EE636

Cost-Minimization Analysis of Acute Bacterial Skin and Skin Structure Infection Treatment with Delafloxacin versus Standard of Care in Jordan

Naser AY.[1], Bilbisi M.[2], Mousa R.[3], Aburmilah A.[4]

Affiliations: 1-Department of Applied Pharmaceutical Sciences and Clinical Pharmacy, Isra'a University, Amman, Jordan. 2-Abdali Medical Center, Amman, Jordan. 3-The University of Jordan, Faculty of Pharmacy, Amman, Jordan. 4-Hikma Pharmaceuticals, Amman, Jordan.

Introduction

Skin and skin structure infections are among the most widespread infectious diseases. Over the past two decades, methicillin-resistant Staphylococcus aureus has become a common cause of purulent skin and soft tissue infections.

Previous studies demonstrated the non-inferiority of delafloxacin compared to “Piperacillin/Tazobactam + Vancomycin” or “Meropenem + Vancomycin” for the treatment of Acute Bacterial Skin and Skin Structure Infections (ABSSSI).

Indirect Comparison	Comparators
Pullman J et al. 1	Baxdela versus Aztreonam/ Vancomycin
O'Riordan W et al. 2	Baxdela versus Aztreonam/ Vancomycin
Fabian TC et al 3	Meropenem versus Imipenem Cilastatin
Tellado J. et al 4	Piperacillin/Tazobactam versus Ertapenem

Objectives

The aim of this study was to determine the relative cost difference between delafloxacin and standard of care for the treatment of ABSSI using a cost-minimization analysis in Jordan.

Methods

We conducted a cost minimization analysis to compare the costs of treating ABSSI with delafloxacin, option A (Piperacillin/ Tazobactam + Vancomycin), and option B (Meropenem + Vancomycin). The cost of medications, the cost of medication administration, and hospitalization costs were calculated as direct costs associated with ABSSSI management. The Jordanian Food and Drug Administration was consulted for information on medication prices. The cost of hospitalization was estimated from four main private hospitals in Jordan.

