EPH240

Epidemiology, Economic Burden, and Humanistic Burden in Multiple Sclerosis Spasticity (MSS): A Systematic Literature Review (SLR)

Caroleen Drachenberg,[1] Gaurang Nazar,[2] Divya Pushkarna,[2] Hema N. Viswanathan,[1] Mir Sohail Fazeli.[2]

[1]Jazz Pharmaceuticals, Palo Alto, California, United States; [2]Evidinno Outcomes Research Inc., Vancouver, British Columbia, Canada

Background

- Multiple sclerosis (MS) is a chronic inflammatory disorder affecting the central nervous system, causing non-traumatic neurological disability in young and middle-aged adults.[1] Globally, 2.8 million people were living with MS in 2020, an estimated 30% increase in the number since 2013.[2] MS adversely affects patients' quality of life and creates significant economic burden because of direct and indirect care costs, even in patients with lower physical disabilities.
- Spasticity is experienced by up to 80% of MS patients.[3] Multiple sclerosis spasticity (MSS) symptoms include spasms, pain, movement difficulties, sleep disturbances, and urinary dysfunction.[4] Despite its high prevalence, MSS is often overlooked and poorly managed.
- Although clinical guidelines for managing MSS have been developed by the Consortium of Multiple Sclerosis (CMSC) and several European countries, there is still a significant lack of evidence on the epidemiology, economic burden, and humanistic burden of MSS.

Objectives

- To identify and characterize the published literature in MSS. Specifically focusing on:
- Epidemiology of MSS (incidence, prevalence, symptoms and risk factors).
- Economic burden of MSS (direct and indirect costs, healthcare resource utilization, and health utilities).
- Humanistic burden of MSS [patient-reported outcomes (PROs), health-related quality of life (HRQoL), and caregiver burden].

Methods

- A systematic literature review (SLR) was conducted by searching Embase, MEDLINE® and EconLit databases from inception to July 8, 2022.
- Grey literature searches were conducted to capture conference abstracts (2020-2022) from relevant conferences.
- Inclusion was based on the following PICO(S) criteria
 - Population: Adults with MSS.
 - Interventions: Approved and investigational therapies, non-pharmaceutical therapies (e.g., physical therapy, surgery), and off-label therapies.
 - Comparators: Any or none.
 - Outcomes: Epidemiology, economic burden, healthcare resource utilization, and humanistic burden.
 - Study design: Observational studies (cohort studies, case-control, cross-sectional studies).
- Data extraction and quality assessment [using the Newcastle-Ottawa Scale (NOS)[5] and Joanna Briggs Institute (JBI) tool[6]] of included studies was undertaken by two independent reviewers. Any discrepancies were resolved by a third investigator. Results were reported as per Preferred Reporting Items for Systematic Reviews and Meta-analysis (PRISMA) guidelines.[7]

Results

Study selection and characteristics

- Of 7,011 abstracts retrieved, 27 unique observational studies were included. The study selection process is presented in **Figure 1.**
- The study sample size ranged from 22 to 20,969 (median: 345). Fifteen studies were conducted in multi-center settings, six in single-center, and six did not report the study setting.
- Most studies were conducted in Germany (n=5) and Spain (n=5), followed by the US (n=4), UK (n=3), and Italy (n=3). Three studies were conducted in multiple countries.
- Study designs included retrospective cohort (n=11), cross-sectional (n=11), prospective cohort (n=5).
- The majority of included studies were rated as low or unclear risk of bias per the NOS and JBI tools. For cohort studies, not having a control group and not specifying response rate and for cross-sectional studies, not having standard measurement criteria were key factors leading to bias.

Patient characteristics

- Mean age was reported in 21 studies (range: 40.1 to 58 years; median: 48.9 years).
- The percentage of female participants was reported in 21 studies and ranged from 40% to 85.7% (median: 69%).
- The most reported MS types were Relapsing-remitting (RRMS), Primary-progressive (PPMS), and Secondary-progressive (SPMS).
 - RRMS: reported in 11 studies, the percentage of included patients with RRMS ranged from 9% to 77% (median: 45%).
 - PPMS: reported in 11 studies, the percentage of included patients with PPMS ranged from 7% to 27% (median: 13%).
 - SPMS: reported in 11 studies, the percentage of included patients with SPMS ranged from 5% to 64% (median: 37%).

