MSR139

Performance of Different Survival Models for Extrapolation of Immature OS Data for 2L+ NSCLC Therapies

Jacob J[1], Vandoulakis M[2], Klint J[3]

[1]IQVIA, London, UK, [2]IQVIA, Athens, Greece, [3]Daiichi Sankyo, Munich, Germany

Introduction

- Overall survival (OS) is a key endpoint to evaluate the cost-effectiveness of oncology therapies.
- For cancer therapies that lead to durable responses and prolonged survival in a subset of patients the potential improvement in OS may only become apparent with longer trial follow-up and large trial sample sizes.
- Long-term extrapolation of immature OS data constitutes a source of high uncertainty, and it is among the cost-effectiveness model components under higher scrutiny by HTA bodies and payers.
- Mean OS provides a measure of how each survival model predicts OS across for the relevant time horizon for cost-effectiveness analyses.
- A retrospective study of published short-term versus corresponding long-term OS data available in the public domain was conducted in 2L+ NSCLC to assess the impact of survival model selection for long-term extrapolations on mean OS estimates.

Objective

Assess the implications of OS data maturity and survival model selection on mean OS

Methods

- A targeted review of clinical trials of monotherapies in 2L+ NSCLC identified 12 studies (with 20 populations) reporting OS with short and long-term follow-up.
- OS Kaplan-Meier curves for each of the population were extracted from the available publications and digitized to generate two datasets for each population (2x20) (**Table 1**).
- 6 conventional parametric models (exponential, Weibull, Gompertz, log-normal, log-logistic, generalized gamma) and 3 spline models (hazard, odds, normal with 3 knots) were fitted to the reconstructed IPD data to extrapolate OS for a lifetime horizon and estimate mean OS.
- All survival extrapolations were adjusted for UK general population mortality. Mean OS was estimated over a lifetime time horizon (i.e., less than 1% survival)
- Survival models were ranked based on goodness-of-fit was based on the Akaike information criterion (AIC) (i.e., the model with lowest AIC was ranked as best fitting model)

Table 1: List of populations included in the analyses

Trial	Therapy	Number of patients
ATLANTIC Cohort 1 (EGFR+/ALK+)	Durvalumab	107
ATLANTIC Cohort 2 (EGFR−/ALK−)	Durvalumab	243
LUX-Lung 8	Afatinib; Erlotinib	398 + 397
POPLAR	Atezolizumab; Docetaxel	144 + 143
KEYNOTE-010	Pembrolizumab; Docetaxel	690 + 343
OAK	Atezolizumab; Docetaxel	613 + 612
CheckMate 017	Nivolumab; Docetaxel	135 + 137
CheckMate 057	Nivolumab; Docetaxel	292 + 290
Pooled CheckMate 017 & 057	Nivolumab; Docetaxel	427 + 427
KEYNOTE-001	Pembrolizumab	449
Pooled NP28673 & NP28761	Alectinib	225
ALTA	Brigatinib	110
AURA	Osimertinib	201

Results

- Results showed an increase in mean OS with longer follow-up, regardless of the choice of survival model. The overall average percentage change in mean OS for short-term versus long-term follow-up was 23%. Log-normal (2%) and log-logistic (5%) were less prone to underestimate OS extrapolated from shorter follow-up datasets. In turn, Gompertz showed the highest increase in mean OS (78%) (**Figure 1**).
- In 19/20 comparisons, mean OS increased with longer follow-up data, with the average change across all survival models ranging between -2% and 65%. An increase in mean OS was observed for all therapy classes: immunotherapy (28%), targeted therapy (25%) and chemotherapy (docetaxel; 14%) (**Figure 2**).
- In 11/20 populations, all survival models tested resulted in an increase in mean OS with longer follow up data. There were only 26/177 survival models that resulted in a decrease in mean OS. (**Figure 3**).
- Considering only the best fitting survival models (based on AIC), the increase in mean OS was higher than the average across all survival models for 14/20 populations (**Figure 4**).
- Average AIC ranking of each survival model did not show a clear correlation with the % change in mean OS (i.e., best fitting models did not result in more consistent mean OS estimates between shorter and longer follow-up data) (**Figure 5**).

Conclusions

- Mean OS estimates from extrapolated data consistently increased with longer follow-up data for all included survival models. An increase in mean OS was observed for all three therapy classes. Chemotherapy showed a smaller increase compared with immunotherapy and targeted therapy, which can be partially explained by the highest level of maturity of the shorter follow-up datasets compared with the other therapy classes.
- Across all populations, log-logistic and log-normal were associated with more consistent results between the short- and long-term datasets. Gompertz resulted in a substantially greater increase than all the remaining models, due mainly to the overestimation of OS in the longer follow-up datasets. This can be explained by the characteristics of the Gompertz distribution (monotone hazard rates that either increase or decrease exponentially with time) which fitted curves with hazards tending to zero in several long-term datasets.
- Goodness of fit based on AIC was not a good guide to reduce underestimation of OS and was not a good guide to the extent of increase in mean OS for shorter vs longer follow-up data
- The differences observed with longer follow-up emphasize the need to complement survival models with external data from other trials, registries or expert elicitation when extrapolating immature data for cost-effectiveness analyses.

Figure 1: % change in mean OS for short- versus long-term follow-up data – Average across all populations

