A Systematic Literature Review to Assess the Level of Evidence in Facioscapulohumeral Muscular Dystrophy

Poster SA74

Lianne Barnieh[1], Rachel Beckerman[1], Helena Emich[1], Katy Eichinger[2], **Adi Eldar-Lissai[3]**

[1]Maple Health Group, LLC, New York, NY, USA; [2]University of Rochester Medical Center, Rochester, NY, USA; [3]Fulcrum Therapeutics, Inc., Cambridge, MA, USA

Background

- Facioscapulohumeral muscular dystrophy (FSHD) is a rare genetic muscle disorder that manifests itself as a progressive weakening and loss of skeletal muscles.[1]
- FSHD is caused by aberrant expression of *DUX4* in skeletal muscle, which leads to death of the muscle and its replacement by fat.[2]
- Patients with FSHD lose upper limb function and mobility, often resulting in challenges performing activities of daily living, loss of independence, and chronic pain.[2]
- Currently, there are no approved therapies for FSHD and disease management focuses on supportive treatments with physical therapy.[3] As new therapies are investigated, their impact on the quality of life of patients with FSHD needs to be considered.

Objective

- This systematic literature review (SLR) assessed the existing evidence base in FSHD, including the humanistic burden of disease and patient reported outcomes (PROs) used in patients with FSHD.

Methods

- The SLR was conducted in accordance with established methodological principles.[4]
- Search strategies were executed from inception to October 11, 2022, in three electronic databases (Embase, MEDLINE, and Cochrane) with no limits on interventions, comparators, outcomes, or study design. Searches were restricted to English publications. Electronic searches were supplemented with manual searches of relevant, non-indexed conference proceedings, limited to the last three years.
- Criteria for study inclusion were adults, adolescents, or children with FSHD type 1 or type 2; studies reporting on one of the topics of interest (efficacy and/or safety; validity of outcome measures in FSHD; humanistic burden; economic burden; disease diagnosis; or disease classification); clinical trials, observational studies (including case reports and case studies), or endpoint validation studies; and English language.
- Study selection, first by title and abstract screening and then by performing full text review, was conducted by two independent reviewers, with conflicts resolved by a third reviewer.

