Cost-effectiveness of mavacamten for the treatment of patients with symptomatic obstructive hypertrophic cardiomyopathy in the Netherlands

Poster number: EE309

Leander Buisman,[1] Maarten Treur,[2] Melissa Verkaik,[1] Gijs Van de Wetering,[2] Linde Michelle Pronk,[2] Marjolein Pompen,[1] Michael Hurst[3]

[1]Bristol Myers Squibb, Utrecht, NL; [2]OPEN Health, Rotterdam, NL; [3]Bristol Myers Squibb, Uxbridge, UK

Introduction

- Hypertrophic cardiomyopathy (HCM) is characterized by primary left ventricular (LV) hypertrophy, with a proportion of patients having obstruction in the outflow tract of the left ventricle (categorized as obstructive HCM)[1-4]
- For patients with obstructive HCM, symptomatic burden can vary significantly (from asymptomatic to breathlessness at rest), and the disease is associated with an increased risk of atrial fibrillation, heart failure, and malignant ventricular arrythmias
- As of 2023, there are no therapies that target the underlying pathophysiology of the disease that are reimbursed in the Netherlands for symptomatic (New York Heart Association [NYHA] class II/III) obstructive HCM
- Mavacamten is a first-in-class, small-molecule, selective allosteric inhibitor of cardiac myosin ATPase that has been developed to target the underlying pathophysiology of HCM, thus reducing contractility and improving myocardial energetics[5]
- Treatment with mavacamten has been shown to lead to improvements in health status and exercise capacity[6] and has recently been incorporated in the treatment pathway in the 2023 European Society of Cardiology (ESC) guidelines[7]

Objectives

- The aim of this work was to present a cost-effectiveness model (CEM) in line with Zorginstituut Nederland (ZIN) health economic guidelines to assess the cost-effectiveness (CE) of mavacamten for the treatment of symptomatic (NYHA class II/III) obstructive HCM in adult Dutch patients

Methods

Model structure

- A de novo CEM was developed to support health technology submissions and was adapted in line with ZIN health economic guidelines[8]
- Mavacamten, in combination with standard of care (SOC) – defined as beta-blockers (BBs)/non-dihydropyridine calcium channel blockers (CCBs) – was compared against SOC alone
- To assess cost-effectiveness, a five-state (NYHA class I–IV and death) Markov model was constructed where patients can transition to any other NYHA health state (or death), or stay in the same health state (**Figure 1**)

Figure 1. Model schematic

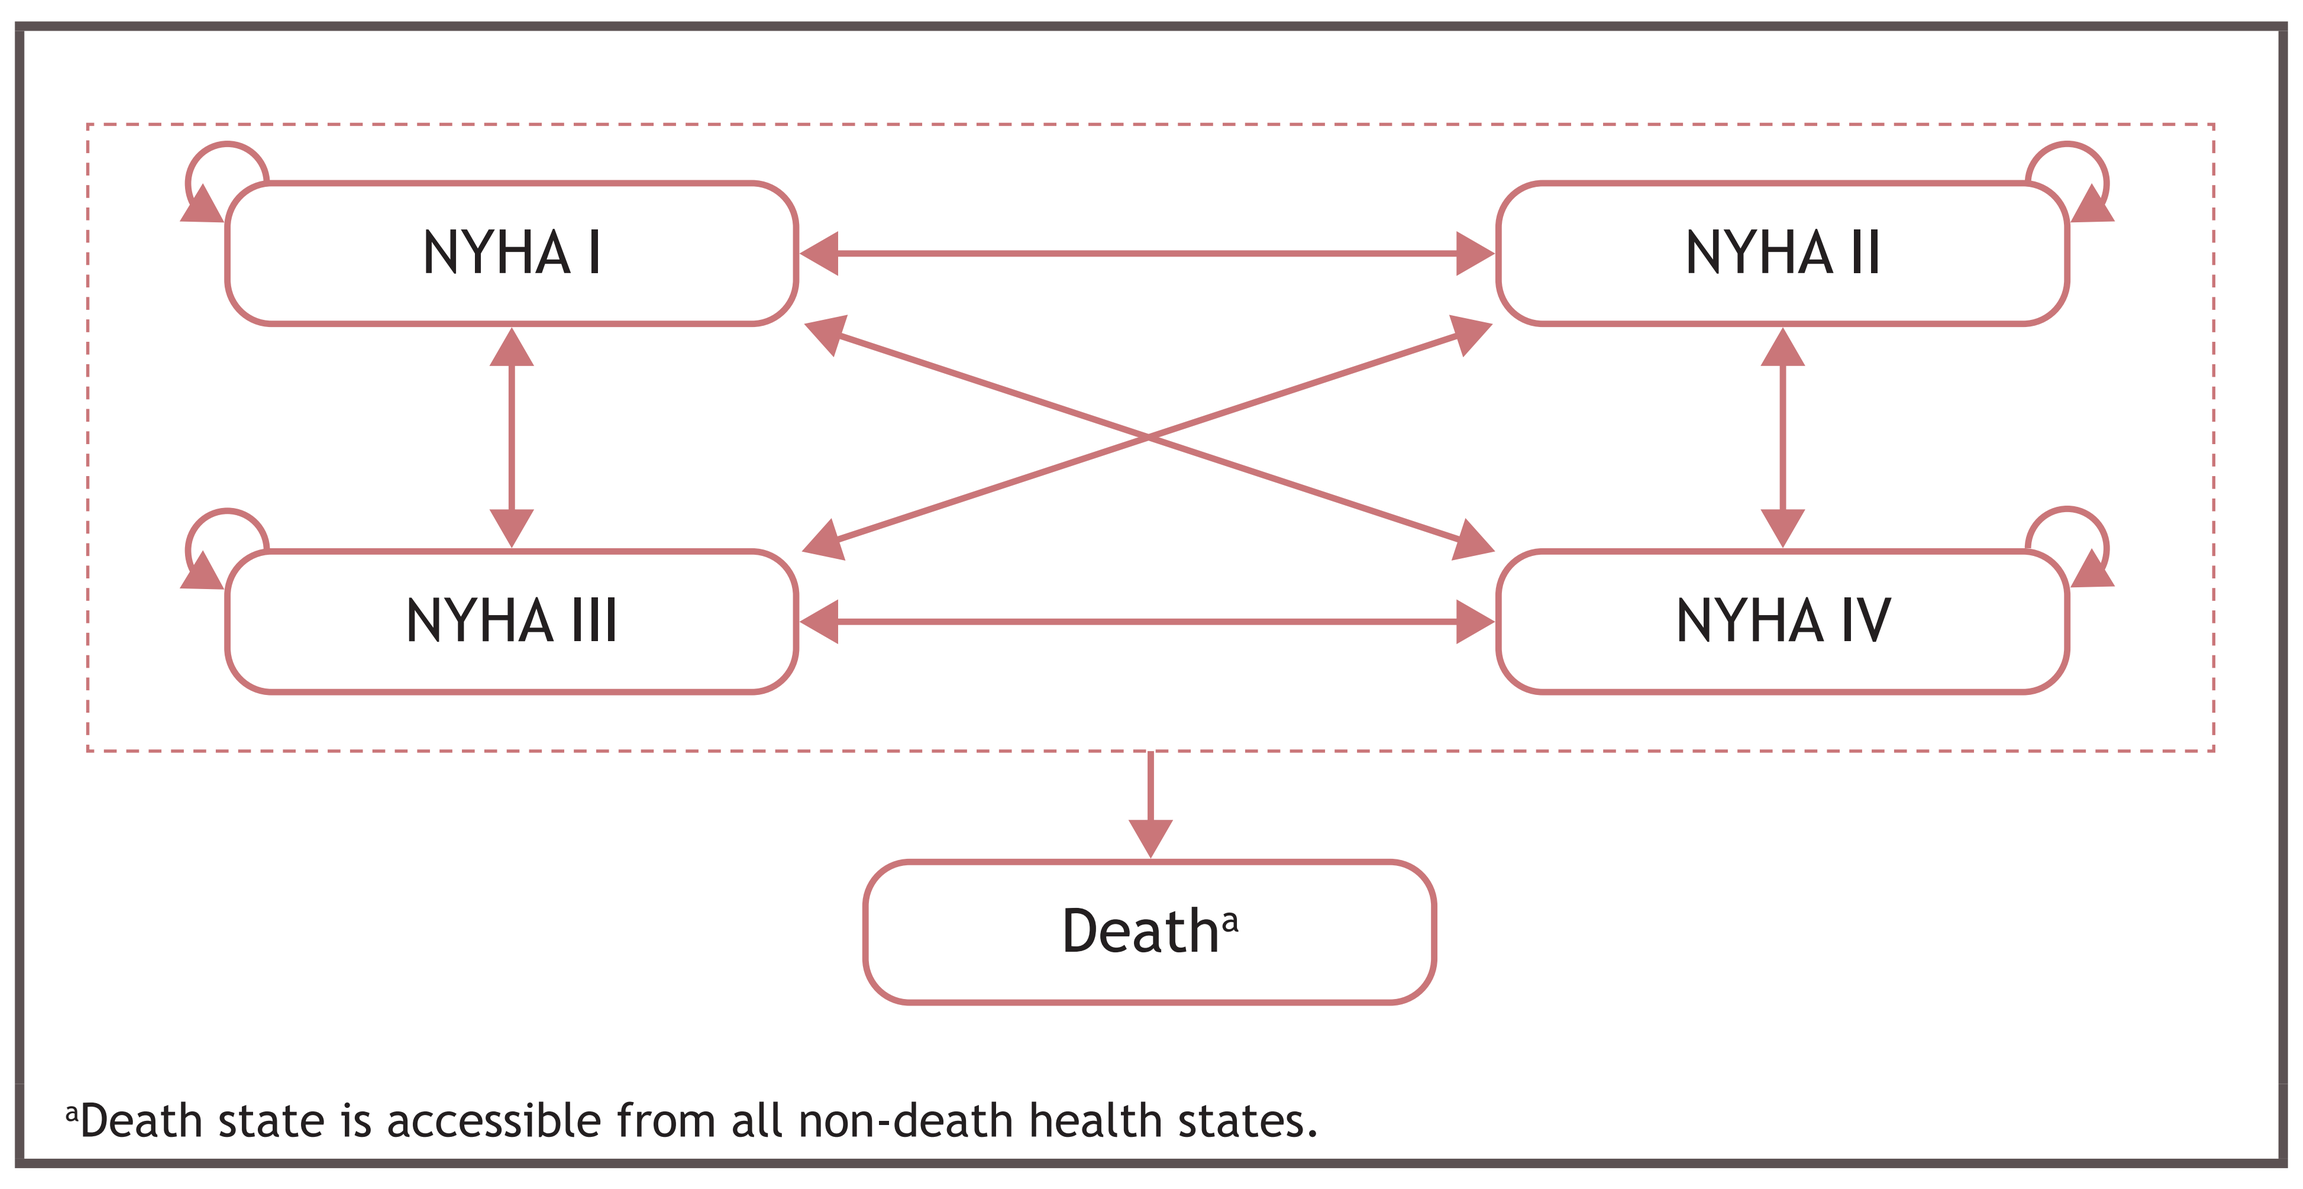
NYHA I
NYHA II
NYHA III
NYHA IV
Death[a]

