EE351

Using Social Return on Investment to capture the broader impact of medicines: advanced Parkinson's disease

A. Stefanie Mikolaizak,[1] Roxanne Maurin,[2] Inez Denham,[2] Irene Deltetto,[2] Jenny Waern,[1] Colman Taylor, [2,3]

[1]AbbVie Pty Ltd, NSW, Australia; [2]**HT**ANALYSTS, Sydney NSW, Australia; [3]The George Institute for Global Health, NSW, Australia

OBJECTIVE

This novel Social Return on Investment (SROI) analysis aimed to evaluate the broader impact created by improving access to levodopa-based device-aided therapies (DATs) for people living with advanced PD (aPD) in Australia

CONCLUSIONS

aPD impacts patients and also their partners, families and broader society, yet less than 25% of aPD patients in Australia are receiving treatment with DATs.

For every $1 invested into access to DATs over $1.79 of social and economic value is created. Over 50% of the value is created for the partners and children of people living with aPD.

Investment into increased access to levodopa-based DATs may create $179.61 million of social value over three years.

AbbVie and the authors thank the participants for their time during the interviews and surveys. The authors would also like to acknowledge the patient advocacy groups, neurologists and nurses, who were essential in providing outreach to patients.

AbbVie Pty Ltd sponsored this research.

AbbVie Pty Ltd assisted in identifying neurologists who were treating patients with aPD and who had experience with levodopa-based DATs. AbbVie Pty Ltd approved all study documents including participant recruitment flyers and interview guides. AbbVie Pty Ltd were not involved in the collection of data to ensure patient privacy was maintained. Patients and health care professionals did not receive payment for their participation in this study

No honoraria or payments were made for authorship. **HT**ANALYSTS collected, analysed, and interpreted the data and developed the manuscript. AbbVie Pty Ltd sponsored the study, contributed to the design, reviewed, and approved the final manuscript.

HTANALYSTS received funding from AbbVie Pty Ltd to conduct this research. ID, ID, RM and CT are all paid employees of **HT**ANALYSTS. ASM and JW are a paid employees of AbbVie Pty Ltd and own shares in AbbVie.

For additional information or to obtain a PDF of this poster

Scan QR code or utilize the following link to download an electronic version of this presentation.

Available here: https://abbv.ie/au-sroi

To submit a medical question, please visit www.abbviemedinfo.com

INTRODUCTION

Social Return on Investment (SROI) methodology is able to capture the broader impact of medicines, including social, environmental and economic outcomes. Patients with Parkinson's disease (PD) become increasingly dependent on family and carers as their disease progresses.

Impact of Parkinson's Disease

- aPD affects not only the person living with the disease but also impacts families, carers and broader society
- CEA only capture patient and payer-related outcomes
- Broader impact not considered in traditional cost-effectiveness analyses (CEA)

METHODS

SROI PRINCIPLES:

1. INVOLVE STAKEHOLDERS
2. UNDERSTAND WHAT CHANGES
3. VAUE WHAT MATTERS
4. ONLY INCLUDE WHAT IS MATERIAL
5. DO NOT OVERCLAIM
6. VERIFY THE RESULTS
7. BE TRANSPARENT
8. BE RESPONSIVE

Forecast SROI analysis over a three-year time horizon

- Qualitative interviews and surveys conducted with people living with aPD and their families
- Supplementary clinical trial data
- Financial proxies were assigned to each final outcome based on willingness-to-pay, economic valuation, and replacement value.
- Cost inputs were sourced from Pharmaceutical Benefits Schedule and Medicare Benefits Scheme prices.

VALUATION AND SROI FILTERS:

1. FINANCIAL VALUATION
2. PROPORTION
3. IMPORTANCE
4. DISPLACEMENT
5. DEADWEIGHT
6. DROPOFF
7. ATTRIBUTION

RESULTS

Significant return on investment with better access

SROI 1.79

For every $1 invested in access to levodopa-based (LD) DATs $1.79 of social value is created

