An exploratory study assessing the utility impact in second informal carers of patients with epidermolysis bullosa

Snell T[1], Stainer L[1], Mather C[2], Hussain S[2], Low E[3], Tolley K[1]

[1]Tolley Health Economics, Buxton, United Kingdom; [2]DEBRA UK; [3]Eric Low Consulting

Poster No. EE553

BACKGROUND

- The impact of disease on carer quality of life is an important consideration in the economic evaluation of health technologies. Increasingly, carer QALYs are a component of ICER estimates in the context of UK health technology appraisals (especially rare diseases assessed under the HST programme) as recommended in guidelines issued by NICE.
- Epidermolysis bullosa (EB) is a rare, genetic skin disease characterised by skin fragility and blistering. For patients and their families, wound care is an important part of living with the disease. In more severe forms of EB, this can involve painful dressing changes that are needed several times per weeks, and may take several hours to perform.
- A submission to NICE was made in November 2022 for the evaluation of Filsuvez® (birch bark extract) gel, a topical ointment that has been shown to increase the speed of wound healing in patients with dystrophic and junctional forms of EB.

 The submission used a transition-based model approach to consider the impact of treatment on movement between EB severity states.
- A series of evidence-gathering studies and elicitation exercises carried out to support the submission informed estimates of the relationship between EB severity and number of informal carers typically involved in routine care. Across the six severity states considered in the economic model, the estimated mean number of carers ranged from 0.5 carers to 1.78 carers per patient (**Figure 1**).
- A time trade-off (TTO) study conducted in the UK general public provided estimates of carer utilities for each health state, ranging from 0.91 to 0.64.
- The focus of the TTO exercise was the quality of life of patients' 'main carer'. Without making assumptions as to the generalisability of the study's findings, no utility estimate for second carers was available.

