FINANCIAL-BASED MANAGED ENTRY AGREEMENTS FOR DRUGS IN ROMANIA – FROM SIMPLE TO COMPLEX

HPR165

Radu CP[1], Dragoi L[2], Udroiu MA[2]

[1]Department of Public Health, University of Medicine and Pharmacy Carol Davila, Bucharest, Romania; [2]Roche Romania SRL, Bucharest, Romania

From **2015** to **2022** the number of **Financial-Based Managed Entry Agreements** (FBMEA) drugs has increased from **4** to **73**, and the number of therapeutic areas with FBMEA drugs has risen from **1** to **11.**

In the same period, the number of pacients treated with FBMEA drugs went up from **932** to **247,418.**

Number of FBMEA drugs for each therapeutic area

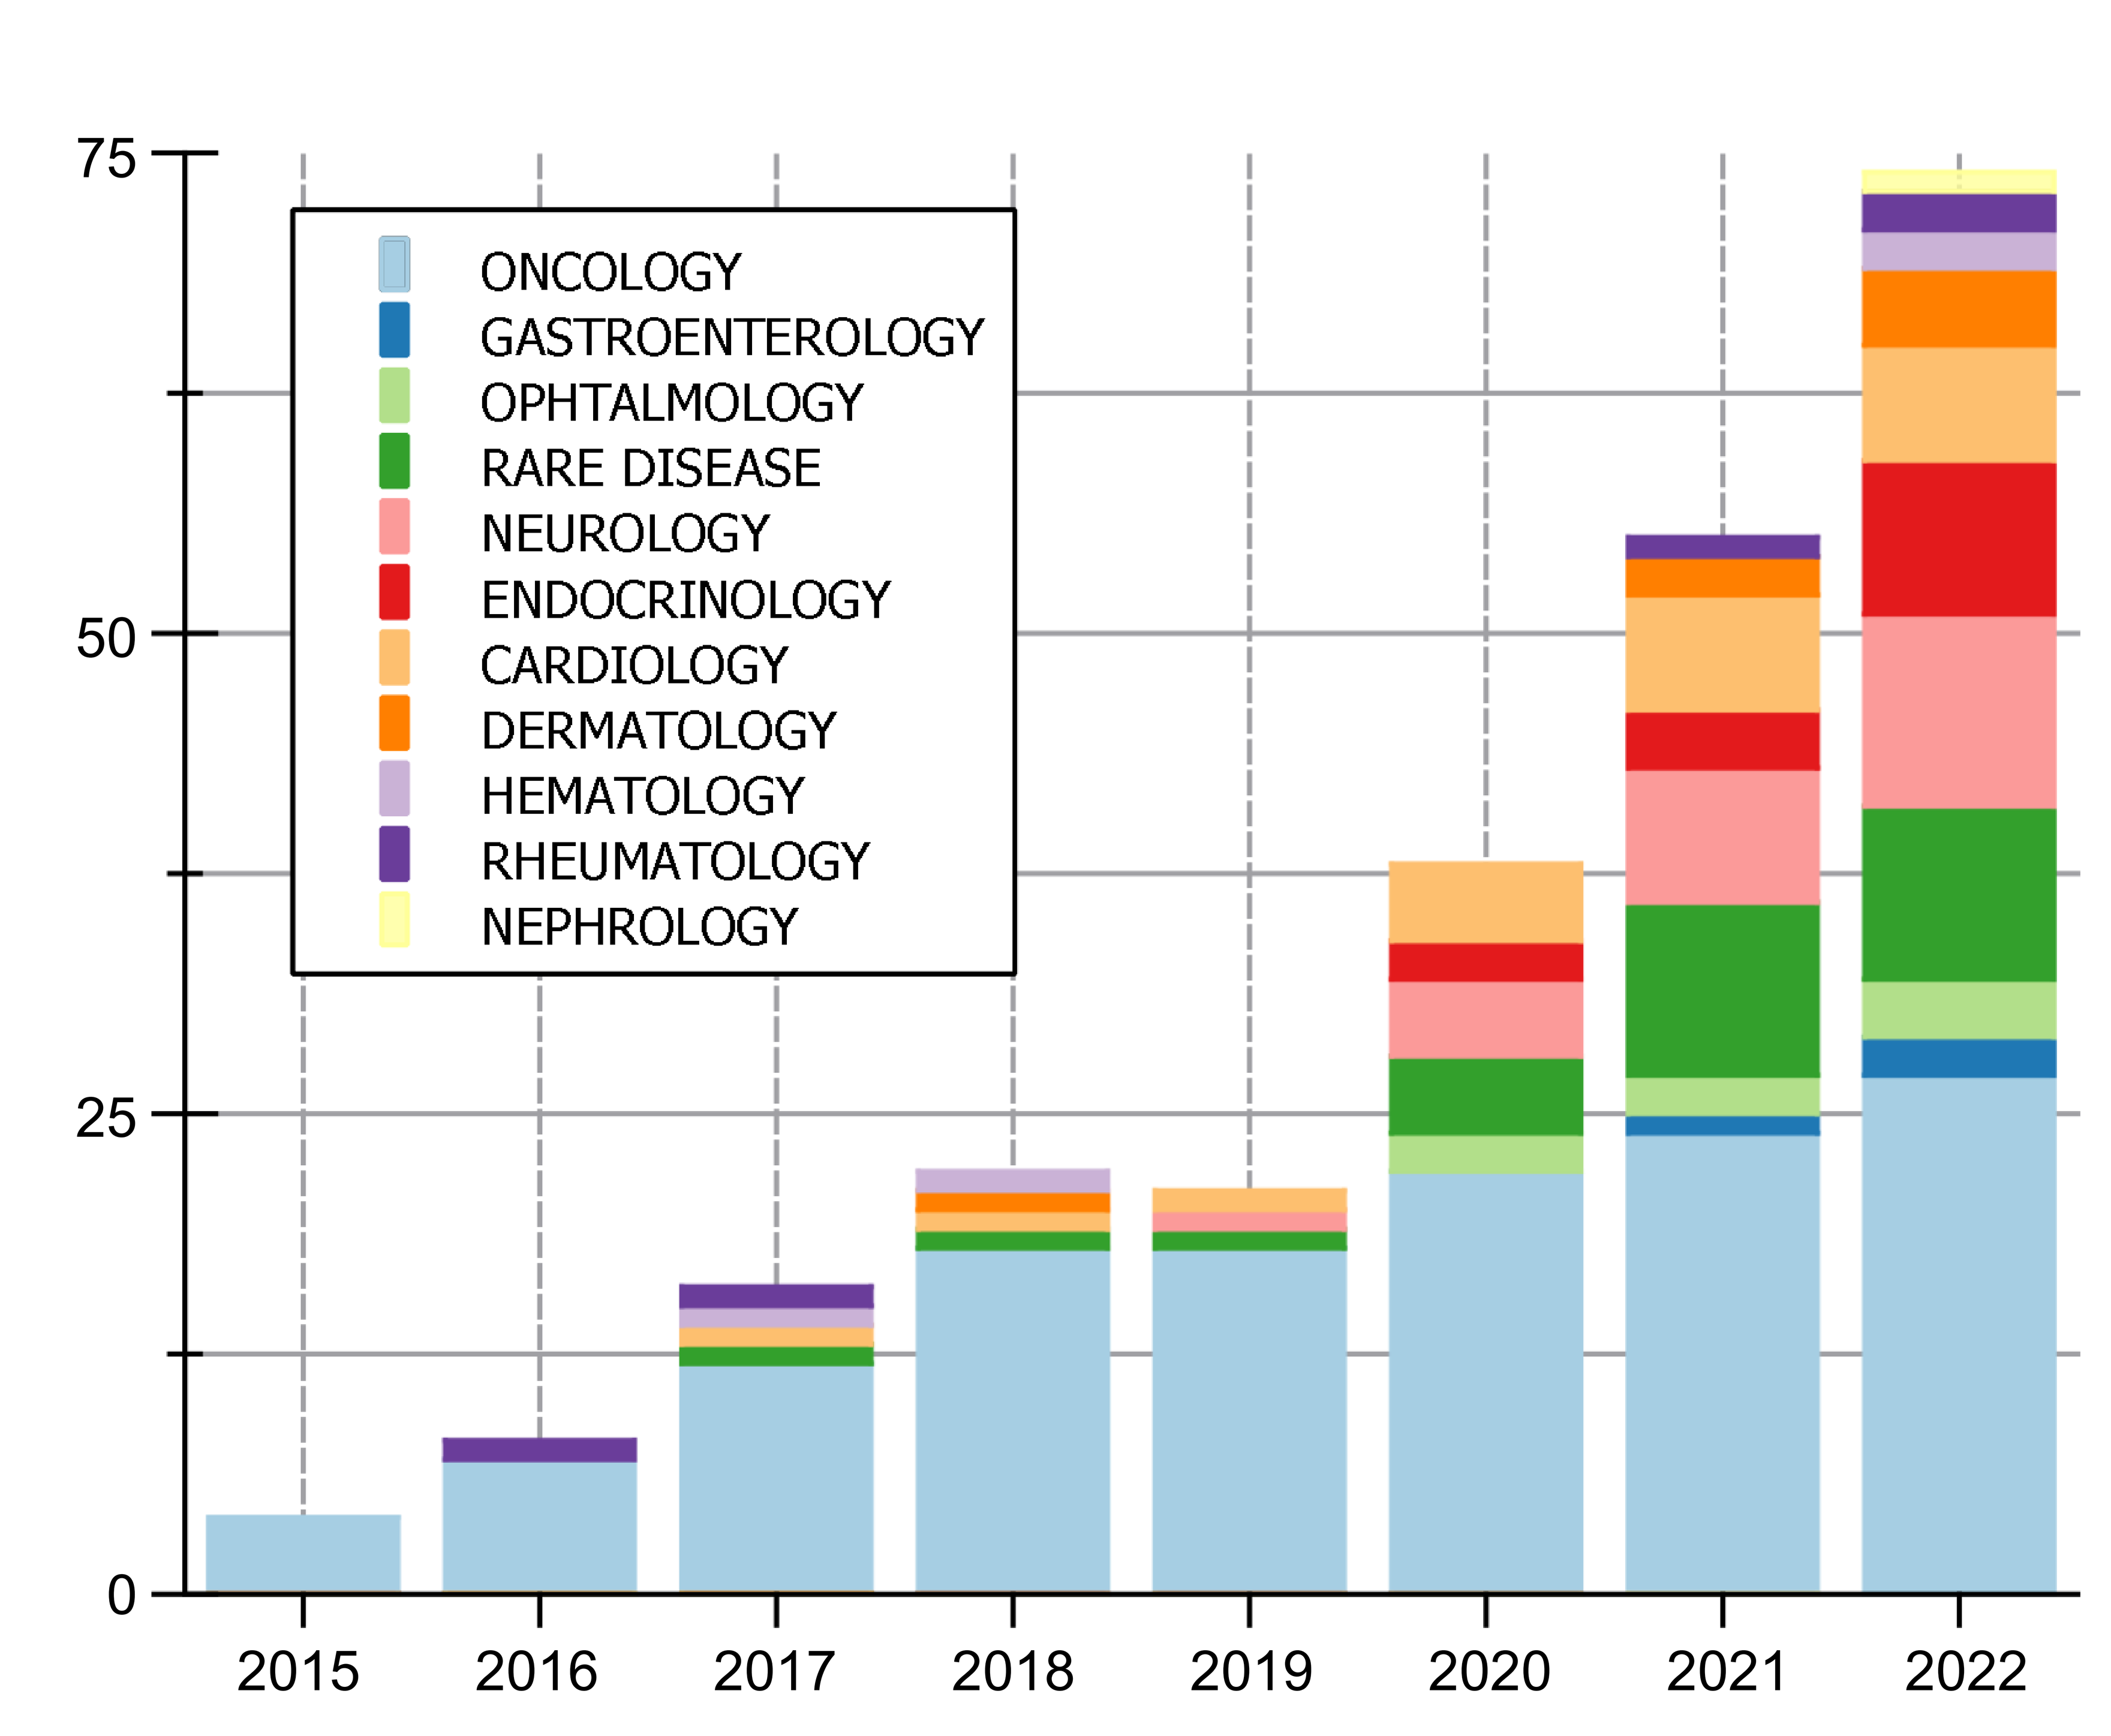
75
50
25
0
ONCOLOGY
GASTROENTEROLOGY
OPHTALMOLOGY
RARE DISEASE
NEUROLOGY
ENDOCRINOLOGY
CARDIOLOGY
DERMATOLOGY
HEMATOLOGY
RHEUMATOLOGY
NEPHROLOGY
2015
2016
2017
2018
2019
2020
2021
2022

