The humanistic and economic burden of illness in progressive fibrosing idiopathic lung disease: a targeted literature review

Istvan Gergo Jozsa[1], Maria Cristina Penaloza Ramos[1], Zoe Marjenberg[2]

[1]Boehringer Ingelheim GmbH, Ingelheim am Rhein, Germany; [2]Maverex Limited, Newcastle, UK

Background

- Progressive fibrosing interstitial lung diseases (PF-ILDs), including idiopathic pulmonary fibrosis (IPF), share common pathophysiologic characteristics and are associated with substantial morbidity and mortality.
- Antifibrotic (AF) therapy aims to manage symptoms and slow disease progression, thereby improving morbidity and life expectancy. However, uptake and adherence are variable.
- An understanding of the burden of PF-ILD in the era of AF therapy is required to inform payers of clinical unmet needs.

Objectives

- To identify the humanistic burden (health-related quality of life [HRQoL] and economic burden (costs and healthcare resource use [HCRU] and indirect economic burden e.g., work productivity) of PF-ILD and IPF in the era of AF therapy.
- To assess the impact of comorbidities, exacerbations, disease subtype, diagnosis delay, initiation of treatment, and treatment discontinuation/dose reduction on humanistic and economic burdens.

Methods

- A pragmatic, targeted literature review was performed to identify relevant and high-impact papers.
- Electronic databases (Embase, MEDLINE, the Cochrane Library) were searched using keywords related to IPF, PF-ILD, antifibrotics, prevalence, HRQoL, HRCU, costs, and other related keywords in the search strategy. Hand-searching (free text searches and reviewing the reference lists/citation tracking of relevant studies) was also performed.

Table 1: Eligibility criteria:

Population	Patients with progressive pulmonary fibrosis including PF-ILD and IPF
Outcomes	Humanistic burden (PROs/HRQoL) for patients and caregivers, economic burden (HCRU, costs) relating to management of patients and indirect costs (e.g., work impairment)
Study design	Non-interventional (observational) studies, RCTs, systematic literature reviews, economic evaluations
Dates published	2017-2022
Countries	France, Germany, Spain, Italy, UK, Canada, USA, Australia, China, and Japan

Results

Humanistic burden

- Fifteen studies were identified; 11 for IPF (7 observational, 4 RCTs) and four (4 RCTs) for PF-ILD (**Table 2**)

Table 2: Humanistic burden of PF-ILD and IPF

Study	PF-ILD/ IPF	Study design and data source	HRQoL instrument	HRQoL reported
Wuyts 2022[1]	IPF	Observational: AIPFR	SGRQ, EQ-5D-5L, EQ-5D VAS, Cough VAS	• HRQoL change over time: worsening in some domains, stable in others
Lancaster 2022[2]	IPF	Observational: Adelphi IPF II DSP 2019 database	EQ-5D-3L, EQ-5D VAS	• HRQoL decreases with increasing disease severity
Hoyer 2022[3]	IPF	Observational: PFBIO cohort	SGRQ, SGRQ-I_{der}, COPD-CAT	• HRQoL change over time: improvement at 6 months, but deteriorated at 12-months • HRQoL significantly worse in patients with diagnostic delay of >1 year vs <1 year
Cox 2021[4]	IPF	Observational: AIPFR	EQ-5D-5L	• Mobility and usual activities impairment, pain/discomfort, and anxiety/depression common • Increasing disease severity and number of comorbidities (>2) significantly associated with a reduction in HSUVs estimated from HRQoL scores • AF use non-significantly associated with higher HSUV estimated from HRQoL scores
Salisbury 2020[5]	IPF	Observational: IPF-PRO registry	SF-12 (MCS and PCS), CASA-Q, EuroQoL, EuroQoL-VAS	• Worsening HRQoL significantly associated with increased odds of being treated with AF
Holtze 2020[6]	IPF	Observational: PFF-PR registry	Fatigue severity score, LCQ, SF-6D, UCSD-SOBQ	• AF-treated patient group had non-significantly poorer HRQoL at baseline compared to non-treatment group
Kreuter 2017[7]	IPF	Observational: INSIGHTS-IPF registry	UCSD-SOBQ, SGRQ, WHO-5, EQ-5D	• Activity impairment and depression common • HRQoL significantly decreases with increasing disease severity and > 2 comorbidities
Glaspole 2021[8] (pooled data from 5 trials)	IPF	RCT: Nintedanib vs placebo trials	SGRQ	• HRQoL decreased over 52 weeks • HRQoL worsened more in patients with ≥5 comorbidities than in those with <5
Kreuter 2020[9] (INPULSIS)	IPF	RCT: Nintedanib vs placebo	SGRQ, UCSD-SOBQ, CASA-Q, EQ-5D-VAS	• Advanced disease and acute exacerbations associated with significant worsening of HRQoL
Moor 2020[10] (NCT03420235)	IPF	RCT: Standard care plus home monitoring vs standard care (all receiving AF)	K-BILD, EQ-5D-5L, HADS EQ-VAS, VAS, GRC	• HRQoL improved over 24 weeks, although anxiety and depression scores remained stable
NCT05321069	IPF	RCT: BI 1015550 (low and high dose) vs placebo	L-PF	• None (trial ongoing)
NCT05321082	PF-ILD	RCT: BI 1015550 (low and high dose) vs placebo	L-PF	• None (trial currently recruiting)
Flaherty 2019[11] (NCT02999178; INBUILD)	PF-ILD	RCT: Nintedanib vs placebo	K-BILD, L-PF	• No difference in HRQoL at 52 weeks compared to baseline
Behr 2021[12] (EudraCT 2014-000861-32; RELIEF)	PF-ILD	RCT: Pirfenidone vs placebo	SGRQ	• No difference in HRQoL at 52 weeks compared to baseline
Maher 2018[13] (NCT03099187)	PF-ILD	RCT: Pirfenidone vs placebo	UCSD-SOBQ, LCQ, cough VAS, SGRQ	• No difference in HRQoL at 24 weeks compared to baseline