Figure 1. PRISMA Flow Diagram

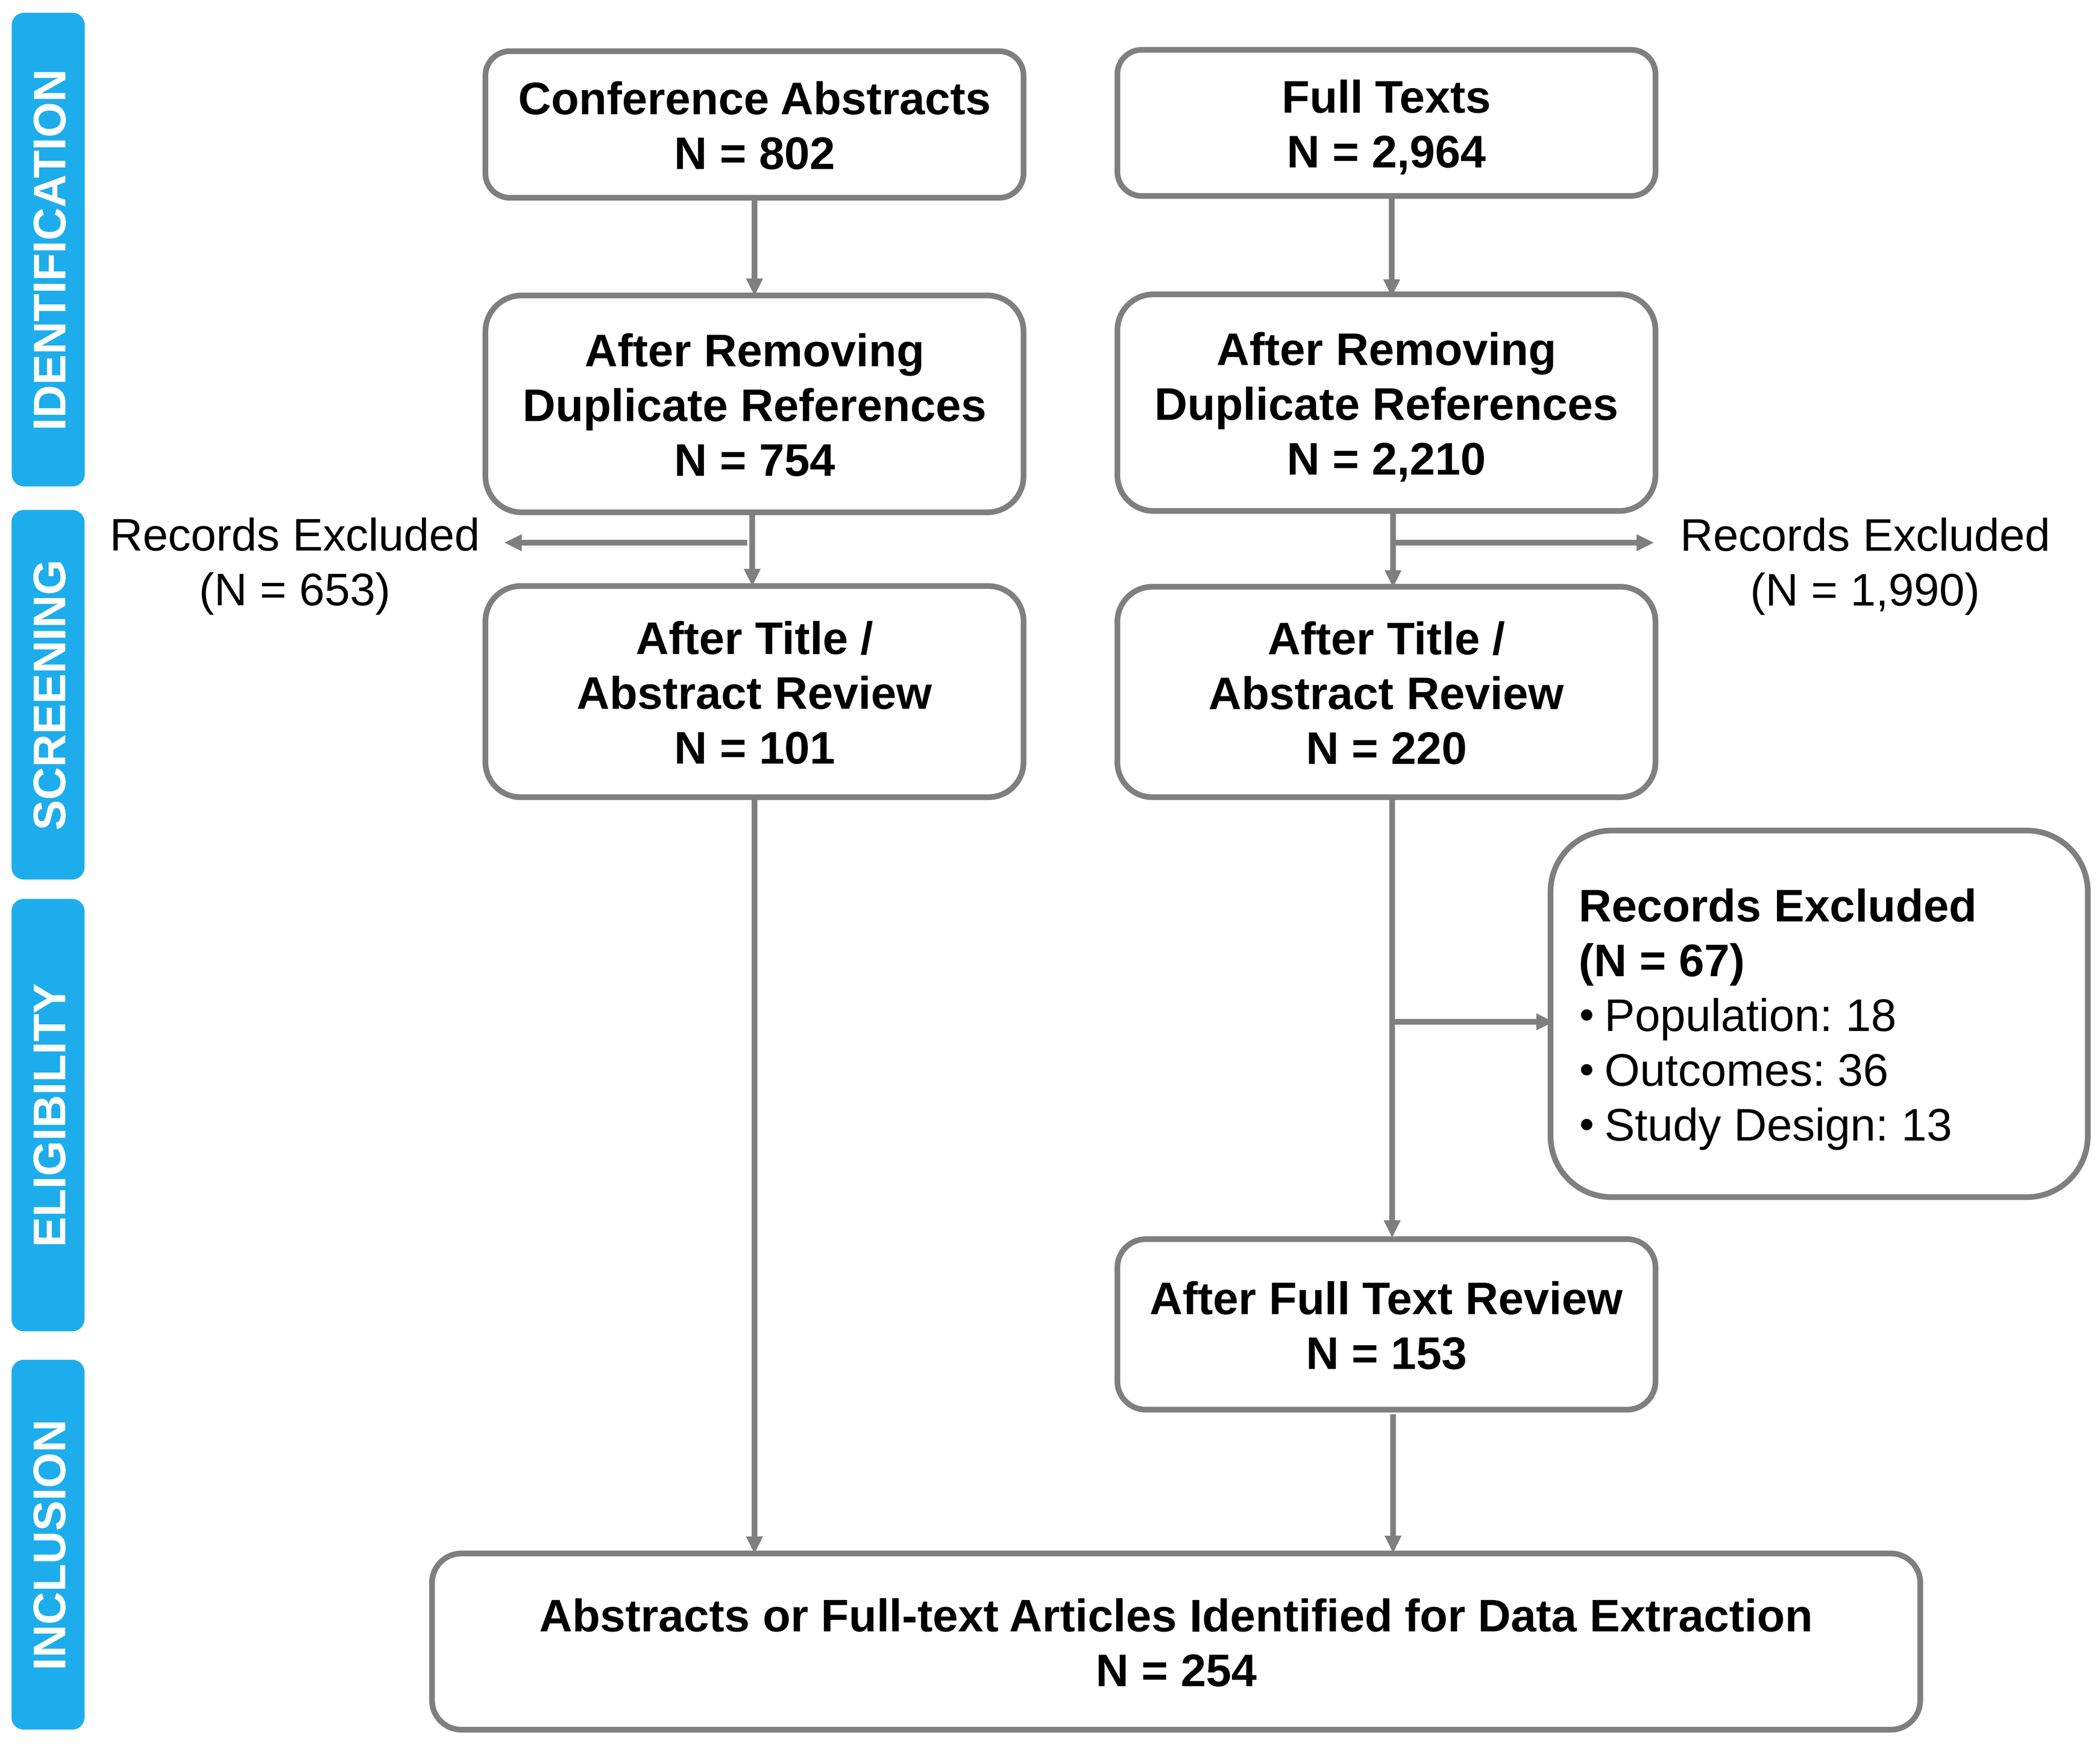
IDENTIFICATION
SCREENING
ELIGIBILITY
INCLUSION
Conference Abstracts
N = 802
Full Texts
N = 2,964
After Removing Duplicate References
N = 754
After Removing Duplicate References
N = 2,210
Records Excluded
(N = 653)
Records Excluded
(N = 1,990)
After Title / Abstract Review
N = 101
After Title / Abstract Review
N = 220
Records Excluded
(N = 67)
• Population: 18
• Outcomes: 36
• Study Design: 13
After Full Text Review
N = 153
Abstracts or Full-text Articles Identified for Data Extraction
N = 254

