EE690

Assessing the relative budget impact of PARP inhibitor combination therapies for mCRPC patients with and without BRCA1/2 mutations

Melissa Gomez Montero[1], Camille Capone[2], Florint Kroi[1], Peter Wigfield[1], Camille Dalibot[3], Paul Robinson[4]

[1] Cytel, Real World Advanced Analytics, Rotterdam, Netherlands;
[2] Janssen EMEA, Issy-les-Moulineaux, France;
[3] Janssen France, Issy-les-Moulineaux, France;
[4] Janssen EMEA, High Wycombe, UK

BACKGROUND

- There is a high unmet need for therapies that improve outcomes in patients with metastatic castration-resistant prostate cancer (mCRPC).
- Two PARP inhibitors (PARPi) have recently been approved in Europe for chemotherapy-ineligible patients with first-line mCRPC:
 - niraparib + abiraterone and prednisone (AAP) dual-action tablet for BRCA 1/2-mutated (BRCA+) patients; and
 - olaparib + AAP free combination for patients irrespective of mutational status.

OBJECTIVES

- To evaluate the potential economic impact of the introduction of niraparib + AAP and olaparib + AAP within their respective licensed indications, we aim to estimate the relative budget impact for BRCA+ compared to non-BRCA first-line mCRPC patients.

METHODS

- An Excel-based model was developed to simulate the BRCA+ and the non-BRCA populations over a five-year time horizon, from a French healthcare perspective (Figure 1).[1]
- The number of patients entering the model was estimated based on prevalence rates for the first year in the model and based on incidence rates for every year thereafter (Table 1).[2-5] Patients remain in the model until death.
- Niraparib + AAP and olaparib + AAP eligible populations were considered in accordance with their EMA labels.[6,7]
- Other modelled first-line mCRPC treatments included AAP monotherapy, enzalutamide, and olaparib monotherapy. Chemotherapies were excluded due to modeled population involving chemotherapy-ineligible patients, as per drug indications.
- Median radiographic progression-free survival and overall survival from pivotal trials were used to estimate pre- and post-progression disease course.[8-12]
- A clinician panel validated background treatment uptake and projected substitution by the PARPi combinations: The reference case simulated the current environment where PARPi treatments are not available, while in the new scenario, PARPi uptake was assumed to increase each year.
- Companion diagnostic costs were considered only for the BRCA+ population.

TABLE 1: Model inputs

Parameter	Non-BRCA	BRCA+
Male population 18+[13]	25,586,034	
Annual growth rate of population[14]	0.30%	
Prevalence rate of mCRPC[2-4]	0.06%	
Incidence rate of mCRPC[3,5]	0.03%	
Percent of patients who received test[5]	NA	100%
Percent of patients with positive BRCA+ mutation test[15]	NA	10%
Mortality rates[16]	Year 1: 8.1%; Year 2: 28.1%; Year 3: 49.9%; Year 4: 66.8%; Year 5: 78.0%	

Abbreviations: mCRPC, metastatic castration-resistant prostate cancer.

