CO142

A retrospective study characterizing age at key clinical disease indicators among patients with Friedreich Ataxia using health administrative claims data in the United States

A Salvucci, DO;[1] C Qian, MSc;[2] L Powell, MPH;[2] D Lynch, MD PhD;[3] G Vasco, MD, PhD;[4] K Johnston, PhD;[2] I Tomazos, PhD MBA[1]

[1]PTC Therapeutics, South Plainfield, NJ, USA; [2]Broadstreet HEOR, Vancouver, BC, Canada; [3]Division of Neurology, CHOP, Phila, PA; [4]Department of Neurorehabilitation and Robotics, Bambino Gesù Children's Hospital, IRCCS, Rome, Italy

1. Background & Objectives:

- **Friedreich ataxia (FA)** is a progressive and degenerative neuromuscular disorder, typically characterized by muscle weakness, worsening ataxia and scoliosis, resulting in loss of ambulation (LOA).[1] In Europe, FA prevalence ranges from ~1:20,000 live births in the southwest to 1 in 250,000 in the north and east.[1]
- Speech is affected, and many patients experience loss of vision and hearing. Cardiomyopathy and diabetes mellitus are also common and serious manifestations of the disease.
- FA management focuses on treatment of symptoms, secondary complications due to FA, and comorbidities, through pharmacotherapy, supportive aids, surgery, and physical therapy.[2,3]
- Currently, there is only one treatment that has been licensed specifically for FA (omaveloxolone, approved in 2023 by the FDA). However, its indication in the United States (US) is restricted to patients with FA who are aged 16 years and older.

This study aimed to characterize the age at key disease severity indicators (scoliosis, cardiomyopathy, and diabetes) among a cohort of patients with FA, using retrospective US claims data.

2. Methods:

- Data source: US Merative MarketScan Commercial database (Aug 2010 to Sept 2020).
- Inclusion criteria: Children and adults with ≥12 months of follow-up and ≥1 inpatient, or ≥2 outpatient visits separated by ≥30 days, with primary or secondary diagnosis of FA (ICD-9 334.0) prior to Oct 2015.* Index visit was defined as 1st inpatient or 2nd outpatient visit with diagnosis of FA.
- All incident patients with FA (ICD-9 334.0) prior to Oct 2015 were identified.
- Incidence was defined with an 18-month washout period with no FA diagnostic codes. Included patients were followed up for 12 months (**Figure 1**).
- This 30-month period was used to observe the age at onset of various disease indicators (cardiomyopathy, scoliosis, and diabetes), through the presence of ≥1 diagnostic, medication, or procedural code for these disorders.
- Age at index date was used as a proxy for age at FA diagnosis. Results were stratified into two categories: <16 years; 16-24 years at FA diagnosis.

*334.0 was replaced by a non-specific ICD-10 code in September 2015

Figure 1. Defining index date, washout period, and 12-month follow-up period in the analysis

