EPH264

The potential public health impact of the adjuvanted respiratory syncytial virus prefusion F protein vaccine among older adults in Italy: preliminary results of a Markov model analysis

Puggina A[1], Calabrò GE[2,3], Santacroce R[1], Rumi F[4], Elliot L[5], Basile M[4], Sharifova M[5], Marijam A[6]

[1]GSK, Verona, Italy; [2]Section of Hygiene, University Department of Life Sciences and Public Health, Università Cattolica del Sacro Cuore, Rome, Italy; [3]VIHTALI (Value in Health Technology and Academy for Leadership and Innovation), Spin-Off of Università Cattolica del Sacro Cuore, Rome, Italy; [4]Graduate School of Health Economics and Management (ALTEMS), Università Cattolica del Sacro Cuore, Rome, Italy; [5]FIECON, London, UK; [6]GSK, Wavre, Belgium.

RSV vaccination of older adults offers the potential to substantially reduce the RSV burden in Italy, and this evidence may help policy makers and clinicians make informed decisions about RSV vaccination among older adults in Italy.

Digital poster

SCAN ME

Introduction

Respiratory syncytial virus (RSV) is a **highly contagious respiratory virus** that has a substantial impact on the health of older adults (OA) aged ≥65 years, particularly for those with underlying comorbidities such as chronic obstructive pulmonary disease (COPD) and asthma, who are at risk of severe outcomes if infected.[1]

RSV infection in OA in Italy is estimated to lead to >291,000 RSV-associated acute respiratory infections, >26,000 hospitalizations, and >1,800 deaths each year,[2] but **the burden of RSV is underestimated due to lack of surveillance and underreporting**.

Objectives

Estimation of public health impact of the adjuvanted RSVPreF3 OA vaccine[3] in comparison with no vaccination in OA in Italy

- In the **influenza vaccination target population** (≥65-year-olds)[4]
- **Two scenario analyses were performed to explore the impact of different vaccination coverage (VC) rates:**
 - **Scenario 1:** VC rate was assumed to be the same as for influenza vaccines in the 2022-2023 season (≥65-year-olds: 56.7%)[5]
 - **Scenario 2:** VC rate was assumed to be the optimal target for influenza vaccines (≥65-year-olds: 95.0%)[4]