Results

- The database and manual searches yielded 2,211 full text articles and 754 conference abstracts after de-duplication. A total of 101 conference abstracts and 153 full text articles met the inclusion criteria for data extraction (**Figure 1**).
- The most reported topics included "Outcome measures and validation" (n=75), "Treatment outcomes and clinical trials (n=67), and "Disease classification" (n=47) (**Figure 2**). A total of 45 publications reported on "Humanistic burden and PROs" (n=30 full texts and n=15 conference abstracts). Please note that numbers do not sum to 254 as some studies reported on two or more topics.

Figure 2. Topics Included in Full Text Articles and Conference Abstracts

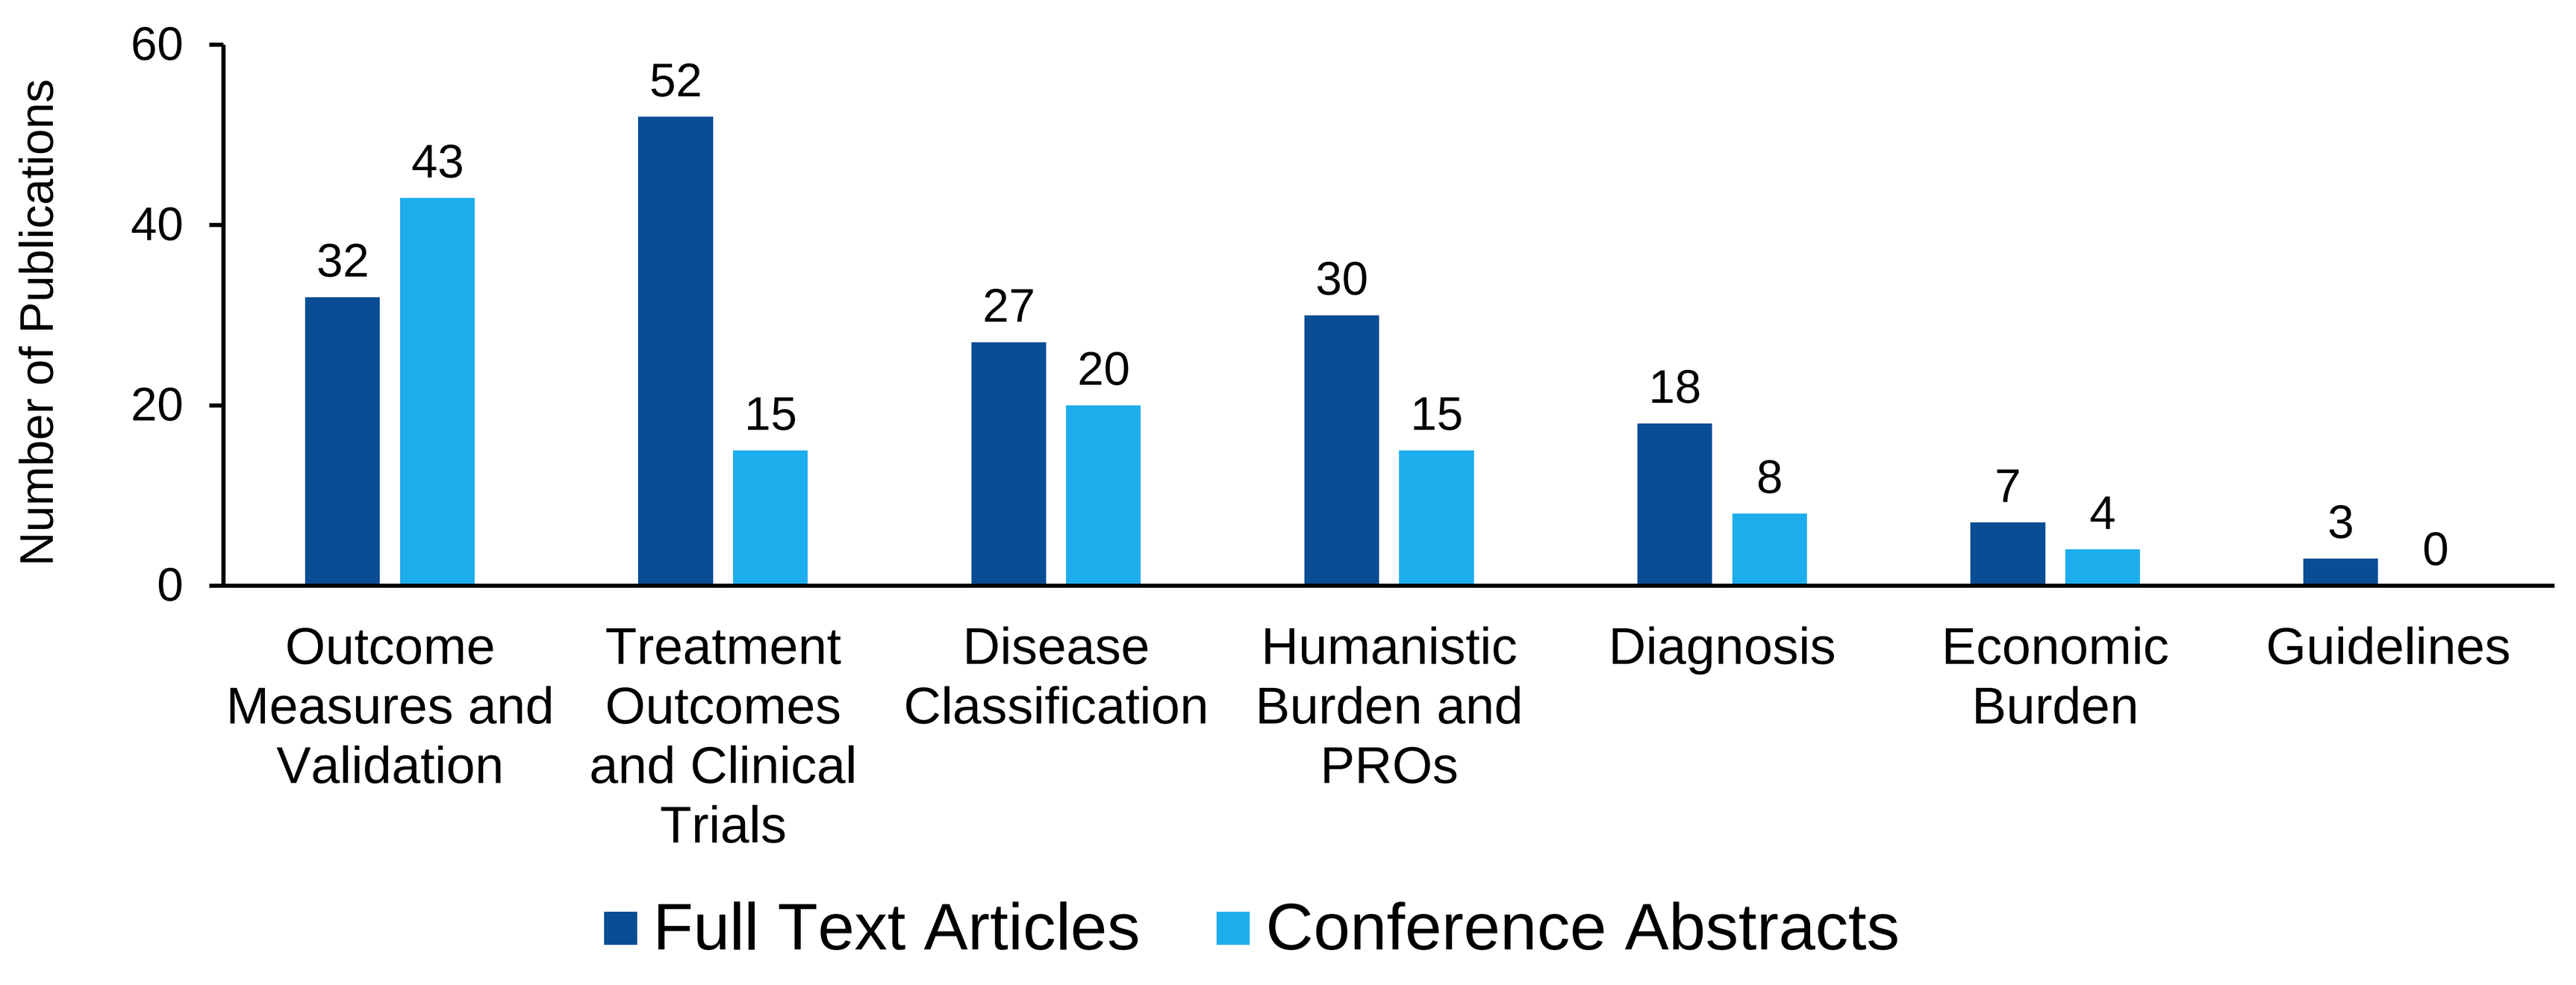
Number of Publications
60
40
20
0
32
43
52
15
27
20
30
15
18
8
7
4
3
0
Outcome Measures and Validation
Treatment Outcomes and Clinical Trials
Disease Classification
Humanistic Burden and PROs
Diagnosis
Economic Burden
Guidelines
Full Text Articles
Conference Abstracts

