A Novel Approach to Capturing Real-World Reasons Behind Use and Satisfaction of NVX-CoV2373 Booster Use Among German Adults: Direct-to-Patient Enrollment

Sarah Calhoun,[1] Kelly Hollis,[1] Lucie Kutikova,[2] Kawitha Helme,[2] Angela Miller,[2] Tino F Schwarz[3]

[1]RTI Health Solutions, Research Triangle Park, NC, USA; [2]Novavax Inc., Gaithersburg, Maryland, USA; [3]Institute of Laboratory Medicine and Vaccination Centre, Klinikum Wuerzburg Mitte, Campus Juliusspital, Wuerzburg, Germany

Acceptance Code: **PCR212**

Contact: **Lucie Kutikova**
lkutikova@Novavax.com

BACKGROUND

- As COVID-19 transitions from a global public health emergency to an endemic disease, individuals at increased risk of severe COVID-19 need continued protection due to the constant presence of the SARS-CoV-2 virus.
- The German Standing Committee on Vaccination (STIKO) recommends all authorized vaccines including NVX-CoV2373, the Novavax COVID-19 vaccine, as a primary series and booster dose. NVX-CoV2373 is the only available protein-based vaccine as all others are mRNA.
- To date, there are no real-world data in Germany describing the use of, reactogenicity, and satisfaction with NVX-CoV2373 boosters. This poster will outline the methodological challenges as well as a recommended approach for capturing data in the dynamic landscape of COVID-19 vaccinations.

As COVID-19 Vaccinations Move to Annual Booster the Key Research Questions Change

