EE262

Indirect Cost Burden for Individuals Living With Sickle Cell Disease With Recurrent Vaso-Occlusive Crises in Europe

LAUREN LILLY,[1] JENNIFER DRAHOS,[1] ADRIANA BOATENG-KUFFOUR,[1] MELANIE CALVERT,[2] ASHLEY VALENTINE,[3] ANTHONY MASON,[4] NANXIN LI,[1] ZAHRA PAKBAZ,[5] FARRUKH SHAH,[6] NICK AINSWORTH,[7] CHUKA UDEZE,[1] ANTONY P. MARTIN[7]

[1]Vertex Pharmaceuticals Incorporated, Boston, MA, USA | [2]Centre for Patient-Reported Outcomes Research, University of Birmingham, Birmingham, UK | [3]Sick Cells, Washington, DC, USA | [4]Sickle Cell Care Manchester, Manchester, UK | [5]University of California Irvine School of Medicine, Division of Hematology Oncology, Orange, CA, USA | [6]NHS Blood and Transplant, London, UK | [7]QC Medica, Liverpool, UK

INTRODUCTION

- Sickle cell disease (SCD) is characterized by vaso-occlusive crises (VOCs), wide-spread organ damage, and reduced life expectancy[1]
- It is well known that individuals with SCD with recurrent VOCs have increased disease severity leading to higher healthcare resource utilization (HCRU) and direct healthcare costs[2,3]
- In the United States, individuals with SCD with recurrent VOCs also have substantial indirect impacts, given higher out-of-pocket (OOP) costs, activities related to disease management, and negative impacts on their ability to work[4,5]
- To date, little is known about the indirect costs incurred by individuals with SCD with recurrent VOCs in Europe

OBJECTIVE

- This analysis used data collected from a quantitative survey of individuals with SCD with recurrent VOCs to quantify the indirect economic impacts of living with SCD in Europe

METHODS

Survey Study Design and Inclusion Criteria

- The overall mixed-methods study consisted of qualitative interviews and a longitudinal online quantitative survey administered at 3 timepoints (Month 0 [baseline, M0], Month 3 [M3], and Month 6 [M6]) in the United States, France, Germany, Italy, and the United Kingdom
- The indirect economic impact results in this study were from the quantitative M3 survey taken by participants living in Europe (France, Germany, Italy, and the United Kingdom)
- Key inclusion criteria were:
 - Aged ⩾18 years
 - Self-reported SCD diagnosis
 - At least 2 severe VOCs/year for 2 consecutive years; a severe VOC was defined as a VOC that requires an interaction with a medical facility and pain medication or a red blood cell transfusion

Survey Design and Administration

- Survey questions were designed based on information gathered from interviews with individuals living with SCD with recurrent VOCs in the United States and United Kingdom and a scientific advisory team for the study consisting of clinicians, patient advocates, and outcomes researchers
- The baseline survey at M0 included items assessing demographic, clinical, and socioeconomic characteristics:
 - Demographics included age, gender, race, and marital status
 - Clinical characteristics included number of severe VOCs and HCRU in the past 12 months
 - Socioeconomic characteristics included geographic region of residence, educational attainment, employment status, annual household income, and primary health insurance
- The survey administered at M3 included questions to assess the time required to manage SCD and indirect medical costs related to the management of SCD
- At M3, the Work Productivity and Activity Impairment (WPAI) questionnaire was included to quantify employment and impairments in paid work

Measures and Calculations Used to Quantify Monthly and Annual Indirect Costs

- Annual OOP costs were calculated by totaling the estimated monthly cost of individual SCD-related expenses and then multiplying the total by 12 months
 - Country-specific currencies were converted to United States dollars (USD) using the following purchasing power parity ratios, as reported by the Organisation for Economic Co-operation and Development (OECD) year-to-date data 2022: France = 0.725, Germany = 0.741, Italy = 0.654, and the United Kingdom = 0.693[6]
- The calculation for annual health-related activity costs was guided by the Human Capital Approach wherein the estimated monthly time spent performing individual activities was totaled, multiplied by 12 months, and then multiplied by the mean hourly wage (24 USD in France, Germany, Italy, and the United Kingdom, as reported by the OECD, 2021)[6]
- Costs associated with absenteeism (days missed from work), presenteeism (reduced productivity while at work), and work productivity loss (overall work impairment) were calculated by multiplying composite scores measured by the WPAI by the mean annual salary across the 4 countries (49,025 USD, as reported by the OECD, 2021)[6]

