Poster Session 2(13 Nov. 2023)

KRPIA Korean Research-based Pharma Industry Association

Exploring the feasibility of indication-based pricing (IBP) in the Korea Healthcare System: A survey study

SA32

Borami Kim[1], Youngshin Lee[1], Saeyeon Ahn[2], Sukhyun Kang[2], Bora Shin[3], Joonsu Kim[4z]

BACKGROUND

- South Korea has a single drug pricing system while there is an increasing number of innovative new drugs approved with multiple indications. And the drug price usually gets lower as it expands additional indications.
- Therefore, multinational companies that are hesitant to expand due to the international reference price will delay the launch in Korea, creating obstacles to Korean patients' access.

OBJECTIVES

- To assess the viability, this study conducts a survey about 'Indication Based Pricing(IBP)' such as weighted average pricing across indications and indication-based rebates.

METHODS

- A focus group interview was conducted first with five experts, divided into five groups: government, healthcare physician, patient advocacy groups & media, academia, and pharmaceutical industry.
- Based on the opinions obtained here, the final questionnaire was organized, and an online survey was conducted on 202 experts and stakeholders from October 5, to November 10, 2022

RESULTS

- As a result of the survey, an average of 62.9% of the respondents said the introduction of a drug pricing system by indication was necessary only for drugs that met certain criteria.
 - Total responders(n=202); Industry(n=56); Academia(n=45); Government officers(n=9); Physician(n=57); Patient(n=35)
- All groups except the government responded that it was most appropriate to pay the government a different refund rate for each indication while leaving the list price intact.

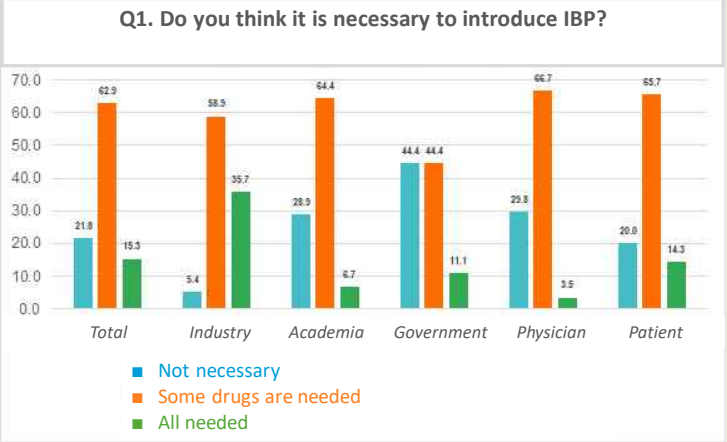
Q1. Do you think it is necessary to introduce IBP?
Total
Industry
Academia
Government
Physician
Patient
Not necessary
Some drugs are needed
All needed

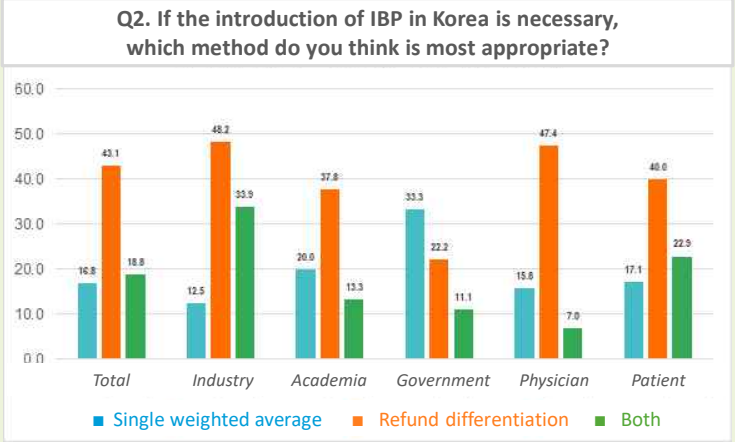
Q2. If the introduction of IBP in Korea is necessary, which method do you think is most appropriate?
Total
Industry
Academia
Government
Physician
Patient
Single weighted average
Refund differentiation
Both

- Opinions on the expected effects and concerns when introducing the drug price system for each indication in Korea are as follows. Expected effects include ‘improving patient access to drugs’, ‘value-based evaluation’, and ‘high feasibility using existing system’.