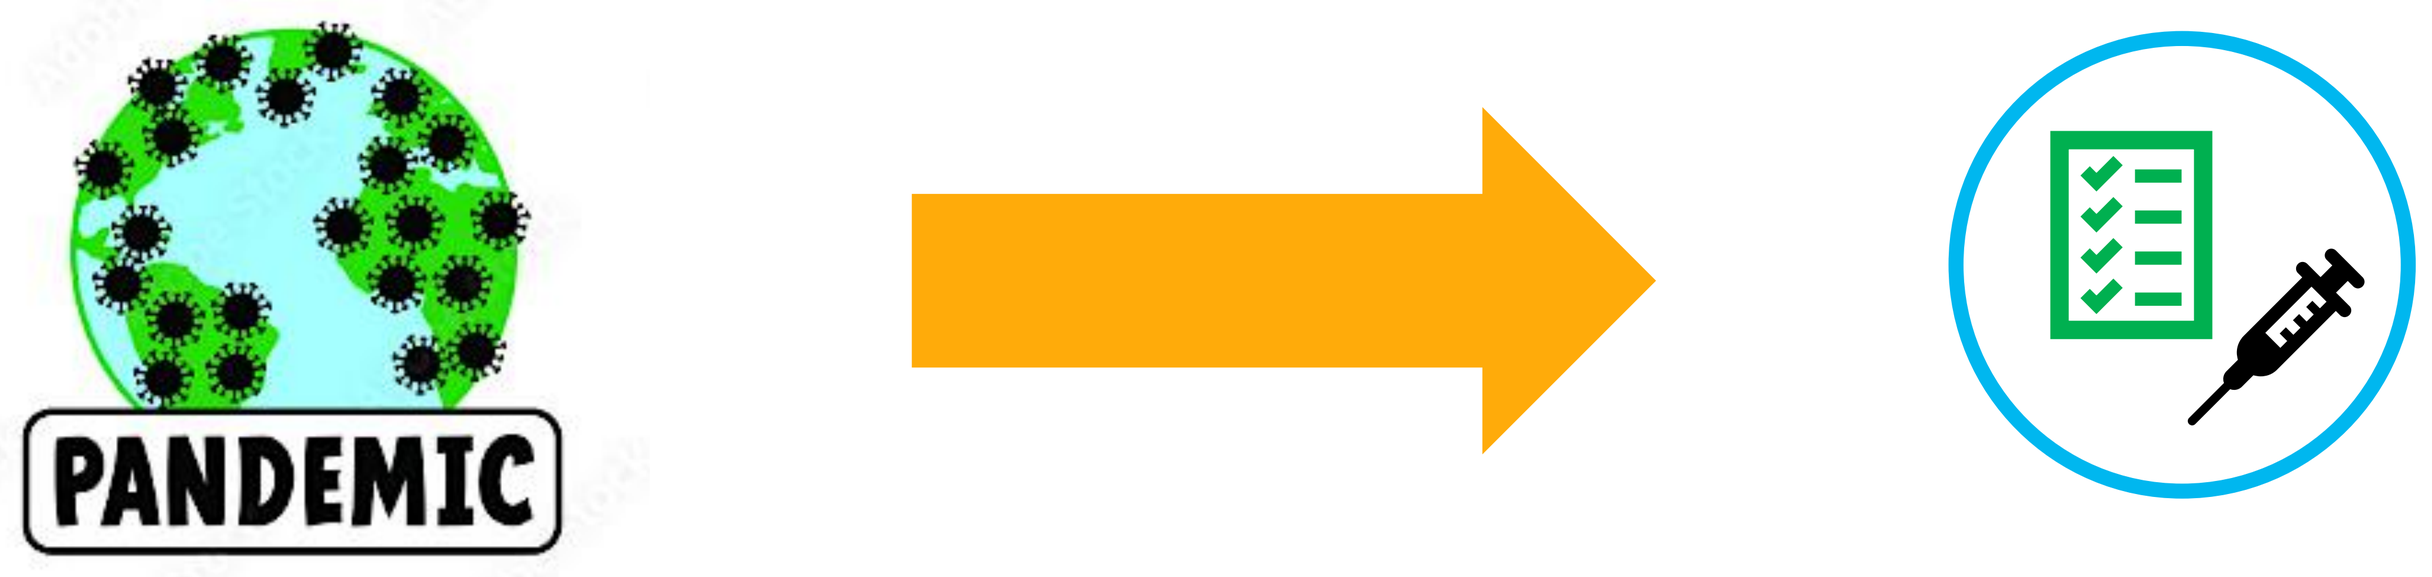
PANDEMIC

Key Questions in the Pandemic:
- What is the vaccine effectiveness against severe hospitalizations?
- What differences are there in protection among vaccines?

Key Questions Today:
- What is the reactogenicity resulting from vaccination?
- What motivates physicians/patients to select a mRNA or a protein-based vaccine?
- What is the patient's satisfaction with the vaccine?

Populations of Interest in Germany Have Also Changed

Previous recommendations were widespread; today more than 95% of the population has been exposed to SARS-CoV-2 antigens through vaccination and/or infection[1]