Statistical Analysis

- Descriptive statistics were conducted for participant characteristics, OOP costs, health-related activities, and WPAI composite scores for absenteeism, presenteeism, and work productivity loss
 - Frequency (n) and proportion (%) were reported for categorical variables, and mean and standard deviation (SD) were reported for continuous variables
- Given the small sample size of individuals in Europe who responded to the WPAI, results from US participants were also included for additional context

RESULTS

Demographic and Clinical Characteristics

- Forty-five adults with SCD with recurrent VOCs living in Europe participated in the survey (**Table 1**)
- Most participants were female (n = 40 [88.9%]) and Black (n = 25 [83.3%]) (**Table 1**)
- Participants experienced a mean of 4.7 (SD: 3.3) severe VOCs, 3.5 (SD: 4.5) hospitalizations, and 4.2 (SD: 5.5) ER visits in the year prior to study entry (**Table 1**)

Table 1. Demographic and Clinical Characteristics

	Adults With SCD With Recurrent VOCs (N = 45)
Age, years, mean	38.3 (11.3)
Gender, n (%)	
Female	40 (88.9)
Race,[a] n (%)	
Black	25 (83.3)
Mixed race	4 (13.3)
Other	1 (3.3)
Marital status, n (%)	
Single	23 (51.1)
Married/domestic partner	11 (24.4)
In a committed relationship	6 (13.3)
Separated/divorced	4 (8.9)
Prefer not to say	1 (2.2)
Severe VOCs[b] in the past 12 months, mean (SD)	4.7 (3.3)
HCRU in the past 12 months	
Hospitalizations with overnight stay, mean (SD)	3.5 (4.5)
Days spent in hospital, median (IQR)	5.5 (1.7–18.3)
A&E visits, mean (SD)	4.2 (5.5)

A&E, accident and emergency; HCRU, healthcare resource utilization; IQR, interquartile range; SCD, sickle cell disease; SD, standard deviation; VOC, vaso-occlusive crisis.

[a]n = 30; [b]A severe VOC was defined as one requiring an interaction with a medical facility and pain medication or an RBCT.

Socioeconomic Characteristics

- Over one-third (36.4%) of participants had a bachelor's degree; 18.2% had a master's degree (**Table 2**)
- Forty-four percent of participants were unemployed and 15.6% worked full-time (**Table 2**)
- Of the 32 participants who reported an annual household income, 90.6% earned 50,000 USD or less, with 40.6% earning less than 10,000 USD (**Table 2**)

Table 2. Socioeconomic Characteristics

Characteristic, n (%)	Adults With SCD With Recurrent VOCs (N = 45)
Geographic region of residence	
Large city	19 (42.2)
City or town[a]	14 (31.1)
Suburb near a large city	6 (13.3)
Small city or town[a,b]	6 (13.3)
Highest level of education achieved[c]	
High school diploma	4 (9.1)
Trade or technical school	7 (15.9)
Bachelor's degree	16 (36.4)
Master's degree	8 (18.2)
Other[d]	9 (20.5)
Employment status[e]	
Unemployed	20 (44.4)
Unable to work	7 (15.6)
Full-time	7 (15.6)
Part-time	4 (8.9)
Other[f]	5 (11.1)
Annual household income (USD)[g,h]	
<10,000	13 (40.6)
10,000 to 25,000	9 (28.1)
25,000 to 50,000	7 (21.9)
50,000 to 75,000	1 (3.1)
75,000 to 100,000	1 (3.1)
100,000 to 200,000	1 (3.1)
Primary health insurance	
NHS/universal/public healthcare	41 (91.1)
No insurance/do not know	4 (8.9)

NHS, National Health Service; SCD, sickle cell disease; SD, standard deviation; VOC, vaso-occlusive crisis; USD, United States dollars.
[a]Not a suburb of a large city; [b]<25K population; [c]n = 44; [d]Other, some college credit (but no degree), doctorate, or prefer not to say; [e]n = 43; [f]Retired, on leave, or unpaid volunteer; [g]Before taxes from all sources; [h]n = 32.