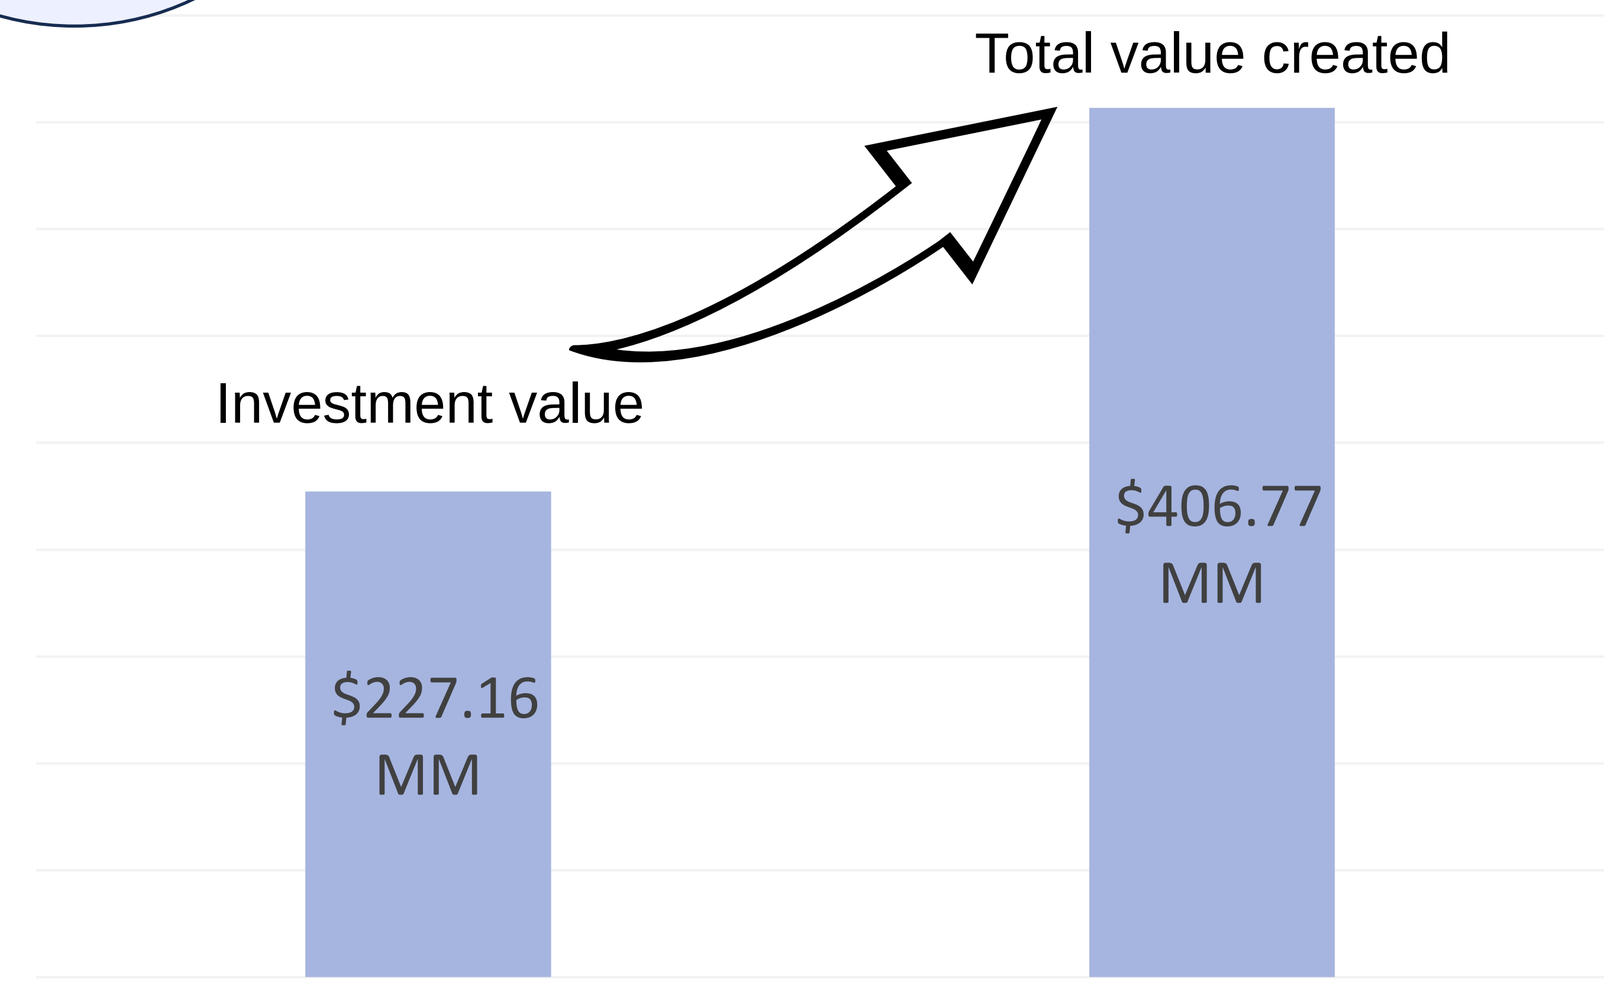

Total value created
Investment value
$227.16 MM
$406.77 MM

1,228 levodopa-responsive people with aPD impacted by access

The **value created** is **shared** between stakeholders

