EE637 - ECONOMIC IMPACT OF MANAGING INVASIVE MOLD DISEASE WITH ISAVUCONAZOLE COMPARED WITH LIPOSOMAL AMPHOTERICIN B IN SPAIN

Moya-Alarcón C[1], Gálvez-Santisteban M[1], Azanza JR[2], Barberán J[3], Ferrer R[4], Kwon M[5], Moreno A[6], Rubio-Terrés C[7], Peral C[1]

1. Pfizer S.L.U, Madrid, Spain; 2. Universidad de Navarra, Pamplona, Spain; 3. Hospital HM Montepríncipe, Madrid, Spain; 4. Hospital Universitari Vall d'Hebron, Barcelona, Spain; 5. Hospital Gral. Univ. Gregorio Marañón, Institute of Health Research Gregorio Marañón, Madrid, Spain; 6. Hospital Universitario Salamanca, Salamanca, Spain; 7. Health Value, Madrid, Spain

INTRODUCTION

- Invasive mold diseases (IMDs) are associated with significant morbidity and mortality[1].
- Isavuconazole (ISAV) is a preferred treatment for certain patient profiles[2].
- European guidelines recommend ISAV and voriconazole as drugs of choice for pulmonary aspergillosis in neutropenic or allo-HSCT patients and in COVID-19-associated pulmonary aspergillosis (CAPA), and amphotericin B as an alternative treatment for pulmonary aspergillosis[3,4].
- In mucormycosis, liposomal amphotericin B (L-AMB) in high doses is recommended in the first line, with alternative treatments being ISAV and posaconazole (POSA)[5].

OBJECTIVES

- An economic model was developed to help inform treatment decisions for adult IMD patients for whom voriconazole is considered unsuitable, to explore the cost with ISAV versus L-AMB followed by POSA (L-AMB→POSA) in Spain.

METHODS

Design

- As indirect comparisons have demonstrated similar efficacy between the comparators[6,7], a cost-minimization approach was taken from the Spanish National Healthcare System (SNHS) perspective.
- The drug administration regimens for each treatment are presented in Figure 1.
- The use of ISAV initial formulation and dosage of L-AMB were based on expert opinion.
- Dosage for ISAV and POSA was based on labels. Duration of treatment (DoT) was based on clinical trials[8,9] and expert opinion.