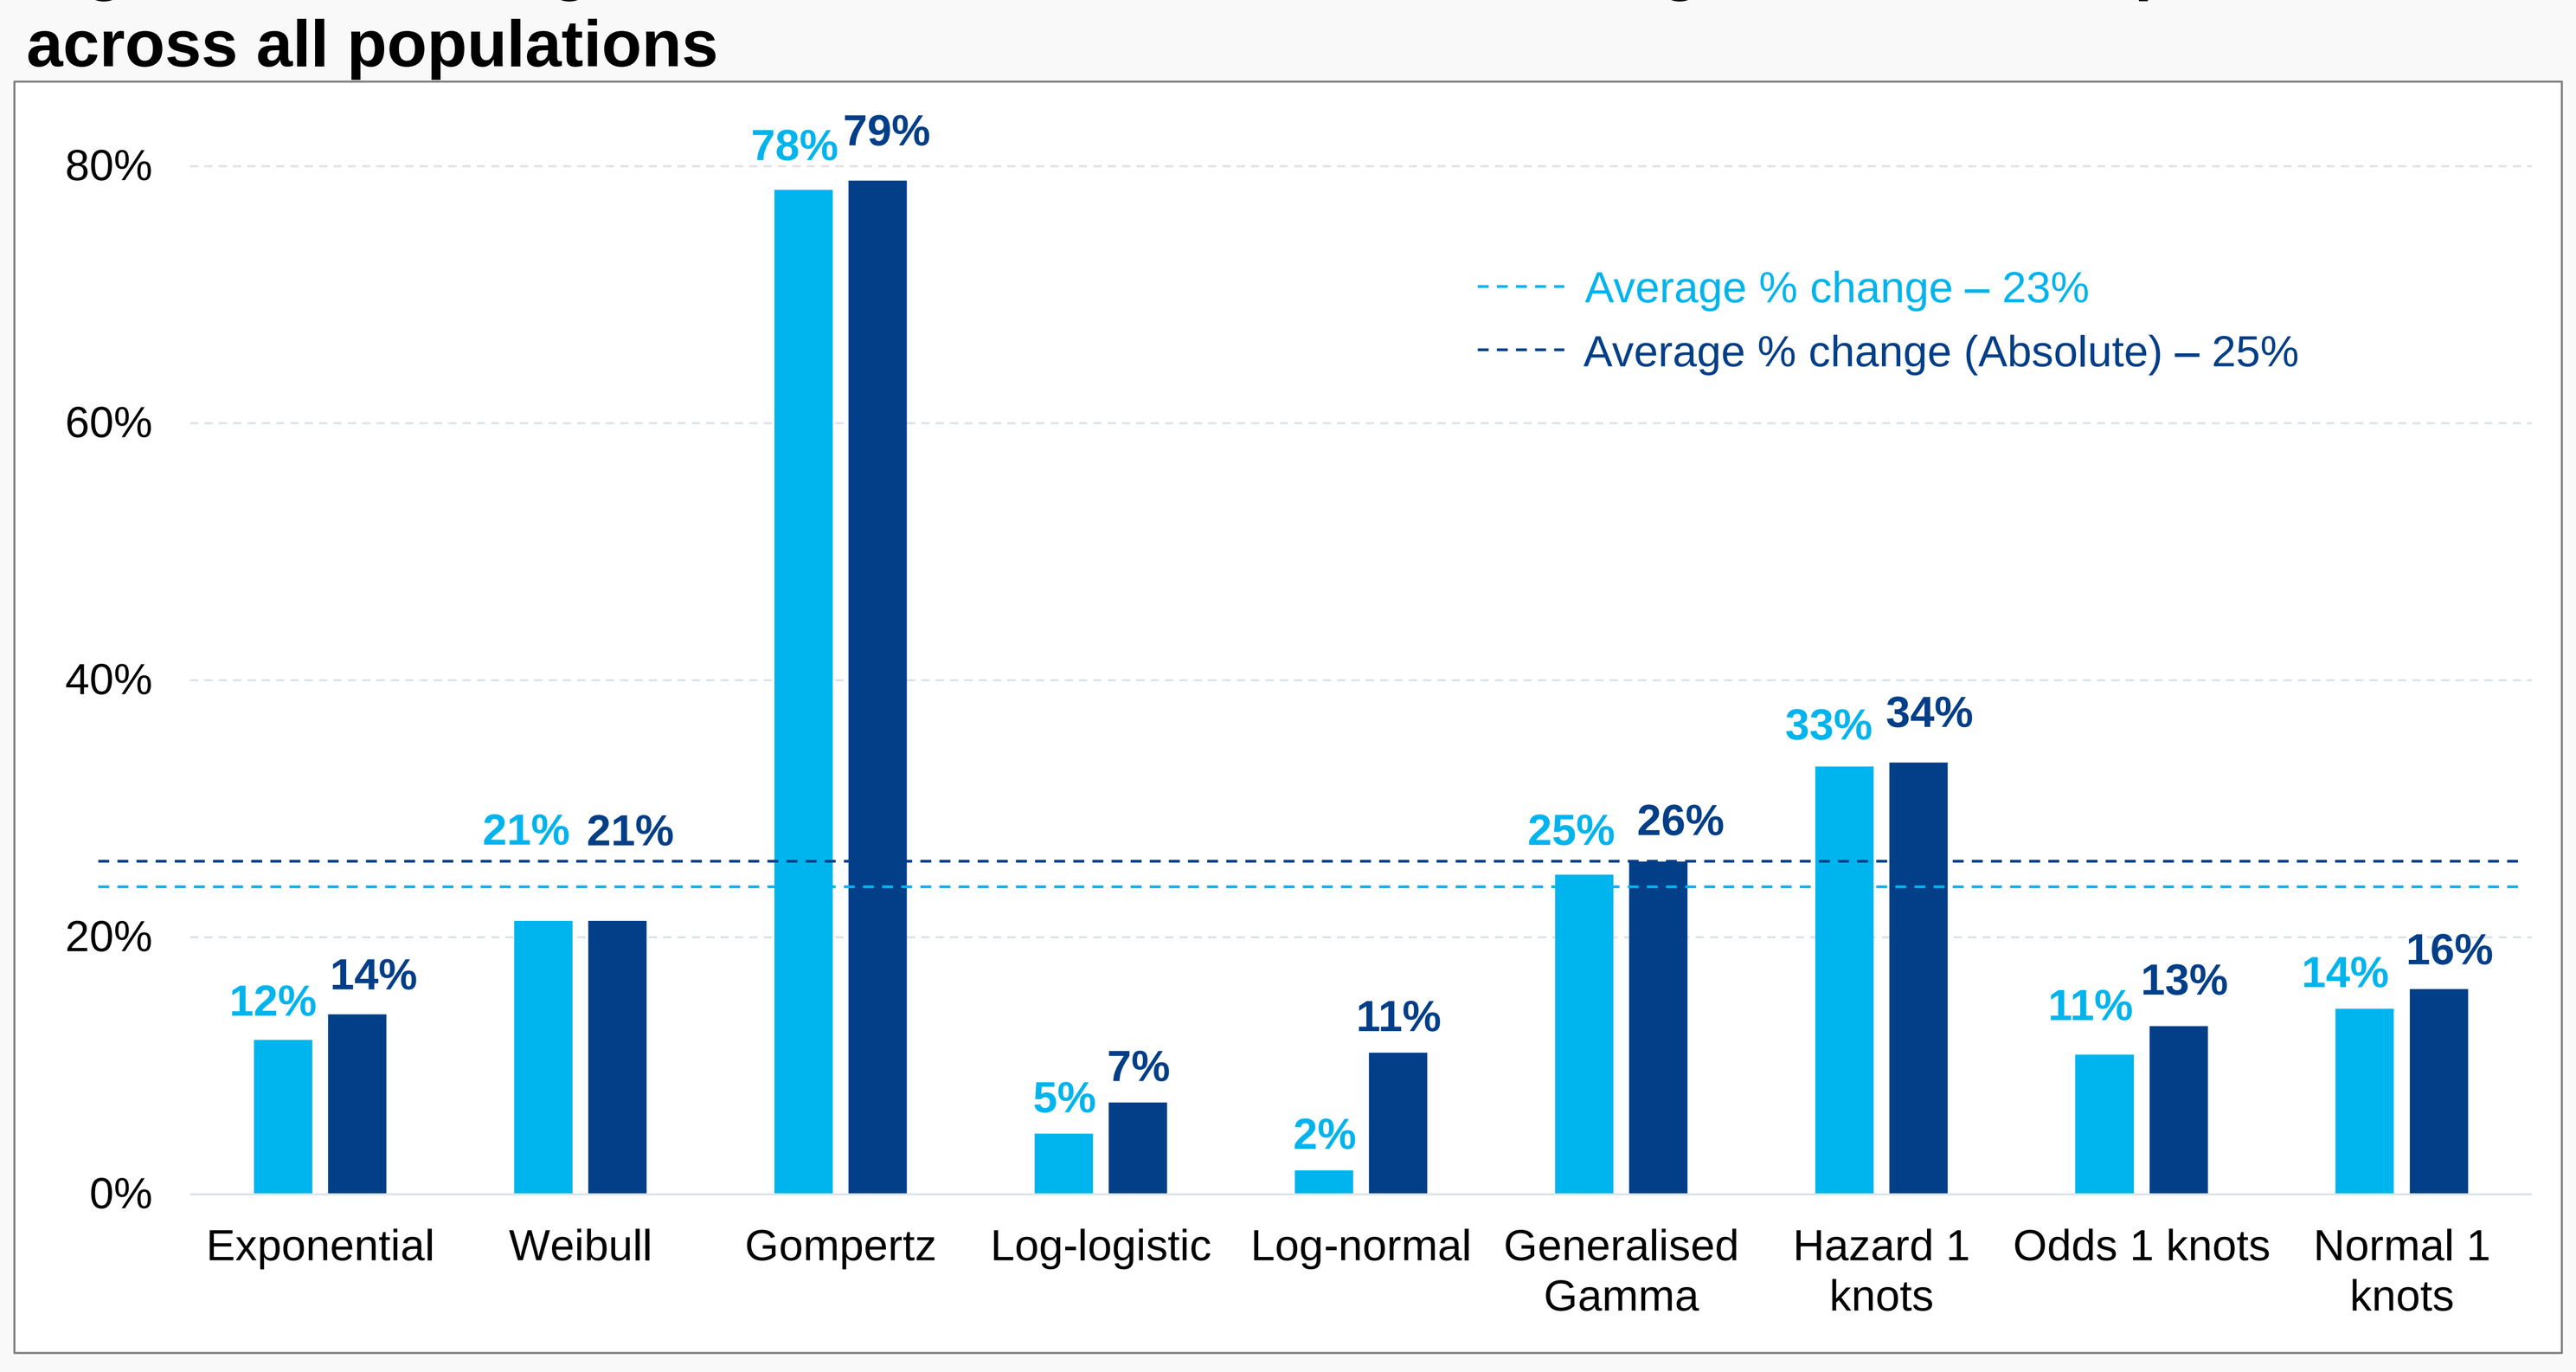
80%
60%
40%
20%
0%
Average % change – 23%
Average % change (Absolute) – 25%
12% 14%
21% 21%
78% 79%
5% 7%
2% 11%
25% 26%
33% 34%
11% 13%
14% 16%
Exponential
Weibull
Gompertz
Log-logistic
Log-normal
Generalised Gamma
Hazard 1 knots
Odds 1 knots
Normal 1 knots

