Poster EE93

Real-world cost-effectiveness of asfotase alfa for treatment of patients with HPP in England

Hazel Dawson[1], Tom O'Connell[2], Shona Fang[1], Katherine Kirkwood[3], Marjorie Crowell[2], Yaser Saraf[1]

[1]Alexion, AstraZeneca Rare Disease, London, UK; [2]Medicus Economics, LLC, Boston, MA; [3]Parexel International, Newton, MA

OBJECTIVES

- In 2017, a managed access agreement (MAA) was implemented providing coverage in England of asfotase alfa, an enzyme-replacement therapy for paediatric-onset hypophosphatasia (HPP).
- The MAA included an outcomes-based agreement (OBA), conditioning payments on realisation of treatment-related benefits.
- In this analysis, benefits observed under the MAA were compared to expectations specified in the OBA, to assess the real-world cost-effectiveness of asfotase alfa.

CONCLUSIONS

- Benefits of asfotase alfa observed under the MAA were consistent with expectations specified in the OBA, confirming real-world cost effectiveness.
- Among patients who initiated treatment at ages 0-1 years, none required invasive ventilation at age 2.
- For patients treated at ages ≥2 years, 102% of the health-related quality of life (HRQoL) gains expected under the MAA were realised.

PLAIN LANGUAGE SUMMARY

Why did we perform this research?
A managed access agreement which incorporated an outcomes based agreement was established between Alexion and the National Health Service (NHS) of England to provide interim funding of asfotase alfa, a therapy used to treat HPP, a rare disease which affects the bones. The analysis was performed to calculate whether a rebate was to be provided to the NHS.

How did we perform this research?
Clinical outcomes of patients treated during the managed access agreement were collected in a HPP registry and compared to the estimates produced in the cost-effectiveness analysis of asfotase alfa versus best supportive care.

What were the findings of this research and what are the implications?
Across the outcomes and age groups assessed, the benefits of asfotase alfa observed during the MAA were consistent with estimates produced in the cost-effectiveness analysis

Where can I access more information?
Information relating to the NICE highly specialised technology assessment of asfotase alfa can be found on: https://www.nice.org.uk/guidance/hst23

This study was funded by Alexion, AstraZeneca Rare Disease.
Poster presented at ISPOR Europe 2023 by Yaser Saraf

INTRODUCTION

- In 2017, a managed access agreement (MAA) was implemented providing coverage in England of asfotase alfa, an enzyme-replacement therapy for paediatric-onset hypophosphatasia, an ultra-rare condition associated with significant mortality in infants and morbidity in children and adults.
- The MAA specified an outcomes-based agreement, conditioning payments on realisation of benefits that were modeled in a cost-effectiveness analysis (CEA) of asfotase alfa vs best supportive care (BSC).
- Such benefits included (i) for patients starting treatment at ages 0-1 years, invasive-ventilation free survival at age 2 and (ii) for patients treated at ages ≥2, quality-adjusted life year (QALY) gains based on health-utility improvements.
- In this analysis, benefits observed under the MAA were compared against cost-effective benefit levels from the CEA, to assess the real-world cost-effectiveness of asfotase alfa.

METHODS

- The MAA's OBA compared actual (achieved) vs. predicted outcomes, with outcomes differing for treatment at ages 0-1 (perinatal/infantile onset) vs. ≥2 years.
 - Data were stored in an MAA Database managed by Alexion Pharmaceuticals, and patients could also elect for their data to be recorded in the HPP Global Registry.
- For treatment at ages 0-1 years, the outcome was ventilation-free survival at 2 years of age. Satisfaction of the outcome was binary:
 - If by the age of 2 years, a patient did not require invasive ventilation (excluding supplementary oxygen), 100% of the outcome was considered achieved.
 - In all other cases, including if a patient did not survive to age 2, 0% of the outcome was achieved.
- For treatment at ages ≥2 years, the outcome was QALY gains.
 - "Actual" outcomes reflected health utility measured with the PedsQL for ages 2-<18 years, and the EQ-5D-5L for ages ≥18 years.
 - "Predicted" outcomes reflected QALY gains estimated in Alexion's CEA of asfotase alfa vs. BSC for paediatric-onset HPP, submitted to England's National Institute for Health and Care Excellence (NICE).
- "Actual" health utility was estimated by mapping PedsQL (Khan et al., 2014) and EQ-5D-5L (van Hout et al., 2012) data to EQ-5D-3L based utilities, reflecting valuation according to Dolan (1997).
 - Change in utility was calculated by modelling "counterfactual" assumptions (i.e., how utility would have changed in the absence of treatment) according to disease progression and utility values modelled in Alexion's CEA submitted to NICE (see Lloyd et al., 2015).

