Healthcare models for Rare Diseases: A systematic literature review for the development of a comprehensive model in a Latin American country

Ossa M[1], Londono S[1] , Rodríguez-Ordóñez P[1]
[1]Sanofi, Bogota, Colombia

Poster # HSD41
ISPOR Europe 2023
November 12-15
Copenhagen, Denmark

INTRODUCTION

The complexity of rare diseases (RD) and healthcare systems represents a challenge for implementing a healthcare model for each disorder as well as a standardized process applicable at global level.

OBJECTIVE

To systematically review evidence about RD healthcare models and identify strategies that should be considered in the development of a comprehensive value-based healthcare model in Colombia.

METHODS

- Systematic literature review of manuscripts in English or Spanish, published from 2018 to 2023 in five electronic databases (Cochrane, Pubmed, Scielo, LILACS and MEDLINE).
- The search strategy included the keywords: Healthcare model, model of care, care pathway, rare diseases, and orphan diseases.
- Duplicated studies were excluded and publications with relevant information in the title or abstract were screened.
- An additional snowball exercise was undertaken.
- All selected studies were subjected to full-text screening.

POSTER HIGHLIGHT: There is a lack of evidence regarding RD healthcare models in Latin America. This SLR reflects the need of a coordinated, multidisciplinary model for RD patients.

RESULTS

- A total of 1,891 studies were identified, 1,045 were excluded. Twenty studies were retrieved for final screening, with eleven making the final selection. Five additional articles from the snowball process were added.
- Eleven studies included models developed in Europe, one in Australia and one in India.
- Three studies reported strategies that could be applied worldwide, with two of these highlighting the importance of caregivers and patient journeys, and one reporting the global goals established for the management of RD.
- No models were identified in Latin America.
- All studies agreed on the need of a coordinated, multidisciplinary model that integrates different components and processes for the benefit of patients and their caregivers.

