EE681

Economic Impact Of Fixed-treatment Duration For Chronic Lymphocytic Leukemia: Budget-Impact Analysis Of Venetoclax Plus Obinutuzumab For First Line Treatment Of Unfit Patients In France

Leblond V[1], **Souihi C[2]**, Ramier J[2], Manzoor BS[3], Lepretre S[4]

[1]Sorbonne University, Paris, France, [2]AbbVie, Rungis, France, [3]AbbVie Inc., North Chicago, USA ,[4]Centre Henri-Becquerel, Rouen, France,,

OBJECTIVE

This study aimed to assess the budget impact (BI) of introducing 12-month fixed-duration venetoclax+obinutuzumab (VEN+O) as first-line (1L) treatment in CLL for patients with or without a *del17p/TP53mut* and ineligible for full dose fludarabine treatment from the French payer perspective.

CONCLUSIONS

Introducing VEN+O, a fixed treatment duration regimen, as treatment of 1L CLL patients offers a lower total economic cost option compared to other available treatments in France.

The budget impact analysis estimated cost savings of 176,632,387 € over 5 years. The fixed 12-month treatment duration of venetoclax was the key driver of these savings, accounting for a reduction of 16% of the total costs of care.

Chemoimmunotherapies should gradually be replaced by VEN+O and BTK inhibitors in the French therapeutic landscape, with further potential savings achievable with VEN+O over BTK inhibitors

AbbVie is the sponsor of this study, and contributed to its design, data collection, analysis and interpretation, and the review and validation of the poster. All authors had access to the relevant data and participated in the writing, review and approval of this poster. The authors received no honorarium for this publication. The authors would like to thank the AbbVie-funded support of IQVIA for the medical writing and budget impact model development.

Stéphane Leprêtre: AbbVie, Amgen, AstraZeneca, Beigene, Janssen; Véronique Leblond: Abbvie, Amgen, AstraZeneca, Beigene, Janssen, Gilead, Lilly, MSD, Pfizer; Julien Ramier: employee of AbbVie and may own AbbVie stock or stock options; Charaf Souihi: employee of AbbVie and may own AbbVie stock or stock options; Beenish Manzoor: employee of AbbVie and may own AbbVie stock or stock options

REFERENCES

1, Defossez G, Le Guyader-Peyrou S, Uhry Z, et al. National Estimates of Cancer Incidence and Mortality in Metropolitan France between 1990 and 2018. Santé Publique France; 2019.
2. Colonna M, Boussari O, Cowppli-Bony A, et al. Time trends and short term projections of cancer prevalence in France. Cancer Epidemiology. 2018
3. AbbVie press release January 31, 2020
4. CT opinion June 29, 2022
5. Choix méthodologiques pour l'analyse de l'impact budgétaire à la HAS
6. Davids MS & al: The economic impact of treatment sequences for chronic CLL in the US
7. Kimberly Guinan & al: The economic impact of treatment sequencing in the management of CLL in Canada

Scan QR code or utilize the following link to download an electronic version of this presentation and other AbbVie 2023 ISPOR scientific presentations:
https://abbvie1.outsystemsenterprise.com/GMAEventPublications/Assets.aspx?ConferenceId=651
Expiration Date: October 12, 2024

To submit a medical question, please visit www.abbviemedinfo.com

For additional information or to obtain a PDF of this poster

INTRODUCTION

- Chronic lymphocytic leukemia (CLL) accounts for 30% of leukemias in French adults, with an estimated incidence of 4,674 new cases in 2018[1] and approximately 23,000 patients living with the condition[2]. CLL can be life-threatening, especially in patients with high-risk genetic alterations such as a *17p* deletion or *TP53* mutation.
- In January 2020, the European Medicine Agency[3] approved venetoclax, an oral BCL-2 inhibitor, in combination with obinutuzumab (VEN+O) for patients with previously untreated CLL, based on the results of the phase III CLL14 trial (NCT02242942). The Haute Autorité de Santé (HAS), the French HTA body, granted a positive opinion[4] for its reimbursement in June 2022 for patients with CLL who were previously untreated with or without a *del17p/TP53mut* and ineligible for full dose fludarabine treatment.
- The fixed duration of VEN+O as compared to existing targeted therapies could relieve the financial burden of CLL for national payer. A budget impact model was developed to assess the economic impact of introducing VEN+O on French market.