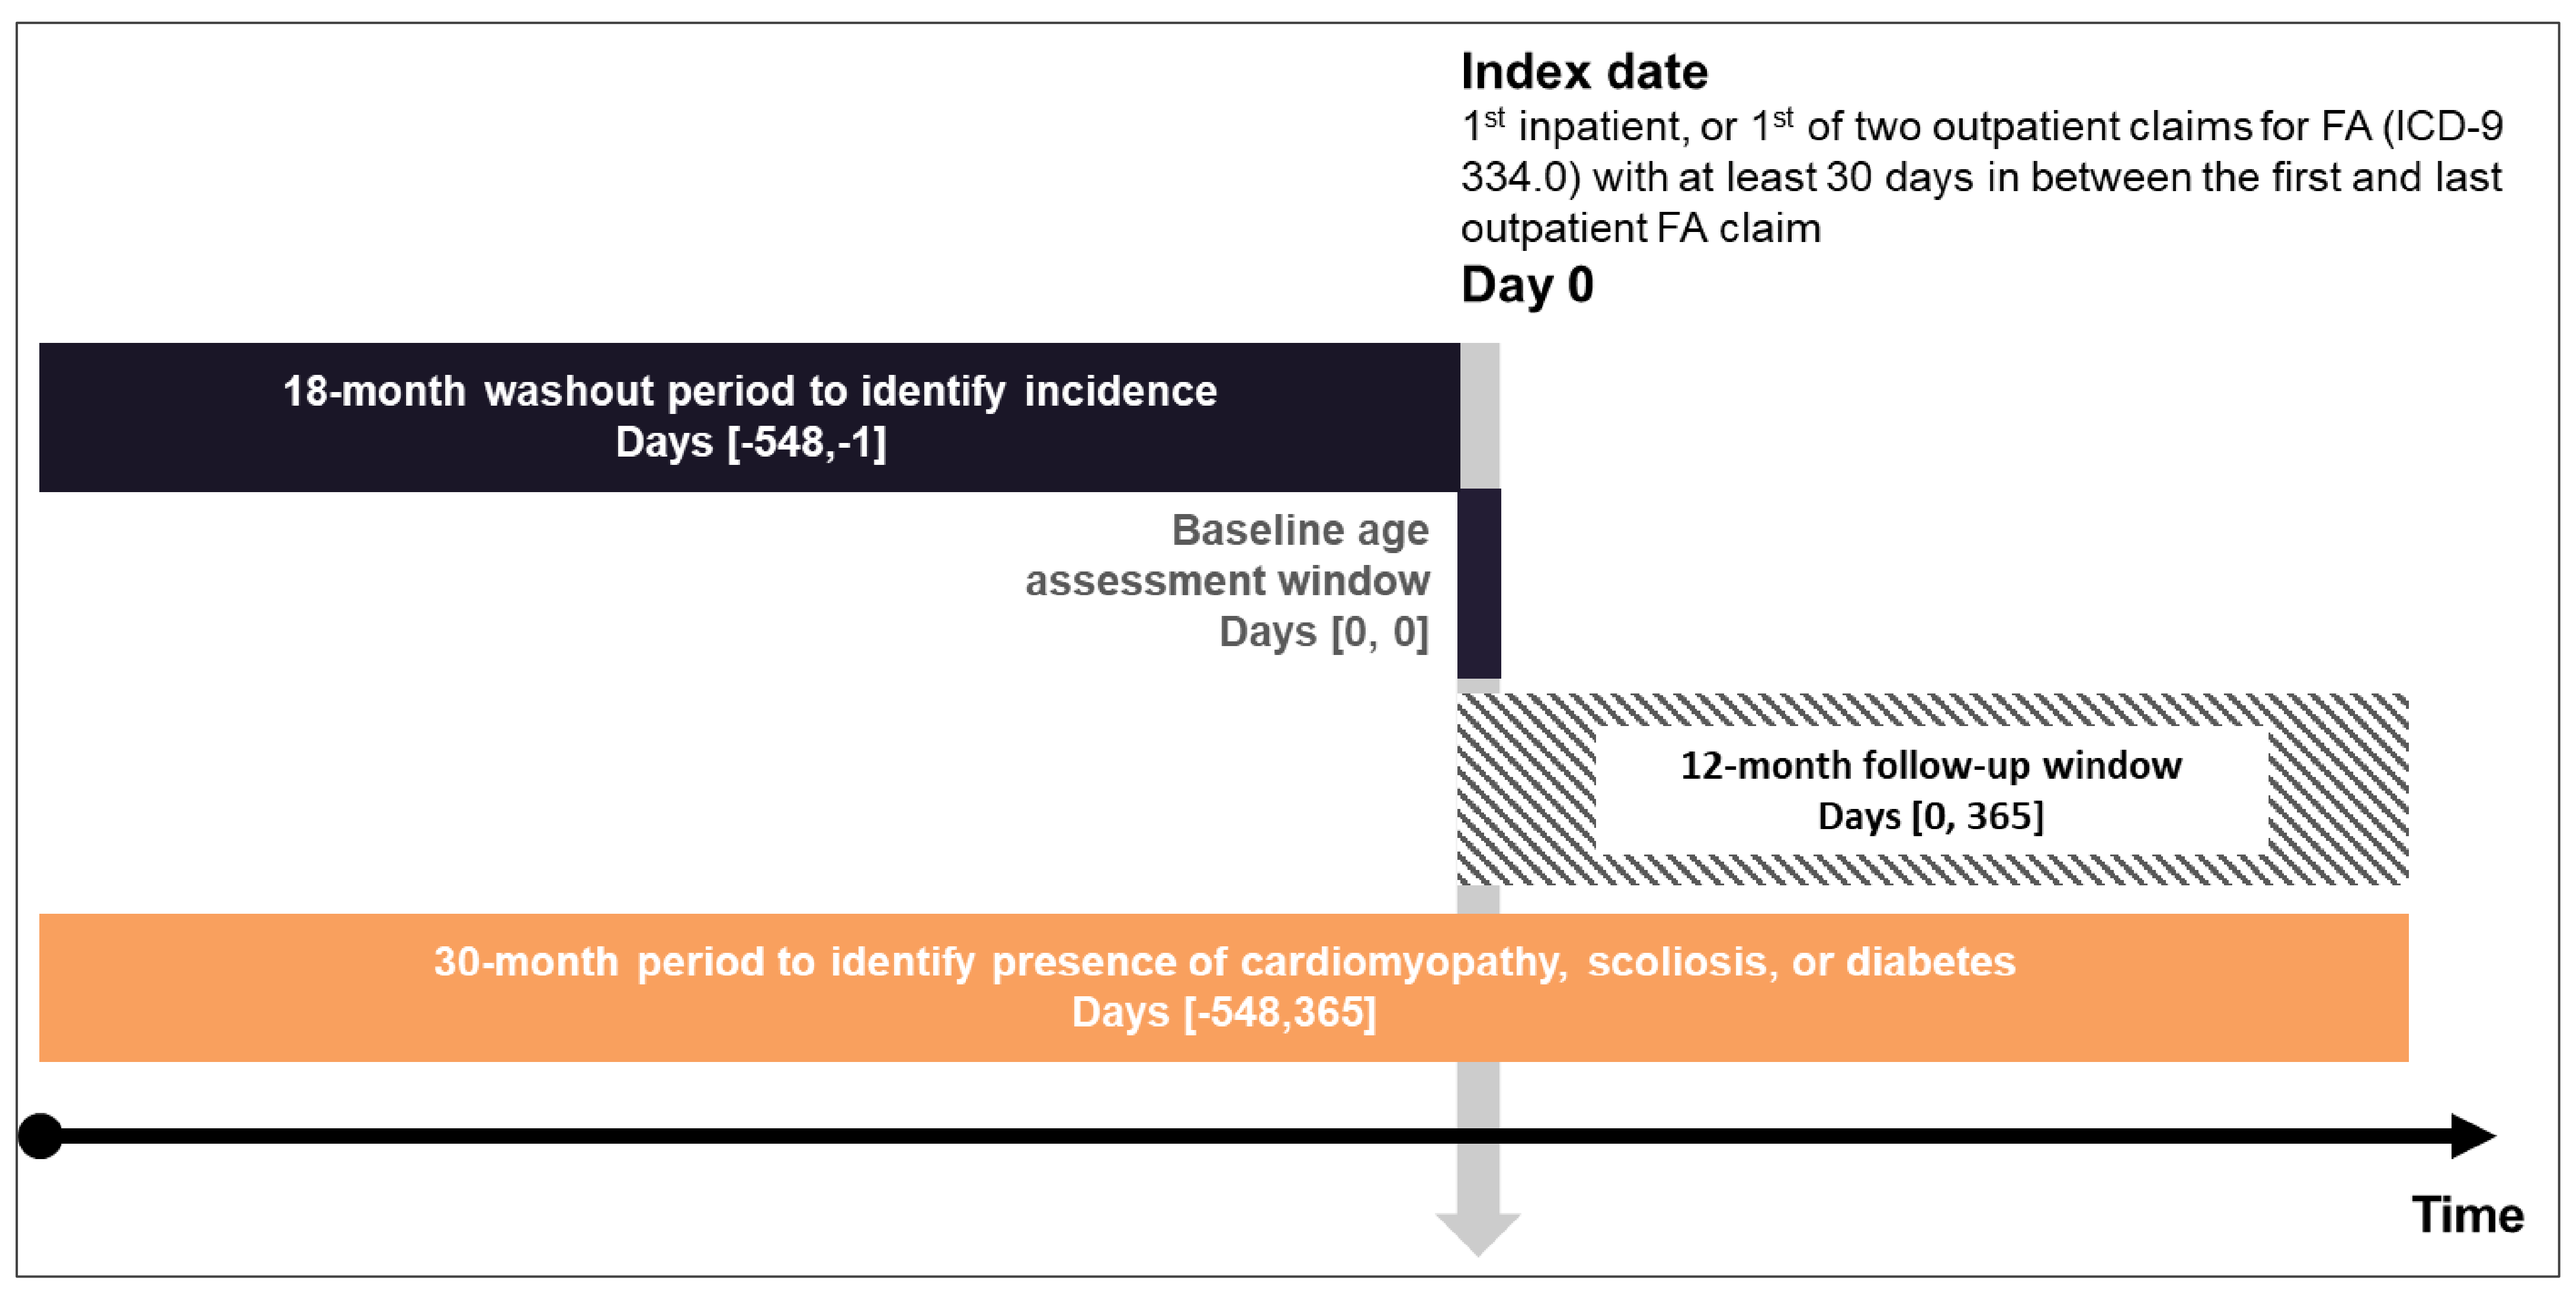
Index date
1st inpatient, or 1st of two outpatient claims for FA (ICD-9 334.0) with at least 30 days in between the first and last outpatient FA claim
Day 0
18-month washout period to identify incidence
Days [-548,-1]
Baseline age assessment window
Days [0, 0]
12-month follow-up window
Days [0, 365]
30-month period to identify presence of cardiomyopathy, scoliosis, or diabetes
Days [-548,365]
Time