OOP Costs

- Mean costs associated with medical appointments (e.g., transportation, childcare) (122 USD [SD: 412]) and prescription medications (72 USD [SD: 258]) were the highest reported monthly OOP expenses among participants (**Table 3**)
- The mean OOP cost per month for all items totaled 461 USD (SD: 753) (**Table 3**)
- Participants reported spending a mean of 5,527 USD on OOP costs annually (**Table 3**)

Table 3. OOP Costs

Category, Mean (SD)	Adults With SCD With Recurrent VOCs (N = 45)
Monthly costs, USD	
Transportation, parking, and other costs associated with attending medical appointments and hospitalizations (e.g., childcare)	122 (412)
Prescription medications	72 (258)
Vitamins or nutritional supplements	53 (72)
Paid caregivers or support services (e.g., personal care, housework)	52 (75)
Medical appointments related to SCD	51 (73)
Holistic care (e.g., massage, acupuncture)	39 (55)
Physical therapy and rehabilitation	30 (68)
Mental health services	26 (68)
Other	16 (46)
Total costs, USD	**461 (753)**
Annual costs, USD	**5,527**

OOP, out-of-pocket; SCD, sickle cell disease; SD, standard deviation; USD, United States dollars; VOC, vaso-occlusive crisis.

Health-Related Activities

- The health-related activities in which participants spent the most time engaging every month (mean [SD]) were attending medical appointments (6.1 hours [5.7]) and traveling to medical appointments (3.6 hours [3.6]) (**Table 4**)
- Participants reported spending an average of 24.7 hours (SD: 23.8) engaging in all health-related activities per month, which translates to 592 USD in unpaid labor managing the disease (**Table 4**)
- Participants reported spending a mean of 296.4 hours engaging in health-related activities annually, which translates to 7,107 USD in unpaid labor managing the disease (**Table 4**)

WPAI

- Thirteen (28.9%) study participants were employed (working for pay) and thus included in the analysis of the WPAI (**Table 5**)
- Employed participants reported substantial absenteeism (28.8%) and presenteeism (40.8%) (**Table 5**)
- Mean lost annual wages due to absenteeism and presenteeism were 14,119 USD and 20,002 USD, respectively (**Table 5**)

WPAI (Continued)

Table 4. Health-Related Activities

Outcome	Adults With SCD With Recurrent VOCs (N = 45)	
	Time, Hours, Mean (SD)	Cost, USD
	Monthly time and costs	
Time at medical appointments	6.1 (5.7)	147
Traveling to medical appointments	3.6 (3.6)	86
Arranging time off from work and school to receive treatment	3.3 (5.6)	78
Preparing and administering treatments at home	3.2 (5.8)	77
Scheduling medical appointments	2.5 (3.3)	60
Organizing payments from health insurance	2.4 (4.6)	58
Arranging refills and picking up medications	2.0 (2.7)	49
Arranging childcare due to healthcare needs	1.6 (3.5)	38
Total	**24.7 (23.8)**	**592**
	Annual time and costs	
Total	**296.4**	**7,107**

SCD, sickle cell disease; SD, standard deviation; USD, United States dollars; VOC, vaso-occlusive crisis.

Table 5. WPAI, Composite Scores

Category	Employed Adults With SCD With Recurrent VOCs in Europe, Mean (SD), % (n = 13)	Corresponding Lost Annual Wages in Europe, USD
Absenteeism	28.8 (34.5)	**14,119**
Presenteeism	40.8 (26.8)	**20,002**
Work productivity loss	57.3 (31.6)	**28,091**

SCD, sickle cell disease; SD, standard deviation; USD, United States dollars; VOC, vaso-occlusive crisis; WPAI, Work Productivity and Activity Impairment.
[a]Activity impairment for employed adults with SCD with recurrent VOCs living in Europe (n = 13) was 58.9% (SD: 27.6). Activity impairment was not costed.

- Scores among European and US participants for absenteeism (28.8% and 32.9%, respectively) and presenteeism (40.8% and 46.2%, respectively) were similar

LIMITATIONS

- The calculation for mean annual OOP and health-related activities costs assumes these costs are consistent from month-to-month for a duration of 1 year
- Indirect impacts for individuals with SCD who were unemployed were not captured using the WPAI; potential additional indirect impacts of lack of employment were not fully captured in this analysis
- Participants were self-selected, which may impact the generalizability of the results
- The majority of study participants were female (88.9%); therefore, care should be taken when applying these conclusions to the population of individuals with SCD as a whole
- All data were self-reported; data eligibility and accuracy were not directly verified with medical records
- Individuals who did not have Internet and email access required to complete the survey were excluded