Figure 1. Drug administration regimens

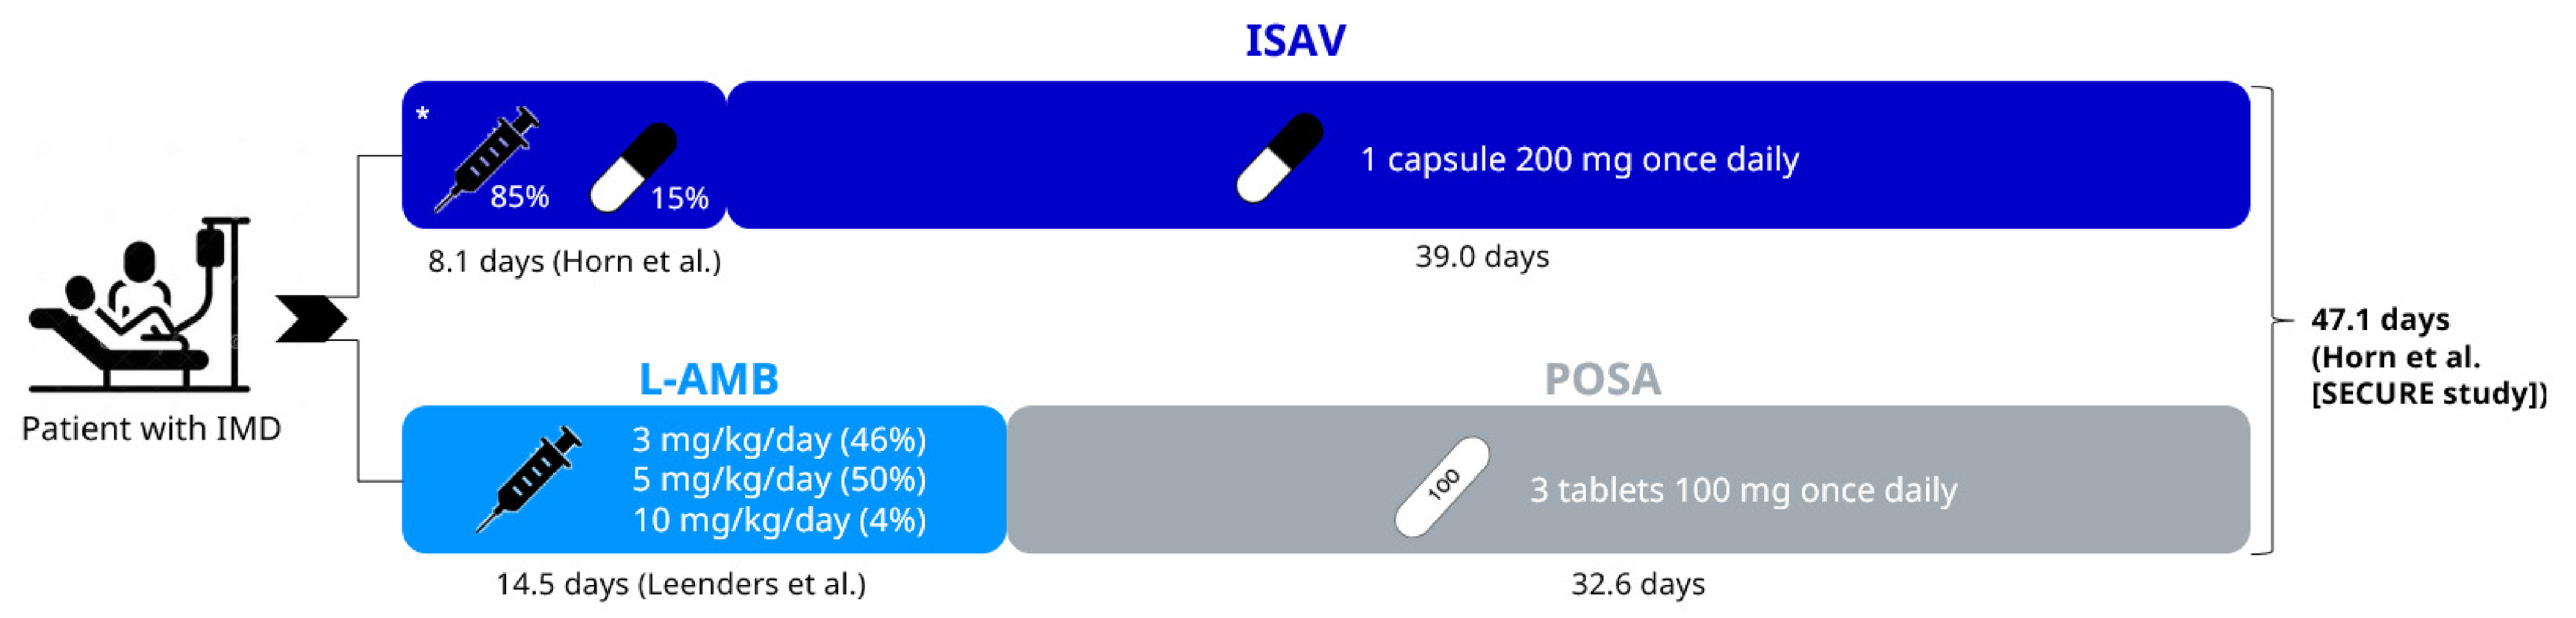
ISAV
*
85%
15%
1 capsule 200 mg once daily
8.1 days (Horn et al.)
39.0 days
Patient with IMD
47.1 days
(Horn et al.
[SECURE study])
L-AMB
POSA
3 mg/kg/day (46%)
5 mg/kg/day (50%)
10 mg/kg/day (4%)
100
3 tablets 100 mg once daily
14.5 days (Leenders et al.)
32.6 days

* Loading dose: 1 vial or 1 capsule 200 mg every 8 hours for the first 2 days (6 administrations in total). Maintenance dose: 1 vial 200 mg once daily.
Switch from intravenous to oral ISAV: On the basis of the high oral bioavailability (98%), switching between intravenous and oral administration is appropriate when clinically indicated.
IMD: Invasive mold disease; ISAV: isavuconazole, L-AMB: liposomal amphotericin B; POSA: posaconazole.

Use of resources and Costs

- Drug acquisition, intravenous (IV) administration, laboratory tests, hospitalization and adverse events (AEs) costs were included in the analysis (Table 1).
- L-AMB dosing was calculated with a mean weight of 68.6 kg in the SECURE study[9], not including unused vial contents[10].
- IV administration and laboratory testing were based on expert opinion[10].
- Time on IV infusion preparation of L-AMB and ISAV was 30 and 10 min, respectively[10].