Abbreviations: FA = Friedreich ataxia; ICD = International Classification of Diseases

3. Results cont'd:

- 269 incident patients with FA were identified (**Figure 2**):
 - 43 (16.0%) were diagnosed <16 years (mean [range] age at diagnosis of 10.9 [1-15]) years
 - 34 (12.9%) between 16-24 years (20.2 [16-24]) years
 - 192 (71.4%) were >24 years (46.1 [25-64]) years.
- Within the 30-month period leading up to and following the initial FA diagnosis, the majority of the earlier onset patients had scoliosis (72.1% among those <16; 67.6% among 16-24; 13.5% among >24 years), with the mean (range) age being 10.6 (1.0-14.7), 18.3 (12.6-24.0), and 41.2 (23.0-61.0) years at first captured incidence, respectively (**Figure 3**, **Table 1**).
- Those who had corrective surgery were also mostly among the <16 age group (11.6% among those <16; 2.9% among those 16-24; 0.5% among those >24 years), with the mean (range) age being 13.1 (12.0-14.3) years at first captured incidence among those <16; age estimates for the other two age groups are not provided due to n=1.
- Cardiomyopathy was most frequently observed among those diagnosed between the ages of 16-24 years (50.0%), followed by those <16 (37.2%) and >24 years (27.1%); with age estimates of 18.8 (12.4-23.5), 10.8 (7.1-14.4), and 39.3 (23.8-62.5) years, respectively (**Figure 3**, **Table 1**).
- Diabetes was not frequently observed among the earlier onset patients (7.0% among those <16; 8.8% among 16-24; 28.1% among >24 years), with age estimates of 8.8 (2.6-12.7), 20.5 (20.2-21.2), 47.4 (21.5-62.2) years, respectively (**Figure 3**, **Table 1**).
- For the youngest age at diagnosis group (<16 years), age at time of key clinical milestones is visualized in **Figure 4**. Scoliosis and cardiomyopathy frequently occurred in childhood, prior to age 16.

Table 1. Proportion of FA patients with each of the disease severity indicators (scoliosis, cardiomyopathy, or diabetes) and age at first indicator during 30-month follow up window

Disease severity indicator	Age at first indicator, years		
	<16 (n=43)	16-24 (n=34)	>24 (n=192)
Scoliosis, n (%)	31 (72.1%)	23 (67.6%)	26 (13.5%)
Age at scoliosis, mean (SD)	10.6 (3.6)	18.3 (3.4)	41.2 (12.6)
Age at scoliosis, median (min-max)	11.6 (1.0-14.7)	17.3 (12.6-24.0)	39.8 (23.0-61.0)
Diabetes, n (%)	3 (7.0%)	3 (8.8%)	54 (28.1%)
Age at diabetes, mean (SD)	8.8 (5.4)	20.5 (0.6)	47.4 (12.2)
Age at diabetes, median (min-max)	11.0 (2.6-12.7)	20.2 (20.2-21.2)	50.5 (21.5-62.2)
Cardiomyopathy, n (%)	16 (37.2%)	17 (50.0%)	52 (27.1%)
Age at cardiomyopathy, mean (SD)	10.8 (2.2)	18.8 (3.1)	39.3 (12.2)
Age at cardiomyopathy, median (min-max)	10.9 (7.1-14.4)	20.4 (12.4-23.5)	36.2 (23.8-62.5)

Abbreviations: FA = Friedreich ataxia; SD = standard deviation

3. Results:

Figure 2. Distribution of age at FA diagnosis for incident patients with FA in the MarketScan commercial dataset