CONCLUSIONS

- Individuals with SCD with recurrent VOCs incur substantial indirect costs due to OOP costs, health-related activities, and work productivity
- Only a small proportion of participants were employed, and these participants reported substantial absenteeism and presenteeism due to SCD
- The substantial indirect costs for individuals living with SCD, as well as society, highlight the unmet needs of this underserved patient population
- Further analyses of indirect costs among caregivers of individuals living with SCD are needed to understand the total burden of disease-related indirect impacts

REFERENCES

1. Piel FB, et al. *N Engl J Med*. 2017;376(16):1561-1573.
2. Darbari DS, et al. *Eur J Haematol*. 2020;105(3):237-246.
3. Udeze C, et al. *Adv Ther*. 2023;40(8):3543-3558.
4. Holdford D, et al. *Value Health*. 2021;24(8):1095-1101.
5. Bradt P, et al. Crizanlizumab, Voxelotor, and L-Glutamine for Sickle Cell Disease: Effectiveness and Value. Evidence Report. Institute for Clinical and Economic Review. Posted March 12, 2020. Updated February 9, 2021. https://icer.org/wp-content/uploads/2021/02/ICER_SCD_Evidence-Report_031220-FOR-PUBLICATION.pdf. Accessed October 2023.
6. Organisation for Economic Co-operation and Development (OECD). Purchasing Power Parities (PPP) Statistics: Monthly comparative price levels. https://stats.oecd.org/Index.aspx?DataSetCode=CPL. Accessed May 2023.

ACKNOWLEDGMENTS

We would like to thank our collaborators, scientific advisory team, patient advocacy group partners, and especially the interviewees and survey participants living with SCD with recurrent VOCs who kindly provided their time and experiences – and without whom this study would not be possible. The study was supported by Vertex Pharmaceuticals Incorporated. Editorial coordination and support were provided by Nathan Blow, PhD, under the guidance of the authors, who may own stock or stock options in the company. Editorial support was provided by Emma-Ruoqi Xu, PhD, and Nicholas Strange of Complete HealthVizion, IPG Health Medical Communications, funded by Vertex Pharmaceuticals Incorporated.

AUTHOR DISCLOSURES

LL, JD, AKB, NL, and CU are employees of Vertex Pharmaceuticals Incorporated and may hold stock or stock options in the company. MC is Director of Birmingham Health Partners Centre for Regulatory Science and Innovation and Centre for Patient Reported Outcomes Research and a National Institute for Health and Care Research (NIHR) senior investigator; has received funding from Anthony Nolan, European Regional Development Fund-Demand Hub and Health Data Research UK, Gilead, GlaxoSmithKline, Janssen, Macmillan Cancer Support, Merck, NIHR, NIHR ARC WM, NIHR Birmingham BRC, NIHR BTRU Precision and Cellular Therapeutics, UCB Pharma, UKRI, and UK SPINE; and has received consultancy fees from Aparito, Astellas, CIS Oncology, Daiichi Sankyo, Gilead, Glaukos, GlaxoSmithKline, Halfloop, Merck, Patient-Centered Outcomes Research Institute, Pfizer, Takeda, and Vertex Pharmaceuticals Incorporated. AV, AM, and NA have nothing to disclose. ZP has received research grants from Amgen, Forma Therapeutics, Global Blood Therapeutics, Novartis, NovoNordisk, and Pfizer; received consulting fees from Agio, Amgen, Dova, Global Blood Therapeutics, Guide point, Novartis, Sanofi, Sobi, and Vertex Pharmaceuticals Incorporated; received honoraria from Cayeene Wellness Center and Child Foundation, Dova, and Global Blood Therapeutics Inc; served as an advisory board or committee member for Alexion, Sanofi, and Sobi; and acted as a CME course director for the Cayenne Wellness Center and Child Foundation and planning committee member for their annual education symposium. FS has received research grants from IQVIA, Novartis Pharma AG, and Vertex Pharmaceuticals Incorporated; received honoraria from Biologix FZ co, Bristol Myers Squibb, Chiesi Ltd, and Novartis Pharma AG; served as an advisory board or committee member for Agios, bluebird bio, Bristol Myers Squibb, Silence Therapeutics Plc, and Vertex Pharmaceuticals Incorporated; and acted as Chair for the UK Forum on Haemoglobin Disorders. APM is a partner of QC Medica who was funded by Vertex Pharmaceuticals Incorporated to perform this research.

Presented at ISPOR Europe 2023, November 12–15, 2023, Copenhagen, Denmark