Epidemiology outcomes

- Four studies reported on overall spasticity prevalence in MS which ranged from 47.5% to 65.7% (median: 51.8%).[8,9,10,11]
- A 2004 US[12] study categorized spasticity prevalence by its severity – minimal spasticity (31%), mild (19%), moderate (17%), severe (need to modify daily activities - 13%), and total (prevents daily activities - 4%); and by types of MS:
 - Relapsing-stable MS: minimal/mild spasticity (35.7%), moderate spasticity (21.3%), and severe/total spasticity (15.9%).
 - Relapsing-worsening MS: minimal/mild spasticity (55.1%), moderate spasticity (72.2%), and severe/total spasticity (74.5%).
 - PPMS: minimal/mild spasticity (9.2%), moderate spasticity (6.5%), and severe/total spasticity (9.6%).
- A 2019 study[11] from Germany reported the prevalence of spasticity by types of MS: RRMS (30.6%), PPMS 74.2%, and SPMS (81.9%) and by the duration of disease (e.g., disease duration less than 2 years (11.1%) and disease duration more than 25 years (76.5%)).
- Muscle spasms were the most common symptom experienced by 58 to 97% of all MSS patients, followed by urinary dysfunction and sleep disturbances.[13,14]
- Fatigue (52%), physical activity (49%), heat (45%), and stress (43%) were identified as triggers for spasticity.[14]

Results (continued)

