Recurrence-free survival as a surrogate endpoint for overall survival in resected stage II/III melanoma: a correlation meta-analysis of randomized controlled trials

CO54

Leung L[1], Kirkwood JM[2], Srinivasan S[3], Dyer M[4], Qian A[1], Pourrahmat MM[1], Kasireddy E[1], May JR[4], Moshyk A[3], Kurt M[3]

[1]Evidinno Outcomes Research Inc., Vancouver, BC, Canada; [2]University of Pittsburgh, Pittsburgh, PA, USA;

[3]Bristol Myers Squibb, Princeton, NJ, USA; [4]Bristol Myers Squibb, Uxbridge, UK

Introduction

- In 2020, melanoma accounted for 4% of all new cancer cases and 1.3% of all cancer deaths in 27 European Union Member States (EU-27), making it the fifth most common malignancy and placing it in the 15 most frequent causes of cancer deaths in the EU-27[1]
- Overall survival (OS) is universally recognized as being an unambiguous and unbiased endpoint with paramount clinical relevance in randomized controlled trials (RCTs) of oncology.[2] However, observing a statistically mature OS benefit may require considerable follow-up time. Therefore, establishing intermediate endpoints that may reach statistical maturity sooner than OS as valid surrogates could expedite drug development and improve patient access to treatments[3,4]
- RFS-OS surrogacy has been previously studied in the literature
 - — One previous study in stage II/III melanoma investigating recurrence-free survival (RFS) as a surrogate endpoint for OS concluded RFS was a valid surrogate for OS; however, this study identified its evidence from a pre-2018 literature review, and restricted the trials to those evaluating adjuvant interferon therapy and those with available individual patient-level data, leading to only 13 trials included in the final analyses[5]
 - — Another study investigating RFS-OS surrogacy in the adjuvant therapy of melanoma with checkpoint inhibitors found a strong association at the patient level but only a moderate association at the trial level[6]

Objectives

- To evaluate the appropriateness of RFS as a surrogate endpoint for OS using the most up-to-date evidence base for patients with resected stage II/III melanoma receiving any adjuvant therapy and using aggregate trial-level data
- To investigate the predictive accuracy of the surrogacy equations for the utility and validity of the model

Methods

Targeted literature review

- A targeted literature review was conducted to search MEDLINE®, Embase, and Cochrane Central Register of Controlled Trials (CENTRAL) from database inception to July 20, 2022. Searches were limited to the English language
- Included articles were randomized or non-randomized clinical trials on patients ≥12 years with resected stage II-III melanoma receiving adjuvant therapy
- Outcomes of interest were OS and RFS (or its analogs such as disease-free survival or disease-free interval). To be included in the analysis, the trials must have reported relative treatment effects for both OS and RFS either in the form of hazard ratios (HR_{OS} and HR_{RFS}) or Kaplan-Meier curves

Data analysis

Trial-level surrogacy models and analysis sets

- The surrogacy of RFS for OS at the trial level was assessed using two meta-analysis models. HRs on each endpoint were log-transformed to be consistent with the linearity assumption for the relationship between the treatment effects
- The first model was based on an alternative bivariate random-effects meta-analysis (BRMA) model proposed by Riley et al 2008,[7] which provides an overall correlation measure between $\log(HR_{RFS})$ and $\log(HR_{OS})$
- The second model was a weighted linear regression (WLR) model where each study was weighted by its corresponding sample size in a regression model estimating HR_{OS} from HR_{RFS}. The association between $\log(HR_{RFS})$ and $\log(HR_{OS})$ was measured by the Pearson correlation coefficient
- In addition to the primary analysis, two sensitivity analyses were conducted by (1) omitting comparisons that failed to satisfy the proportional hazards assumption, and (2) restricting the evidence base to RCTs published after the year 2000. The purpose behind the second sensitivity analysis was to investigate the impact of the mechanism of action and changes in the subsequent treatment landscape on the surrogacy relationship, as studies published prior to 2000 mostly evaluated non-interferon interventions (e.g., levamisole, melanoma vaccine, transfer factor)
- A meta-regression was conducted by including disease stage at baseline as a binary covariate with two levels (0 – stage II; 1 – stage III)

Assessing the surrogacy equation and the correlation estimates

- The validity of the model was assessed by using a leave-one-out cross-validation (LOOCV) approach based on WLR models
- The National Institute for Health and Care Excellence (NICE) Decision Support Unit (DSU) Technical Support Document 20 was used as a guide to assess model validity.[8] A surrogacy model can be deemed valid if the observed HR_{OS}'s were covered by the 95% prediction interval (PI) for at least 95% of the comparisons
- The surrogate threshold effect (STE), which is defined as the minimum treatment effect on RFS that would translate into a statistically significant and positive treatment effect on OS at a 95% significance level, was estimated
 - — In statistical terms,[9] STE corresponds to the HR_{RFS} at which the upper bound of 95% PI of the HR_{OS} crosses 1
 - — As the STE could change based on the sample size of a prospective RCT, it was estimated for two hypothetical trial sample sizes of 500 and 800 patients
- German Institute of Quality and Efficiency in Health Care (IQWiG) guidelines were used to assess the strength of the correlation estimate[10]
- According to the IQWiG criteria, a correlation is considered high if the lower limit of the 95% confidence interval (CI) of the estimated correlation coefficient ≥ 0.85, low if the upper limit of the 95% CI of the estimated correlation coefficient ≤ 0.7, and medium otherwise

Results

Targeted literature review

- Of 4,029 records identified, 31 publications pertaining to 30 unique RCTs (stage II/III: n = 8/11; mixed: n = 11), published between 1978 to 2022 (median: 2013) were included in the literature review and subsequent correlation meta-analysis (**Figure 1**)
- The RCTS included 45 to 1,670 patients (median: 519)
- Treatments studied in the RCTs involved mostly interferon-alfa (n = 17), followed by other immunotherapy-containing regimens (n = 10), immune checkpoint inhibitors (ICIs; n = 3), and targeted therapies (n = 2). The RCTs included in the evidence base and the therapies they investigated are listed in **Table 1**

