EE722

Cost-Utility Analysis for Haemate-P® in the Treatment of von Willebrand Disease

S. Linden[1], P. Méndez Valera[1], R. Tomic[2], Marco Panebianco[2], L. Vidal Barrientos[1], S. Yan[3], M. Wilson[4]

[1]CSL Iberia SA, Barcelona, Spain. [2]CSL Behring S.p.A, Milan, Italia. [3]CSL Behring, King of Prussia, PA, United States. [4]RTI Health Solutions, Research Triangle Park, NC, United States

Introduction

- Von Willebrand Disease (vWD) is a blood disease caused by a deficiency of von Willebrand Factor (vWF), a fundamental protein in blood coagulation (1).
- vWD is the most common congenital bleeding disorder, and it is estimated to have a prevalence of symptomatic disease between 0,01% and 0,1% (2).
- Different treatment alternatives are currently available. Economic evaluation, particularly cost-utility analysis, will assist evidence-based decision-making.

Objective

This study aimed to compare the **cost-utility** of **Haemate-P®** versus **Fanhdi® and Wilate®** in the treatment of vWD **under different treatment regimes** (long-term prophylaxis, intermittent prophylaxis and on-demand) from the **Spanish National Health System** perspective.

Methods

- A cost-utility analysis was performed using a Markov model with 6-month cycles over a patient's lifetime. Health states considered were: "no joint surgery", "joint surgery", and "death". Moreover, two possible bleed events were included (Figure 1).

