HSD62

The Emotional and Information Seeking Experience of Locally Advanced Cervical Cancer: Qualitative Research Findings from Brazil

Elizabeth Szamreta[a], Emily Mulvihill[b], Katherine Aguinaga[b], Kaitlan Amos[c], Ritu Salani[d]

[a]Merck & Co., Inc., Rahway, NJ 07065, USA

[b]Cerner Enviza, 2800 Rock Creek Pkwy, North Kansas City, MO 64117, USA

[c]Independent Researcher, Dover, New Hampshire, USA

[d]Division of Gynecologic Oncology, Department of Obstetrics and Gynecology, David Geffen School of Medicine, University of California, Los Angeles, 10833 Le Conte Avenue, Los Angeles, CA 90095, USA

Objectives

Little attention has been devoted to the emotional journey of women with locally advanced cervical cancer (LACC) in Brazil. This study sought to understand the emotional states and information-seeking behaviors reported by these women.

Background

Cervical cancer patients struggle with complex decisions affecting their health, fertility, and sexuality. A treatment decision may **offer hope for a good outcome** while causing psychological distress (Germeni, 2014).

Informative resources can alleviate this burden, **helping patients comprehend their diagnosis, plan ahead and assess treatment choices.** Information can motivate patients to actively engage in treatment decisions.

Numerous studies show that most cancer patients prefer involvement and being well-informed; such knowledge can improve:

- Clinical outcomes
- Mental health
- Treatment satisfaction (Hillen 2011, Kane 2014).

This research builds on our previous study of information seeking needs among women with metastatic, persistent, and recurrent CC (Szamreta 2022).

Methods

Semi-structured **60-minute telephone interviews** were conducted from March to November 2022 among 25 women diagnosed with LACC within the past 2 years in Brazil.

This research elicited voices representing diverse demographic & socioeconomic groups.

Trained moderators asked open-ended questions about experience and emotions during the journey from diagnosis to treatment decision, including information seeking. In order to emphasize those elements that are important to the patient, not every participant was required to discuss every topic. Transcripts were coded using NVivo qualitative analysis software to identify key themes that emerged from the data.

The study included high-risk LACC patients (FIGO 2014 stage IB2-IIB with node-positive disease or stage III-IVA, self report) who had selected a treatment within the past two years. Patients who had completed treatment were also part of the study to ensure accurate treatment information. The study was reviewed by the Pearl IRB and received Exemption Status on February 14, 2022.