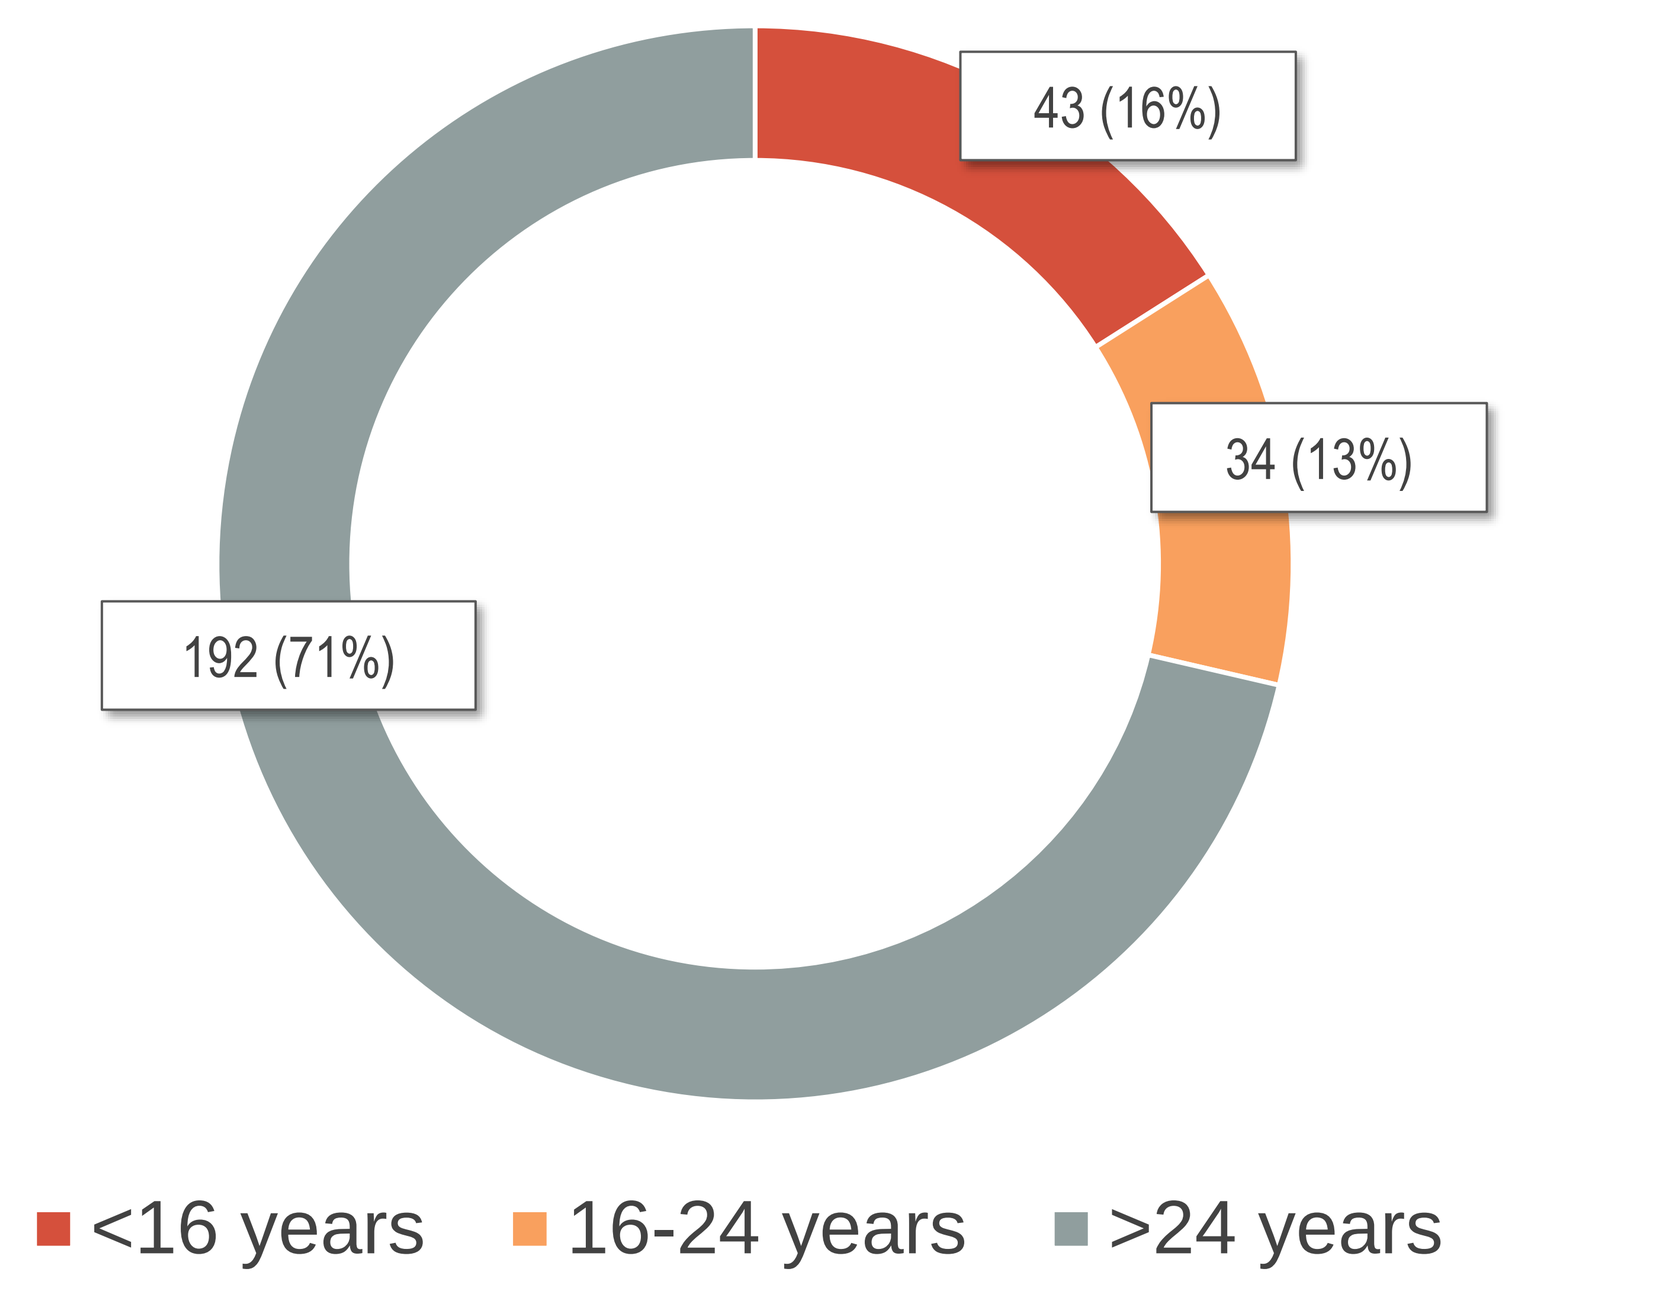
43 (16%)
34 (13%)
192 (71%)
<16 years
16-24 years
>24 years