METHODS

Model

- A budget impact model was developed to estimate two different scenarios (with and without VEN+O) over a 5-year time horizon, from the French payer perspective. The model has been designed in accordance with the HAS methodological guidance for budget impact analysis[5].

Comparators

- Comparators included were chemoimmunotherapies (bendamustine+rituximab [BR] and obinutuzumab+chlorambucil [Oclb]) and BTK inhibitors (ibrutinib [I], acalabrutinib [A], and acalabrutinib+obinutuzumab [A+O]).

Target population and market shares

- The model used an incidence-based, open multi-cohort approach. The eligible population was estimated by using French national registries and expert's opinion. The market share growth dynamic of VEN+O included competition with an expected growth of A/A+O and a gradual decline of chemoimmunotherapies market shares.

Costs items

- The model calculated the total cost of care (TCC) for each regimen and included 1L treatment costs (drug [list price] and administration), tumor lysis syndrome (TLS) prevention costs, adverse event (AE) costs, monitoring costs, subsequent treatment costs (drug [list price] and administration) and transportation costs.

Clinical inputs

- Clinical data (**Table 1**) were obtained from pivotal clinical trials (for time on treatment [ToT], progression-free survival [PFS] and AE) and validated with clinical experts.

Table 1. Clinical inputs

	VEN+O	OClb	A	A+O	I	BR
Median ToT (months)	11,1/5,5	11,1/5,5	59	58.1/5,5	57	5.5
Median PFS (months)	Not reached	36	Not reached	Not reached	Not reached	44
Source	CLL14	CLL14	ELEVATE-TN	ELEVATE-TN	RESONATE 2	ALLIANCE

Sensitivity analysis

- Deterministic sensitivity analyses on cost and clinical inputs were performed to explore parametric uncertainty.

RESULTS

Treated population

- Over the 5-year time horizon, a total of 7,500 patients were treated, of which 2,670 by VEN+O in the world WITH.

Total cost of care

- Total cost per patient, by regimen and costs item are presented in **Figure 1**.
- The TCC in the scenario without VEN+O was 1,107,105,361€, while the TCC was 930,472,974 € in the scenario with VEN+O (**Figure 2**). First-line drug acquisition costs accounts for 92% and 88% of the TCC in scenario without and with VEN+O respectively. Subsequent-line treatment acquisition costs ranked second, with less than 5% of the TCC in both scenarios.

Figure 1. TCC per patient, by regimen and costs item

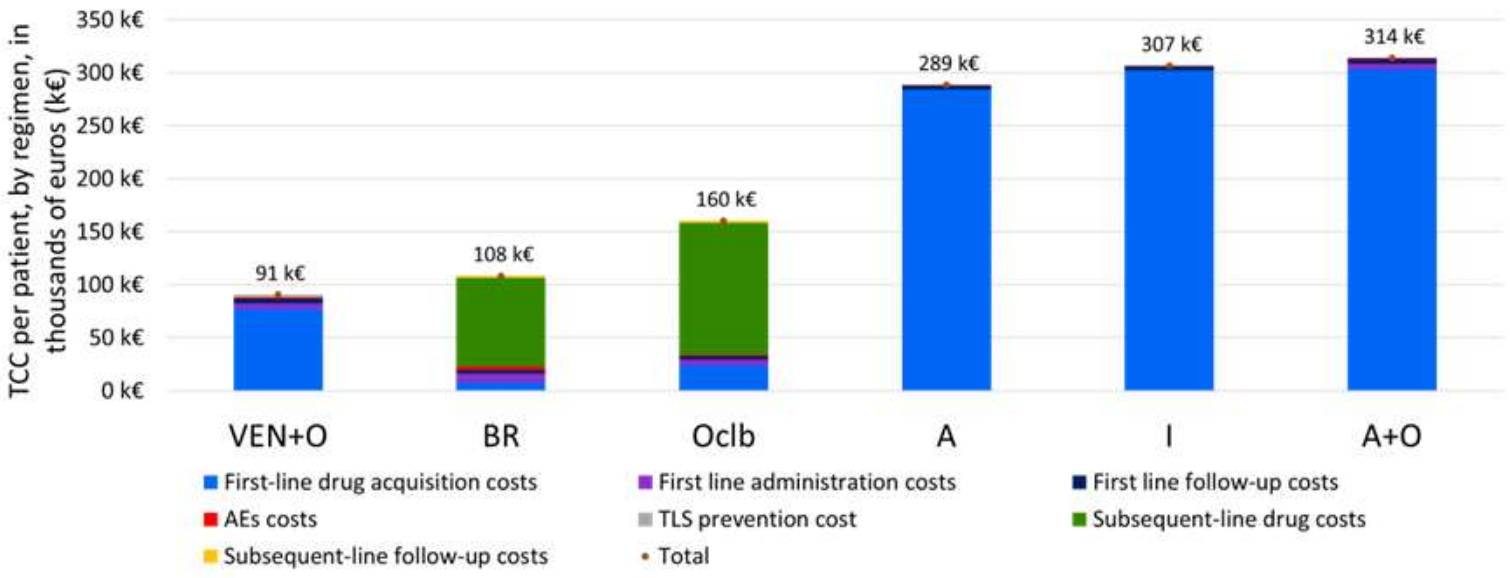
TCC per patient, by regimen, in thousands of euros (k€)
350 k€
300 k€
250 k€
200 k€
150 k€
100 k€
50 k€
0 k€
91 k€
108 k€
160 k€
289 k€
307 k€
314 k€
VEN+O
BR
Oclb
A
I
A+O
First-line drug acquisition costs
First line administration costs
First line follow-up costs
AEs costs
TLS prevention cost
Subsequent-line drug costs
Subsequent-line follow-up costs
Total

