Cost-Effectiveness Analysis of iGlarLixi Compared with Basal Bolus Insulin Regimen in Triple Metabolic Goal in Type 2 Diabetes Mellitus in Latin America

Londono S[1], Garcia W[1], Salamanca C[1], Silva L[1], Santos V[1,2], Upegui A[1]

[1]Sanofi, Bogotá, Colombia [2]Universidad Externado de Colombia

Poster # EE621
ISPOR Europe 2023
November 12-15
Copenhagen, Denmark

INTRODUCTION

iGlarLixi has shown to be effective in decreasing Hb1AC, with a neutral effect on body weight, and no increased risk of hypoglycemic events (Triple Metabolic Goal [TMG]) in adults with type 2 diabetes (T2DM).

OBJECTIVE

This study aims to calculate the achievement of the composite endpoints (CE) costs with iGlarLixi compared to Basal-Bolus (rapid-acting insulin plus Gla-U100 [B/B]) in 5 Latin-American countries: Panama, Costa Rica, Salvador, Guatemala, and Honduras.

METHODS

- A cost-effectiveness analysis was done to determine the CE achievement costs in T2DM adults in one-year time horizon considering the proportion of patients that achieve the triple metabolic goal (TMG).
- The cost-effectiveness index (CEI) was calculated as the quotient between the cost per patient achieving the CE with B/B and iGlarLixi.
- The percentage of patients reaching the TMG was extracted from published literature.
- Analysis is made over a hypothetical cohort of 1 patient.
- Costs were extracted from local published literature and are expressed in 2022 USD.

POSTER HIGHLIGHT: To achieve the triple metabolic goal (TMG) in Latin America, USD$ 1.59 must be invested in a basal-bolus treatment for every USD$ 1 invested in iGlarLixi.

Table 1. Percentage of patients reaching therapeutic goals

Therapeutic Goal	iGlarLixi	B/B	Source
HbA1c <7%	55.2%	36.6%	Tabák et al, 2020
HbA1c <7% and no hypoglycemia	44.8%	24.2%	
HbA1c <7% and no weight gain	33.5%	16.5%	
Triple Metabolic Goal	26.8%	12.4%	

Table 2. Total annual treatment cost per country

	iGlarLixi	B/B	Difference %
Panamá	$2,221	$1,686	32%
Costa Rica	$2,083	$1,589	31%
El Salvador	$1,819	$1,314	38%
Guatemala	$2,668	$1,790	49%
Honduras	$1,923	$1,467	31%