Figure 2: % change in mean OS for short- versus long-term follow-up data by population – Average across all survival models

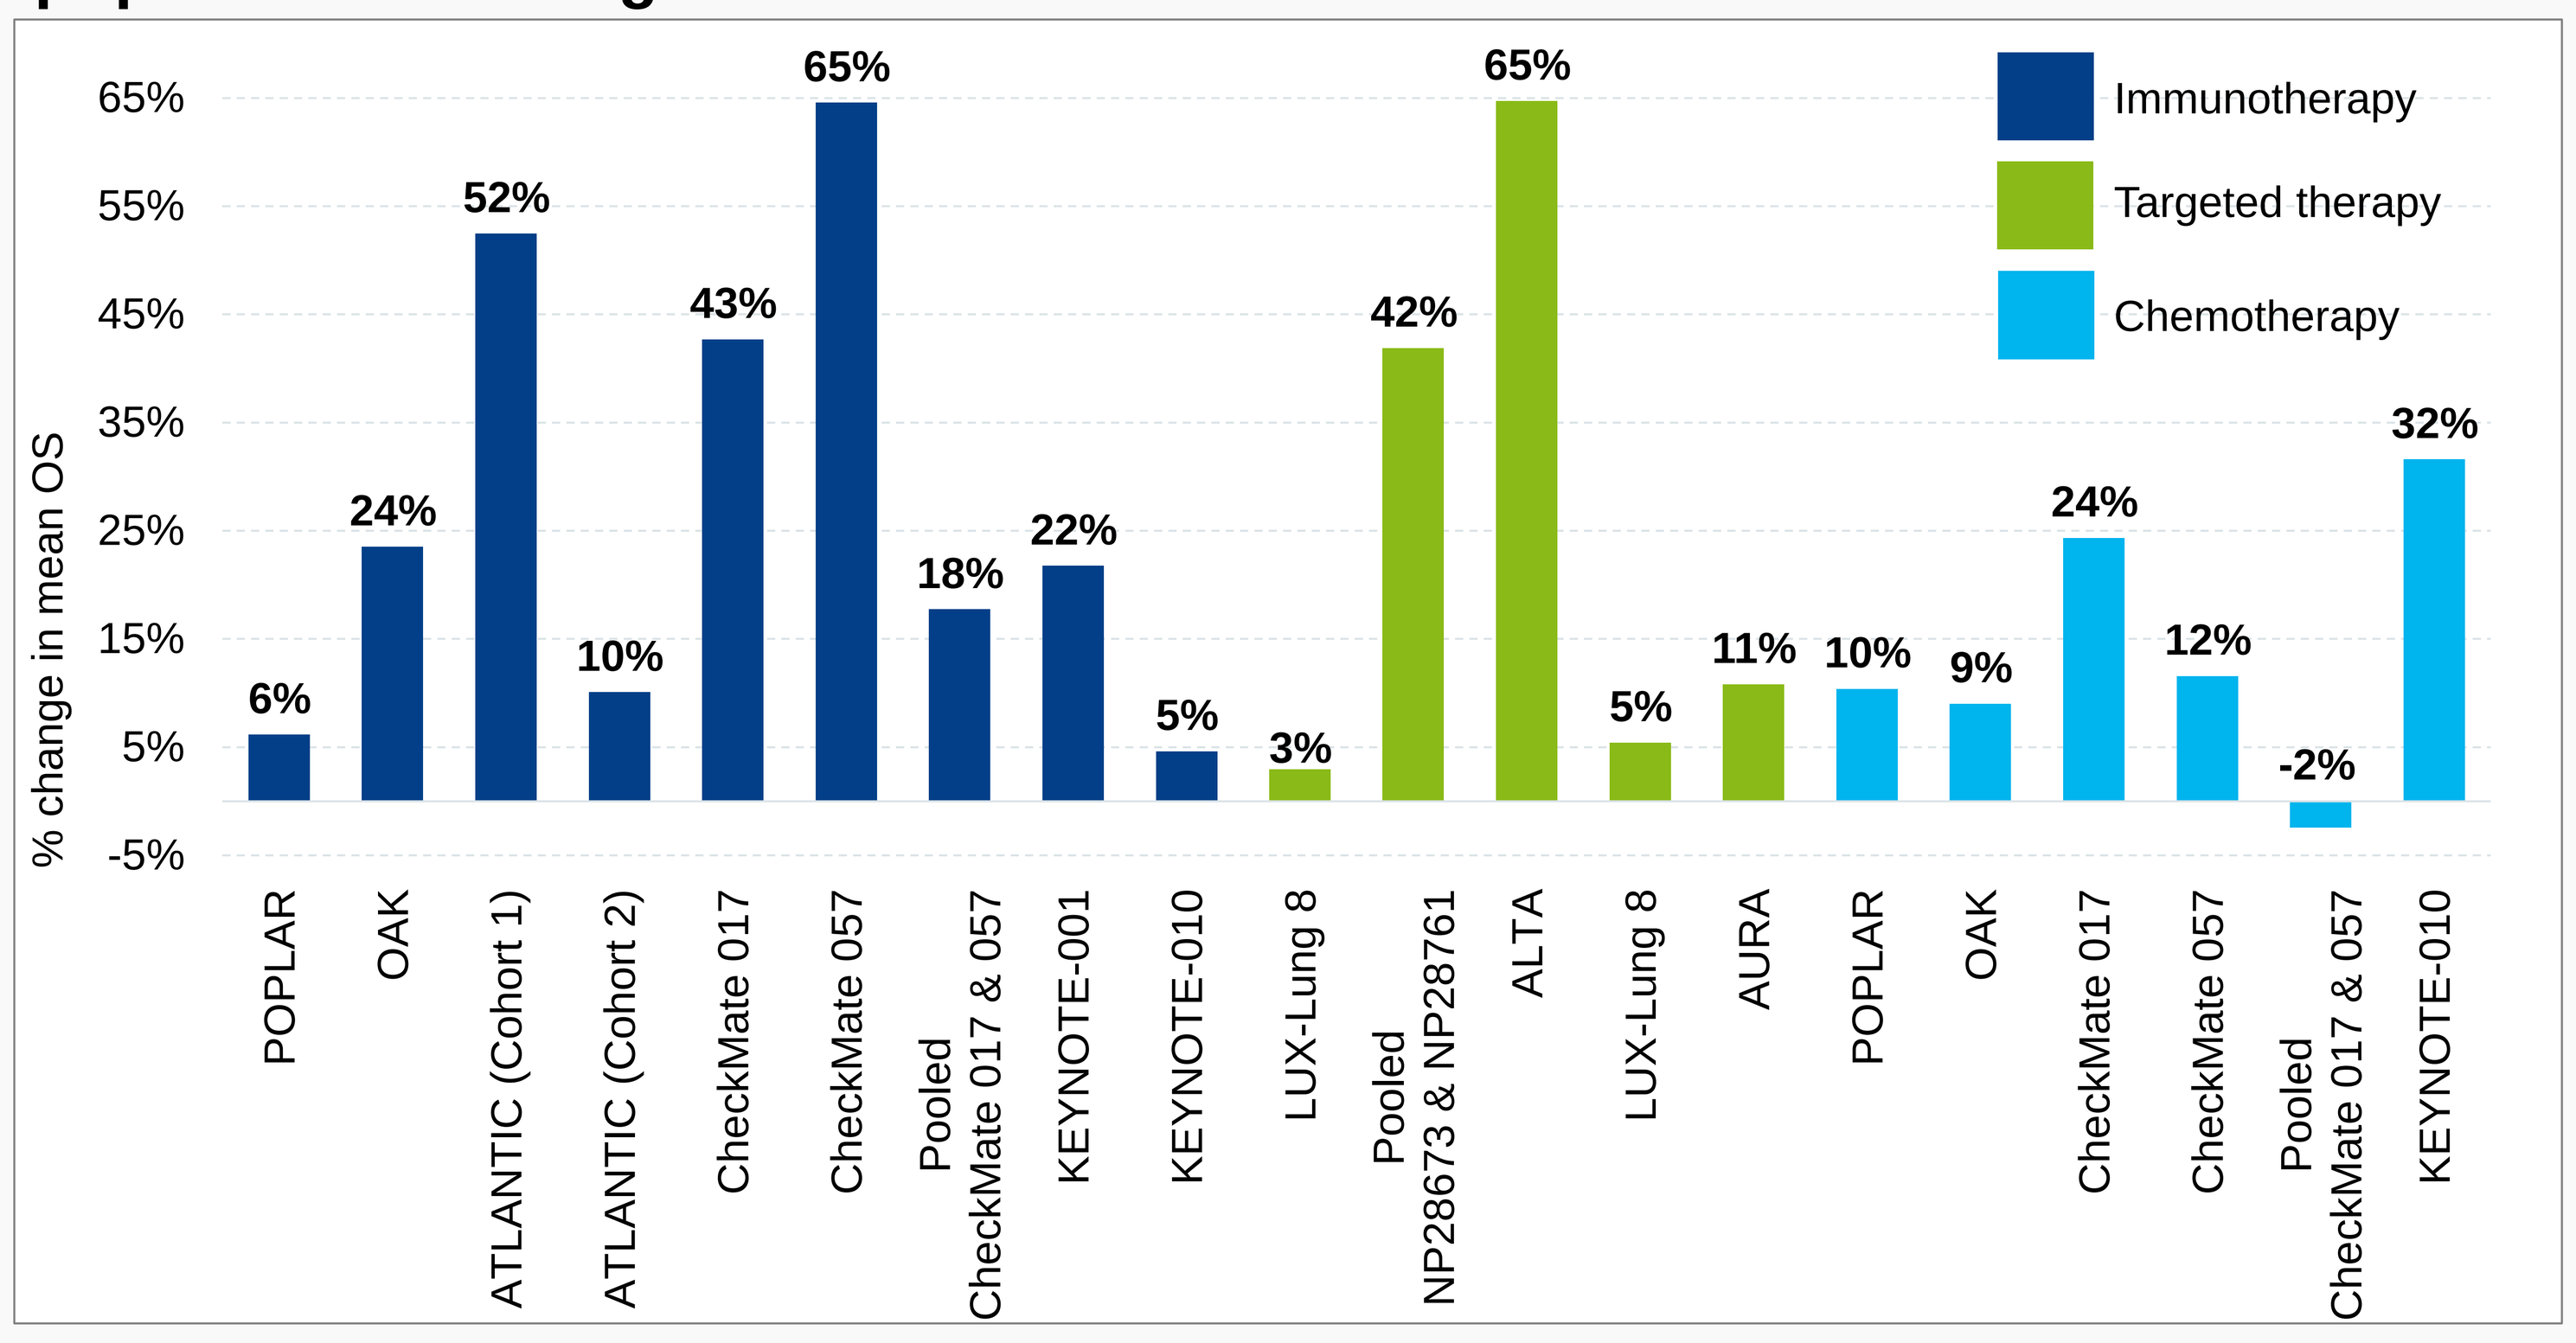
% change in mean OS
Immunotherapy
Targeted therapy
Chemotherapy
6% 24% 52% 10% 43% 65% 18% 22% 5% 3% 42% 65% 5% 11% 10% 9% 24% 12% -2% 32%
POPLAR
OAK
ATLANTIC (Cohort 1)
ATLANTIC (Cohort 2)
CheckMate 017
CheckMate 057
Pooled CheckMate 017 & 057
KEYNOTE-001
KEYNOTE-010
LUX-Lung 8
Pooled NP28673 & NP28761
ALTA
LUX-Lung 8
AURA
POPLAR
OAK
CheckMate 017
CheckMate 057
Pooled CheckMate 017 & 057
KEYNOTE-010