PROSTATE CANCER

FIGURE 1: Model structure

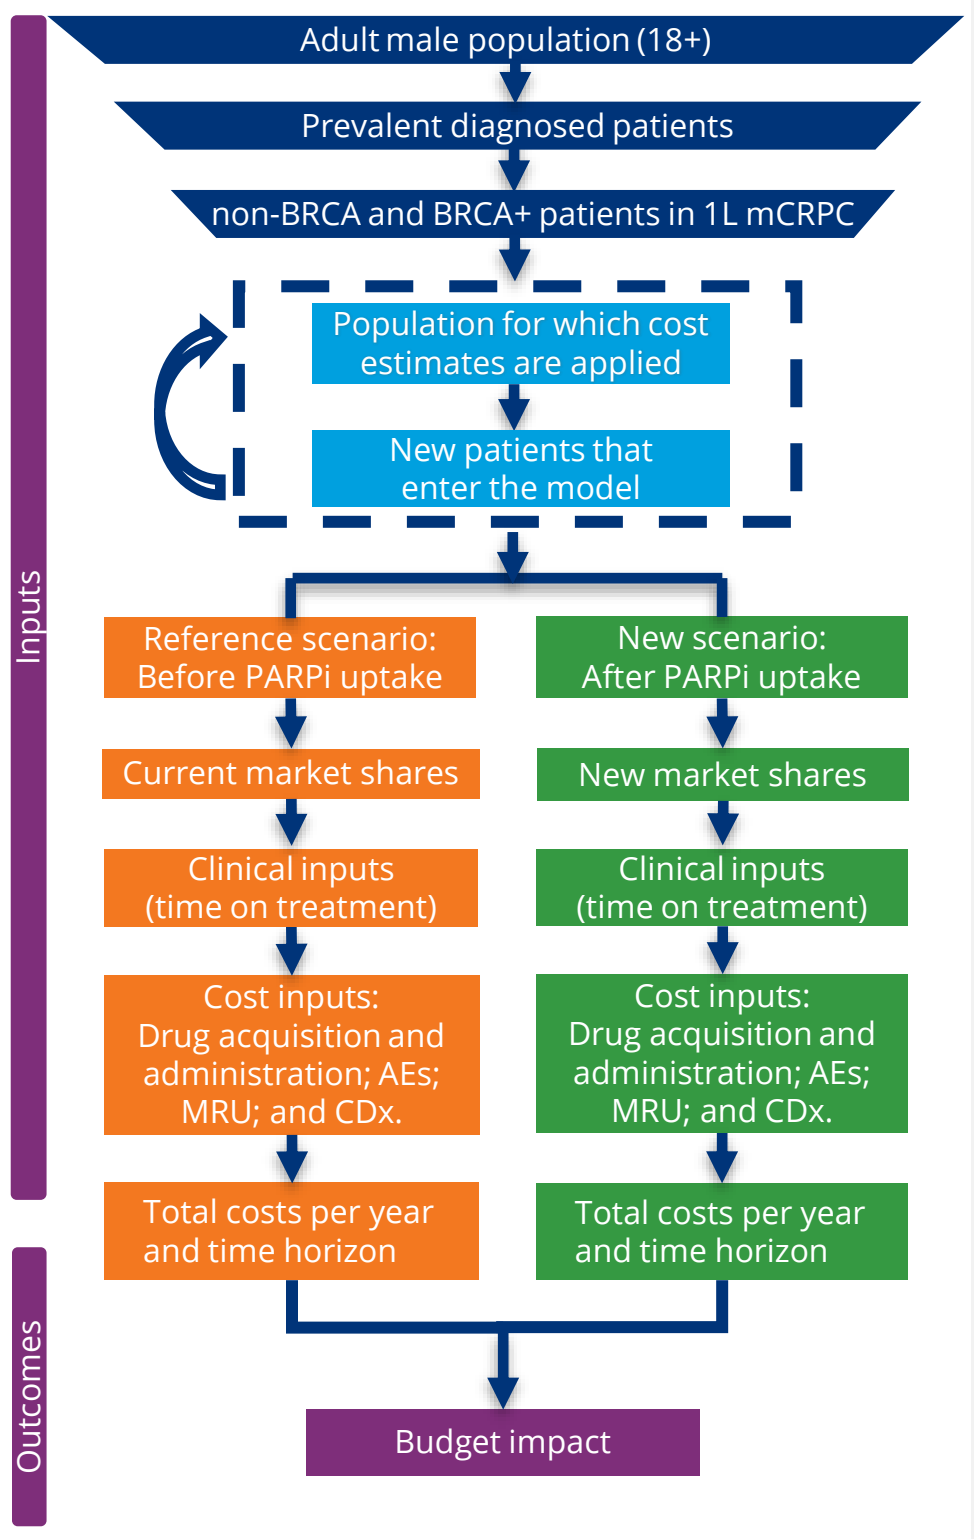
Inputs
Outcomes
Adult male population (18+)
Prevalent diagnosed patients
non-BRCA and BRCA+ patients in 1L mCRPC
Population for which cost estimates are applied
New patients that enter the model
Reference scenario: Before PARPi uptake
New scenario: After PARPi uptake
Current market shares
New market shares
Clinical inputs (time on treatment)
Clinical inputs (time on treatment)
Cost inputs: Drug acquisition and administration; AEs; MRU; and CDx.
Cost inputs: Drug acquisition and administration; AEs; MRU; and CDx.
Total costs per year and time horizon
Total costs per year and time horizon
Budget impact

Abbreviations: 1L, first-line; AEs, adverse events; CDx, companion diagnostic; mCRPC, metastatic castration-resistant prostate cancer; MRU, medical resource use.