Figure 1: Mean expected number of carers per patient with EB

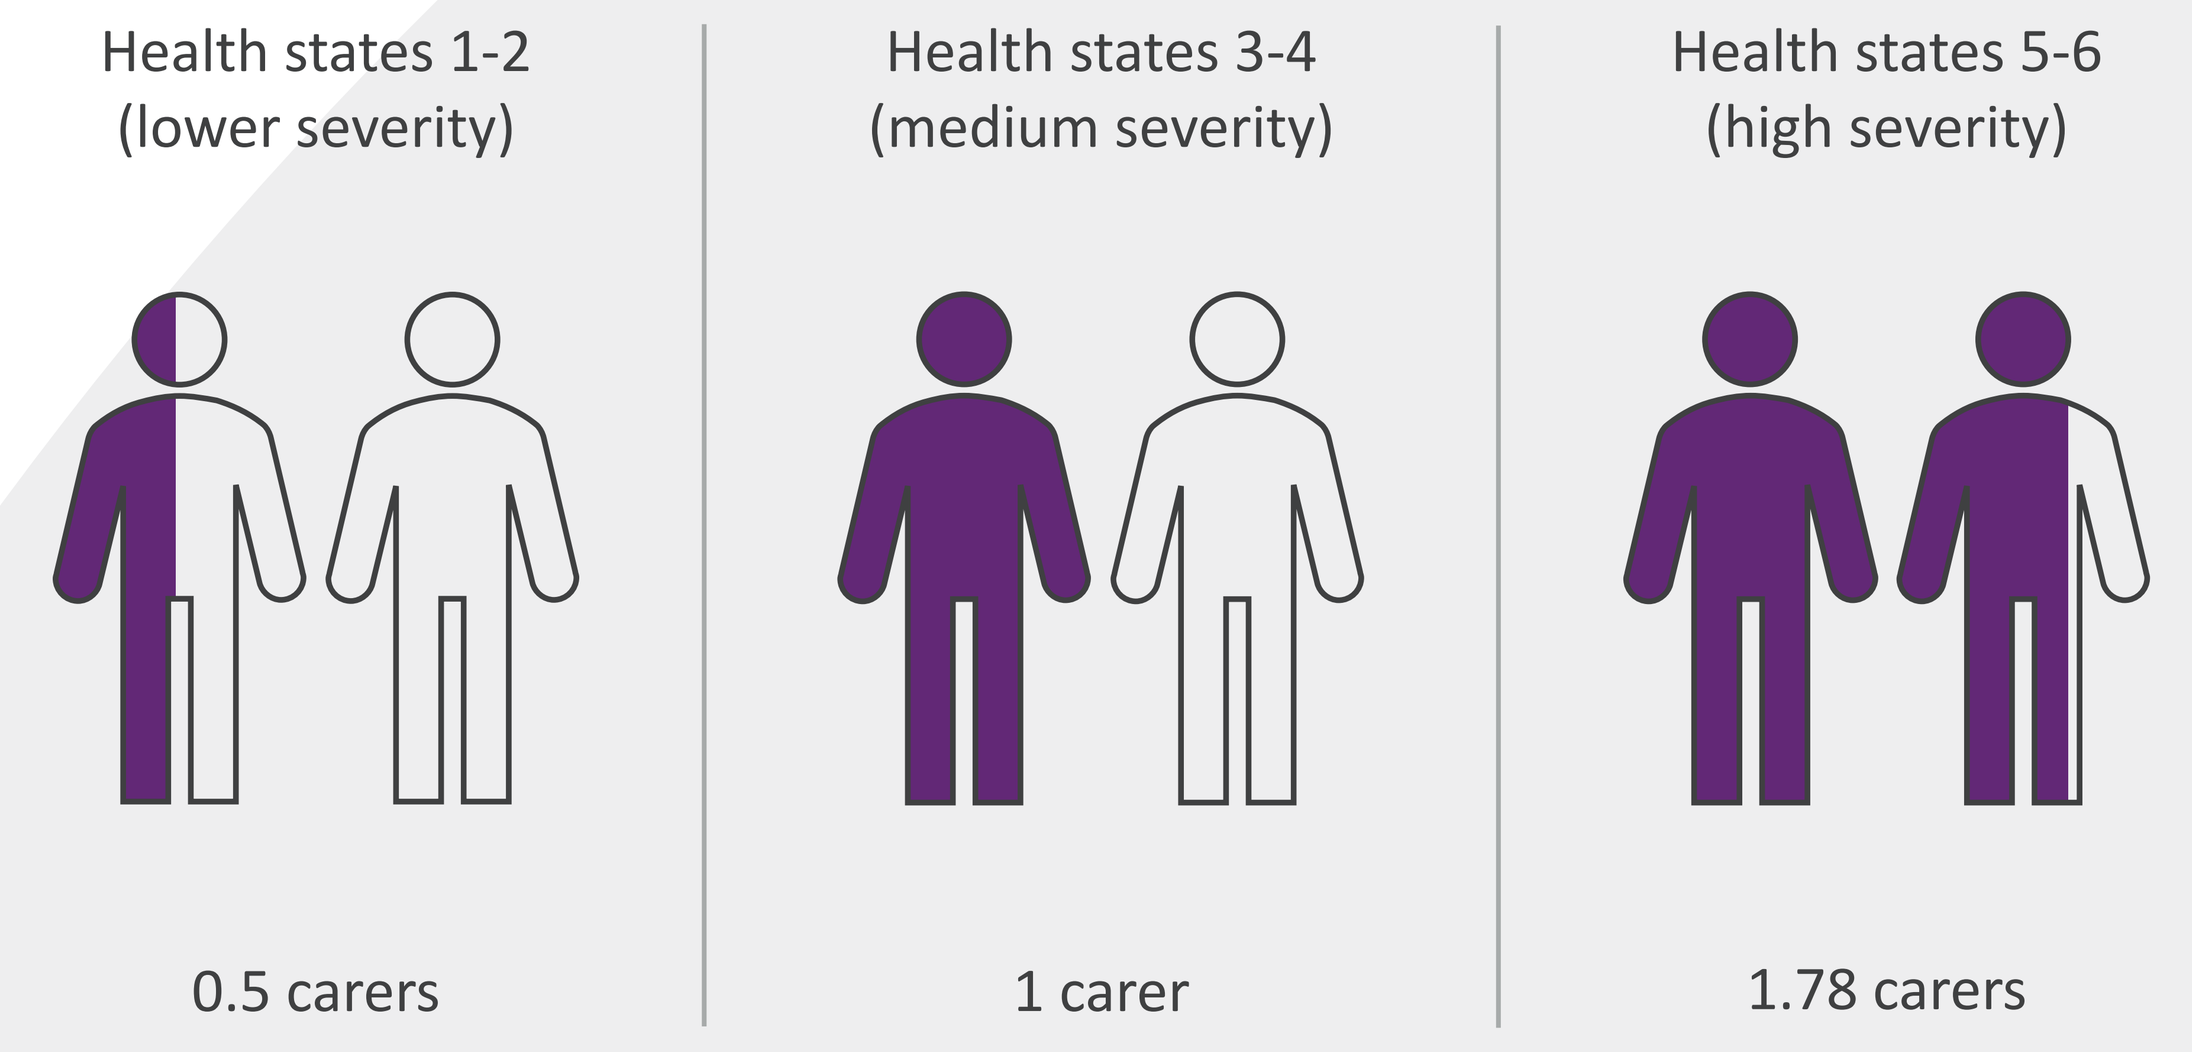

Health states 1-2
(lower severity)
Health states 3-4
(medium severity)
Health states 5-6
(high severity)
0.5 carers
1 carer
1.78 carers

OBJECTIVES

- A pragmatic, targeted survey was developed to elicit estimates from patients, patient representatives and/or health professionals familiar with EB around the relative quality of life impact on second carers of patients with more severe forms of the disease.