Budget impact results

- Introducing VEN+O resulted in total cost savings of €176,632,387 over the 5-year time horizon, which represents almost a 16% reduction of the total cost of care.
- First line treatment acquisition costs was the most impacted item by introducing VEN+O (-198,3 M€, -19.5% of the related costs). This is attributable to the fixed 12-month treatment duration of venetoclax, compared with continued use until progression or toxicity for BTK inhibitors, and the significant market share uptake of VEN+O on other targeted therapies (**Figure 3**).

Figure 2. TCC in worlds with and without VEN+O

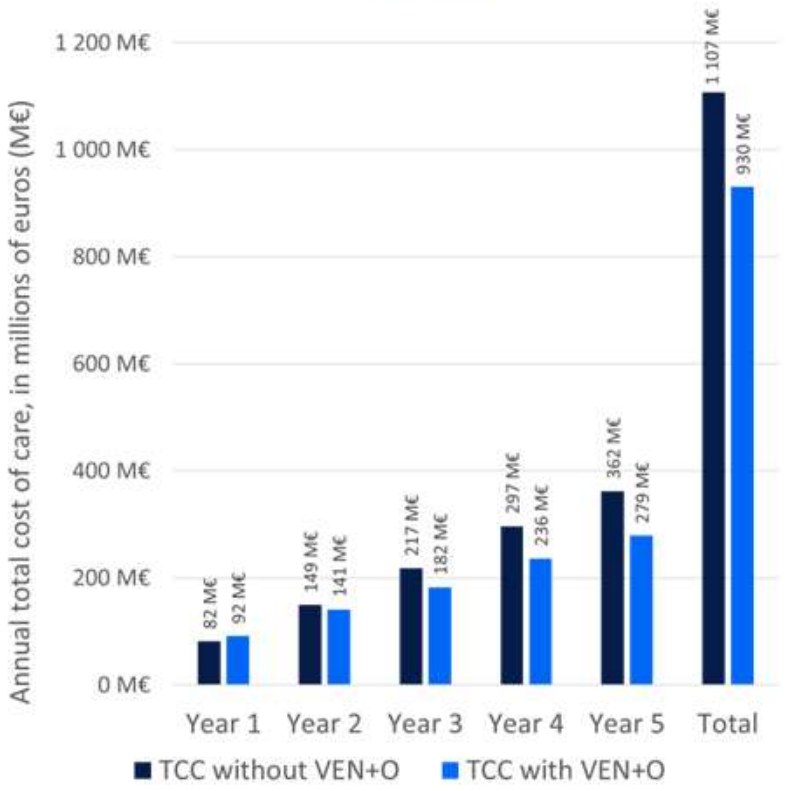
Annual total cost of care, in millions of euros (M€)
1 200 M€
1 000 M€
800 M€
600 M€
400 M€
200 M€
0 M€
82 M€
92 M€
149 M€
141 M€
217 M€
182 M€
297 M€
236 M€
362 M€
279 M€
1 107 M€
930 M€
Year 1
Year 2
Year 3
Year 4
Year 5
Total
TCC without VEN+O
TCC with VEN+O

