Poster number: HTA304

Presented at ISPOR Europe 2023, 12–15 November 2023, Copenhagen, Denmark

The impact of different HTA frameworks on time to patient access: a case study assessing the first commercial launch indications for five oncology medicines

Barry Crean[1], David Parry[1], Alison Horsfield[1], James Ryan[1] and Nektarios Oraiopoulos[2]

[1]AstraZeneca, Cambridge, UK; [2]University of Cambridge, Cambridge, UK

Objectives

- Assess the health technology assessment (HTA) processes in six different countries for five AstraZeneca cancer medicines following regulatory approval.
- Quantify the time from regulatory approval to patient access, and the subsequent impact on patients.
- Identify potential improvements for the medicine access pathway when long-term clinical benefit is uncertain.

Conclusions

- Time to patient access for five cancer medicines across six countries was dependent on the duration of the HTA process, which varied widely despite the evidence package for each medicine being the same in each country.
 - Most HTA processes were longer than the 6-month European Commission target.
- Prolonged time to access led to potential life-years lost for patients who were unable to benefit from these medicines during the HTA process.
 - This was emphasized by the long-term data confirming that these medicines lead to meaningful survival benefits.
- During periods of data uncertainty (e.g. immature survival data), coverage with evidence development, including outcome-based access agreements between the health technology developer and payers, can accelerate access and minimize lost outcome opportunities.
- Reimbursement systems must continue to evolve and align with scientific advances to realize the benefit of new medicines for health technology developers, regulators, payers and, most importantly, patients, in a timely manner.
 - Delays also have implications for competition and Europe as an innovation hub because small health technology developers may be unable to survive the wait for revenue.

Plain language summary

Why did we perform this research?

- After a new medicine has been approved by regulators, many countries use health technology assessments (HTAs) to decide if or how much they will pay for the medicine.
 - In countries with an HTA process, patients must wait for governments to agree to pay for medicines before they can use the medicine – we can think of this as an **HTA time gap**.
- We wanted to assess the HTA process in Spain, Italy, France, Germany, England and Canada, and the impact it has on patients.

How did we perform this research?

- We calculated the HTA time gap for five cancer medicines.
- We looked at how these medicines improved survival.
- We estimated how many years of life were lost for patients because of the HTA time gap.

What were the findings of this research?

- On average, the HTA time gap was over 15 months for cancer medicines – the time from approval to being routinely available to patients.
 - This was substantially longer than the 6-month target set by the European Commission.
- Across the countries and medicines analysed, over 2000 years of life were potentially lost because these medicines were not routinely available to patients during the HTA time gap.

What are the implications of this research?

- There is a compelling need to reduce the time to patient access for new medicines.
- All groups involved in healthcare access have a shared responsibility and should urgently consider how to improve time to patient access, including for new medicines with uncertainty about their long-term effects (e.g. using managed entry agreements).

Where can I access more information?

- Information about the HTA process can be found here:
 - Canada: https://www.cadth.ca/cadth-framework-patient-engagement-health-technology-assessment
 - England: https://www.nice.org.uk/about/what-we-do/our-programmes/nice-guidance/nice-technology-appraisal-guidance
 - EU: https://health.ec.europa.eu/health-technology-assessment/overview_en

Please scan this quick response (QR) code with your smartphone camera or app to obtain a copy of these materials. Alternatively, please click on the link below:

https://bit.ly/3QA1Nw4

Copies of this poster obtained through this QR code are for personal use only and may not be reproduced without permission from the authors of this poster.

Introduction

- The value of pharmaceutical innovation is only realized when patients access treatment; however, following regulatory approval, patients often have to wait for reimbursement, which is usually decided via an HTA process, before they can access the new medicine.
 - In oncology, the full long-term benefit of new medicines may not be known at the time of the HTA process.

Methods

- **Figure 1** describes our methodology.
 - The supplementary materials contain additional methodology.