Methods

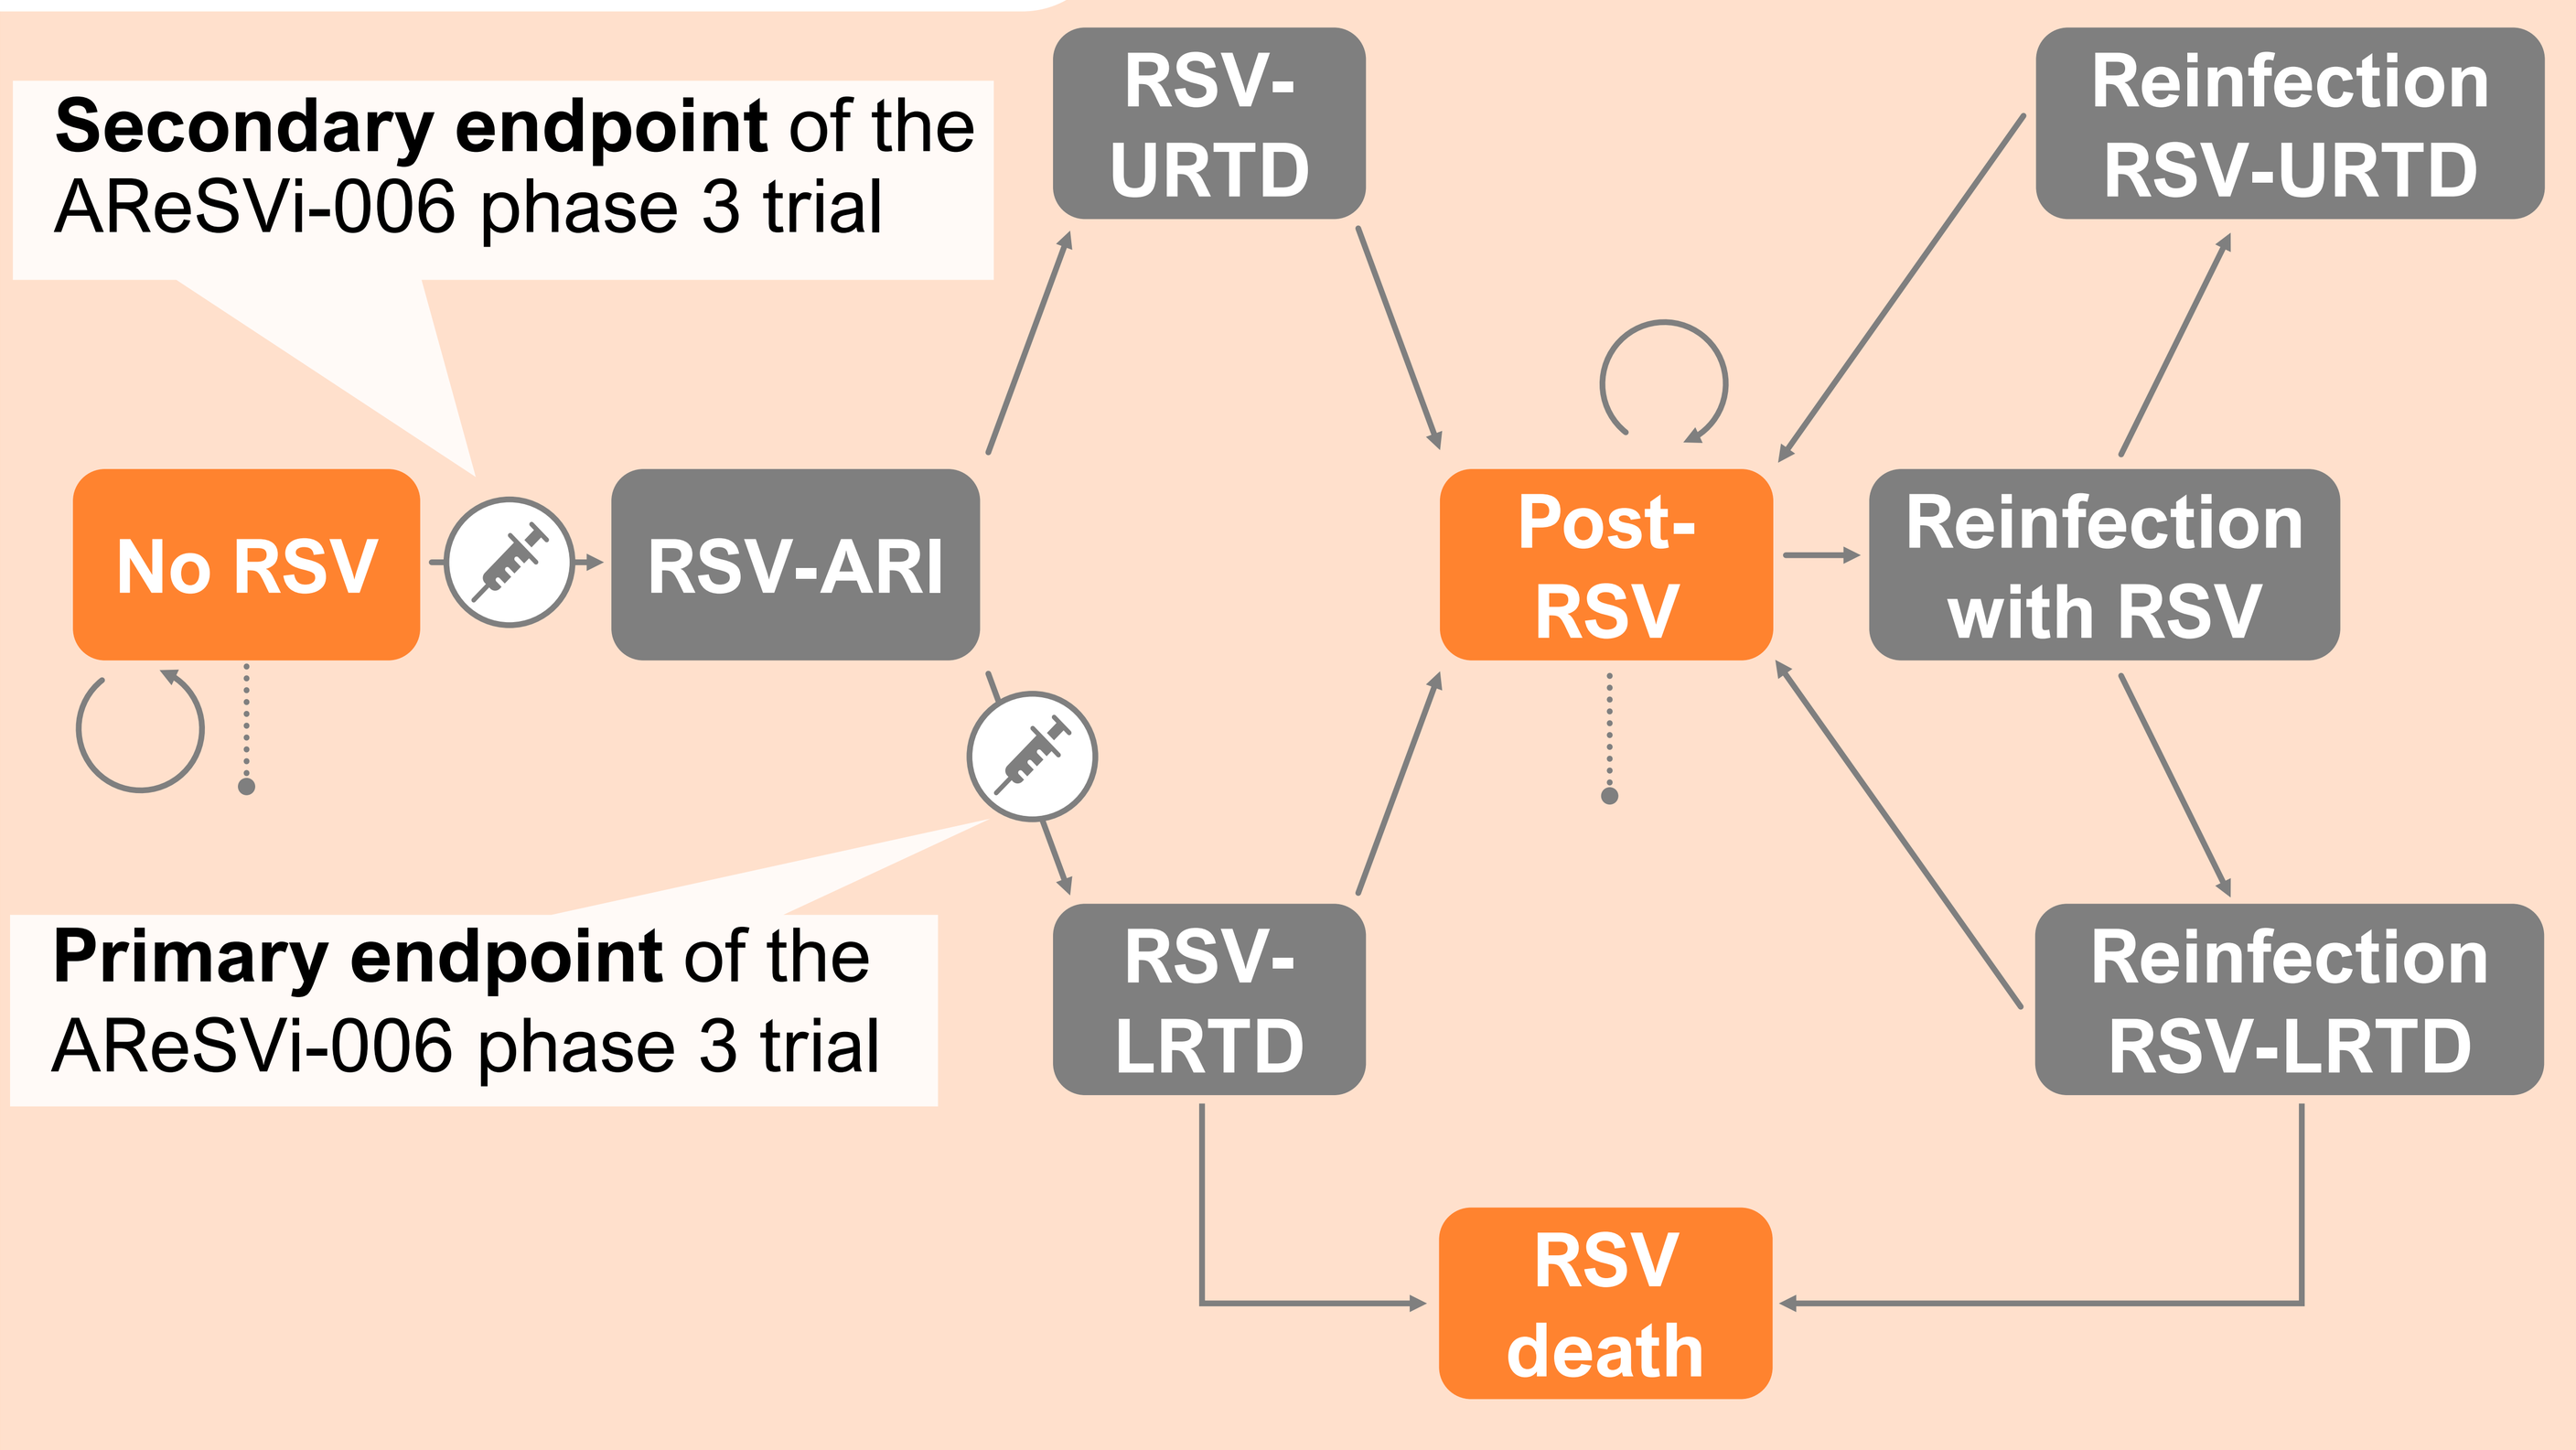
Secondary endpoint of the AReSVi-006 phase 3 trial
Primary endpoint of the AReSVi-006 phase 3 trial
No RSV
RSV-ARI
RSV-URTD
RSV-LRTD
Post-RSV
Reinfection with RSV
Reinfection RSV-URTD
Reinfection RSV-LRTD
RSV death
Death from other causes
Health state
Disease transition event