OBJECTIVES: The objective of this research is to present the outcomes of the FBMEA in Romania performed during 8 years of implementation.

METHODS:

- evolution of the legislation on FBMEA
- analysis of the existing FBMEA in Romania looking at public data sources from 2015 to 2022
- outcomes of FBMEA development in terms of number of drugs reimbursed, therapeutic areas covered, treated patients and the resources allocated

RESULTS: In Romania, the Managed Entry Agreements (MEA) are performed as Cost-Volume contracts (financial-based contracts with discounts based on market share - FBMEA) or Cost-Volume-Result contracts (outcome-based contracts - OBMEA).

	2015		2022
Drugs reimbursement via FBMEA contracts	4	↗	73
Number of patients treated via FBMEA contracts	932	↗	247,418
FBMEA budget (EUR)	5.3 mil	↗	426 mil.
Share of FBMEA buget of total drugs with public reimbursement	0.29%	↗	13.30%

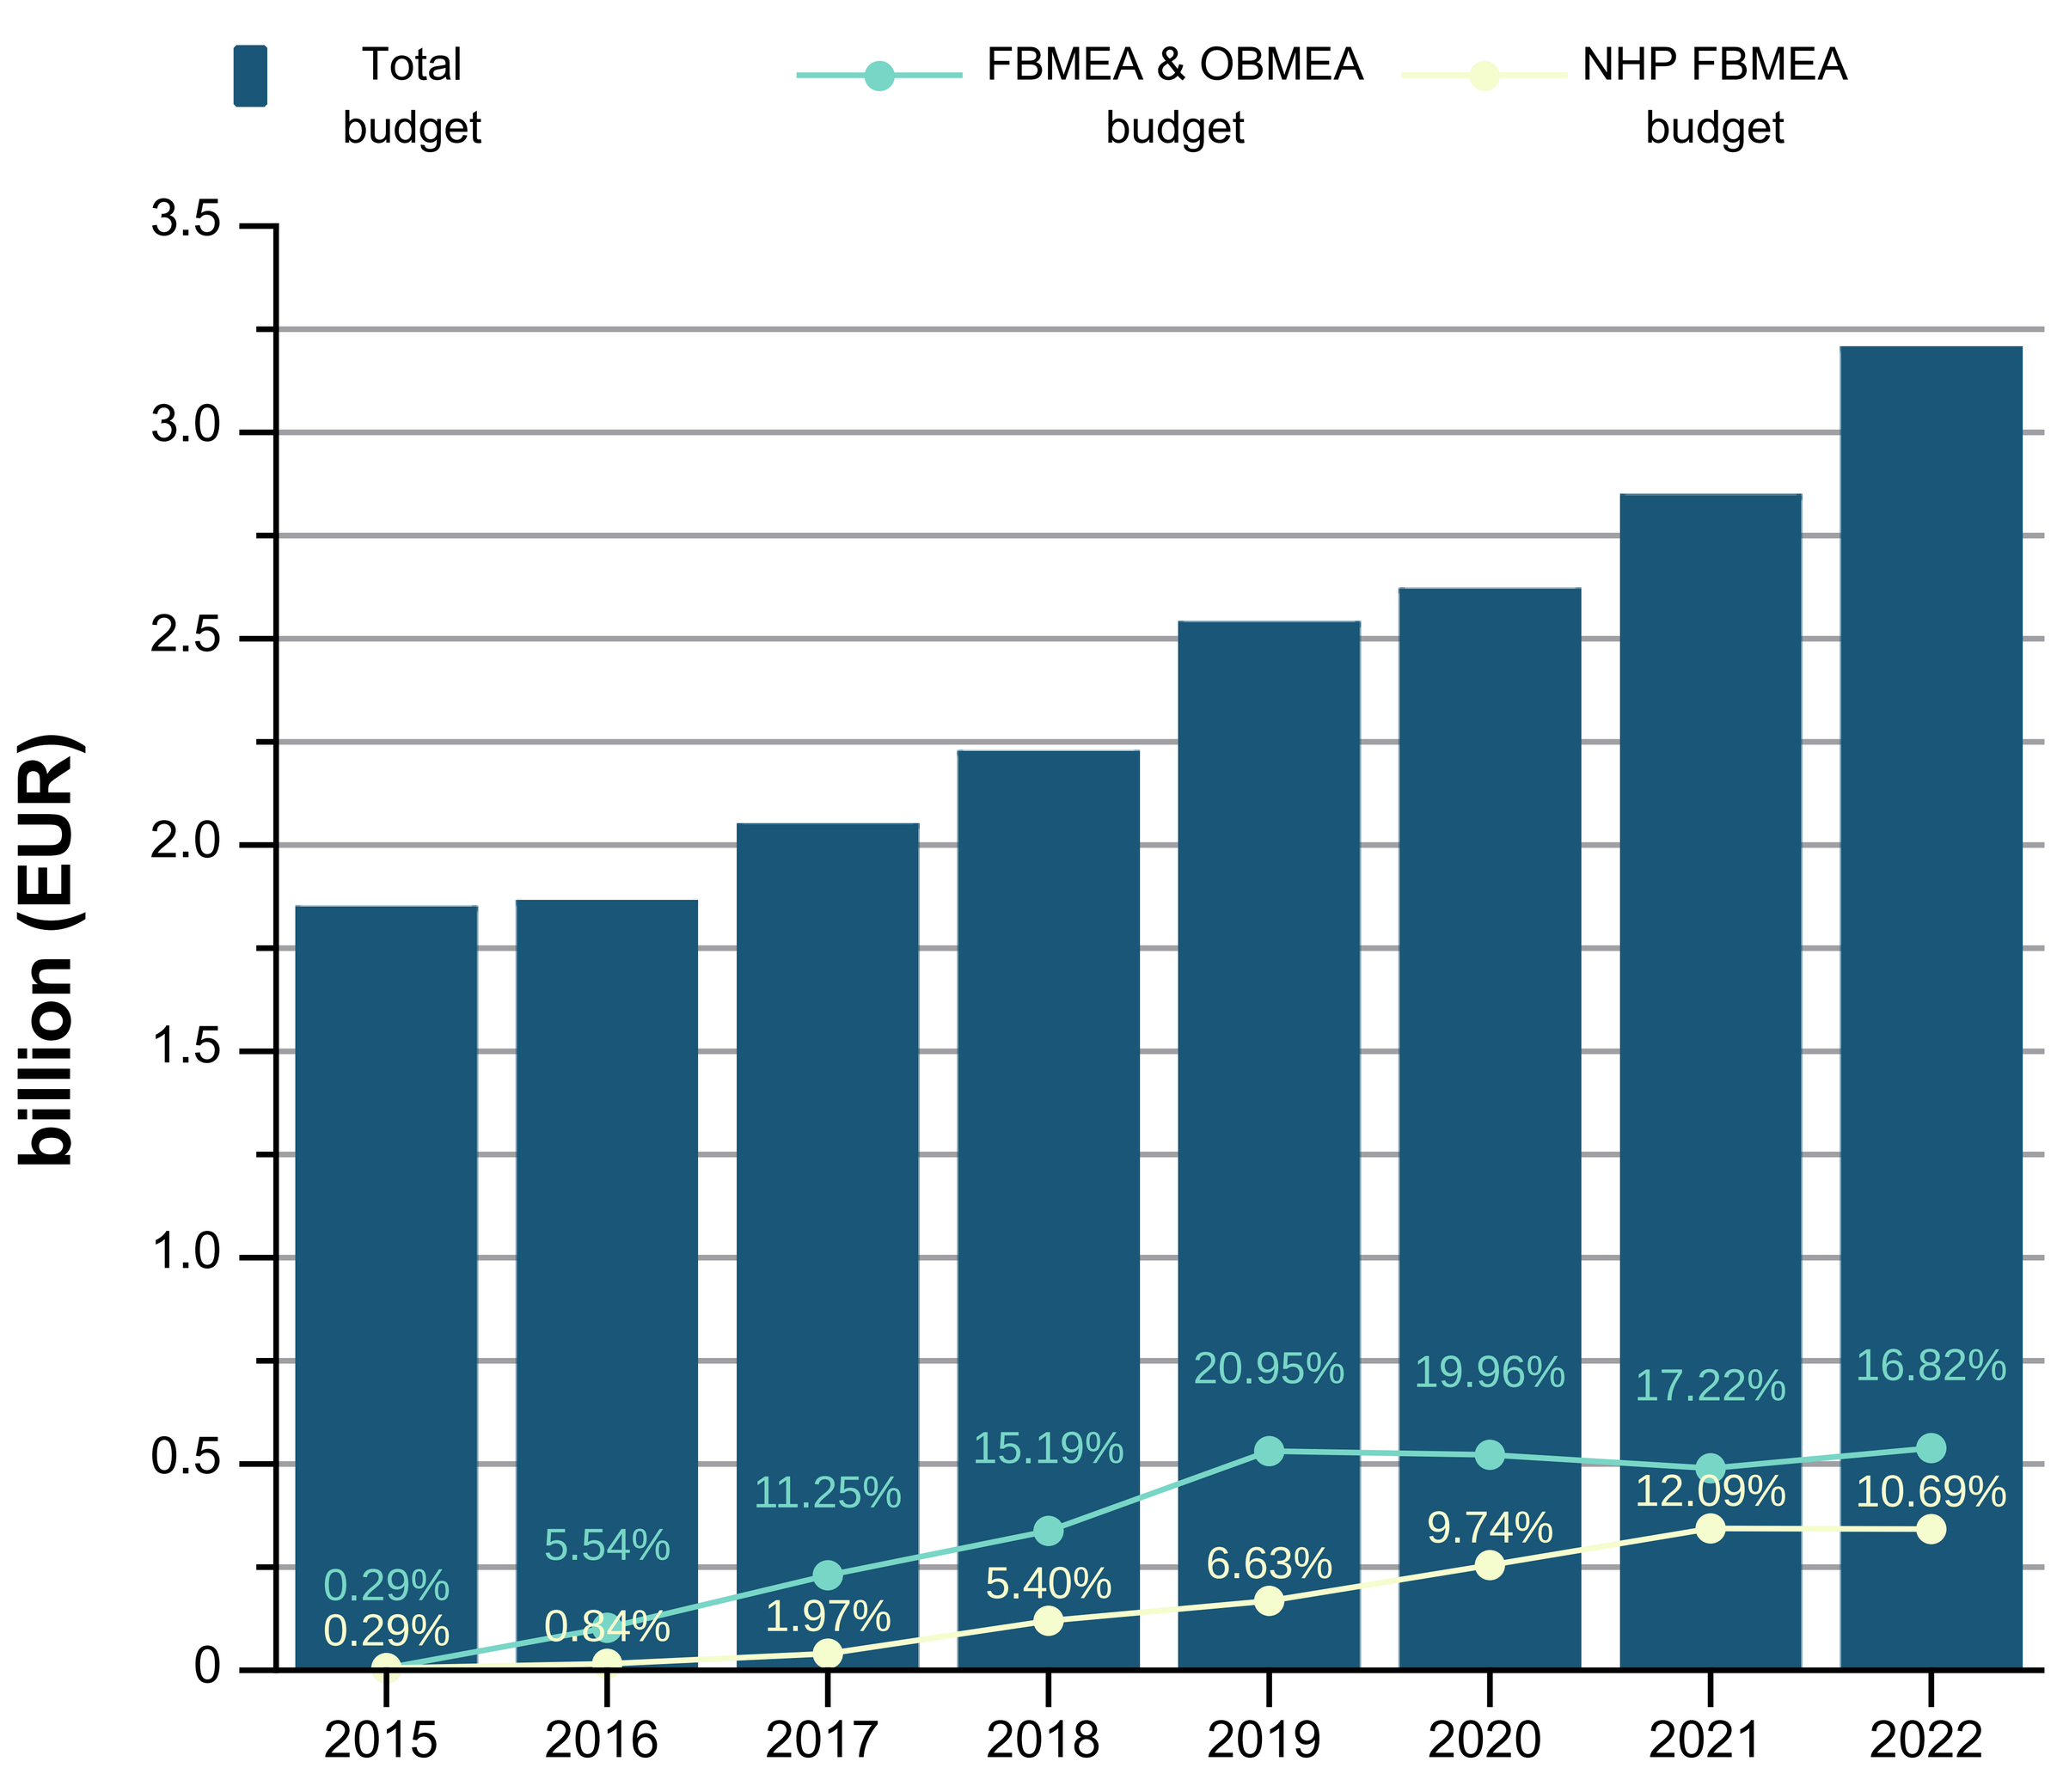
Total budget
FBMEA & OBMEA budget
NHP FBMEA budget
billion (EUR)
3.5
3.0
2.5
2.0
1.5
1.0
0.5
0
0.29%
0.29%
5.54%
0.84%
11.25%
1.97%
15.19%
5.40%
20.95%
6.63%
19.96%
9.74%
17.22%
12.09%
16.82%
10.69%
2015
2016
2017
2018
2019
2020
2021
2022