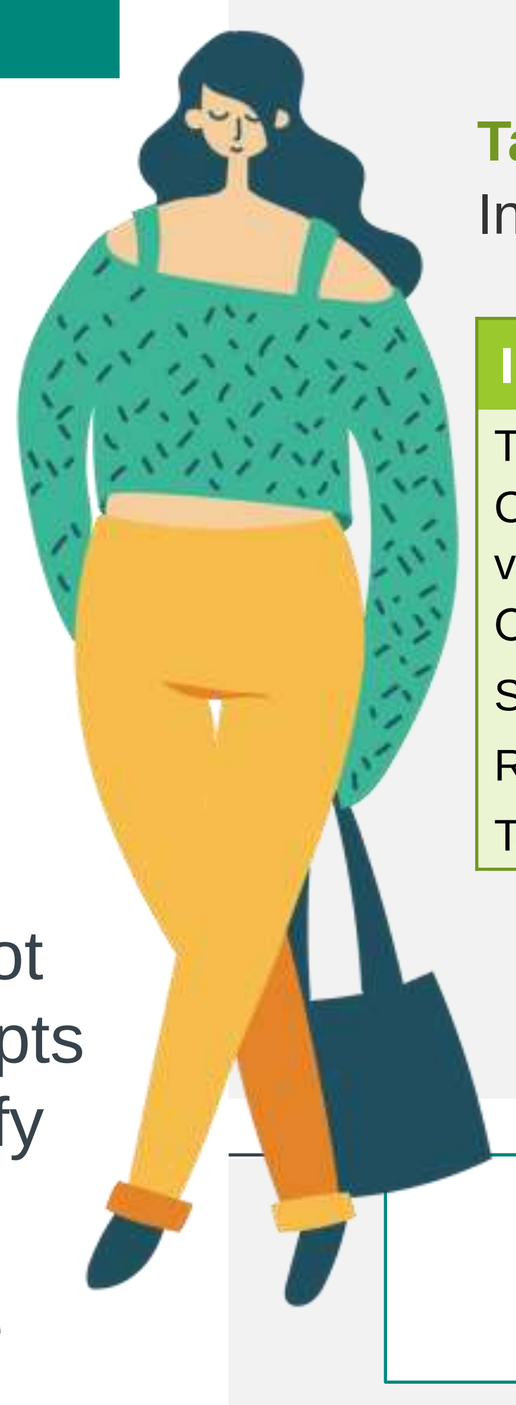

Demographics and Patient Characteristics

25 women participated representing multiple race/ethnicities, states, and health systems:

- 2 had completed primary education,
- 11 had completed secondary education, and
- 12 had completed higher education.
- 8 did not wish for children/more children; however, 10 wanted children but didn't pursue fertility preservation and 3 were uncertain about their wish for children.

Table 1. Participant characteristics (count)

Characteristic	LACC Patients	Characteristic	LACC Patients
Age		Race and Ethnicity	
Mean	35.7	Black	1
Min	26	White	13
Max	49	Pardas	11
Setting		Insurance	
Urban	23	Public	11
Rural	2	Private	14

