Preliminary results from a teleassistance-based program in a real-life diabetes Colombian patient cohort

Mejia-Torres S[1], Camacho-Misol C[1], Londono S[2], García W[2], Silva L[2], Upegui A[2]

[1]Viva1A, Bogotá, Colombia. [2]Sanofi, Bogotá, Colombia

Poster # HSD57
ISPOR Europe 2023
November 12-15
Copenhagen, Denmark

POSTER HIGHLIGHT: Collaborative work and joint efforts between different Healthcare System actors create a positive impact on patients, translating into better health outcomes, increasing patient quality of life, and eliminating access barriers.

INTRODUCTION

- "Viva takes care of You" is a program implemented by Viva1A, an HCO with over 3 million affiliates that includes around 350,000 individuals living with chronic non-communicable diseases (NCD).
- This teleassistance program sponsored by Sanofi seeks to recover adherence to the cardiovascular risk program (focused on reaching disease control) in patients with more than 6 months without follow-up consultation.

OBJECTIVE

This study aims to describe preliminary clinical and operational results for the diabetic population in the teleassistance program.

METHODS

- The program monitors non-adherent NCD patients (over six months without follow-up consultation) over a period of 18 months.
- During follow-up, psychosocial and environmental factors were assessed to identify access barriers, create alerts, and implement supportive actions (scheduling medical appointments and administrative task guidance) to ensure program adherence.
- The primary outcome was program adherence defined as participation in at least one learning and educational activity, attendance in outpatient consultations, and disease-related testing.
- Other outcomes included glycemic control at follow-up (HbA1c<7%), and healthcare services quality perception measured by a third party at outpatient consultations.