Figure 1: PRISMA flow diagram

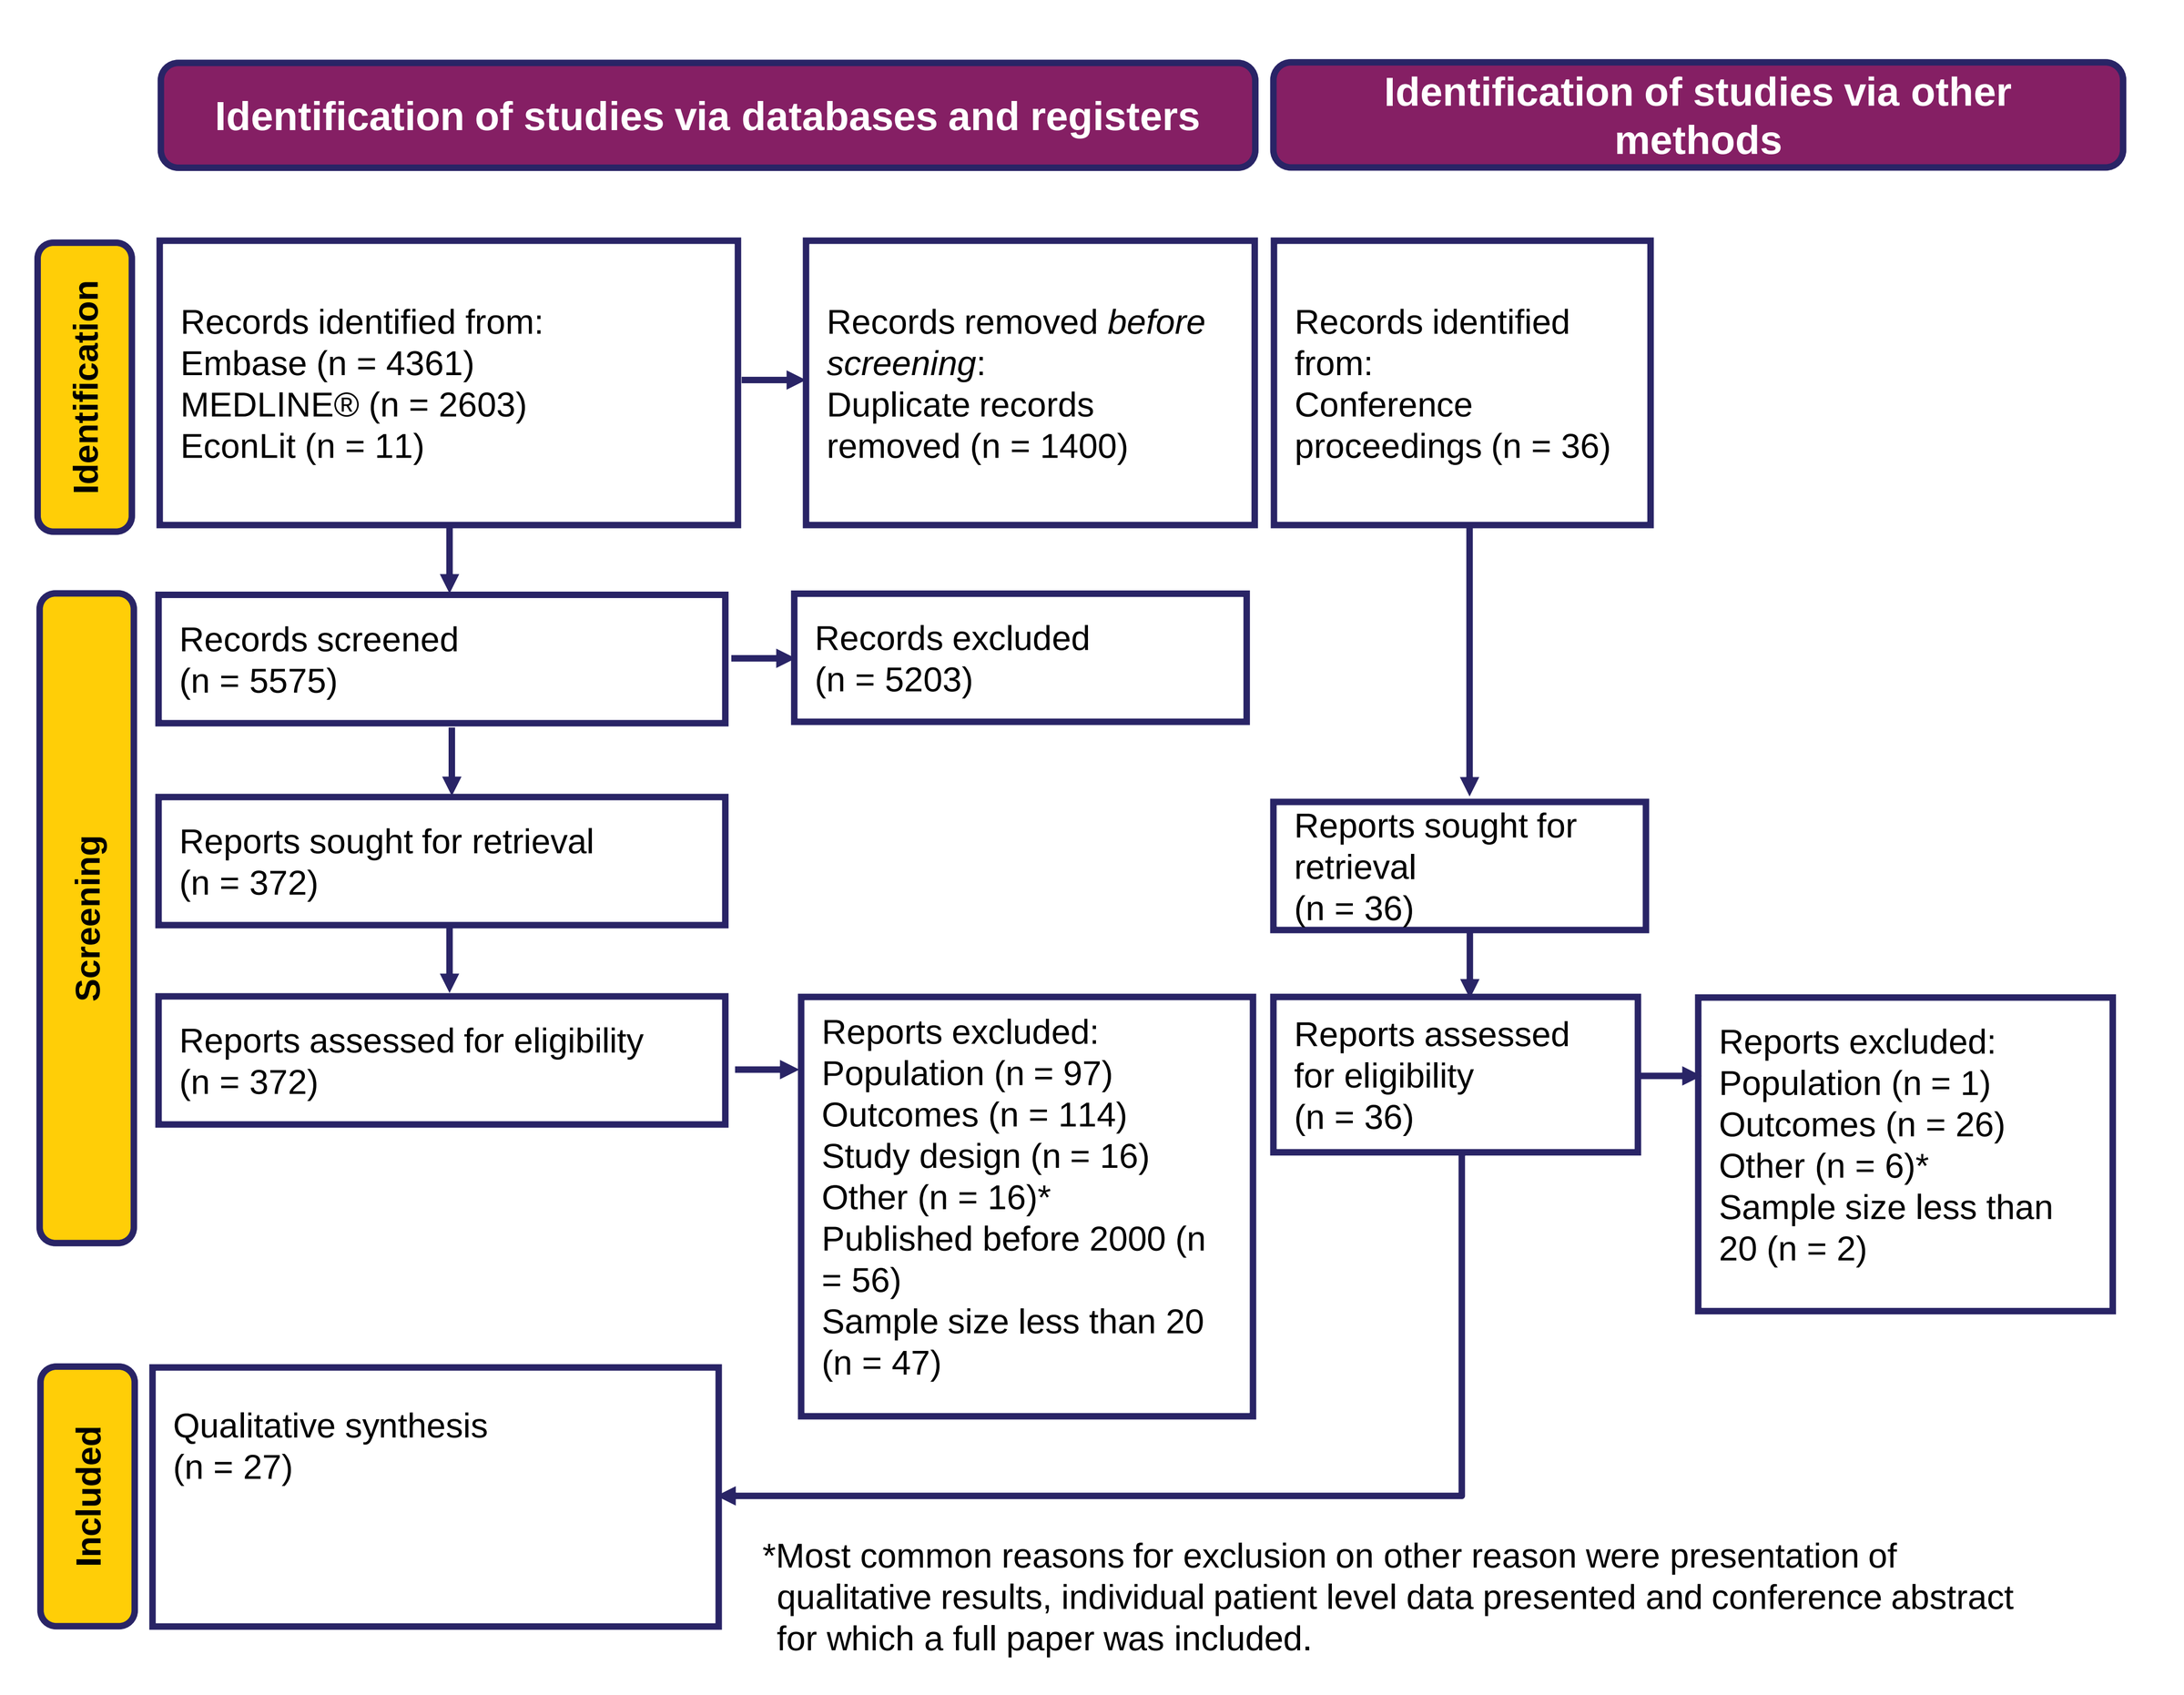
Identification of studies via databases and registers
Identification of studies via other methods
Identification
Records identified from:
Embase (n = 4361)
MEDLINE® (n = 2603)
EconLit (n = 11)
Records removed before screening:
Duplicate records removed (n = 1400)
Records identified from:
Conference proceedings (n = 36)
Screening
Records screened (n = 5575)
Records excluded (n = 5203)
Reports sought for retrieval (n = 372)
Reports sought for retrieval (n = 36)
Reports assessed for eligibility (n = 372)
Reports excluded:
Population (n = 97)
Outcomes (n = 114)
Study design (n = 16)
Other (n = 16)*
Published before 2000 (n = 56)
Sample size less than 20 (n = 47)
Reports assessed for eligibility (n = 36)
Reports excluded:
Population (n = 1)
Outcomes (n = 26)
Other (n = 6)*
Sample size less than 20 (n = 2)
Included
Qualitative synthesis (n = 27)
*Most common reasons for exclusion on other reason were presentation of qualitative results, individual patient level data presented and conference abstract for which a full paper was included.