Figure 1. PRISMA flow diagram

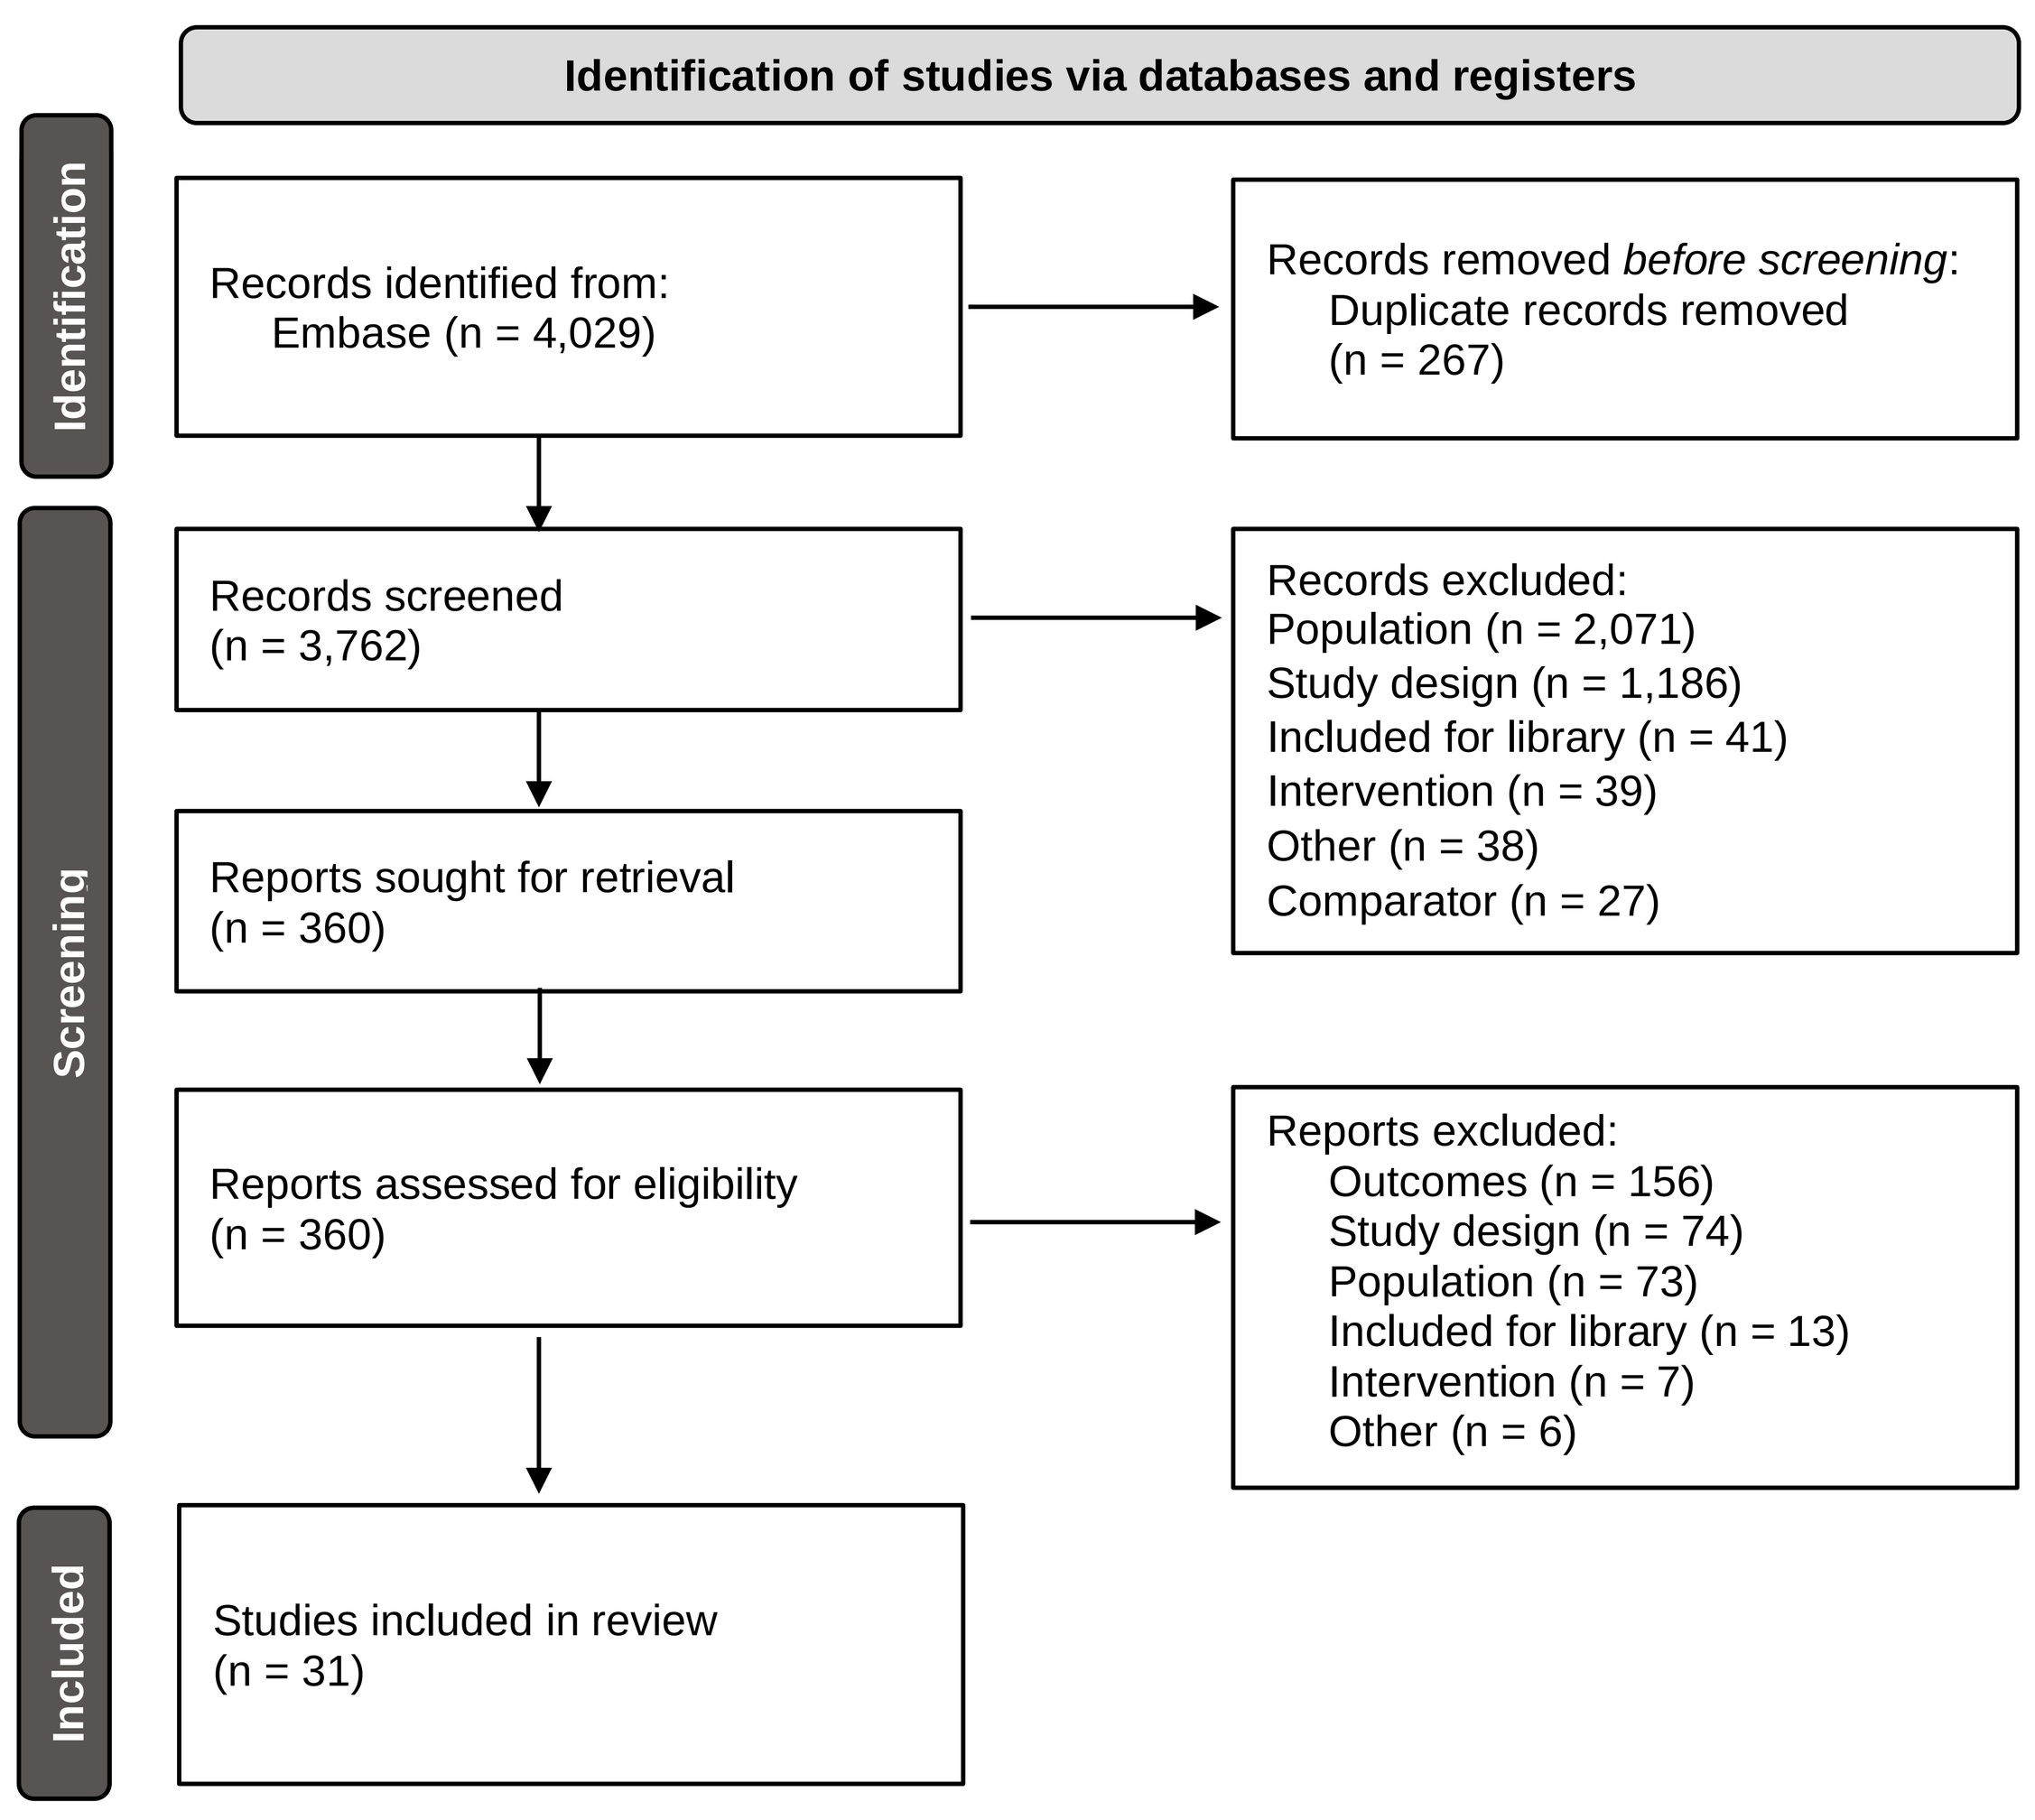
Identification of studies via databases and registers
Identification
Records identified from:
Embase (n = 4,029)
Records removed *before screening*:
Duplicate records removed
(n = 267)
Screening
Records screened
(n = 3,762)
Records excluded:
Population (n = 2,071)
Study design (n = 1,186)
Included for library (n = 41)
Intervention (n = 39)
Other (n = 38)
Comparator (n = 27)
Reports sought for retrieval
(n = 360)
Reports assessed for eligibility
(n = 360)
Reports excluded:
Outcomes (n = 156)
Study design (n = 74)
Population (n = 73)
Included for library (n = 13)
Intervention (n = 7)
Other (n = 6)
Included
Studies included in review
(n = 31)

