PCR149

Determination of Ranges of HiSQOL Scores Defining Clinically Meaningful Within-Patient Improvement Thresholds and Severity Levels

Joslyn S. Kirby,[1] John R. Ingram,[2] Jérémy Lambert,[3] Robert Rolleri,[4] Edward Muller,[5] Ingrid Pansar,[6] Christopher G. Pelligra[7]

Objective

To determine clinically meaningful within-patient change thresholds for improvement (often used in the responder definition) and severity thresholds, to categorise patients across disease activity/impact bands, for Hidradenitis Suppurativa (HS) Quality of Life (HiSQOL) questionnaire total and subscale scores.

Background

- HS is a chronic inflammatory skin disease, characterised by painful and recurrent skin lesions.[1]
- The 17-item HiSQOL questionnaire provides a valid, reliable assessment of HS patients' health-related quality of life, an area where instruments are lacking.[2]
- Here, we determined the clinically meaningful within-patient improvement and severity thresholds to guide interpretation of HiSQOL total and subscale scores for patients with moderate to severe HS.

Methods

- Pooled, blinded data from two randomised phase 3 trials, BE HEARD I & II, of bimekizumab 320 mg every 2/4 weeks or placebo were used to estimate HiSQOL score thresholds.[3,4]
- The 17 HiSQOL item scores range from 0 (not at all) to 4 (extremely) and are summed to generate a total score (range 0–68). Three subscale scores were also evaluated:
 - Symptoms (0–16);
 - Psychosocial (0–20);
 - Activities-adaptations (0–32).
- Higher scores correspond to higher symptomology or impact on health-related quality of life.
- Threshold analyses were conducted on observed scores for all randomised patients with ≥1 non-missing HiSQOL subscale score at any scheduled assessment visit.
- Thresholds for clinically meaningful within-patient improvement for Week 16 were determined and assessed by triangulating threshold estimates from the following analyses:
 - Anchor-based analyses, using Patient Global Impression of Severity of HS (PGI-S-HS), to divide patients into response groups and describe HiSQOL score changes from baseline as a basis for the thresholds;
 - Empirical cumulative distribution function (eCDF) plot of changes in HiSQOL scores from baseline to Week 16 to support selection of the final thresholds;
 - Distribution-based analyses (one standard error of measurement and half of the baseline standard deviation [SD]) to support the relevance of the thresholds.
- Disease activity thresholds were determined using the maximum Youden index values from the receiver operating characteristic (ROC) analyses with PGI-S-HS as the anchor.

Results

- The HiSQOL analysis set included 1,010 patients with HS from BE HEARD I & II.
- The mean age and duration of disease for included patients were 36.7 years and 8.0 years, respectively (**Table 1**). Most patients were female (56.6%) and 44.4% had Hurley Stage III disease at baseline.
- For HiSQOL total score, a 20- to 21-point decrease was identified as a clinically meaningful within-patient improvement.
- For HiSQOL subscales, the following clinically meaningful within-patient improvement thresholds were identified:
 - Symptoms: 5- to 6-point decrease;
 - Psychosocial: 4- to 5-point decrease;
 - Activities-adaptations: 10- to 11-point decrease.
- Findings from the eCDF plot supported the use of the aforementioned improvement threshold estimates; a plot of changes in HiSQOL total score is shown in **Figure 1**.
- The ROC curves used to determine disease severity thresholds for the HiSQOL total score, using PGI-S-HS response categories, are shown in **Figure 2**.
- Disease severity thresholds of none, mild, moderate, severe and very severe were identified for the HiSQOL total score and subscale scores (**Figure 3**).

Conclusions

This analysis defined clinically meaningful within-patient improvement and severity thresholds for HiSQOL total and subscale scores. These thresholds can be used to guide interpretation of scores and assess treatment effects on disease burden in patients with HS.