- Among studies included in the topic "Humanistic burden and PROs", the majority of the studies were cross-sectional (n=30); sample size ranged from 4 to 690 patients. FSHD patients included in "Humanistic burden and PRO" studies were primary of type 1 FSHD, only two studies included individuals with type 2 FSHD. Multiple aspects of humanistic burden were reported in the literature. The total number of each PRO outcome reported, by domain, is shown in **Figure 3.** The most reported outcome was depression / anxiety followed by general quality of life (QoL).

Figure 3. Number of times each PRO outcome was reported in the published FSHD literature

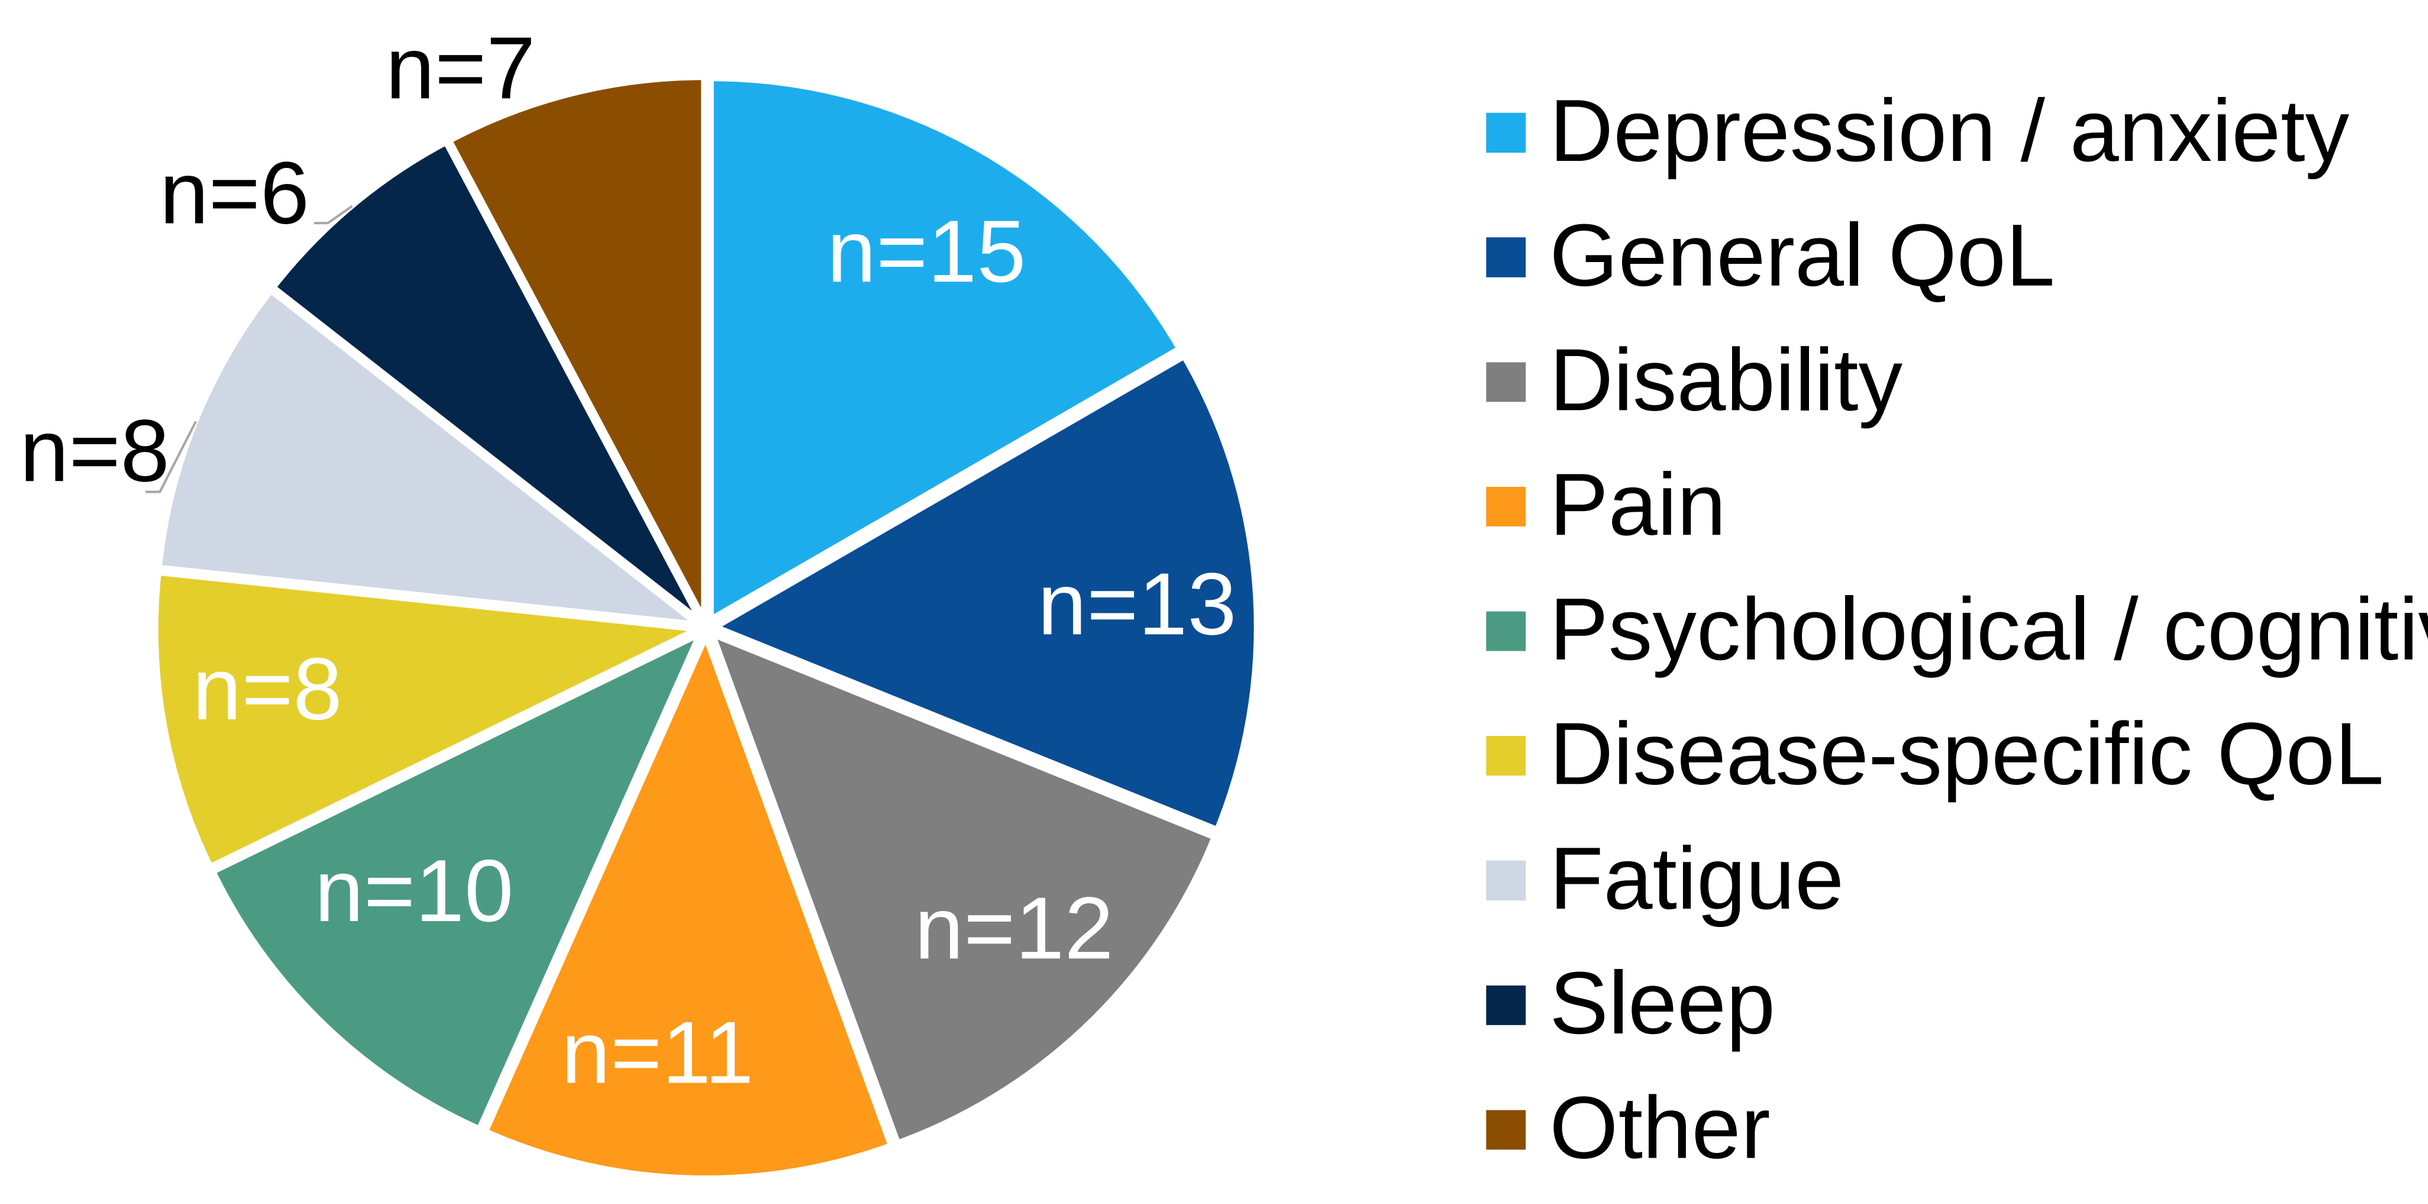
n=15
n=13
n=12
n=11
n=10
n=8
n=8
n=6
n=7
Depression / anxiety
General QoL
Disability
Pain
Psychological / cognitive
Disease-specific QoL
Fatigue
Sleep
Other