Figure 3. Cumulative budget impact between worlds with and without VEN+O and breakdown by cost-item

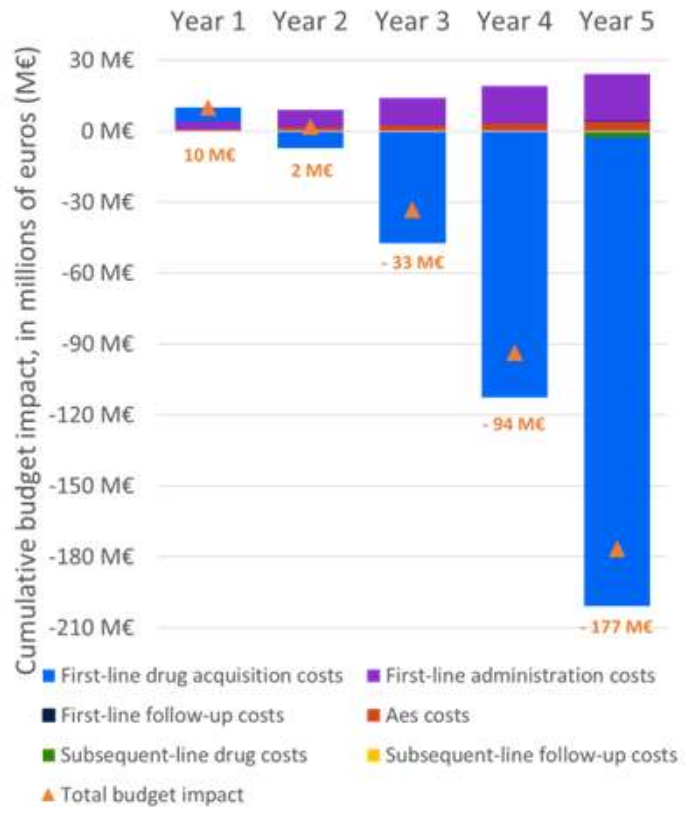
Year 1
Year 2
Year 3
Year 4
Year 5
Cumulative budget impact, in millions of euros (M€)
30 M€
0 M€
-30 M€
-60 M€
-90 M€
-120 M€
-150 M€
-180 M€
-210 M€
10 M€
2 M€
- 33 M€
- 94 M€
- 177 M€
First-line drug acquisition costs
First-line administration costs
First-line follow-up costs
Aes costs
Subsequent-line drug costs
Subsequent-line follow-up costs
Total budget impact

Sensitivity analysis results

- Sensitivity analyses confirmed the base-case findings, savings were observed in all sensitivity analysis. The model was most sensitive to parameters related to targeted therapy (I, A, VEN) prices and ToT, with absolute variations of 5 to 25% compared to base-case results.

DISCUSSION

- The economic impact of shifting the therapeutic strategy from traditional immunochemotherapies to targeted therapies is limited by the introduction of VEN+O, which appears to be the least expensive strategy, yet as effective as other targeted therapies, over the time horizon considered (**Table 1, Figure 2**).
- These results are consistent with those found in the literature. Several studies conducted in other Western countries[6,7] have indeed shown the relatively high economic burden of continuous targeted therapies in CLL, and thus the opportunity to reduce associated acquisition costs when introducing fixed-duration treatment such as venetoclax.
- One limitation of the analysis was the unavailability of robust estimates of the relative efficacy of all regimens (notably ToT data). The pragmatic choice to use crude data from different trials was made, validated with clinical experts, and extensive sensibility analysis on ToT parameters didn't affect the findings.
- Furthermore, one limitation of this analysis is the requirement for a more extended time horizon, spanning approximately 10 years. It would facilitate an assessment of the effectiveness of both first-line and second-line treatments, as well as an evaluation of the costs associated with subsequent therapies in scenarios post-BTKi and post-VEN+O.

Acronyms : 1L : First-line; A: acalabrutinib: A+O: acalabrutinib + obinutuzumab; BR: bendamustine + rituximab; CLL: chronic lymphocytic leukemia; I: ibrutinib; HAS: Haute Autorité de Santé; M€: millions of euros; Oclb: obinutuzumab + chlorambucil; TCC: total cost of care; ToT: time-on-treatment; VEN+O: venclyxto + obinutuzumab

ISPOR 2023, November 12-15, Copenhagen, Denmark