Image 1. Effectiveness Index

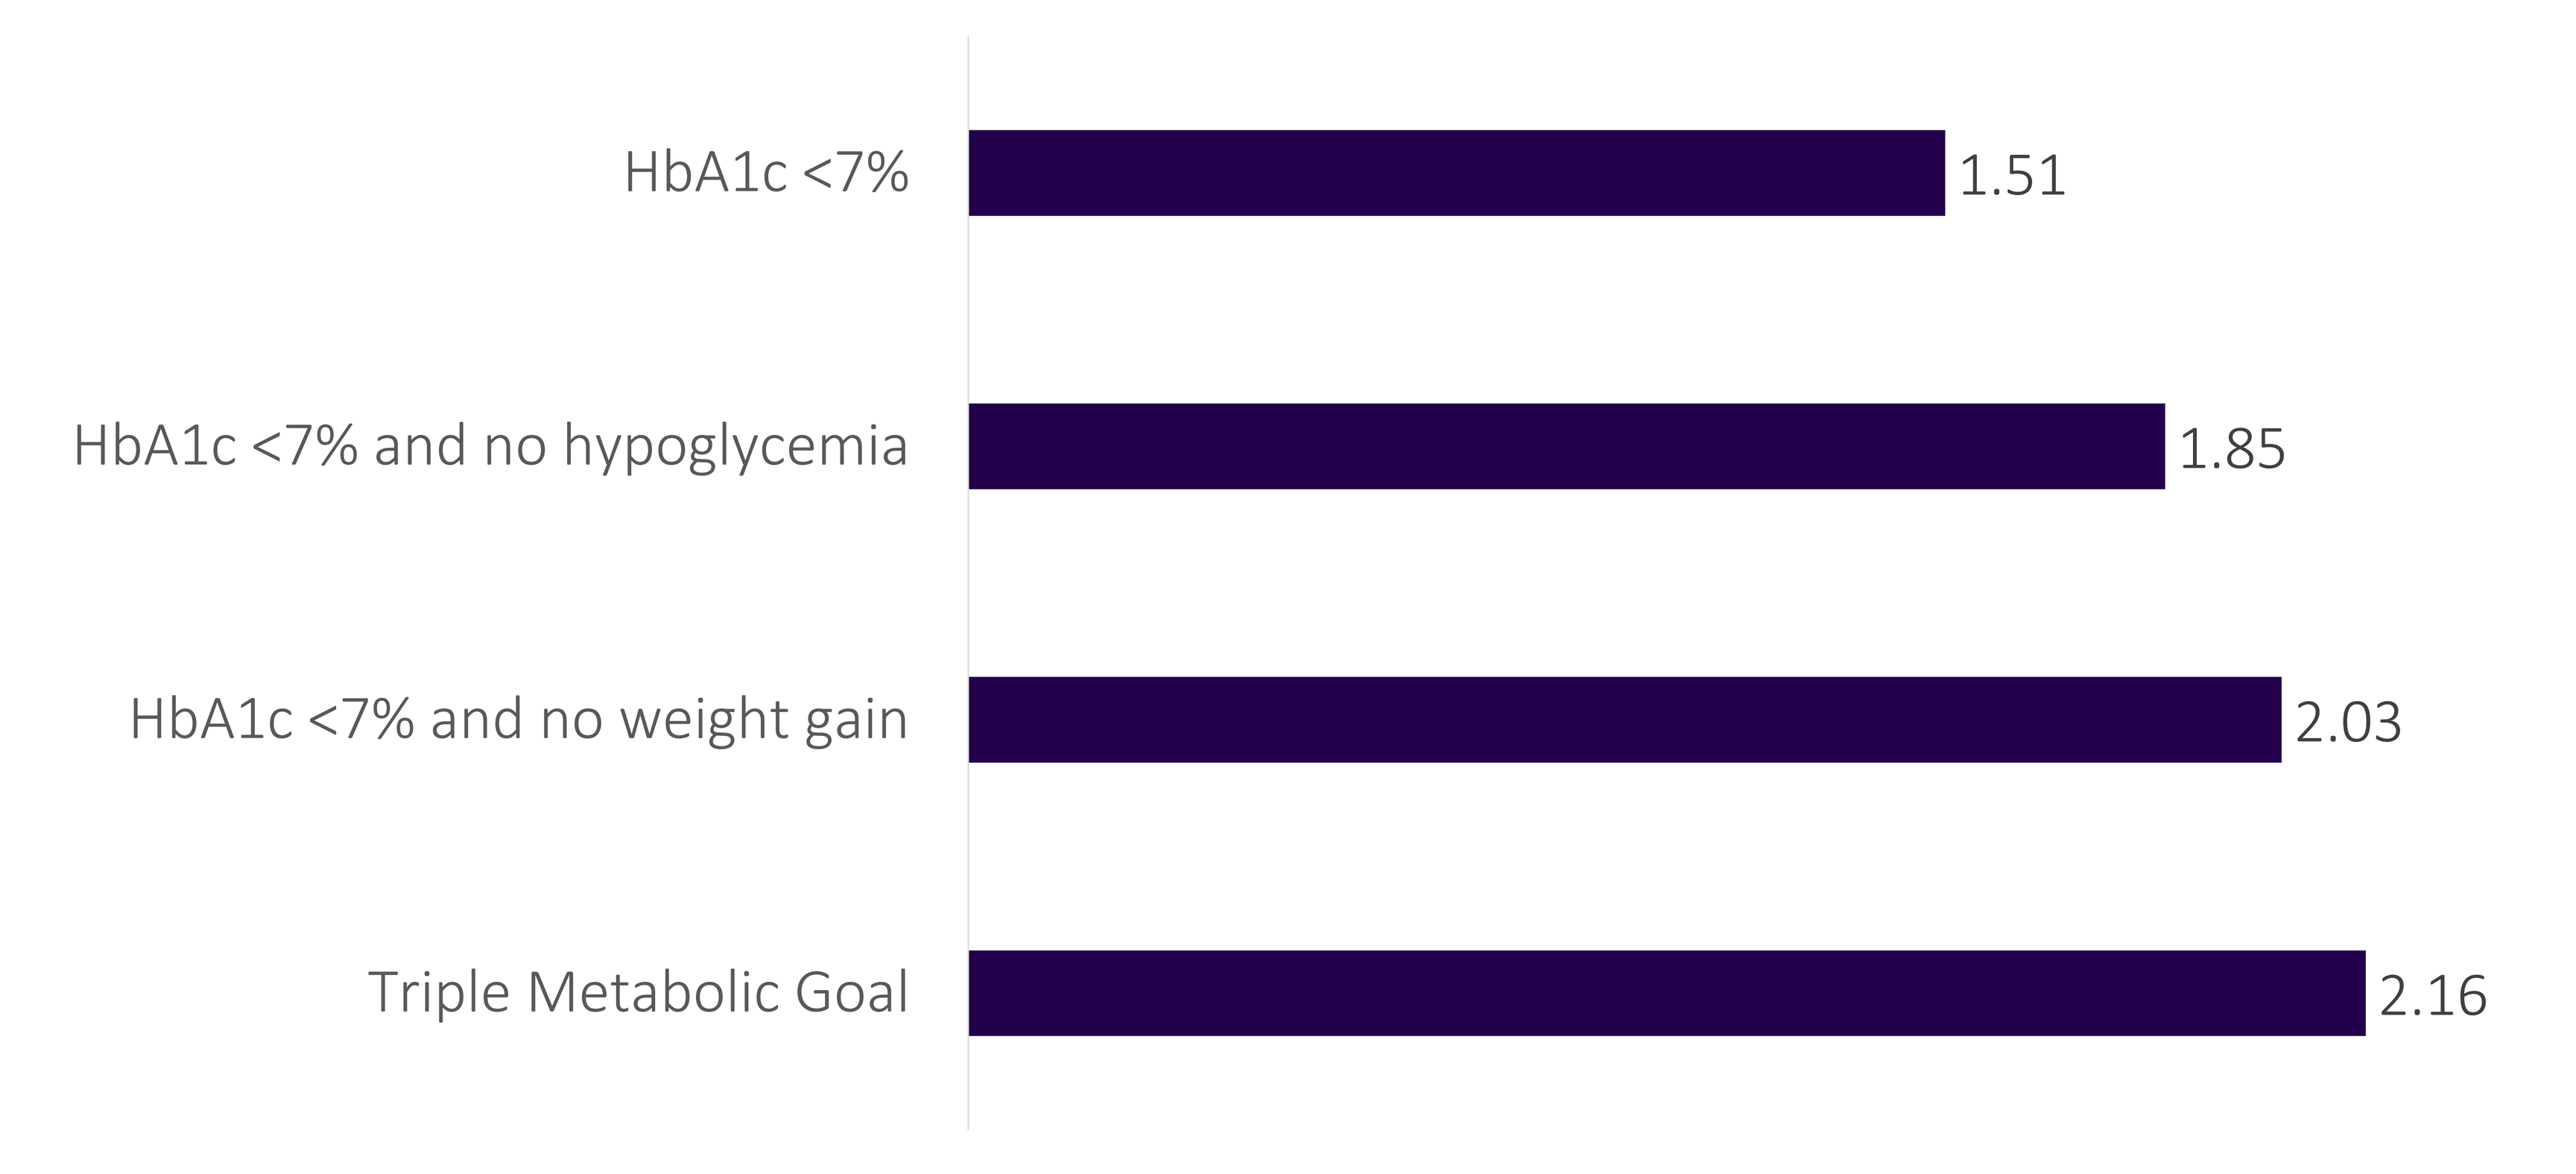
HbA1c <7%
1.51
HbA1c <7% and no hypoglycemia
1.85
HbA1c <7% and no weight gain
2.03
Triple Metabolic Goal
2.16