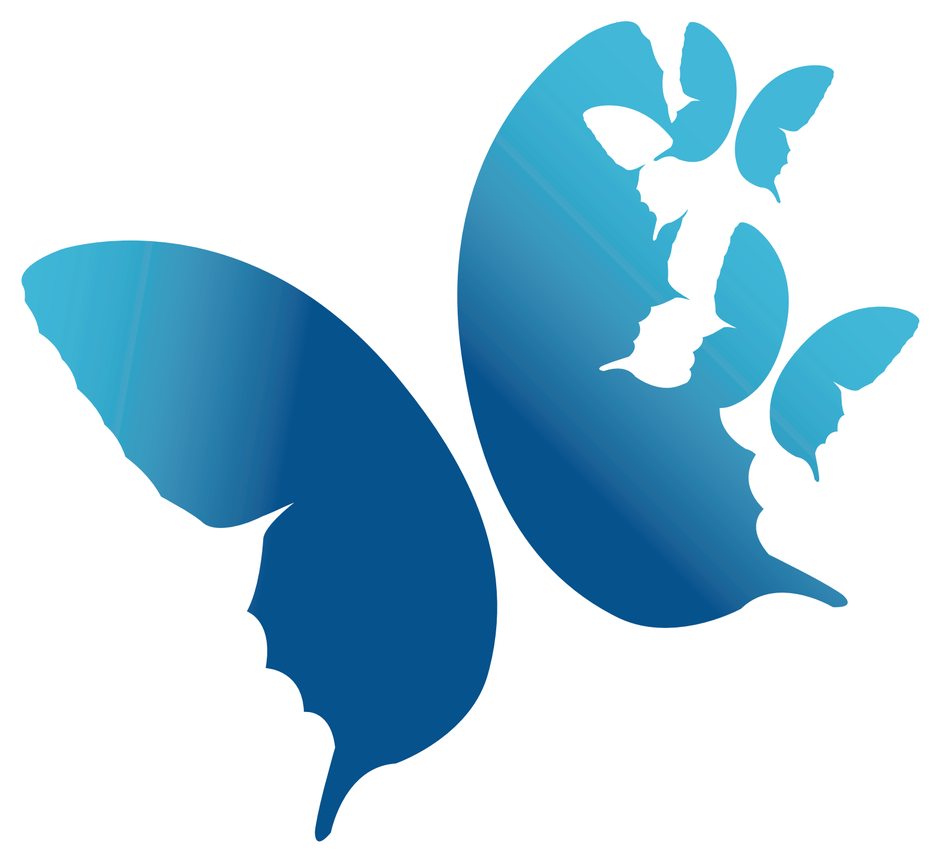

- The purpose of the survey was to supplement existing evidence to allow for carer impact to be estimated with greater certainty. This meant that new evidence had to be framed carefully within the context of the underlying evidence and assumptions already used to inform decision-making.
- A key challenge was in communicating relatively complex questions around disease burden, the measurement and valuation of quality-of-life measures, while minimising response burden to ensure that interpretable results could be obtained in a short time window.

METHODS

- A short online questionnaire was developed by THE and piloted with representatives at DEBRA UK, a national charity supporting members including EB patients, their relatives, partners and carers, and professionals and researchers working with EB.
- Upon finalisation of the materials, the survey was rolled out to members of DEBRA UK (including patients, carers, and health professionals) to complete via a link (hosted by SurveyMonkey) on the members area of the DEBRA UK website.
- The final survey included two core questions, both of which related to the carer vignette (Table 1) used in an earlier TTO exercise to describe care of patients with the high EB severity in terms of body surface area percentage (BSAP).
- Respondents were first asked to consider the validity of the assumption that more than one carer would typically be involved in providing unpaid care.
- Respondents were then asked to consider circumstances in which a second carer was involved, and asked to estimate the extent to which caregiving affected the quality of life impact of the second carer relative to the 'main' carer. Responses were collected using a sliding scale where 0% indicates that the secondary carers quality of life is not impacted at all, and 100% means that their quality of life is impacted by at least as much as the main caregiver.

Table 1: Vignette description provided to survey respondents

Wounds and other symptoms
• You are the main caregiver of a person with wounds covering 25% or more of their body. These wounds cover a significant area of their limbs and a significant area of their chest and abdomen (see diagram). The person develops blisters easily and has a lot of skin crusting, scabbing or erosions on their body.
Disease management
• You dress the person's wounds daily, so that they heal. The process takes 4 or more hours daily.
• The person with EB has severe acute (temporary) skin pain or discomfort with itching, especially when dressings are changed.
• You regularly administer treatment for pain, itch and nutritional supplements to the person with EB. They require a high dose of painkillers (daily) for their wounds.
Impact on your life
• The person with EB is unable to eat or drink normally– you aid their feeding multiple times a day. They are unable to sleep well and your sleep will be very impacted.
• You accompany the person with EB to their frequent medical visits and in-hospital treatment for anaemia; you are unable to work. Due to their inability to move around or use their hands, you must always aid the person with EB to adjust to their daily activities.
• Often, you experience negative emotions (such as anxiety or frustration) due to the nature and burden of the condition.