Summary

This analysis defined **clinically meaningful within-patient improvement** and **severity thresholds** for HiSQOL total and subscale scores

The thresholds were identified by evaluating HiSQOL scores and changes in scores against the severity levels from established patient-reported disease severity anchor measures

Clinically meaningful within-patient improvement thresholds:

- **Total** score: 20- to 21-point decrease
- **Symptoms** subscale: 5- to 6-point decrease
- **Psychosocial** subscale: 4- to 5-point decrease
- **Activities-adaptations** subscale: 10- to 11-point decrease

Disease severity thresholds:

- Thresholds of **none, mild, moderate, severe** and **very severe** were identified

These thresholds can be used to **guide interpretation of scores** and **assess treatment effects** on disease burden in patients with HS

Table 1 Baseline characteristics

	HiSQOL Analysis Set (N=1,010)
Age, years, mean (SD)	36.7 (12.2)
Female, n (%)	572 (56.6)
Race, n (%)	
White	771 (76.3)
Black or African American	106 (10.5)
Asian	41 (4.1)
Other or Mixed	42 (4.2)
Region, n (%)	
North America	385 (38.1)
Western Europe	290 (28.7)
Central and Eastern Europe	260 (25.7)
Asia and Australia	75 (7.4)
BMI, kg/m², mean (SD)	33.1 (8.1)
Duration of disease, years, mean (SD)	8.0 (7.8)
AN count, mean (SD)	16.2 (16.1)
DT count, mean (SD)	3.6 (4.3)
Hurley Stage, n (%)[a]	
II	562 (55.6)
III	448 (44.4)
HiSQOL baseline scores, mean (SD)	
Total score	25.2 (13.4)
Symptoms score	7.9 (3.5)
Psychosocial score	5.3 (4.4)
Activities-adaptations score	12.0 (7.2)

Data presented for HiSQOL analysis set (all randomised patients with ≥1 non-missing HiSQOL subscale score at any scheduled assessment visit). Percentages may not sum to 100 due to rounding. [a]Only patients with Hurley Stage II and III were included at baseline, as per the BE HEARD I & II eligibility criteria.

Figure 1 eCDF plot of changes from baseline to Week 16 in HiSQOL total score by PGI-S-HS response category