Figure 1. Patient sample included in HRQoL analyses

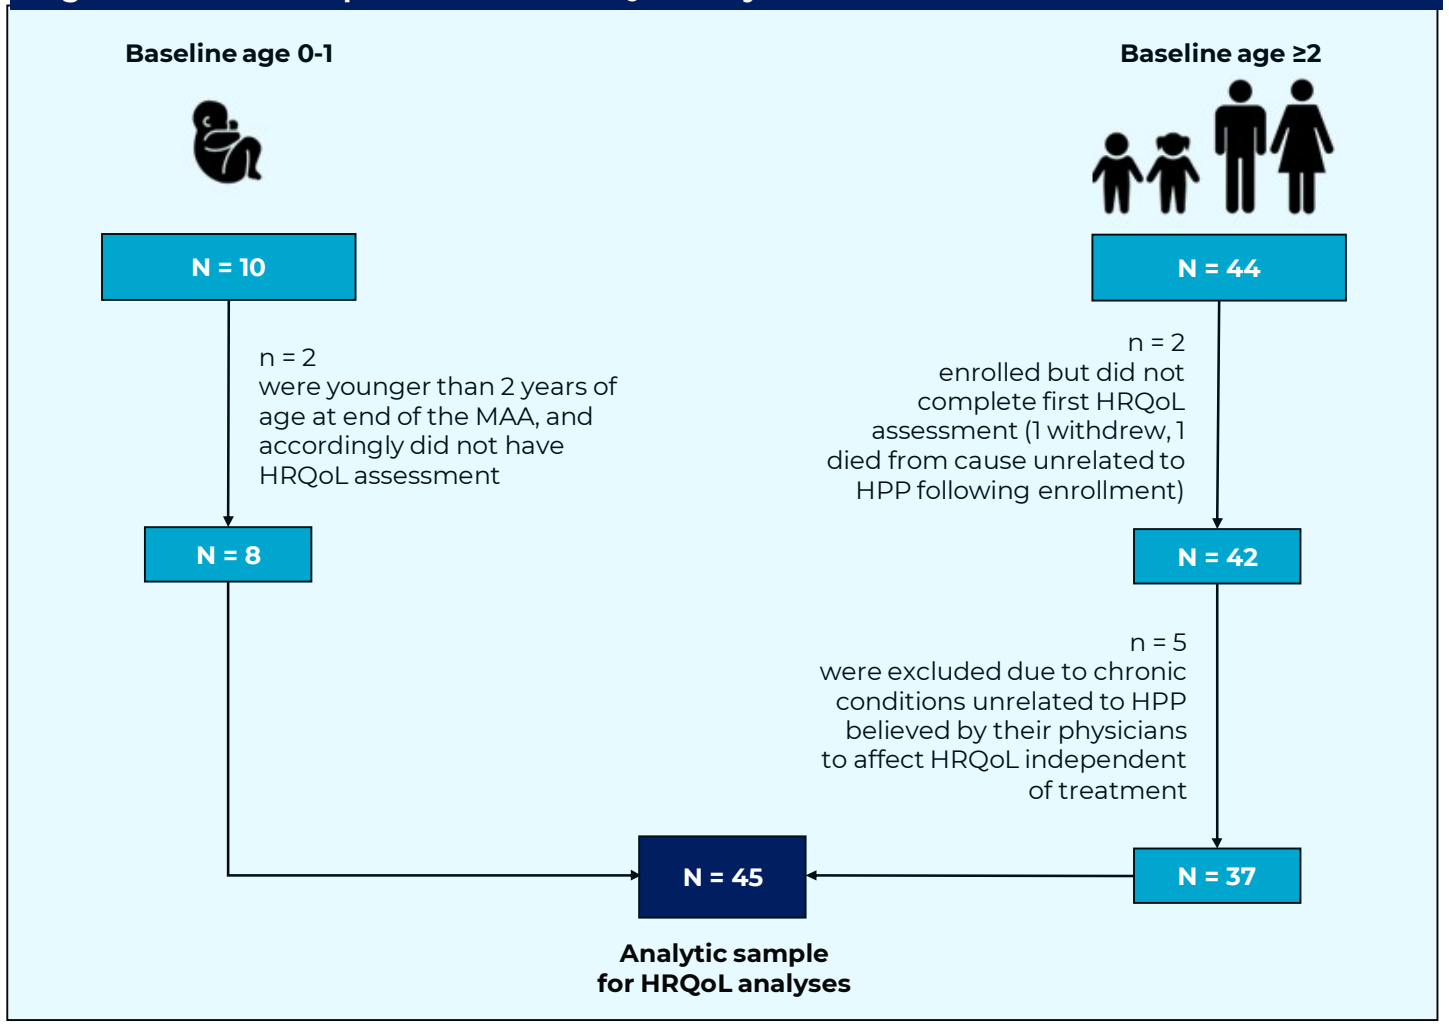
Baseline age 0-1
Baseline age ≥2
N = 10
n = 2
were younger than 2 years of age at end of the MAA, and accordingly did not have HRQoL assessment
N = 8
N = 44
n = 2
enrolled but did not complete first HRQoL assessment (1 withdrew, 1 died from cause unrelated to HPP following enrollment)
N = 42
n = 5
were excluded due to chronic conditions unrelated to HPP believed by their physicians to affect HRQoL independent of treatment
N = 37
N = 45
Analytic sample for HRQoL analyses