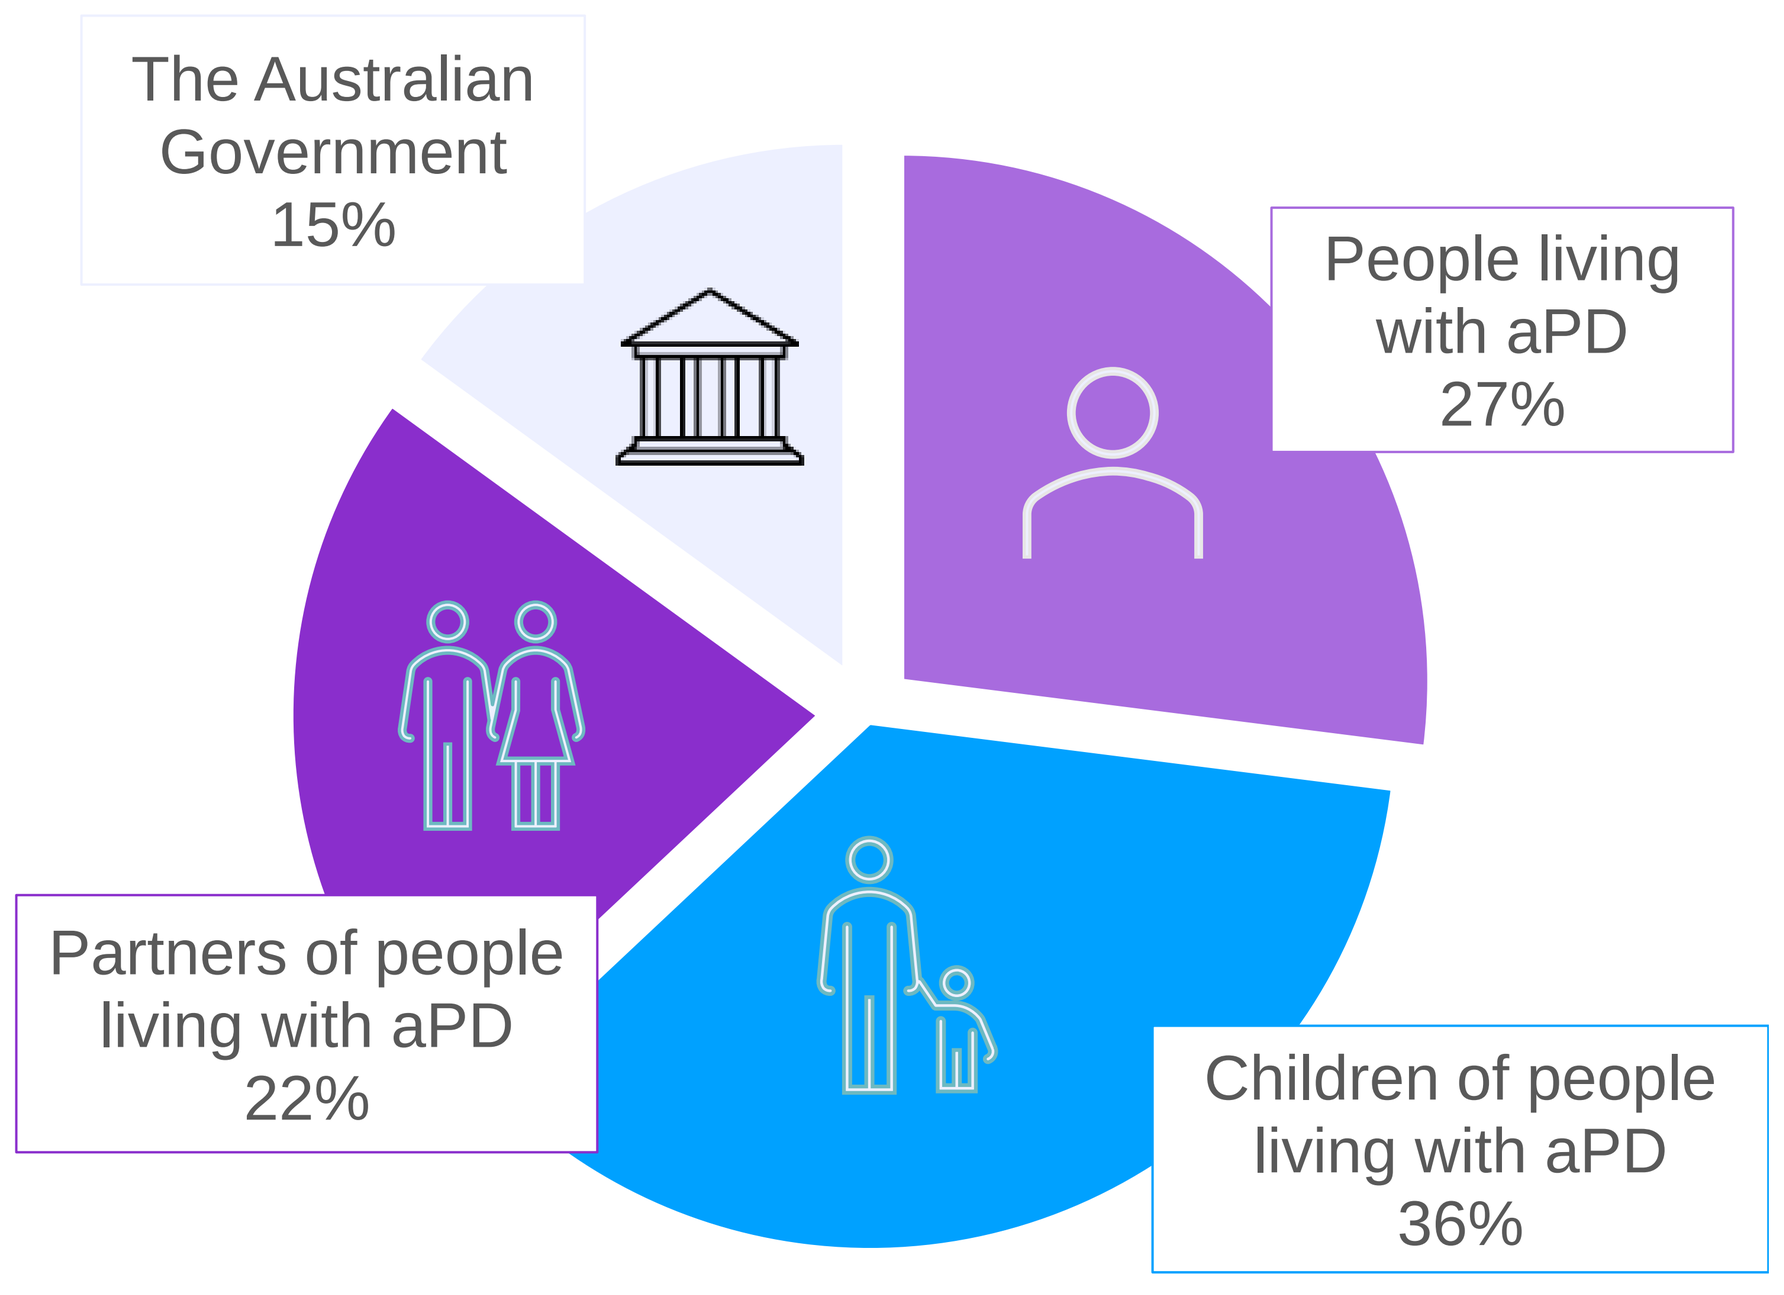

The Australian Government 15%
People living with aPD 27%
Partners of people living with aPD 22%
Children of people living with aPD 36%