Abbreviations: FA = Friedreich ataxia

Figure 3. Proportion of FA patients with each of the disease severity indicators (scoliosis, cardiomyopathy, or diabetes), by age at diagnosis (years)

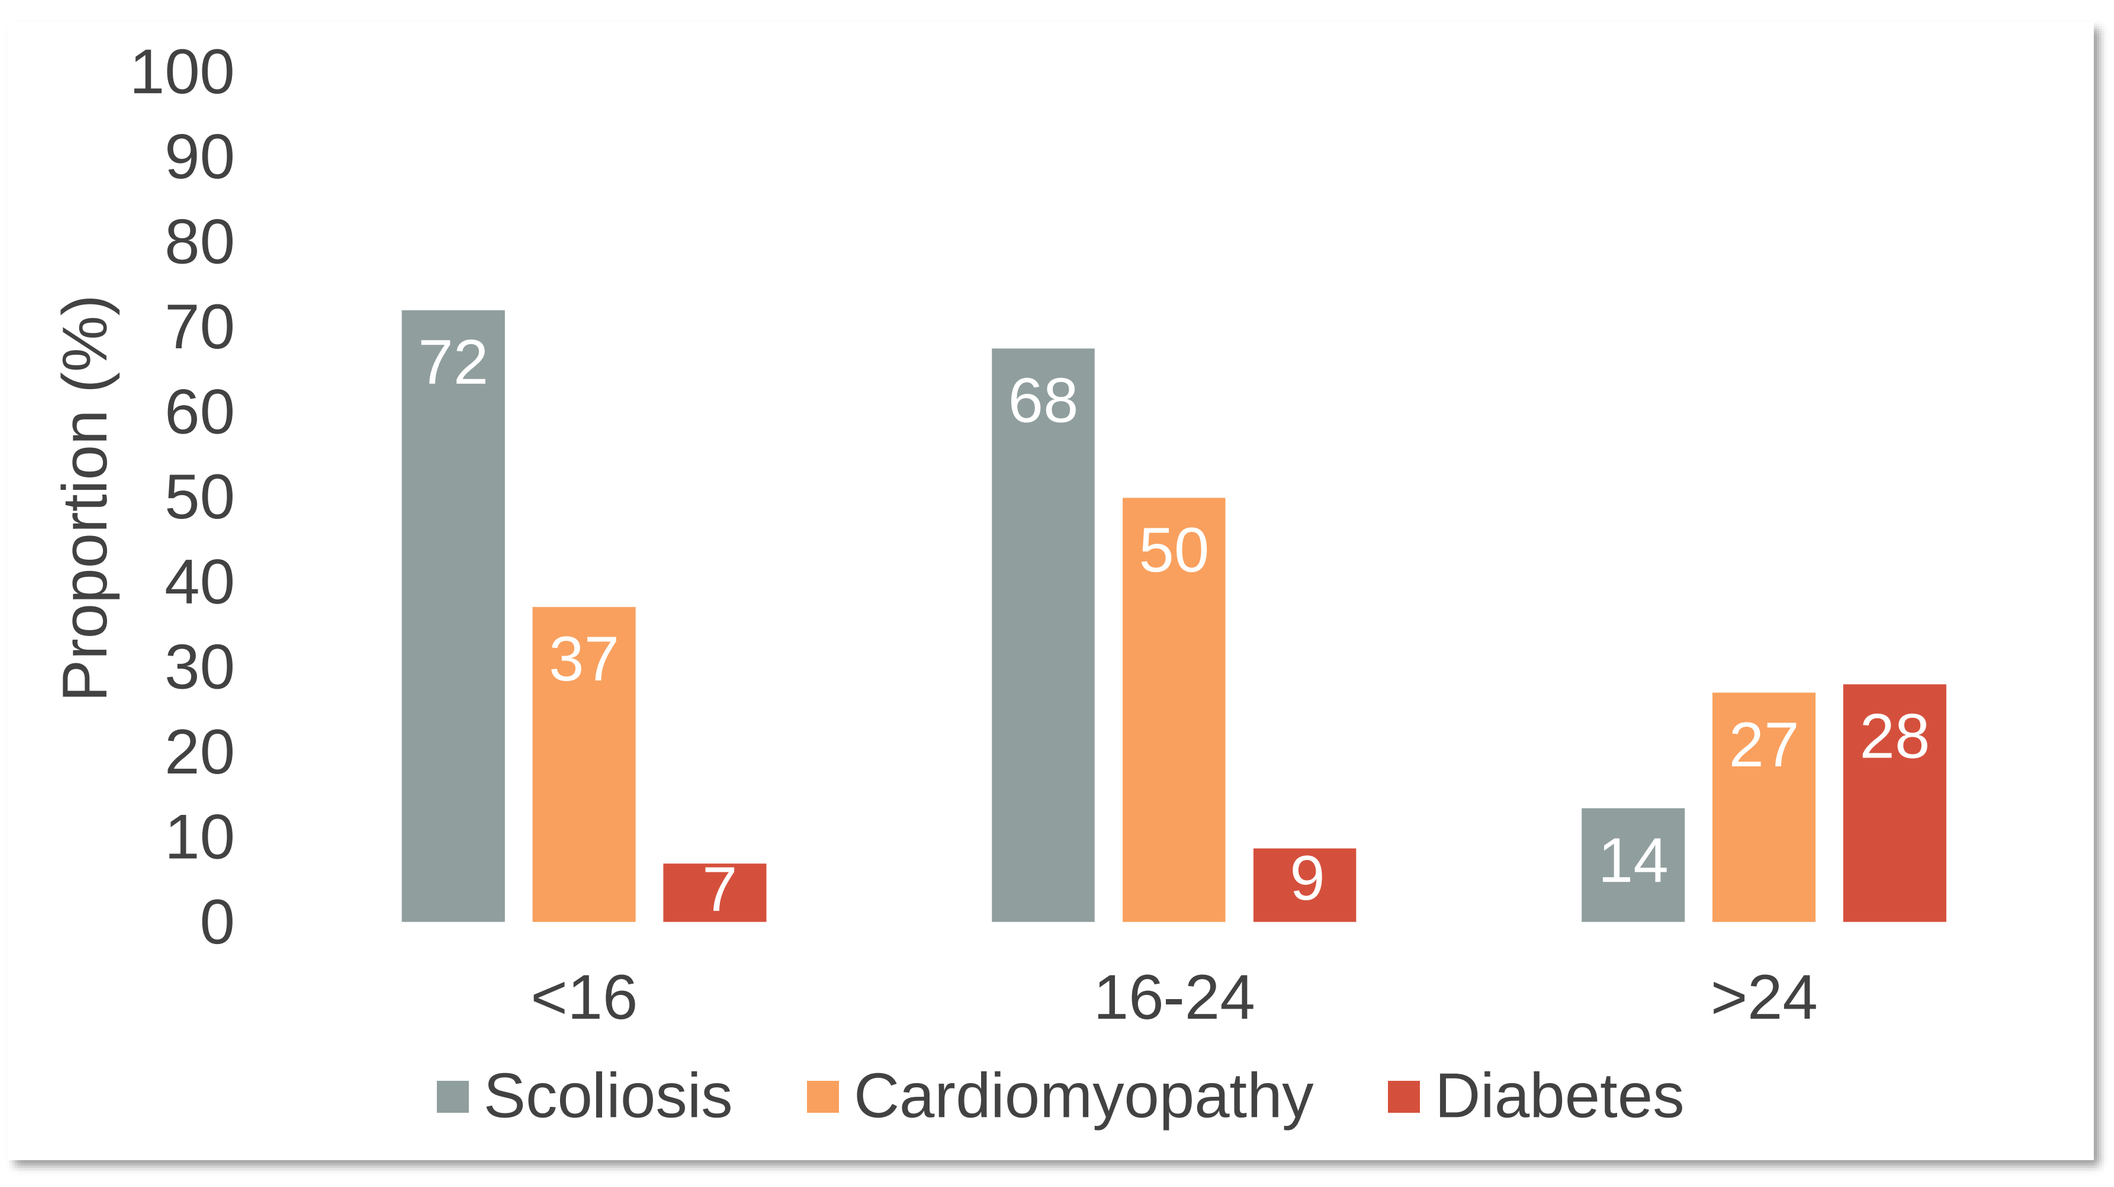
Proportion (%)
100
90
80
70
60
50
40
30
20
10
0
72
37
7
68
50
9
14
27
28
<16
16-24
>24
Scoliosis
Cardiomyopathy
Diabetes