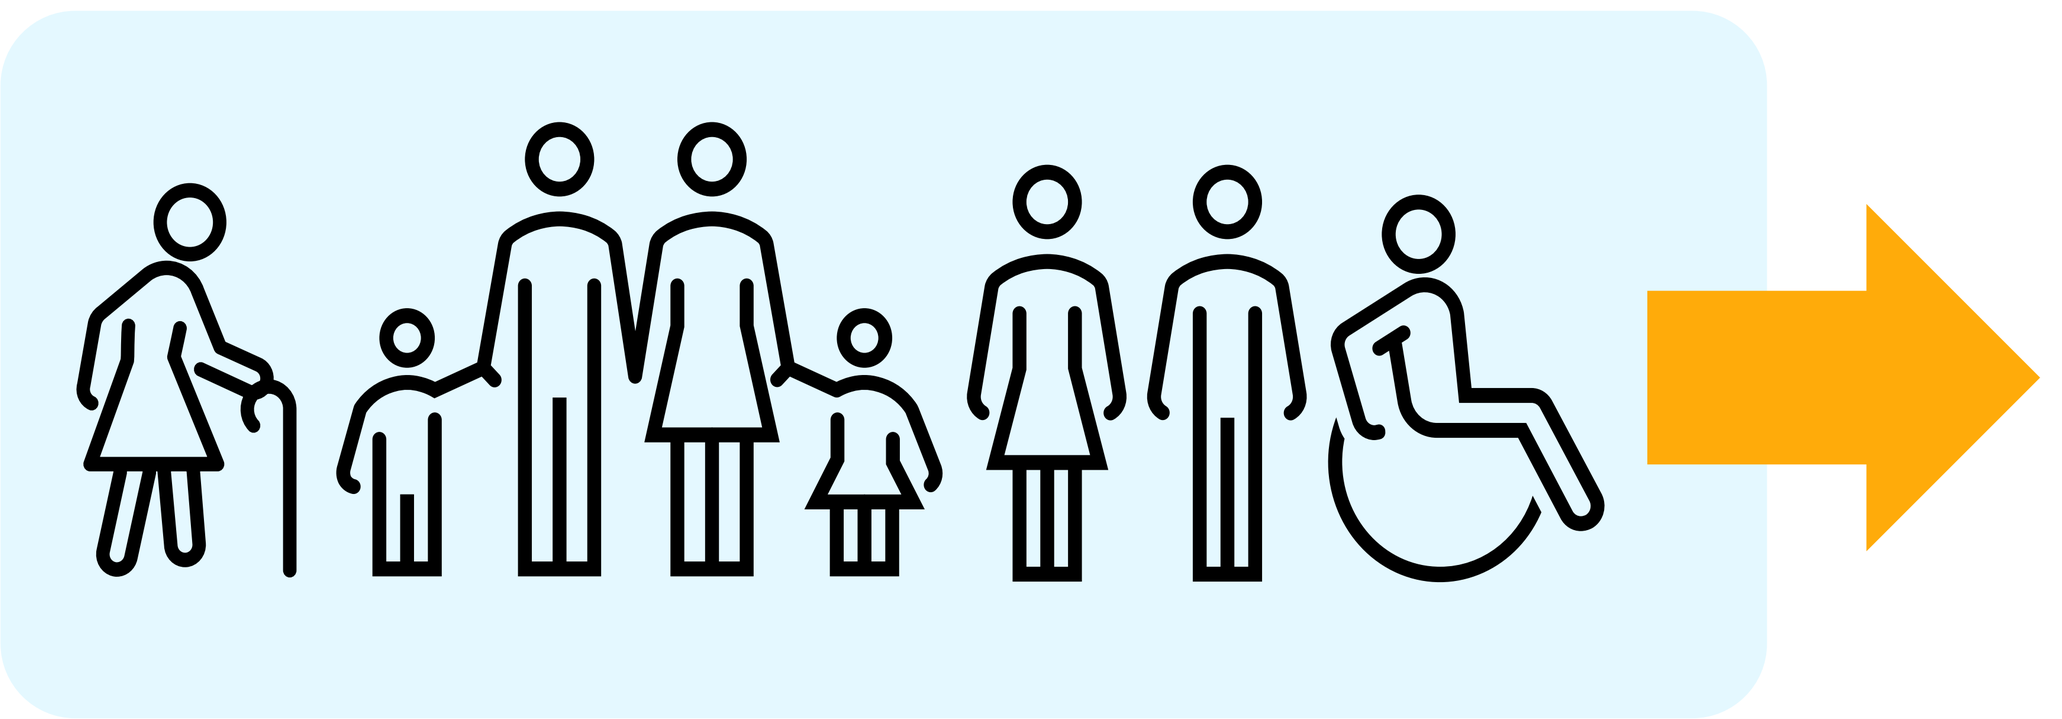

STIKO 2023 recommends an annual booster dose for the following high-risk groups:

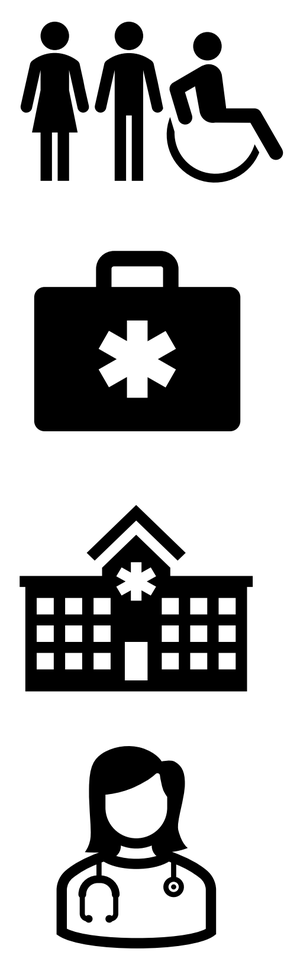

- Adults > 60 years
- Adults with chronic medical conditions
- Persons being cared for in nursing homes
- Healthcare workers