Figure 1. Markov Model Structure

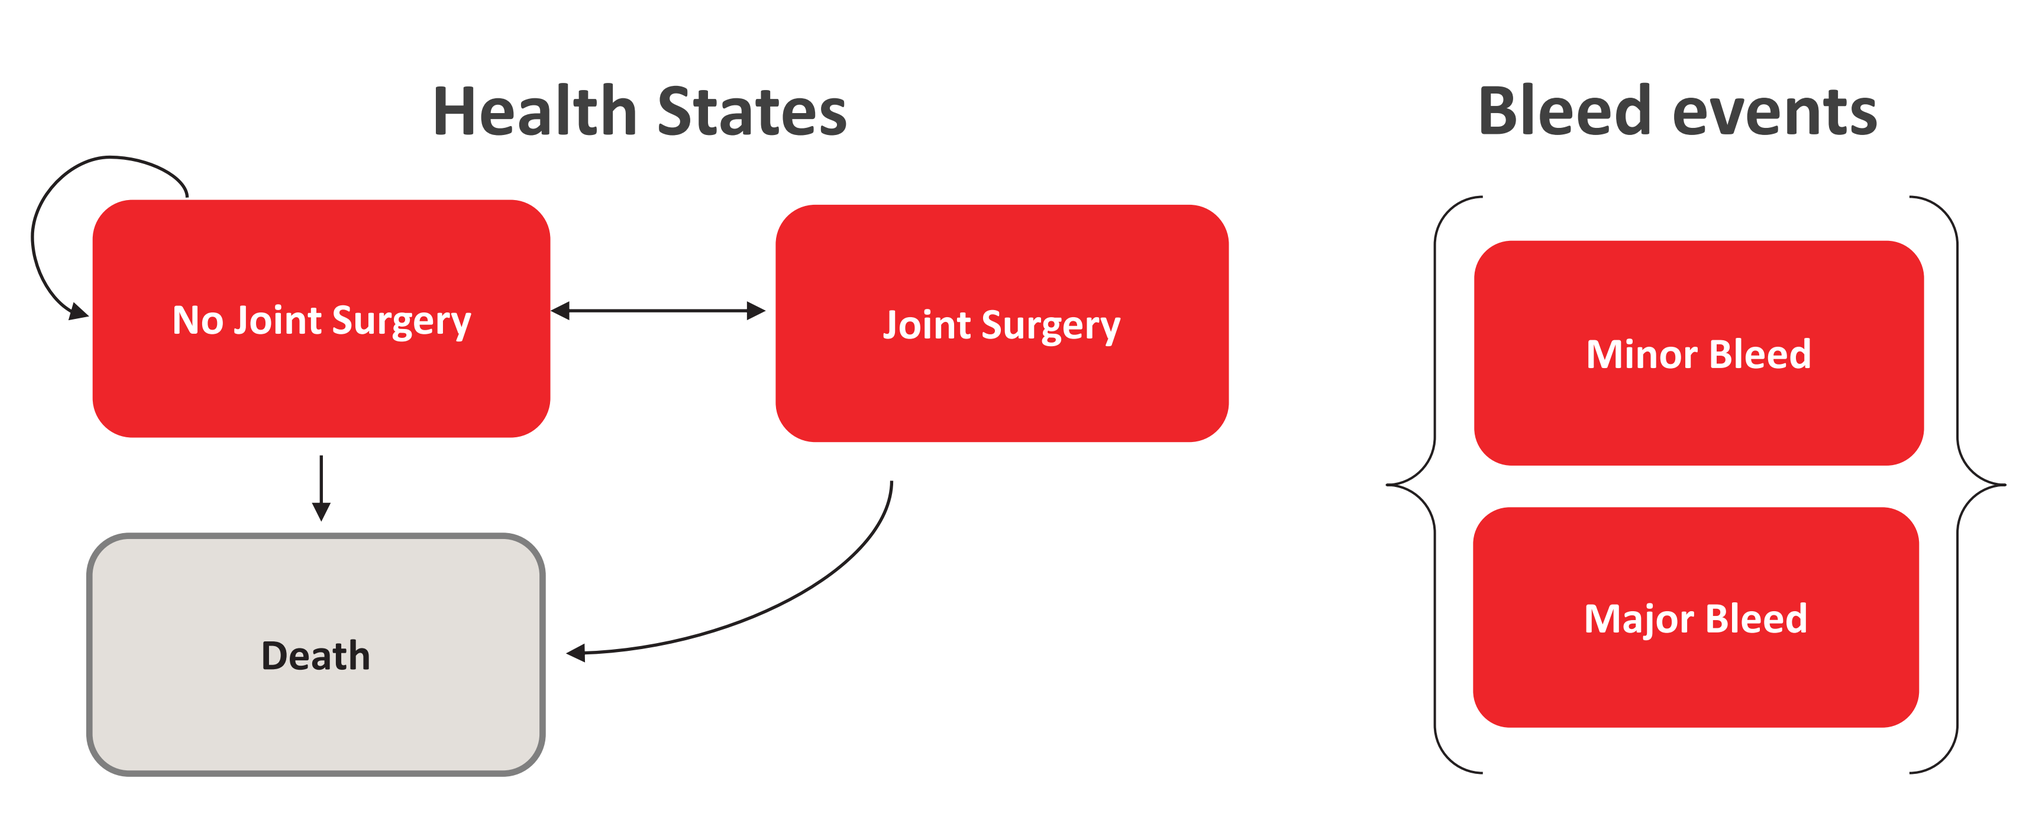
Health States
Bleed events
No Joint Surgery
Joint Surgery
Death
Minor Bleed
Major Bleed

- The characteristics of the vWD population are summarized in Table 1:

Table 1. Population characteristics

Average patient age (3)	43 years old
Percentage type 3 vWD (3)	5%
Percentage females (4)	51.01%
Pettersson score at baseline†	14.00

† Due to lack of data in patients with vWD, it was assumed the same as patients with hemophilia (5)

Efficacy and transition probabilities

- Patients experienced bleed events according to literature probabilities and treatment regimes (Table 2).