Figure 1. Five AstraZeneca[a] medicines were evaluated in six countries following regulatory approval for their first launched indications

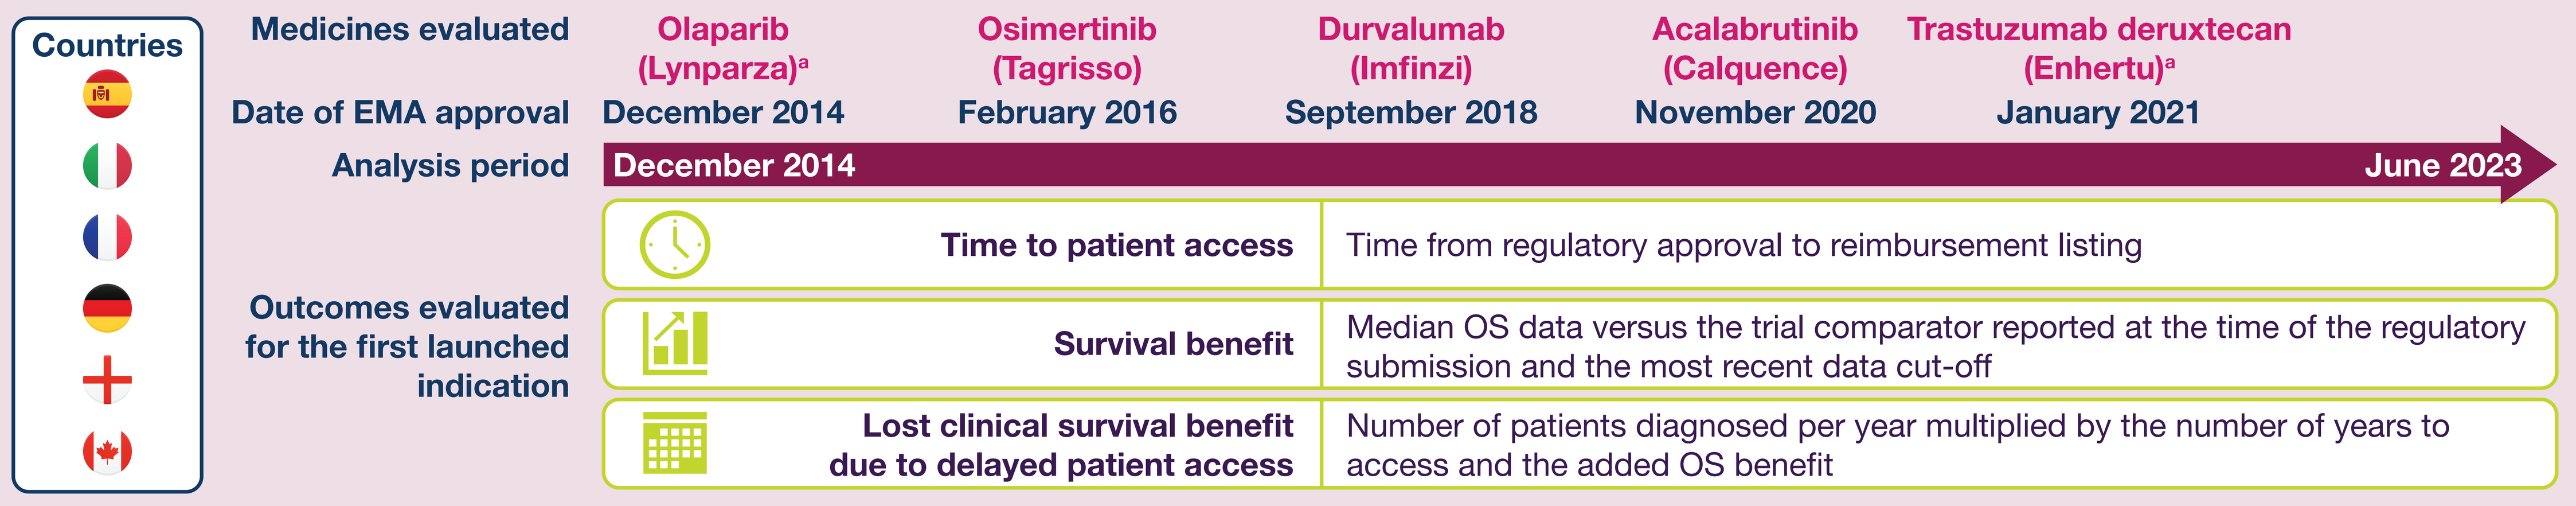
Countries
Medicines evaluated
Olaparib (Lynparza)a
Osimertinib (Tagrisso)
Durvalumab (Imfinzi)
Acalabrutinib (Calquence)
Trastuzumab deruxtecan (Enhertu)a
Date of EMA approval
December 2014
February 2016
September 2018
November 2020
January 2021
Analysis period
December 2014
June 2023
Outcomes evaluated for the first launched indication
Time to patient access
Time from regulatory approval to reimbursement listing
Survival benefit
Median OS data versus the trial comparator reported at the time of the regulatory submission and the most recent data cut-off
Lost clinical survival benefit due to delayed patient access
Number of patients diagnosed per year multiplied by the number of years to access and the added OS benefit