RESULTS AND INTERPRETATION

- The MAA enrolled 54 patients from November 2017 to October 2022.
 - N = 47 contributed to this analysis of effectiveness under the OBA, as outlined in **Figure 1**.
- Treatment was started at ages 0-1 by 10 patients.
 - All 8 patients who turned age 2 during the MAA were invasive-ventilation free (**Figure 2**).
- QALY gains were analysed in the 37 patients who started treatment at ages ≥2 and the 8 who turned 2 during the MAA.
 - Mean follow-up for HRQoL measurement was 29 months in the overall population.
 - In total, actual QALY gains exceeded predicted by 2.2% (48.7 vs. 47.7, **Figure 2**).
- Across the outcomes and age groups assessed, benefits of asfotase alfa observed under the MAA were consistent with the CEA estimates accepted by the NICE Evaluation Committee, confirming cost-effectiveness of outcomes achieved in terms of incremental costs of asfotase alfa vs. BSC.

Figure 2. Actual versus predicted benefits achieved in the MAA, by age group

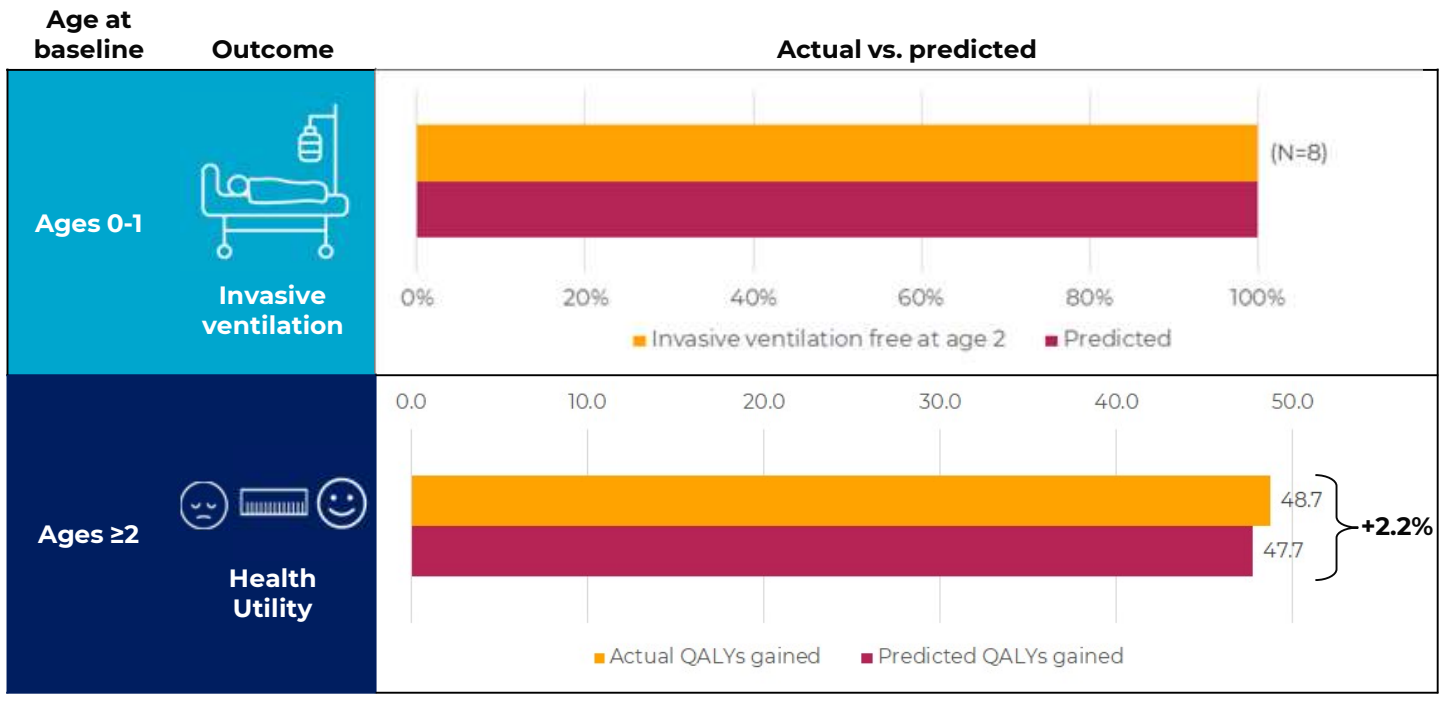
Age at baseline
Outcome
Actual vs. predicted
Ages 0-1
Invasive ventilation
(N=8)
0%
20%
40%
60%
80%
100%
Invasive ventilation free at age 2
Predicted
Ages ≥2
Health Utility
0.0
10.0
20.0
30.0
40.0
50.0
48.7
47.7
+2.2%
Actual QALYs gained
Predicted QALYs gained

Ages 0-1 years

Survival and respiratory outcomes

In the natural history of perinatal / infantile onset HPP, respiratory compromise and mortality (estimated 73% by age 5[5]) is common.

Of the 10 patients treated during the MAA:

- All survived through end of follow-up
- None required invasive ventilation at age 2

Ages ≥2 years

Health-related quality of life (HRQoL)

Paediatric-onset HPP significantly affects HRQoL of patients (estimated to reduce annual utility/QALYs by >0.6 on a 0-1 scale[4]).

Among the 45 patients with HRQoL data collected during the MAA:

- Over mean follow-up of 29 months, mean QALY gains were 1.1 per patient (0.44 per year), as reflected in **Figure 3**