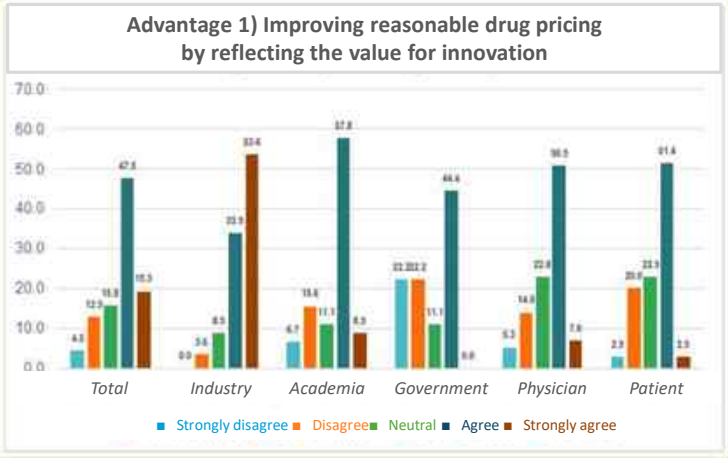
Advantage 1) Improving reasonable drug pricing by reflecting the value for innovation
Total
Industry
Academia
Government
Physician
Patient
Strongly disagree
Disagree
Neutral
Agree
Strongly agree

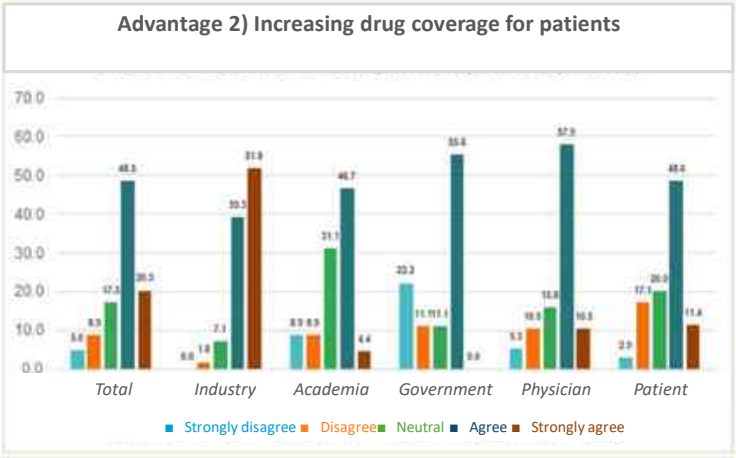
Advantage 2) Increasing drug coverage for patients
Total
Industry
Academia
Government
Physician
Patient
Strongly disagree
Disagree
Neutral
Agree
Strongly agree

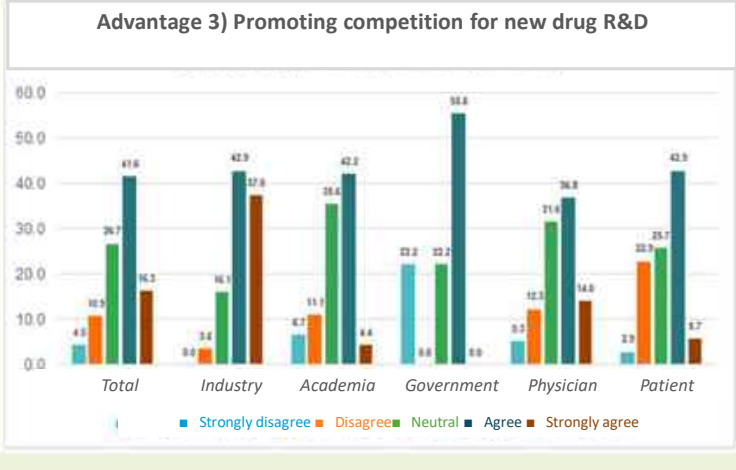
Advantage 3) Promoting competition for new drug R&D
Total
Industry
Academia
Government
Physician
Patient
Strongly disagree
Disagree
Neutral
Agree
Strongly agree