Table 1: Results for studies reporting EQ-5D

Author and Year	EQ-5D outcome	Population	Estimate Type	Value/Score
Flachenecker 2013[17]	Health utilities	Mild spasticity	mean	0.6
		Moderate spasticity		0.5
		Severe spasticity		0.3
Barin 2018[16]	Health utilities	All MS	median	77.6*
		RRMS		83.5*
		Progressive MS		59*
	VAS	All MS		78
		RRMS		80
		Progressive MS		60
D'Hooghe 2021[20]	VAS	Patients using Sativex	mean	39 (at baseline) to 64 (at 12 months)
Vermersch 2016[21]	VAS	Patients using THC:CBD Oromucosal spray	mean	49.8 (at baseline) to 54.3 (at 3 months)
Vermersch 2014[19]	Health utilities	European MSS population	mean	53.3*
Svensson 2014[18]	Health utilities	Mild spasticity	mean	0.5
		Moderate spasticity		0.3
		Severe spasticity		0.03
	VAS	Mild spasticity		58.4
		Moderate spasticity		44.7
		Severe spasticity		34.9

*Re-scaled from 0-worst health to 100-best health

Table 2: Results for studies reporting MSQOL-54

Author and Year	MSQOL-54 Outcome	Population	Estimate Type	Value/Score
Meca-Lallana 2012[23]	Physical health composite	RRMS	Mean	62
	Mental health composite	RRMS	Mean	66.9
	Mental health composite	mild spasticity	Mean	59.6
	Mental health composite	moderate spasticity	Mean	53.7
	Mental health composite	severe spasticity	Mean	48.7
Flachenecker 2013[17]	Physical health composite	mild spasticity	Mean	54.9
	Physical health composite	moderate spasticity	Mean	46
	Physical health composite	severe spasticity	Mean	39.5

Conclusions

- Over half of the patients with MS, particularly those in the middle-aged group, suffer from spasticity and these patients experience high economic costs as well as poorer quality of life, both of which increase with increasing severity of spasticity. There is a lack of high-quality studies on epidemiology and economic burden in MSS, warranting further research.

Results (continued)

Humanistic burden outcomes

- EuroQoL 5-Dimension (EQ-5D), 54-item Multiple Sclerosis Quality of Life (MSQOL-54) and Activities of Daily Living (ADL) were among the commonly reported humanistic burden measures. This SLR found poorer health-related quality of life (HRQoL) with increasing spasticity severity.
- EQ-5D[15] is used to measure mobility, self-care, usual activities, pain/discomfort, and anxiety/depression. EQ-5D index score (0=death and 1=full health) and EQ-VAS [(self-reported measure of health and well-being) score of 0=worst health state and 100=best health state] were reported by six studies.
 - Two cross-sectional studies reported EQ-5D index scores.[16,17] One study reported mean index scores of 0.6, 0.5, and 0.3 for mild, moderate, and severe MSS respectively.[17] The other study reported median index scores of 77.6, 83.5, and 59 for RRMS, progressive MS, and the total MS population, respectively (**Table 1**).[16]
 - Two retrospective cohort studies reported EQ-5D index scores.[18,19] One study reported mean index scores of 0.5, 0.3, and 0.03 for mild, moderate, and severe MSS, respectively.[18] The other study reported a mean EQ-5D index score of 53.3 in the general European MS population (**Table 1**).[19]
 - Four studies reported EQ-VAS scores.[16,18,20,21] First study reported a VAS change from 39 to 64 over 12 months in patients using Sativex (nabiximols) cannabinoid oromucosal spray.[20] Second study reported a significant VAS change from 49.8 to 54.3 over 3 months in patients using THC:CBD oromucosal spray.[21] Third study reported EQ-VAS scores were 58.4, 44.7, and 34.9, respectively.[18] Fourth study reported lower EQ-VAS scores in MSS patients with progressive MS (60), compared with RRMS (80), and all MS (78) (**Table 1**).[16]
- MSQOL-54, combines generic and MS-specific items and contains subscales including physical function, role limitations, pain, emotional well-being, energy, health perceptions, social function, cognitive function, health distress, overall quality of life, and sexual function.[22] MSQOL-54 subscale scores range from 0 to 100 and higher scores indicate better quality of life.
 - A retrospective cohort study reported the overall physical and mental composite scores at six months in patients with RRMS switching from interferon-beta to glatiramer acetate, as 62 and 66.9 respectively, with significant improvements vs. baseline scores (**Table 2**).[23]
 - One cross-sectional study reported the overall mental health composite scores in mild, moderate, and severe spasticity patients, respectively, as 59.6, 53.7 and 48.7. It also reported the overall physical health composite scores in mild, moderate, and severe spasticity patients as 54.9, 46.0, and 39.5, respectively (**Table 2**).[17]
- Four studies reported ADL as an outcome measure. ADL scores are used to measure an individual's ability to independently care for themselves (ADL score of 0=total dependency and 100=full independence).[24,25]
 - A retrospective cohort study found significant improvements in four of the 16 ADL items with cannabidiol oromucosal spray, and in mean total score for functional impairment (36.2 vs 20.2; p = 0.022).[26]
 - A prospective cohort study reported a mean overall Global attainment scaling (GAS) score increase from 32.1 at baseline to 43.6 at month 3 with Sativex, equivalent to a mean relative gain of +35.8%.[27]
 - A cross-sectional study found that 24 out of 25 MSS patients were easier to position in bed and their wheelchairs after intrathecal phenol injection.[28]
 - Another cross-sectional study reported that spasticity moderately or greatly interfered with stair climbing (41.1%), walking (39.7%), and sleep (34.5%), and least likely to interfere with eating (7.7%). As the severity of spasticity increased, interference with ADL also increased (Spearman rho ranging from 0.39 for eating to 0.67 for walking).[29]