Table 1. Use of resources and unit costs

Drug	Costs (€2023)	ISAV	L-AMB	POSA
Pack (€)	-	200 mg 1 vial / 100 mg 14 tablets	50 mg 10 vials	100 mg 24 tablets
List Price per pack	-	400.00€ / 700.00 €	1,300.61 €	266.02 €
Intravenous (IV) administration (min)				
IV administration	1.07	121	435	NA
Hospitalization (days)				
Emergency room	1,760.00	3.1	3.1	0.0
General ward	653.91	16.6	16.6	0.0
Laboratory tests (per week)				
Urinalysis	3.00	0.0	7.0	
Serum creatinine test	3.00	0.0	7.0	
Magnesium test	17.00	0.0	6.0	
Liver function test	3.00	7.0	7.0	
Adverse events (%)				
Vision impared or blurred	59.91	0.40%	0.00%	1.00%
Photopsia	59.91	0.00%	0.00%	0.70%
Hyperbilirubinemia	153.90	0.40%	0.00%	3.00%
Abnormal hepatic function	153.90	0.80%	0.00%	1.40%
Increased GGT	181.82	2.30%	0.00%	3.80%
Increased ALP	181.82	1.90%	0.00%	2.40%
Increased AST	181.82	1.90%	15.70%	6.30%
Increased ALT	181.82	1.60%	0.00%	7.60%
QT prolonged	181.82	0.40%	0.00%	4.00%
Hallucination	180.13	0.40%	0.00%	1.40%
Dyspnoea	146.90	3.10%	0.00%	0.00%
Nephrotoxicity	853.66	0.00%	10.60%	0.00%
Hypokalaemia	237.33	17.50%	60.80%	28.50%
Hypomagnesemia	780.00	5.40%	28.10%	0.00%
Infusion reaction	76.17	4.30%	9.50%	0.00%
Dialysis following treatment	196.46	0.00%	3.80%	0.00%

- L-AMB→POSA patients required urinalysis, serum creatinine tests and liver-function tests once weekly (7 in total), plus 6 magnesium tests; while ISAV patients required 7 liver-function tests[10].
- Total hospital stay was 19.7 days[8] and included 15.6% days of treatment in the emergency rooms[11].
- AEs for ISAV and POSA were obtained from their EMA Assessment Reports and L-AMB in Wade et al[12].
- Drug costs were consulted in Botplus 2.0 and costs for resource use were obtained from e-Salud database and adjusted for inflation (€2023).

RESULTS

Base case analysis

- ISAV treatment cost compared to L-AMB→POSA showed a saving of € 4,029.25 (€ 8,195.50 versus € 12,224.75), representing a saving of 33.0%.

Table 2. Deterministic base case results

	ISAV		L-AMB → POSA		ISAV vs L-AMB → POSA Cost difference (€ and %)	
	Use resources	Cost (€)	Resource use	Cost (€)		
IV formulation	8.1 days	4,295.50	14.5 days	11,074.22	-6,778.72	-61.2%
Oral formulation	39 days	3,900.00	32.6 days	1,150.54	2,749.46	239.0%
Total Treatment	47.1 days	**8,195.50**	47.1 days	**12,224.75**	**-4,029.25**	**-33.0%**
IV administration	121 min	109.67	435 min	463.84	-354.17	-76.4%
Hospital stay	19.7 days	16,281.26	19.7 days	16,281.26	0.00	0.0%
Laboratory tests	7 tests	21.00	27 tests	165.00	-144.00	-87.3%
Adverse Events	39.8%	108.11	188.8%	618.67	-510.56	-82.5%
Total Use of resources		**16,520.04**		**17,528.78**	**-1,008.74**	**-5.8%**
TOTAL		**24,715.54**		**29,753.53**	**-5,037.99**	**-16.9%**

Sensitivity analysis

- All the variables analyzed showed savings in the cost of treatment with ISAV compared to L-AMB/POSA (Figure2).
- Savings ranged from € 326.59 to € 7,968.89 with ISAV.
- The most influential parameter in the outcome was IV administration time and the least influential was drug price.