Budget analysis

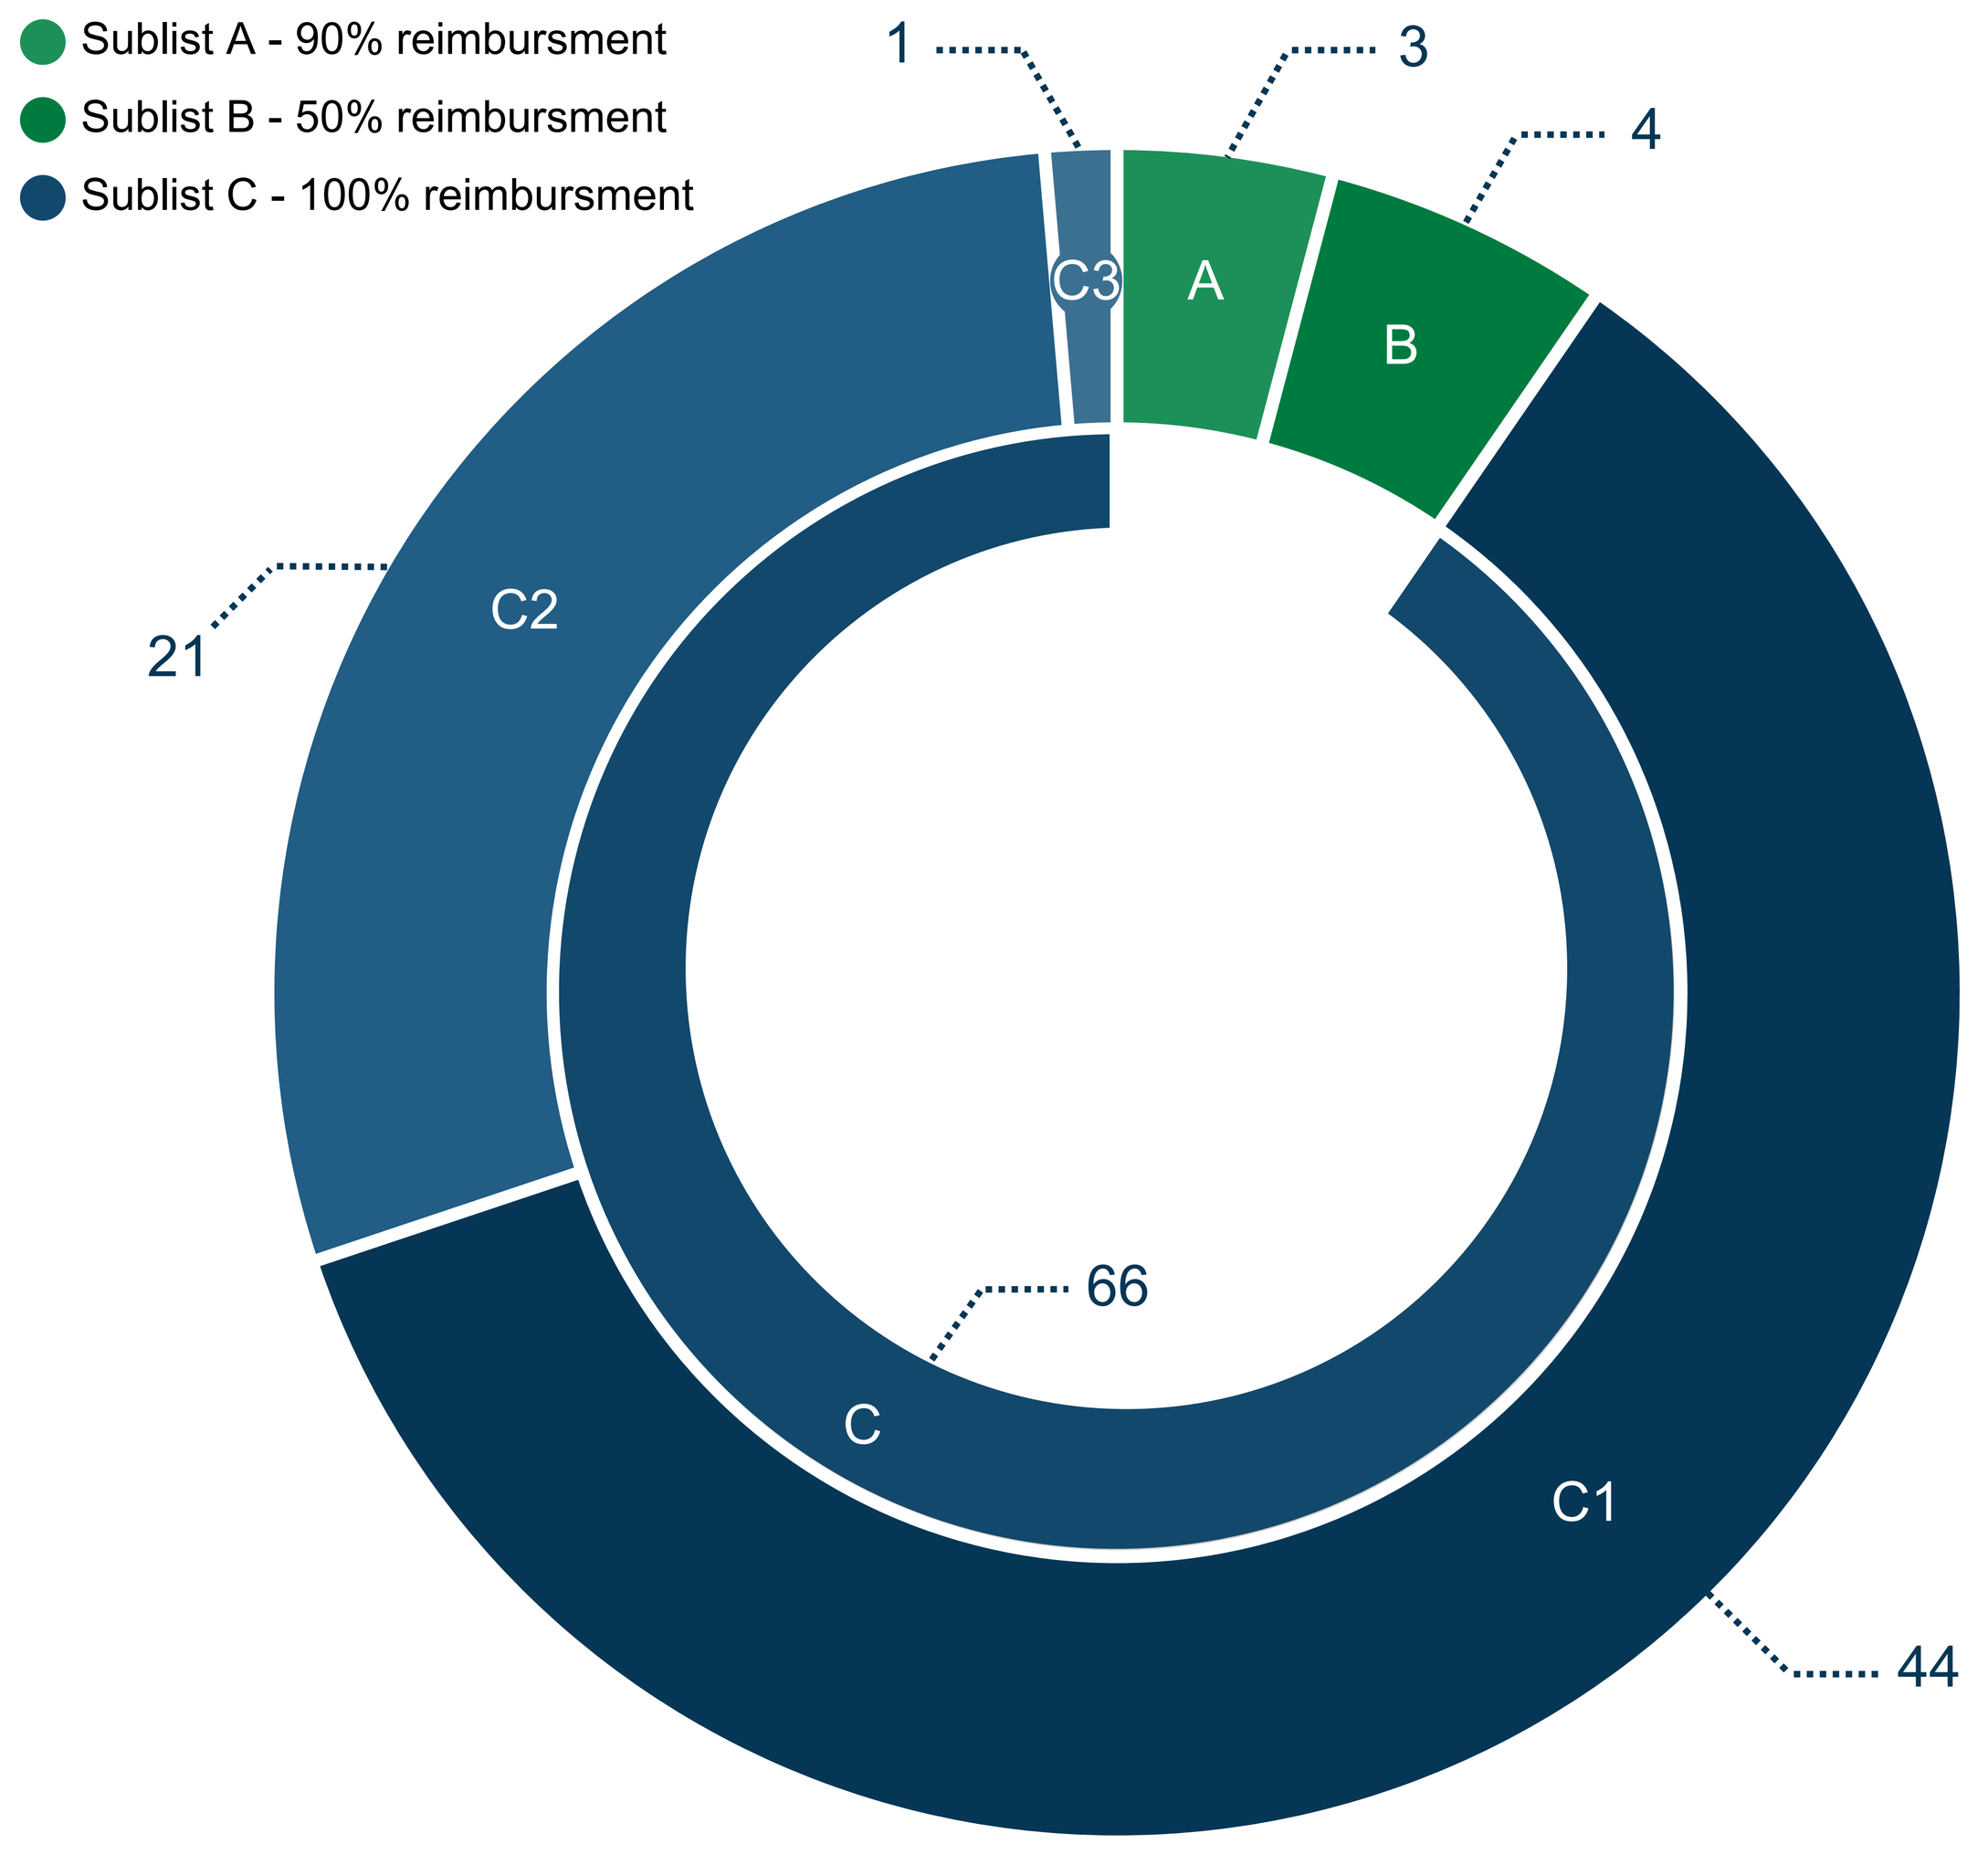
Sublist A - 90% reimbursment
Sublist B - 50% reimbursment
Sublist C - 100% reimbursment
1
3
4
21
66
44
C3
A
B
C2
C
C1