TABLE 2: Budget impact model results

Budget impact	
Non-BRCA	€ 367,447,186
BRCA+	€ 34,553,588
Budget impact per patient	
Non-BRCA	€ 26,990
BRCA+	€ 22,895

FIGURE 3: Total budget impact per year

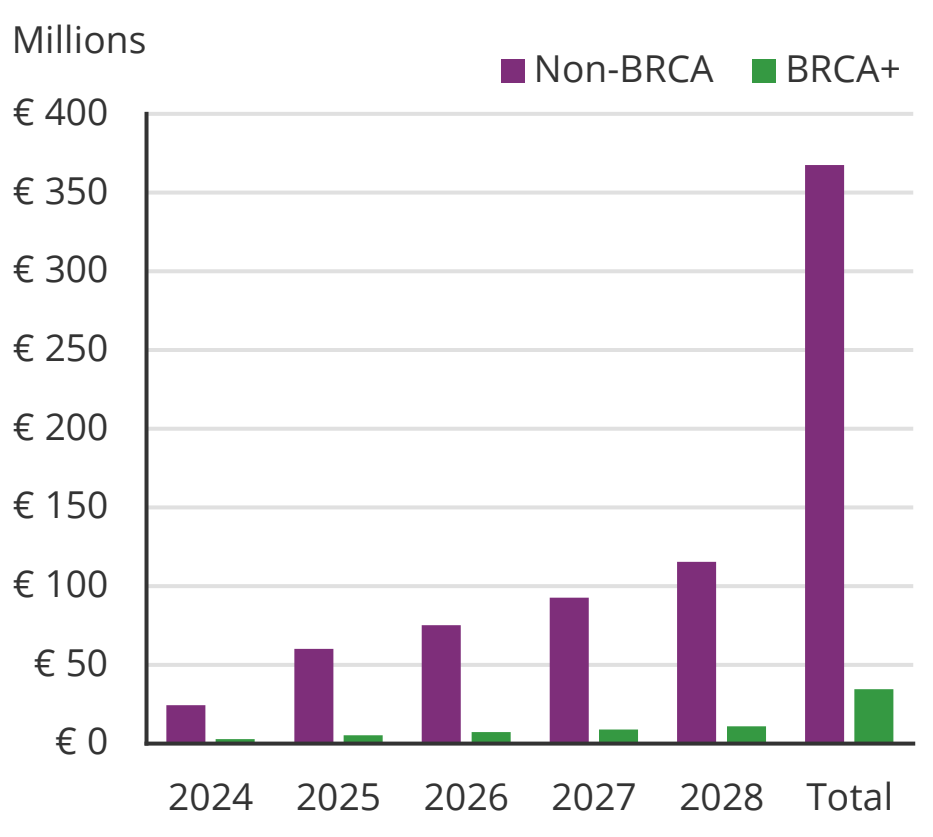
Millions
Non-BRCA
BRCA+
€ 400
€ 350
€ 300
€ 250
€ 200
€ 150
€ 100
€ 50
€ 0
2024
2025
2026
2027
2028
Total