Figure 4. Incident cohort timing of disease severity indicators during follow-up (<16-year incident cohort subgroup)

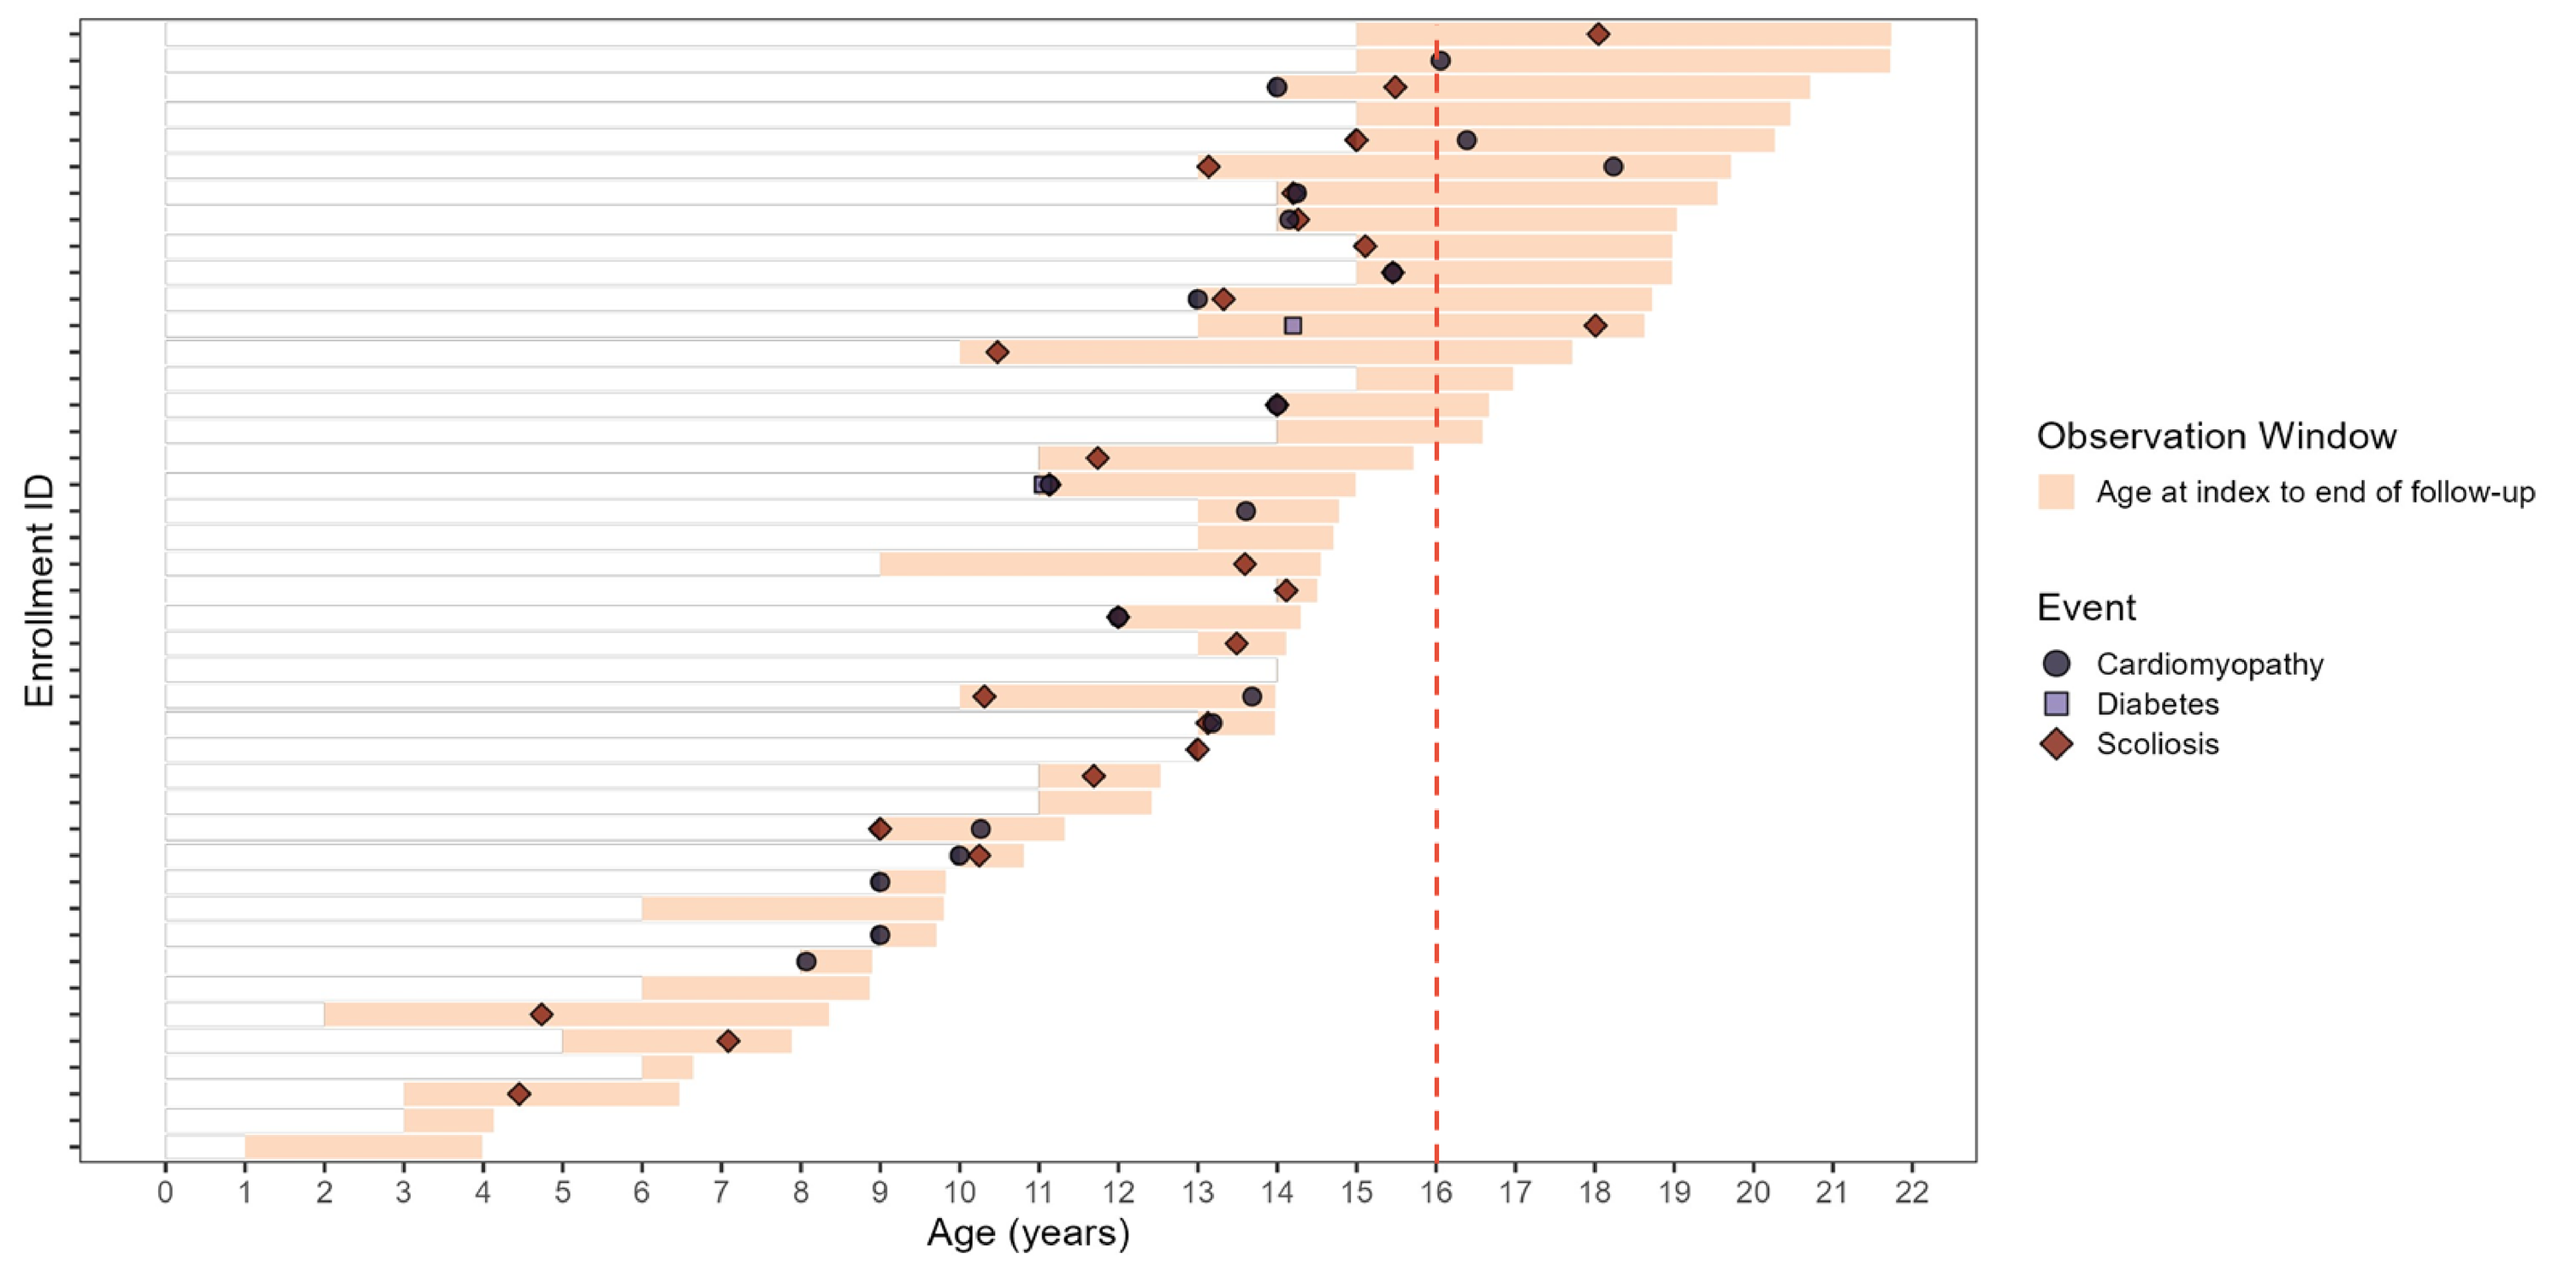
Enrollment ID
Age (years)
0 1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20 21 22
Observation Window
Age at index to end of follow-up
Event
Cardiomyopathy
Diabetes
Scoliosis