Figure 3: Number of survival models for which longer follow-up data resulted in an increase in mean OS

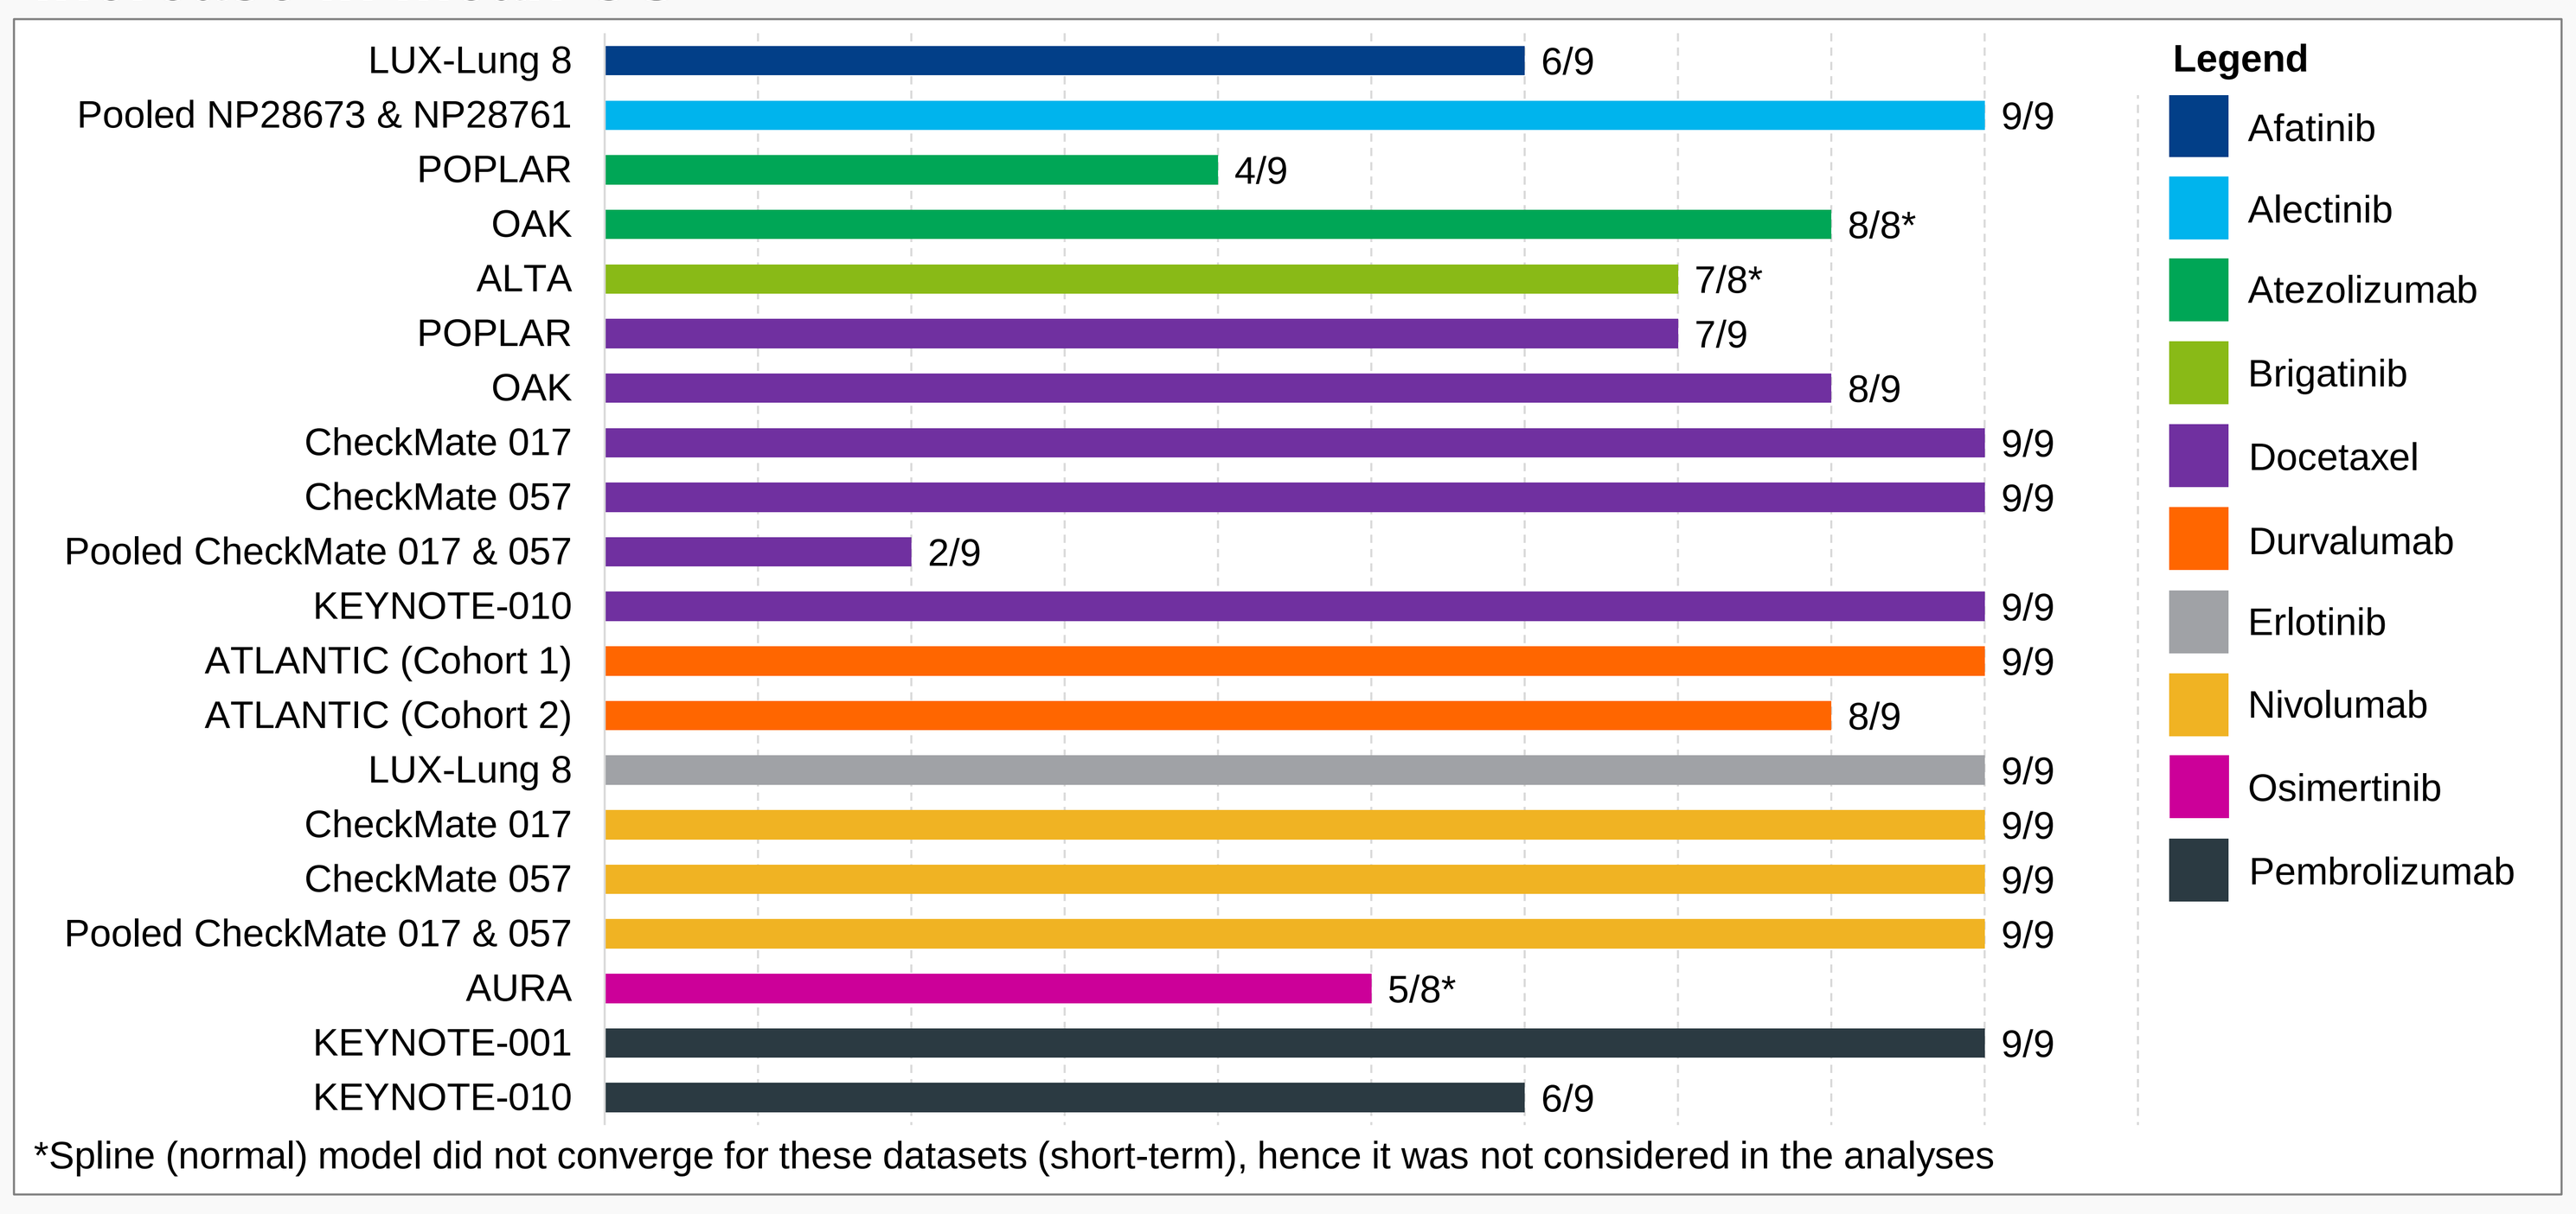
LUX-Lung 8 6/9
Pooled NP28673 & NP28761 9/9
POPLAR 4/9
OAK 8/8*
ALTA 7/8*
POPLAR 7/9
OAK 8/9
CheckMate 017 9/9
CheckMate 057 9/9
Pooled CheckMate 017 & 057 2/9
KEYNOTE-010 9/9
ATLANTIC (Cohort 1) 9/9
ATLANTIC (Cohort 2) 8/9
LUX-Lung 8 9/9
CheckMate 017 9/9
CheckMate 057 9/9
Pooled CheckMate 017 & 057 9/9
AURA 5/8*
KEYNOTE-001 9/9
KEYNOTE-010 6/9
Legend
Afatinib
Alectinib
Atezolizumab
Brigatinib
Docetaxel
Durvalumab
Erlotinib
Nivolumab
Osimertinib
Pembrolizumab

*Spline (normal) model did not converge for these datasets (short-term), hence it was not considered in the analyses

Figure 4: % change in mean OS for short- versus long-term follow-up data by population – Difference between best fitting models (based on AIC)