[a]Death state is accessible from all non-death health states.

- To align with EXPLORER-HCM, cycle length was variable in the first 30 weeks of the model (herein referred to as the short-term), with a subsequent fixed cycle length of 28 days to align with the anticipated dosage of mavacamten
- To account for transitions occuring at times within a cycle, a half-cycle correction was implemented within the economic model
- Model outcomes included incremental costs, quality-adjusted life years (QALYs), life-years (LYs), and CE/utility ratios

Treatment sequencing

- In the CEM, patients either start on mavacamten + SOC or on SOC only, with patients who discontinue mavacamten treatment switching to SOC only. This assumption is supported by Dutch clinical experts[9]
- While in the SOC pathway, the model included treatment sequencing – as suggested by local experts and aligned with the ESC guidelines; including progression to septal reduction therapies – for each treatment arm[7]
- Discontinuation of mavacamten occurs at week 30 if no improvement in NYHA class was observed (validated by Dutch clinical experts), or due to serious adverse event, with rates obtained from EXPLORER-HCM[6]

Model inputs

- The starting patient population was modeled according to the patients who were observed in the EXPLORER-HCM clinical trial. 59.4% of patients were male, with an average age of 59 years, and a NYHA distribution at baseline of 72.9% and 27.1% for NYHA class II and III, respectively[6]
- Progression through and patterns of treatment were informed by transition probabilities based on data from EXPLORER-HCM,[6] and supplemented by literature and expert opinion[9]
- NYHA class deterioration for all treatments was permitted through the modeling of natural disease progression, informed by literature[10,11] and local expert opinion[9]
- Treatment acquisition costs were derived from the 2023 Z-index and hospital diagnosis related group codes (NZa) for pharmacological and surgical interventions, respectively[12]
- Indirect costs encompassing caregiver, travel, and productivity costs (calculated with friction cost methodology) were based on tariffs, prior cardiovascular reimbursement submissions, and local clinical expertise[13]
- Utilities were derived from EXPLORER-HCM based EQ-5D-5L data,[6] applying Dutch tariffs
- The adapted economic model represents the perspective of the Dutch healthcare system, adopting a societal perspective over a lifetime horizon. In line with ZIN guidelines, both costs and effects, respectively, were discounted at 4.0% and 1.5% annually
- The robustness of results was tested with a deterministic sensitivity analysis (DSA) and a probabilistic sensitivity analysis (PSA)

Results

- Mavacamten + SOC was found to be cost-effective compared to SOC at a willingness to pay (WTP) threshold of €20,000/QALY (**Table 1**)

Table 1. Deterministic base case results

Treatment arm	Total costs	Total life years	Total QALYs	ICER (incremental cost/LYs)	ICUR (incremental cost/QALYs)
Mavacamten + SOC	€228,793	16.92	14.88		
SOC only	€179,968	13.59	11.43		
Incremental	€48,825	3.34	3.45	€14,638	€14,170

ICER, incremental cost-effectiveness ratio; ICUR, incremental cost-utility ratio.