Image 2. Cost-effectiveness Index per country

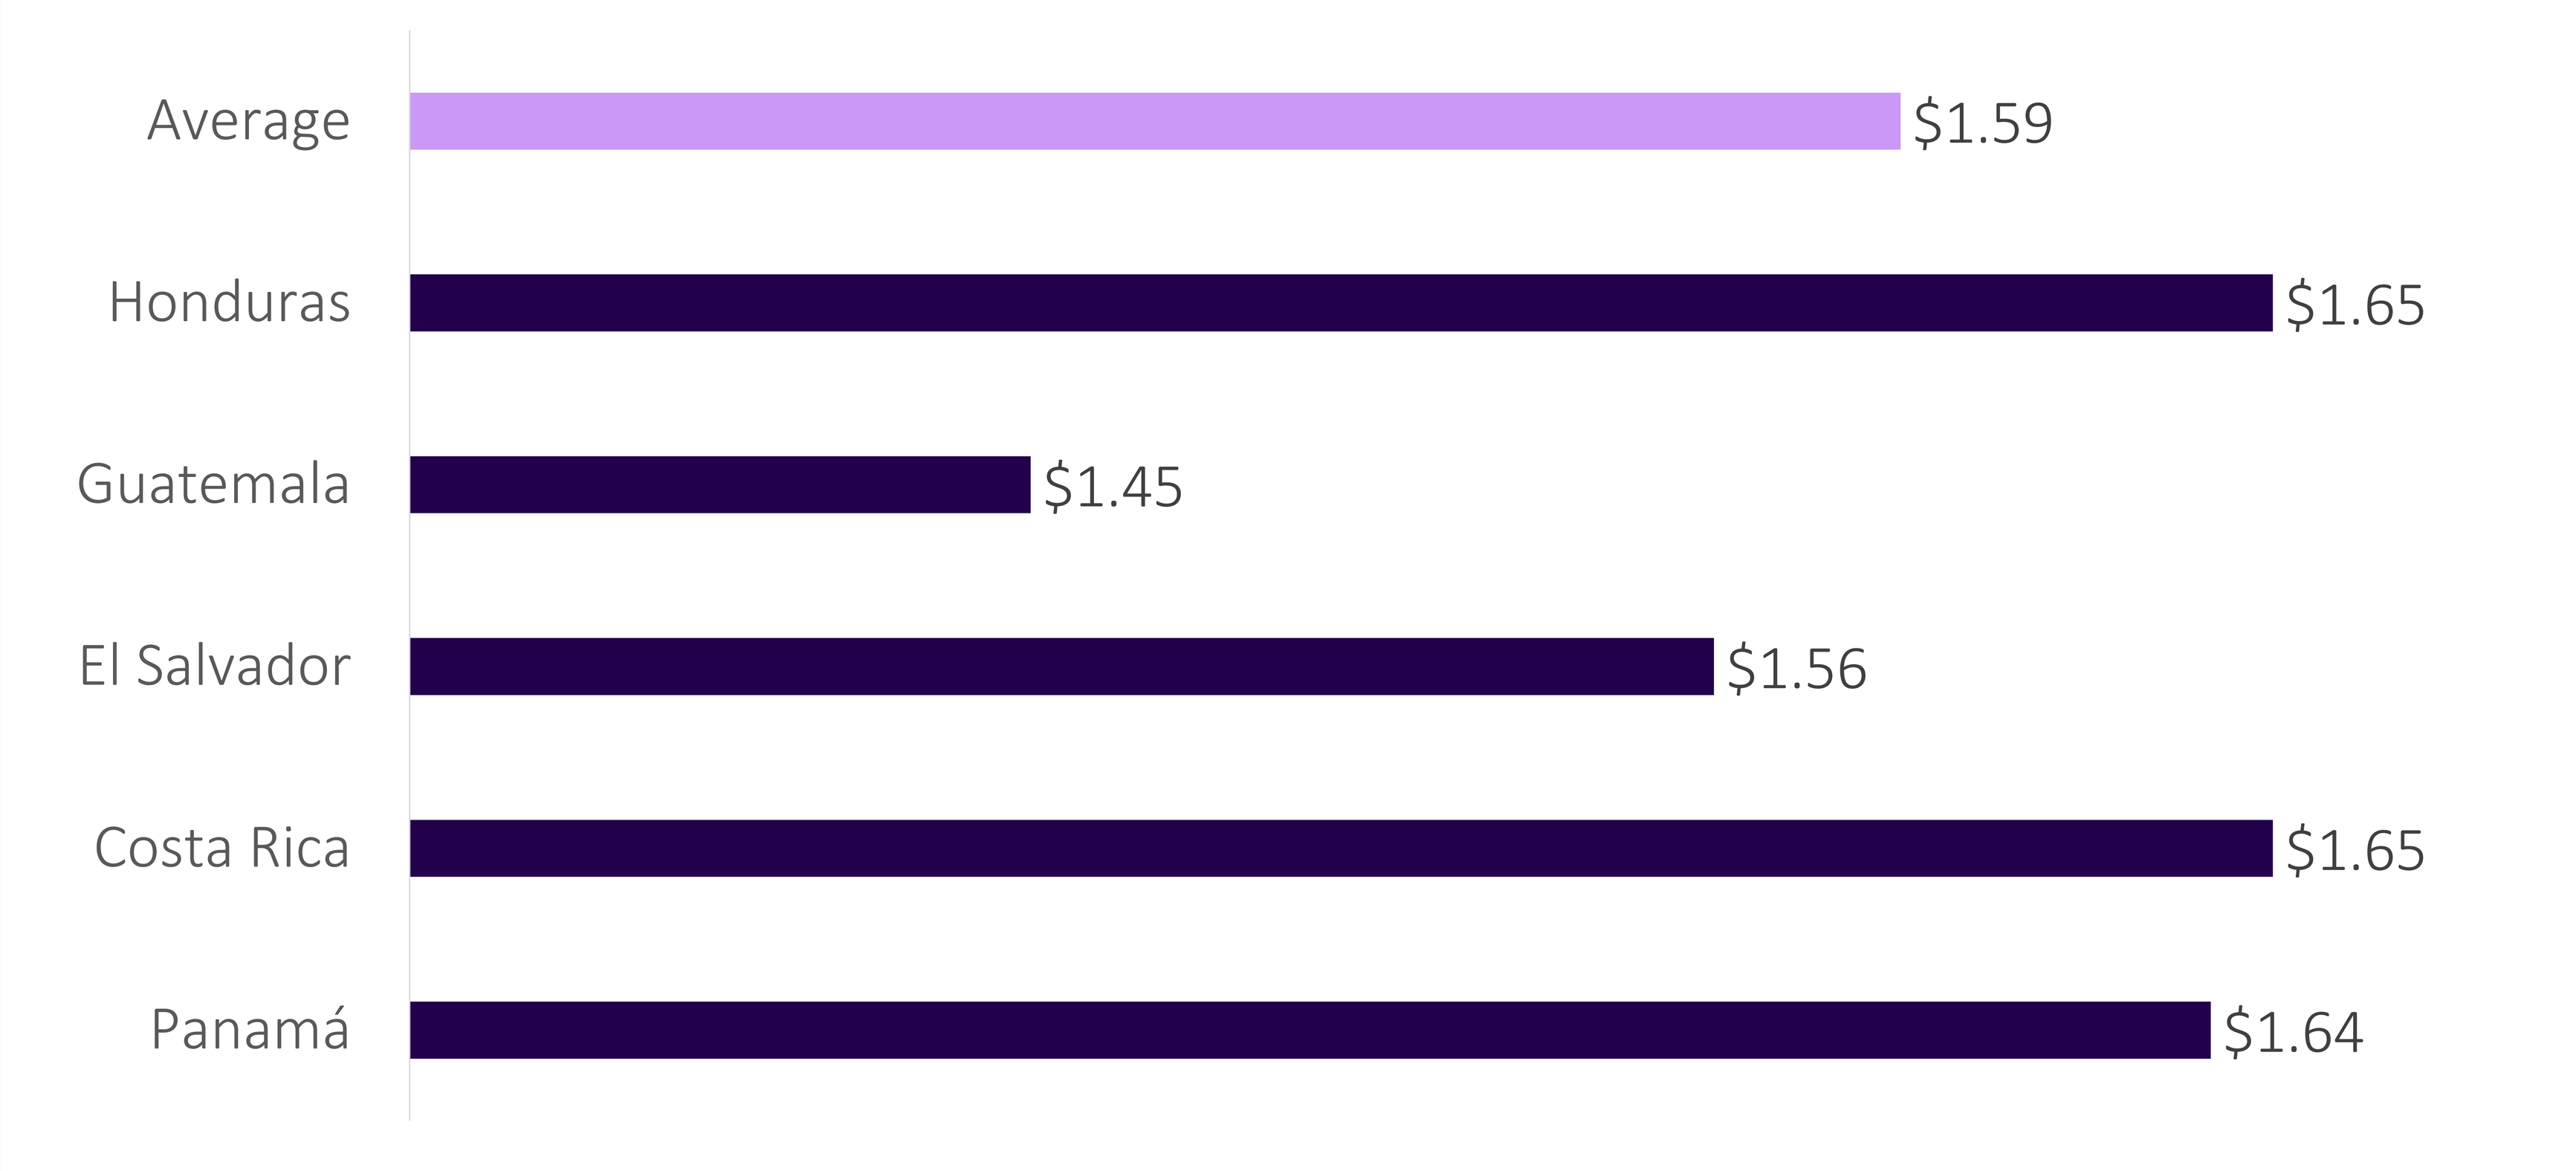
Average
$1.59
Honduras
$1.65
Guatemala
$1.45
El Salvador
$1.56
Costa Rica
$1.65
Panamá
$1.64

