EPH 73

Modelling the wider socio-economic costs of problematic opioid use in Germany

George Keel[1], Susanna Meyner[2], Daniel Mogford[2], Mike Burrows[3], Stojan Zavisic[2]

[1]Department of Learning, Informatics, Management, and Ethics, Medical Management Center, Karolinska Institutet, Stockholm, Sweden; [2]Camurus AB, Lund, Sweden; [3]Bury GP Federation, Bury, Greater Manchester, UK

ihe.se

AIM

To develop and apply a costing model to estimate the wider socio-economic costs, stratified by sector and liable payer, of problematic opioid users† (POUs) both in and out of treatment in Germany and the savings gained through recruitment or retention of POUs into treatment.

BACKGROUND

- Problematic opioid use is associated with substantial harm to public health and safety, with considerable costs to society.
- Studies consistently show that most of these costs fall outside the healthcare sector.[1,2] →
- A country's ability to successfully control the cost of opioid dependence is largely correlated with the capacity to get individuals into treatment.[1,2]
- Treatment is multi-faceted, and includes pharmacological and psychosocial therapy.[3] →
- In Germany, approximately 50% of all POUs are not in treatment.[4]

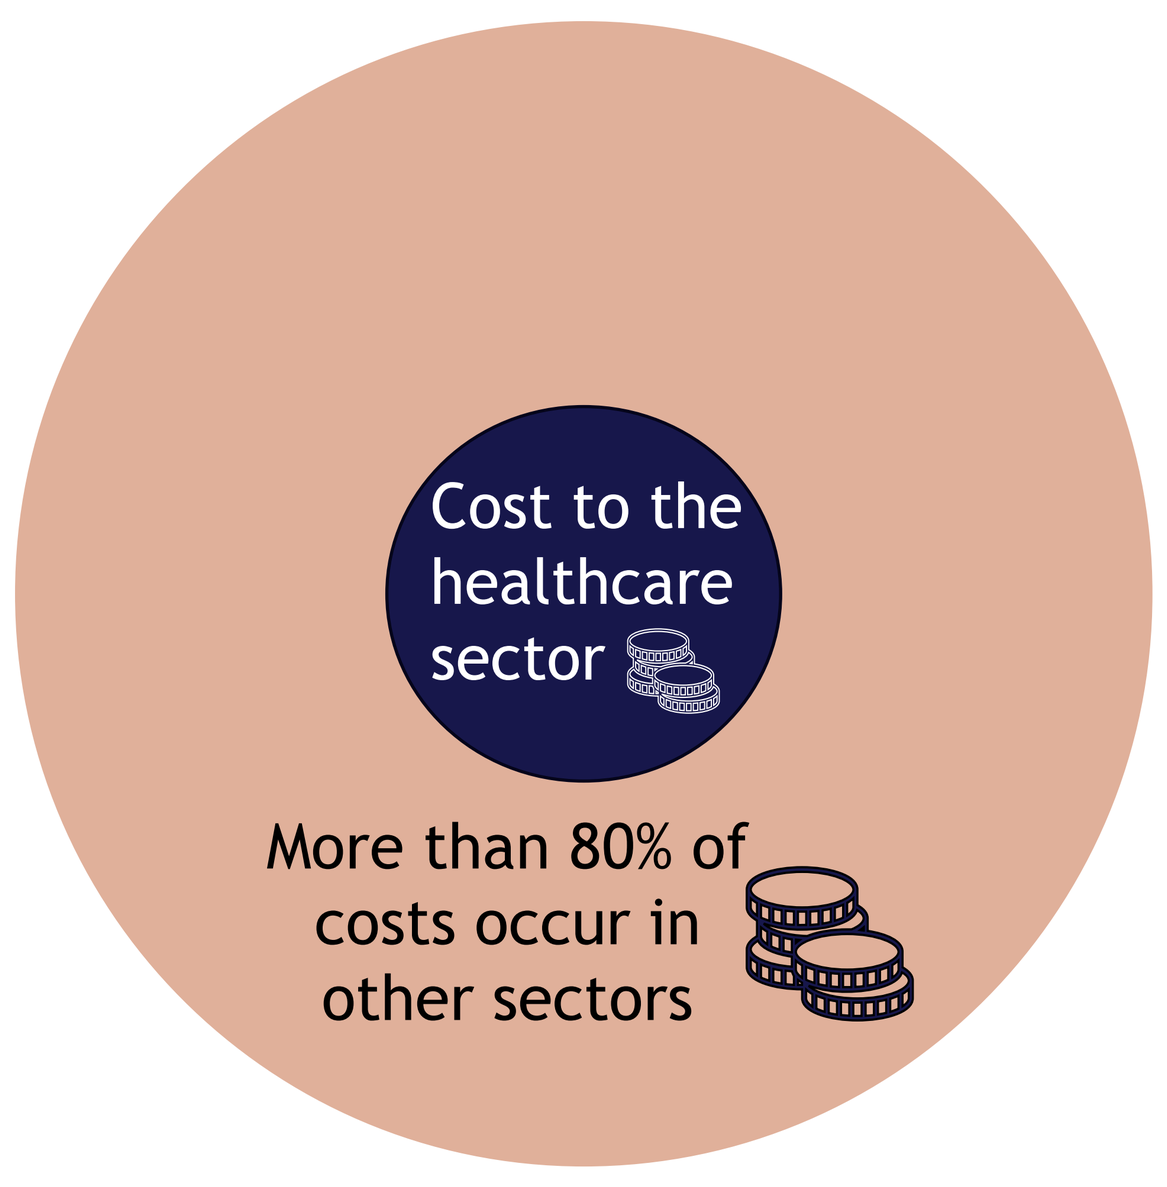
Societal costs of POUs
Cost to the healthcare sector
More than 80% of costs occur in other sectors