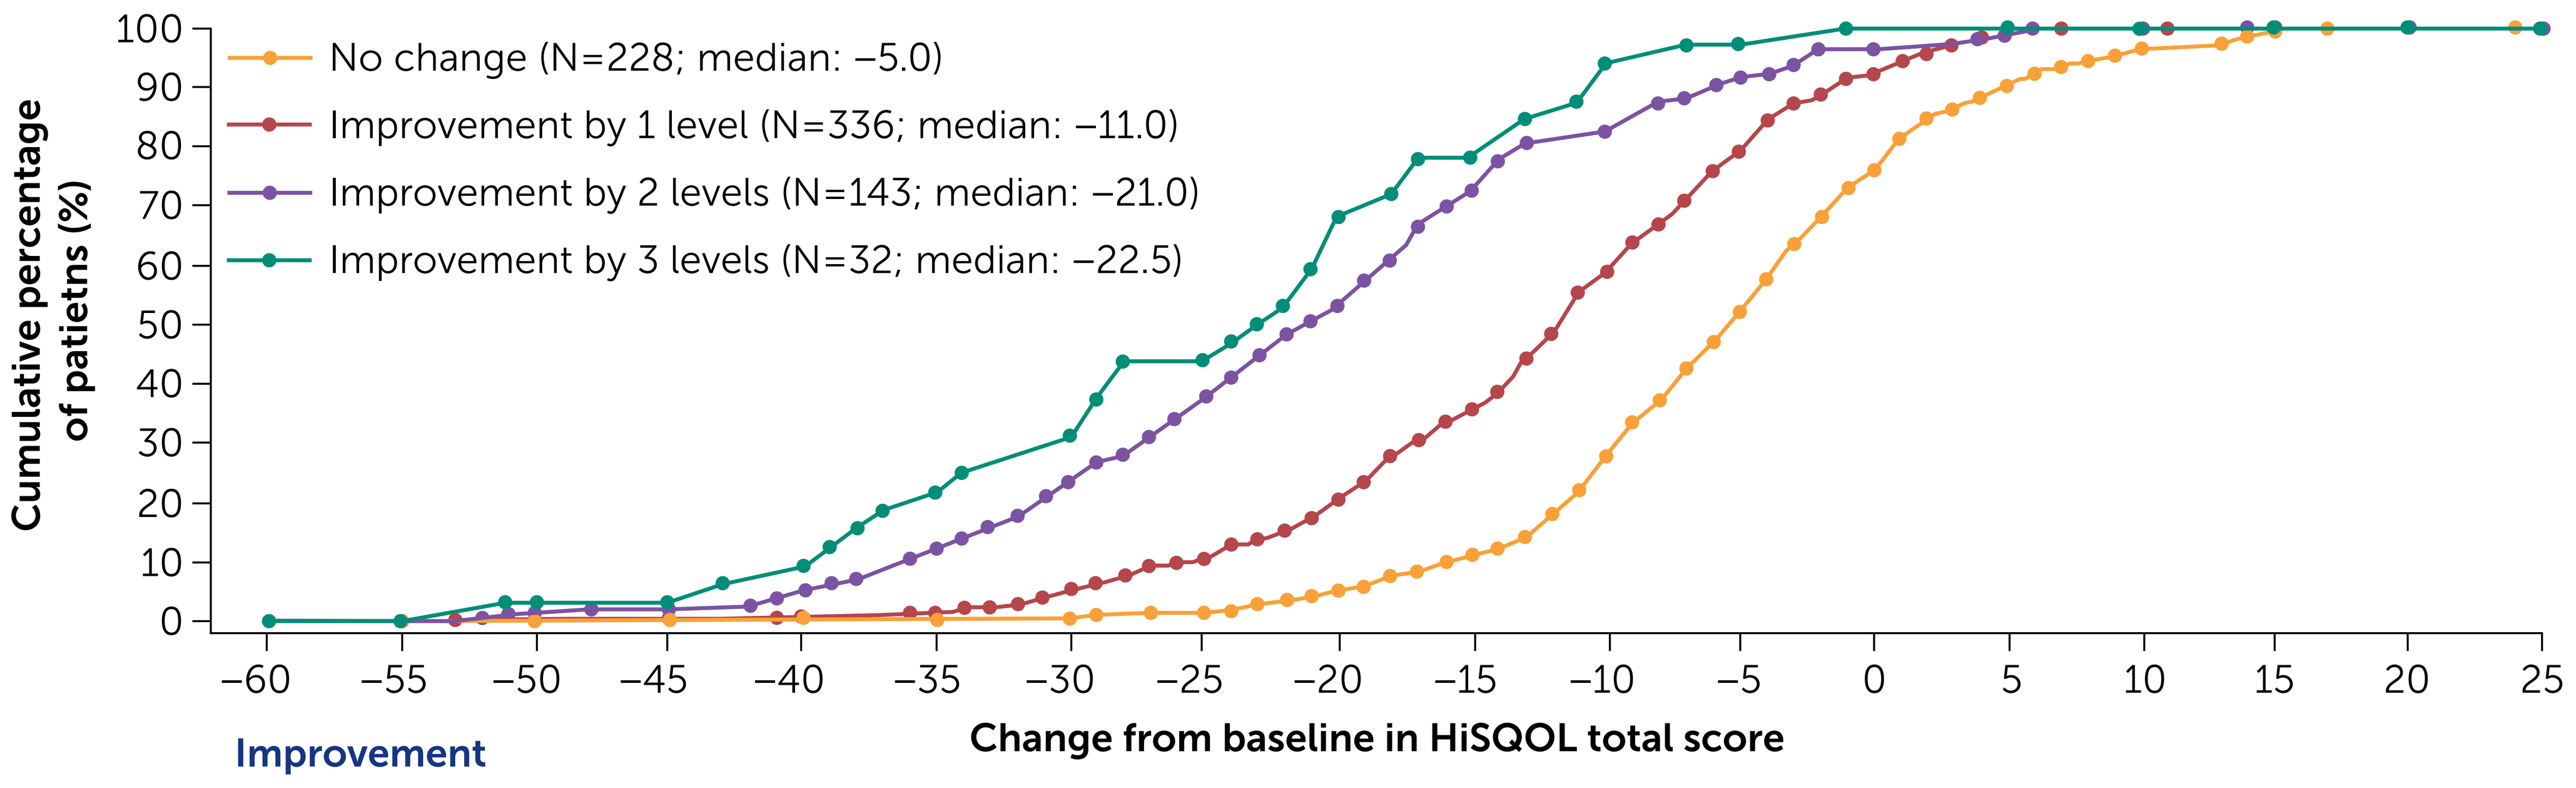

No change (N=228; median: −5.0)
Improvement by 1 level (N=336; median: −11.0)
Improvement by 2 levels (N=143; median: −21.0)
Improvement by 3 levels (N=32; median: −22.5)
Cumulative percentage of patietns (%)
Change from baseline in HiSQOL total score
Improvement