Table 2. Bleed events rates and probabilities

Without prophylaxis	
Annual number of bleeds (6)	26.50
Probability of minor bleed (7)	61.84%
Probability of major bleed (7)	38.16%
Intermittent prophylaxis	
RR bleed (vs. no prophylaxis): Haemate-P®, Fanhdi®, Wilate® (7)	0.77, 0.81, 0.86
Probability of minor bleed (7)	70.00 %
Probability of major bleed (7)	30.00 %
Long-term prophylaxis	
RR bleed (vs. no prophylaxis): Haemate-P®, Fanhdi®, Wilate® (7)	0.09, 0.24, 0.42
Probability of minor bleed (7)	70.00 %
Probability of major bleed (7)	30.00 %

RR: Relative risk

- Each bleeding event carried a probability of being a joint bleed. Also, each joint bleed increased the probability of raising the Pettersson score. The patient required surgery at a specific Pettersson score level (Table 3).

Table 3. Joint bleed events and surgery probabilities

Percentage of bleeds that are joint bleeds (9)	24.00 %
Joint bleeds required to increase Petterson score 1 point (10)	12.60
Petterson score at which surgery occurs (5)	28.00
Limit age (years) for arthropathy surgery (8)	80

Adverse events

- Only thromboembolic events (Table 2) were considered as adverse events, as the rest of AE were assumed to be the same for all therapies.

Costs and resources use

- Each state was associated with a cost (Table 4), including disease management costs (outpatient visits, hospital stays, bleeding treatment and surgeries) and drug acquisition costs (€, 2023).
- Unit costs were obtained from local sources (11,12).

Table 4. Costs & resource use

Resource use unit costs	
Outpatient visits (11)	€ 200.87
Inpatient stay for bleed (12)	€ 1,203.95
Minor bleed†	€ 50.22
Major bleed†	€ 702.41
Joint surgery (11)	€ 3,061.59
Death†	€ 0.00
Drug costs	
Haemate-P® (13)	0.16 €/IU
Fahndi® (13)	0.33 €/IU
Wilate® (13)	0.39 €/IU
On-demand treatment for bleeds	
Haemate-P®: Major bleed‡	Dose of 66.67 IU/kg, 2 doses per day, 4 days
Fahndi®: Major bleed§	Dose of 45 IU/kg, 2 doses per day, 4 days
Wilate®: Major bleed§	Dose of 45 IU/kg, 2 doses per day, 4 days
Haemate-P®: Minor bleed‡	Dose of 30 IU/kg, 2 doses per day, 1 days
Fahndi®: Minor bleed§	Dose of 30 IU/kg, 2 doses per day, 1 days
Wilate®: Minor bleed§	Dose of 30 IU/kg, 2 doses per day, 1 days
Long-term and intermittent prophylaxis*	
Haemate-P®‡	Dose of 30 IU/kg, 2.5 doses per week
Fanhdi®§	Dose of 30 IU/kg, 2.5 doses per week
Wilate®§	Dose of 30 IU/kg, 2.5 doses per week

IU: International units. * Intermittent prophylaxis was received only 25% of weeks (1 every 4 weeks). † Assumption. ‡ SmPC guidance for Voncento due to the lack of data for long-term and intermittent prophylaxis for Haemate-P, § SmPC guidance for Wilate.

Utility values

- Each health state was associated with a utility value from which disutility were subtracted when an AE occurred.

Table 5. Utility and disutility values

Utility value	
0 – 30 years old PS = 0 / PS = 1-27 (5)	0.940 / 0.820
31 – 40 years old PS = 0 / PS = 1-27 (5)	0.840 / 0.740
41 – 50 years old PS = 0 / PS = 1-27 (5)	0.860 / 0.690
51 – 60 years old PS = 0 / PS = 1-27 (5)	0.830 / 0.540
Disutility value	
Minor bleed (14)	-0.000
Major bleed (14)	-0.003

PS: Pettersson score

Analysis

- The incremental cost-utility ratio (ICUR) per quality-adjusted life year (QALY) gained was estimated.
- One-way (OWSA) and probabilistic sensitivity analysis (PSA) were performed by varying the parameters.

Results

Cost-utility analysis

Long-term prophylaxis

- In long-term prophylaxis Haemate-P® was dominant vs Fanhdi® and Wilate® generating less costs with a QALYs gain.