RESULTS

- A total of 67,305 non-BRCA patients and 7,478 BRCA+ patients were estimated to be treated for first-line mCRPC over five years (Figure 2).
- The overall budget impact was estimated at €367.4M for non-BRCA patients treated with olaparib + AAP, compared to €34.5M for BRCA+ patients treated with either niraparib + AAP or olaparib + AAP over a five-year time horizon (Figure 3).
- The corresponding per-patient budget impact was estimated at €26,990 for non-BRCA patients, compared to €22,895 for BRCA+ patients (Table 2).
- Non-BRCA patients reflected 90% of mCRPC patients in terms of population, which generated 91% of the budget impact costs.
- In comparison, BRCA+ patients represented 10% of mCRPC patients, leading to 9% of total costs.
- Treatment related costs (i.e., drug acquisition and administration costs) in the pre- and post-progression states were the main drivers of the budget impact for both non-BRCA and BRCA+ patients.
- Non-BRCA patients experience a less aggressive disease course compared to BRCA+ patients. Therefore, given that treatment continues until progression, non-BRCA patients will have longer treatment exposure.[17]
- This leads to increased per-patient drug costs and AE-related medical resource use costs in the non-BRCA group receiving olaparib + AAP.
- Non-treatment related costs (i.e., adverse event, medical resource use and companion diagnostic costs) increased in the new scenario for both non-BRCA and BRCA+ patients. (Figure 4).

FIGURE 4: Non-treatment related costs

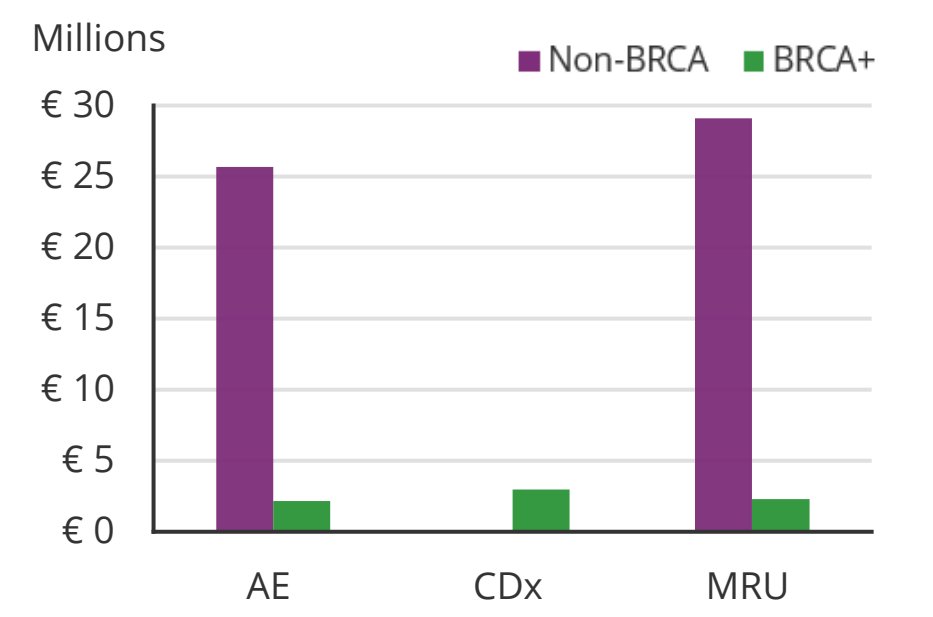
Millions
Non-BRCA
BRCA+
€ 30
€ 25
€ 20
€ 15
€ 10
€ 5
€ 0
AE
CDx
MRU

Abbreviations: AEs, adverse event; CDx, companion diagnostic; MRU, medical resource use.

FIGURE 2: Treatment breakdown of non-BRCA and BRCA+ patients after PARPi combination uptake