	−60	−55	−50	−45	−40	−35	−30	−25	−20	−15	−10	−5	0	5	10	15	20	25
No change (%)	0.0	0.0	0.0	0.0	0.0	0.3	0.3	1.4	5.2	11.1	27.8	52.1	75.7	90.3	96.5	99.3	99.7	100
Improvement by 1 level (%)	0.0	0.0	0.3	0.3	0.6	1.2	5.1	10.4	20.5	35.4	58.9	78.9	92.3	99.4	99.7	100	100	100
Improvement by 2 levels (%)	0.0	0.0	1.4	2.1	5.6	11.9	23.1	37.8	53.1	72.7	82.5	91.6	96.5	98.6	99.3	100	100	100
Improvement by 3 levels (%)	0.0	0.0	3.1	3.1	9.4	21.9	31.3	43.8	68.8	78.1	93.8	96.9	100	100	100	100	100	100

HiSQOL analysis set (all randomised patients with ≥1 non-missing HiSQOL subscale score at any scheduled assessment visit). Patients were divided into different response groups based on the PGI-S-HS response from baseline to Week 16.

Figure 2 ROC curves for determination of disease severity thresholds for HiSQOL total score using PGI-S-HS response categories

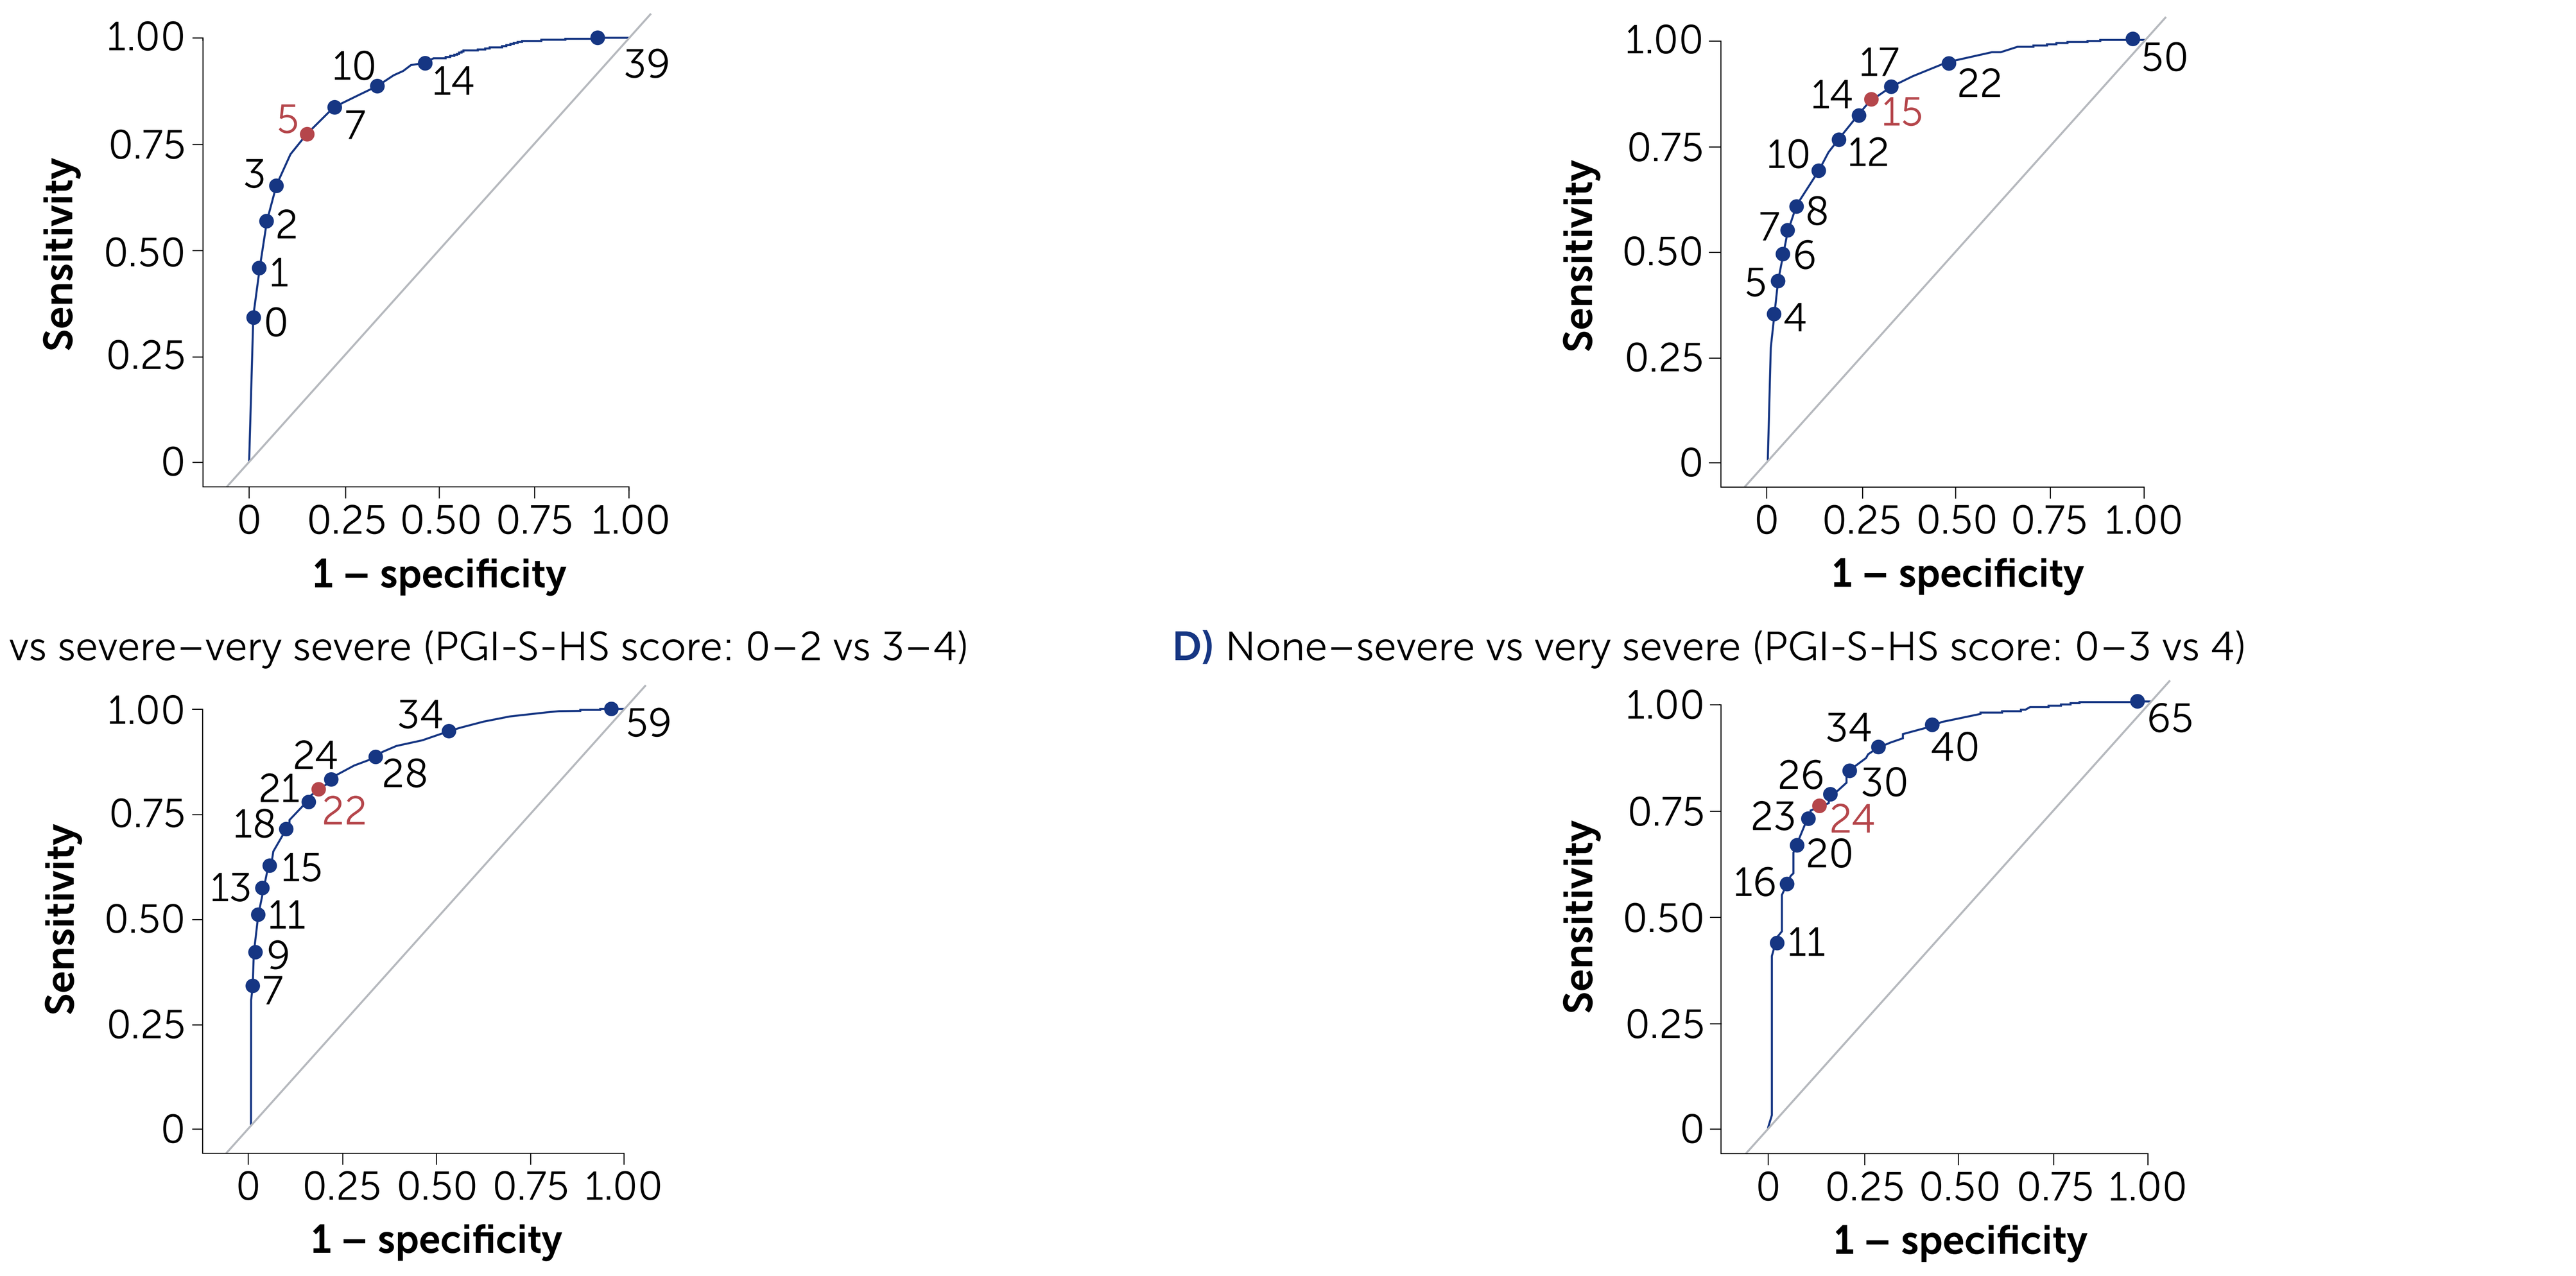

A) None vs mild–very severe (PGI-S-HS score: 0 vs 1–4)
B) None–mild vs moderate–very severe (PGI-S-HS score: 0–1 vs 2–4)
C) None–moderate vs severe–very severe (PGI-S-HS score: 0–2 vs 3–4)
D) None–severe vs very severe (PGI-S-HS score: 0–3 vs 4)
Sensitivity
1 – specificity

HiSQOL analysis set (all randomised study patients with ≥1 non-missing HiSQOL subscale score at any scheduled assessment visit). Each target scale cut-off threshold for a given severity level was estimated from the highest Youden index of the ROC curve, using data pooled across visits at baseline, Week 4, Week 16, Week 32 and Week 48. Red markers indicate cut-offs corresponding to the maximum Youden index values.