Outcomes for the Australian Government

Economic outcomes including avoided costs

$60.32 MILLION total value for the Australian Government

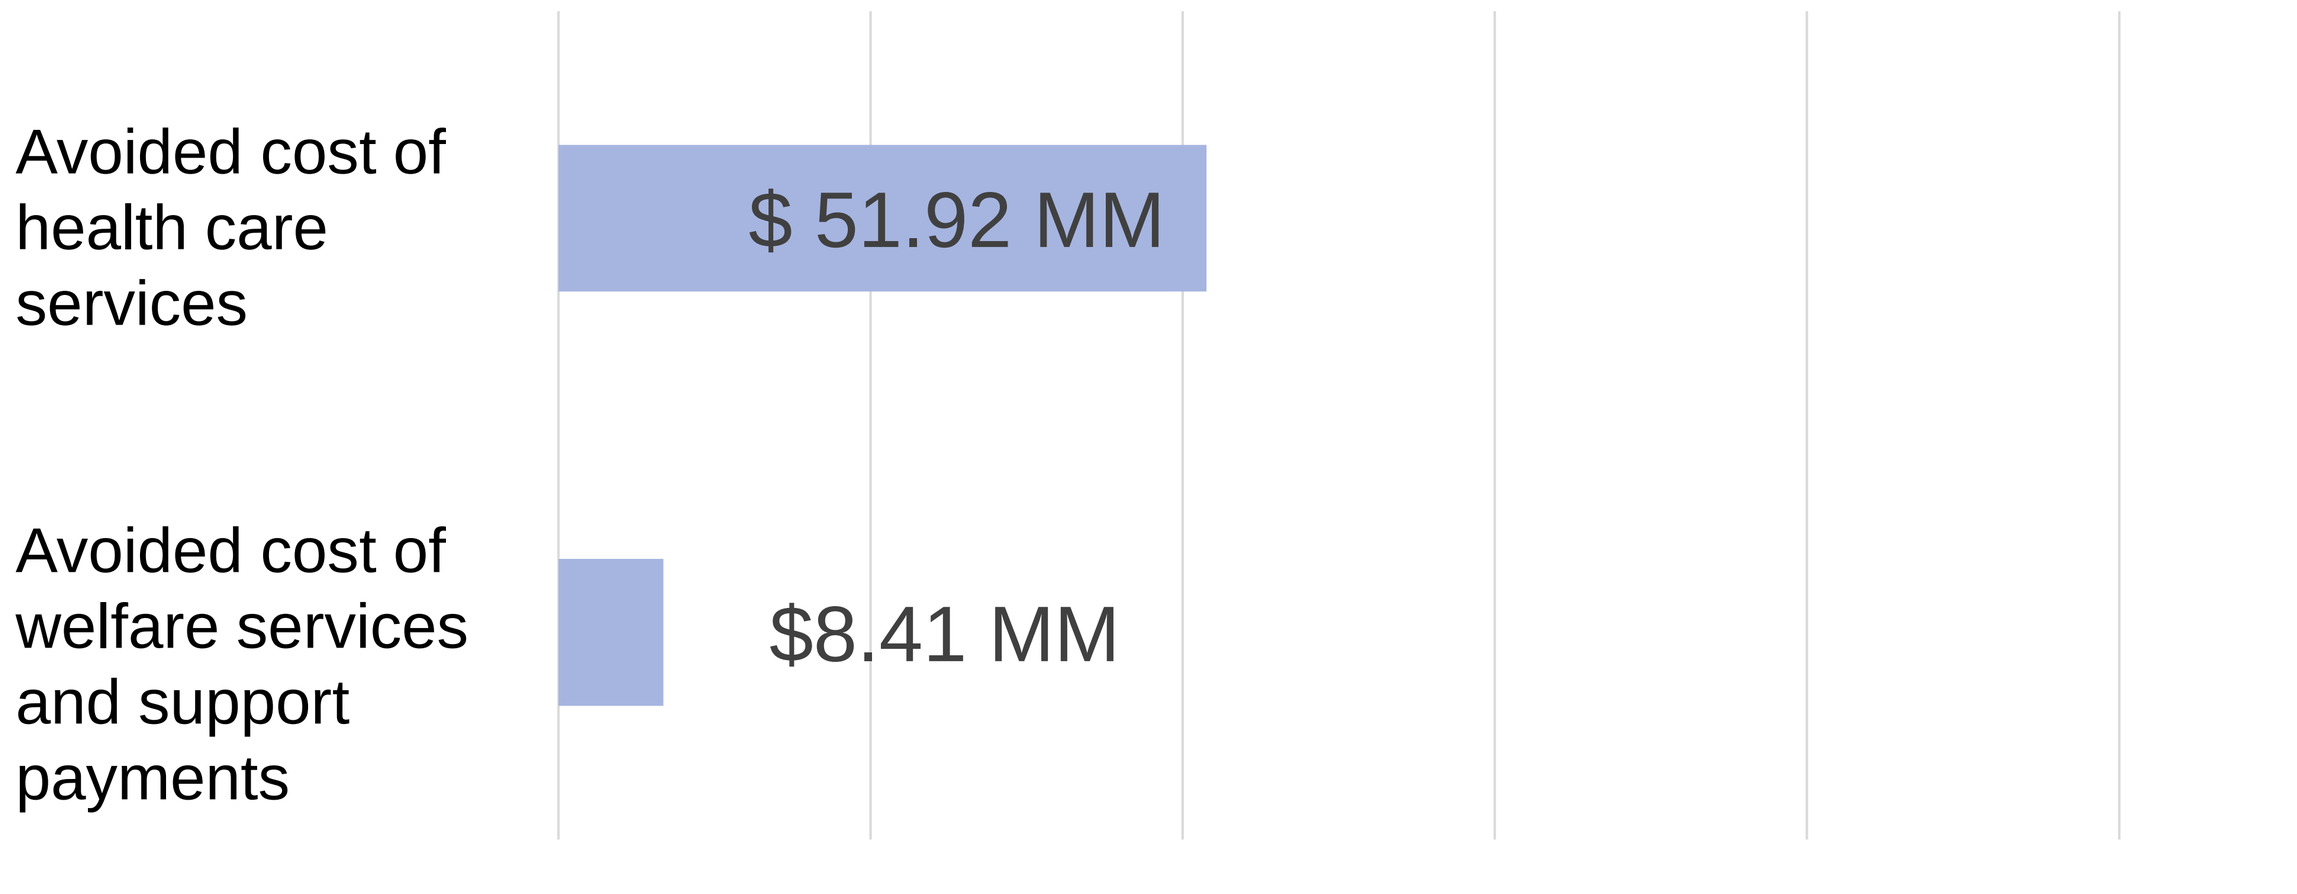

Avoided cost of health care services
$ 51.92 MM
Avoided cost of welfare services and support payments
$8.41 MM

Outcomes for children of people living with aPD

Social and emotional outcomes as a result of increased access to DATs.