Figure 1. Sociodemographic characteristics

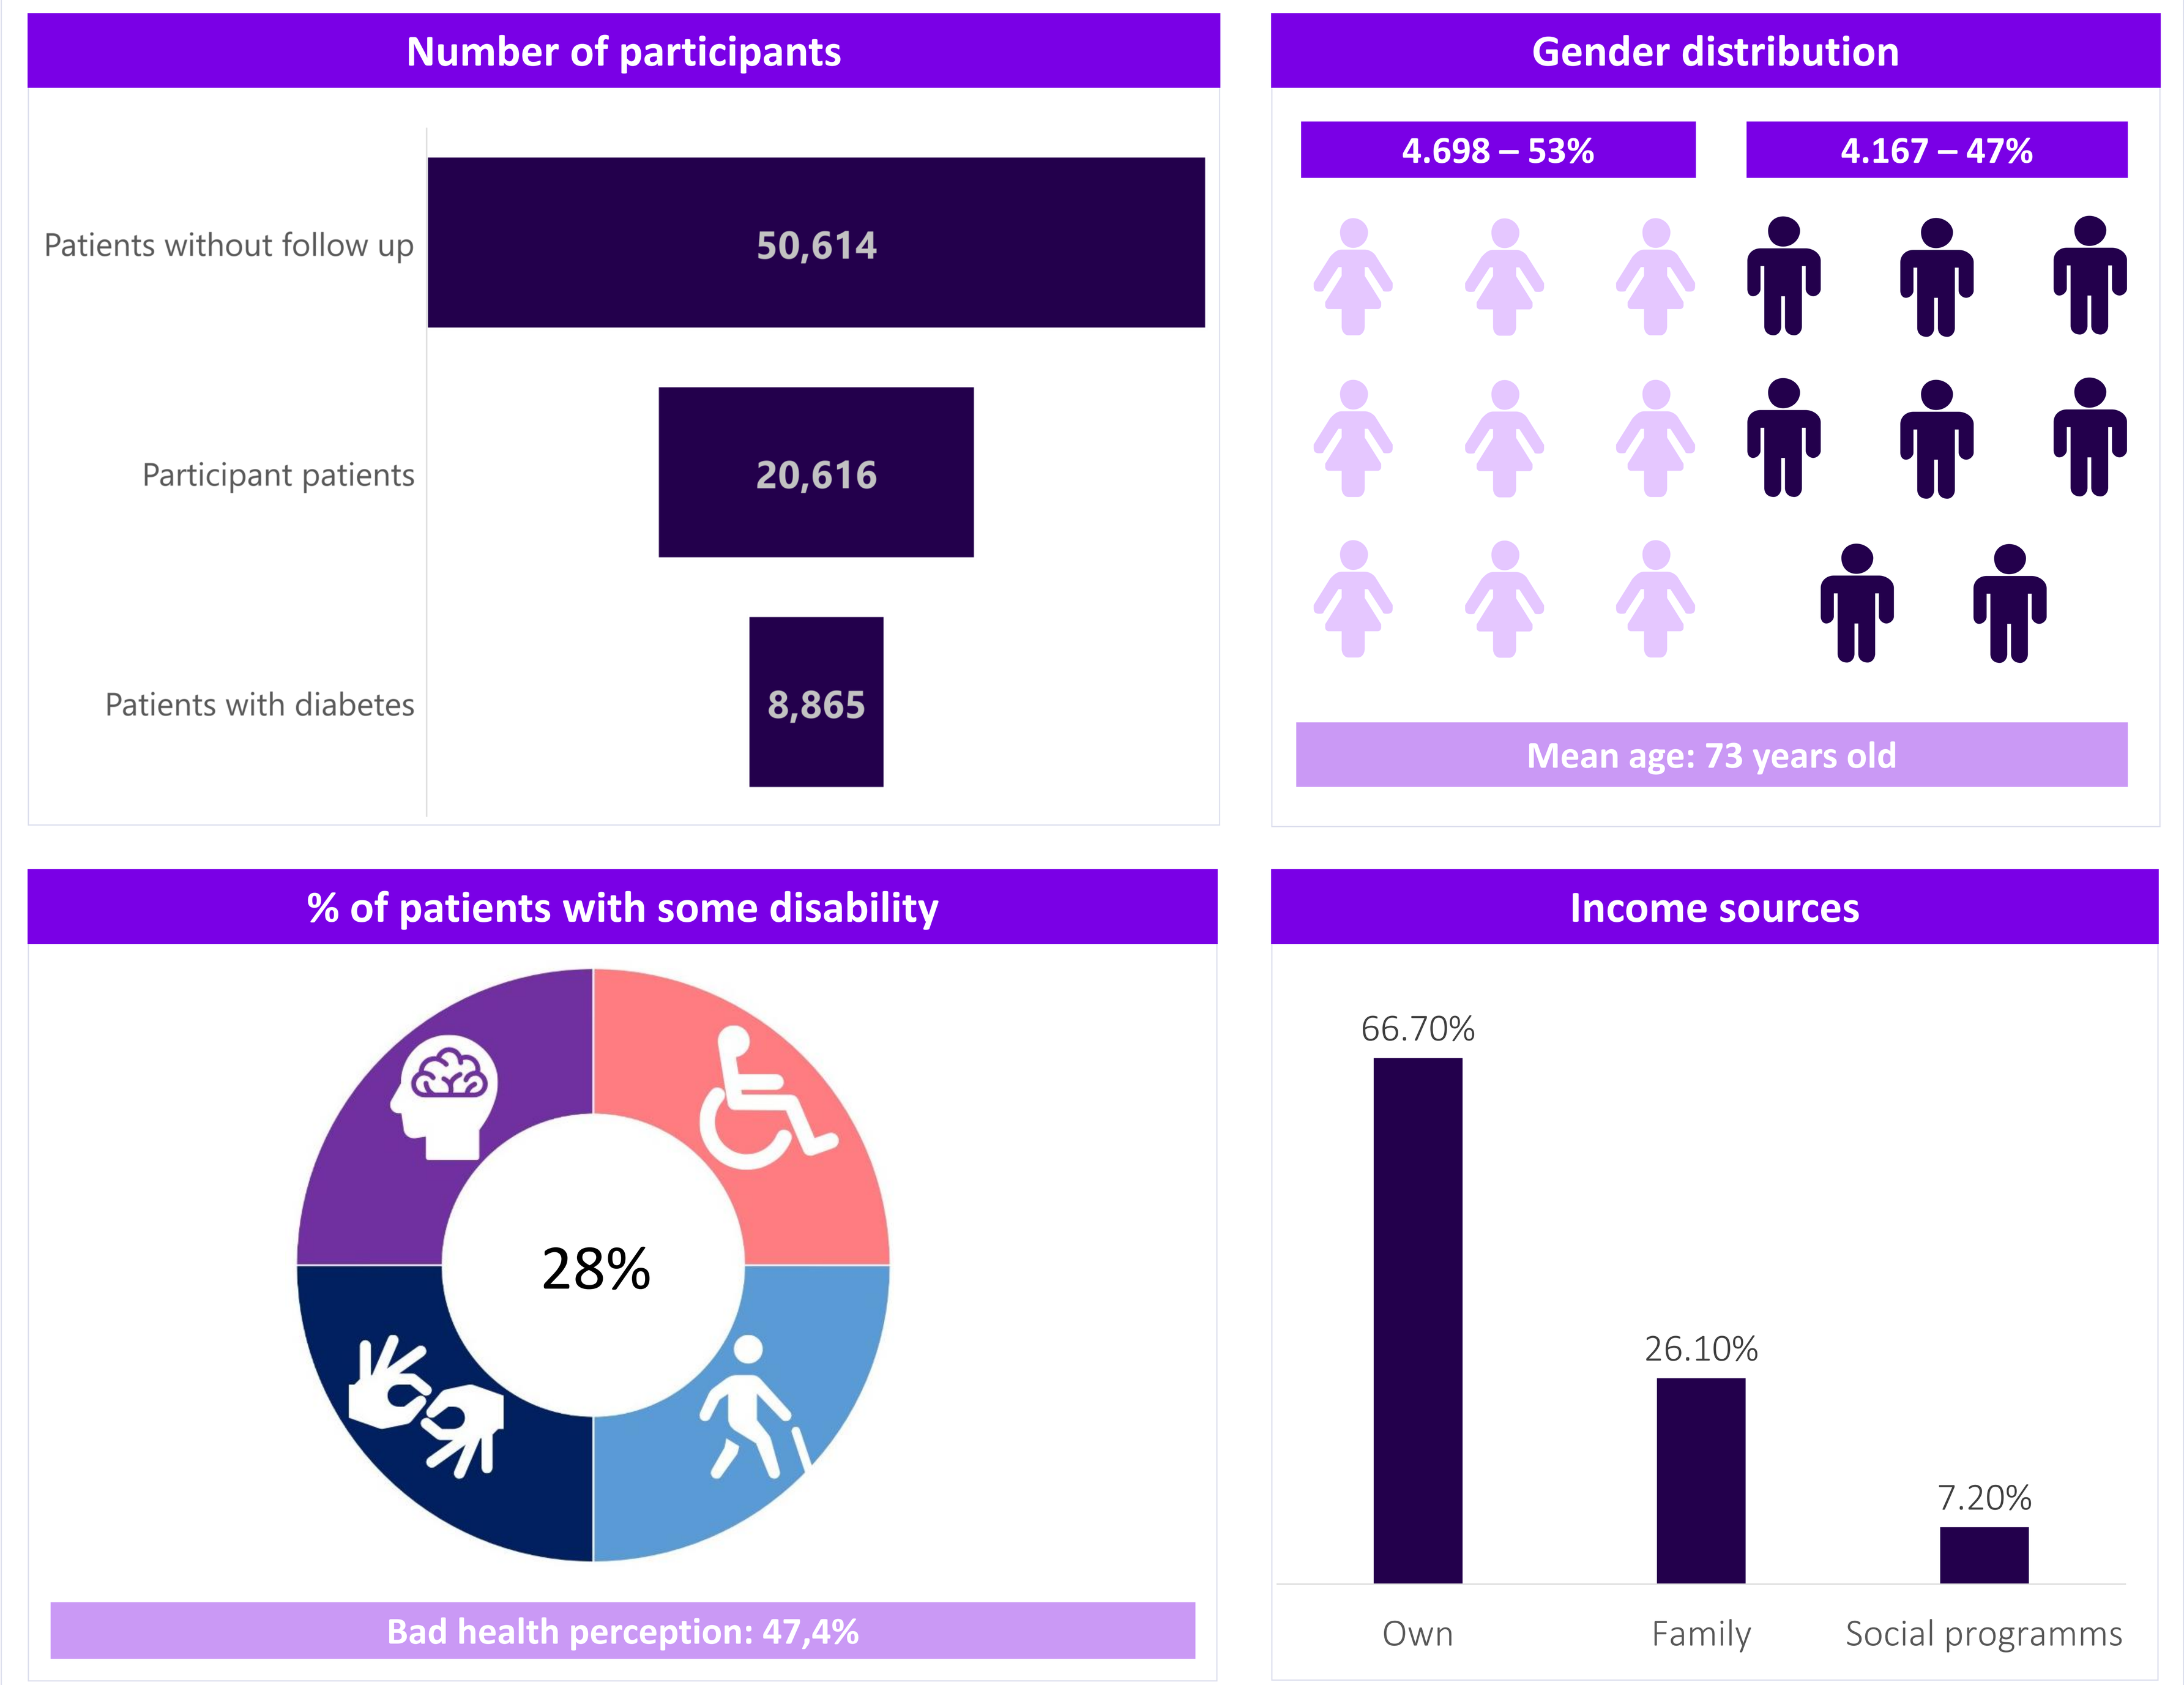
Number of participants
Patients without follow up
50,614
Participant patients
20,616
Patients with diabetes
8,865
Gender distribution
4.698 – 53%
4.167 – 47%
Mean age: 73 years old
% of patients with some disability
28%
Bad health perception: 47,4%
Income sources
66.70%
26.10%
7.20%
Own
Family
Social programms