Figure 1. Residence

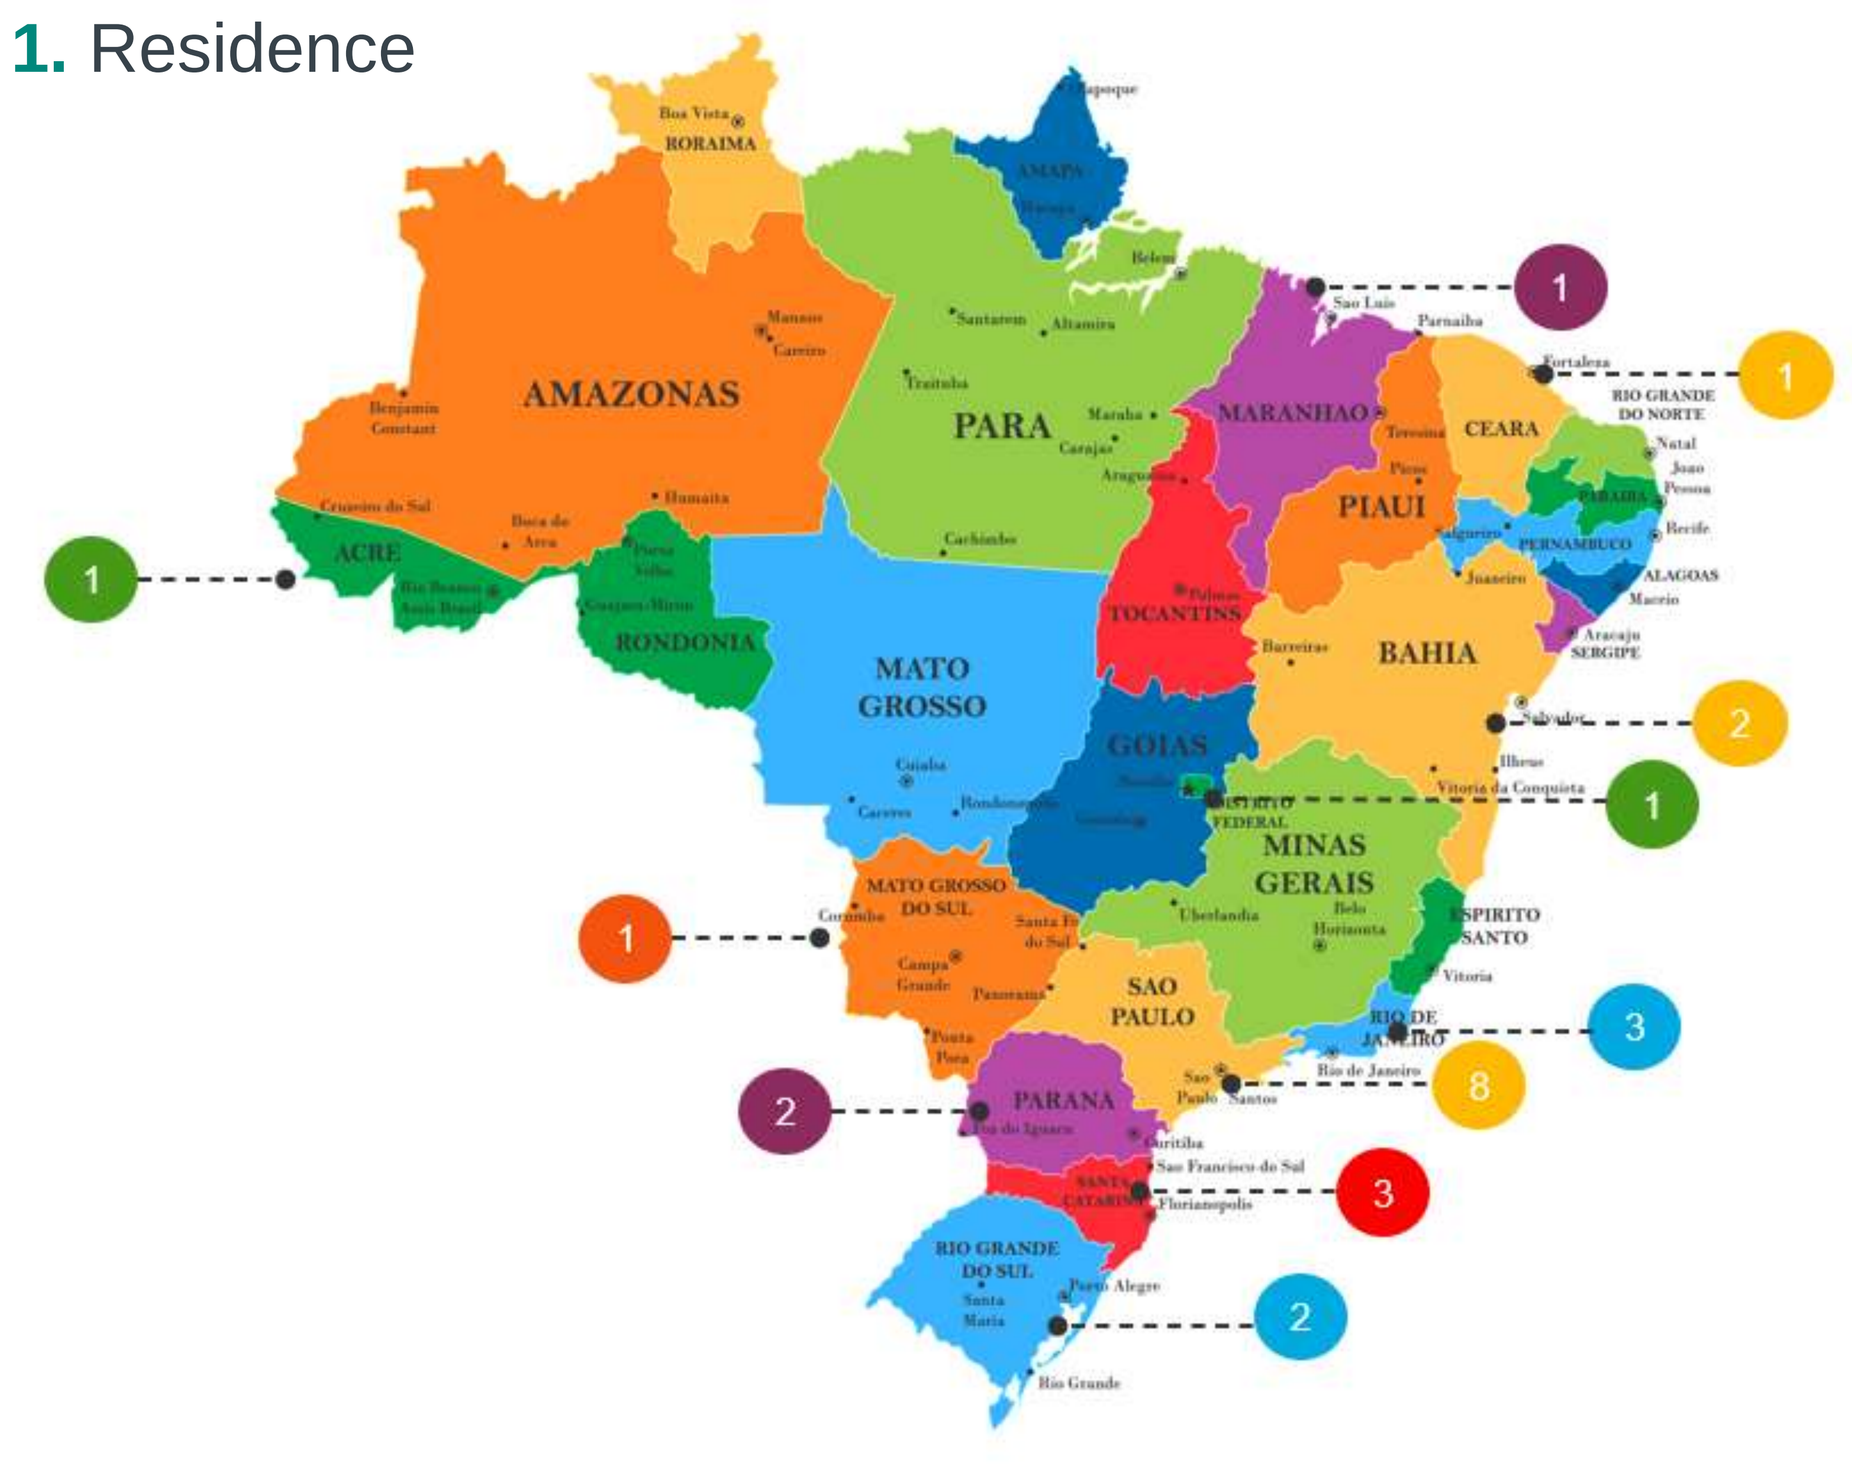

RORAIMA
AMAZONAS
PARA
MARANHAO
CEARA
PIAUI
ACRE
RONDONIA
TOCANTINS
BAHIA
MATO GROSSO
GOIAS
MINAS GERAIS
MATO GROSSO DO SUL
SAO PAULO
PARANA
RIO GRANDE DO SUL

Results

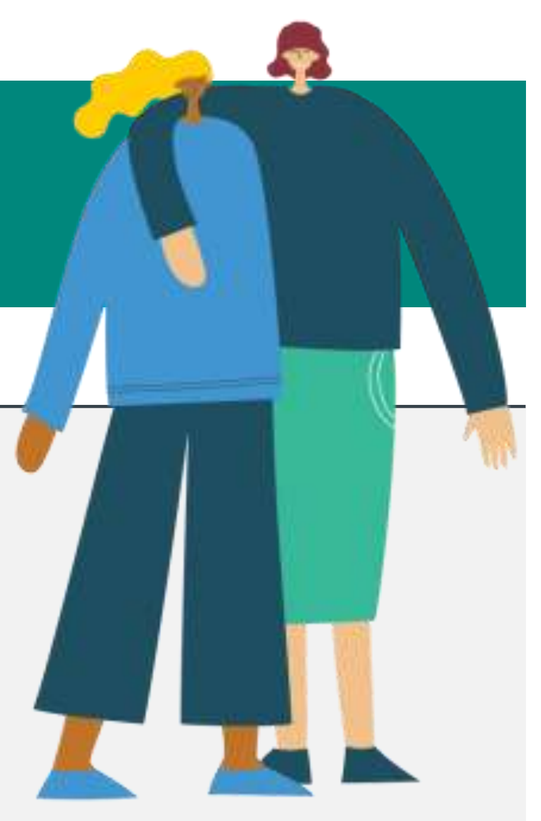

Key Learning 1

The LACC journey is complex and emotional, highlighting unmet needs for patients despite treatment.

DIAGNOSIS

Once receiving their diagnosis, women experienced **fear and anxiety**, including feeling lost, helpless, shocked, overwhelmed and in denial. Several (7) experienced **stigma** associated with CC.