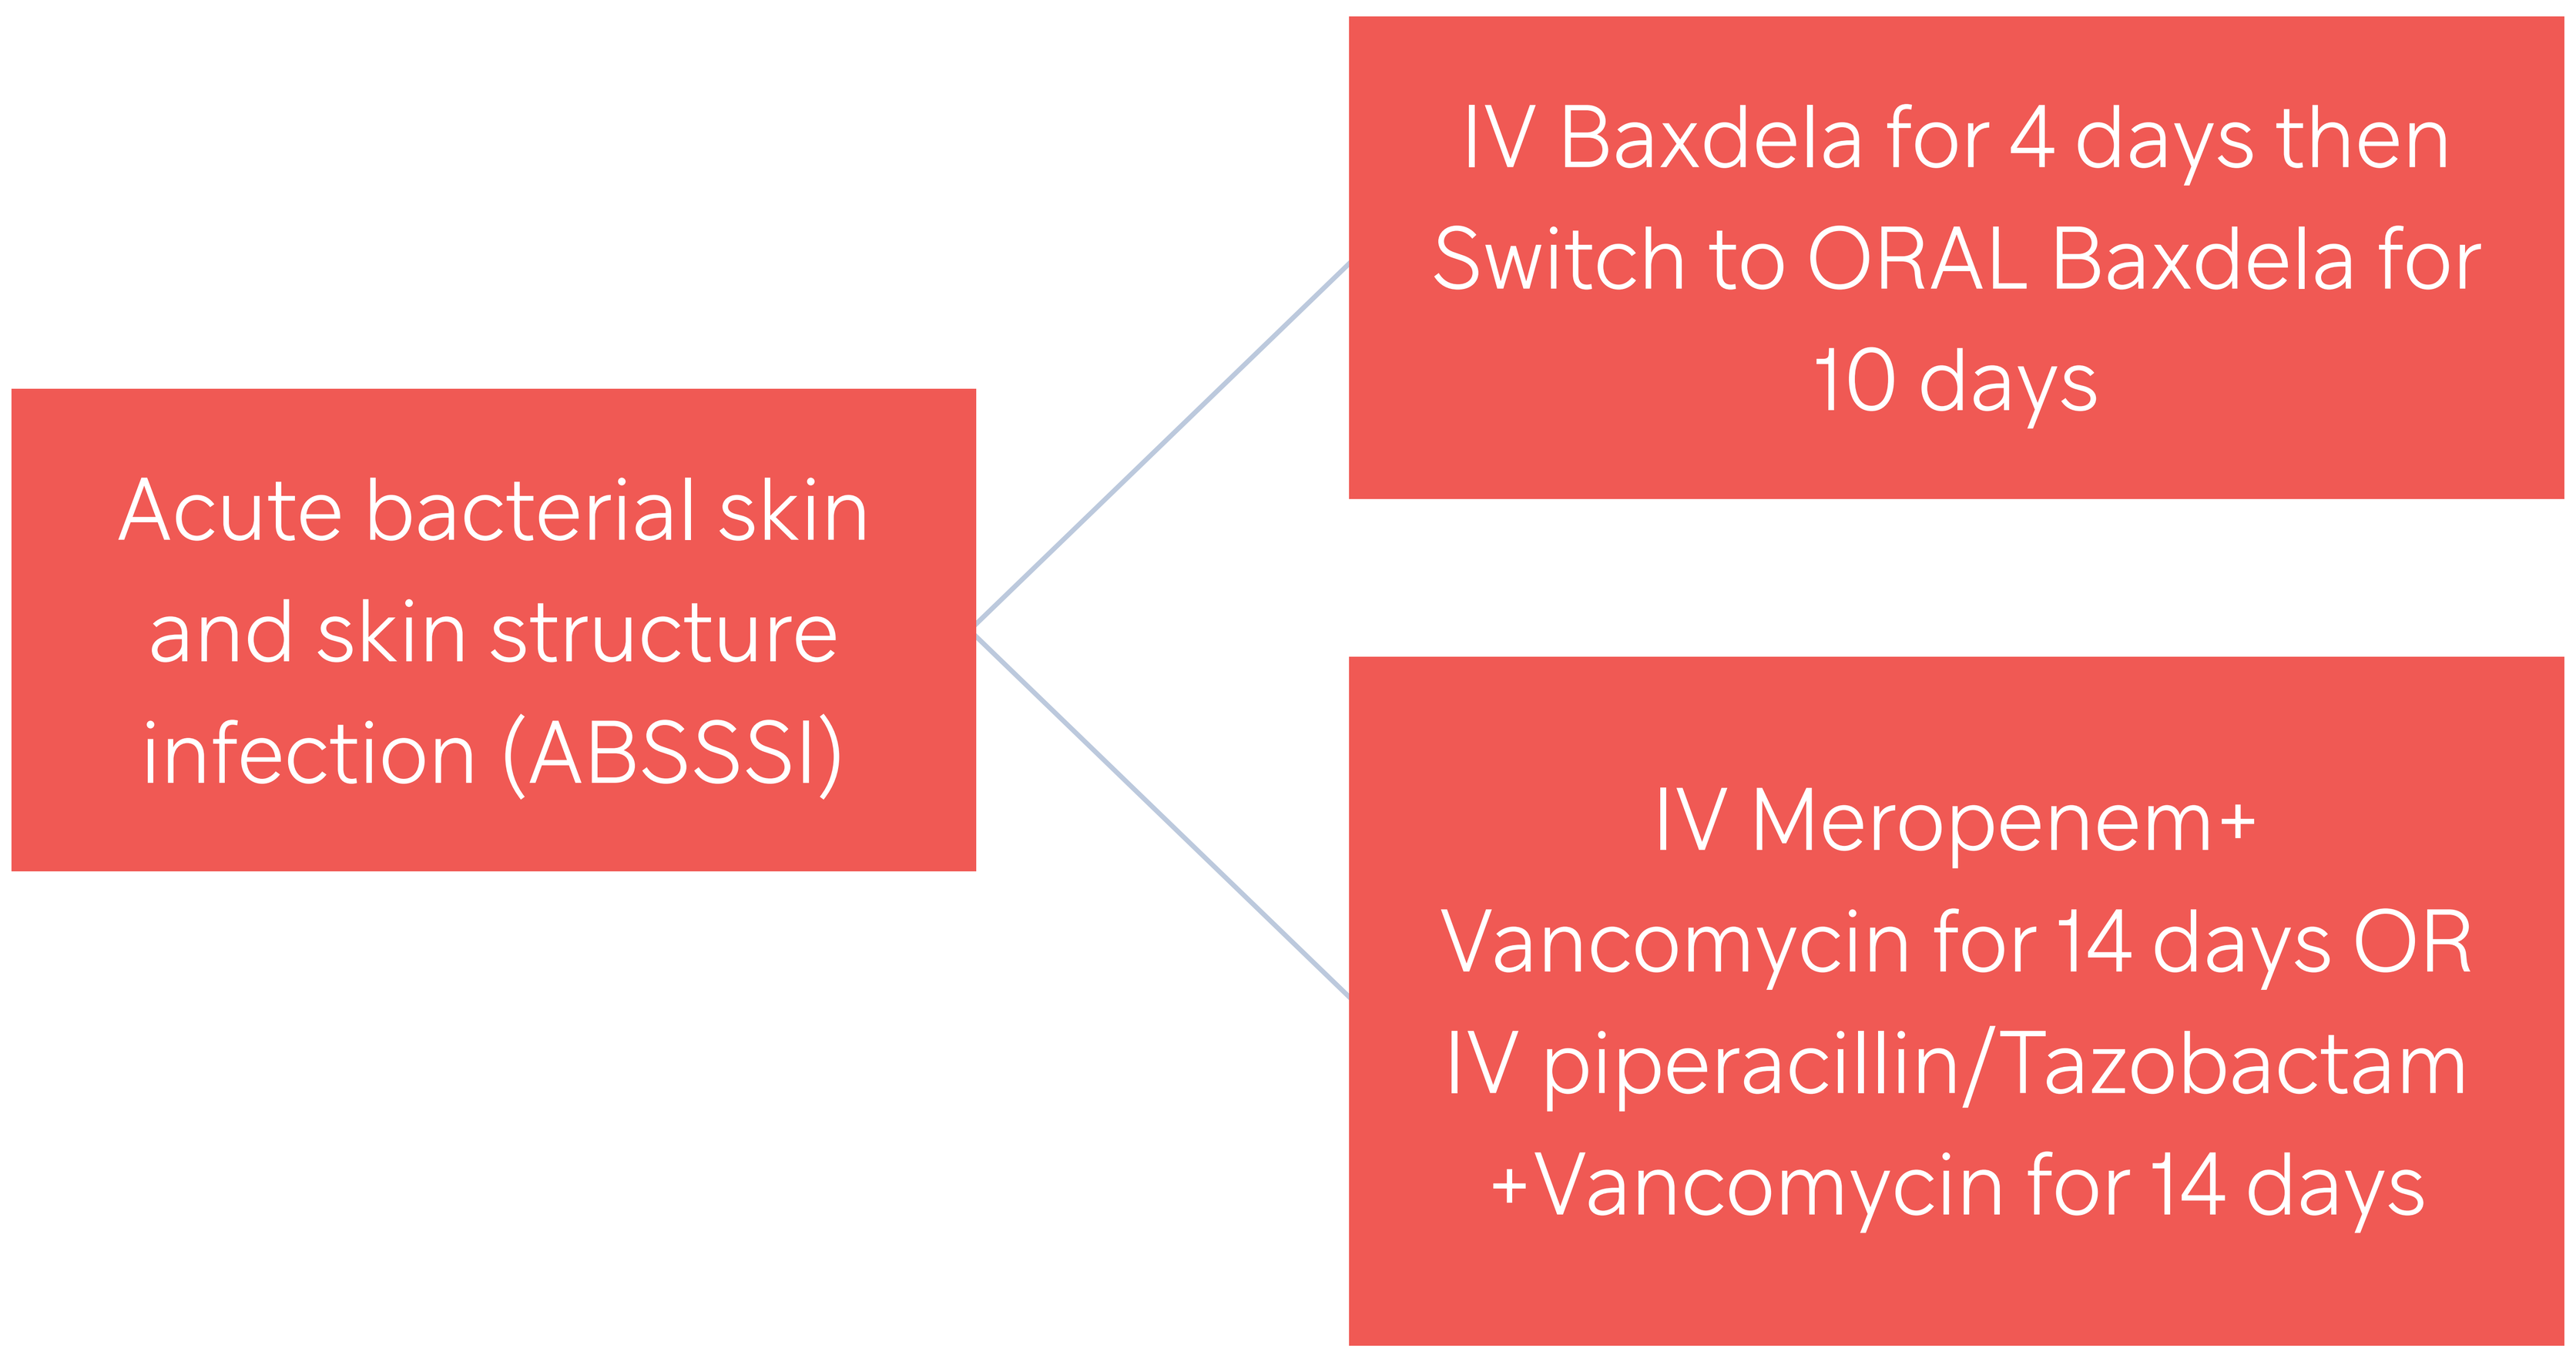
Acute bacterial skin and skin structure infection (ABSSSI)
IV Baxdela for 4 days then Switch to ORAL Baxdela for 10 days
IV Meropenem+ Vancomycin for 14 days OR IV piperacillin/Tazobactam +Vancomycin for 14 days