Figure 2. Results

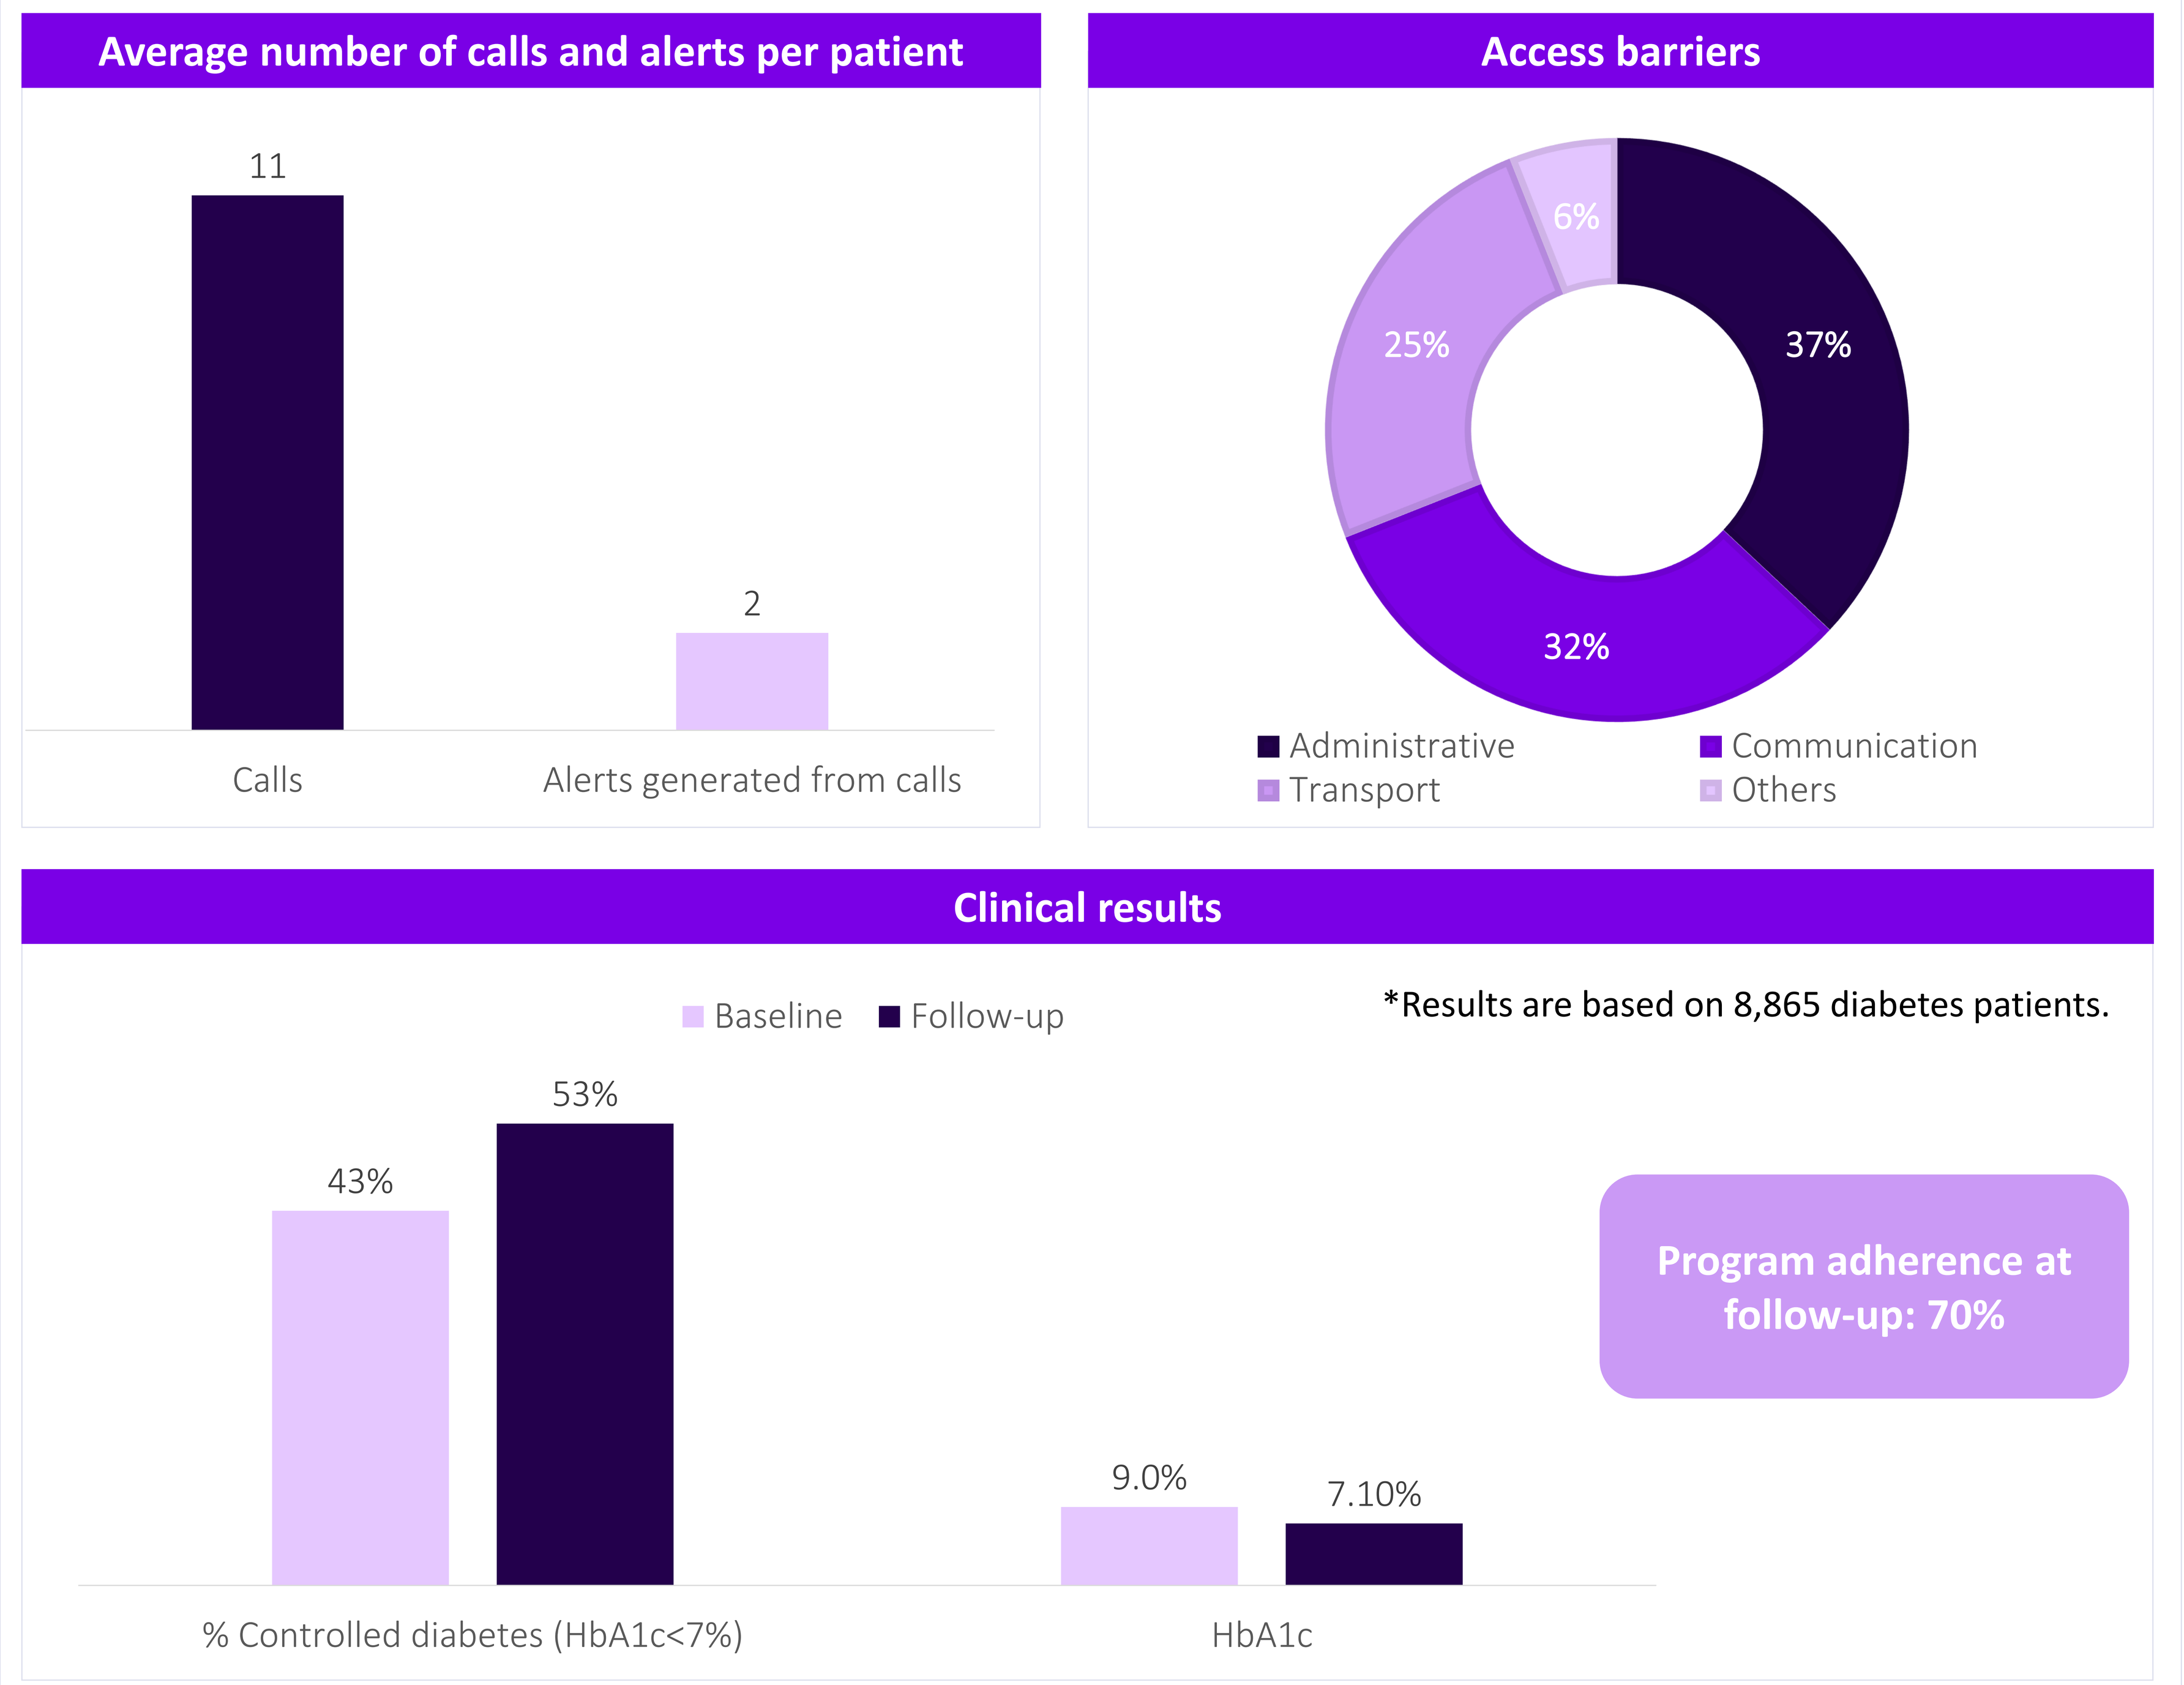
Average number of calls and alerts per patient
11
2
Calls
Alerts generated from calls
Access barriers
6%
25%
37%
32%
Administrative
Communication
Transport
Others
Clinical results
Baseline
Follow-up
*Results are based on 8,865 diabetes patients.
43%
53%
9.0%
7.10%
% Controlled diabetes (HbA1c<7%)
HbA1c
Program adherence at follow-up: 70%