Intermittent prophylaxis

- In intermittent prophylaxis, Haemate-P® was dominant vs Fanhdi® and Wilate® generating fewer costs with a QALYs gain.

On-demand

- Finally, on demand, Haemate-P® had the same QALYs as Fahndi® and Wilate® but was less costly.

Table 6. Cost-utiliy analysis

	Long-term prophylaxis	*Intermittent prophylaxis*	*On demand*
Haemate-P® vs. Fanhdi®			
Δ Costs	-1,313,845€	-1,001,510€	-696,857€
Δ QALYs	+0.13	+0.03	0,00
ICUR	**Dominant**	**Dominant**	**Less costly**
Haemate-P® vs. Wilate®			
Δ Costs	-2,233,940€	-1,520,998€	-1,145,780€
Δ QALYs	+0.29	+0.07	0,00
ICUR	**Dominant**	**Dominant**	**Less costly**

Sensitivity analysis

- The OWSA found no parameter whose change modifies the conclusions obtained in the deterministic analysis.
- The PSA showed that Haemate-P dominates Fanhdi and Wilate in most cases.

Table 7. Scenario distribution in probabilistic sensitivity analysis

	Long-term prophylaxis	*Intermittent prophylaxis*	*On demand**
Haemate-P® vs. Fanhdi®			
More cost, more QALYs	**5.32%**	0.00%	0.00%
More cost, less QALYs	0.00%	0.12%	
Less cost, more QALYs	**94.68%**	**64.22%**	**100%**
Less cost, less QALYs	0.00%	35.60%	
Haemate-P® vs. Wilate®			
More cost, more QALYs	0.00%	0.00%	0.00%
More cost, less QALYs	0.00%	0.00%	
Less cost, more QALYs	**99,98%**	**75.48%**	**100%**
Less cost, less QALYs	0.02%	24.52%	

* Patients in *on demand* regime obtain the same QALYs no matter the treatment used.

Conclusions

Haemate-P® presents **better ICUR results than any other treatment strategy**, especially when used as a **long-term prophylactic treatment** but also under intermittent prophylaxis and on demand treatment.

References

1. Sharma, R. et al. Hematology 2014, the American Society of Hematology Education Program Book (2017): 379-384. 2. Bowman, M., et al. Journal of Thrombosis and Haemostasis 8.1 (2010): 213-216. 3. CSL-B data on file. February (2021). 4. Instituto Nacional de Estadística (2023). 5. ICER. Valoctocogene roxaparvovec and emicizumab for hemophilia A: Effectiveness and value Final Report. (2020). 6. Lissitchkov, Toshko J., et al. Blood Coagulation & Fibrinolysis 28.2 (2017): 152. 7. Lethagen, S. Thrombosis Research. (2006) 118, S9-S11. 8. Earnshaw, S. R., et al. Haemophilia. (2015): 310-319. 9. Holm, E. et al. Blood coagulation & fibrinolysis. (2015) 26(4), 383-388. 10. Fischer, Kathelijn, et al. Haematologica (2011): 738. 11. Gisbert R, et al. Base de datos de costes sanitarios y ratios coste efectividad españoles: eSalud. Barcelona: Oblikue Consulting, S. L. 12. Ministerio de Sanidad. Conjunto Mínimo Básico de Datos de las Altas Hospitalarias. (2021). 13. Consejo General de Colegios Oficiales de Farmacéuticos. Botplusweb. 14. Nolous NS, et al. Blood, The Journal of the American Society of Hematology. (2021): 1677-1690

Acknowledgements

Medical writing support was provided by Outcomes'10. All authors reviewed the results and approved the final version of the poster.

Disclosures

SL, PMV, RT, MP, LVB, SY and MW: are employees of CSL Behring.

Funding

This study was funded by CSL Behring.

Presented at the ISPOR Europe 2023 Congress, Copenhagen, Denmark, 11–12/15-2023.

CSL Behring