- Multiple instruments were used to assess humanistic burden across similar domains. Across the 45 studies included, a total of 53 different instruments were used to assess PRO outcomes. **Figure 4** displays the number of different instruments used for each PRO outcome identified in the FSHD literature.

Figure 4. Number of different instruments used, by domain, in the published FSHD literature

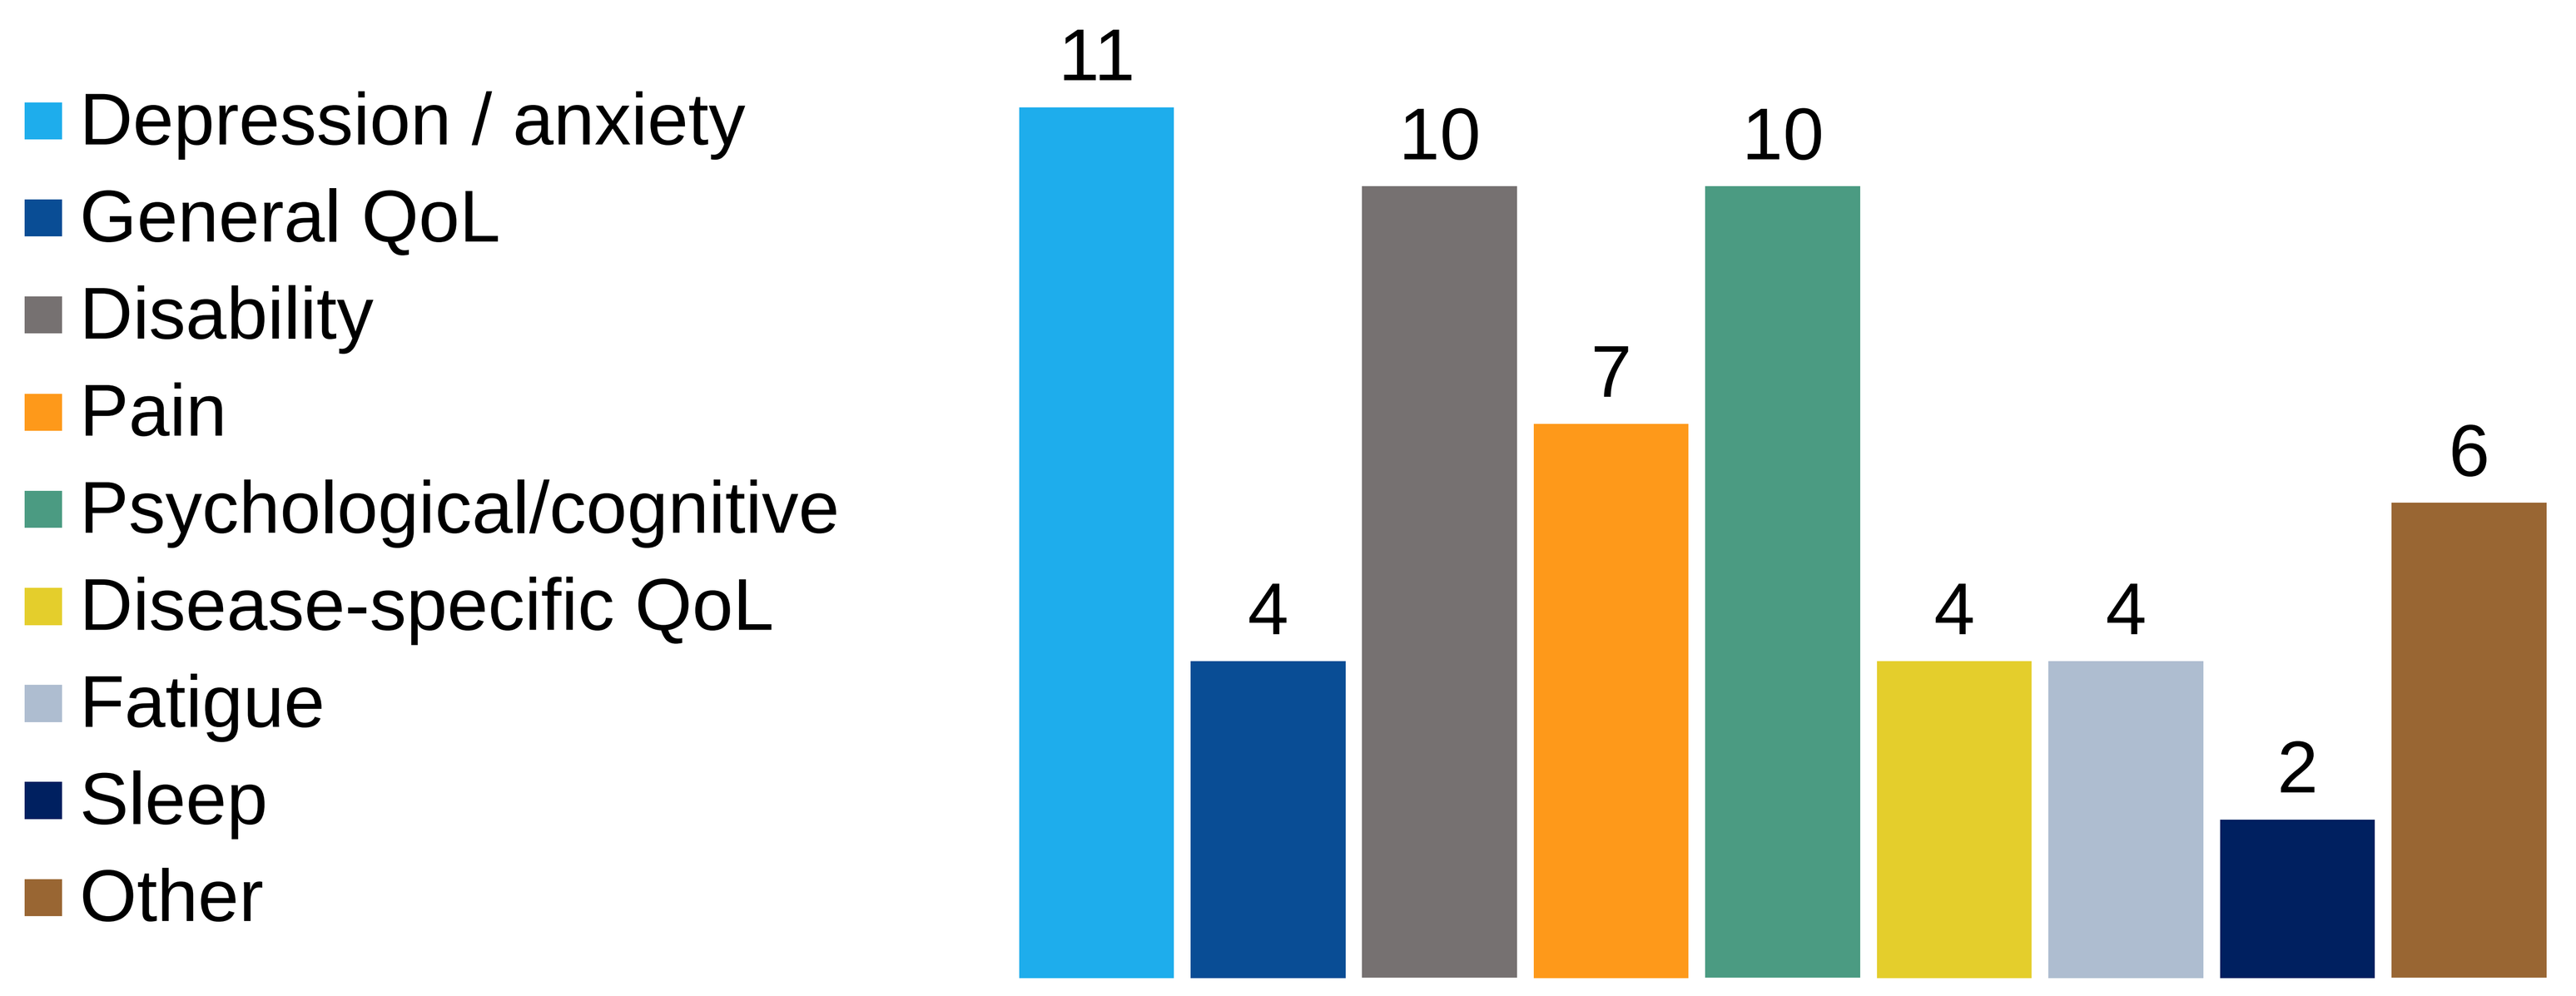
Depression / anxiety
General QoL
Disability
Pain
Psychological/cognitive
Disease-specific QoL
Fatigue
Sleep
Other
11
4
10
7
10
4
4
2
6