Abbreviations: Australian IPF registry (AIPFR); Cough And Sputum Assessment Questionnaire (CASA-Q); Chronic Obstructive Pulmonary Disease Assessment Test (COPD-CAT); Disease Specific Programme (DSP); EuroQol-5 dimensions-3 levels/5 levels (EQ-5D-3L/5L); Global Rating of Change (GRC); Hospital Anxiety and Depression Scale (HADS); Investigating Significant Health Trends in Idiopathic Pulmonary Fibrosis (INSIGHTS-IPF); Idiopathic Pulmonary Fibrosis-PRospective Outcomes (IPF-PRO); King's Brief Interstitial Lung Disease (KBILD); Leicester Cough Questionnaire (LCQ); Living with Pulmonary Fibrosis (L-PF); Pulmonary Fibrosis Biomarker (PFBIO); Pulmonary Fibrosis Foundation Patient Registry (PFF-PR); Short form-6 dimensions (SF-6D); Short form 12 mental and physical component score (SF-12 MCS and PCS); St. George's Respiratory Questionnaire (SGRQ); University of California, San Diego Shortness of Breath Questionnaire (UCSD-SOBQ); Visual Analogue Scale (VAS); World Health Organisation- Five Well-Being Index (WHO-5)

HRQoL assessment and burden

16 different PROs used to assess HRQoL in patients

Activity & mobility were reported to be particularly affected (2 studies)[4,7]

The most commonly utilised PROs were SGRQ, UCSD-SOBQ, EQ-5D and variants, HADS, and K-BILD

Whilst these PRO are used in clinical trials the validity and appropriateness was outside the scope of this study

Depression was also demonstrated as common with two studies reporting almost half of patients showing depression symptoms[4,7]

Impact of disease severity and exacerbations

4 studies — Increasing disease severity was associated with a deterioration in HRQoL[2,4,7,9] and was reported significant (p<0.05) by three of these studies[4,7,9]

1 study — One study reported that acute exacerbations were associated with significantly poorer HRQoL (p<0.05)[9]

Impact of comorbidities

3 studies — Increasing numbers of comorbidities were associated with worsening in HRQoL[4,7,8]

2 observational studies — Reported a significant (p<0.05) reduction in HRQoL (or health state utility values [HSUVs] estimated from HRQoL scores) with 2 or more comorbidities[4,7]

Diagnosis

Early diagnosis is important for reducing the humanistic burden

Patients with a >1 year delay in diagnosis had consistently worse HRQoL compared to a delay of <1 year[3]

- There is a lack of published data on caregiver burden and the humanistic impact of different IPF subtypes

Economic burden

- Eleven studies were identified: 10 for IPF (9 observational, 1 RCT) and one for PF-ILD (1 RCT) (**Table 3**).
- Studies were identified for Europe, USA, Israel, and Australia.