Multi-cohort Markov model with a 3-year time horizon, representing 3 RSV seasons

Adaptation to the Italian healthcare setting

Demographic, epidemiologic, and cost data were sourced from **published literature**

Vaccine efficacy (VE) and waning rates were informed by the **AReSVi-006 phase 3 clinical trial.**[6]

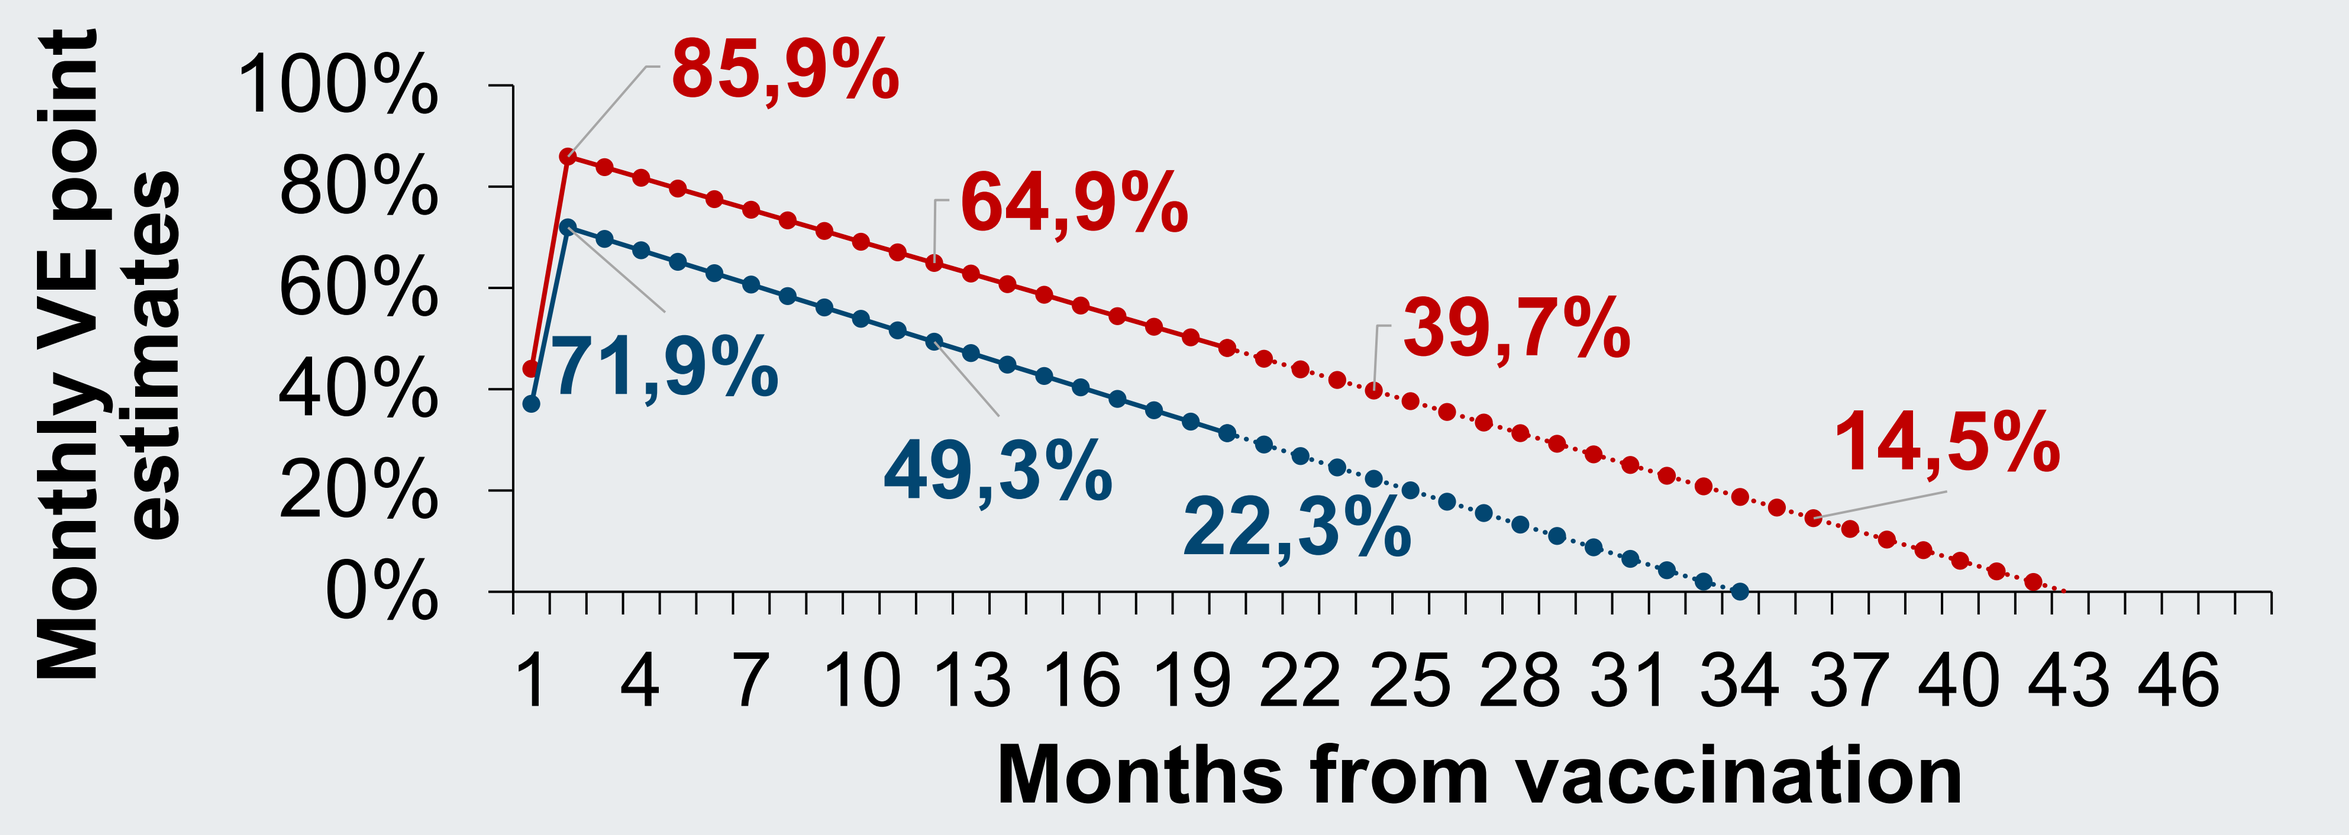
Monthly VE point estimates
100%
80%
60%
40%
20%
0%
85,9%
64,9%
39,7%
14,5%
71,9%
49,3%
22,3%
1 4 7 10 13 16 19 22 25 28 31 34 37 40 43 46
Months from vaccination