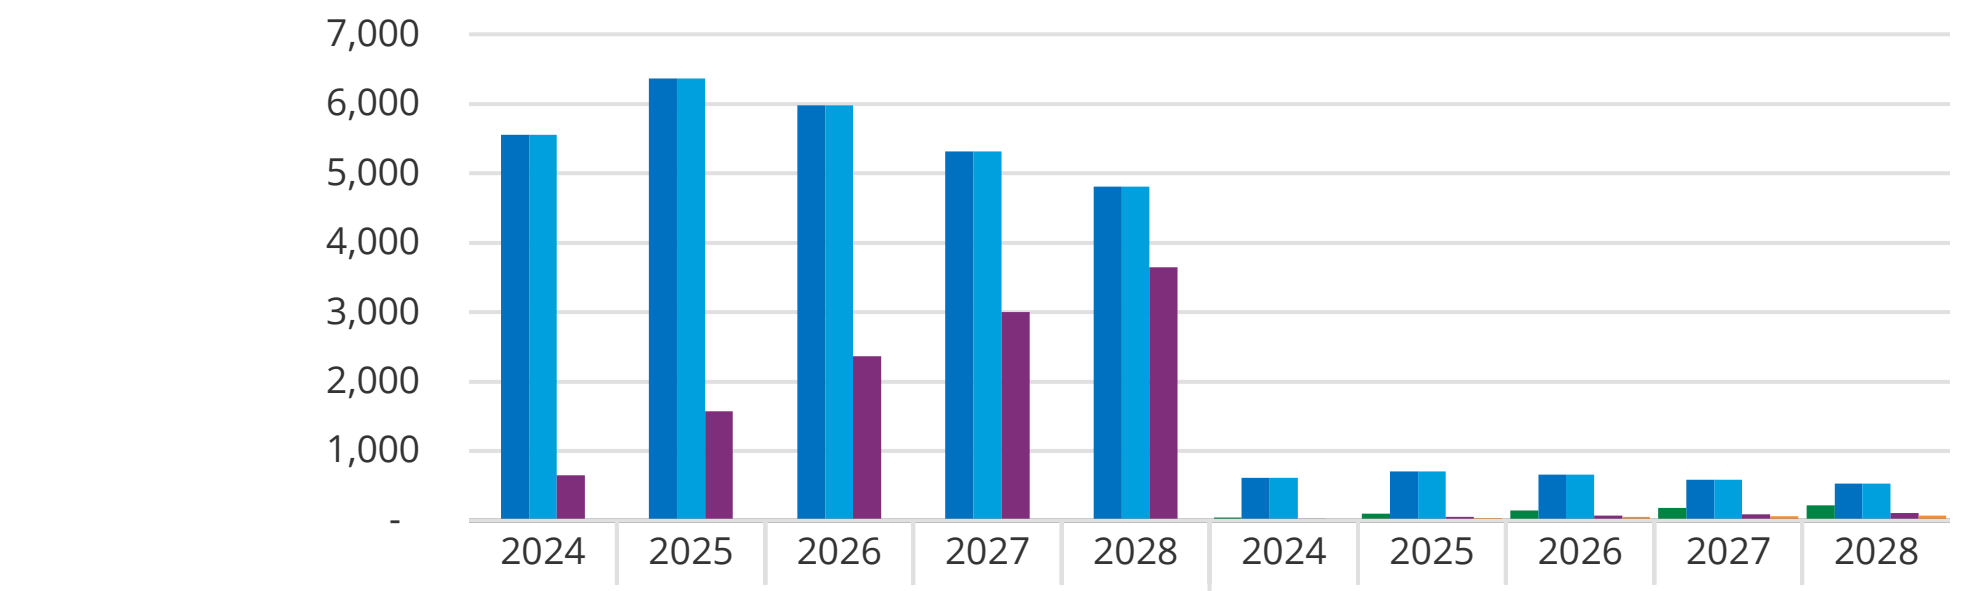
7,000
6,000
5,000
4,000
3,000
2,000
1,000
-

	non-BRCA 2024	non-BRCA 2025	non-BRCA 2026	non-BRCA 2027	non-BRCA 2028	BRCA 2024	BRCA 2025	BRCA 2026	BRCA 2027	BRCA 2028
Niraparib + AAP	-	-	-	-	-	39	95	143	182	221
AAP	5,556	6,371	5,980	5,317	4,812	617	708	664	591	535
Enzalutamide	5,556	6,371	5,980	5,317	4,812	617	708	664	591	535
Olaparib + AAP	647	1,575	2,363	2,999	3,650	20	48	72	91	111
Olaparib monotherapy	-	-	-	-	-	13	32	48	61	74

KEY TAKEAWAY

Uncertain clinical benefit in non-BRCA patients, compounded with a high cost burden, highlights the importance of ensuring PARPi combinations are reserved for those patients at greatest unmet need and the most cost-efficient use of healthcare resources.

