EE105

Ghetti G[1], **Porta C**[1], Pradelli L[1], Fascì A[2]

ANTI-VEGF USE IN NEOVASCULAR AGE-RELATED MACULAR DEGENERATION: AN ANALYSIS OF THE NATIONAL REPORT ON MEDICINES USE IN ITALY

[1] AdRes - Health Economics and Outcome Research, Turin, Italy; [2] Roche spa, Monza, Italy

Objective

- Neovascular age-related macular degeneration (nAMD) is the leading cause of vision loss in the elderly, negatively affecting patients' quality of life.
- Anti–vascular endothelial growth factor therapies (anti-VEGFs) are the current standard of care for nAMD. However, real-world studies revealed suboptimal outcomes due to the frequent need for therapeutic interventions, burdening patients and healthcare systems [1].
- The objective of this study was to analyze the consumption of anti-VEGFs in Italy and present potential strategies to enhance treatment appropriateness.

Methods

Medicine use in Italy

- The consumption of on-label anti-VEGFs in the first year of treatment was analyzed through the 2020/2021 national reports produced by the National Observatory on the Use of Medicines (OsMed) [2,3].
- Approved on label anti-VEGFs were classified based on their ability to provide extended treatment intervals: <7 (IVT<7), 7 (IVT7), and >7 (IVT>7) injections/year, according to Nota 98 prescription appropriateness report (Table 1) [4].
- Appropriateness was defined as the ratio of the observed annual mean injections number with Nota 98 report and Summary of Product Characteristics (SmPC) value.

Table 1 – Approved anti-VEFGs

Category	Injection/year*
IVT<7	6.00
IVT7	7.00
IVT>7	9.95

* According to Nota 98 report and SmPC

Optimization problem

- Two constrained optimization models were developed in Microsoft Excel, using a linear solver. These models were designed to identify the optimal Treatment Allocation (TA) (Table 2), with the following objectives:
 - **M1** To maximize the appropriateness without increasing the budget (S1).
 - **M2** To minimize the expenditure while achieving an 80% appropriateness level (S2).
- Additional scenario analyses were carried out to evaluate M2 outcomes, assuming:
 - a 65% price reduction of IVT>7 due to the introduction of biosimilars (S3);
 - the future availability of a novel anti-VEGF treatment requiring less than 6 injections/year (IVT<6) as faricimab (S4).

Table 2 – Optimization models and scenarios

	Objective function	Predefined constraints	Scenarios
M1	Difference between theoretical and real-world appropriateness	Total costs ≤ current budget 5% ≤ anti-VEGFs share ≤ 95%*	**(S1)** fixed expenditure ceiling
M2	Expenditure at 80% appropriateness level	5% ≤ anti-VEGFs share ≤ 95%*	**(S2)** 80% appropriateness level **(S3)** S2+65% price reduction of IVT>7 **(S4)** S2+introduction of IVT<6

* It was assumed that the utilization percentages of anti-VEGFs would not decrease to zero

Economic inputs

National on label treatment acquisition and administration costs were considered (Table 3).

Table 3 – Unit costs

Cost items	Value (€)
Drug cost	
Brolucizumab	613.70
Aflibercept	667.85
Ranibizumab	669.66
Faricimab	631.92
Administration cost	268.15

- For drugs ex-factory price net of mandatory discounts was considered.
- Administration cost was derived from national tariffs [5].

Results

Anti-VEFG consumption

- **27,401 eyes** started an approved anti-VEGF therapy in Italy in 2021: 33.5% received IVT>7, 57.0% received IVT7, and 9.5% received IVT<7.
- The estimated **appropriateness** was **46.6%** (Figure 1), with a total **expenditure** of **€93.9** million for **100,851 injections** (Figure 2).