Table 1. List of RCTs included in the evidence base and the treatments they investigated

Trial	Intervention	Comparator
AIM HIGH	IFN-α-2a	PBO
AVAST-M	Bevacizumab	PBO
Cameron et al (2001)	IFN-α-2b	PBO
Cascinelli et al (2001)	IFN-α-2b	PBO
Checkmate 238	Nivolumab	Ipilimumab
COMBI-AD	Dabrafenib + trametinib	PBO
Creagan et al (1995)	IFN-α-2a	PBO
ECOG-ACRIN E1609	Ipilimumab	High-dose IFN-α
ECOG 1690	IFN-α-2b (low dose)	PBO
Eigentler et al (2016)	Pegylated IFN-α-2a	IFN-α-2a
EORTC 18071	Ipilimumab	PBO
EORTC 18081	Pegylated IFN-α-2b	PBO
EORTC 18871	Iscador-M®	PBO
EORTC 18952	IFN-α-2b (lower dose)	PBO
EORTC 18961	GM2-KLH/QS-21 vaccine	PBO
EORTC 18991	Pegylated IFN-α-2b	PBO
EORTC E1697	IFN-α-2b	PBO
Flaherty et al (2014)	Biochemotherapy (dacarbazine, cisplatin, vinblastine, interleukin-2, IFN-α-2b and granulocyte colony-stimulating factor)	High-dose IFN
Garbe et al (2008)	IFN-α-2a	PBO
Gonzalez et al (1978)	Levamisole	PBO
Khammari et al (2020)	Adoptive tumor-infiltrating lymphocytes therapy and interleukin-2	Abstention (did not receive any other melanoma treatments prior to inclusion)
Kim et al (2009)	Biochemotherapy (cisplatin, vinblastine, dacarbazine, IFN-α-2b, interleukin-2)	IFN-α-2b (high or intermediate dose)
Lian et al (2013)	Temozolomide + cisplatin	IFN-α-2b (high dose)
MAVIS	Seviprotimut-L	PBO
Miller et al (1988)	Transfer factor	PBO
Mohr et al (2015)	Intermittent high-dose IFN-α-2b	IFN-α-2b (high dose)
Nordic IFN trial	IFN-α-2b (3 years)	PBO
Oratz et al (1991)	Melanoma antigen vaccine + cyclophosphamide	Melanoma antigen vaccine
SWOG-9035	Melacine cell lysate + DETOX	PBO
Wallack et al (1995)	Vaccinia melanoma oncolysate	Vaccinia vaccine virus - PBO

DETOX: detoxified Freund adjuvant, containing mycobacterial cell wall skeleton plus monophosphoryl lipid A; IFN: interferon; PBO: this term encompasses placebo, no treatment, or observation.