Table 3: Economic burden of PF-ILD and IPF

Study	PF-ILD/IPF	Study design description	Economic burden reported
Wuyts 2022[1]	IPF	Observational: PROOF registry	HCRU at follow-up
Lancaster 2022[2]	IPF	Observational: Adelphi IPF II Disease Specific Programme (DSP 2019 database)	HCRU in past 12 months, impact of disease severity, work impairment
Cox 2022[14]	IPF	Observational: Australian IPF registry (AIPFR)	HCRU in past 12 months, direct costs incurred in past 12 months, impact of disease severity and comorbidities on direct costs in past 12 months
Singer 2022[15]	IPF	Observational: Optum Research Database administrative claims	Direct costs incurred in past 12 months, impact of AF treatment delay on hospitalisation risk
Corral 2020a[16]	IPF	Observational: Medicare beneficiaries	Monthly HCRU, monthly direct costs
Corral 2020b[17]	IPF	Observational: IBM MarketScan databases (Commercial, Medicare, Early View)	Direct costs incurred 12 months post-index, HCRU in 12 months post-index, HCRU post-index
Dempsey 2019[18]	IPF	Observational: OptumLabs data warehouse administrative claims	Impact of AF therapy on risk of acute hospitalisation
Nagar 2018[19]	IPF	Observational: administrative claims	Monthly direct costs
Blanc 2020[20]	IPF	Observational: retrospective cohort	HCRU per year at end of treatment period
Moor 2020[10] (NCT03420235)	IPF	RCT: Standard care plus home monitoring vs standard care (all receiving AF)	HCRU at 24-week follow-up
Maher 2018[13] (NCT03099187)	PF-ILD	RCT: Pirfenidone vs placebo	HCRU at 24-week follow-up

Healthcare resource use

Hospitalisation is common in IPF patients

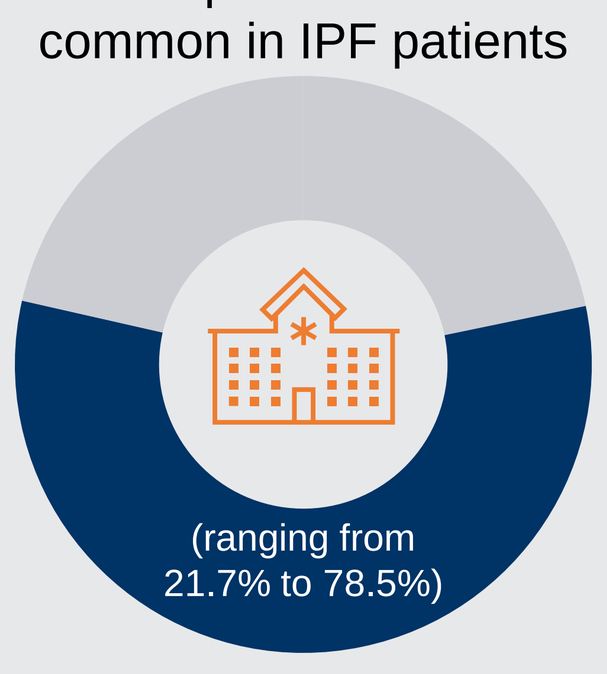

(ranging from
21.7% to 78.5%)

Patients frequently use other healthcare resources such as:

Outpatient services

GPs

Pharmacy services

Work impairment

Impairment of ability to work in IPF[2]

