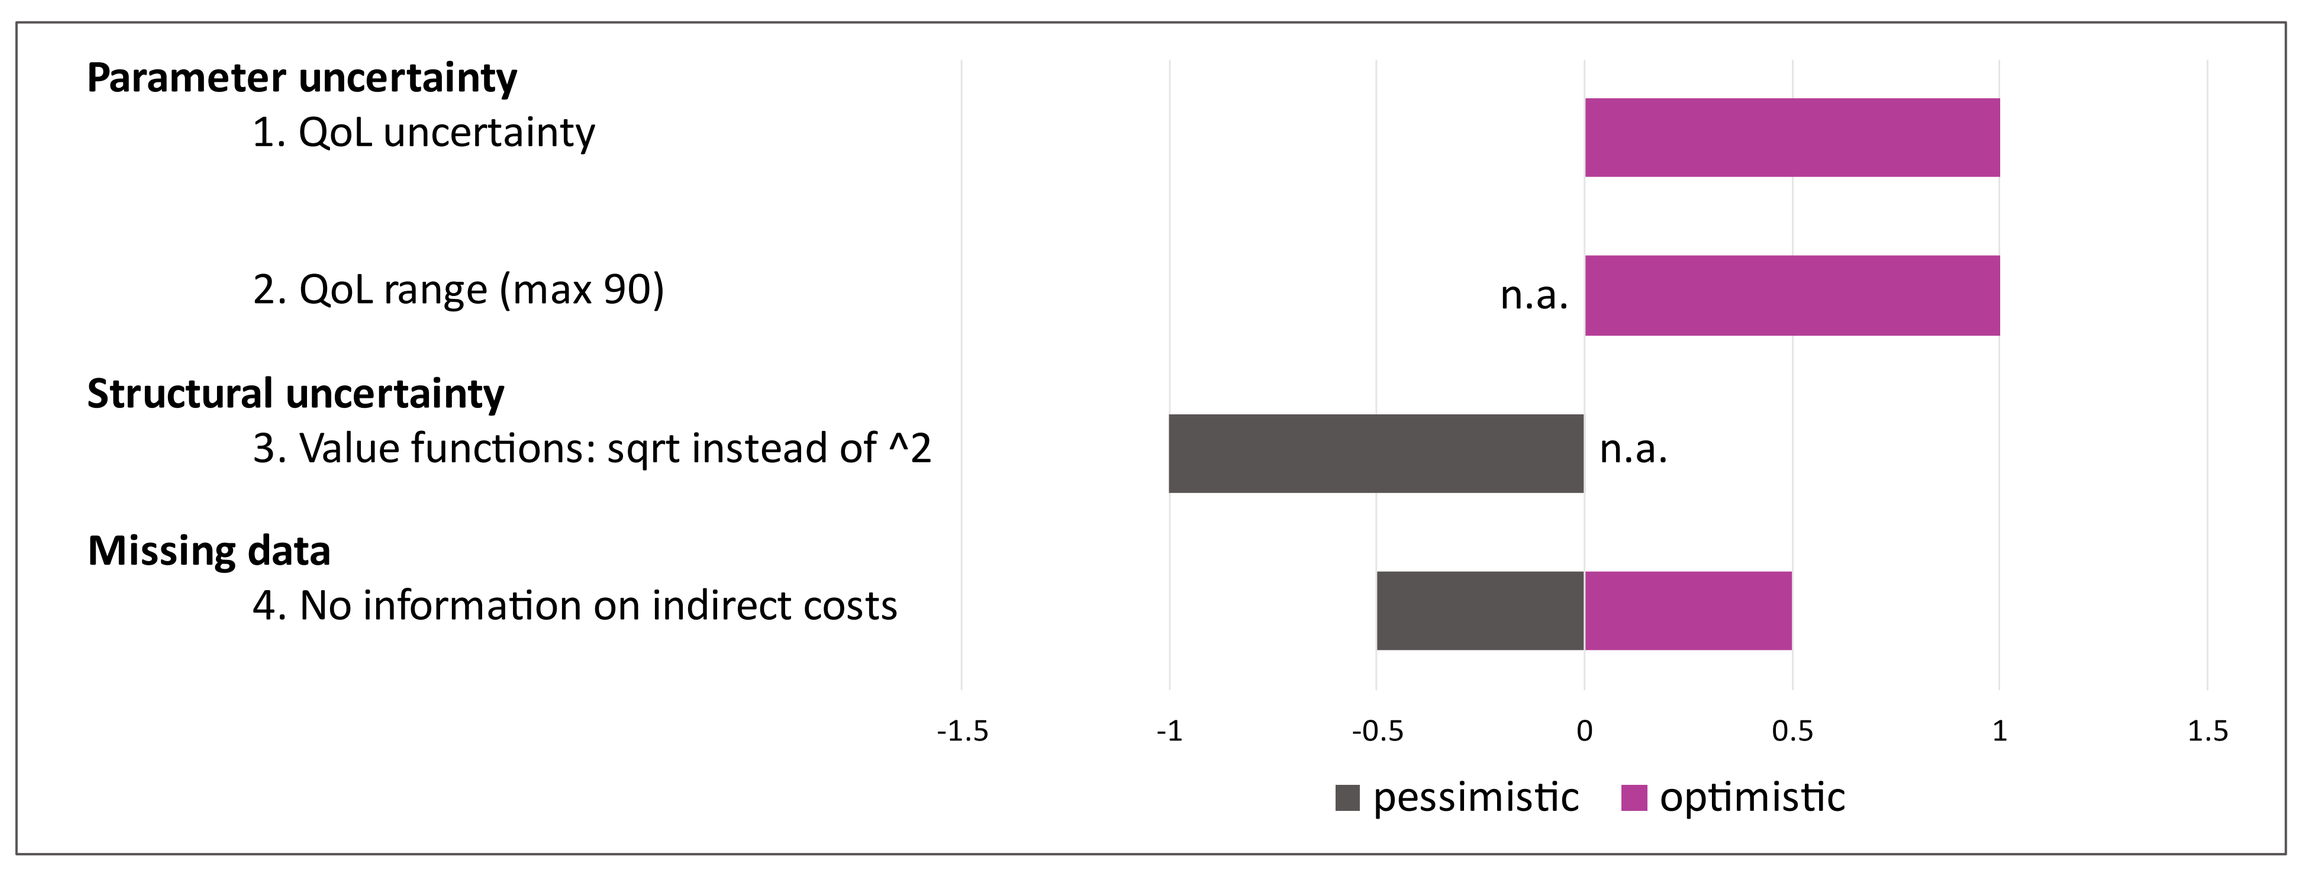
Parameter uncertainty
1. QoL uncertainty
2. QoL range (max 90)
n.a.
Structural uncertainty
3. Value functions: sqrt instead of ^2
n.a.
Missing data
4. No information on indirect costs
-1.5 -1 -0.5 0 0.5 1 1.5
pessimistic optimistic