"In the beginning, I was extremely worried. In general, when we receive the diagnosis of cancer, it is something that we do not have knowledge about. So, you do not know what will happen to you exactly. In Brazil, when we receive the diagnosis of cancer, it seems like a death sentence." **BR01**

"When I talked with my mother, she did not want me to tell people. She said, "You will not tell anyone." I said, "No, I will talk. If God gave me these purposes, he wants something from me." So, there was no reason to hide it. We must share even more when it comes to cervical cancer. Although people think it is an easier cancer, it is not. It is not." **BR05**

"What happened? Why do I have cervical cancer?". People associate cervical cancer with dirty, being dirty. I said, "For the love of God"." **BR03**

TREATMENT

While women were **hopeful and confident** after deciding on their treatment plan**, they at the same time expressed concern and worried** about side effects of treatment. Twelve of traveling to treatment. mentioned troubles with **distance or expense.**

"It was a mix of feelings, to be honest. It was the fear, the relief of knowing something that was bad for me was being removed. That was it." **BR20**

"The second barrier was finding specialists in the field. I live in a city and region where there is no specialist in gynecology oncology. So, there is no health professional in this field here. I had to look for them in two capitals of my state and states next to mine." **BR01**

IMPACT ON HEALTH

Most (24) discussed untoward effects of treatment including long term effects; among these, 7 expressed **unpreparedness and/or regret**, including 3 who regretted that children were no longer possible.

"It was missing, no one told me, for example, that I would have all these sequelae from radiotherapy, or chemotherapy, nothing… I developed a fistula…I developed a nerve injury called peripheral neuropathy, which is something that hurts a lot, a lot, a lot, a lot." **BR11**

"This was a very negative part, the fact of not being able to live that, that anguish. The fact I could not be a mother impacted me a lot. The scar impacted me aesthetically but also left me destroyed." **BR07**

Key Learning 2

As they embarked on their CC journey, women sought empowerment by seeking knowledge, and often a second opinion.

Women's information seeking made them feel at ease/relaxed/relieved (4), and happy/prepared (2), but also uncertain/anxious (4), and shameful/sad (1). The top resources women used were online search engines (18) and social media message board forums (6). **Women sought information about treatment, the experience of other patients, and survival and cervical cancer generally.**

Table 2.
Information sought by LACC patients

Information sought	n=23
Treatment info, side effects	10
Cancer info (causes, symptoms, terms, vaccine, staging)	11
Other patients' experiences	8
Survival	1
Relapse, metastasis, cancer progression	1
Test results	2

"There is no information about this, there's nothing; people helping each other regarding the sequelae that remain. The sequelae that we're going to have, how to prepare for them, and how to deal with them — that's also unavailable. It doesn't exist, and I missed it a lot. There are people saying it is cancer and so on, but nothing regarding how to go through with it — this didn't exist." **BR14**

"I found out that cancer can be cured. To me, this information was the most meaningful." **BR02**

"Everything that brought me emotional conformity was more meaningful. For example, "This medication and this protocol have a healing effect." Healing and cure are what I am searching for all the time. So, everything that moved me emotionally impacted me more when deciding and choosing. So, there was this argument. For example, if the answer impacts me emotionally, making me feel comfortable, it would be more effective." **BR01**

"What impacted me the most was the number of people with the same disease. I did not imagine it was the number of people. When I started to research on Facebook, I did not imagine there were so many women with the same disease I had. I thought there were few cases, but there are many." **BR20**