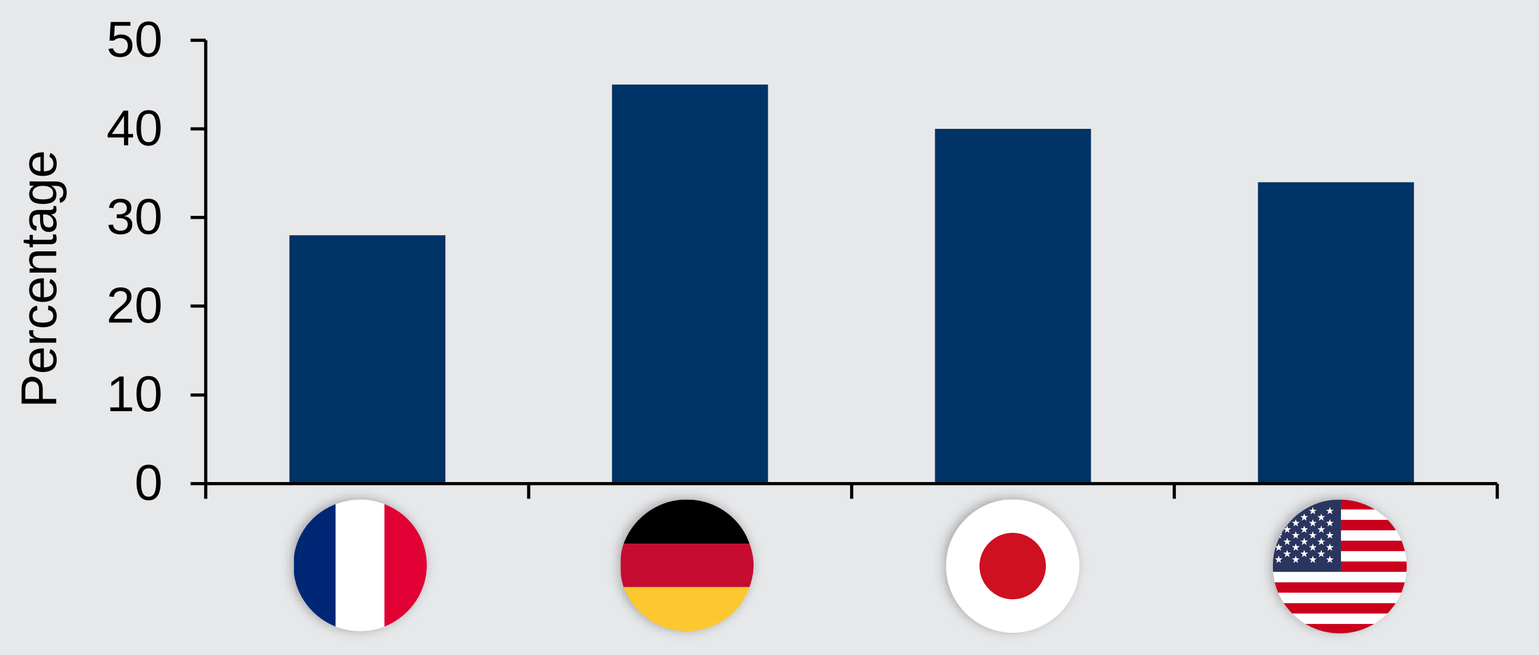

Percentage
50
40
30
20
10
0

Direct costs

1 study[14]

Total project annual cost based on prevalence estimate: $299 M

Yearly mean costs per patient: **AUD $31,655**

3 studies[15–17]

Mean total healthcare costs for AF-treated IPF[16,17]

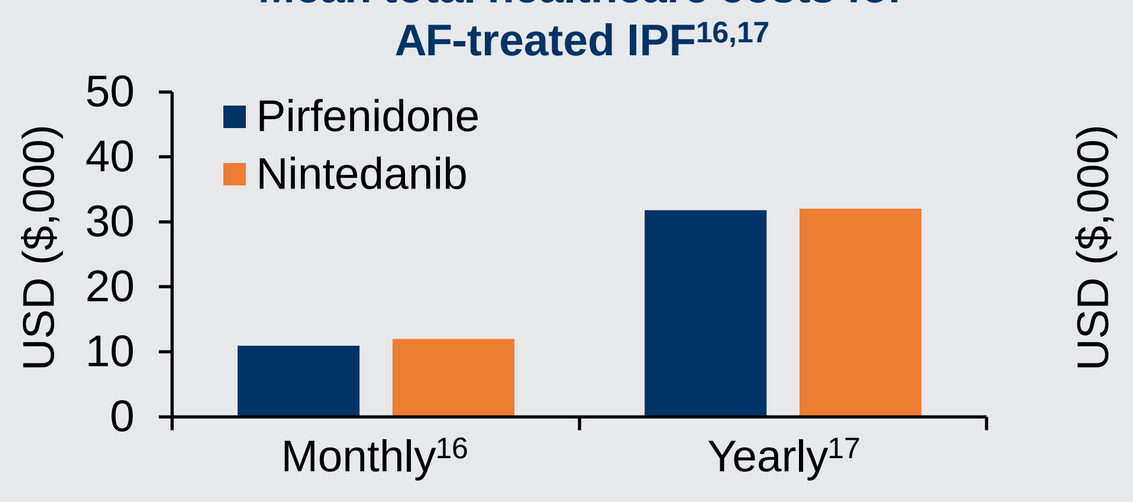

USD ($,000)
50
40
30
20
10
0
Pirfenidone
Nintedanib
Monthly16
Yearly17

12-month medical costs are higher when AF initiation is delayed[15]