Limitations

The model is only as good as its input. Although we use weights that correspond to the preferences of the Swiss population, other aspects are less well supported: Some input values were outside the defined ranges, or the data quality itself is not optimal.

Our proposal is not yet fully developed in every detail. Hence, the absolute values should not be directly interpreted – instead, we recommend focusing on the difference in values across categories.

Conclusion

- The valuation of MCDA attributes through the general population offers a solid foundation for quantifying the value of innovative treatments.
- This tool can assist Swiss payers when evaluating the value of innovative medicines for advanced cancer and its results might be used for reimbursement decisions.
- The case study serves as a proof-of-concept. It quantifies the value gain from treating advanced renal cell carcinoma with nivolumab plus ipilimumab instead of sunitinib.

References

1. Albiges, L., Tannir, N.M., Burotto, M., McDermott, D., Plimack, E.R., Barthélémy, P., Porta, C., Powles, T., Donskov, F., George, S., Kollmannsberger, C.K., Gurney, H., Grimm, M.-O., Tomita, Y., Castellano, D., Rini, B.I., Choueiri, T.K., Saggi, S.S., McHenry, M.B., Motzer, R.J., 2020. Nivolumab plus ipilimumab versus sunitinib for first-line treatment of advanced renal cell carcinoma: extended 4-year follow-up of the phase III CheckMate 214 trial. ESMO Open 5, e001079. https://doi.org/10.1136/esmoopen-2020-001079
2. Cella, D., Grünwald, V., Escudier, B., Hammers, H.J., George, S., Nathan, P., Grimm, M.-O., Rini, B.I., Doan, J., Ivanescu, C., Paty, J., Mekan, S., Motzer, R.J., 2019. Patient-reported outcomes of patients with advanced renal cell carcinoma treated with nivolumab plus ipilimumab versus sunitinib (CheckMate 214): a randomised, phase 3 trial. Lancet Oncol. 20, 297–310. https://doi.org/10.1016/S1470-2045(18)30778-2
3. Marsh, K., IJzerman, M., Thokala, P., Baltussen, R., Boysen, M., Kaló, Z., Lönngren, T., Mussen, F., Peacock, S., Watkins, J., Devlin, N., 2016. Multiple Criteria Decision Analysis for Health Care Decision Making—Emerging Good Practices: Report 2 of the ISPOR MCDA Emerging Good Practices Task Force. Value Health 19, 125–137. https://doi.org/10.1016/j.jval.2015.12.016
4. McFadden, D., 2001. Economic Choices. Am. Econ. Rev. 91, 351–378.
5. Motzer, R.J., Tannir, N.M., McDermott, D.F., Arén Frontera, O., Melichar, B., Choueiri, T.K., Plimack, E.R., Barthélémy, P., Porta, C., George, S., Powles, T., Donskov, F., Neiman, V., Kollmannsberger, C.K., Salman, P., Gurney, H., Hawkins, R., Ravaud, A., Grimm, M.-O., Bracarda, S., Barrios, C.H., Tomita, Y., Castellano, D., Rini, B.I., Chen, A.C., Mekan, S., McHenry, M.B., Wind-Rotolo, M., Doan, J., Sharma, P., Hammers, H.J., Escudier, B., 2018. Nivolumab plus Ipilimumab versus Sunitinib in Advanced Renal-Cell Carcinoma. N. Engl. J. Med. 378, 1277–1290. https://doi.org/10.1056/NEJMoa1712126
6. Oniangue-Ndza, C., Schneider, R., Malcolm, B., May, J., Gooden, K.M., Klijn, S., Cakar, E., 2019. Cost-effectiveness of Nivolumab in combination with Ipilimumab compared to Sunitinib for the first-line treatment of advanced/metastatic intermediate- or poor-risk renal carcinoma in Switzerland. Value Health 22, S462. https://doi.org/10.1016/j.jval.2019.09.336

Acknowledgments

This study was supported by Bristol Myers Squibb. All authors contributed to and approved the presentation.

Presented at ISPOR Europe 2023; November 12-15, 2023; Copenhagen, Denmark

Email: florian.kuhlmey@polynomics.ch

Copies of this poster are for personal use only and may not be reproduced without written permission of the authors.