	Peak VE	Monthly Waning
RSV LRTD	88.0% (LB: 65.8%; UB: 99.2%)	-2.1% (LB: -0.1%; UB: -4.3%)
RSV ARI	74.2% (LB: 56.4%; UB: 94.0%)	-2.2% (LB: -0.3%; UB: -4.3%)

Results

The model estimates that, compared to no vaccination, adjuvanted RSVPreF3 OA vaccine would substantially reduce the burden of RSV among Italian adults aged ≥65 years by preventing RSV-LRTD events and their associated complications, hospitalizations, and deaths.

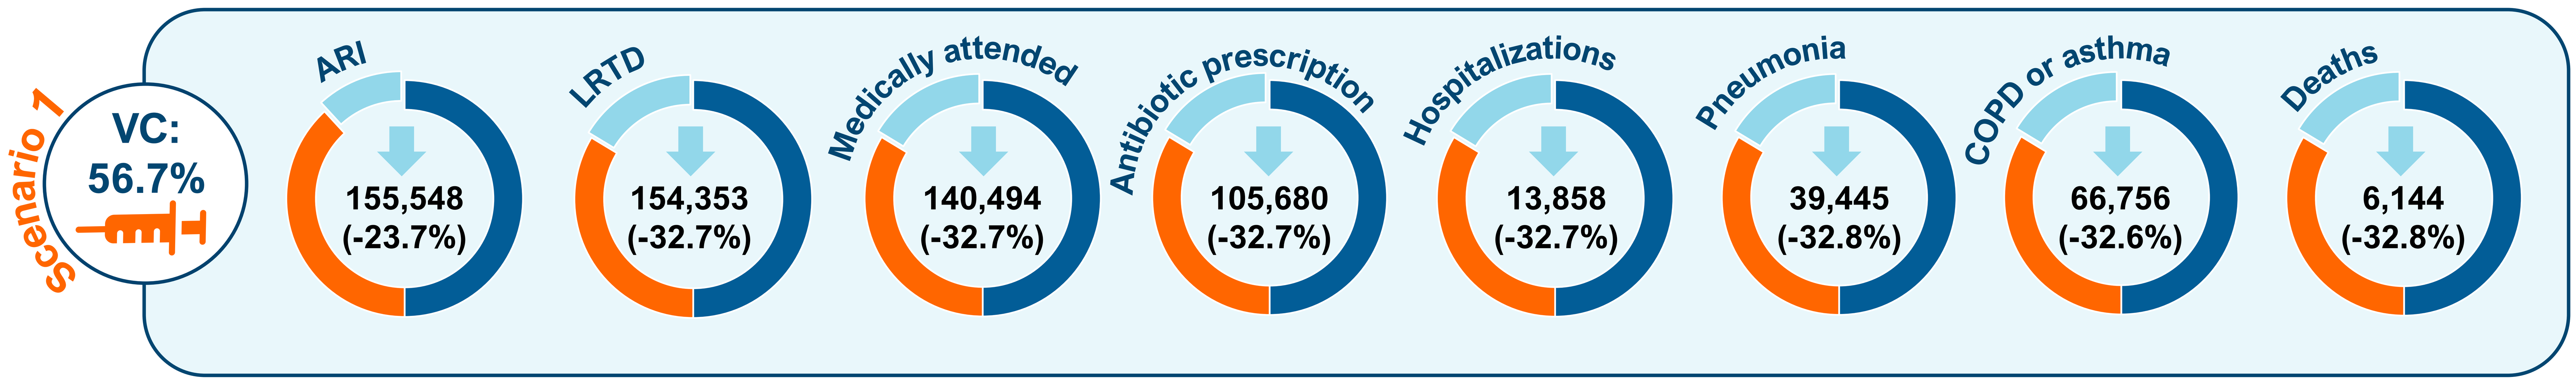
scenario 1
VC: 56.7%
ARI
155,548 (-23.7%)
LRTD
154,353 (-32.7%)
Medically attended
140,494 (-32.7%)
Antibiotic prescription
105,680 (-32.7%)
Hospitalizations
13,858 (-32.7%)
Pneumonia
39,445 (-32.8%)
COPD or asthma
66,756 (-32.6%)
Deaths
6,144 (-32.8%)