- The costs for the mavacamten + SOC arm were mainly driven by drug acquisition costs (€129,397) whereas SOC costs were mainly driven by caregiver costs (€142,986)
- Patients within the mavacamten + SOC arm (vs SOC alone) accumulated the majority of LYs and QALYs in NYHA class I, accumulating 9.18 (vs 1.60) and 8.34 (vs 1.45), respectively
- When adjusting up and down for each parameter individually as part of the DSA, no analyses returned an adjustment in each direction which breached the €20,000/QALY WTP threshold (**Figure 2**)

Figure 2. The impact of individual parameters on the ICUR (cost/QALY) as determined by the DSAs

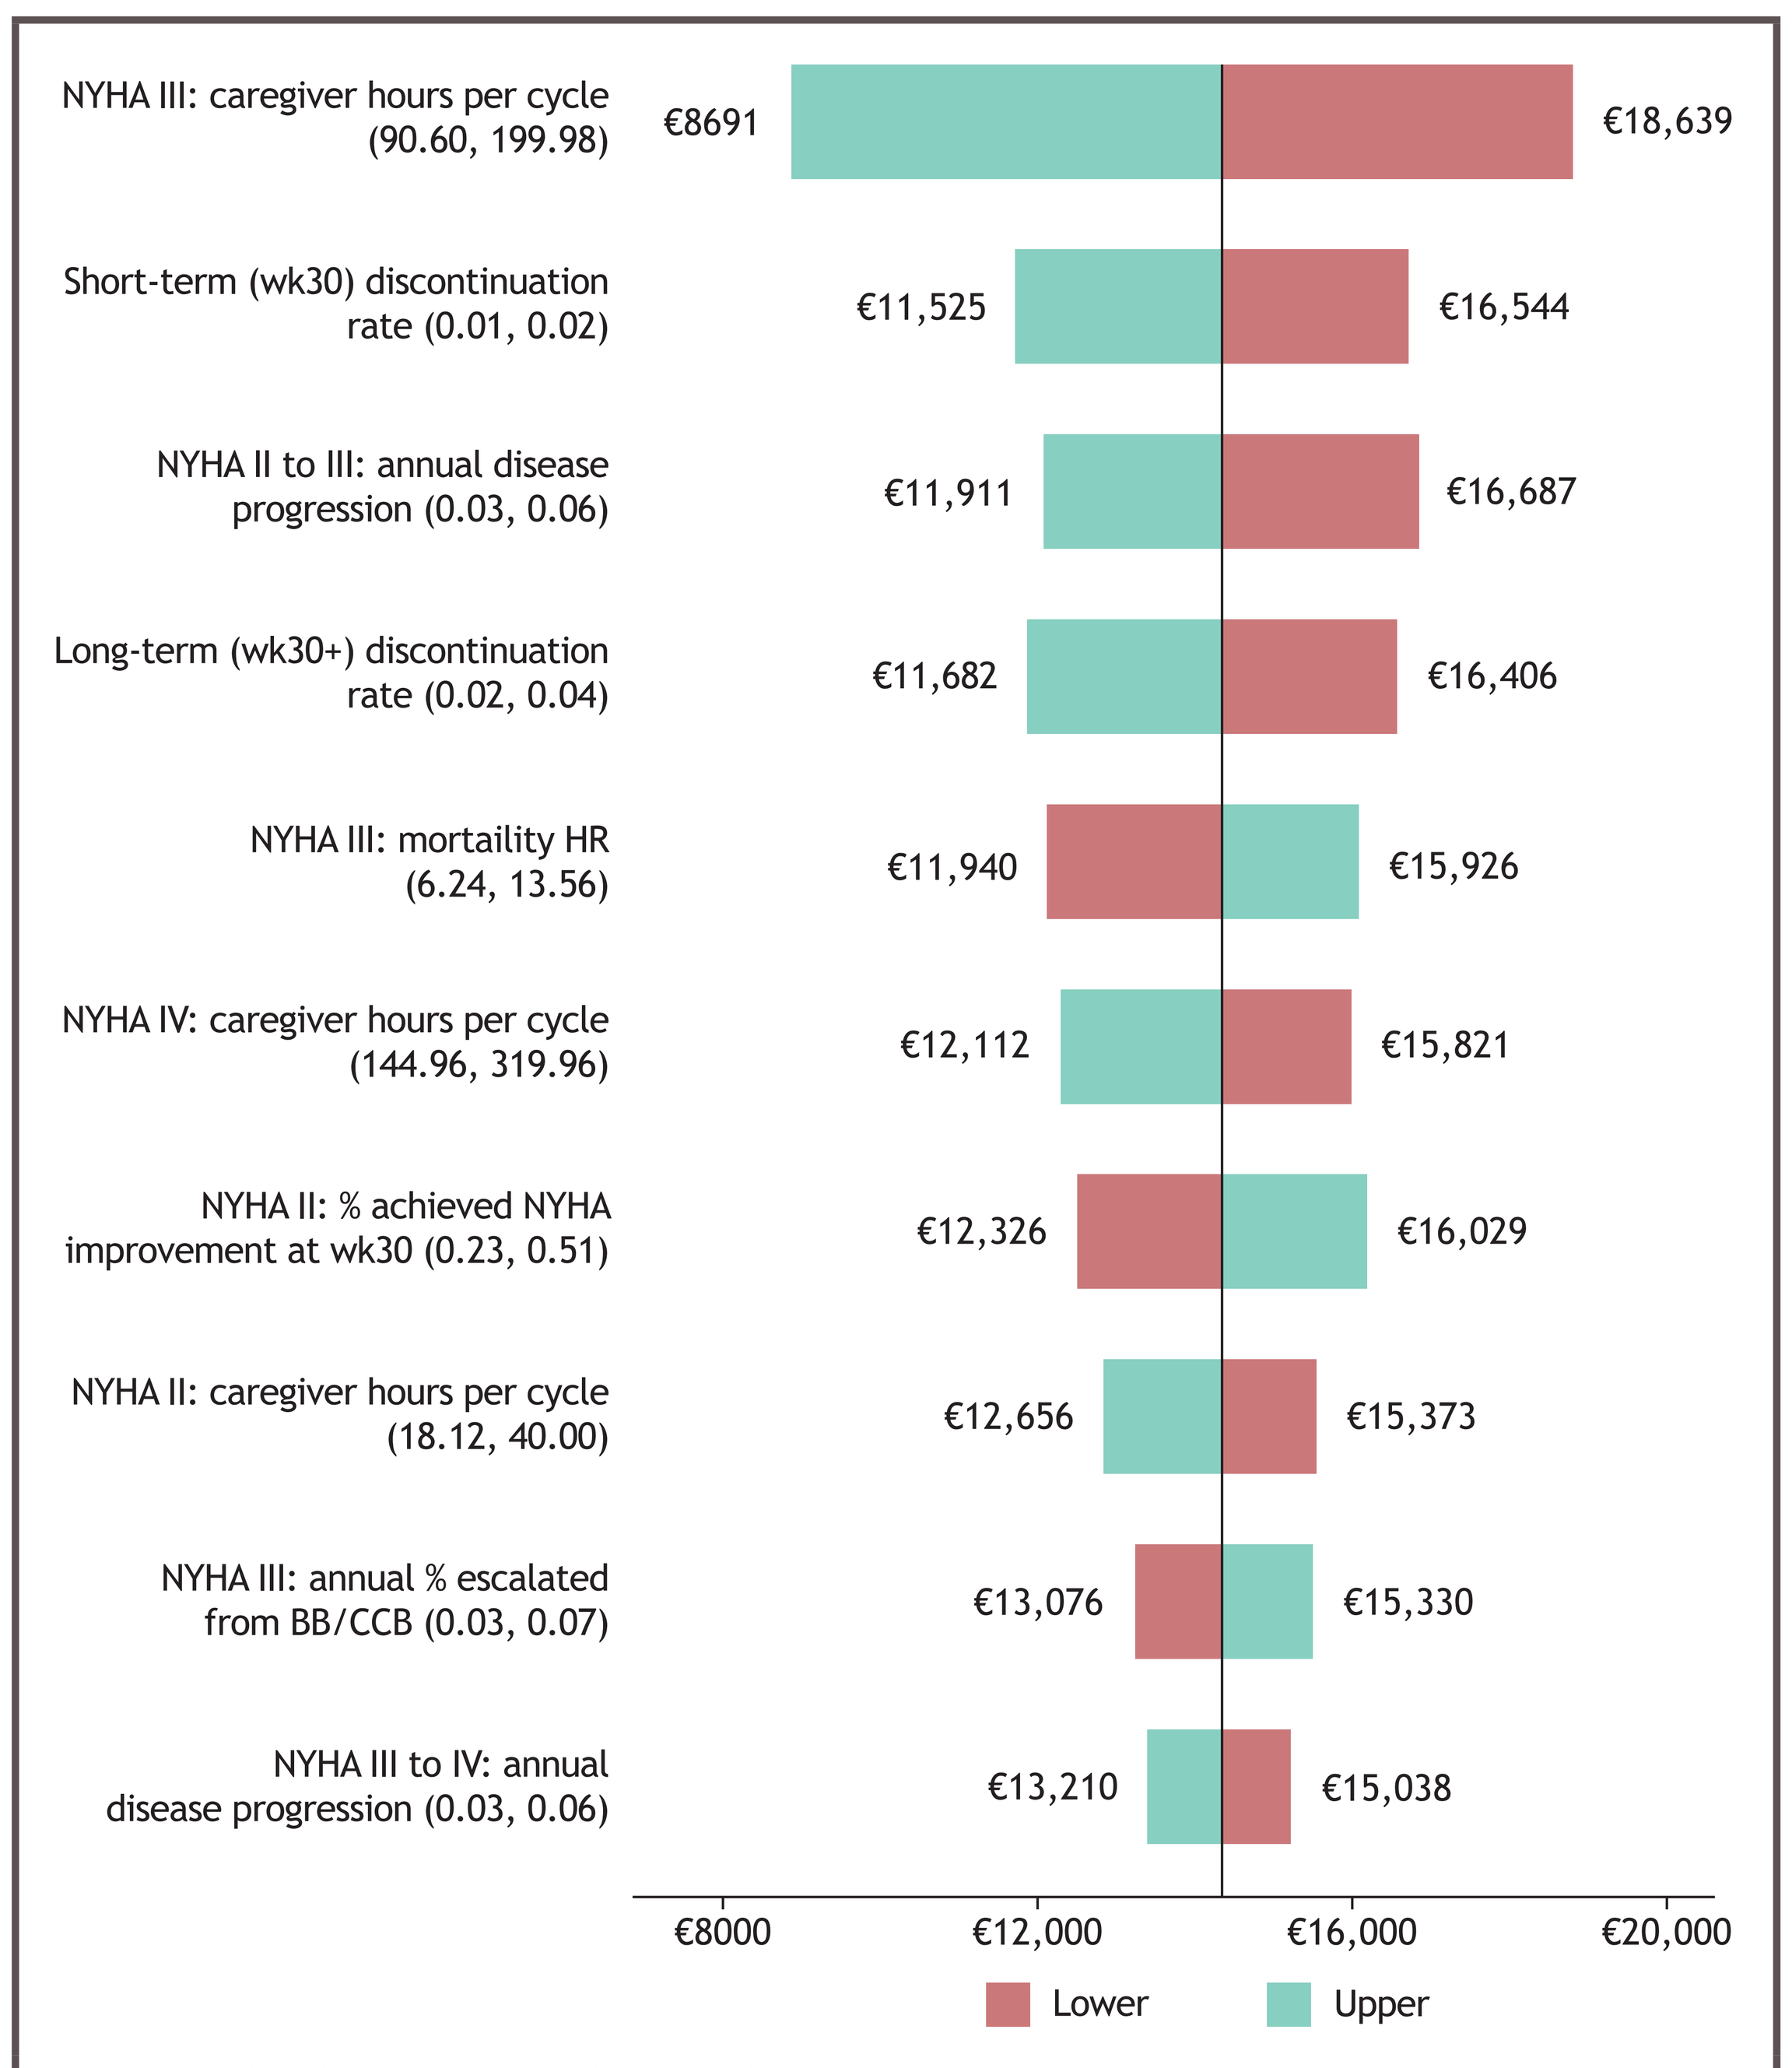
NYHA III: caregiver hours per cycle (90.60, 199.98)
€8691
€18,639
Short-term (wk30) discontinuation rate (0.01, 0.02)
€11,525
€16,544
NYHA II to III: annual disease progression (0.03, 0.06)
€11,911
€16,687
Long-term (wk30+) discontinuation rate (0.02, 0.04)
€11,682
€16,406
NYHA III: mortaility HR (6.24, 13.56)
€11,940
€15,926
NYHA IV: caregiver hours per cycle (144.96, 319.96)
€12,112
€15,821
NYHA II: % achieved NYHA improvement at wk30 (0.23, 0.51)
€12,326
€16,029
NYHA II: caregiver hours per cycle (18.12, 40.00)
€12,656
€15,373
NYHA III: annual % escalated from BB/CCB (0.03, 0.07)
€13,076
€15,330
NYHA III to IV: annual disease progression (0.03, 0.06)
€13,210
€15,038
€8000
€12,000
€16,000
€20,000
Lower
Upper