Figure 1: SLR Prisma Diagram

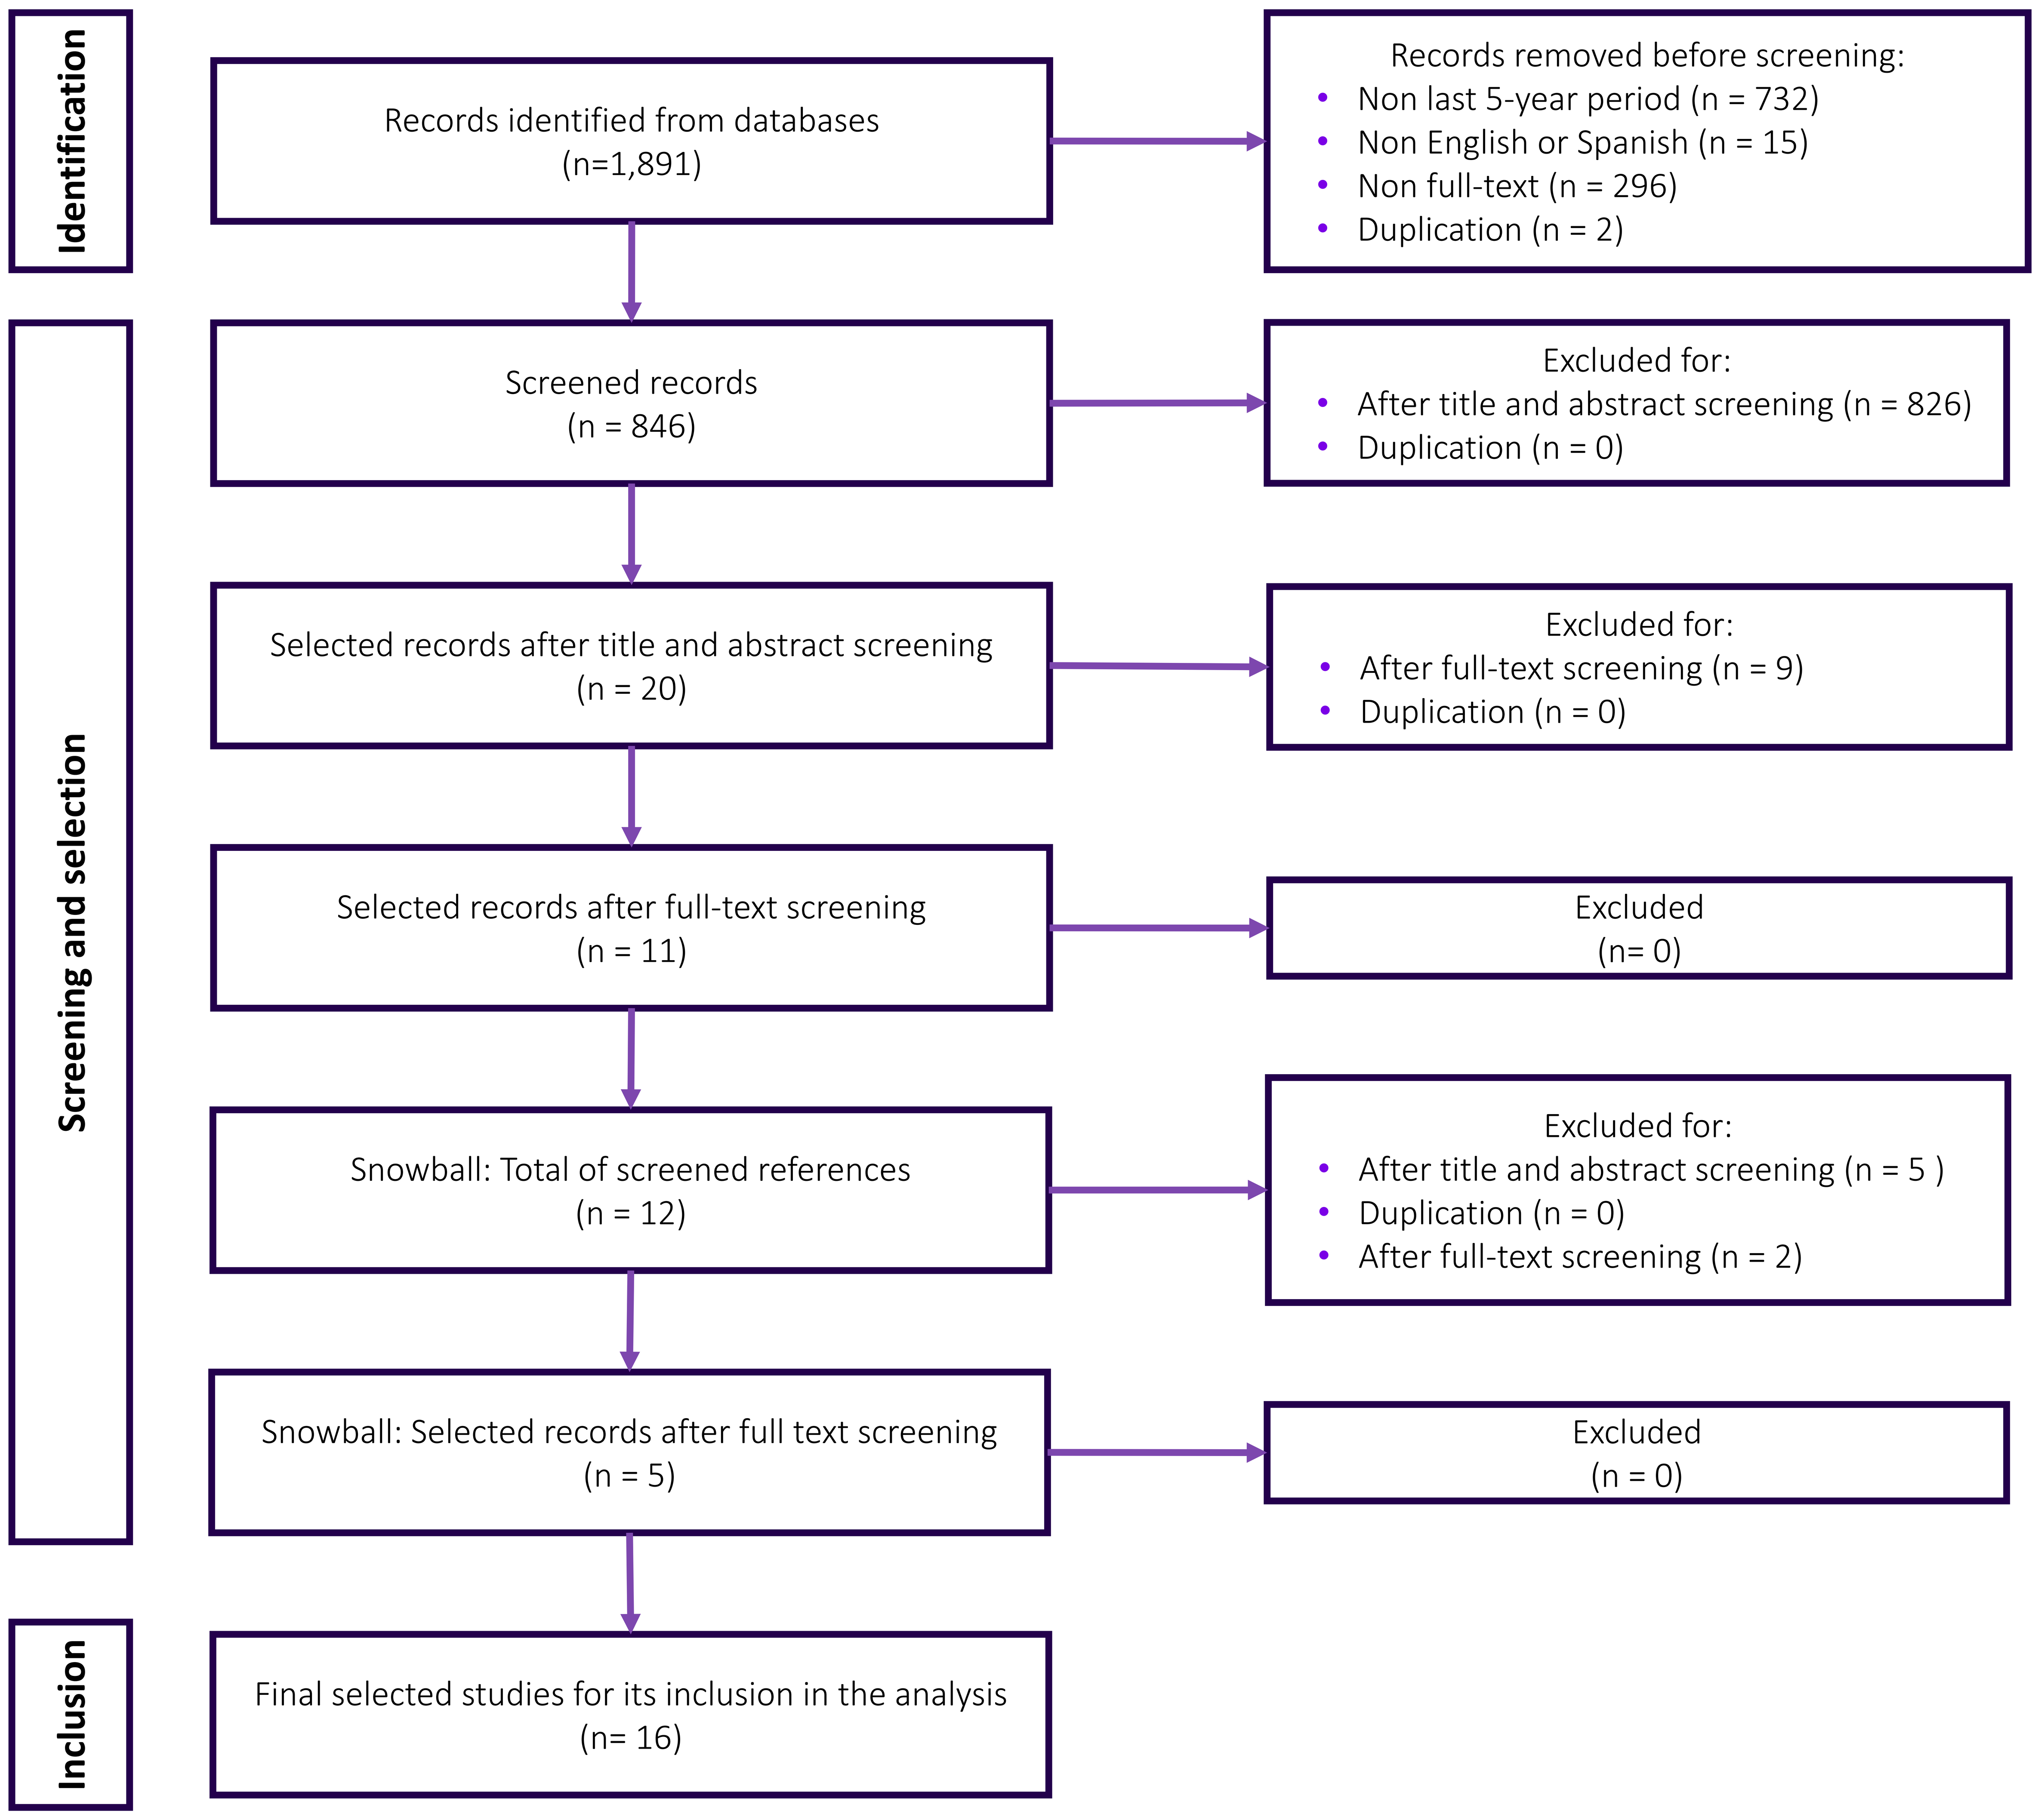
Identification
Records identified from databases (n=1,891)
Records removed before screening:
Non last 5-year period (n = 732)
Non English or Spanish (n = 15)
Non full-text (n = 296)
Duplication (n = 2)
Screening and selection
Screened records (n = 846)
Excluded for:
After title and abstract screening (n = 826)
Duplication (n = 0)
Selected records after title and abstract screening (n = 20)
Excluded for:
After full-text screening (n = 9)
Duplication (n = 0)
Selected records after full-text screening (n = 11)
Excluded (n= 0)
Snowball: Total of screened references (n = 12)
Excluded for:
After title and abstract screening (n = 5)