Figure 2. Surrogacy equation derived from WLR model for the primary analysis and the STEs for RCTs with 500 and 800 patients

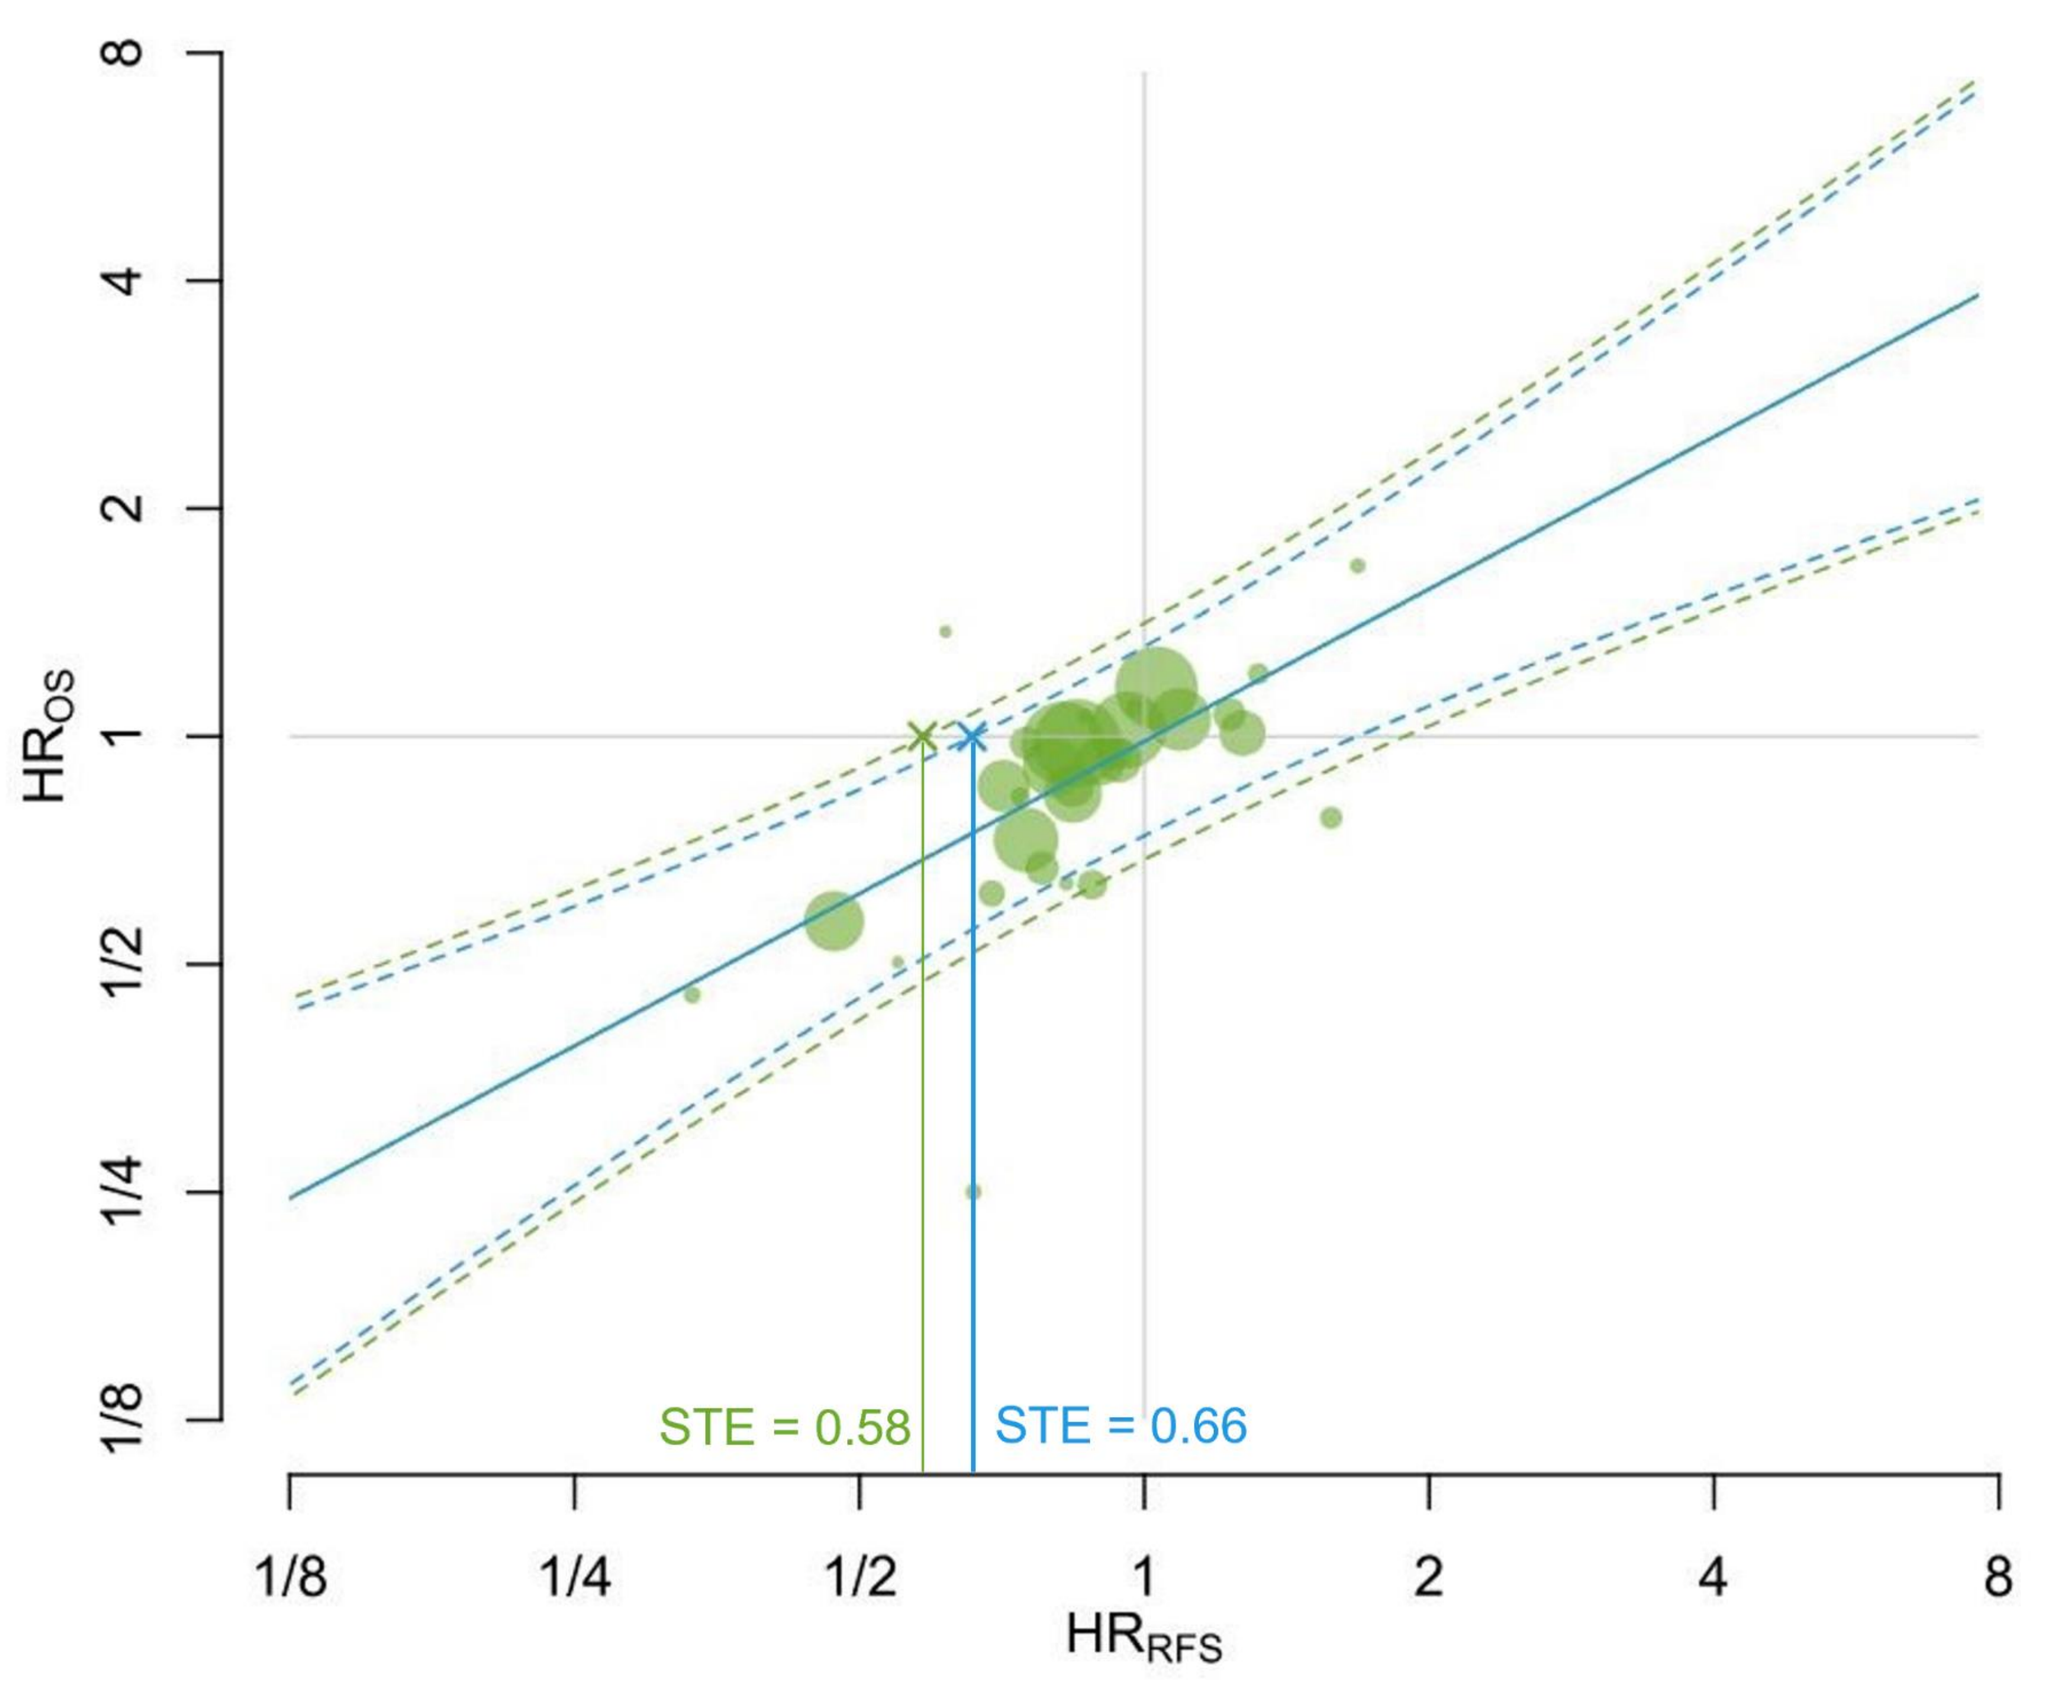
8
4
2
1
1/2
1/4
1/8
HROS
STE = 0.58
STE = 0.66
1/8
1/4
1/2
1
2
4
8
HRRFS