Our Understanding of Reactogenicity is Evolving

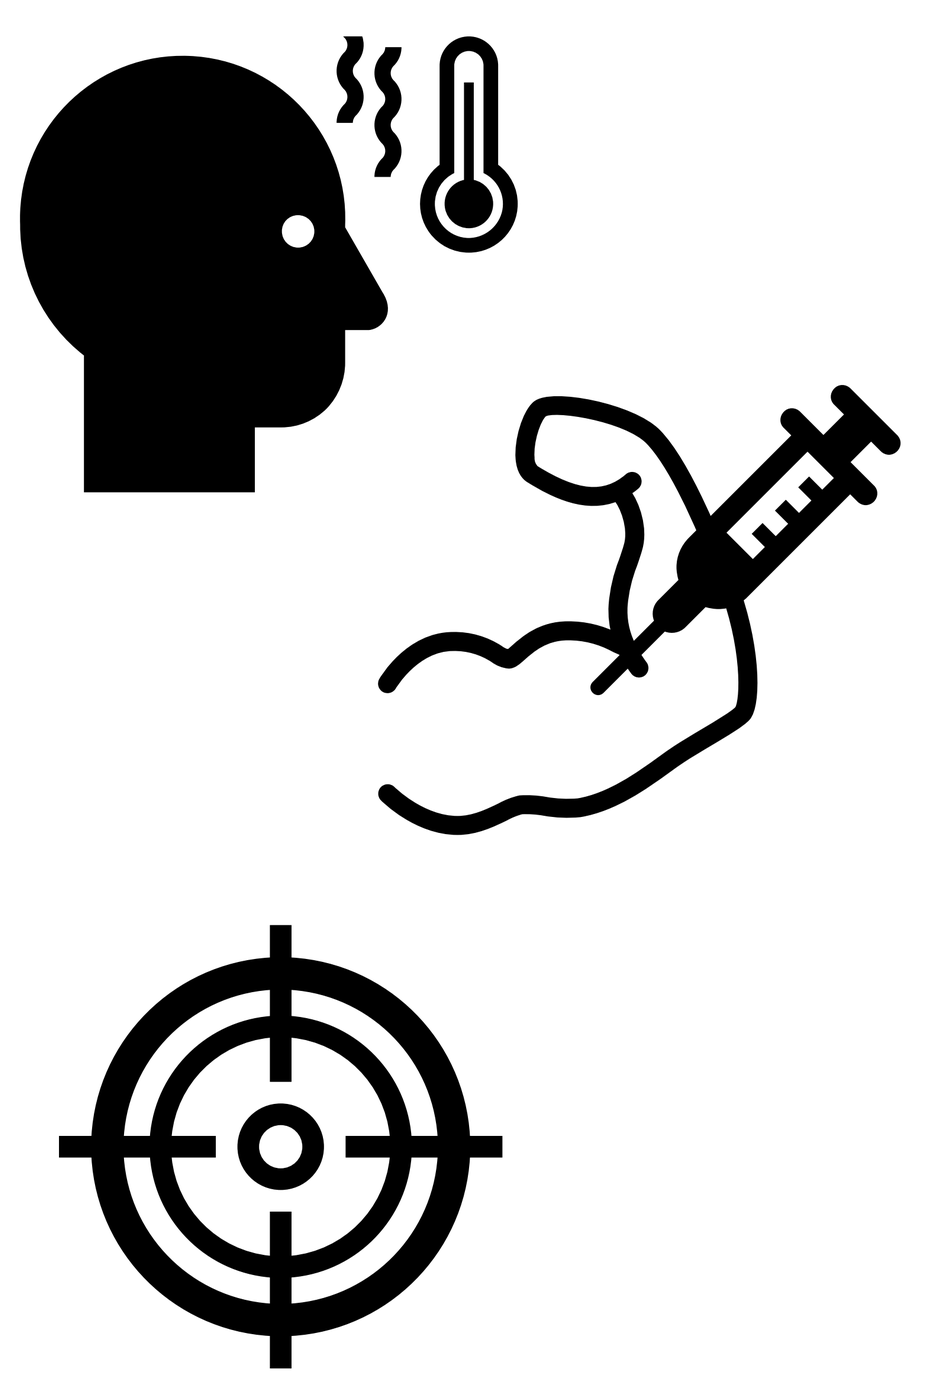

- There is a lack of consistency in reported events; however, the most frequently reported are fever, fatigue, headache, pain at injection site, redness and swelling **(Table 1)**
- There is a difference in occurrence of reactogenicity events by vaccine type **(Table 1)**
- Factors that effect reactogenicity include dose, age, and comparator group **(Table 1)**
- In a post hoc analysis of data from a descriptive study of NVX-CoV2373 booster recipients compared to mRNA, it was observed that the overall percentage of NVX-CoV2373 booster recipients experiencing any reactogenicity was 77.6% vs. 95.9% for mRNA recipients; incidences of local and systemic symptoms were lower for all events reported for the NVX-CoV2373 cohort.[3] As the data are observational only, these findings do not themselves support comparative conclusions but provide a rationale for further hypothesis-driven research

Table 1. Published Descriptive COVID Vaccine Reactogenicity Data

Author	Journal	Year	DOI Number