Figure 2. One way sensitivity analysis on cost difference ISAV vs L-AMB → POSA (€)

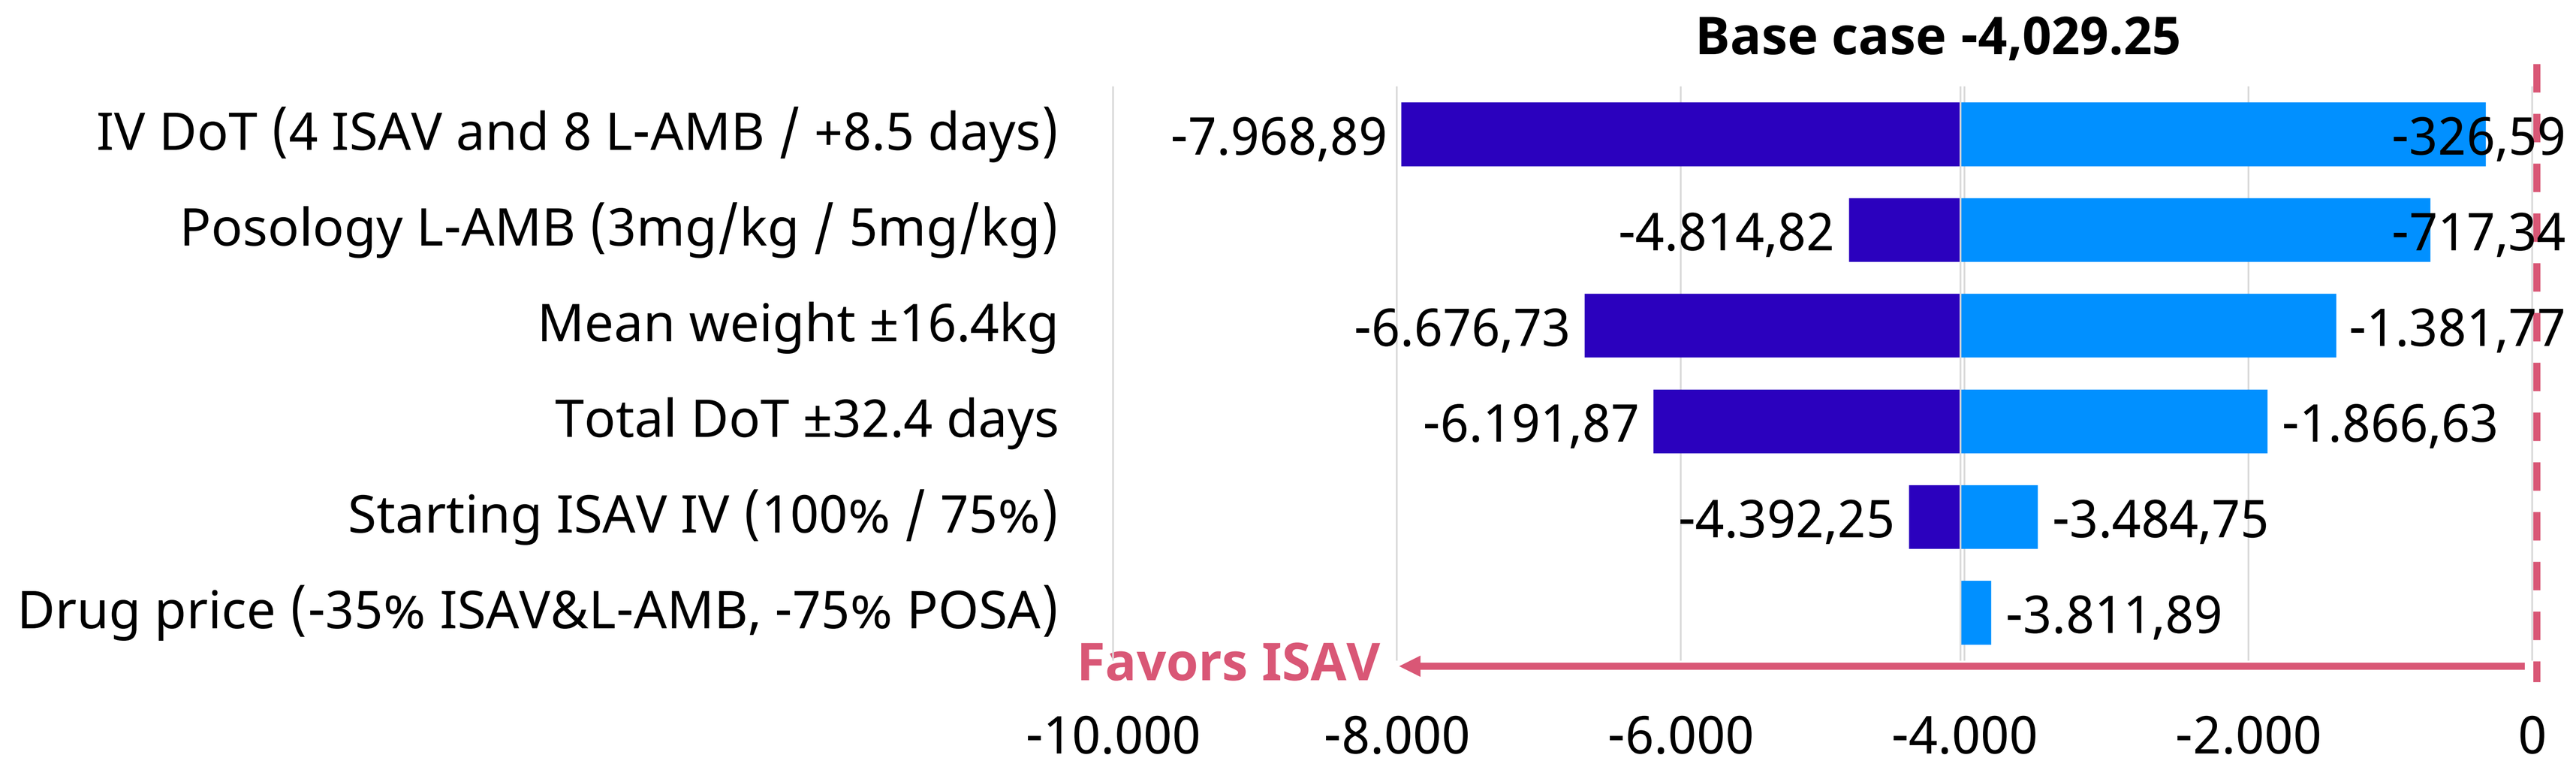
Base case -4,029.25
IV DoT (4 ISAV and 8 L-AMB / +8.5 days)
Posology L-AMB (3mg/kg / 5mg/kg)
Mean weight ±16.4kg
Total DoT ±32.4 days
Starting ISAV IV (100% / 75%)
Drug price (-35% ISAV&L-AMB, -75% POSA)
-7.968,89
-326,59
-4.814,82
-717,34
-6.676,73
-1.381,77
-6.191,87
-1.866,63
-4.392,25
-3.484,75
-3.811,89
Favors ISAV
-10.000
-8.000
-6.000
-4.000
-2.000
0