Legend: The WLR is graphed as a solid blue straight line with its corresponding 95% predictive interval boundaries as dotted curved lines (green for the sample size of 500 patients, and blue for the sample size of 800 patients). The green dots represent relative treatment effects from the trials in the evidence base and their sizes represent their relative weights (i.e., sample size) in the WLR.

HR: hazard ratio; OS: overall survival; RFS: recurrence-free survival; STE: surrogate threshold effect; WLR, weighted linear regression.

Primary analysis

- The estimated surrogacy equation was $\log(HR_{OS}) = -0.01 + 0.67 \times \log(HR_{RFS})$
- Using LOOCV, 29 out of 31 (93.5%) comparisons were captured by their 95% PIs
- The estimated STEs ranged between 0.58-0.66 for RCTs including 500-800 patients (the higher the sample size the higher the STE)
- The WLR model and LOOCV on the primary analysis are presented in **Figure 2** and **Figure 3**, respectively

Sensitivity analysis 1: omitting studies or comparisons that failed to satisfy the proportional hazards assumption

- Based on 22 comparisons from 21 trials, the estimated surrogacy equation was $\log(HR_{OS}) = 0.02 + 0.83 \times \log(HR_{RFS})$
- Using LOOCV, in 21/22 (95.5%) comparisons the observed OS HRs were captured by their 95% PIs generated from the model
- The STEs were calculated to be 0.58 (for a hypothetical RCT of 500 patients) and 0.65 (for a hypothetical RCT of 800 patients)