Figure 1 – Estimated appropriateness and mean number of injections

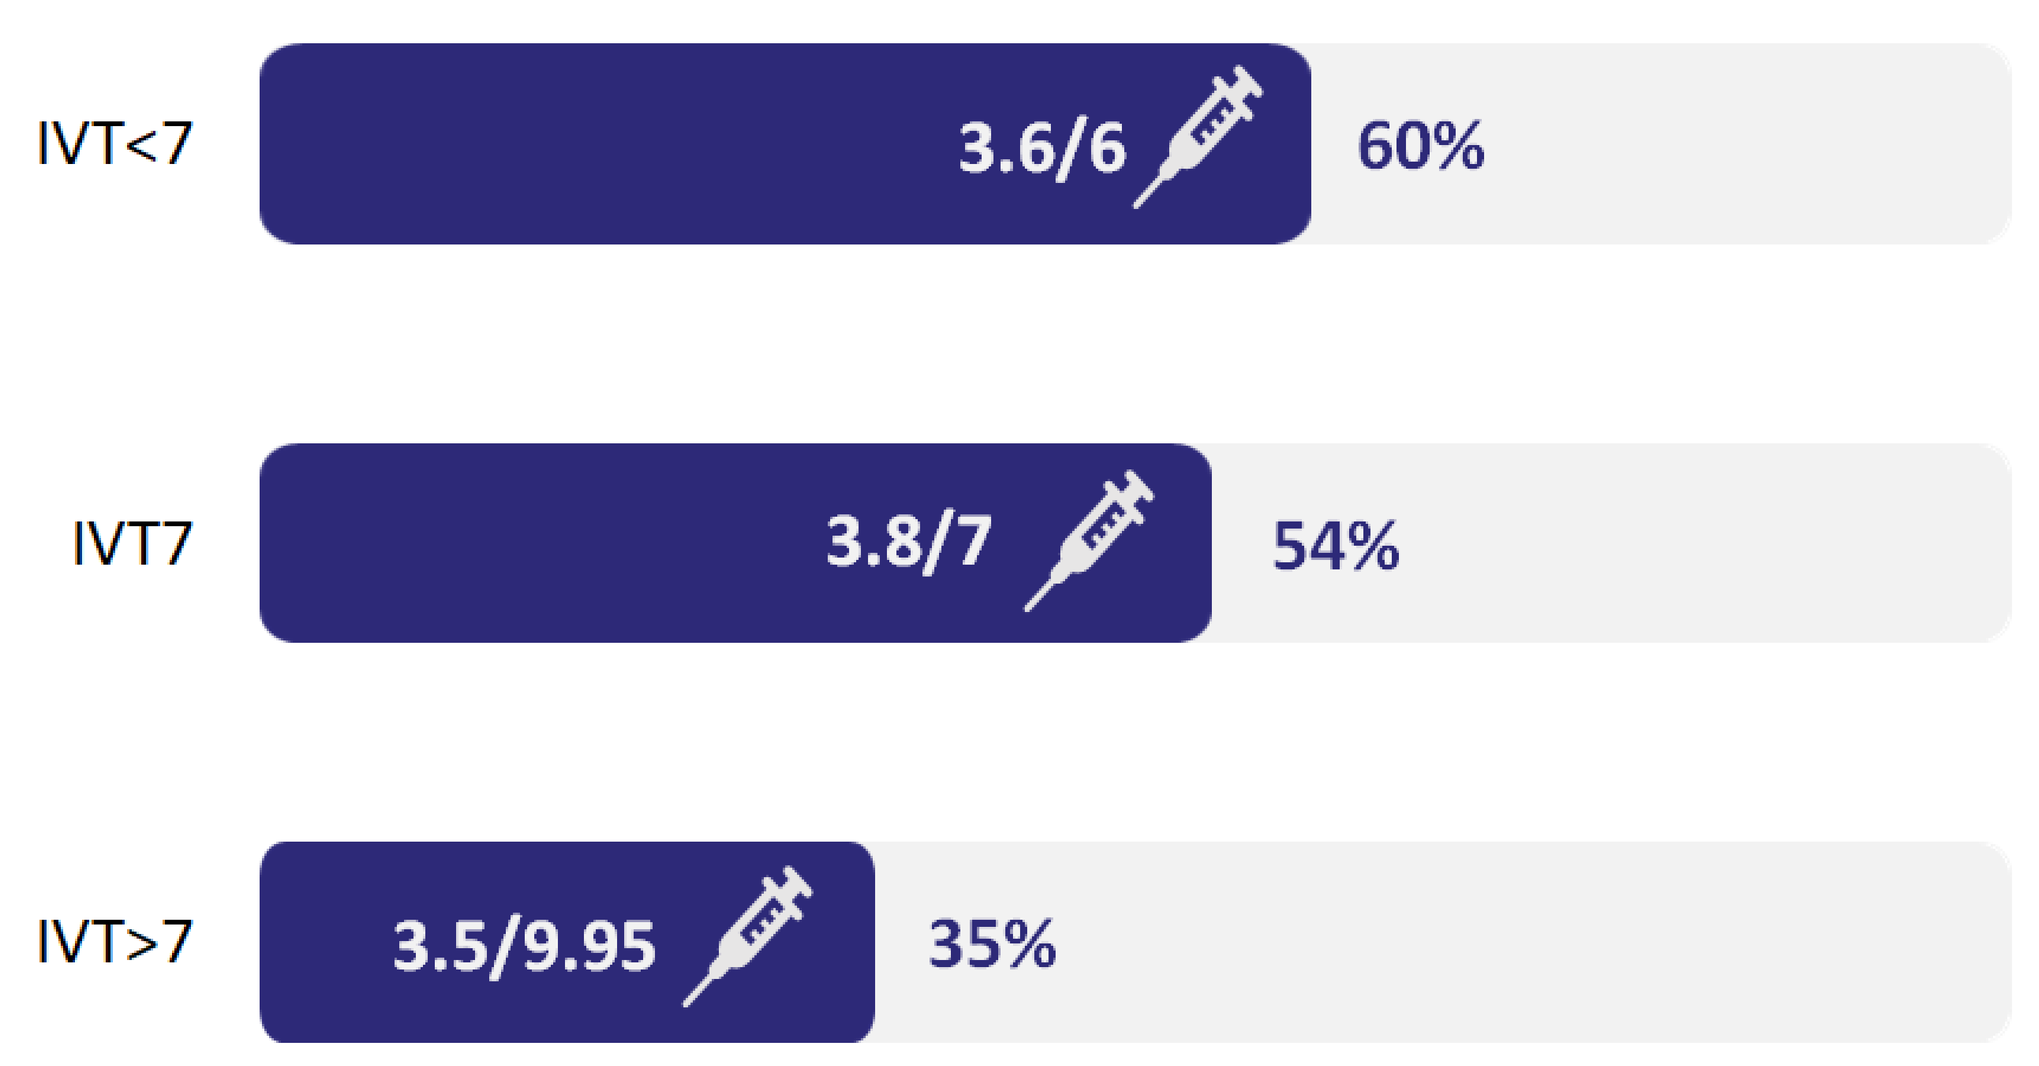
IVT<7
3.6/6
60%
IVT7
3.8/7
54%
IVT>7
3.5/9.95
35%