Economic burden outcomes

- The mean total annual MSS treatment costs were high (e.g., EUR 114,293 in Sweden)[18] and varied across countries. In general, the direct costs (total, outpatient, and inpatient), indirect costs, and healthcare resource utilization (physiotherapy and outpatient rehabilitation) increased with increasing severity of spasticity.
- Direct costs: Three studies reported total annual direct costs,[18,30,31] three studies reported outpatient costs,[30,31,32] and two studies reported inpatient costs.[30,32]
 - Total direct costs: Mean total annual costs per patient was EUR 41,399 for mild spasticity and EUR 134,853 for severe spasticity in a Swedish study.[18] A German study reported similar trends with costs ranging from EUR 2,268 for mild spasticity to EUR 8,688 for severe spasticity.[30] A Spanish study reported mean total annual costs of treatment for MSS patients to be EUR 15,405.[31]
 - Outpatient costs: A German study[30] reported mean annual outpatient costs per patient of EUR 76 for mild spasticity, EUR 77 for moderate spasticity, and EUR 87 for severe spasticity. A Spanish study[31] reported annual per-patient costs of EUR 200 for resistant MSS. A Swedish study[32] reported costs ranging from GBP 144 (No/mild spasticity) to GBP 2,652 (severe/extremely severe spasticity).
 - Inpatient costs: One study[30] reported mean annual hospital stay costs of EUR 194 for mild spasticity and EUR 672 for severe spasticity. Another study[32] reported hospital admission costs of GBP 7 for no/mild spasticity and GBP 885 for severe/extremely severe spasticity.
- Indirect costs: Two studies reported on absenteeism.[23,30] One study reported the mean duration of sick leave (25.9 days - mild spasticity, 40.4 days - moderate spasticity, 16.2 days - severe spasticity).[30] Another study with follow up at 3 and 6 months reported that 13 patients missed work by month-3, with 5 due to spasticity, while by month-6, 9 patients missed work but none due to spasticity.[23] Further, absenteeism at work due to illness was 26.5 days over 6 months, and higher for patients with moderate vs. mild spasticity. One study reported indirect costs by spasticity level, with mean annual costs of EUR 33,840 for mild spasticity, EUR 39,700 for moderate spasticity, and EUR 45,726 for severe spasticity.[18]
- Healthcare Resource Utilization: A German study[30] found that a lower proportion of patients with mild MSS (vs. moderate vs. severe MSS) reported home visits for physiotherapy (2.7 vs. 5.5 vs. 22.5%) and outpatient rehabilitation (0 vs. 3.1 vs. 8.0%), but a greater proportion reported conventional physiotherapy (18.9 vs. 12.9 vs. 24.5%), physiotherapy on a neurophysiological basis (78.4 vs. 81.6 vs. 70.6%), or inpatient rehabilitation (100 vs. 96.9 vs. 96%). A greater proportion of moderate spasticity patients reported use of physiotherapy on a neurophysiological basis, while a greater proportion of severe spasticity patients reported home visits for physiotherapy.

Disclosures

- This SLR was funded by Jazz Pharmaceuticals and executed by Evidinno Outcomes Research Inc.
- CD and HNV are employees of Jazz Pharmaceuticals. GN, DP and MSF are employees of Evidinno Outcomes Research Inc.

Scan this code to access this poster online. This code is not for promotional purposes.

Presented at ISPOR Europe 2023; November 12-15; Copenhagen, Denmark

Email: mfazeli@evidinno.com

Copies of this poster are for personal use only and may not be reproduced without written permission of the authors.