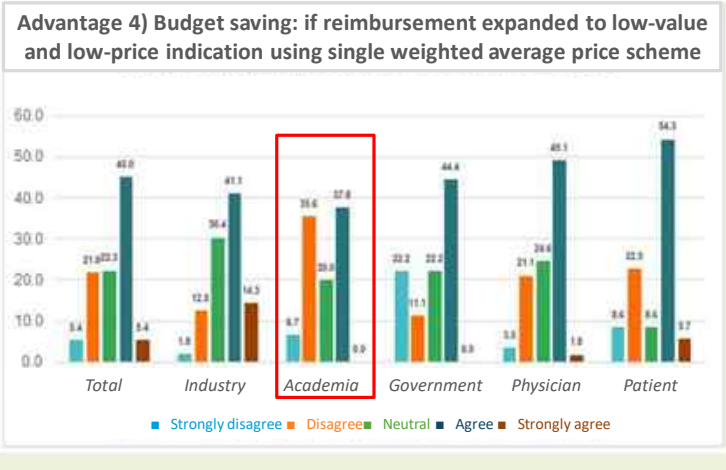
Advantage 4) Budget saving: if reimbursement expanded to low-value and low-price indication using single weighted average price scheme
Total
Industry
Academia
Government
Physician
Patient
Strongly disagree
Disagree
Neutral
Agree
Strongly agree

- Concerns include ‘administrative cost and infrastructure construction’, ‘the burden of repetitive cost-effectiveness assessment’, ‘equality and capacity of social members among patients’, ‘disturbing drug claims’, ‘supplier-oriented drug price calculation’, and ‘excessive drug price’

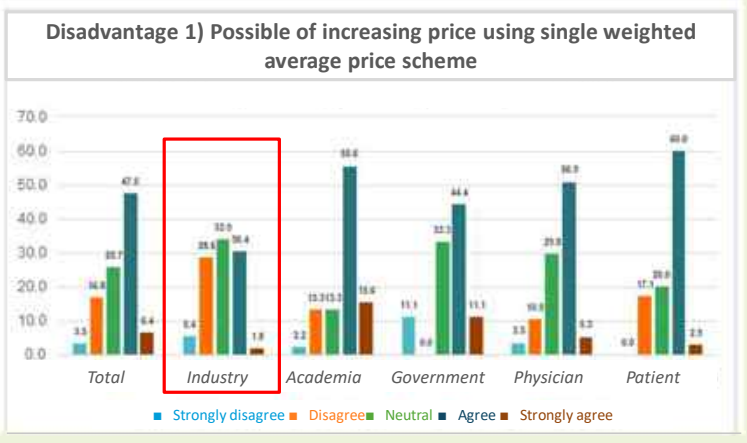
Disadvantage 1) Possible of increasing price using single weighted average price scheme
Total
Industry
Academia
Government
Physician
Patient
Strongly disagree
Disagree
Neutral
Agree
Strongly agree

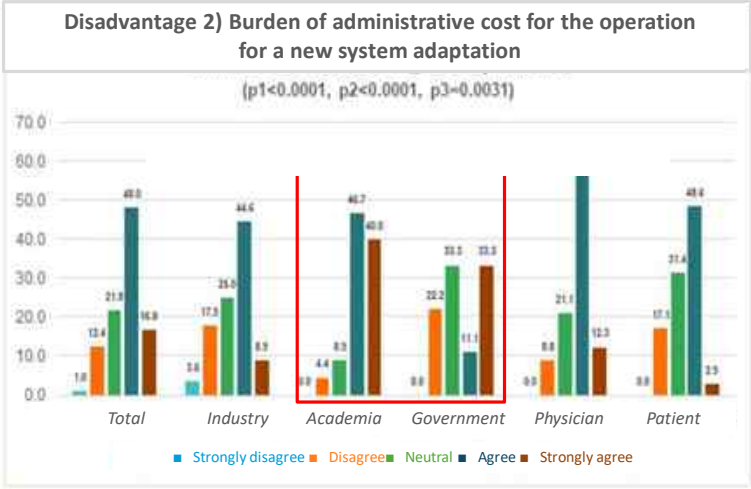
Disadvantage 2) Burden of administrative cost for the operation for a new system adaptation
(p1<0.0001, p2<0.0001, p3=0.0031)
Total
Industry
Academia
Government
Physician
Patient
Strongly disagree
Disagree
Neutral
Agree
Strongly agree