Sensitivity analysis 2: using RCTs published after the year 2000

- Based on 26 comparisons from 25 trials, the estimated surrogacy equation was $\log(HR_{OS}) = 0.00 + 0.75 \times \log(HR_{RFS})$
- Using LOOCV, in 25 out of 26 (96.2%) comparisons, the observed OS HRs were captured by their 95% PIs generated from the model
- The STEs were calculated to be 0.61 (for a hypothetical RCT of 500 patients) and 0.68 (for a hypothetical RCT of 800 patients)

Meta-regression with disease stage as a covariate

- Disease stage was treated as a separate binary covariate in the WLR with two levels (stage II, stage III)
- Based on 28 comparisons from 23 trials, the estimated surrogacy equation was $\log(HR_{OS}) = -0.06 + 0.77 \times \log(HR_{RFS}) + 0.04 \times (\text{stage} = \text{III}) - 0.08 \times \log(HR_{RFS}) \times (\text{stage} = \text{III})$
- For stage II patients, the above equation becomes $\log(HR_{OS}) = -0.06 + 0.77 \times \log(HR_{RFS})$ while for stage III patients it becomes $\log(HR_{OS}) = -0.02 + 0.69 \times \log(HR_{RFS})$. The two equations are similar, meaning that under the meta-regression model, staging has little effect on the relationship between $\log(HR_{RFS})$ and $\log(HR_{OS})$
- Using LOOCV, in 25 out of 28 (89.3%) comparisons, the observed OS HRs were captured by their 95% PIs generated from the model
- The STEs were calculated to be 0.53 and 0.56 for stages II and III, respectively (for a hypothetical RCT of 500 patients) and 0.61 and 0.63 (for a hypothetical RCT of 800 patients). The STEs were marginally sensitive to the disease stage