Figure 3 Severity thresholds for HiSQOL total and subscale scores

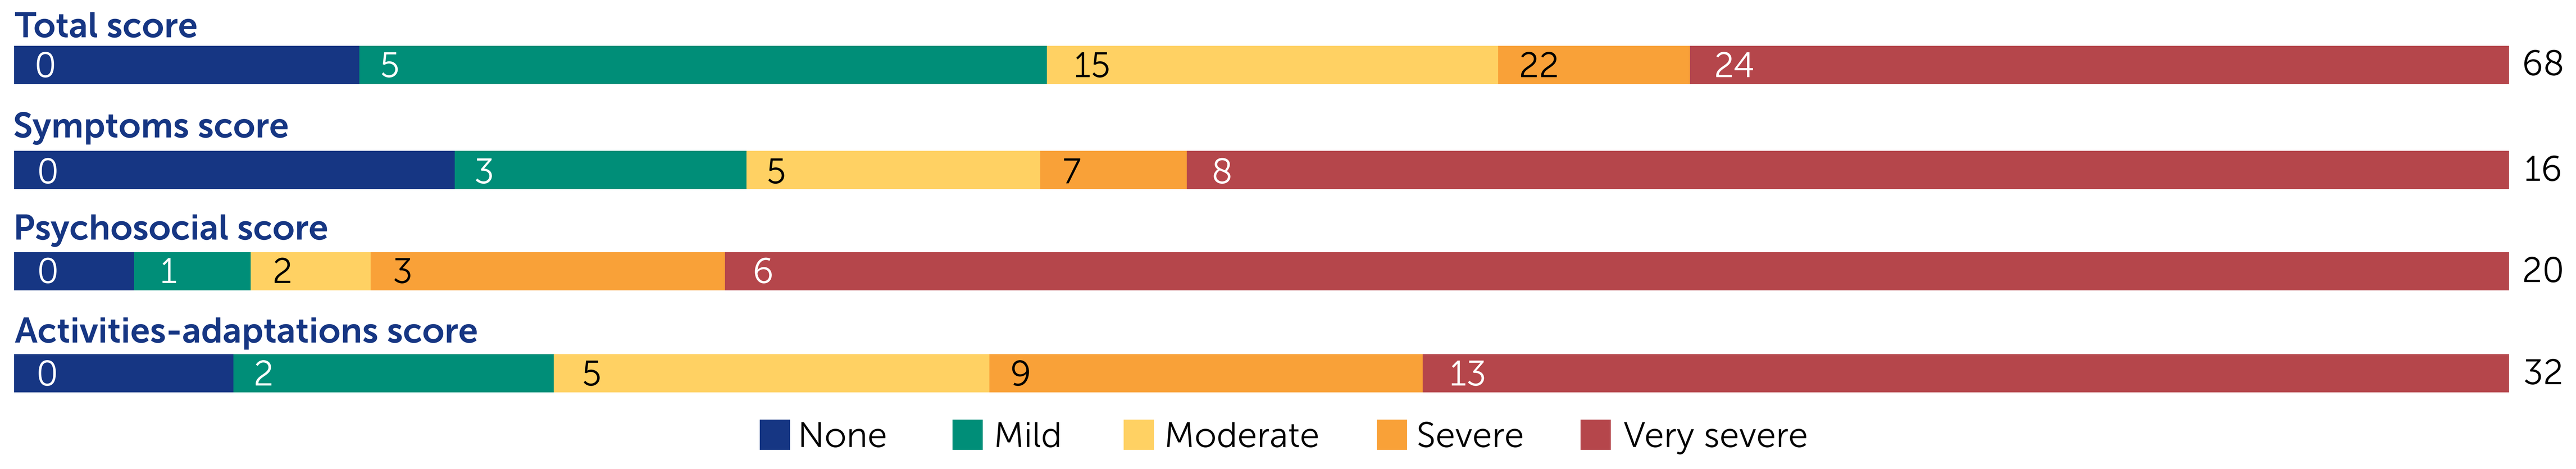

Total score
0 5 15 22 24 68
Symptoms score
0 3 5 7 8 16
Psychosocial score
0 1 2 3 6 20
Activities-adaptations score
0 2 5 9 13 32
None Mild Moderate Severe Very severe

Bars are not to scale. Numbers within the coloured bars indicate the lower score for each severity threshold.