- After 1,000 second-order Monte Carlo simulations, a mean total savings per patient treated with ISAV versus L-AMB→POSA of € 5,029 were obtained, with a 95% confidence interval (CI) of € -4,088 and € -6,107 (Table 3).
- The probability of savings with ISAV was 100%.

Table 3. Probabilistic analysis results

	ISA	L-AMB→POSA	Diference	Probability of savings
Mean	24,671 €	29,700 €	-5,029 €	100%
SD	2,602 €	3,132 €	-	
Inferior Lim CI95%	20,057 €	24,145 €	-4,088 €	
Superior Lim CI95%	29,961 €	36,068 €	-6,107 €	

CONCLUSIONS

ISAV has the potential to substantial save costs on the IMD management relative to L-AMB→POSA.

ISAV maintains the same efficacy and tolerability during the whole treatment duration without switching to another therapeutic class.

This analysis would enable healthcare professionals to make informed therapeutic decisions based on most efficient therapy.

REFERENCES

1. Rodríguez-Tudela JL et al. Clin Microbiol Infect. 2015 Feb;21(2):183-9. 2. Natesan et al. Infection and Drug Resistance 2016:9 291–300. 3. Ullmann AJ et al. Clin Microbiol Infect. 2018; 24 (Suppl 1): e1-e38. 4. Koehler P et al. Lancet Infect Dis. 2021; 21: e149-62. 5. Cornely OA et al. Lancet Infect Dis. 2019; 19: e405-e421. 6. Herbrecht R, et al. Curr Med Res Opin. 2018;34(12):2187-2195. 7. Marty FM et al. Lancet Infect Dis. 2016; 16:828-837. 8. Horn D et al. J. Med. Econ. 19(7), 728–734 (2016). 9. Leenders A et al. Br. J. Haematol. 103(1), 205–212 (1998). 10. Bagshaw E et al. FutureMicrobiol. (2018) 13(11), 1283–1293. 11. Batista MV et al. J Fungi (Basel). 2023 Jan 27;9(2):166. 12. Wade RL et al. Diagnostic Microbiology and Infectious Disease 76 (2013) 361–367.

DISCLOSURE

- This study was sponsored by Pfizer S.L.U. The authors JRA, JB, RF, MK and AM were the study coordinators and received research support and honoraria from Pfizer S.L.U. CRT is employee of Health Value which received funding from Pfizer S.L.U. CMA, MGS and CP are a employees of Pfizer S.L.U.

Pfizer

ISPOR Europe 2023 | 11-15 November
Bella Center Copenhagen, Copenhagen, Denmark