Figure 3. WLR model for the primary analysis

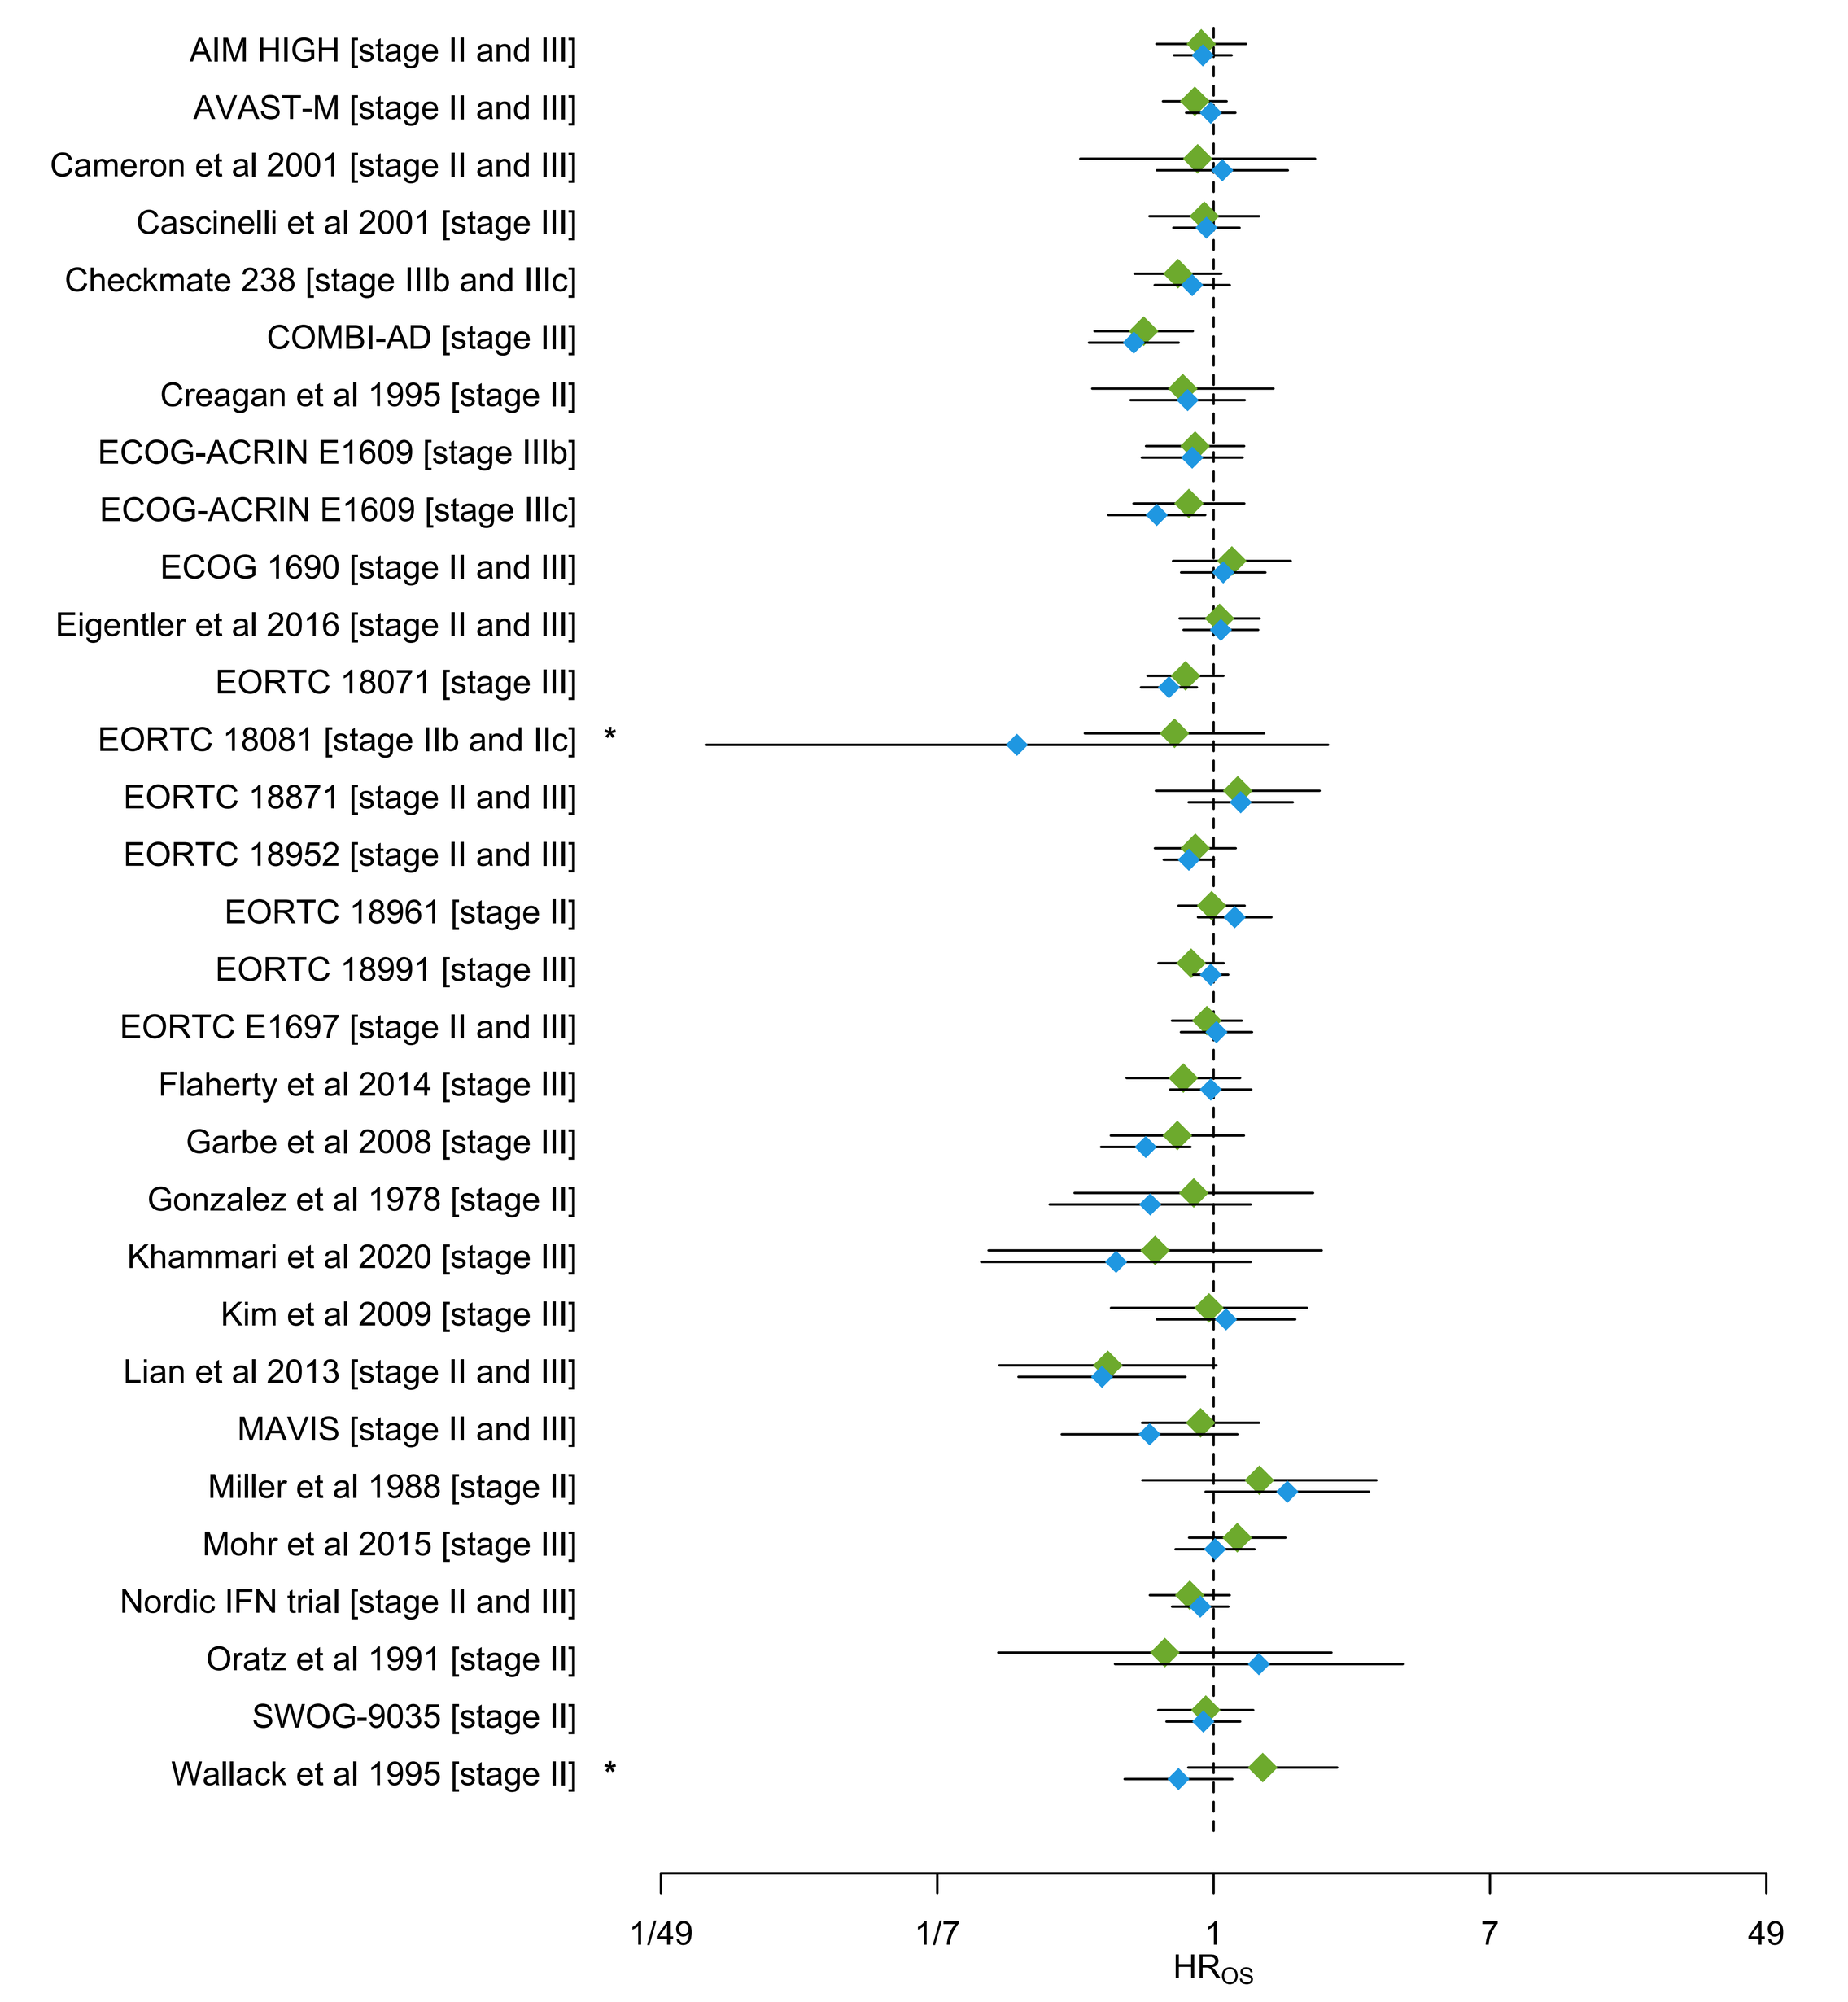
AIM HIGH [stage II and III]
AVAST-M [stage II and III]
Cameron et al 2001 [stage II and III]
Cascinelli et al 2001 [stage III]
Checkmate 238 [stage IIIb and IIIc]
COMBI-AD [stage III]
Creagan et al 1995 [stage II]
ECOG-ACRIN E1609 [stage IIIb]
ECOG-ACRIN E1609 [stage IIIc]
ECOG 1690 [stage II and III]
Eigentler et al 2016 [stage II and III]
EORTC 18071 [stage III]
EORTC 18081 [stage IIb and IIc] *
EORTC 18871 [stage II and III]
EORTC 18952 [stage II and III]
EORTC 18961 [stage II]
EORTC 18991 [stage III]
EORTC E1697 [stage II and III]
Flaherty et al 2014 [stage III]
Garbe et al 2008 [stage III]
Gonzalez et al 1978 [stage II]
Khammari et al 2020 [stage III]
Kim et al 2009 [stage III]
Lian et al 2013 [stage II and III]
MAVIS [stage II and III]
Miller et al 1988 [stage II]
Mohr et al 2015 [stage III]
Nordic IFN trial [stage II and III]
Oratz et al 1991 [stage II]
SWOG-9035 [stage II]
Wallack et al 1995 [stage II] *
1/49
1/7
1
7
49
HROS