RESULTS

- A link to the survey was loaded to the members page of the DEBRA UK website on 30th March 2023, with a request for responses by the 5th April. The survey link was kept live by DEBRA UK beyond this date to accommodate additional responses up to the deadline for new evidence to inform the technical engagement process.
- Key questions (Q1 and Q2) had been responded to by six individuals at the point of analysis (13th April 2023). All were either friends or family members of a patient with EB (n=3) or had professional experience with EB (n=3).
- All respondents agreed that more than one informal carer would typically be involved in the management of an EB patient with EB of the severity described.
- The estimated decrement overall average HRQoL impact of 77% (range 60%-100%) relative to the main carer (Figure 2).
- The utility score of 0.64 estimated for main carers of DEB and JEB patients in the highest severity state corresponded to a utility decrement of 0.27 relative to age-adjusted general population levels.
- Reflecting the assumption that second carers' utility decrement is 77% of this level, a utility score of 0.70 was estimated for the second carers of EB patients in the highest health state.
- The QALY impact on primary and secondary carers as well as patients was accepted as suitable for NICE decision making in the positive recommendation of birch bark extract as the first approved treatment of partial thickness wounds associated with dystrophic EB and junctional EB in the UK.

Figure 2: Summary of Q2 responses by respondent type

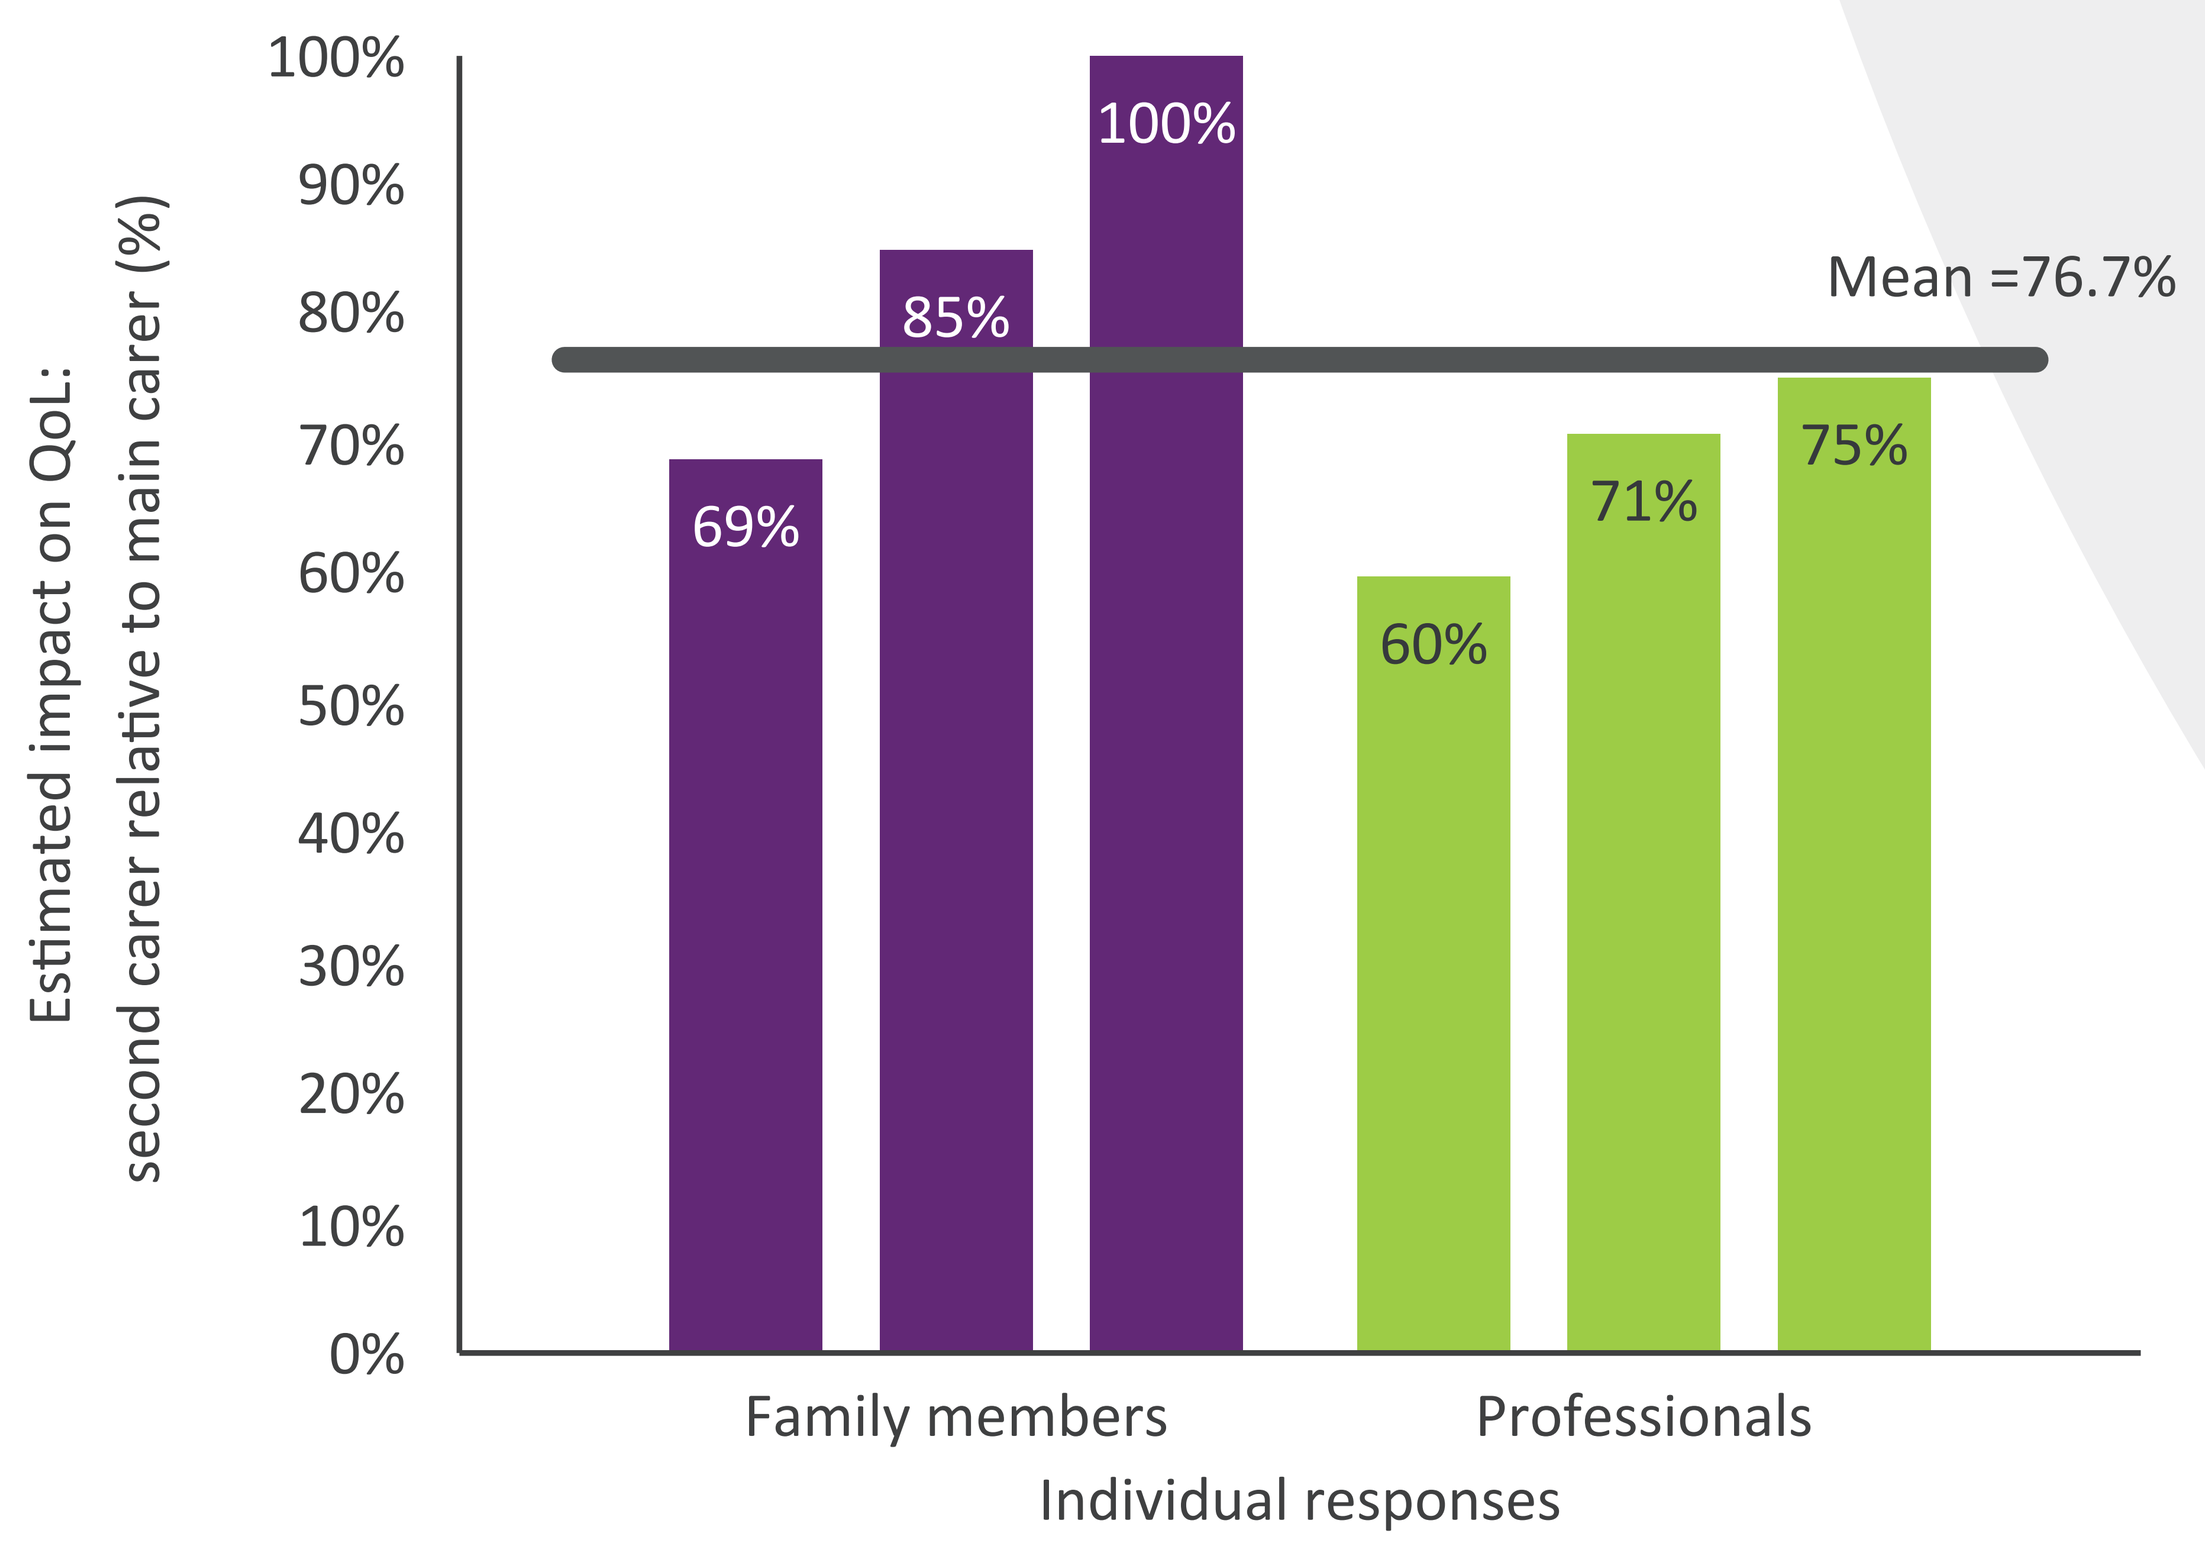

Estimated impact on QoL:
second carer relative to main carer (%)
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
69%
85%
100%
60%
71%
75%
Mean =76.7%
Family members
Professionals
Individual responses