Note: early access schemes in France and England were not included in this analysis because patients' access to medicines via early access schemes was restricted based on specific criteria and data on how many patients were able to access the medicines were not available.
[a]Olaparib was a joint venture with Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA, and trastuzumab deruxtecan was a joint venture with Daiichi Sankyo Company, Limited.
EMA, European Medicines Agency; OS, overall survival.

Results

Time to patient access

- **Figure 2A** shows the time to patient access.
 - As of June 2023, trastuzumab deruxtecan had not yet been submitted in Italy or Canada or reimbursed in France for its first regulatory-approved indication, so time to access is likely to have been underestimated.
 - In Germany, the HTA submission for trastuzumab deruxtecan was delayed until confirmatory trial data were routinely available because the HTA in Germany has a low acceptance of indirect comparisons using single-arm trial data.
- Some countries have adopted pathways to avoid or reduce the delay to patient access during HTAs (see supplementary materials).

- Reasons for long HTA processes include delays in starting the HTA and the speed of the process.
 - This is often linked to different evidence requirements versus regulators, particularly related to uncertainty over long-term outcomes.[1]

Survival benefit

- The survival benefit is the difference in median overall survival (OS) between the medicine and the control arm (**Figure 2B**).
 - This was estimated using adjusted synthetic control arms for single-arm trials (i.e. for DB01).

Lost clinical survival benefit

- **Figure 2C** shows the potential loss in overall life-years caused by the increased time to patient access.
- Our results may be uncertain, related to the following factors.
 - Real-world outcomes estimated in our study were assumed to match those seen in both arms of the clinical trials.
 - All eligible patients were assumed to have received the medicine.
 - Treatment switching within the trials may have led to an underestimation of the survival benefit.

Figure 2. (A) Time to patient access, (B) median OS benefit versus comparator and (C) population life-years potentially lost because of time to access