Follow-up period: 6 months

RESULTS

- Out of 50,614 patients without follow-up, 20,616 (41%) agreed to participate, with 8,865 being diagnosed with diabetes.
- Patients' mean age was 73 years, 53% were women, and 28% reported having some disability.
- Over 30% of patients require economic support from family and social programs.
- A total of 239,442 calls were made. Among participants, an average of 11 calls and 2 alerts were reported.
- Delays in medical appointments (32%) and transportation (28%) were the main access barriers identified. All of these were completely solved through the program assistance; resulting in 70% of patients becoming adherent to the program.
- At baseline, 57% of patients had inadequate diabetes control (average HbA1c 9.0%). In the second measurement, uncontrolled patients decreased to 47%, (Average HbA1c 7.1%).
- Healthcare quality perception increased from 90% to 93%.

CONCLUSIONS

Preliminary results of this program show that patient supporting tools help to identify and solve main treatment barriers, hence improving treatment access and adherence to programmed appointments and procedures, acting as a complementary strategy to pharmacological treatment to improve disease control.

Author contact information: William Garcia – william.garcia@sanofi.com
Study sponsored by Sanofi.
LS, GW, SL and UA are employees of Sanofi and may own shares and/or stock options in the company.
MTS and CMC are employees of VIVA1A.