Legend: The blue diamond and its error bars are the observed HR_{OS} and their 0.95% CI, respectively. The green diamond and its error bars are the predicted HR_{OS} and their 95% PI, respectively. The asterisks indicate the trials where the observed HR_{OS} was not covered by the 95% PI.

HR: hazard ratio; OS: overall survival.

Table 2. Summary of results from BRMA and WLR models

Analysis set	N	Correlation coefficient from BRMA (95% CI)	Correlation coefficient from WLR (95% CI)	LOOCV (% RCTs captured)	STE 1	STE 2
0	31	0.68 (0.45, 0.82)	0.71 (0.42, 0.87)	29/31 (93.5%)	0.58	0.66
1	22	0.74 (0.48, 0.89)	0.65 (0.25, 0.86)	21/22 (95.5%)	0.58	0.65
2	26	0.81 (0.62, 0.91)	0.77 (0.50, 0.90)	25/26 (96.2%)	0.61	0.68
3	28	0.69 (0.46, 0.84)	0.72 (0.39, 0.89)	25/28 (89.3%)	0.53 (stage II) 0.56 (stage III)	0.61 (stage II) 0.63 (stage III)

LOOCV: leave-one-out cross-validation; STE 1/2: surrogate threshold effect calculated using a hypothetical RCT of 500/800 patients; Analysis set 0: overall population; Analysis set 1: omitting studies or comparisons that failed to satisfy the proportional hazards assumption; Analysis set 2: using RCTs published after the year 2000; Analysis set 3: meta-regression analysis adjusting for staging.

- The BRMA correlation estimate between log-transformed HR_{RFS} and HR_{OS} for the primary analysis was 0.68 (95% CI: 0.45, 0.82)
- In the sensitivity analyses and the meta-regression, the correlation estimates from the BRMA ranged between 0.69-0.81
- According to the IQWiG criteria, correlation estimates from both the BRMA and the WLR were medium across all analyses
- A summary of the results of all the analyses is presented in **Table 2**

Conclusion

- Statistically meaningful correlations between the treatment effects on RFS and OS in patients with resected stage II/III melanoma receiving adjuvant therapy were observed across all analyses
- The surrogacy equation between the treatment effects on RFS and OS may enable earlier assessments of OS benefit from the RFS benefit for patients with resected stage II/III melanoma receiving adjuvant therapy
- Sensitivity analyses produced similar correlation estimates to those in the primary analysis, indicating the robustness of the analyses undertaken
- Overall, while findings in this study provide insights into the strength of RFS-OS surrogacy in broader stage II/III melanoma settings including the latest treatments in stage II-III melanoma (including ICIs and targeted therapies) to date, the majority of evaluable treatments are still non-ICIs. Therefore, future work is required to confirm the predictions from the surrogacy equations and the strength of the RFS-OS correlation in the ICI setting when further data from these trials is available
- To our knowledge, this is the most recent RFS-OS surrogacy study in the literature developed on the broadest evidence base

References

1. Koczkodaj P, Sulkowska U, Didkowska J, Rutkowski P, Mańczuk M. Melanoma Mortality Trends in 28 European Countries: A Retrospective Analysis for the Years 1960-2020. *Cancers*. 2023;15(5).
2. Driscoll J, Rixe O. Overall Survival: Still the Gold Standard Why Overall Survival Remains the Definitive End Point in Cancer Clinical Trials. *Cancer journal (Sudbury, Mass)*. 2009;15(5):401-405.
3. Chen EY, Joshi SK, Tran A, Prasad V. Estimation of Study Time Reduction Using Surrogate End Points Rather Than Overall Survival in Oncology Clinical Trials. *JAMA Internal Medicine*. 2019;179(5):642-647.
4. FDA. Clinical Trial Endpoints for the Approval of Cancer Drugs and Biologics Guidance for Industry. December 2018. https://www.fda.gov/media/71195/download [Accessed September 21, 2023].
5. Coart E, Suciu S, Squifflet P, et al. Evaluating the potential of relapse-free survival as a surrogate for overall survival in the adjuvant therapy of melanoma with checkpoint inhibitors. *Eur J Cancer*. 2020;137:171-174.
6. Suciu S, Eggermont AMM, Lorigan P, et al. Relapse-Free Survival as a Surrogate for Overall Survival in the Evaluation of Stage II-III Melanoma Adjuvant Therapy. *J Natl Cancer Inst*. 2018;110(1).
7. Riley RD, Thompson JR, Abrams KR. An alternative model for bivariate random-effects meta-analysis when the within-study correlations are unknown. *Biostatistics*. 2008;9:172-186.
8. Bujkiewicz S, Achana F, Papanikos T, Riley R, Abrams K. Multivariate meta-analysis of summary data for combining treatment effects on correlated outcomes and evaluating surrogate endpoints. *NICE DSU technical support document*. 2019;20.
9. Burzykowski T, Buyse M. Surrogate threshold effect: an alternative measure for meta-analytic surrogate endpoint validation. *Pharm Stat*. 2006;5(3):173-186.
10. Validity of surrogate endpoints in oncology: Executive summary of rapid report A10-05, Version 1.1. *Institute for Quality and Efficiency in Health Care: Executive Summaries [Internet]* 2011; https://www.ncbi.nlm.nih.gov/books/NBK198799/. Accessed Mar 14, 2023.

Acknowledgments

- This study was funded by Bristol Myers Squibb and conducted by Evidinno Outcomes Research Inc. S.S., M.D., J.R.M., A.M., and M.K. report employment with Bristol Myers Squibb. E.K., A.Q., L.L., M.M.P. report employment with Evidinno Outcomes Research Inc. J.M.K. is affiliated with the University of Pittsburgh. Authors report no other conflicts of interest.

Presented at ISPOR Europe 2023; November 12-15, 2023; Copenhagen, Denmark

Email: ekasireddy@Evidinno.com

Copies of this poster are for personal use only and may not be reproduced without written permission of the authors.