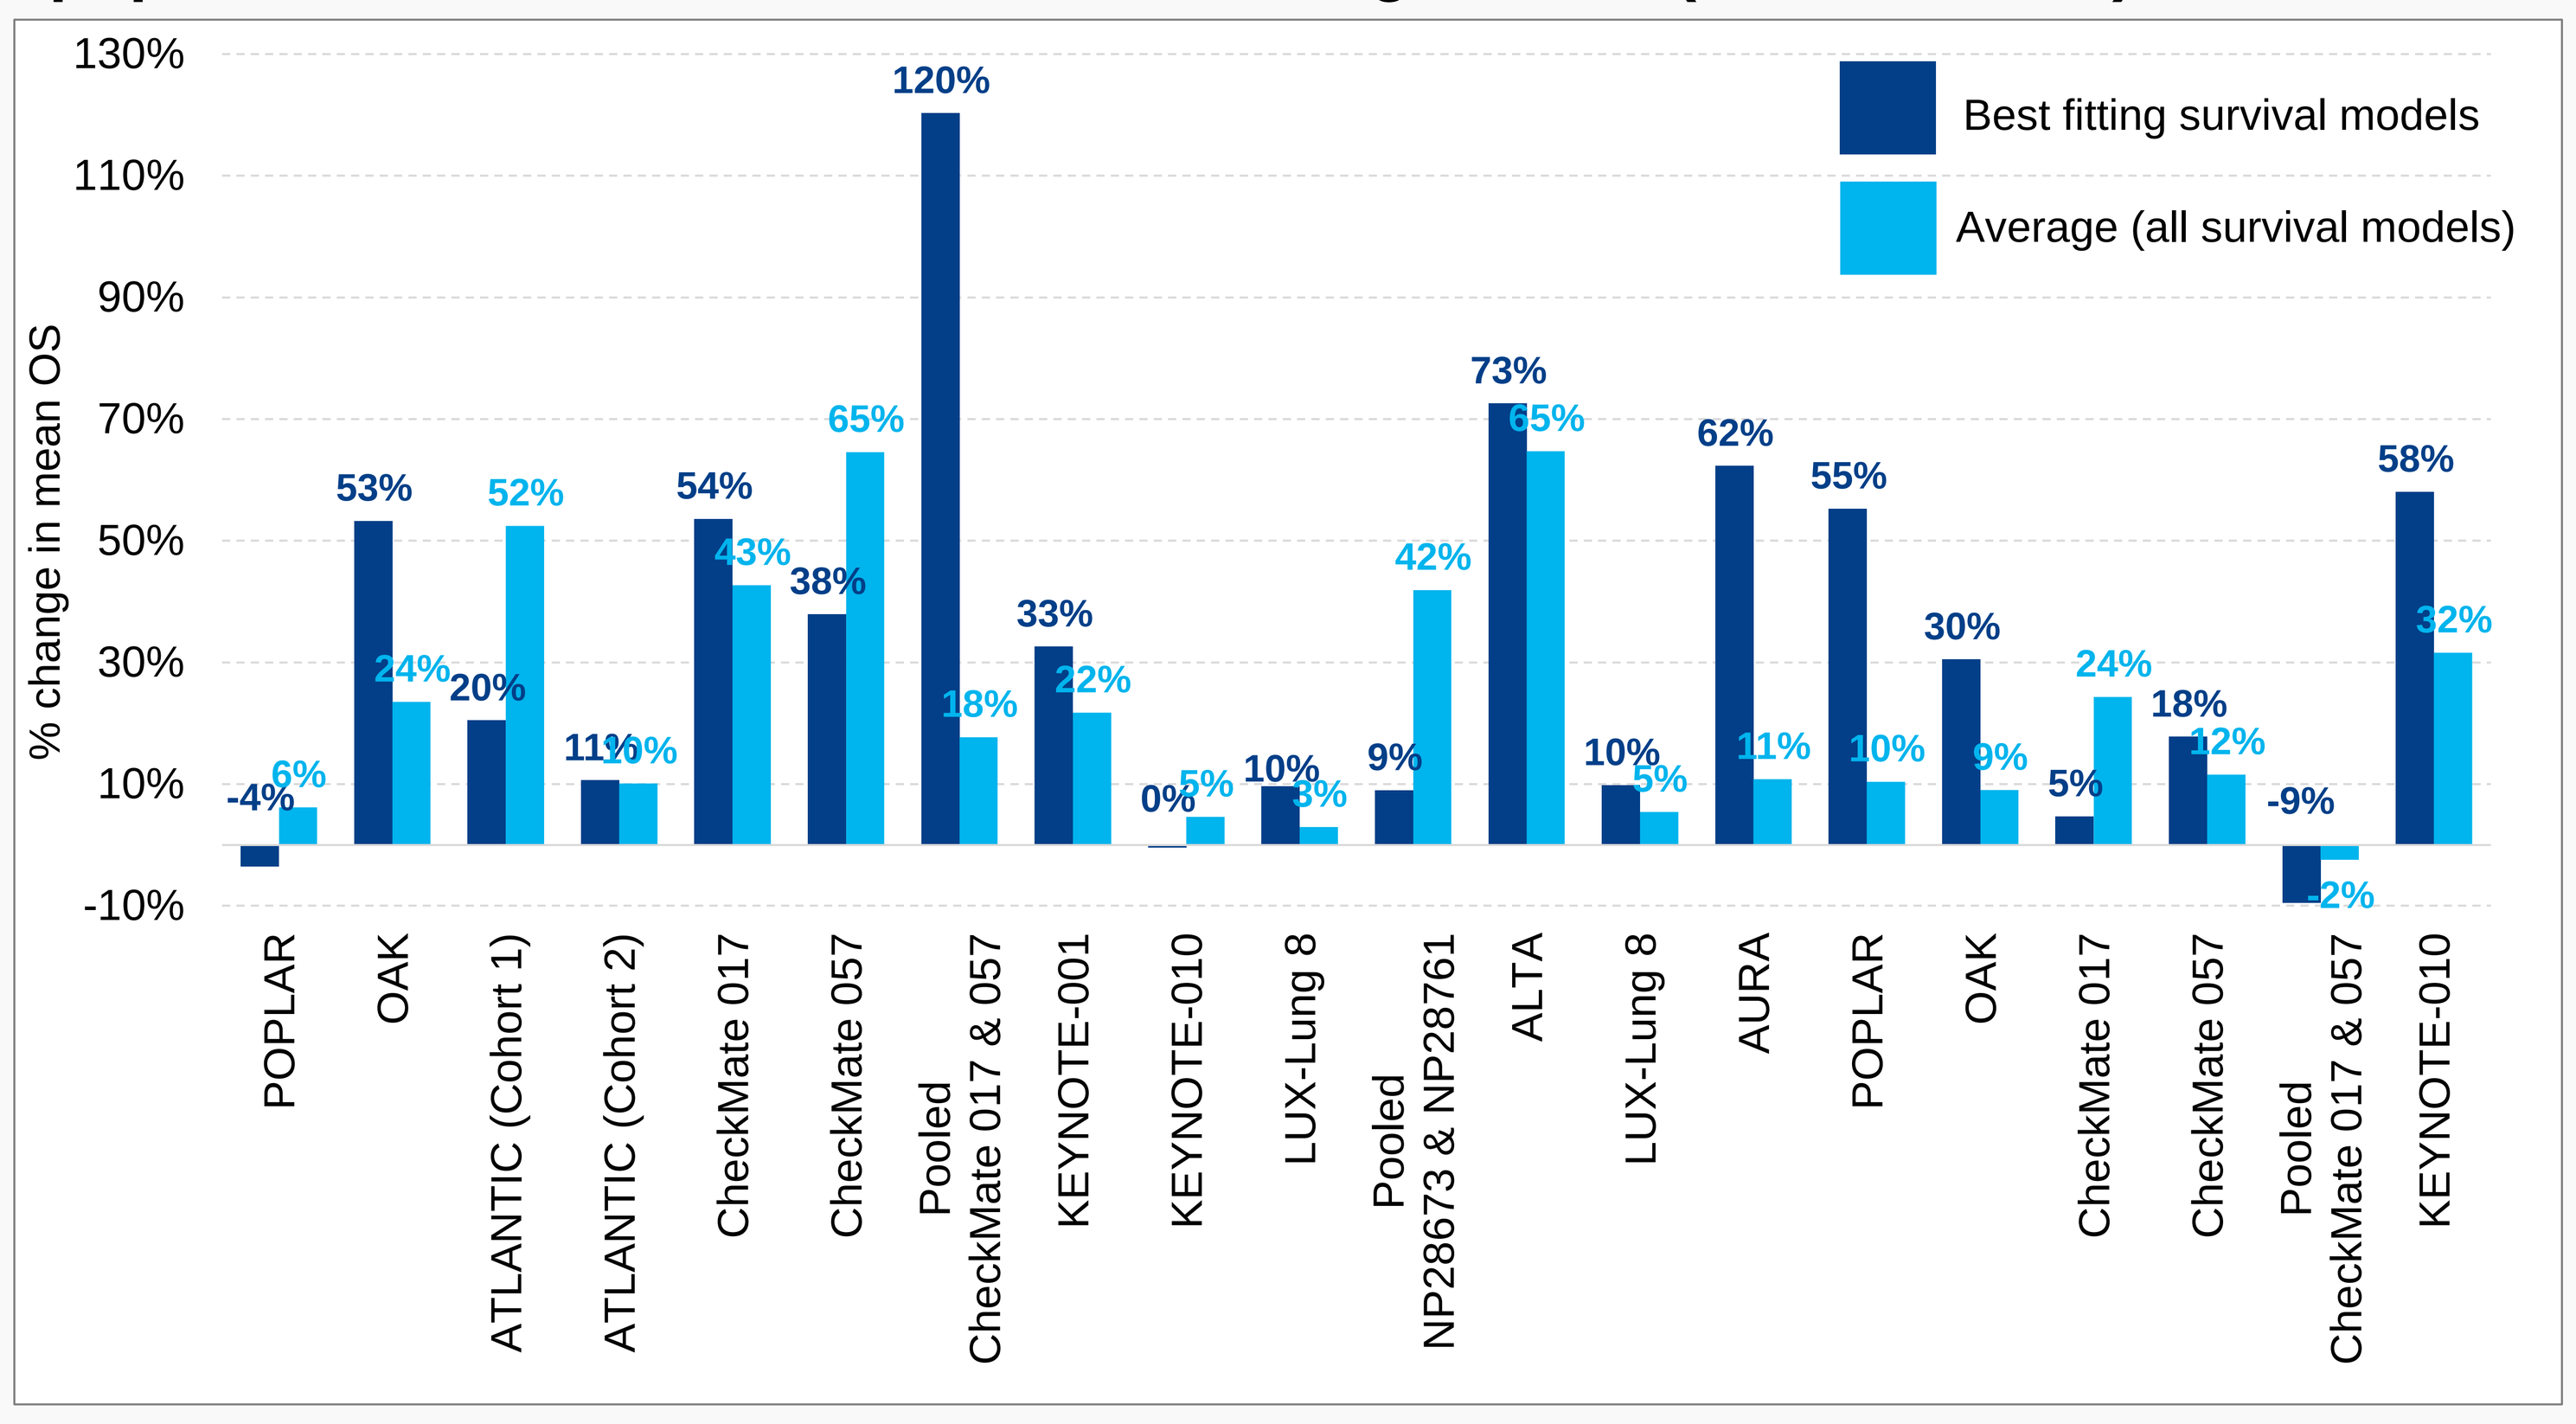
% change in mean OS
Best fitting survival models
Average (all survival models)
130%
110%
90%
70%
50%
30%
10%
-10%
POPLAR -4% 6%
OAK 53% 24%
ATLANTIC (Cohort 1) 20% 52%
ATLANTIC (Cohort 2) 11% 10%
CheckMate 017 54% 43%
CheckMate 057 38% 65%
Pooled CheckMate 017 & 057 120% 18%
KEYNOTE-001 33% 22%
KEYNOTE-010 0% 5%
LUX-Lung 8 10% 3%
Pooled NP28673 & NP28761 9% 42%
ALTA 73% 65%
LUX-Lung 8 10% 5%
AURA 62% 11%
POPLAR 55% 10%
OAK 30% 9%
CheckMate 017 5% 24%
CheckMate 057 18% 12%
Pooled CheckMate 017 & 057 -9% -2%
KEYNOTE-010 58% 32%