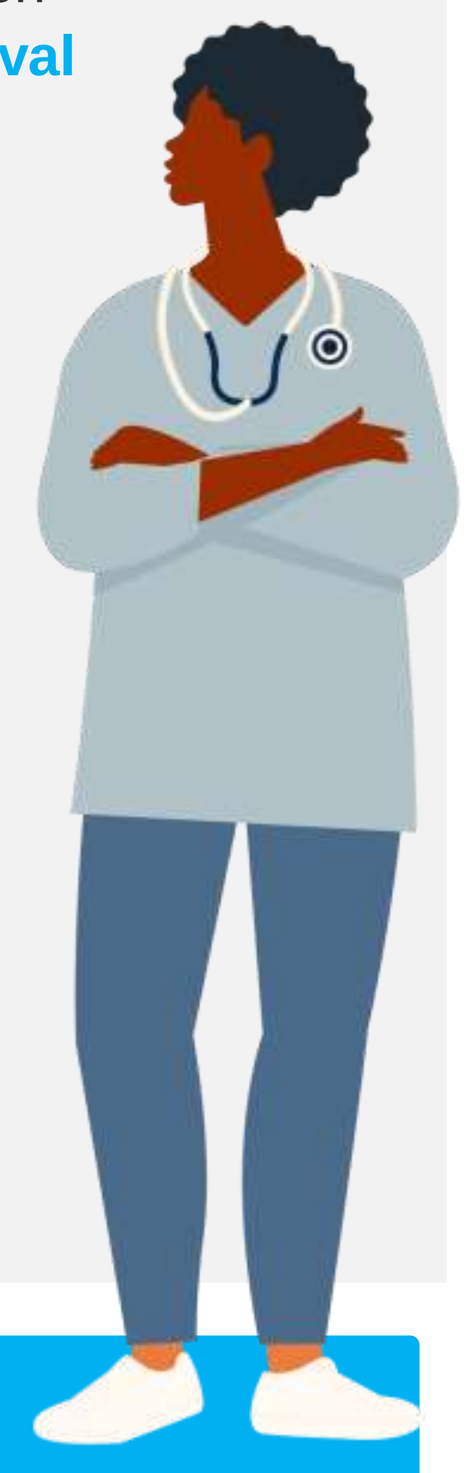

Key Learning 3

LACC patients rely on their healthcare team as an important source of information.

Physicians were an information source for 20 women, and 8 mentioned not seeking a second opinion due to **trust in their doctor**. Asking detailed questions can be a form of self-advocacy; most women felt their questions were answered and came away with **positive feelings and confidence**.

Physicians most frequently shared general information about CC. **Several reported their physician sharing confidence in treatment and raising the chance to be cured after surgery/treatment.**

Table 3.
Information shared by physicians about LACC

Information shared	n=23
General treatment info, general side effects	15
Chance of a cure	12
Efficacy, survival, expectations, next steps	7
Confidence in treatment	4

"He made me feel confident about what he was going to do and thinking to do. However, he has always said to me it was my decision. In this case, I think it is hard to make a decision against the doctor, mainly when you imagine he is doing the right thing. However, if I said, "No, I do not want to do surgery," it was not going to happen. If I said, "No, I do not want to do chemotherapy," it would be okay. He sent me what was best, and I believed him and did it." **BR04**

"She said there is no recipe when it comes to cancer. For example, do this to get this result. However, in stage II, chances of getting cured are high." **BR04**

"They discussed with me the best options. When we saw it had decreased he said, "The whole team voted for surgery. However, if you do not want to do it, it is your choice. You can start another treatment." **BR07**

"She did; she explained everything to me. I left the place with more knowledge." **BR26**

"No, he always explained everything; he was very clear, telling me what happened, what could happen, that if I wanted to have children, I wouldn't be able to anymore because of the treatment." **BR10**

"I made a lot of questions. They answered all of them. I think that is why I trusted both doctors a lot." **BR02**

When interacting with their healthcare providers, 13 women opted for in-person appointments. 12 (5 with public insurance, 7 with private insurance) utilized messaging apps and among these, 11 favored messaging due to prompt responses and the convenience of avoiding travel.

However, 8 women (7 with public insurance, 1 with private insurance) indicated that they can only engage with their physicians through scheduled appointments and turn to outpatient or emergency services for urgent concerns.

"Nowadays, it is WhatsApp. It is hardly a call. I would keep it this way because I have always received a quick answer every time I needed it." **BR06**

"No, I don't have anything; I have to go to the clinic, and that's it – I schedule an appointment and have the consultation. I don't have a WhatsApp contact or something like that." **BR13**

"No, I only get in touch with him at appointments; it is the schedule. If something happens, like pain, I need to look for the hospital outpatient service." **BR20**

Conclusions

The diagnosis and treatment for LACC impose a heavy emotional and physical burden on patients, potentially leaving women feeling unprepared or regretful.

However, successful information seeking can help turn this daunting journey into an empowering one, where trusted healthcare providers emerge as crucial pillars of support and knowledge, particularly when communication is facilitated by messaging apps.