Numbers in brackets denote the lower and upper parameters, respectively.
HR, hazard ratio; wk, week.

- The greatest impact on the incremental cost–utility ratio (ICUR) observed in the DSA was the number of hours of informal care (per cycle) in NYHA III (base: 145.29; lower: 90.60; upper: 199.98) ranging from ICURs of €18,639 – €8691
- Of all of the model simulations conducted in the PSA, 99.5% fall in the north-eastern quadrant of the CE plane, with 0.5% of simulations falling in the south-eastern quadrant where mavacamten + SOC is dominant (**Figure 3**)
- The probability for each treatment to be cost-effective over a range of WTPs is visualized in the cost-effectiveness acceptability curve (CEAC) (**Figure 4**)
- The CEAC calculated that the probability that mavacamten + SOC is cost-effective (vs SOC) at a WTP threshold of €20,000, €50,000 and €80,000/QALY is 86.4%, 100.0% and 100.0% respectively
- Where simulations fell into the north-east quadrant (increased costs and QALYs), ICURs within the PSA ranged between €1950/QALY and €37,464/QALY

Figure 3. Scatter plot from the PSA for mavacamten + SOC versus SOC only treatment

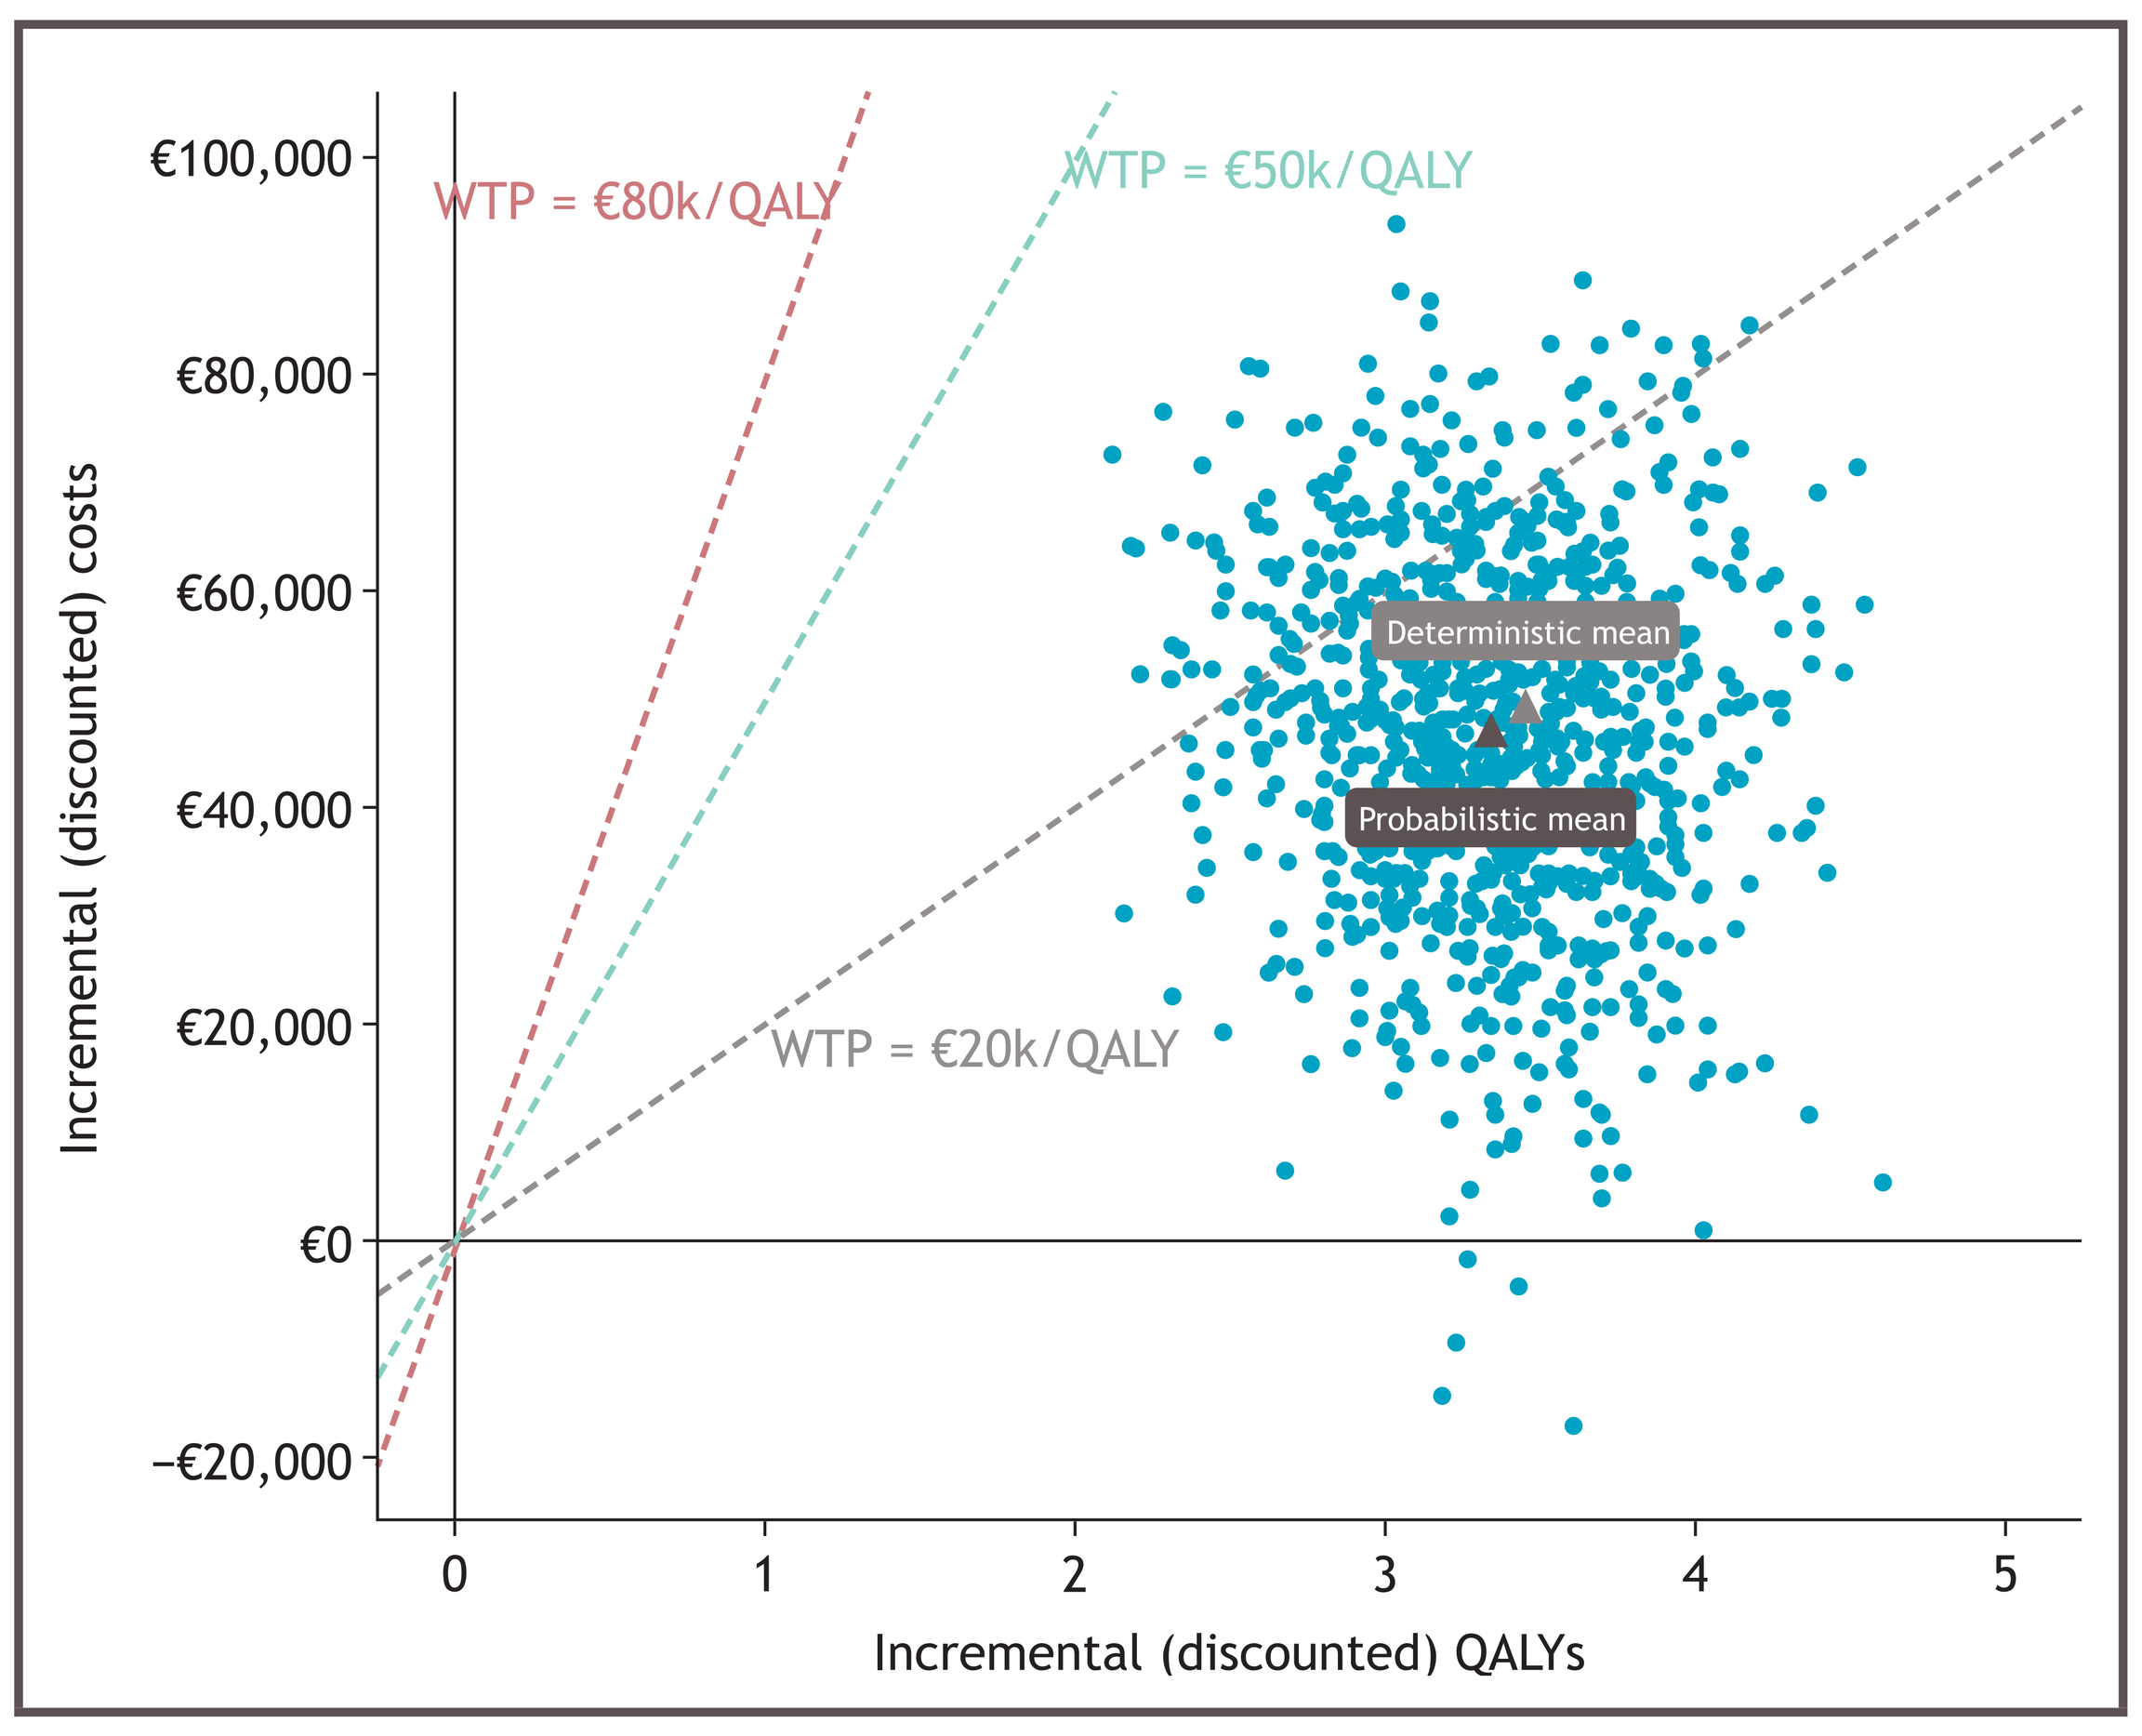
WTP = €80k/QALY
WTP = €50k/QALY
WTP = €20k/QALY
Deterministic mean
Probabilistic mean
Incremental (discounted) costs
€100,000
€80,000
€60,000
€40,000
€20,000
€0
−€20,000
0
1
2
3
4
5
Incremental (discounted) QALYs