Conclusion

- **Patients with FSHD experience multidimensional detriments on quality of life, including depression, anxiety, disability and pain.**
- **There is a lack of consensus on instruments used to assess this humanistic burden among FSHD patients.**
- **Efforts should be maintained on harmonizing assessments on the impact of the debilitating and progressive nature of FSHD, as research on treatment options continues.**

References

1. Huml *et al.*, *Ther Innov Regul Sci* 2020; 54(1): 144-50.
2. FULCRUM Therapeutics. About FSHD. 2022. https://www.fulcrumtx.com/therapeutic-focus/fshd/.
3. Preston *et al.,* Facioscapulohumeral Muscular Dystrophy. In: [Internet] G, editor.; 2020.
4. University of York Centre for Reviews and Dissemination. Systematic Reviews: CRD's guidance for undertaking reviews in health care, 2008.

Abbreviations: FSHD = facioscapulohumeral muscular dystrophy; PRO = patient-reported outcome; QoL = quality of life; SLR = systematic literature review

Disclosures: AE-L is a current employee and shareholder at Fulcrum Therapeutics. KE has served on advisory boards or has received consulting fees from Biogen, Acceleron, PTC Therapeutics, Ionis Pharmaceuticals, Fulcrum Therapeutics, Dyne Therapeutics, Avidity Biosciences, and Roche. Maple Health Group received consulting fees for this work.