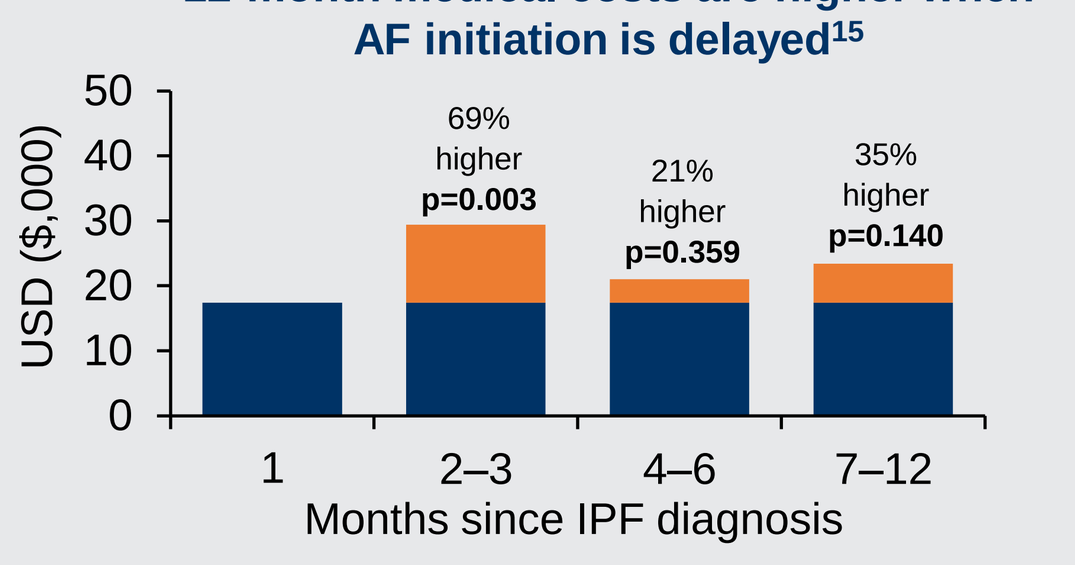

USD ($,000)
69% higher p=0.003
21% higher p=0.359
35% higher p=0.140
1
2–3
4–6
7–12
Months since IPF diagnosis

AF medications are a large cost driver of IPF direct costs

Disease severity and presence of comorbidities

2 studies — IPF disease severity and presence of comorbidities significantly (p<0.05) increases direct costs[14]

Increasing IPF disease severity increases hospitalisation risk (significance not reported), although hospitalisation is still common in mild functional impairment[2]

Diagnosis and initiation of AF treatment

1 study — Delays between IPF diagnosis and AF treatment initiation raises hospitalisation risk and all-cause medical costs[15]

Hospitalisation risk of patients untreated in months after diagnosis compared to treated patients