RESULTS

- The proportion of patients achieving the TMG for iGlarLixi and B/B was 26.8% and 12.4% respectively, indicating an efficacy index of 2.16.
- On average, the total annual cost of iGlarLixi treatment was USD$2,142 (USD$1,819-USD$2,668) vs USD$1,569 (USD$1,314-USD$1,790) for B/B, representing a difference of 36% (31%-49%).
- However, the cost per patient achieving the TMG with iGlarLixi was lower compared to B/B: USD$7,994 (USD$6,786-USD$9,954) vs USD$12,655 (USD$11,830-USD$14,433), respectively, equivalent to a 37% (31%-39%) lower cost favoring iGlarLixi.
- The average CEI was $1.59, with Honduras and Costa Rica exhibiting the highest index ($1.65), followed by Panama ($1.64), Salvador ($1.56) and finally Guatemala ($1.45).
- The larger the difference in treatment cost, the lower the CEI observed as can be seen for Guatemala with B/B having 49% lower treatment costs than iGlarLixi and having the lowest CEI.

CONCLUSIONS

- iGlarLixi has a lower cost per patient reaching the TMG compared to B/B. The study suggests that per each USD$1 invested in iGlarLixi to reach the TMG, it is necessary to invest, on average, USD$1,59 in B/B treatment to obtain the same therapeutic results in the different Latin-American countries included in this analysis.
- These clinical benefits and overall lower costs, can encourage developing nations in LATAM to invest in iGlarLixi instead of a B/B scheme.

REFERENCES

1. Tabák, Á. G., Anderson, J., Aschner, P., Liu, M., Saremi, A., Stella, P., & Meier, J. J. (2020). Efficacy and safety of iGlarLixi, fixed-ratio combination of insulin glargine and lixisenatide, compared with basal-bolus regimen in patients with type 2 diabetes: propensity score matched analysis. Diabetes Therapy, 11, 305-318.

2. Aroda, V. R., et al. (2016). Efficacy and safety of LixiLan, a titratable fixed-ratio combination of insulin glargine plus lixisenatide in type 2 diabetes inadequately controlled on basal insulin and metformin: the LixiLan-L randomized trial. Diabetes care, 39(11), 1972-1980.

Author contact information: Angie Upegui – angiegisseth.upeguipachon@sanofi.com
Study sponsored by Sanofi.
LS, GW, CS, SL and UA are employees of Sanofi and may own shares and/or stock options in the company.
SV worked as an intern in Sanofi at the time of this study.