Number of FBMEA drugs per type of reimbursement list

CONCLUSIONS:

In Romania, the FBMEA is an important pathway to public reimbursement of new drugs, especially in areas like Oncology and Neurology. There are still some problems for adjustment, but the FBMEA proved to be a solid ground for the reimbursement of new drugs. The future steps for FBMEA are already envisaged in terms of Paying for Performance mechanisms, which could bring additional benefits for the health care system.

1. National Health Insurance House website (www.cnas.ro), available online: https://cnas.ro/wp-content/uploads/2023/06/evol-buget.pdf (accessed on 30. August 2023); 2. Government Decision No. 720/2008, Published in Official Gazette No. 523/2008. Available online: https://legislatie.just.ro/Public/DetaliiDocumentAfis/95556 (accessed on 14 September 2023); 3. Ministry of Health Order No. 861/2014 Published in Official Gazette No. 557/2014. Available online: https://legislatie.just.ro/Public/DetaliiDocumentAfis/160204 (accessed on 14 September 2023); 4. Carlson, J.J.; Sullivan, S.D.; Garrison, L.P.; Neumann, P.J.; Veenstra, D.L. Linking payment to health outcomes: A taxonomy and examination of performance-based reimbursement schemes between healthcare payers and manufacturers. Health Policy 2010, 96, 179–190. https://doi.org/10.1016/j.healthpol.2010.02.005; 5. Government Emergency Ordinance No. 77/2011 Published in Official Gazette No. 680/2011. Available online: https://legislatie.just.ro/Public/DetaliiDocumentAfis/131707 (accessed on 22 September 2023); 6. NHIH Activity Report for 2022. Available online: https://cnas.ro/wp-content/uploads/2023/06/Raport-de-activitate-CNAS-2022.pdf (accessed on 29 September 2023); 7. Ministry of Health Order No. 735/976/2018, Published in Official Gazette No. 487/2018. Available online: https://legislatie.just.ro/Public/DetaliiDocument/216470 (accessed on 25 September 2023).

ISPOR Europe 2023 Conference, November 12-15, Copenhagen, Denmark