Figure 5: % change in mean OS vs AIC ranking

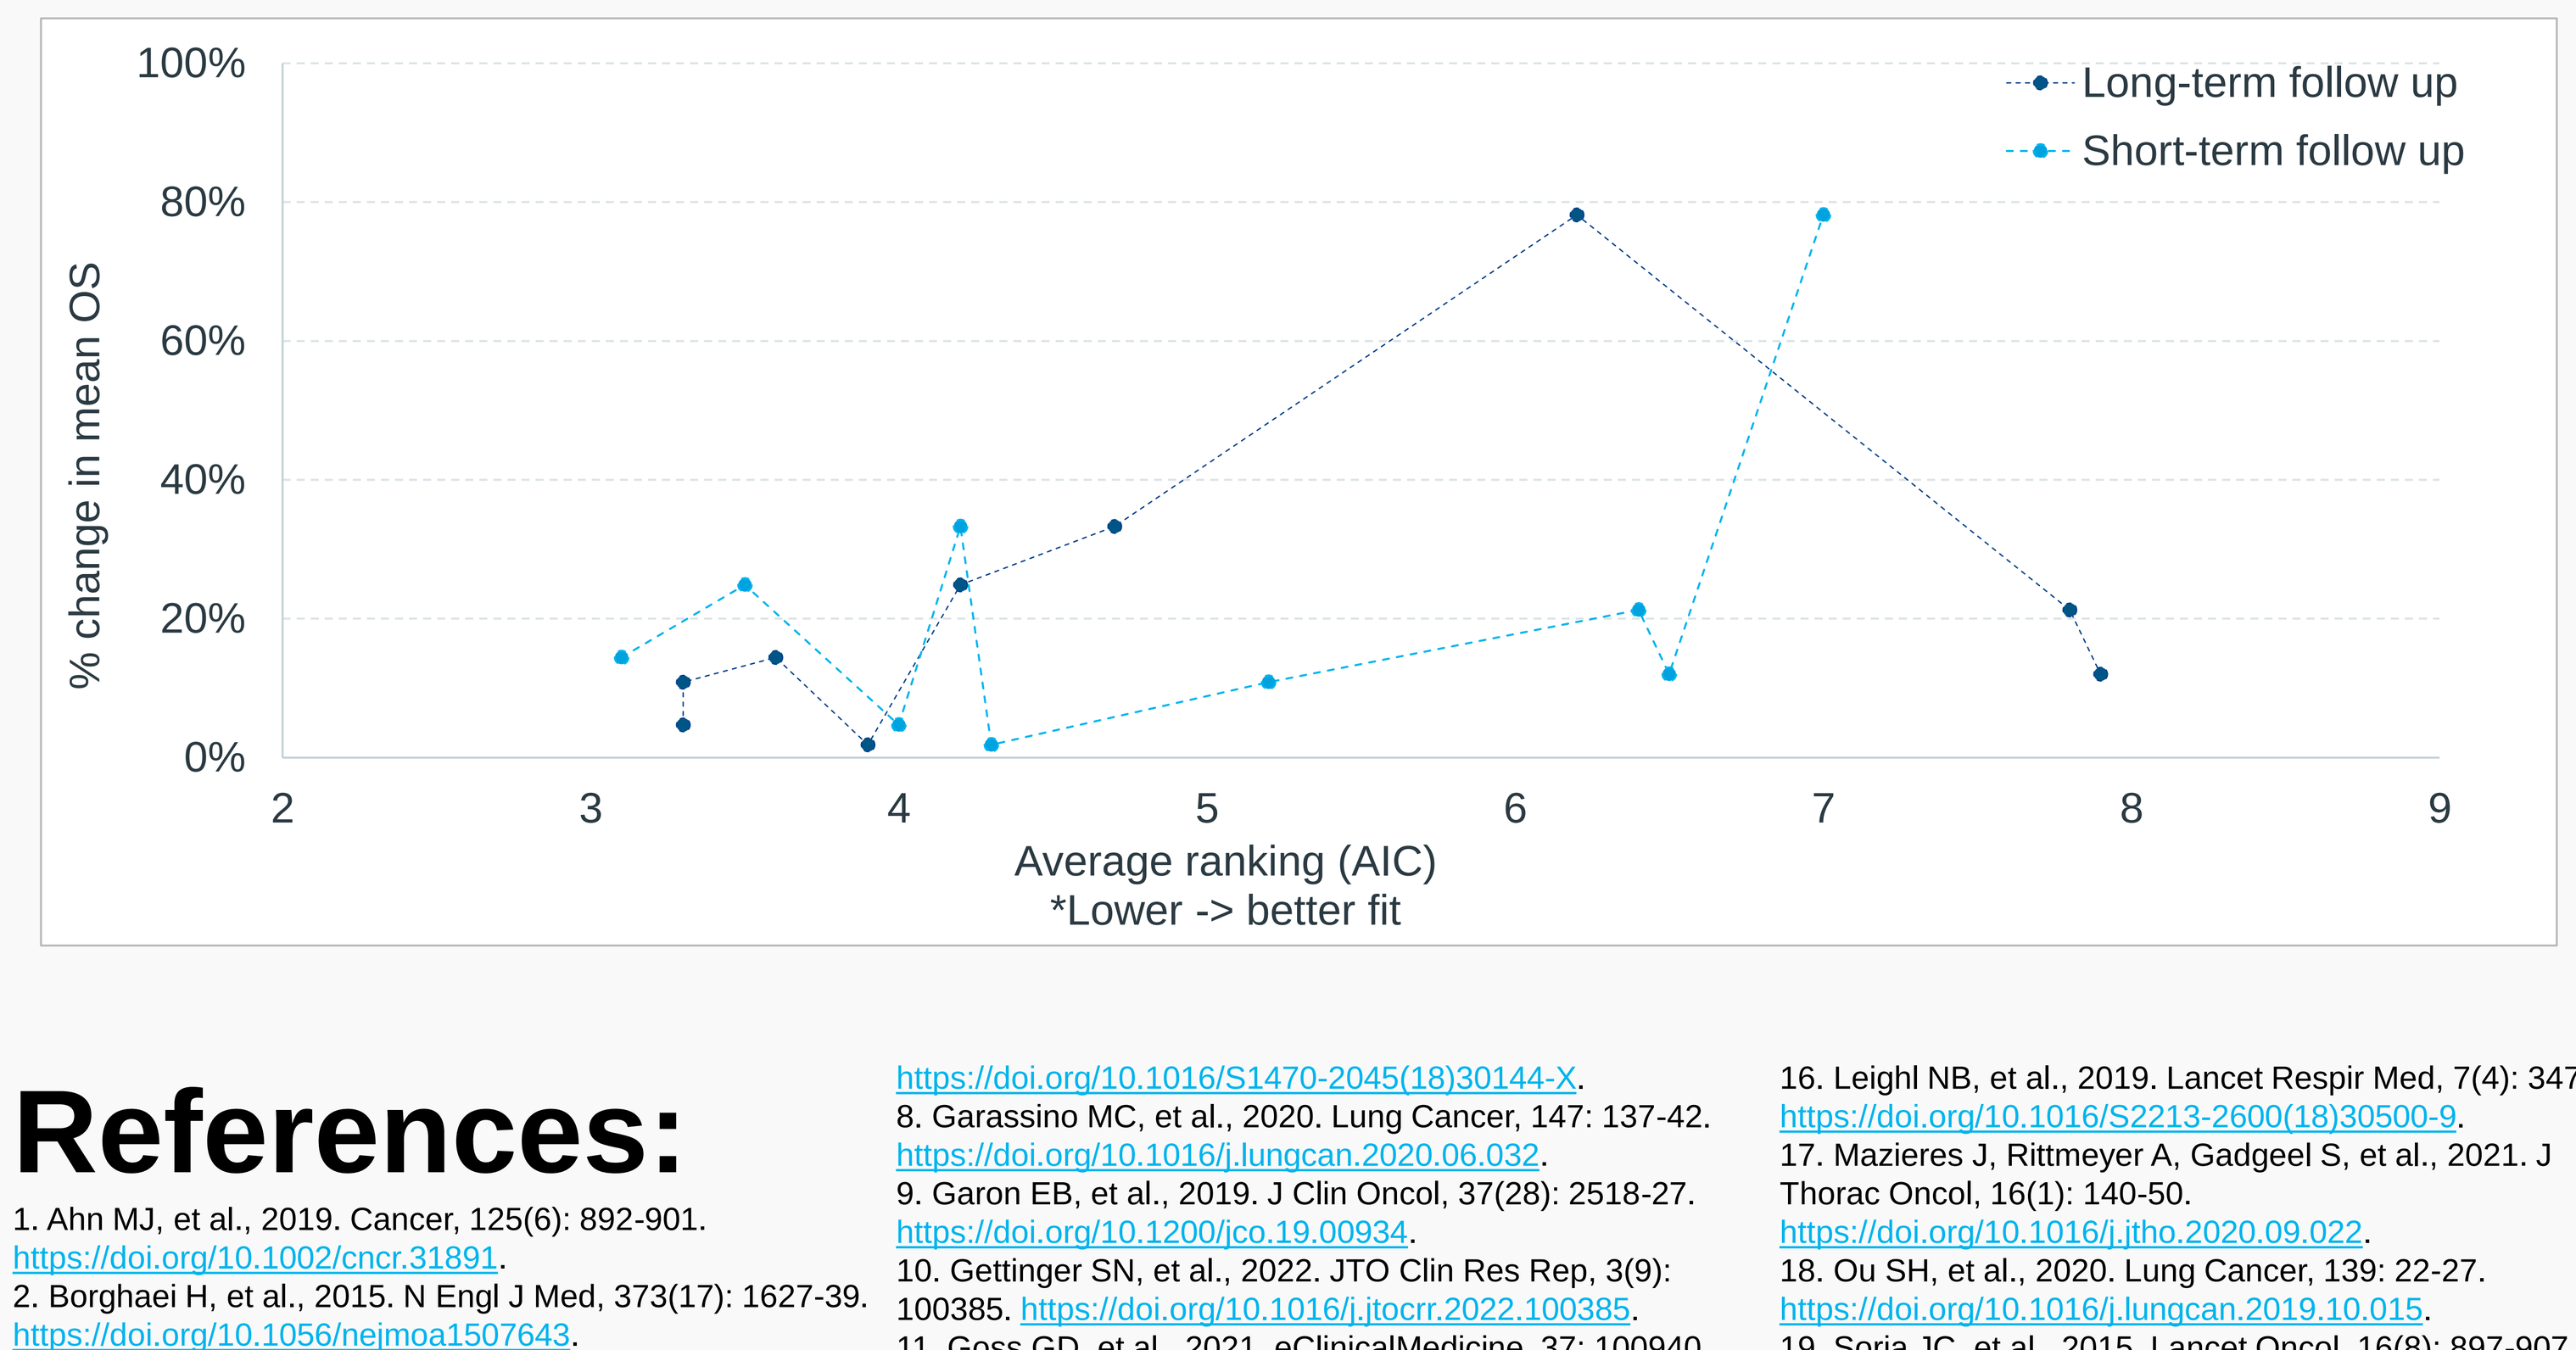
% change in mean OS
Long-term follow up
Short-term follow up
100%
80%
60%
40%
20%
0%
2
3
4
5
6
7
8
9
Average ranking (AIC)
*Lower -> better fit