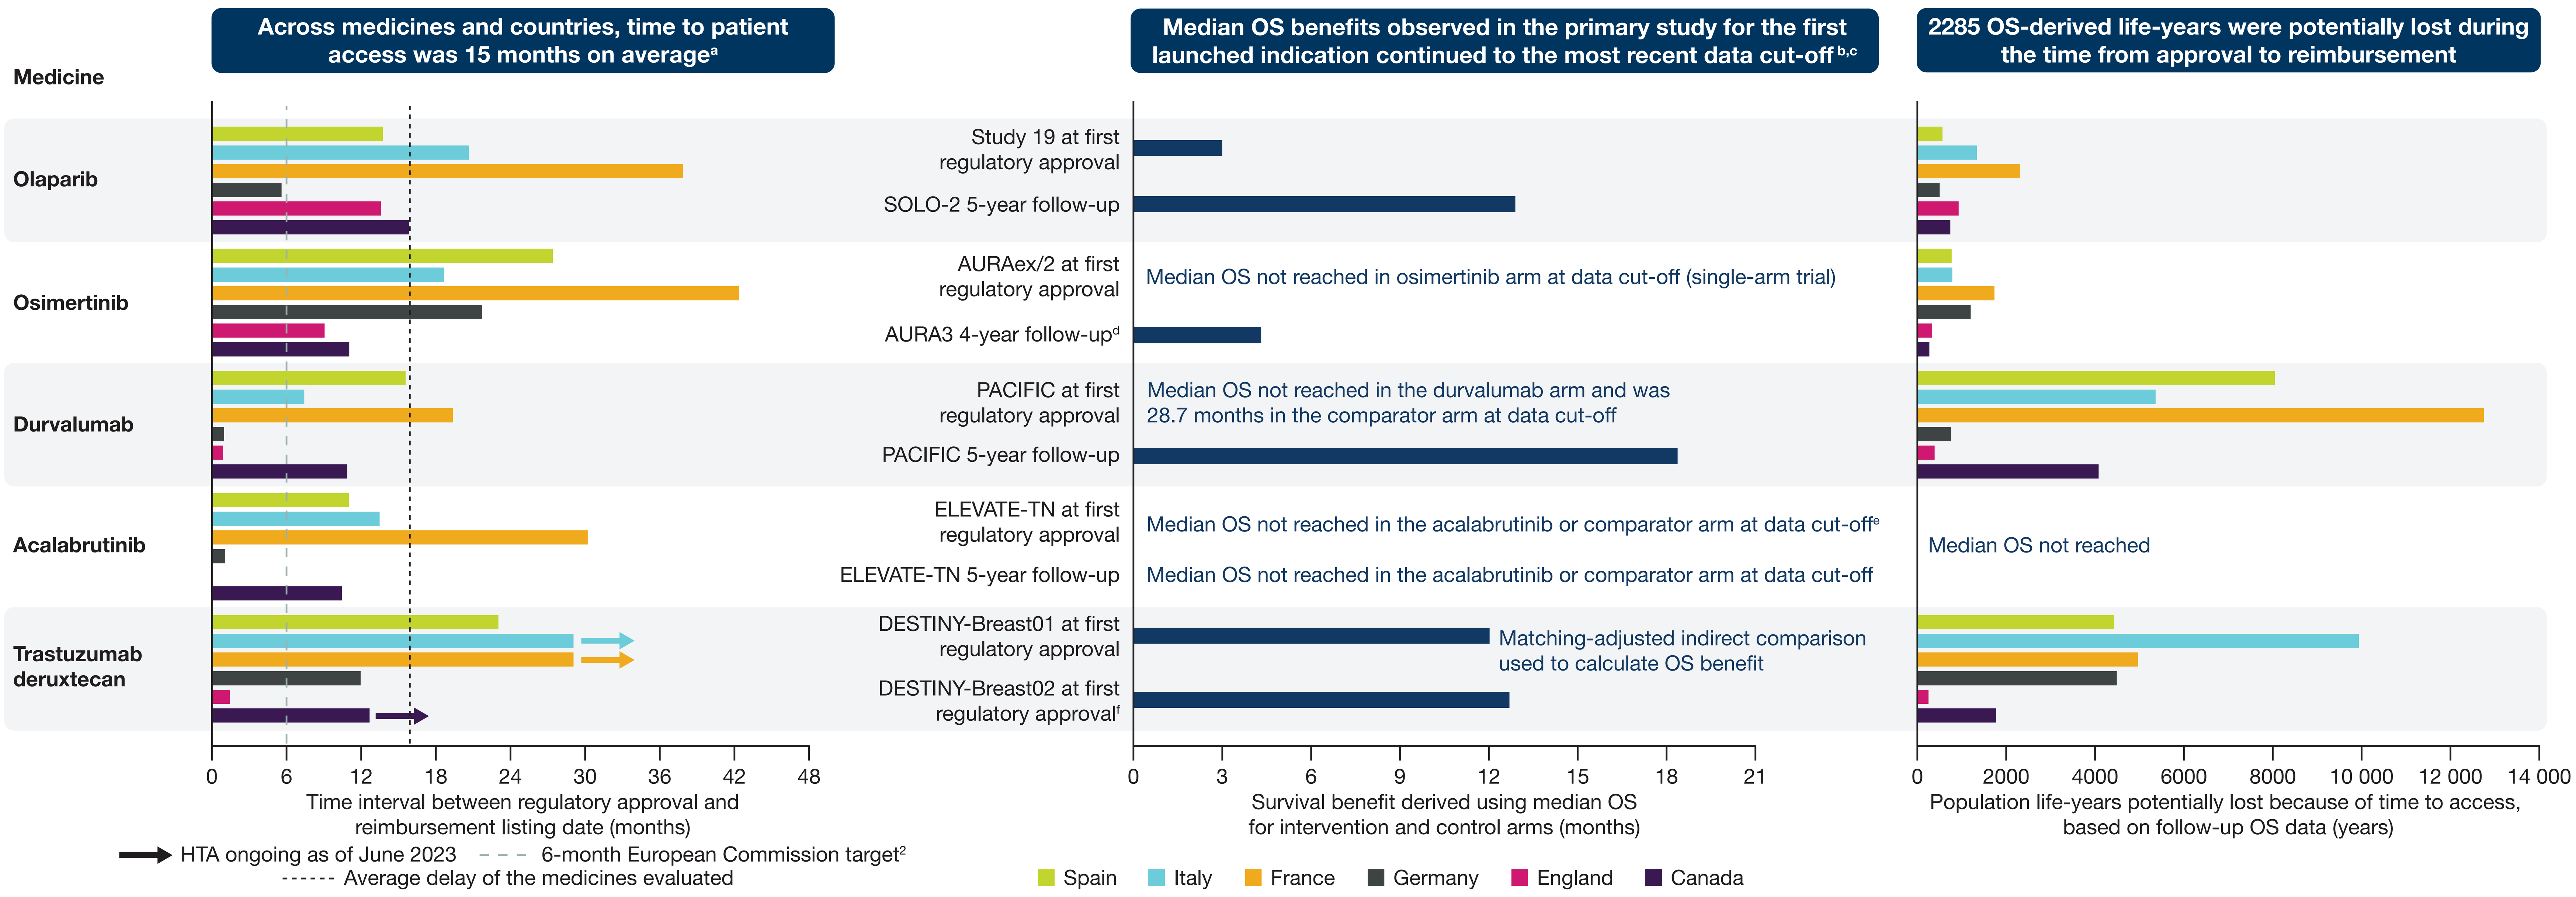
Across medicines and countries, time to patient access was 15 months on averagea
Median OS benefits observed in the primary study for the first launched indication continued to the most recent data cut-off b,c
2285 OS-derived life-years were potentially lost during the time from approval to reimbursement
Medicine
Olaparib
Osimertinib
Durvalumab
Acalabrutinib
Trastuzumab deruxtecan
Study 19 at first regulatory approval
SOLO-2 5-year follow-up
AURAex/2 at first regulatory approval
AURA3 4-year follow-upd
PACIFIC at first regulatory approval
PACIFIC 5-year follow-up
ELEVATE-TN at first regulatory approval
ELEVATE-TN 5-year follow-up
DESTINY-Breast01 at first regulatory approval
DESTINY-Breast02 at first regulatory approvalf
Median OS not reached in osimertinib arm at data cut-off (single-arm trial)
Median OS not reached in the durvalumab arm and was 28.7 months in the comparator arm at data cut-off
Median OS not reached in the acalabrutinib or comparator arm at data cut-offe
Median OS not reached in the acalabrutinib or comparator arm at data cut-off
Matching-adjusted indirect comparison used to calculate OS benefit
Median OS not reached
0 6 12 18 24 30 36 42 48
Time interval between regulatory approval and reimbursement listing date (months)
0 3 6 9 12 15 18 21
Survival benefit derived using median OS for intervention and control arms (months)
0 2000 4000 6000 8000 10 000 12 000 14 000
Population life-years potentially lost because of time to access, based on follow-up OS data (years)
HTA ongoing as of June 2023
6-month European Commission target2
Average delay of the medicines evaluated
Spain
Italy
France
Germany
England
Canada