Opioid dependency treatment helps POUs in many areas

Gainful Employment[6]	Health complications and healthcare utilization[5]
Quality of life	Encounters with the criminal justice system[7]
Stable housing	
Sustained recovery	Infectious disease[7]

METHOD

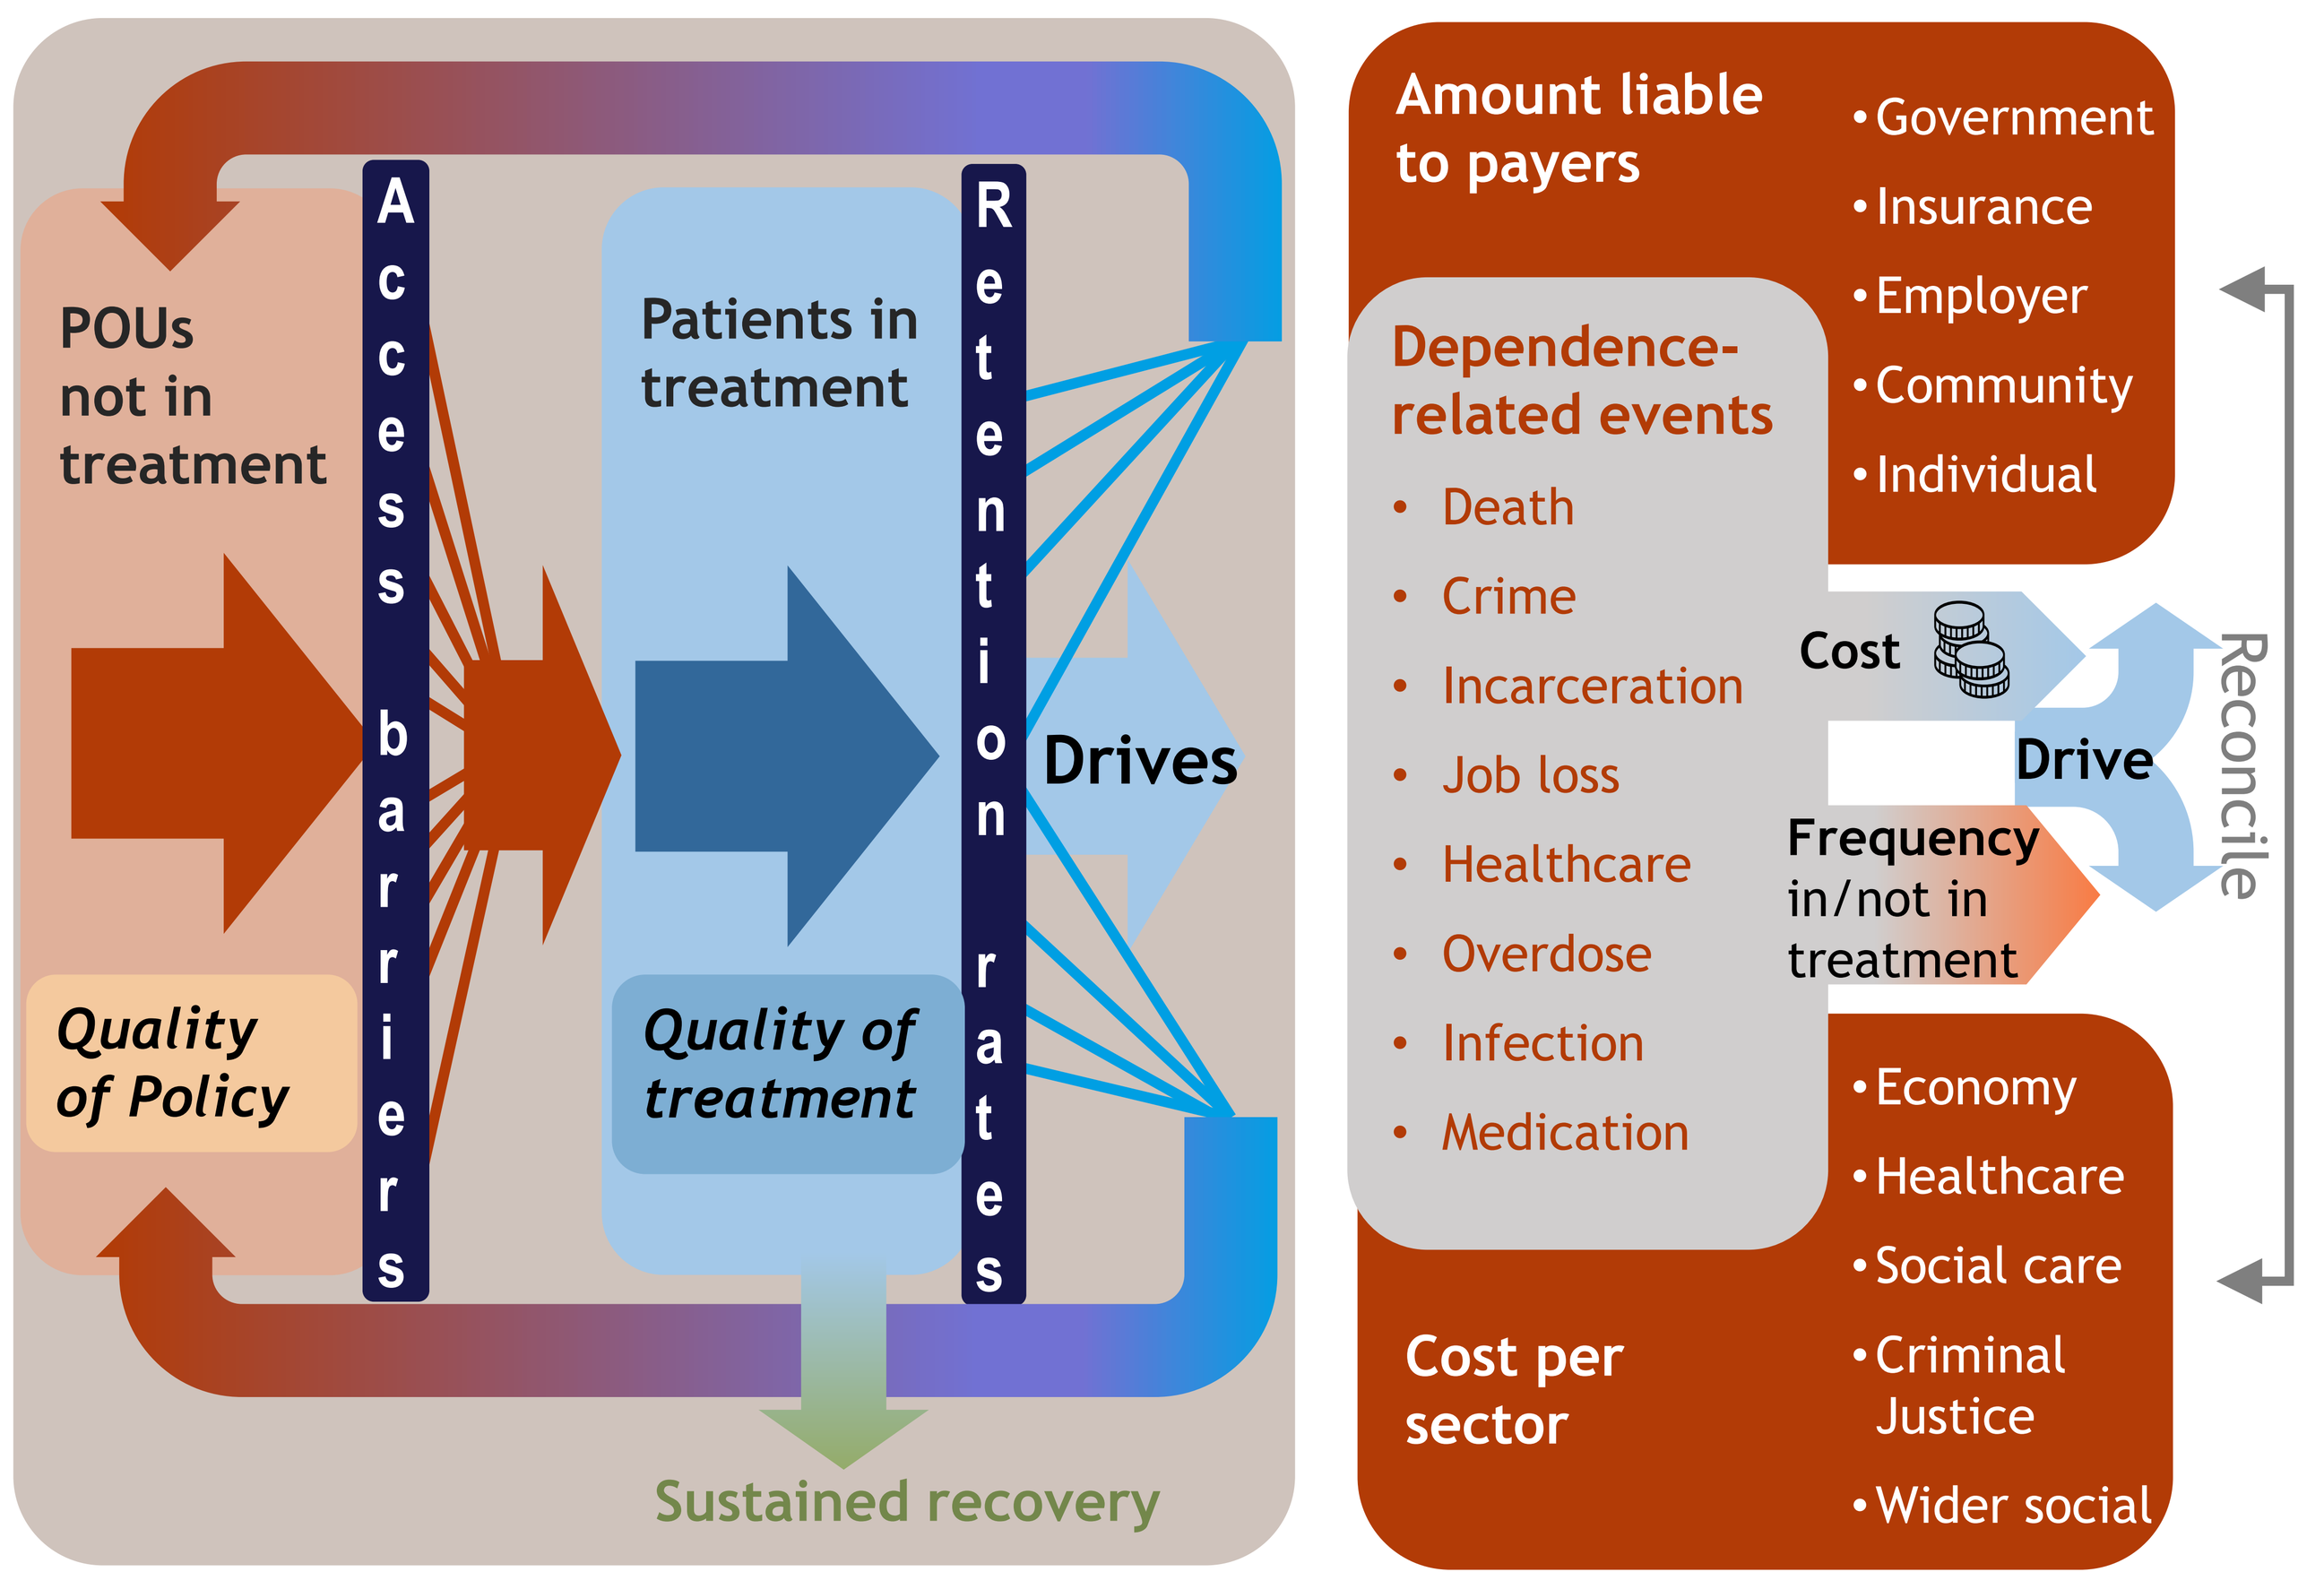
The Model Concept
POUs not in treatment
Quality of Policy
Access barriers
Patients in treatment
Quality of treatment
Retention rates
Drives
Sustained recovery
Amount liable to payers
•Government
•Insurance
•Employer
•Community
•Individual
Dependence-related events
Death
Crime
Incarceration
Job loss
Healthcare
Overdose
Infection
Medication
Cost
Drive
Frequency in/not in treatment
Reconcile
Cost per sector
•Economy
•Healthcare
•Social care
•Criminal Justice
•Wider social