Results

The total cost of the delafloxacin treatment option was 1,631.8 Jordanian Dinar (JOD) (one JOD equals 1.41 USD), compared with 3,675.4 JOD and 3,703.6 JOD for treatment options A and B, respectively. The main cost driver that contributed to the most significant difference in treatment cost among different treatment options was the infusion administration cost related to longer length of hospital stay associated with other treatment options (Piperacillin/Tazobactam + Vancomycin or Meropenem + Vancomycin). The overall cost savings associated with the use of delafloxacin are estimated to be around 2,043.7 JOD and 2,071.8 JOD for options A and B, respectively.

Treatment Options	Product Name	Price in Jordan per unit	Length of treatment in days	Doses per length of stay	Total drug cost	Hospitalization cost	Infusion administration	Total cost for options (drug and hospitalization)	Cost saving
Option 1	Delafloxacin IV 300mg/ vial	JOD 55.64	4	8	JOD 445.12	JOD 280.00	JOD 240.00		
	Delafloxacin Oral 450mg 20 tabs	JOD 666.64	10	20	JOD 666.64	JOD -	JOD -	JOD 1,631.76	-
	Total for option 1				JOD 1,111.76				
Option 2	Vancomycin IV 1g/ vial	JOD 8.14	14	28	JOD 227.92	JOD 980.00	JOD 840.00		
	Piperacillin/Tazobacatm IV 4.5g/vial	JOD 8.75	14	42	JOD 367.50	JOD -	JOD 1,260.00	JOD 3,675.42	JOD 2,043.66
	Total for option 2				JOD 595.42				
Option 3	Vancomycin IV 1g/ vial	JOD 8.14	14	28	JOD 227.92	JOD 980.00	JOD 840.00		
	Meropenem IV 1g/ vial	JOD 9.42	14	42	JOD 395.64	JOD -	JOD 1,260.00	JOD 3,703.56	JOD 2,071.80
	Total for option 3				JOD 623.56				

Conclusions

Delafloxacin is a cost-saving treatment option for patients with ABSSSI in Jordan. Our study findings can help clinicians and decision makers in the healthcare sector prescribe medications for patients with ABSSSI at lower costs while maintaining the efficacy.

References:

1. Pullman J, Gardovskis J, Farley B, Sun E, Quintas M, Lawrence L, Ling R, Cammarata S; PROCEED Study Group. Efficacy and safety of delafloxacin compared with vancomycin plus aztreonam for acute bacterial skin and skin structure infections: a Phase 3, double-blind, randomized study. J Antimicrob Chemother. 2017 Dec 1;72(12):3471-3480. doi: 10.1093/jac/dkx329. PMID: 29029278; PMCID: PMC5890686.
2. O'Riordan W, McManus A, Teras J, Poromanski I, Cruz-Saldariagga M, Quintas M, Lawrence L, Liang S, Cammarata S; PROCEED Study Group. A Comparison of the Efficacy and Safety of Intravenous Followed by Oral Delafloxacin With Vancomycin Plus Aztreonam for the Treatment of Acute Bacterial Skin and Skin Structure Infections: A Phase 3, Multinational, Double-Blind, Randomized Study. Clin Infect Dis. 2018 Aug 16;67(5):657-666. doi: 10.1093/cid/ciy165. PMID: 29518178; PMCID: PMC6093995.
3. Fabian TC, File TM, Embil JM, Krige JE, Klein S, Rose A, Melnick D, Soto NE. Meropenem versus imipenem-cilastatin for the treatment of hospitalized patients with complicated skin and skin structure infections: results of a multicenter, randomized, double-blind comparative study. Surg Infect (Larchmt). 2005 Fall;6(3):269-82. doi: 10.1089/sur.2005.6.269. PMID: 16201937.
4. Kohei Takimoto, Qianzhi Wang, Daisuke Suzuki, Mitsuya Katayama & Yoshiro Hayashi (2017) Clinical efficacy of piperacillin/tazobactam in the treatment of complicated skin and soft tissue infections, Expert Opinion on Pharmacotherapy, 18:10, 1027-1034, DOI: 10.1080/14656566.2017.1341491