Figure 3. Mean actual QALY gains, by duration of HRQoL follow-up

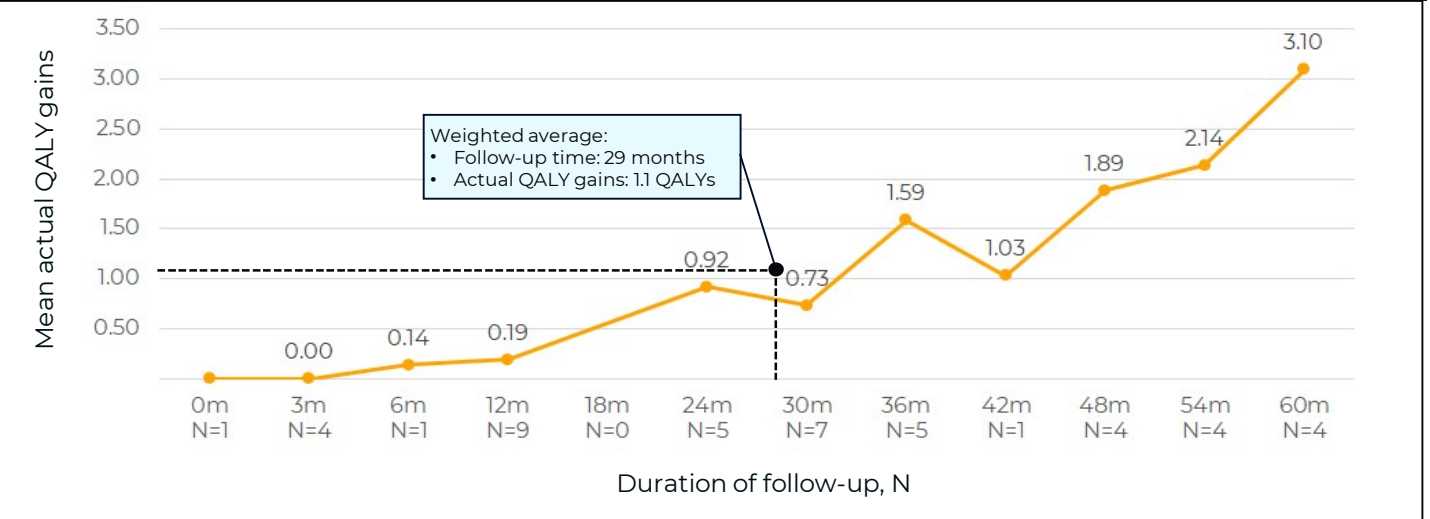
Mean actual QALY gains
3.50
3.00
2.50
2.00
1.50
1.00
0.50
Weighted average:
• Follow-up time: 29 months
• Actual QALY gains: 1.1 QALYs
0.00
0.14
0.19
0.92
0.73
1.59
1.03
1.89
2.14
3.10
0m N=1
3m N=4
6m N=1
12m N=9
18m N=0
24m N=5
30m N=7
36m N=5
42m N=1
48m N=4
54m N=4
60m N=4
Duration of follow-up, N

Actual QALY gains were calculated as observed utility (estimated from PedsQL or EQ-5D-5L responses) minus "counterfactual" utility. For ages <2 at baseline, counterfactual utility was 0.00/0.24 if the patient did/did not require invasive ventilation before treatment (see Lloyd et al., 2015). For ages ≥2 at baseline, counterfactual utility was modelled as a linear reduction over time, based on change from baseline to 5 years for a BSC patient in the NICE CEA. For patients whose six-minute walk test (6MWT) at baseline corresponded to the most severe health state of the CEA (i.e., 74% of enrolled patients who started treatment at age ≥2), no reduction could be modelled; baseline utility was therefore carried forward as the counterfactual.

References

1. van Hout B, et al. Value Health. 2012 Jul-Aug;15(5):708-15.
2. Khan KA, et al. Pharmacoeconomics. 2014 Jul;32(7):693-706.
3. Dolan P. Med Care. 1997 Nov;35(11):1095-108.
4. Lloyd A, et al. Value Health. 2015;18(7):A651.
5. Whyte MP, et al. J Clin Endocrinol Metab. 2016 Jan; 101(1): 334–342

Acknowledgements
We extend our gratitude to the clinical experts who collected the data to inform the HPP registry, NHS England and NICE for their support in the MAA process, and the patients who participated in the MAA program. Their dedication and collaboration have been instrumental in improving healthcare outcomes for patients with HPP.

Please scan this quick response (QR) code with your smartphone camera or app to obtain a copy of these materials.

Copies of this poster obtained through this QR code are for personal use only and may not be reproduced without permission from the authors of this poster.

Poster presented at ISPOR Europe 2023, 12-15 November, Copenhagen, Denmark

Corresponding author email address: yaser.saraf@alexion.com