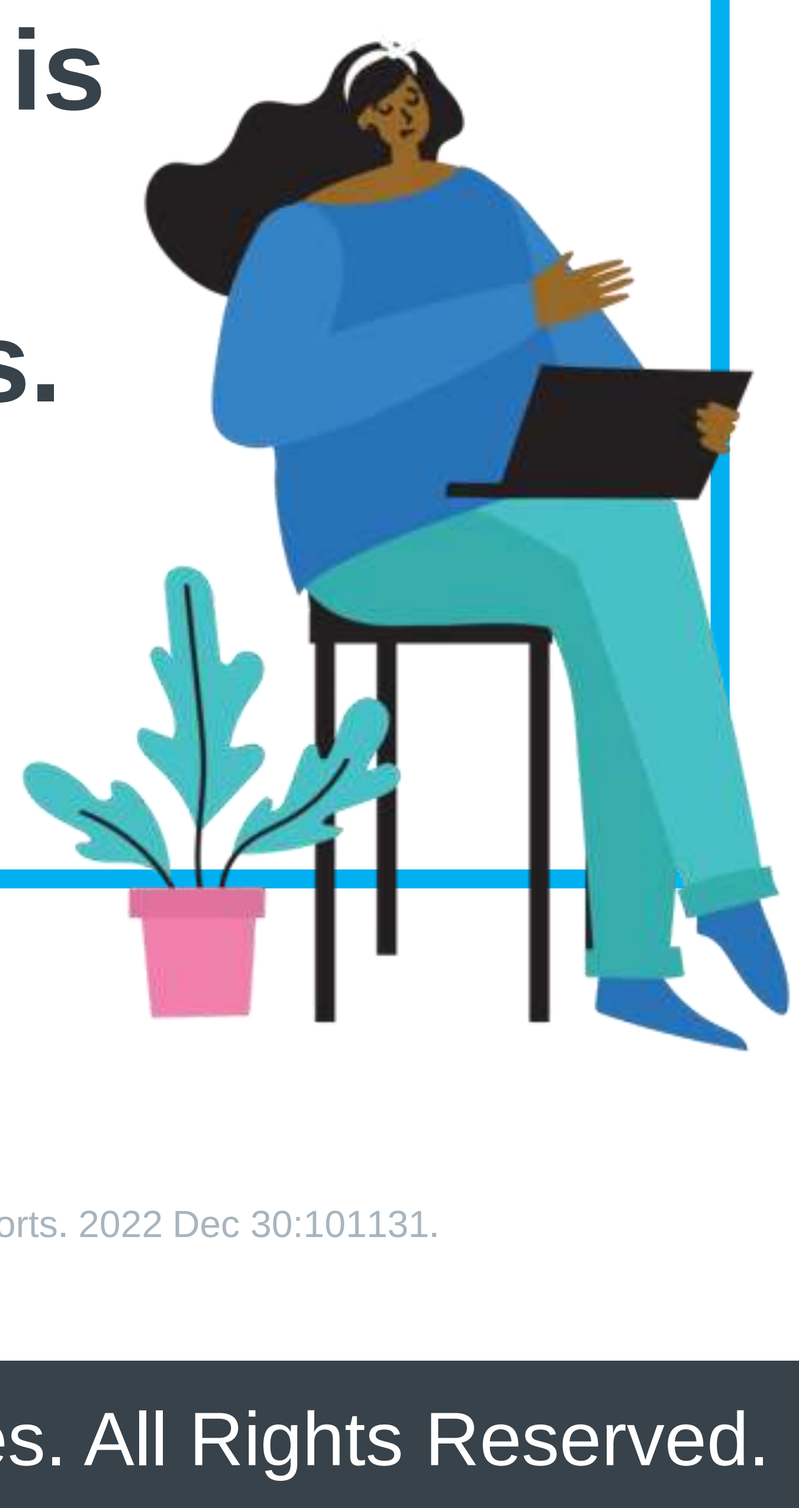

1. Germeni E, Schulz PJ. Information seeking and avoidance throughout the cancer patient journey: two sides of the same coin? A synthesis of qualitative studies. Psycho-Oncology. 2014 Dec;23(12):1373-81.
2. Hillen MA, De Haes HC, Smets EM. Cancer patients' trust in their physician—a review. Psycho-oncology. 2011 Mar;20(3):227-41.
3. Kane HL, Halpern MT, Squiers LB, Treiman KA, McCormack LA. Implementing and evaluating shared decision making in oncology practice. CA: a cancer journal for clinicians. 2014 Nov;64(6):377-88.
4. Szamreta EA, Wayser GR, Prabhu VS, Mulvihill E, Aguinaga K, Salani R. Information needs during cancer care: qualitative research with advanced cervical cancer patients in Brazil, China, Germany, and the United States. Gynecologic Oncology Reports. 2022 Dec 30:101131.

Presented at ISPOR Europe, Copenhagen, 12-15 November 2023

Copyright © 2023 Merck & Co., Inc., Rahway, NJ, USA and its affiliates. All Rights Reserved.