$146.91 MILLION total value for children of people living with aPD

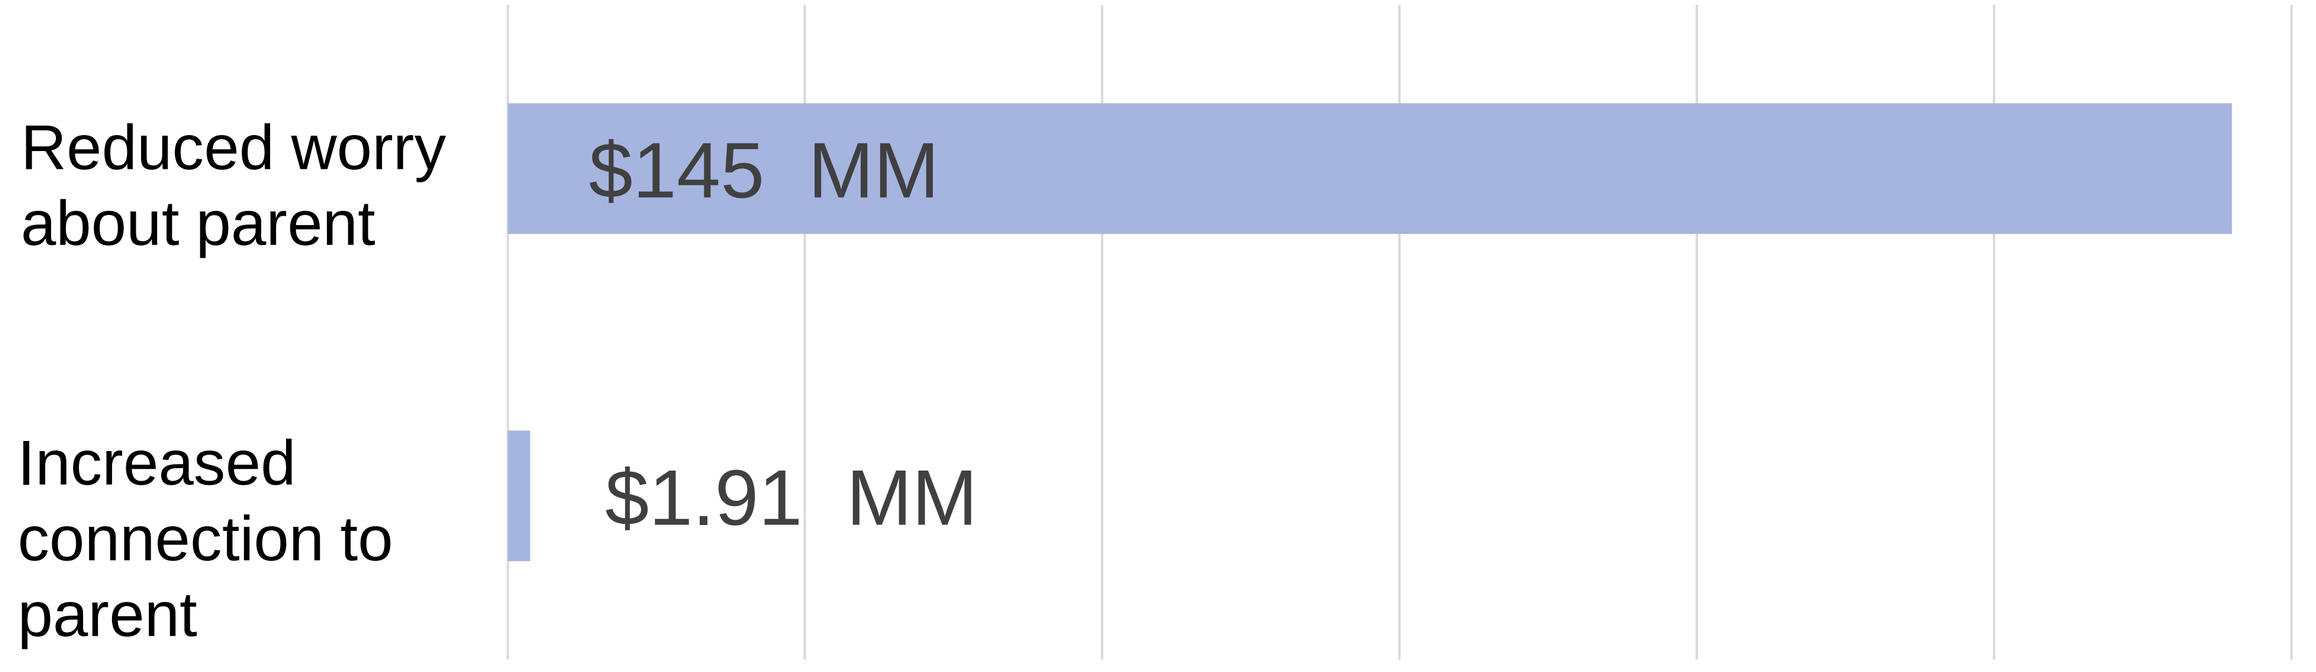

Reduced worry about parent
$145 MM
Increased connection to parent
$1.91 MM

"His sons were really blown away by having these great long conversations with their dad. – Nurse caring for people with aPD receiving treatment with a LD-based DAT

Outcomes for people living with aPD

Drug and initiation costs are shared by the people living with aPD and the Australian Government.

High initial investment however results in wide-ranging social and economic benefits.