Figure 4. CEAC for mavacamten + SOC versus SOC only treatment

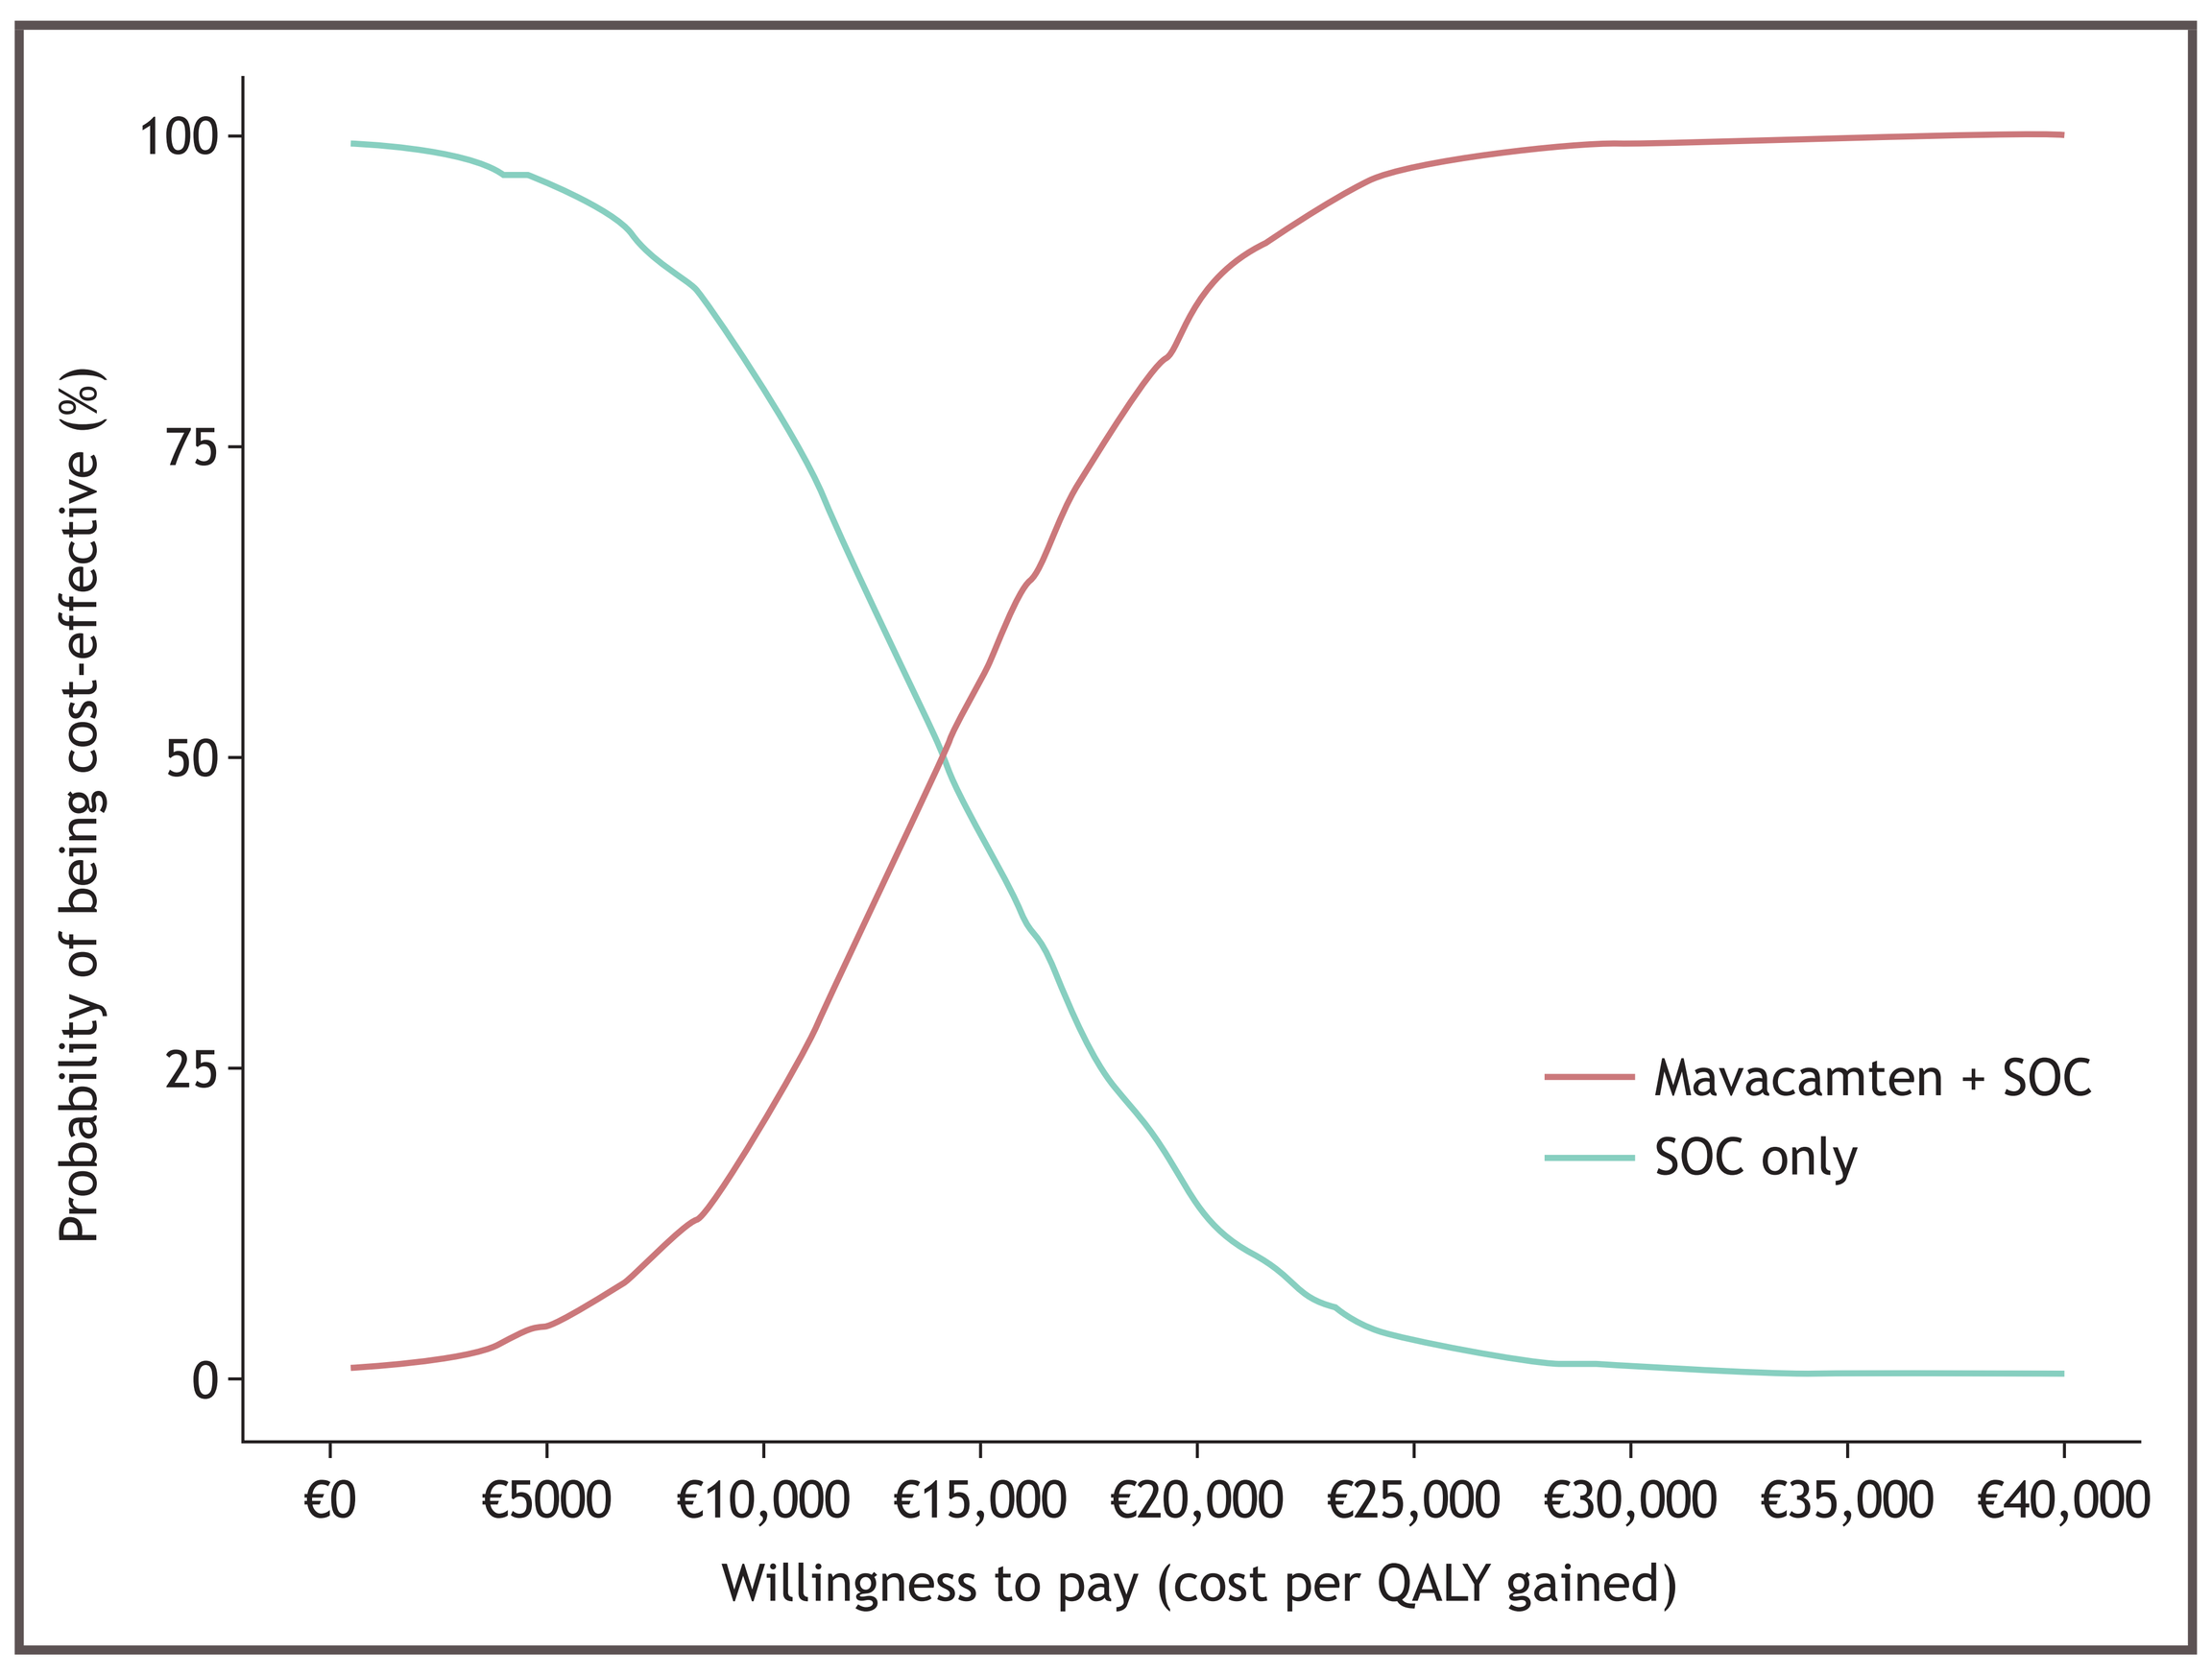
Probability of being cost-effective (%)
100
75
50
25
0
€0
€5000
€10,000
€15,000
€20,000
€25,000
€30,000
€35,000
€40,000
Willingness to pay (cost per QALY gained)
Mavacamten + SOC
SOC only