[a]Early access schemes in France and England were not included in the analysis because patients' access to medicines via early access schemes was restricted based on specific criteria and data on how many patients were able to access the medicines were not available. [b]The studies are in different indications and different populations and outcomes should not be compared. [c]OS was not a primary endpoint for Study 19, AURAex/2, ELEVATE-TN or DESTINY-Breast01. [d]OS benefit has not been adjusted for crossover. [e]24-month OS was 95% with acalabrutinib plus obinutuzumab, 95% with acalabrutinib monotherapy and 92% with obinutuzumab plus chlorambucil. [f]Long-term follow-up ongoing.
HTA, health technology assessment; OS, overall survival.

Acknowledgements

This study was funded by AstraZeneca, UK. The authors thank Helen Fishpool of PharmaGenesis Oxford Central, Oxford, UK, for medical writing support, which has been funded by AstraZeneca, UK, in accordance with Good Publication Practice (GPP 2022) guidelines (www.ismpp.org/gpp-2022).

Disclosures

BC, DP, AH and JR are employed by, and own shares in, AstraZeneca, whose medicines are discussed in this research. NO has no disclosures.

References

1. European Federation of Pharmaceutical Industries and Associations. 2020. Available from: https://www.efpia.eu/media/554527/root-causes-unvailability-delay-cra-final-300620.pdf (Accessed 13 September 2023).
2. European Parliament and Council of the European Union. 1998. Available from: http://data.europa.eu/eli/dir/1998/34/oj/eng (Accessed 11 October 2023).