4. Discussion & Conclusions:

- In this cross-sectional analysis, cardiomyopathy and scoliosis were observed in patients with FA starting in childhood and adolescence, with a substantial number of patients experiencing cardiomyopathy and scoliosis prior to the age of 16 years.
- To slow progression of cardiomyopathy and scoliosis, initiation of disease modifying treatment at a young age is critical.
- Diabetes was observed more frequently in the older age at onset group (>24 years) and was less frequent among the younger patients.
- This study confirms that a significant number of patients with FA are affected by scoliosis and cardiomyopathy at a young age. There is need for disease modifying treatments that can be initiated at a young age and effectively delay the progression of FA.

This study confirms that a significant number of patients with FA are affected by scoliosis and cardiomyopathy at a young age (<16 years). There is a need for disease modifying treatments that can be initiated at a young age and effectively delay the progression of FA.

5. Limitations:

- This study used a large, real-world, US dataset which allowed for identification of a substantial number of patients with FA for robust analyses.
- Nevertheless, some limitations of this study included:
 - Given that the data source is composed of patients on commercial insurance, patients with FA with later-onset disease may be over-represented in the cohort, as observed through the larger proportion of patients between the ages of 16-24 years at index FA diagnosis; however, limitations may exist in ascertaining incidence with a washout period of eighteen months, and therefore the representativeness of this cohort will need to be better assessed upon comparison to real-world studies with longitudinal data.
 - This may have led to under-representation of the early-onset, most severe patients with FA, and underestimation of the frequency of key disease indicators.
 - Due to the lack of a specific ICD-10 code for FA between September 2015 and September 2020, the results from this study are limited to a cohort of patients identified in or before 2015, and generalizability to cohorts diagnosed since then is uncertain.

6. References:

1. Cook A & Giunti. British medical bulletin. 2017:1-12
2. de Silva et al. Orphanet journal of rare diseases. 2019;14(1):1-10.
3. Schulz et al. Nat Rev Neurol. 2009;5(4):222-234

FUNDING & DISCLOSURES: This study was funded by PTC Therapeutics. AS and IT are employees of PTC Therapeutics. CQ, LP, and KJ are employees of Broadstreet HEOR, which received funds from PTC Therapeutics for this work, and GV received consulting fees from PTC Therapeutics. Medical writing and editorial support was provided by Broadstreet HEOR and was funded by PTC Therapeutics.

CONTACT INFORMATION: Ioannis Tomazos, ytomazos@ptcbio.com

© 2023 PTC Therapeutics, Inc. All rights reserved.

THE INTERNATIONAL SOCIETY FOR PHARMACOECONOMICS AND OUTCOMES RESEARCH (ISPOR) EUROPEAN CONFERENCE, COPENHAGEN, DENMARK, NOVEMBER 12-15, 2023