$109.11 MILLION total value for people living with aPD

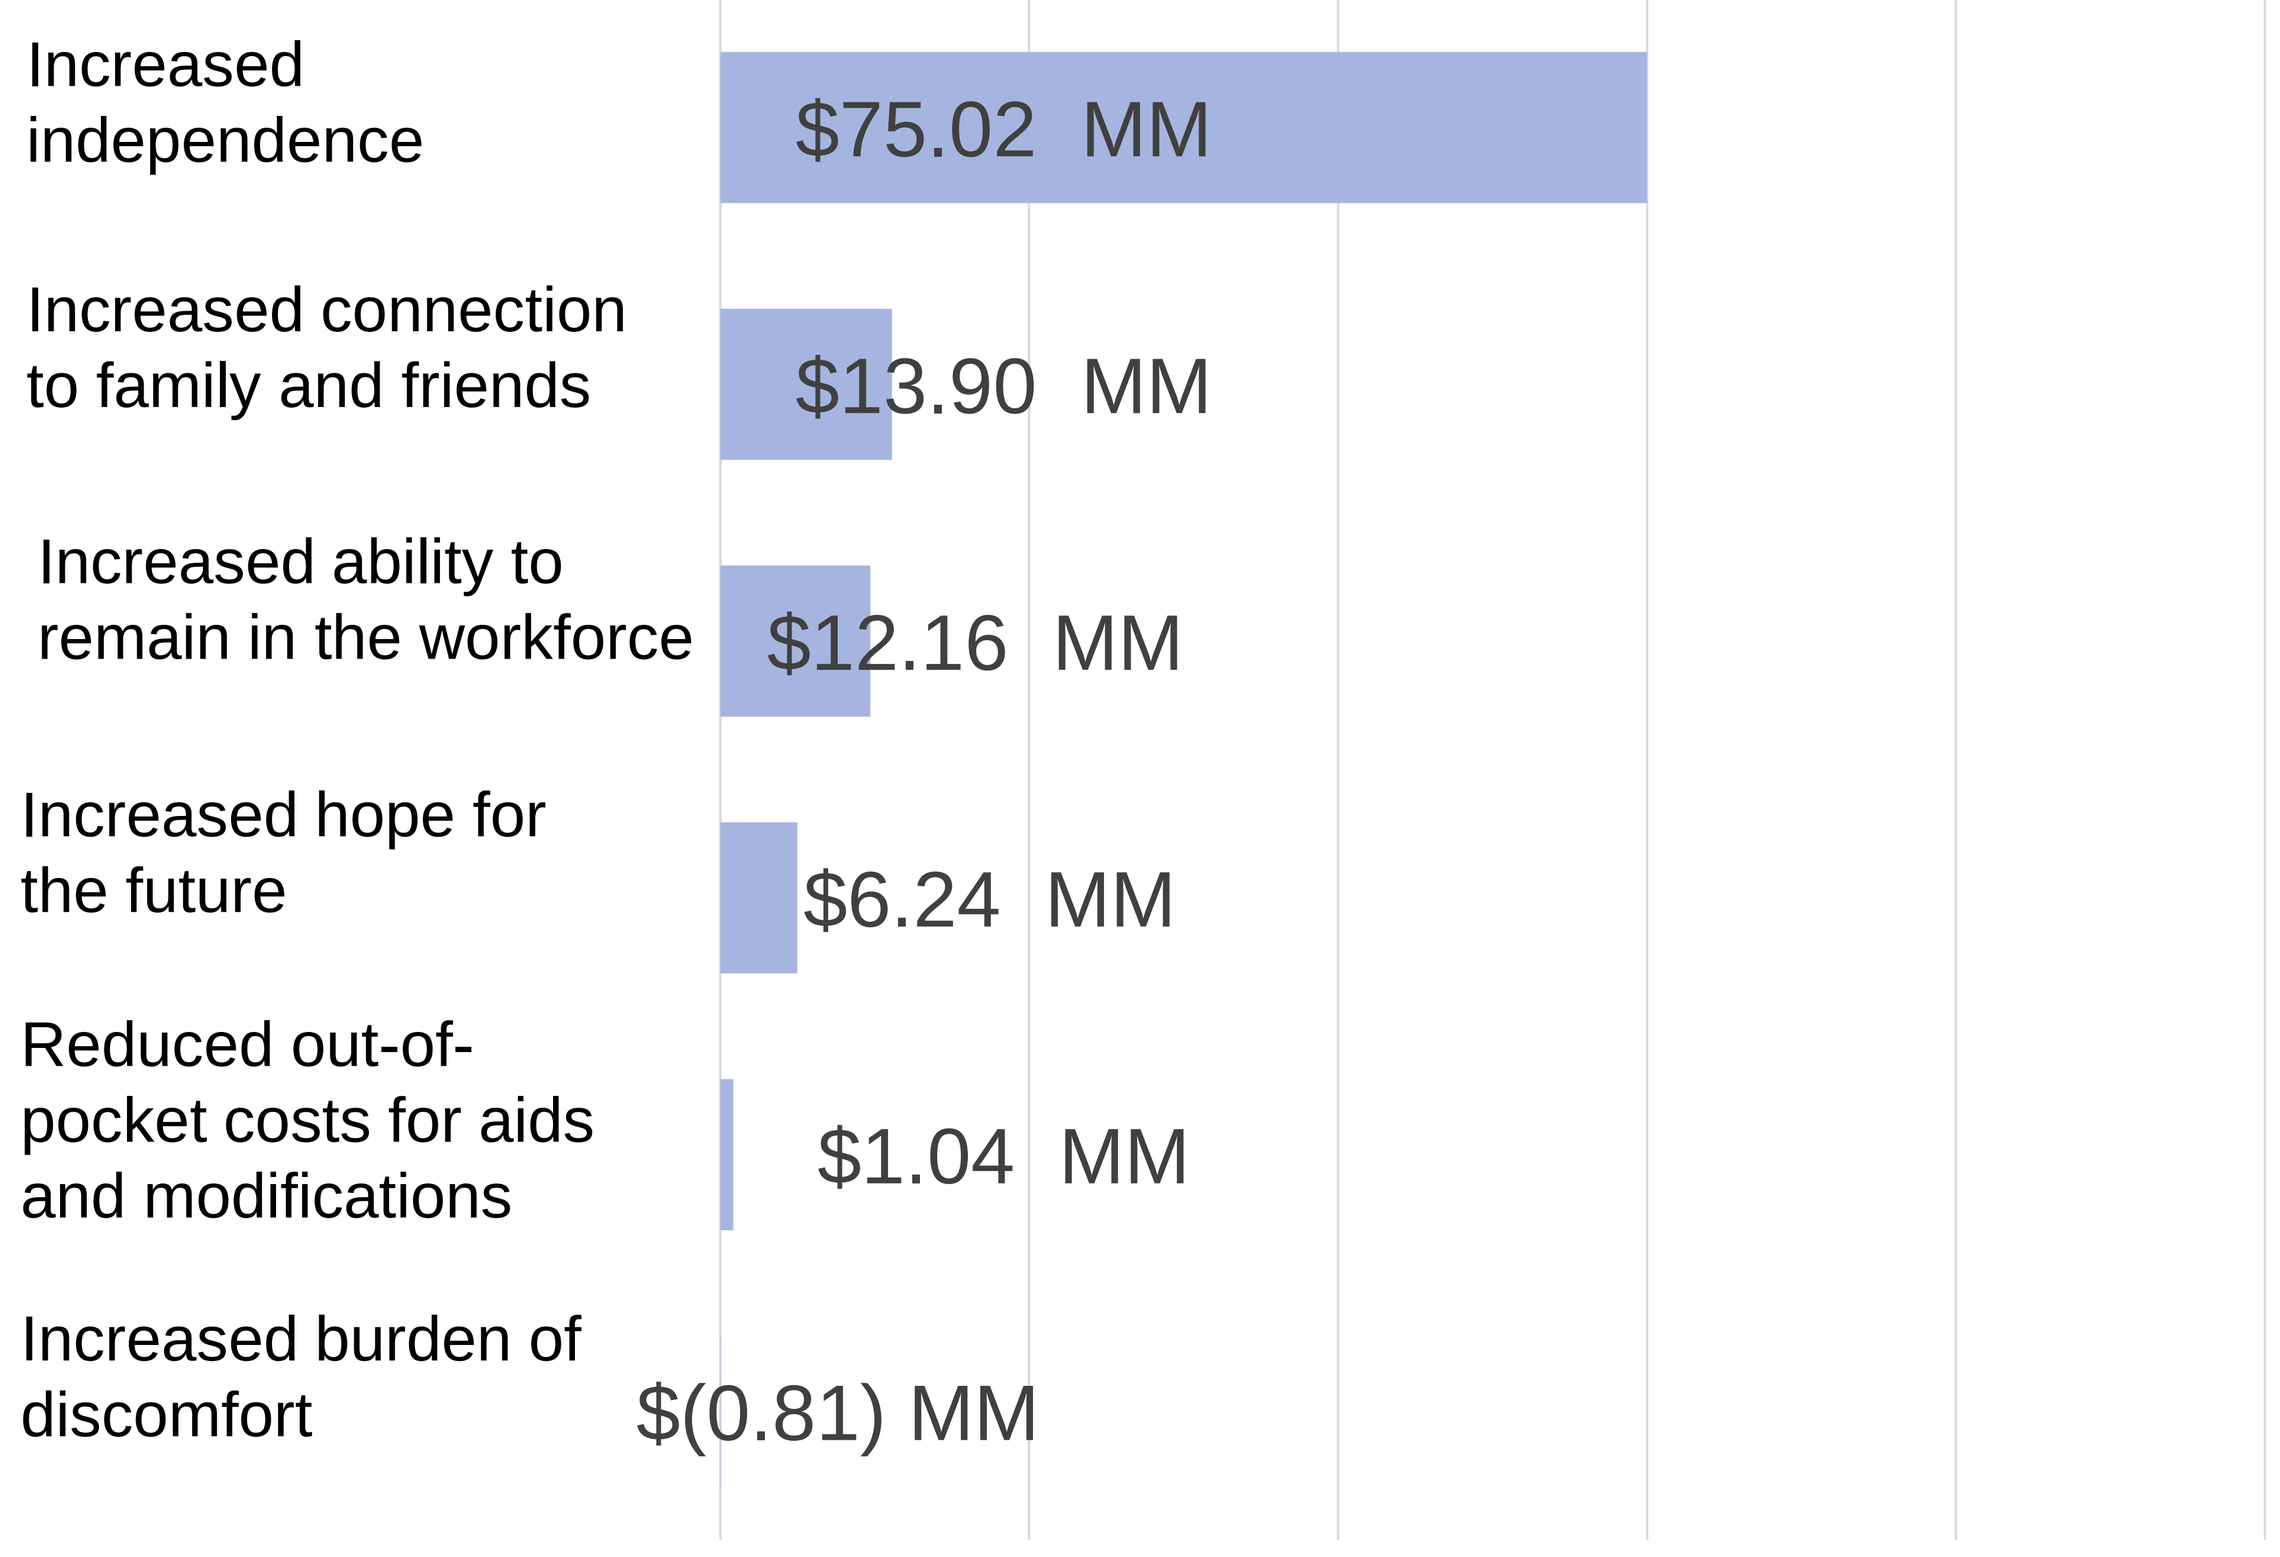

Increased independence
$75.02 MM
Increased connection to family and friends
$13.90 MM
Increased ability to remain in the workforce
$12.16 MM
Increased hope for the future
$6.24 MM
Reduced out-of-pocket costs for aids and modifications
$1.04 MM
Increased burden of discomfort
$(0.81) MM

"The best part is we have a social life again! Reconnecting with my friends and spending time with my family has brought me so much joy and happiness. – Person living with aPD receiving treatment with a LD-based DAT [1]

Outcomes for partners of people living with aPD

Social and emotional outcomes as a result of increased access to DATs.