AN: abscess and inflammatory nodule; **BMI:** body mass index; **DT:** draining tunnel; **eCDF:** empirical cumulative distribution function; **HiSQOL:** Hidradenitis Suppurativa Quality of Life; **HS:** hidradenitis suppurativa; **PGI-S-HS:** Patient Global Impression of Severity of Hidradenitis Suppurativa; **ROC:** receiver operating characteristic; **SD:** standard deviation.

Institutions: [1]Department of Dermatology, Penn State University, Hershey, Pennsylvania, USA; [2]Department of Dermatology & Academic Wound Healing, Division of Infection and Immunity, Cardiff University, Cardiff, UK; [3]UCB Pharma, Colombes, France; [4]UCB Pharma, Morrisville, North Carolina, USA; [5]UCB Pharma, Slough, UK; [6]UCB Pharma, Brussels, Belgium; [7]Evidera, Medellín, Colombia.

References: [1]Ingram JR et al. J Eur Acad Dermatol Venereol 2022;36:1597–605; [2]Kirby et al. Br J Dermatol 2020;183:340–8; [3]BE HEARD I: www.clinicaltrials.gov/study/NCT04242446; [4]BE HEARD II: www.clinicaltrials.gov/study/NCT04242498.
Author Contributions: Substantial contributions to study conception/design, or acquisition/analysis/interpretation of data: **JSK**, **JRI**, **JL**, **RR**, **EM**, **IP**, **CGP**; Drafting of the publication, or reviewing it critically for important intellectual content: **JSK**, **JRI**, **JL**, **RR**, **EM**, **IP**, **CGP**; Final approval of the publication: **JSK**, **JRI**, **JL**, **RR**, **EM**, **IP**, **CGP**. **Author Disclosures: JSK:** Reports personal fees from AbbVie, ChemoCentryx, CSL Behring, DermTech, Incyte, Insmed, Janssen, MoonLake Immunotherapeutics, Novartis, and UCB Pharma; personal fees and grants from Incyte. Co-copyright holder of HiSQOL©; consultant for and received honoraria from AbbVie, Alumis, DermTech, Incyte, Insmed, Janssen, MoonLake Immunotherapeutics, Novartis, and UCB Pharma; has been a speaker for AbbVie, Janssen, Novartis, and UCB Pharma. **JRI:** Receives a stipend as Editor-in-Chief of the British Journal of Dermatology and an authorship honorarium from UpToDate; consultant for AbbVie, Boehringer Ingelheim, ChemoCentryx, Citryll, MoonLake Immunotherapeutics, Novartis, UCB Pharma, and Union Therapeutics; has served on advisory boards for Insmed, Kymera Therapeutics, and Viela Bio; co-copyright holder of HiSQOL© and HS-IGA; his department receives income from copyright of the Dermatology Life Quality Instrument (DLQI) and related instruments. **JL**, **RR**, **EM**, **IP:** Employees and shareholders of UCB Pharma. **CGP:** Employee of Evidera, a part of Thermo Fisher Scientific that receives funding for research from UCB Pharma. **Acknowledgements:** This study was funded by UCB Pharma. We thank the patients and their caregivers in addition to the investigators and their teams who contributed to this study. The authors acknowledge Susanne Wiegratz, MSc, UCB Pharma, Monheim, Germany for publication coordination, Jack Wardle, MSc, Costello Medical, Cambridge, UK for medical writing and editorial assistance and the Creative team at Costello Medical for graphic design assistance. All costs associated with development of this poster were funded by UCB Pharma.

To receive a copy of this poster, scan the QR code or visit:
UCBposters.com/ISPOREU2023
Poster ID: PCR149
Link expiration: 29 November 2023

Presented at ISPOR Europe 2023 | Copenhagen, Denmark | 12–15 November 2023