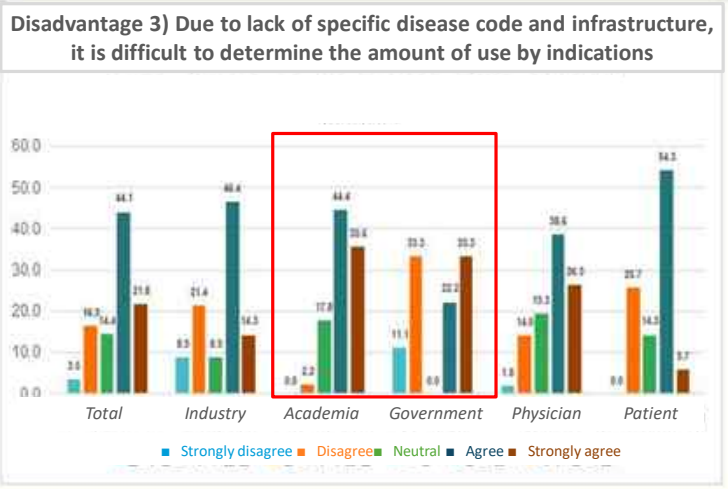
Disadvantage 3) Due to lack of specific disease code and infrastructure, it is difficult to determine the amount of use by indications
Total
Industry
Academia
Government
Physician
Patient
Strongly disagree
Disagree
Neutral
Agree
Strongly agree

- Opinions on the regulations and operational matters of the drug price system for each indication in Korea are as follows.
 - Submission data: 'cost-effectiveness evaluation by indication', 'total amount CAP by pharmaceutical company', and 'economic evaluation data' are mentioned.
 - The system construction method is divided according to two operational plans.
 1) Single weighted average: 'weighted average formula', 'using existing billing system’
 2) Refund rate differential applied: 'Post-listing system’
 - Commonly needed, 'Tracking usage through establishment of registry by indication', 'Evaluation of post-use and update of weighted average' and 'requires training system for prescribing medication by medical staff'

CONCLUSIONS

- If the operational aspects of the academic and the government can handle the indication-based pricing indication-based pricing will be an alternative to increase access to innovative new drugs desired by the industry and patients.

LIMITATIONS

- Government agencies such as the National Health Insurance Service (NHIS) and Health Insurance Review and Assessment (HIRA) tried to get them involved in the survey, but there were not enough respondents.
- Therefore, government groups may have too few respondents and lack representation, so there is a limitation in that caution is required when compared to other groups..

REFERENCES

- Preckler, Víctor, and Jaime Espín. "The role of indication-based pricing in future pricing and reimbursement policies: a systematic review." Value in Health 2022; 25(4): 666-675.
- Flume, Mathias, et al. "Feasibility and attractiveness of indication value-based pricing in key EU countries." Journal of Market Access & Health Policy 2016; 4(1): 30970.

ACKNOWLEDGEMENTS AND CONTACT INFORMATION

- The research presented in this Europe ISPOR 2023, was based on a research funded by KRPIA and submitted on December 30, 2022, by Professor Kang Hye-young's research team at Yonsei University's College of Pharmacy. I would like to take this opportunity to express my gratitude.
- **KRPIA** is a major Korean pharmaceutical industry association that represents the interests of global pharmaceutical companies aimed at developing new drugs that are operating in Korea and strives to improve the pricing and regulation system through the partnerships with the government so that Korean patients can access innovative new drugs. If you have any questions, please contact 1st author, Borami Kim, the co-chair of Market Access committee(borami.kim@krpia.or.kr).

1. Korea Research based Pharmaceutical Industry Association(KRPIA), Seoul, Republic of Korea, 2. MSD, Seoul, Republic of Korea, 3. AstraZeneca, Seoul, Republic of Korea, 4. AbbVie, Seoul, Republic of Korea

RESEARCH POSTER PRESENTATION DESIGN © 2022
www.PosterPresentations.com