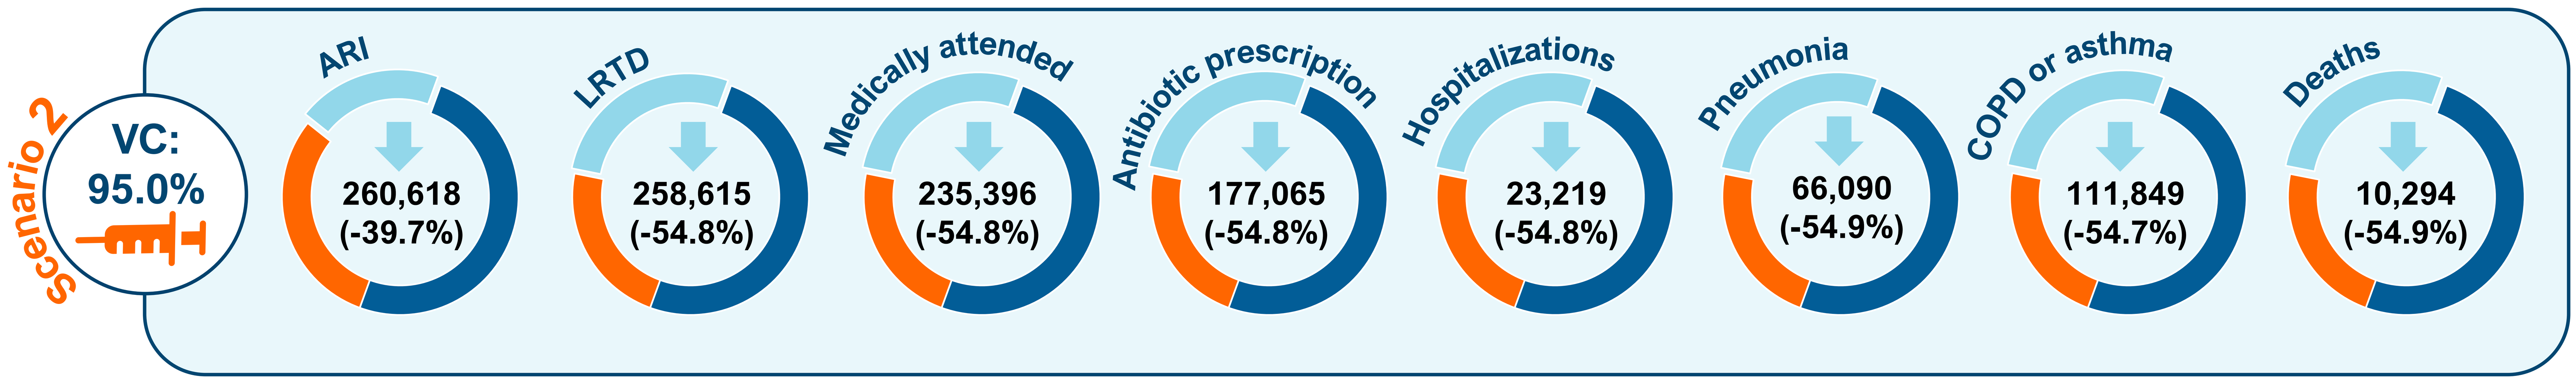
scenario 2
VC: 95.0%
ARI
260,618 (-39.7%)
LRTD
258,615 (-54.8%)
Medically attended
235,396 (-54.8%)
Antibiotic prescription
177,065 (-54.8%)
Hospitalizations
23,219 (-54.8%)
Pneumonia
66,090 (-54.9%)
COPD or asthma
111,849 (-54.7%)
Deaths
10,294 (-54.9%)