Duplication (n = 0)
After full-text screening (n = 2)
Snowball: Selected records after full text screening (n = 5)
Excluded (n = 0)
Inclusion
Final selected studies for its inclusion in the analysis (n= 16)

CONCLUSIONS

The development of a RD healthcare model for Colombia should include a coordinated, multidisciplinary care based on a patient-centered approach, involving the role of caregivers, and technological tools such as telemedicine, to facilitate patient access to early diagnosis, correct treatment, and adherence and persistence.

REFERENCES

1. Ward AJ et al. Designing rare disease care pathways in the Republic of Ireland: a co-operative model. Orphanet J Rare Dis. 2022 Apr 11;17(1):162.
2. Talarico R et al. RarERN Path: a methodology towards the optimisation of patients' care pathways in rare and complex diseases developed within the European Reference Networks. Orphanet J Rare Dis. 2020 Dec 14;15(1):347. Erratum in: Orphanet J Rare Dis. 2021 Mar 22;16(1):146.
3. Csanádi M et al. Prioritization of implementation barriers related to integrated care models in Central and Eastern European countries. Health Policy. 2022 Nov;126(11):1173-1179.

Author contact information: Sergio Londoño – sergio.londono@sanofi.com
Study sponsored by Sanofi.
OM, LS and RP are employees of Sanofi and may own shares and/or stock options in the company.