Munro et al.	*Lancet*	2021	10.1016/S0140-6736(21)02717-3
Stuart et al.	*Lancet*	2021	10.1016/S0140-6736(21)02718-5
Atmar et al.	*NEJM*	2022	10.1056/NEJMoa2116414
Salter et al.	*Vaccines*	2022	10.3390/vaccines10122017
Sutton et al.	*Exp Rev of Vaccines*	2022	10.1080/14760584.2022.2098719
Lyke et al.	*npj Vaccines*	2023	10.1038/s41541-023-00693-z
Rousculp et al.	*MedRxiv pre-print*	2023	10.1101/2023.05.31.23290594

- Additional data on COVID Vaccine reactogenicity, and potential platform specific differences is needed.

METHODS*

Challenge #1: Traditional Site-Based Data Capture is Not Optimal

- As the new vaccines with the current variants enter the German market, real-world data capture using sites is difficult due to

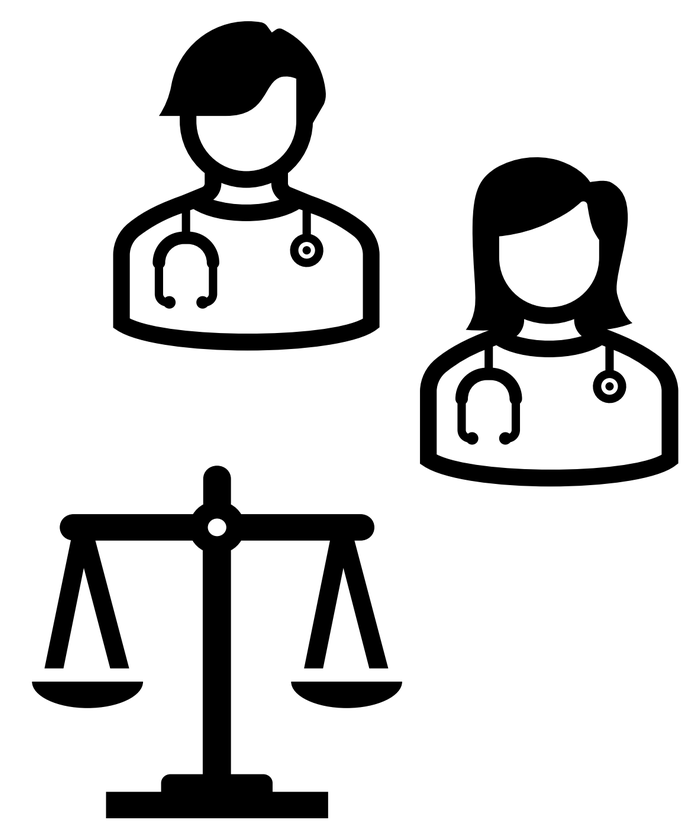

- General practitioners typically do not participate in pharmaceutical trials in Germany
- Infrequent use of NVX-CoV2373 creates an imbalance in recruitment that could result in bias in geography and time
- The number of sites that would be required to achieve the sample size would be extensive due to the specific patient population eligible for the vaccine