Based on modelled population size and estimated RSV vaccine coverage, 8,038,611 OA and 13,468,573 OA were assumed to receive RSVPreF3 OA in the vaccinated arm of the analysis.

No vaccine RSVPreF3 OA Averted

Conclusions

- Assuming the same vaccination coverage as for influenza vaccines in the 2022-2023 season, the analysis predicts that, compared to no vaccination, the adjuvanted RSVPreF3 OA vaccine would approximately prevent 32.7% of RSV LRTD cases over 3 years among Italian OA aged ≥65 years, resulting in a €231M cost reduction related to avoided healthcare resource utilization and associated costs.
- The model predicts that vaccinating 95.0% of the Italian OA population with the adjuvanted RSVPreF3 OA vaccine would prevent 54.8% of RSV-LRTD events and their associated hospitalizations vs. no RSV vaccination, amounting to a reduction of €388M in direct clinical costs over the 3 years.
- The number needed to vaccinate (NNV) with a single dose of the adjuvanted RSVPreF3 OA vaccine to prevent one RSV-LRTD event among the Italian OA population is estimated to be 52 over a 3-year time horizon, representing 3 RSV seasons.
- RSV vaccination offers the potential to substantially reduce the RSV burden in Italy, and this evidence may help policy makers and clinicians make informed decisions about RSV vaccination among OA in Italy.

Abbreviations

AReSVi006: AReSVi006 (Adult Respiratory Syncytial Virus) phase III clinical trial; **ARI:** acute respiratory illness; **COPD:** Chronic obstructive pulmonary disease; **VC:** Vaccine Coverage; **LRTD:** lower respiratory tract disease; **OA:** older adults aged ≥65 years; **RSV:** respiratory syncytial virus; **RSVPreF3:** Respiratory Syncytial Virus Prefusion F Protein Vaccine disease; **URTD:** upper respiratory tract disease; **VE:** Vaccine Efficacy

References

1. Falsey AR, Walsh EE. Drugs Aging 2005; 22: 577–587.
2. Savic M, et al. Influenza Other Respi Viruses. 2022;1–10.
3. https://www.ema.europa.eu/en/medicines/human/EPAR/arexvy access date: 01 October 2023
4. Link at: renderNormsanPdf (salute.gov.it) access date: 01 October 2023
5. Link at: https://www.epicentro.iss.it/vaccini/dati_ita#flu access date: 01 October 2023
6. Papi A. et al., N Engl J Med 2023. 388: 595-608.

Acknowledgements

The authors would like to thank Eleftherios Zarkadoulas for his contributions to this study. Business & Decision Life Sciences platform provided editorial assistance and publications coordination, on behalf of GSK.

Disclosures

Funding by GlaxoSmithKline Biologicals SA (GSK study identifier: VEO-000627). A. Puggina, R. Santacroce and A. Marijam are GSK employees and hold shares in GSK. G.E. Calabro is an employee of VIHTALI. L. Elliot and M. Sharifova are employees of FIECON, entity that received payment from GSK for this work. These authors declare no other financial and non-financial relationships and activities. R. Filippo and M. Basile declare no financial or non-financial relationships and activities and no conflicts of interest.

ISPOR Europe | 12-15 November 2023 | Copenhagen, Denmark

Presenting Author: Anna Puggina anna.x.puggina@gsk.com

© 2023 GSK