Figure 3: Carer utility estimates incorporating carer 2 response

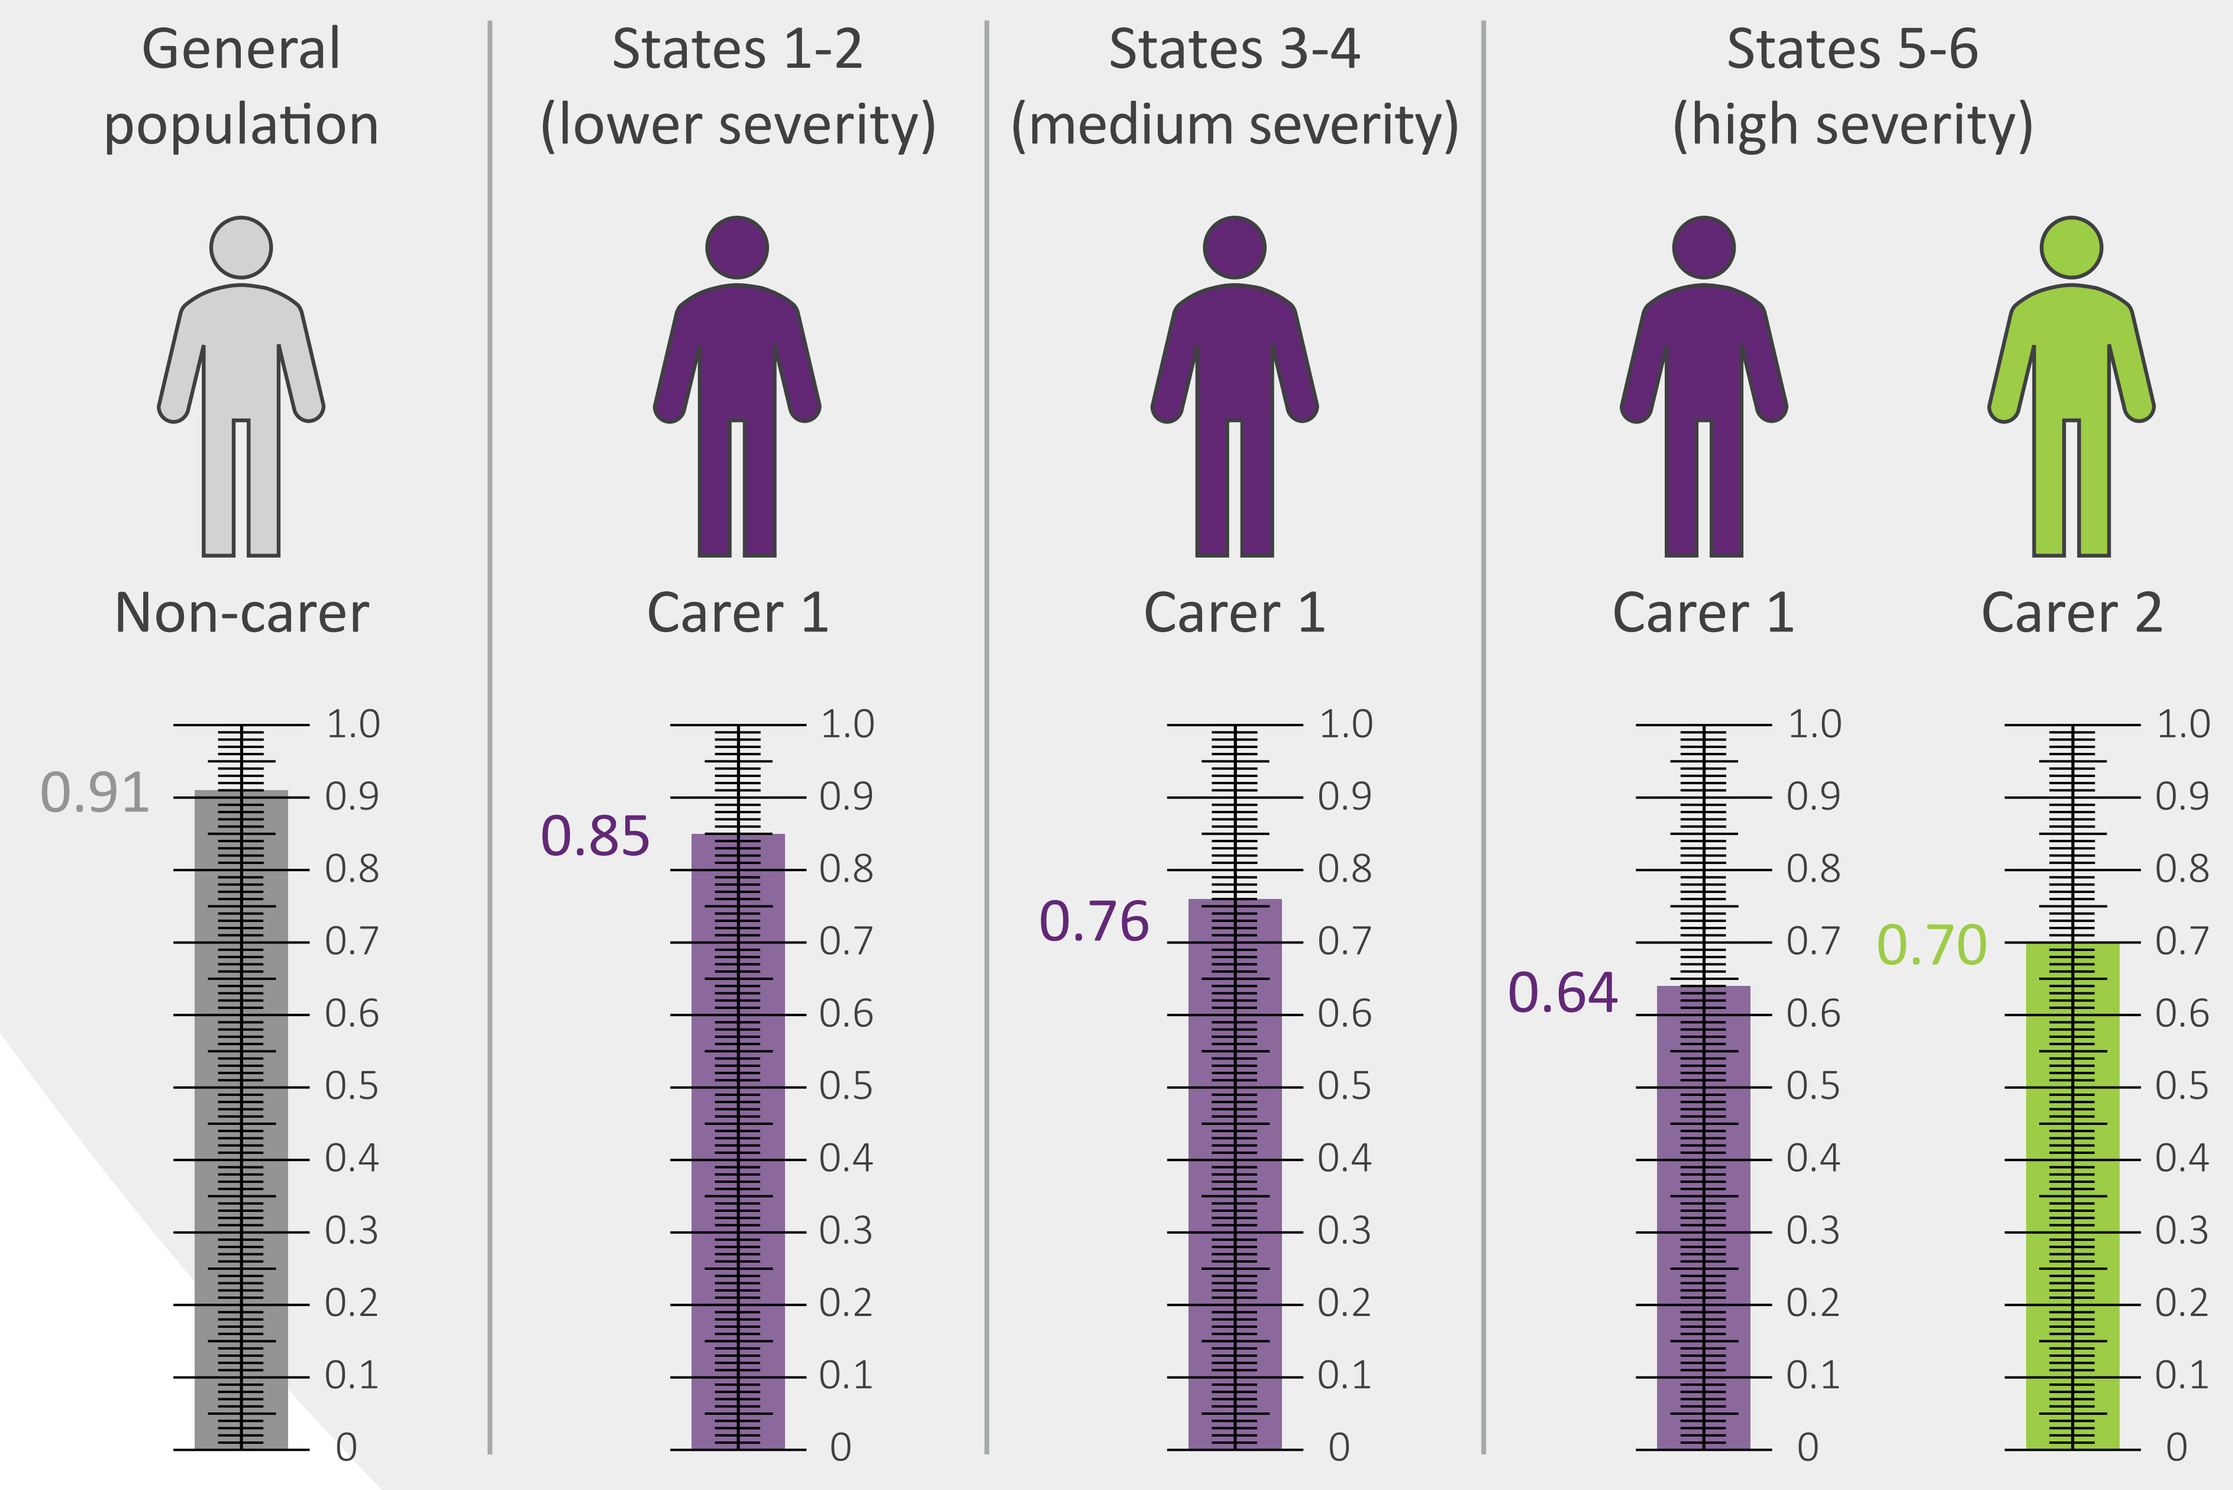

General
population
States 1-2
(lower severity)
States 3-4
(medium severity)
States 5-6
(high severity)
Non-carer
Carer 1
Carer 1
Carer 1
Carer 2
0.91
0.85
0.76
0.64
0.70
1.0
0.9
0.8
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0

CONCLUSION

- The study provides an important first step in exploring the relative quality of life impact of disease where informal care is received from more than one source. More research is needed to validate estimates using alternative valuation methods, to explore potential substitution effects between carers and to assess the generalisability of results to other disease areas in which multiple carers may be involved.
- The positive outcome achieved in spite of constraints in timeframe and reach to stakeholders demonstrates the value of targeted, pragmatic evidence generation for addressing HTA uncertainties.

This work was funded by Amryt Pharmaceuticals Ltd.

ISPOR Europe, 12-15 November 2023, Copenhagen

ABBREVIATIONS

BSAP, body surface area percentage; EB, Epidermolysis Bullosa; HRQoL, health-related quality of life; HTA, health technology assessment; ICER, incremental cost-effectiveness ratio; QALY, quality-adjusted life year; UK, United Kingdom; TTO, time trade-off.

REFERENCES

1. Noble-Longster, J; Russell, M; Ryder, S; Tolley, K (2022). Informal care disutility: how many caregivers are captured in cost-utility analyses of treatments for rare diseases submitted for NICE highly specialised technologies?

debra
THE BUTTERFLY SKIN CHARITY
Help stop the pain of EB

Tolley Health Economics
A bespoke approach to Health Technology Assessment