Figure 2 – Total expenditure and number of injections: current TA

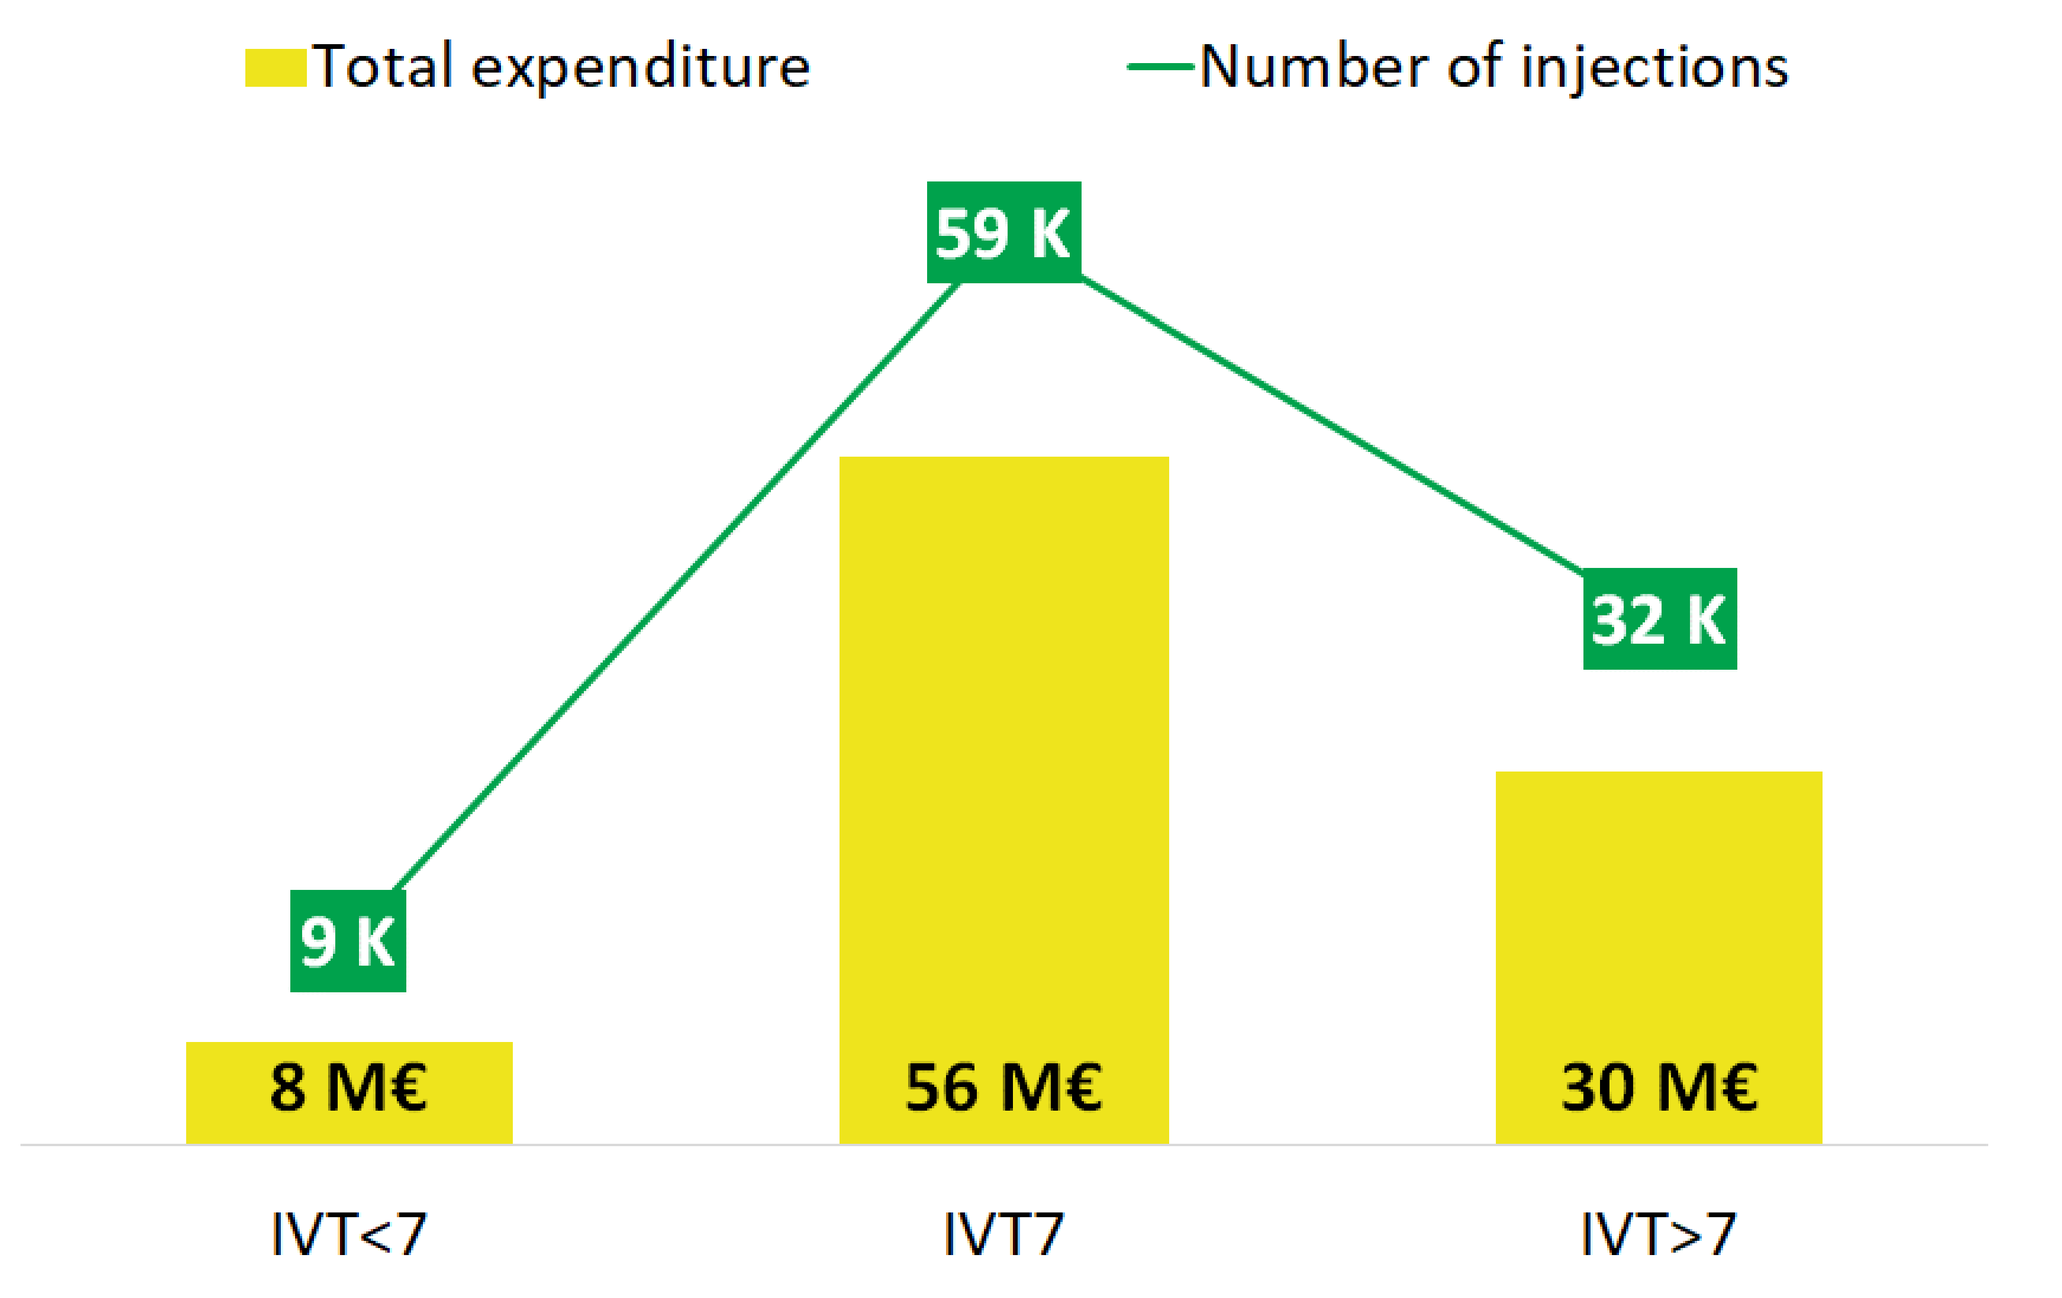
Total expenditure
Number of injections
9 K
59 K
32 K
8 M€
56 M€
30 M€
IVT<7
IVT7
IVT>7