Challenge #2: Capturing Reactogenicity Data

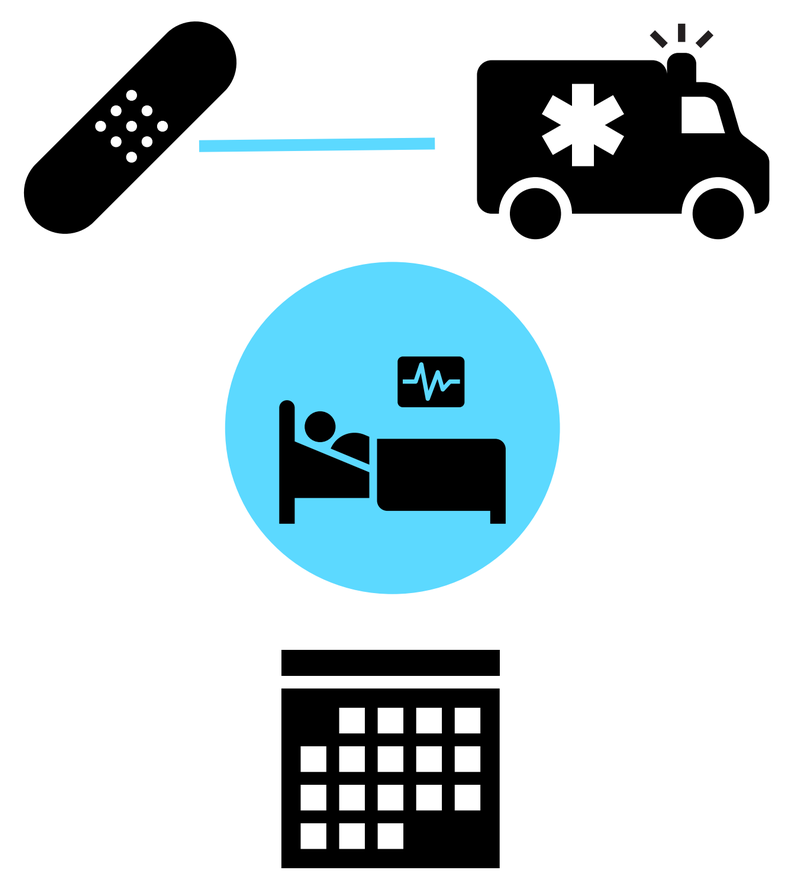

- Local and systemic reactogenicity events are categorized as none, mild, moderate and severe
- Research has shown that severe reactogenicity events resulting in a hospitalization or medical visits are rare; therefore, the patient is the most reliable source for reactogenicity data
- Research has also shown that the bulk of reactogenicity events occur within 2 to 3 days of vaccination; thus it is critical to capture data in a narrow recall window

Novel Methodology

- Direct-to-patient enrollment approach through online technology using study-specific algorithms that evaluate web traffic across more than 1,500 websites, social media platforms, mobile applications, and patient advocacy groups.
- If an individual's online activity aligns with the algorithm, a study advertisement will appear, allowing the individual to indicate interest. Interested individuals will receive an email detailing the study design and a link to electronically consent and answer eligibility questions.
- After consenting, participants will then complete a survey to assess reactogenicity, their perceptions of motivators of vaccine choice, and overall satisfaction with the vaccine.