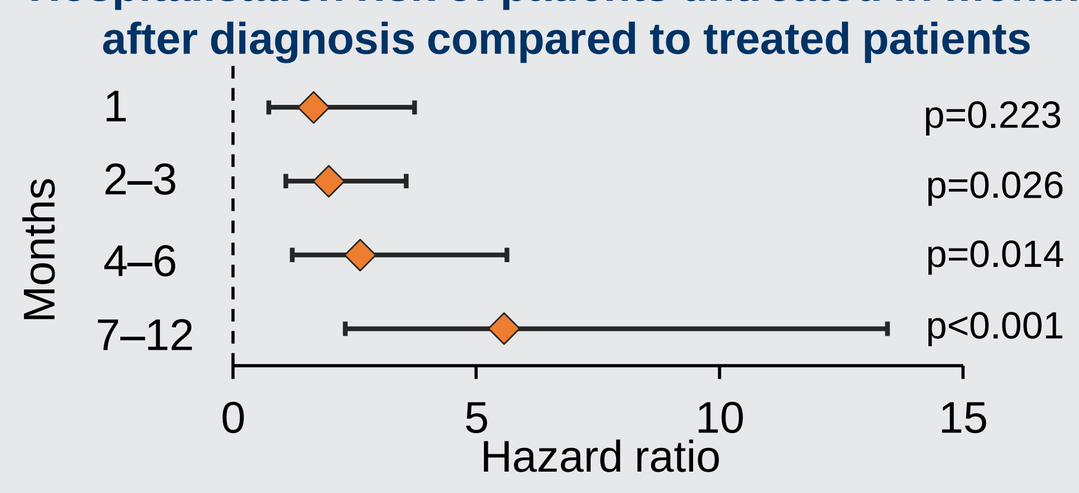

Months
1
2–3
4–6
7–12
p=0.223
p=0.026
p=0.014
p<0.001
0
5
10
15
Hazard ratio

- No studies were identified reporting:
 - Differences between treatment-naïve and AF-treated patients
 - Differences between stable and progressing patients treated with AF

Conclusions

Humanistic burden

- HRQoL is impaired for many IPF patients in the antifibrotic era, with studies indicating particular problems with mobility, activity, pain or discomfort, and anxiety or depression. Increasing comorbidities, exacerbations, and severity of disease are associated with a worsening of HRQoL.
- A number of different PROs for capturing HRQoL are used in PF-ILD and IPF studies; future studies should consider which PROs are capturing the most important aspects from a patient's point of view. Further research may be required into the most suitable PRO to capture the salient aspects of the disease.

Economic burden

- There is a lack of studies assessing the economic impact of PF-ILD, and costs and healthcare resource use in IPF are only published for Australia and USA.
- Delays between disease diagnosis and start of AF treatment significantly increase medical costs and healthcare resource use, emphasising the importance of prompt initiation of AF therapy.

References

1. Wuyts et al. *Pulm Ther.* 2022; 8(2):181–94
2. Lancaster et al. *Respirology.* 2022; 27(1):66
3. Hoyeret al. *BMJ Open Respir Res.* 2022; 9(1):e001276
4. Cox et al. *Qual Life Res.* 2021; 30(9):2615–32
5. Salisbury et al. *Ann Am Thorac Soc.* 2020; 17(11):1413–23
6. Holtze et al. *Respir Res.* 2020; 21(1)
7. Kreuter et al. *Respir Res.* 2017; 18(1):1–10
8. Glaspole et al. *Respir Res.* 2021; 22(1)
9. Kreuter et al. *Respir Res.* 2020; 21(1):1–10
10. Moor et al. *Am J Respir Crit Care Med.* 2020; 202(3):393–401
11. Flaherty et al. *N Engl J Med.* 2019; 381(18):1718–27
12. Behr et al. *Lancet Respir Med.* 2021; 9(5):1902279
13. Maher et al. *BMJ Open Respir Res.* 2018; 5(1)
14. Cox et al. *Euro J Health Econ.* 2022; 1:1–19
15. Singer et al. *J Med Econ.* 2022; 25(1):532–40
16. Corral et al. *J Comp Eff Res.* 2020a; 9(13):933–43
17. Corral et al. *BMC Pulm Med.* 2020b; 20(1)
18. Dempsey et al. *Am J Respir Crit Care Med.* 2019; 200(2):168–74
19. Nagar et al. *J Manag Care Pharm.* 2018; S71
20. Menárguez Blanc et al. *Euro J Hosp Pharm.* 2020; 27(Suppl 1):A197

Abbreviations

Antifibrotics (AF); general practitioners (GPs) healthcare resource use (HCRU); health-related quality of life (HRQoL); health state utility values (HSUVs); idiopathic pulmonary fibrosis (IPF); progressive fibrosing interstitial lung diseases (PF-ILD); randomised controlled trial (RCT).

Sponsorship

The study was supported and funded by Boehringer Ingelheim International Gmbh

Conflict of interest statement

- The authors meet criteria for authorship as recommended by the International Committee of Medical Journal Editors (ICMJE).
- The authors did not receive payment related to the development of the poster.
- Istvan Gergo Jozsa and Maria Cristina Penaloza Ramos are employees of Boehringer Ingelheim.
- BI was given the opportunity to review the manuscript for medical and scientific accuracy as well as intellectual property considerations.

ISPOR Europe, Copenhagen, Denmark; 12 – 15 November 2023

Scan QR code or visit URL for a device-friendly version of this poster including slides

URL: https://bit.ly/3PRmS3V

Scan QR code or visit URL for a webpage featuring all BI-supported publications at ISPOR Europe 2023

URL: https://bit.ly/3Lzks8n