CONCLUSIONS

Treatment with PARPi in the non-BRCA population results in a ten-fold increase in budget impact relative to the BRCA+ population.

Olaparib + AAP is associated with disproportionately higher healthcare costs compared to niraparib + AAP when considered in accordance to their EMA labels.

Given the high AE-related costs, the risk-benefit of olaparib + AAP treatment in non-BRCA patients has been questioned.[17]

The current clinical data and the estimated budget impact highlight the benefit and responsibility to use PARPi combination treatments in the BRCA+ subgroup of mCRPC patients.

REFERENCES

1. Sullivan et al. 2014, Budget impact analysis-principles of good practice: report of the ISPOR 2012 Budget Impact Analysis Good Practice II Task Force; PMID: 24438712.
2. Santé Publique France. Synthèse - Estimations nationales de l'incidence et de la mortalité par cancer en France métropolitaine entre 1990 et 2018.
3. Office parlementaire d'évaluation des politiques de santé. Rapport sur le dépistage et le traitement du cancer de la prostate; 2009.
4. HAS. Avis de la Commission de la Transparence de LYNPARZA. 5 mai 2021.
5. Cerner Enviza. Prise en charge du cancer de la prostate metastatique resistant a la castration en vie réelle: Rapport clinique. Février 2022.
6. European medicines agency (EMA) Lymparza product information available at: https://www.ema.europa.eu/en/documents/product-information/lynparza-epar-product-information_en.pdf
7. European medicines agency (EMA) Akeega product information available at: https://www.ema.europa.eu/en/documents/product-information/akeega-epar-product-information_en.pdf
8. Chi et al. 2023, Niraparib and Abiraterone Acetate for Metastatic Castration-Resistant Prostate Cancer; PMID: 36952634
9. Saad et al. 2023, Olaparib plus abiraterone versus placebo plus abiraterone in metastatic castration-resistant prostate cancer (PROpel): final prespecified overall survival results of a randomised, double-blind, phase 3 trial; PMID: 37714168
10. Fizazi et al. 2023, TALAPRO-2: Phase 3 study of talazoparib (TALA) + enzalutamide (ENZA) versus placebo (PBO) + ENZA as first-line (1L) treatment for patients (pts) with metastatic castration-resistant prostate cancer (mCRPC) harboring homologous recombination repair (HRR) gene alterations; DOI: 10.1200/JCO.2023.41.16_suppl. 5004
11. Armstrong et al. 2020, Five-year Survival Prediction and Safety Outcomes with Enzalutamide in Men with Chemotherapy-naïve Metastatic Castration-resistant Prostate Cancer from the PREVAIL Trial; PMID: 32527692
12. Bono et al. 2010, Prednisone plus cabazitaxel or mitoxantrone for metastatic castration-resistant prostate cancer progressing after docetaxel treatment: a randomised open-label trial; PMID: 20888992
13. INSEE. Population by sex and age group, data available on January 1st, 2023. Men ≥ 18 years old.
14. INSEE. Bilan démographique 2021. https://www.insee.fr/fr/statistiques/6024136 .
15. MAGNITUDE CSR FA July 2023 - Data on file.
16. MAGNITUDE cost-effectiveness model v3.20, September 2023.
17. Oncologic Drugs Advisory Committee (ODAC) Meeting. Lynparza (Olaparib) in Combination with Abiraterone and Prednisone or Prednisolone for the Treatment of Adult Patients with Metastatic Castration-Resistant Prostate Cancer. Sponsor Briefing Document. April 2023. Retrieved from: https://www.fda.gov/media/167485/download

DISCLOSURES

The figures in this poster presentation were based on inputs from final analysis data of the MAGNITUDE study. This differs from the abstract submission, which used data from interim analysis 2 datacut, since final data were not available at the time of submission.

This study was sponsored by Janssen EMEA.

Presented at ISPOR Europe 2023; 15 November 2023; Copenhagen, Denmark.