$90.42 MILLION total value for partners of people living with aPD

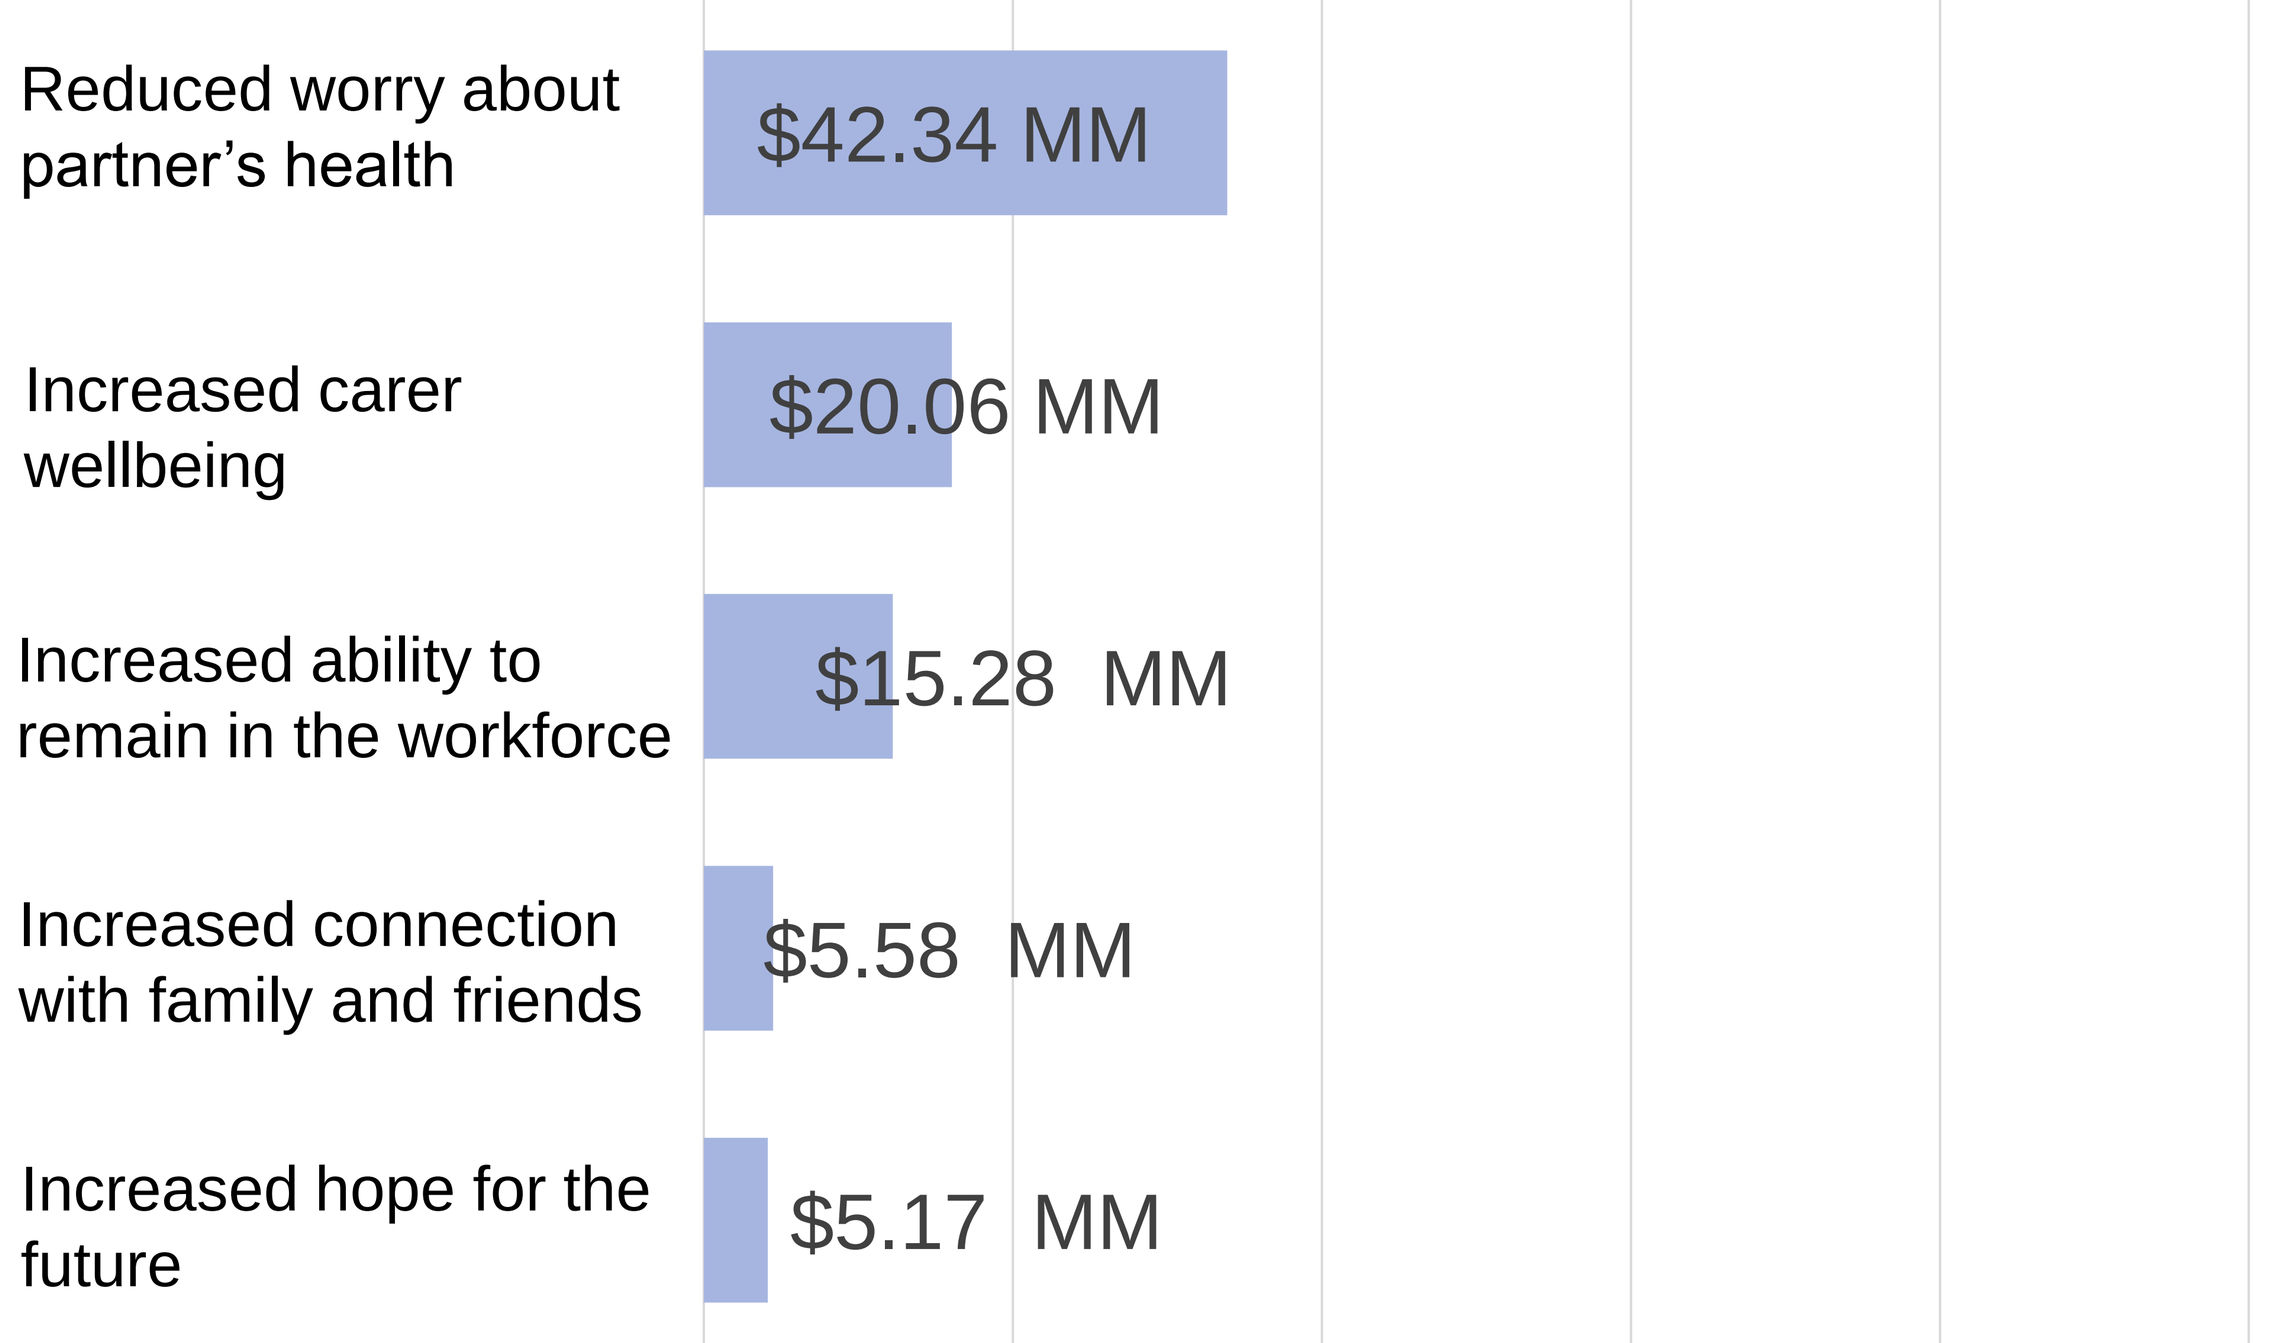

Reduced worry about partner's health
$42.34 MM
Increased carer wellbeing
$20.06 MM
Increased ability to remain in the workforce
$15.28 MM
Increased connection with family and friends
$5.58 MM
Increased hope for the future
$5.17 MM

"We are so much happier. We were given life back. My wife doesn't have to worry anymore. – Partner of a person living with aPD receiving treatment with a LD-based DAT [1]

Source: (1) Canadian Agency for Drugs and Technologies in Health (CADTH). CADTH Common Drug Review: Patient Input. Levodopa/Carbidopa (Duodopa) Indication: Parkinson's Disease.

The Professional Society for Health Economics and Outcomes Research (ISPOR EU); November 2023

N.B. All dollar values are in Australian Dollars.