- **Model conceptualization** - definition of relevant opioid dependence events per POU and categorization to sector and payer was based on a targeted literature review and input from experts in the field.
- **Key model inputs** - POU prevalence & demographic data, frequency & cost estimates of key events. German data where available, otherwise sourced internationally. Recovery was not included in this analysis.
- **Key model output** – Cost-per-POU in treatment & not in treatment

RESULTS

1. Cumulative annual costs per sector and actor

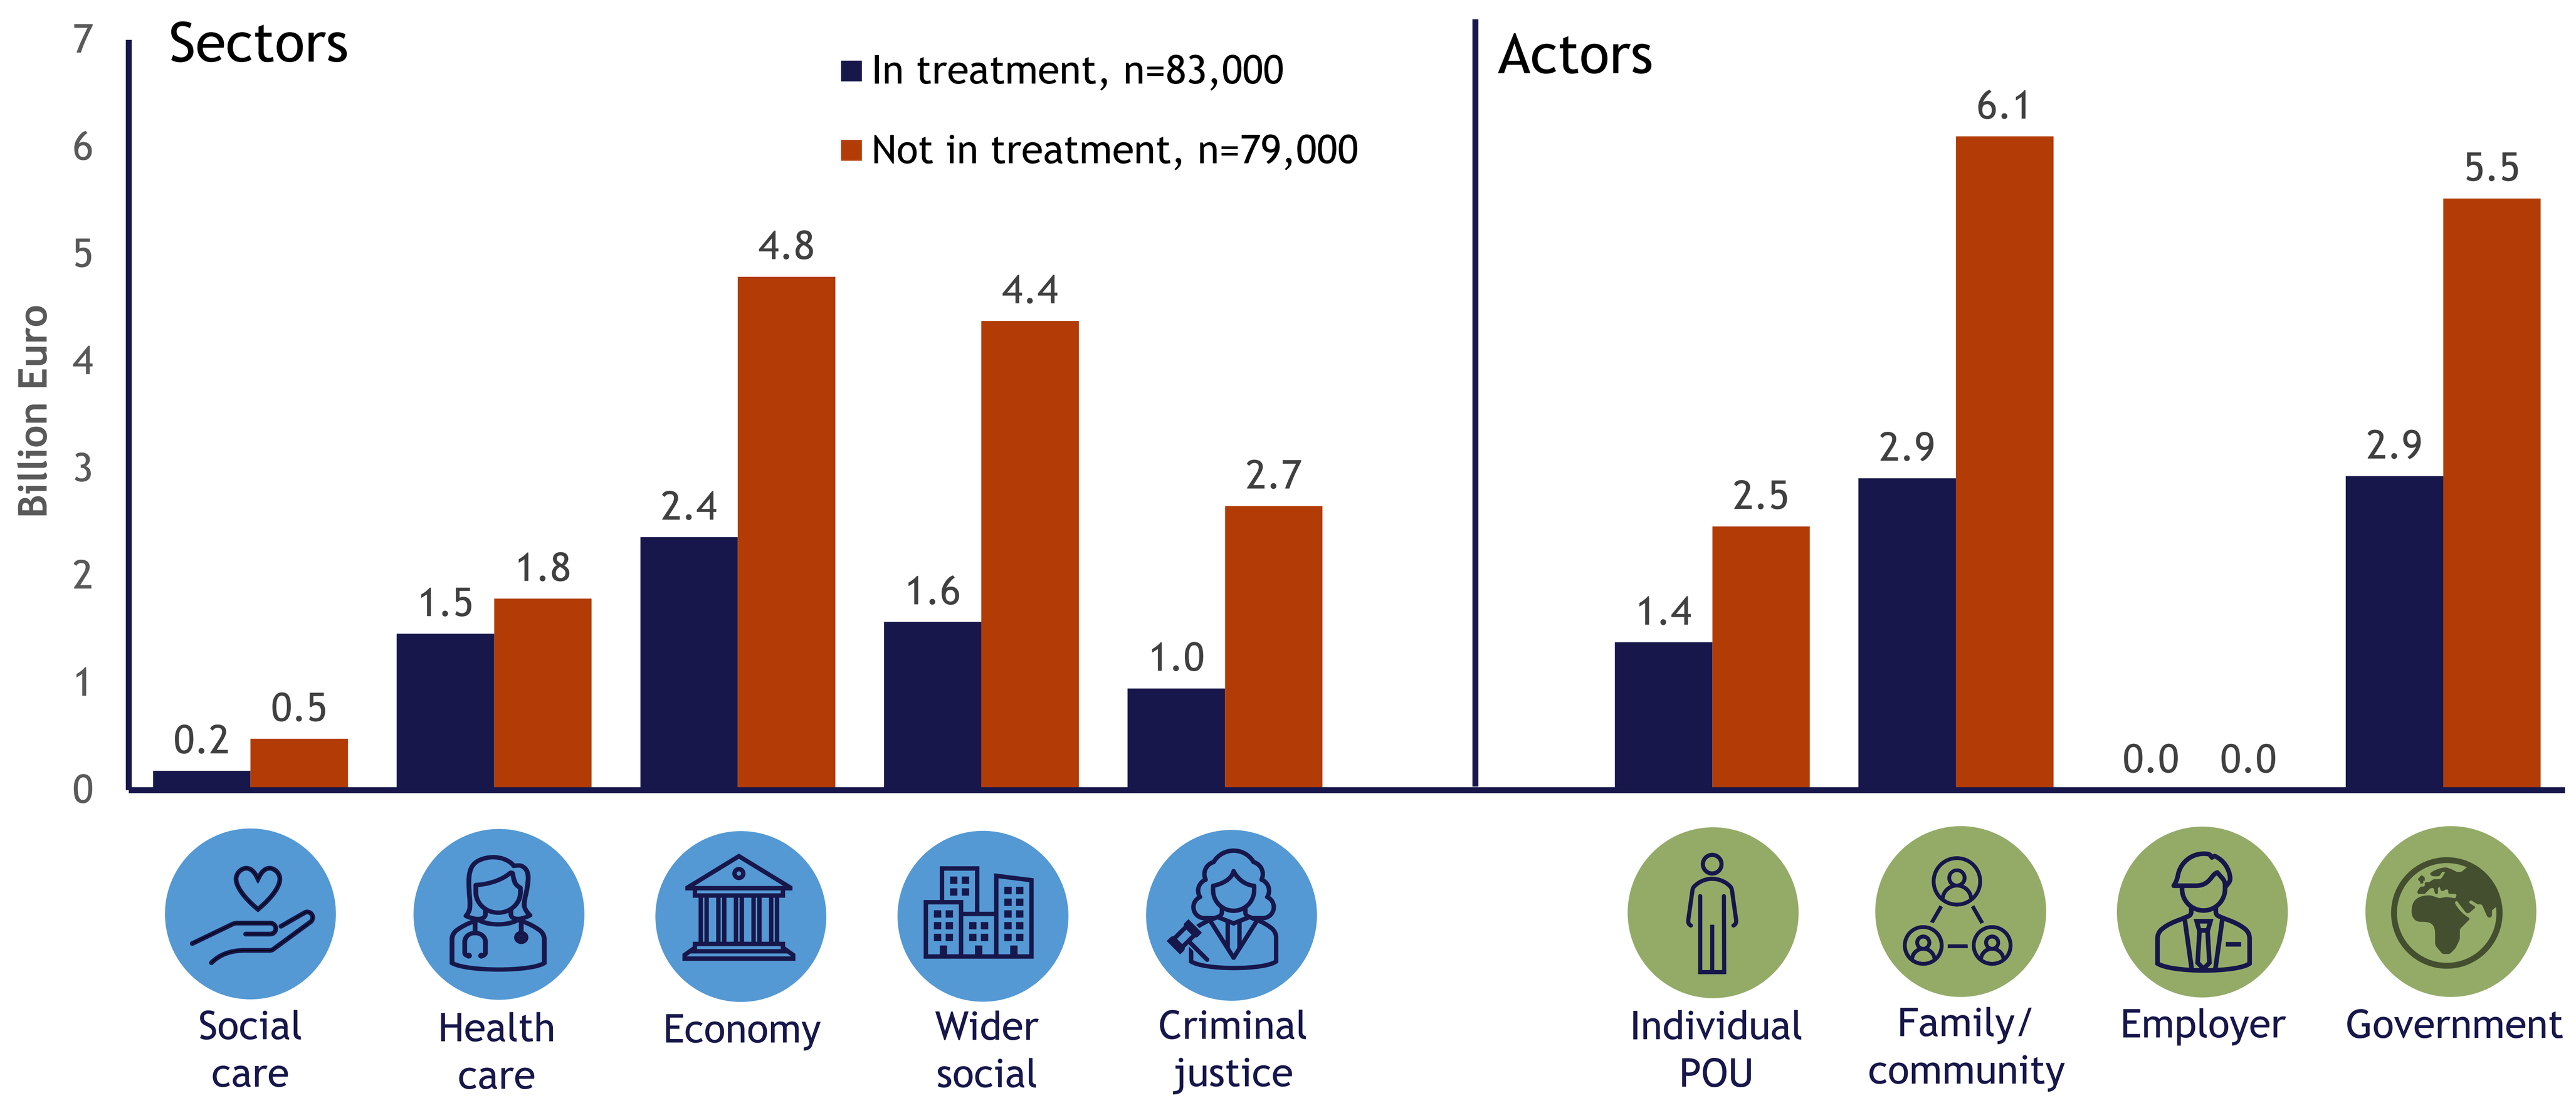
Sectors
Actors
Billion Euro
0
1
2
3
4
5
6
7
In treatment, n=83,000
Not in treatment, n=79,000
0.2
0.5
1.5
1.8
2.4
4.8
1.6
4.4
1.0
2.7
1.4
2.5
2.9
6.1
0.0
0.0
2.9
5.5
Social care
Health care
Economy
Wider social
Criminal justice
Individual POU
Family/ community
Employer
Government