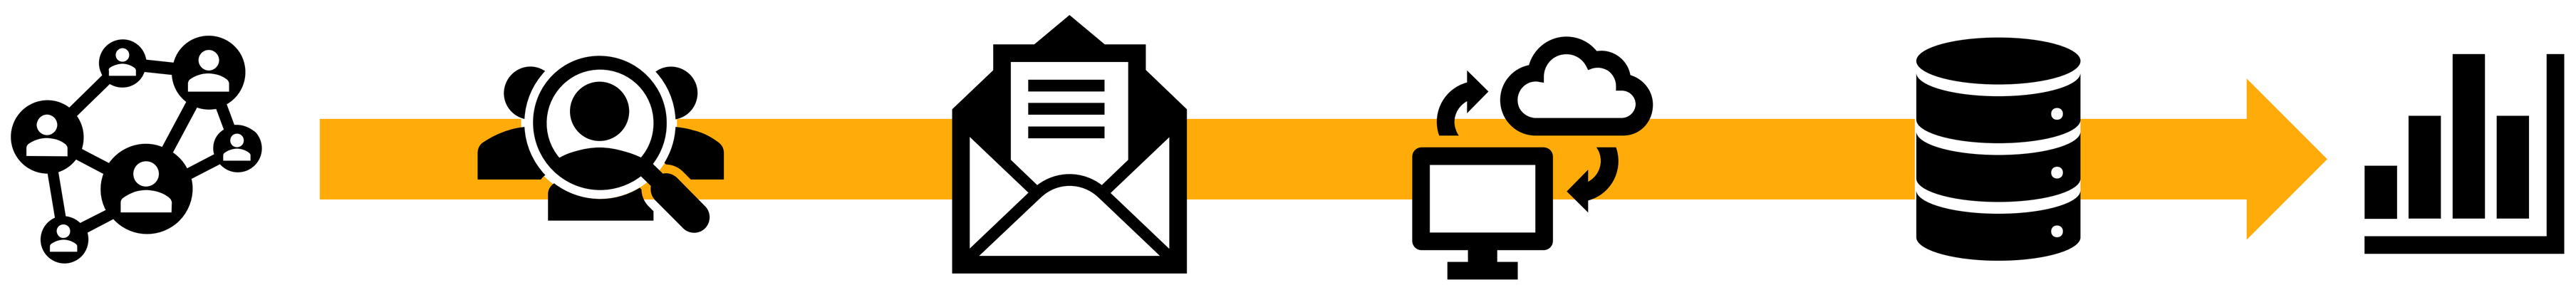

Advantages to Recruitment Via Social Media

- Allows widespread recruiting to capture a diversity of patient populations to be more representative of a real-world population
- Real-time data capture
- Quick and easy participant enrollment to capture prospective data on vaccinations

REFERENCES

1. Statement of the Standing Commission on Vaccination (STIKO) at the Robert Koch Institute Decision on the implementation of the COVID-19 vaccination into the general recommendations of the STIKO 2023; available at https://www.rki.de/EN/Content/infections/Vaccination/recommandations/implementation_covid-19_vaccination.pdf? __blob=publicationFile.
2. Sutton N, et al. (2022) Comparing reactogenicity of COVID-19 vaccines: a systematic review and meta-analysis, Expert Review of Vaccines, 21:9,1301-1318, DOI: 10.1080/14760584.2022.2098719.
3. Rousculp M, Ziemiecki R, Marchese A. Protein Vaccine Demonstrates Less Reactogenicity than mRNA – A Real World Study. MedRxiv preprint doi: https://doi.org/10.1101/2023.05.31.23290594.

CONCLUSIONS

- Data collection will begin in fall 2023. This cross-sectional study will enroll adults residing in Germany who received an NVX-CoV2373 or mRNA booster within 5 days of enrollment.
- Analysis will descriptively characterize study participants, evaluate reactogenicity and describe reasons for receiving the type of vaccine.
- Analysis will be powered for comparison of systemic reactogenicity between mRNA and Novavax vaccine.

**Methodology has changed since the submission of the abstract.*

Disclosures and Acknowledgments: Study was funded by Novavax Inc. SC and KH are employees of RTI. LK and AM are employees of Novavax Inc. KH is a former employee of Novavax Inc. TS is an employee of Klinikum Wuerzburg Mitte.
Poster layout support was provided by Allison Saviano of Sephirus Communications.