Strategies to enhance treatment appropriateness

S1 **Greater use of IVT<7 products** can increase **appropriateness** to **57.7%** (Figure 3), resulting in a reduction of both injections (-2.1%) and expenditure (-6.7%) (Figure 4).

S2 **Greater use of IVT<7 products** can lead to **achieving 80% appropriateness,** requiring **higher injection capacity (+35.8%)** and **increased expenditure (+29.6%)**.

S3 In the event of **IVT>7 price reduction**, their increased utilization could **mitigate the rise in expenditure (+18.8%).** However, achieving 80% appropriateness would require a **massive** increase in **injection capacity (+108.8%)**.

S4 Considering anti-VEGF with <6 injections/year (**IVT<6**), achieving 80% appropriateness required **less investments (+22.9%)** and only a **modest increase of injections (+26.6%)**.

Figure 3 – Anti-VEGF distribution (current TA vs. best TA) and appropriateness by scenario

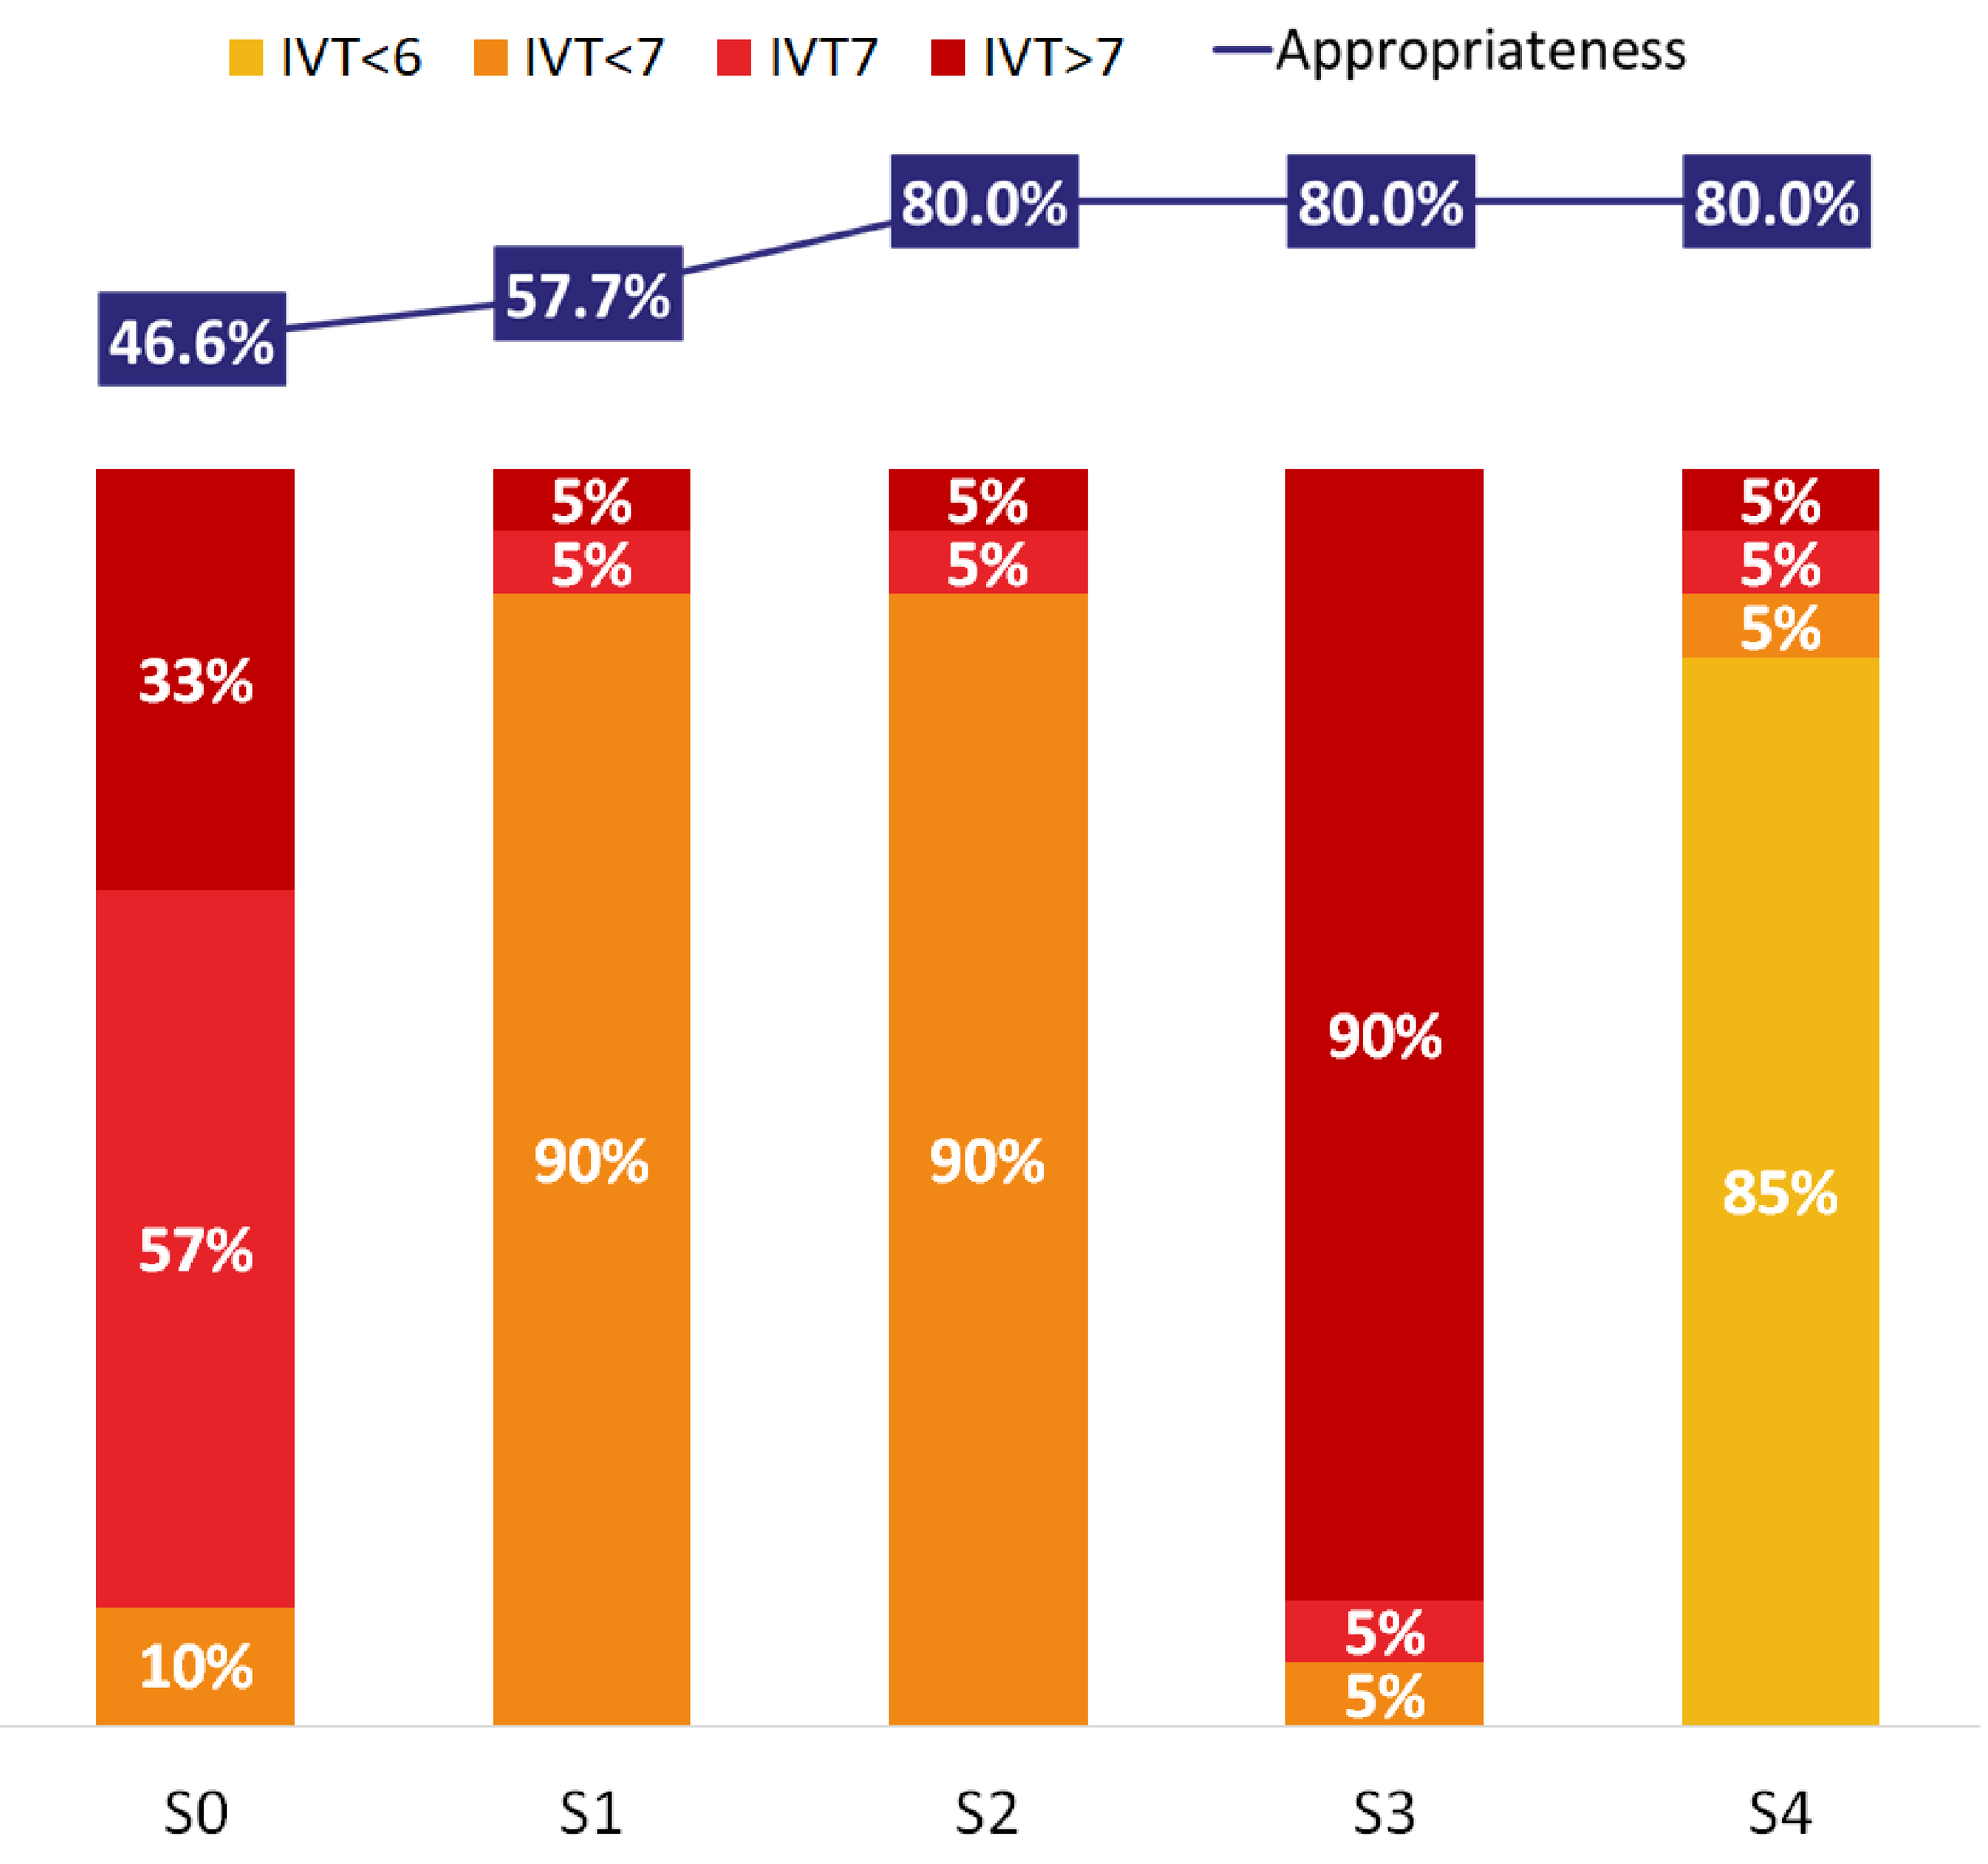
IVT<6
IVT<7
IVT7
IVT>7
Appropriateness
46.6%
57.7%
80.0%
80.0%
80.0%
33%
57%
10%
5%
5%
90%
5%
5%
90%
90%
5%
5%
5%
5%
5%
85%
S0
S1
S2
S3
S4