- The total costs of POUs in Germany are €6.5 billion for those in treatment and €14.1 billion for those not in treatment.
- Government, wider social sector and families/communities are the most financially affected by POUs not in treatment.
- Productivity loss associated with POUs not in treatment has major financial implications for the economy.
- The criminal justice sector and government also experience heavy financial costs for POUs not in treatment
- The healthcare and social care sectors have far less costs related to POU compared to other sectors in society

2. Annual savings per POU in treatment

Actors ↓ / Sectors →	Social care	Health care	Economy	Wider social	Criminal justice	Total
Individual POU	-	-	€13,987	-	-	€13,987
Family/ community	-		€14,130	€35,886	-	€50,017
Employer	-	-	€15	-	-	€15
Government	€4,009	€4,618	€3,215	-	€21,705	€33,547
Insurance	-	-	-	-	-	-
Total	€4,009	€4,618	€31,348	€35,886	€21,705	€97,566

- For every POU successfully recruited into treatment programs, German society saves around €100 thousand/year.
- Approximately 37% of these savings fall within the wider social sector, entirely represented by a reduction in costs for families and communities.
- The government also make large saving from the criminal justice system
- Insurance sector costs are zero, as these costs largely fall under German social insurance

CONCLUSIONS

- **The model quantifies savings asssocaited with getting POUs on treatment and maintains previous findings that doing so reduces costs to society and heavily reduces the financial burden on families and communities.**
- **Substantially larger savings occur outside the healthcare sector — a sector that faces heavy treatment costs, and thus has a monetary counterincentive to implementation.**
- **Considerably high savings occur in the criminal justice sector, generating a risk of policy displacement, where criminal justice interventions are more heavily prioritized than healthcare interventions.**
- **Wide-perspective policy is necessary to launch and sustain interventions that increase access to treatment and treatment retention.**

References: **1.** Casey J. Assessing the scale and impact of illicit drug markets in Scotland. Edinburgh: Scottish Government Social Research; 2009; **2.** Wall R, et al. Social costs of untreated opioid dependence. J Urban Health. 2000;77(4):688-722; **3.** Michels, II, et al. Substitution treatment for opioid addicts in Germany. Harm Reduct J. 2007;4:5; **4.** EMCDDA. Statistical Bulletin 2022; **5.** Mattick RP, et al. Methadone maintenance therapy versus no opioid replacement therapy for opioid dependence. Cochrane Database Syst Rev. 2009;2009(3):Cd002209; **6.** Kidorf M, et al. Increasing employment of opioid dependent outpatients: an intensive behavioral intervention. Drug Alcohol Depend. 1998;50(1):73-80; 7. National Institute on Drug Abuse. Medications to Treat Opioid Use Disorder Research Report. NIH; 2021.

† According to The European Monitoring Centre for Drugs and Drug Addiction: "*…injecting drug use or long duration or regular use of opioids, …[]… Existing estimates of problem drug use are often limited to opioid and poly-drug use.*"

For presentation by George Keel at ISPOR Europe, Copenhagen, Denmark,12-15 November 2023. Contact: george.keel@ihe.se