Limitations

- Where data were scarce, model assumptions were kept conservative and they have been informed by published literature or clinical expert opinion
 - The model does not assume additional long-term benefits over and above that observed in the short-term from treatment with mavacamten, where data from the MAVA-LTE long-term extension trial did show additional benefit beyond week 30[14,15]

Conclusions

- At a WTP of €20,000 per QALY gained, the CEM demonstrated that mavacamten added to SOC (vs SOC alone) is a cost-effective treatment for adult patients with symptomatic (NYHA class II and III) obstructive HCM in the Netherlands
- The impact of assumptions made in the model have been evaluated in sensitivity analyses where the ICUR remained below the WTP threshold of €20,000/QALY for most analyses
- Within the model, the introduction of mavacamten has a significant impact on the reduction of social care costs, showing a considerable indirect benefit economically but also addressing unmet needs for both patients and carers
- These findings support a recommendation based on ZIN health-economic guidelines

References

1. Maron BJ, et al. *J Am Coll Cardiol* 2014;64:83–99.
2. Priori SG, et al. *Eur Heart J* 2015;36:2793–2867.
3. Semsarian C, et al. *J Am Coll Cardiol* 2015;65:1249–1254.
4. Royal Brompton & Harefield Hospitals Specialist Care. https://www.rbhh-specialistcare.co.uk/news/our-new-hypertrophic-cardiomyopathy-service. Accessed September 6, 2023.
5. Lim GB. *Nat Rev Cardiol* 2020;17:677.
6. Olivotto I, et al. *Lancet* 2020;396:759–769.
7. Arbelo E, et al. *Eur Heart J* 2023;44:3503–3626.
8. Zorginstituut Nederland. https://english.zorginstituutnederland.nl/publications/reports/2016/06/16/guideline-for-economic-evaluations-in-healthcare. Accessed September 6, 2023.
9. Bristol Myers Squibb. Expert Meeting: Mavacamten in obstructieve hypertrofische cardiomyopathie in Nederland. April 25, 2022. Data on file.
10. Maron BJ, et al. *JACC Heart Fail* 2018;6:353–363.
11. Maron MS, et al. *J Am Coll Cardiol* 2016;67:1399–1409.
12. Nederlandse Zorgautoriteit. https://www.opendisdata.nl. Accessed September 14, 2023.
13. Zorginstituut Nederland. https://www.zorginstituutnederland.nl/publicatiespublicatie/2016/02 /29/richtlijn-voor-het-uitvoeren-van-economische-evaluaties-in-de-gezondheidszorg. Accessed September 14, 2023.
14. Rader F, et al. *J Am Coll Cardiol* 2021;77(18 Supplement_1):532.
15. Jacoby DL, et al. *Circulation* 2021;144:A10201.

Acknowledgments

- The authors would like to acknowledge the contributions of the following:
 - Manikanta Dasari and Varun Ektare from OPEN Health, and Kasper Johannesen, Marta Contente, Taryn Krause, Yuanhui Zhang and Xiaoyan Li from Bristol Myers Squibb, for their contributions towards the strategic insights of the modeling methodology.
 - Zoe Cheah and Melissa Ho from OPEN Health for their support in developing various materials relating to this poster.
- The study was supported by Bristol Myers Squibb. All authors contributed to and approved the poster; editorial assistance was provided by Kate Ward MBiochem and Grant Womack of Oxford PharmaGenesis, Oxford, UK, funded by Bristol Myers Squibb.

Disclosures

- LB, MV, MP and MH are employees of Bristol Myers Squibb and may own Bristol Myers Squibb stock or stock options. MT, GVdW and LMP are employees of OPEN Health, which has received consultancy fees from Bristol Myers Squibb. The authors declare having no competing interests beyond those reported.

Presented at ISPOR Europe 2023; November 12-15, 2023; Copenhagen, Denmark

Correspondence to: Leander Buisman; Email: Leander.Buisman@bms.com

Copies of this poster are for personal use only and may not be reproduced without written permission of the authors