Figure 4 – Total expenditure and number of injections: current TA vs. best TA

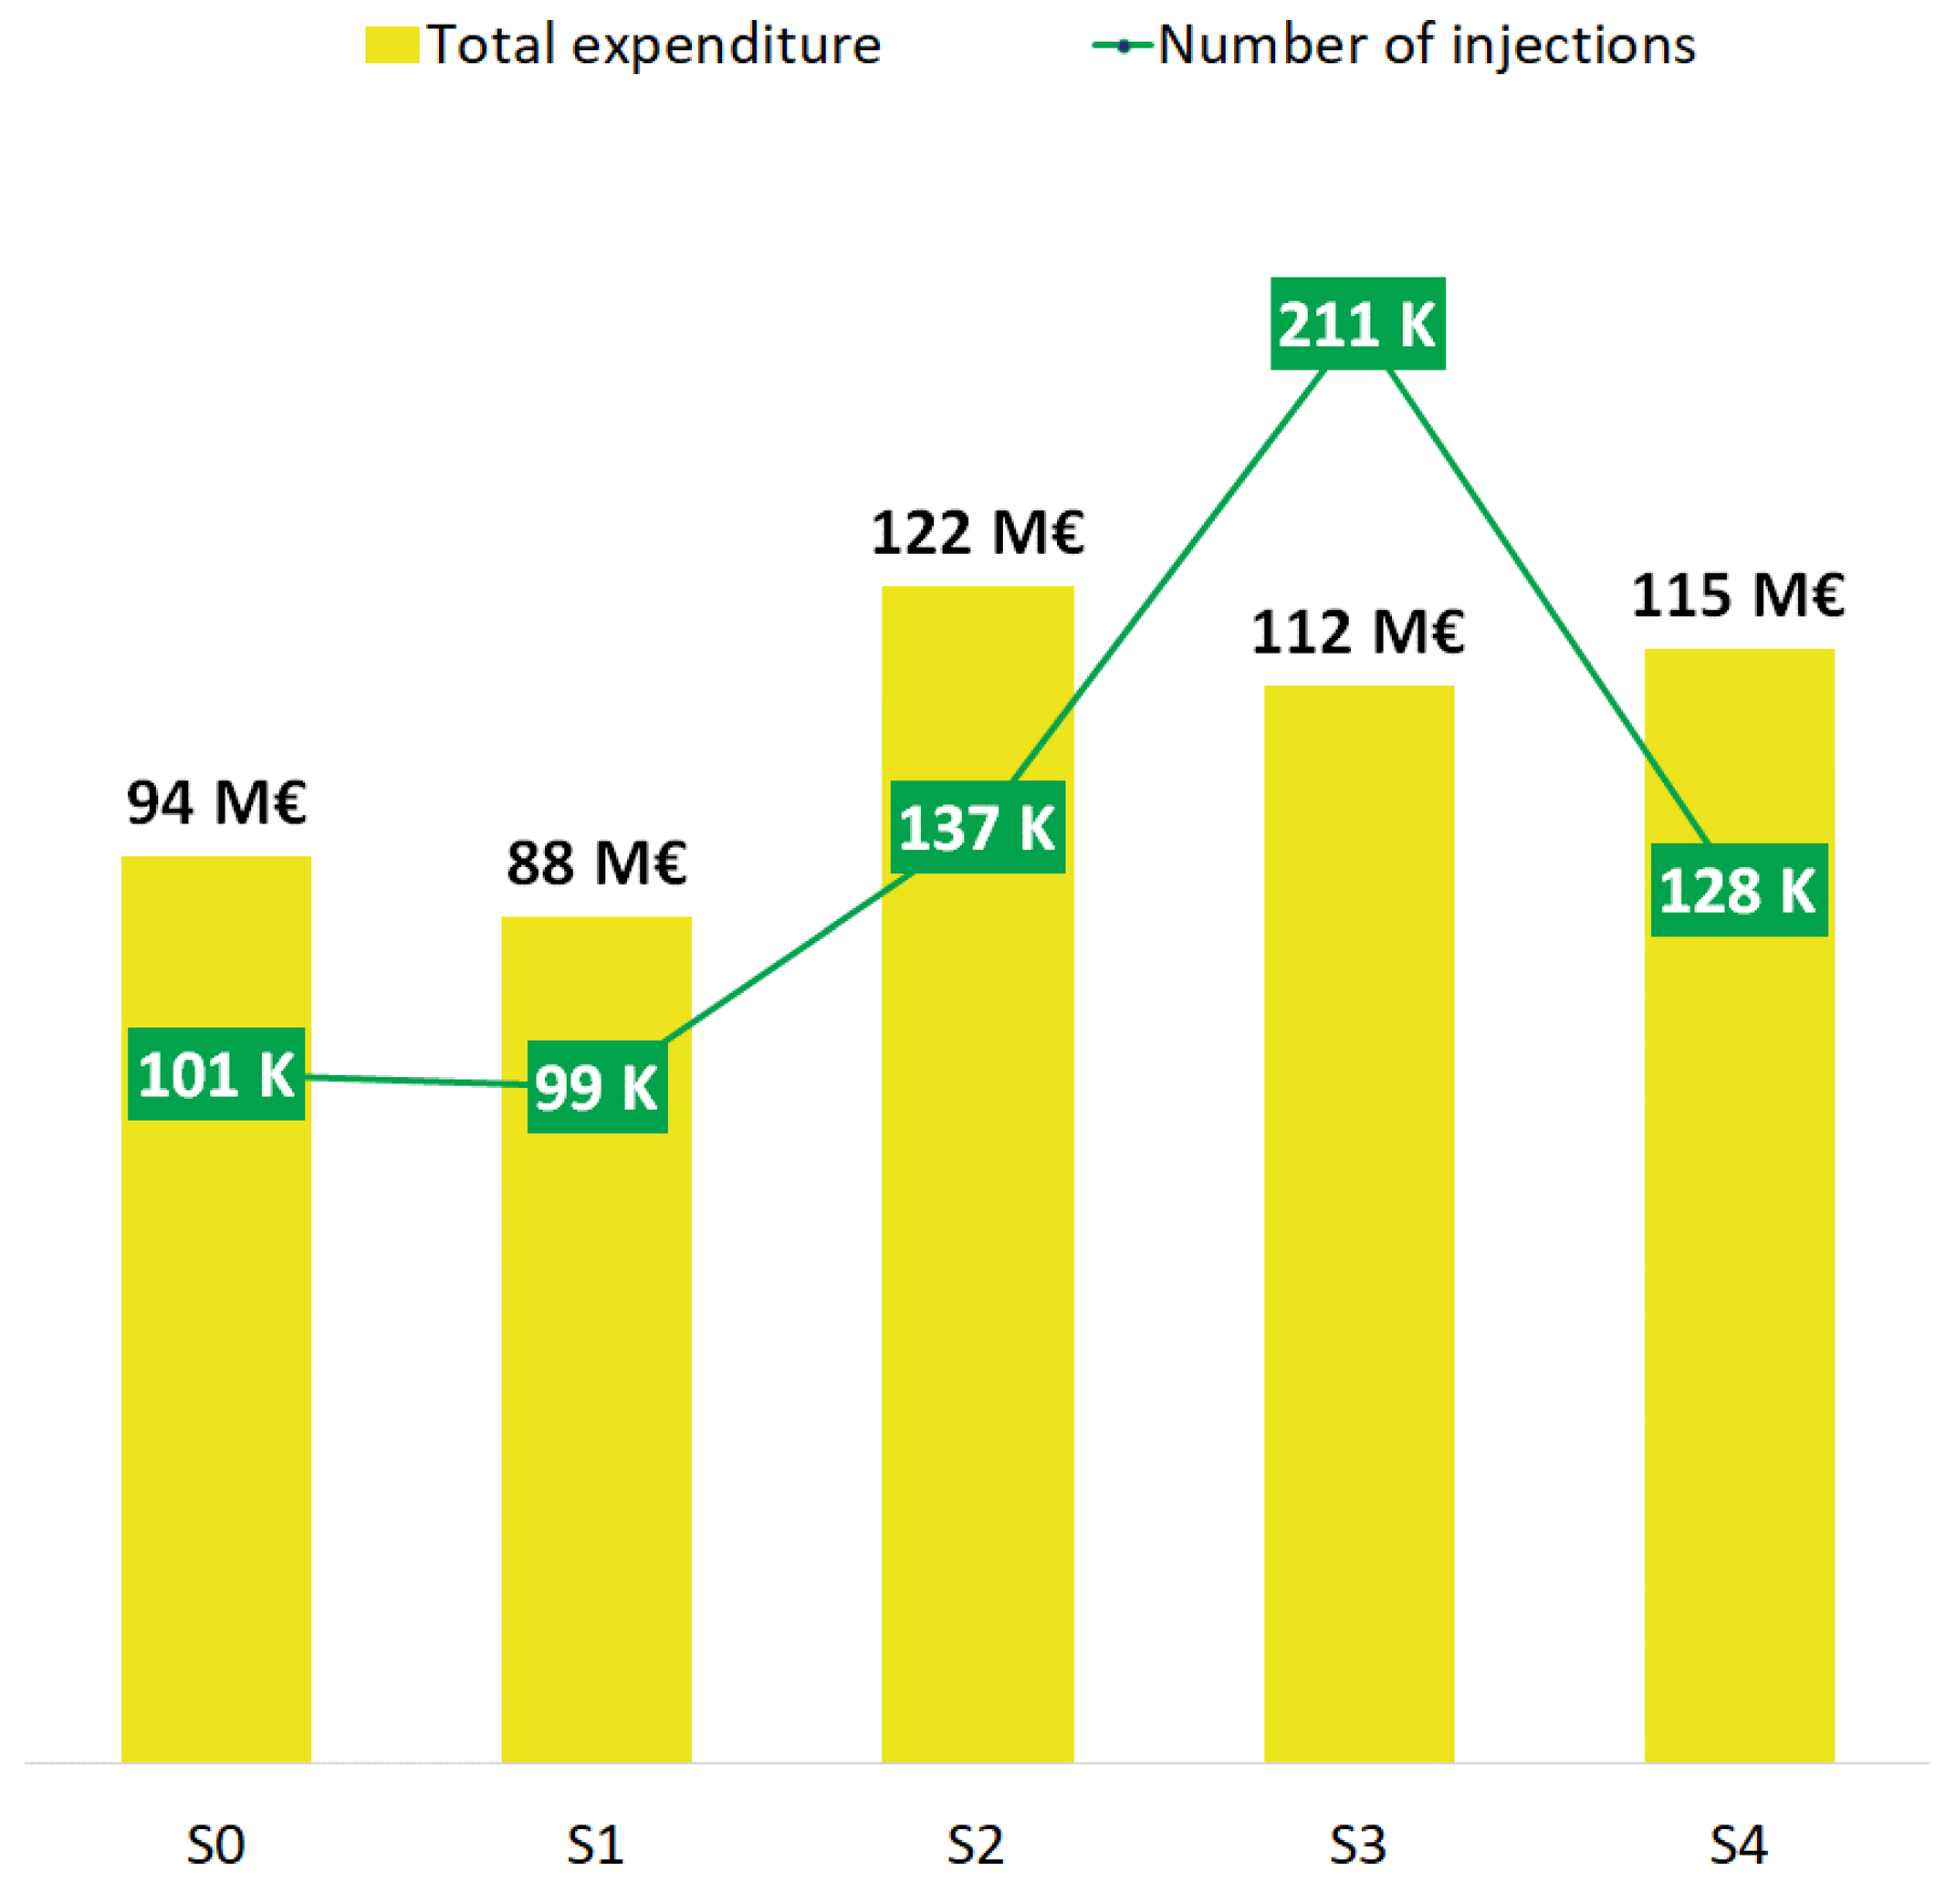
Total expenditure
Number of injections
94 M€
88 M€
122 M€
112 M€
115 M€
101 K
99 K
137 K
211 K
128 K
S0
S1
S2
S3
S4

Conclusions

A sub-optimal appropriateness was observed across all anti-VEGF drugs, with an uneven distribution of patients favouring IVT7 and IVT>7 drugs.
Greater use of IVT<7 products can enhance appropriateness, up to an additional 11.1% in the fixed expenditure ceiling scenario.
Achieving 80% appropriateness depends on the **capacity of treatment centers** and the **financial feasibility** for payers, with **durable products** potentially favouring patients' adherence.

- While using **discounted IVT>7 products** may appear economically advantageous, it may be unfeasible due to the **excessive number of injections needed**.
- The **availability of IVT<6 products** resulted in achieving 80% appropriateness with a lower economic effort and a limited increase in injections.

References

1. Holz FG et al. Br J Ophthalmol. 2015;99(2):220-6.
2. OsMed 2020. Medicine use in Italy.
3. OsMed 2021. Medicine use in Italy.
4. AIFA. Nota 98.
5. Intesa Conferenza Stato e Regioni. Seduta 14/04/2023

ISPOR Copenhagen 2023

Acknowledgements This analysis was funded by Roche spa, Italy