References:

1. Ahn MJ, et al., 2019. Cancer, 125(6): 892-901. https://doi.org/10.1002/cncr.31891.
2. Borghaei H, et al., 2015. N Engl J Med, 373(17): 1627-39. https://doi.org/10.1056/nejmoa1507643.
3. Borghaei H, et al., 2021. J Clin Oncol, 39(7): 723-33. https://doi.org/10.1200/jco.20.01605.
4. Brahmer J, et al., 2015. N Engl J Med, 373(2): 123-35. https://doi.org/10.1056/nejmoa1504627.
5. Fehrenbacher L, et al., 2016. Lancet, 387(10030): 1837-46. https://doi.org/10.1016/s0140-6736(16)00587-0.
6. Fehrenbacher L, et al., 2018. J Thorac Oncol, 13(8): 1156-70. https://doi.org/10.1016/j.jtho.2018.04.039.
7. Garassino MC, et al., 2018. Lancet Oncol, 19(4): 521-36. https://doi.org/10.1016/S1470-2045(18)30144-X.
8. Garassino MC, et al., 2020. Lung Cancer, 147: 137-42. https://doi.org/10.1016/j.lungcan.2020.06.032.
9. Garon EB, et al., 2019. J Clin Oncol, 37(28): 2518-27. https://doi.org/10.1200/jco.19.00934.
10. Gettinger SN, et al., 2022. JTO Clin Res Rep, 3(9): 100385. https://doi.org/10.1016/j.jtocrr.2022.100385.
11. Goss GD, et al., 2021. eClinicalMedicine, 37: 100940. https://doi.org/10.1016/j.eclinm.2021.100940.
12. Herbst RS, et al., 2016. Lancet, 387(10027): 1540-50. https://doi.org/10.1016/S0140-6736(15)01281-7.
13. Herbst RS, et al., 2019. Ann Oncol, 30(2): 281-89. https://doi.org/10.1093/annonc/mdy545.
14. Herbst RS, et al., 2021. J Thorac Oncol, 16(10): 1718-32. https://doi.org/10.1016/j.jtho.2021.05.001.
15. Kim DW, et al., 2017. J Clin Oncol, 35(22): 2490-98. https://doi.org/10.1200/jco.2016.71.5904.
16. Leighl NB, et al., 2019. Lancet Respir Med, 7(4): 347-57. https://doi.org/10.1016/S2213-2600(18)30500-9.
17. Mazieres J, Rittmeyer A, Gadgeel S, et al., 2021. J Thorac Oncol, 16(1): 140-50. https://doi.org/10.1016/j.jtho.2020.09.022.
18. Ou SH, et al., 2020. Lung Cancer, 139: 22-27. https://doi.org/10.1016/j.lungcan.2019.10.015.
19. Soria JC, et al., 2015. Lancet Oncol, 16(8): 897-907. https://doi.org/10.1016/s1470-2045(15)00006-6.
20. Vokes EE, et al., 2018. Ann Oncol, 29(4): 959-65. https://doi.org/10.1093/annonc/mdy041.
21. Yang JC, et al., 2017. J Clin Oncol, 35(12): 1288-96. https://doi.org/10.1200/jco.2016.70.3223.
22. Yang JC, et al., 2017. J Thorac Oncol, 12(10): 1552-60. https://doi.org/10.1016/j.jtho.2017.06.070.

Presented at ISPOR EU 2023 Copenhagen, DK | 12-15 November 2023