ISPOR Europe, 12-15 November 2023, Copenhagen, Denmark SA83

{maicplus}: an R package to support analysis and reporting of matching adjusted indirect treatment comparisons (MAIC) for HTA dossiers

X. Gregory Chen[1], Michael Seo[2], Miranta Antoniou[2] , Rossella Belleli[2] , Chrysostomos Kalyvas[1] , Isaac Gravestock[2]

[1] MSD Switzerland, Zurich, Switzerland, [2] F. Hoffman-La Roche, Basel, Switzerland

Introduction

Sponsors are required to submit evidence of relative effectiveness of their treatment comparing to relevant comparators that may not be included in their clinical trial, for health technology assessment (HTA) in different countries. Matching-adjusted indirect treatment comparison (MAIC) [1,2] is a prevalent and well-accepted method [3] to derive population-adjusted treatment effect when there is no connected network and/or the distribution of effect modifiers differs between two trials, one of which has Individual patient data (IPD) and the other has only aggregate data (AgD).

However, there is lack of an open-source R package following good software engineering practices for conducting and reporting MAIC analyses. Currently in CRAN, {maic} performs only the matching step with limited features in preparing data and reporting. {maicChecks} [4] performs some useful checks on numerical feasibility of the matching step and provides alternative weighting schemes.

The implementation of MAIC needs to account for anchored or unanchored cases, different clinical endpoint types (time-to-event, binary and continuous), and two common approaches to address estimation uncertainty (sandwich estimator and bootstrapping).

Objectives

Introduce an R package {maicplus} developed jointly in a **cross-industry workgroup** (**HTA-R workstream**). Dev team members set up the Github repo and merge the codebase of previous internal tools, {MAIC} from Roche (available in Github) and {maicplus} from MSD, in July 2023 and started the collaboration of creating this open-source R package.

Good Open-Source Software Principles

Open-source does not guarantee quality, it depends on how it is done.

If there is good software engineering practice

- Clean code, good product design, agile development style
- Sound version control and change management
- Informative roxygen header and annotation for co-development
- Relevant unit tests (or UAT if products are apps) with clear documentation, to ensure the correctness of the desired functionality and reproducibility

If there is enough traffic

- Best test of quality by users in real applications
- Build credibility via continuously resolving publicly reported issues in Github

If there is good documentation

- Transparency of method, their implementation
- Stimulate discussion about “cross-industry” solutions on various topics

These outline the key aims and principles of the dev team of {maicplus}.

Typical Workflow with {maicplus} with Example Output

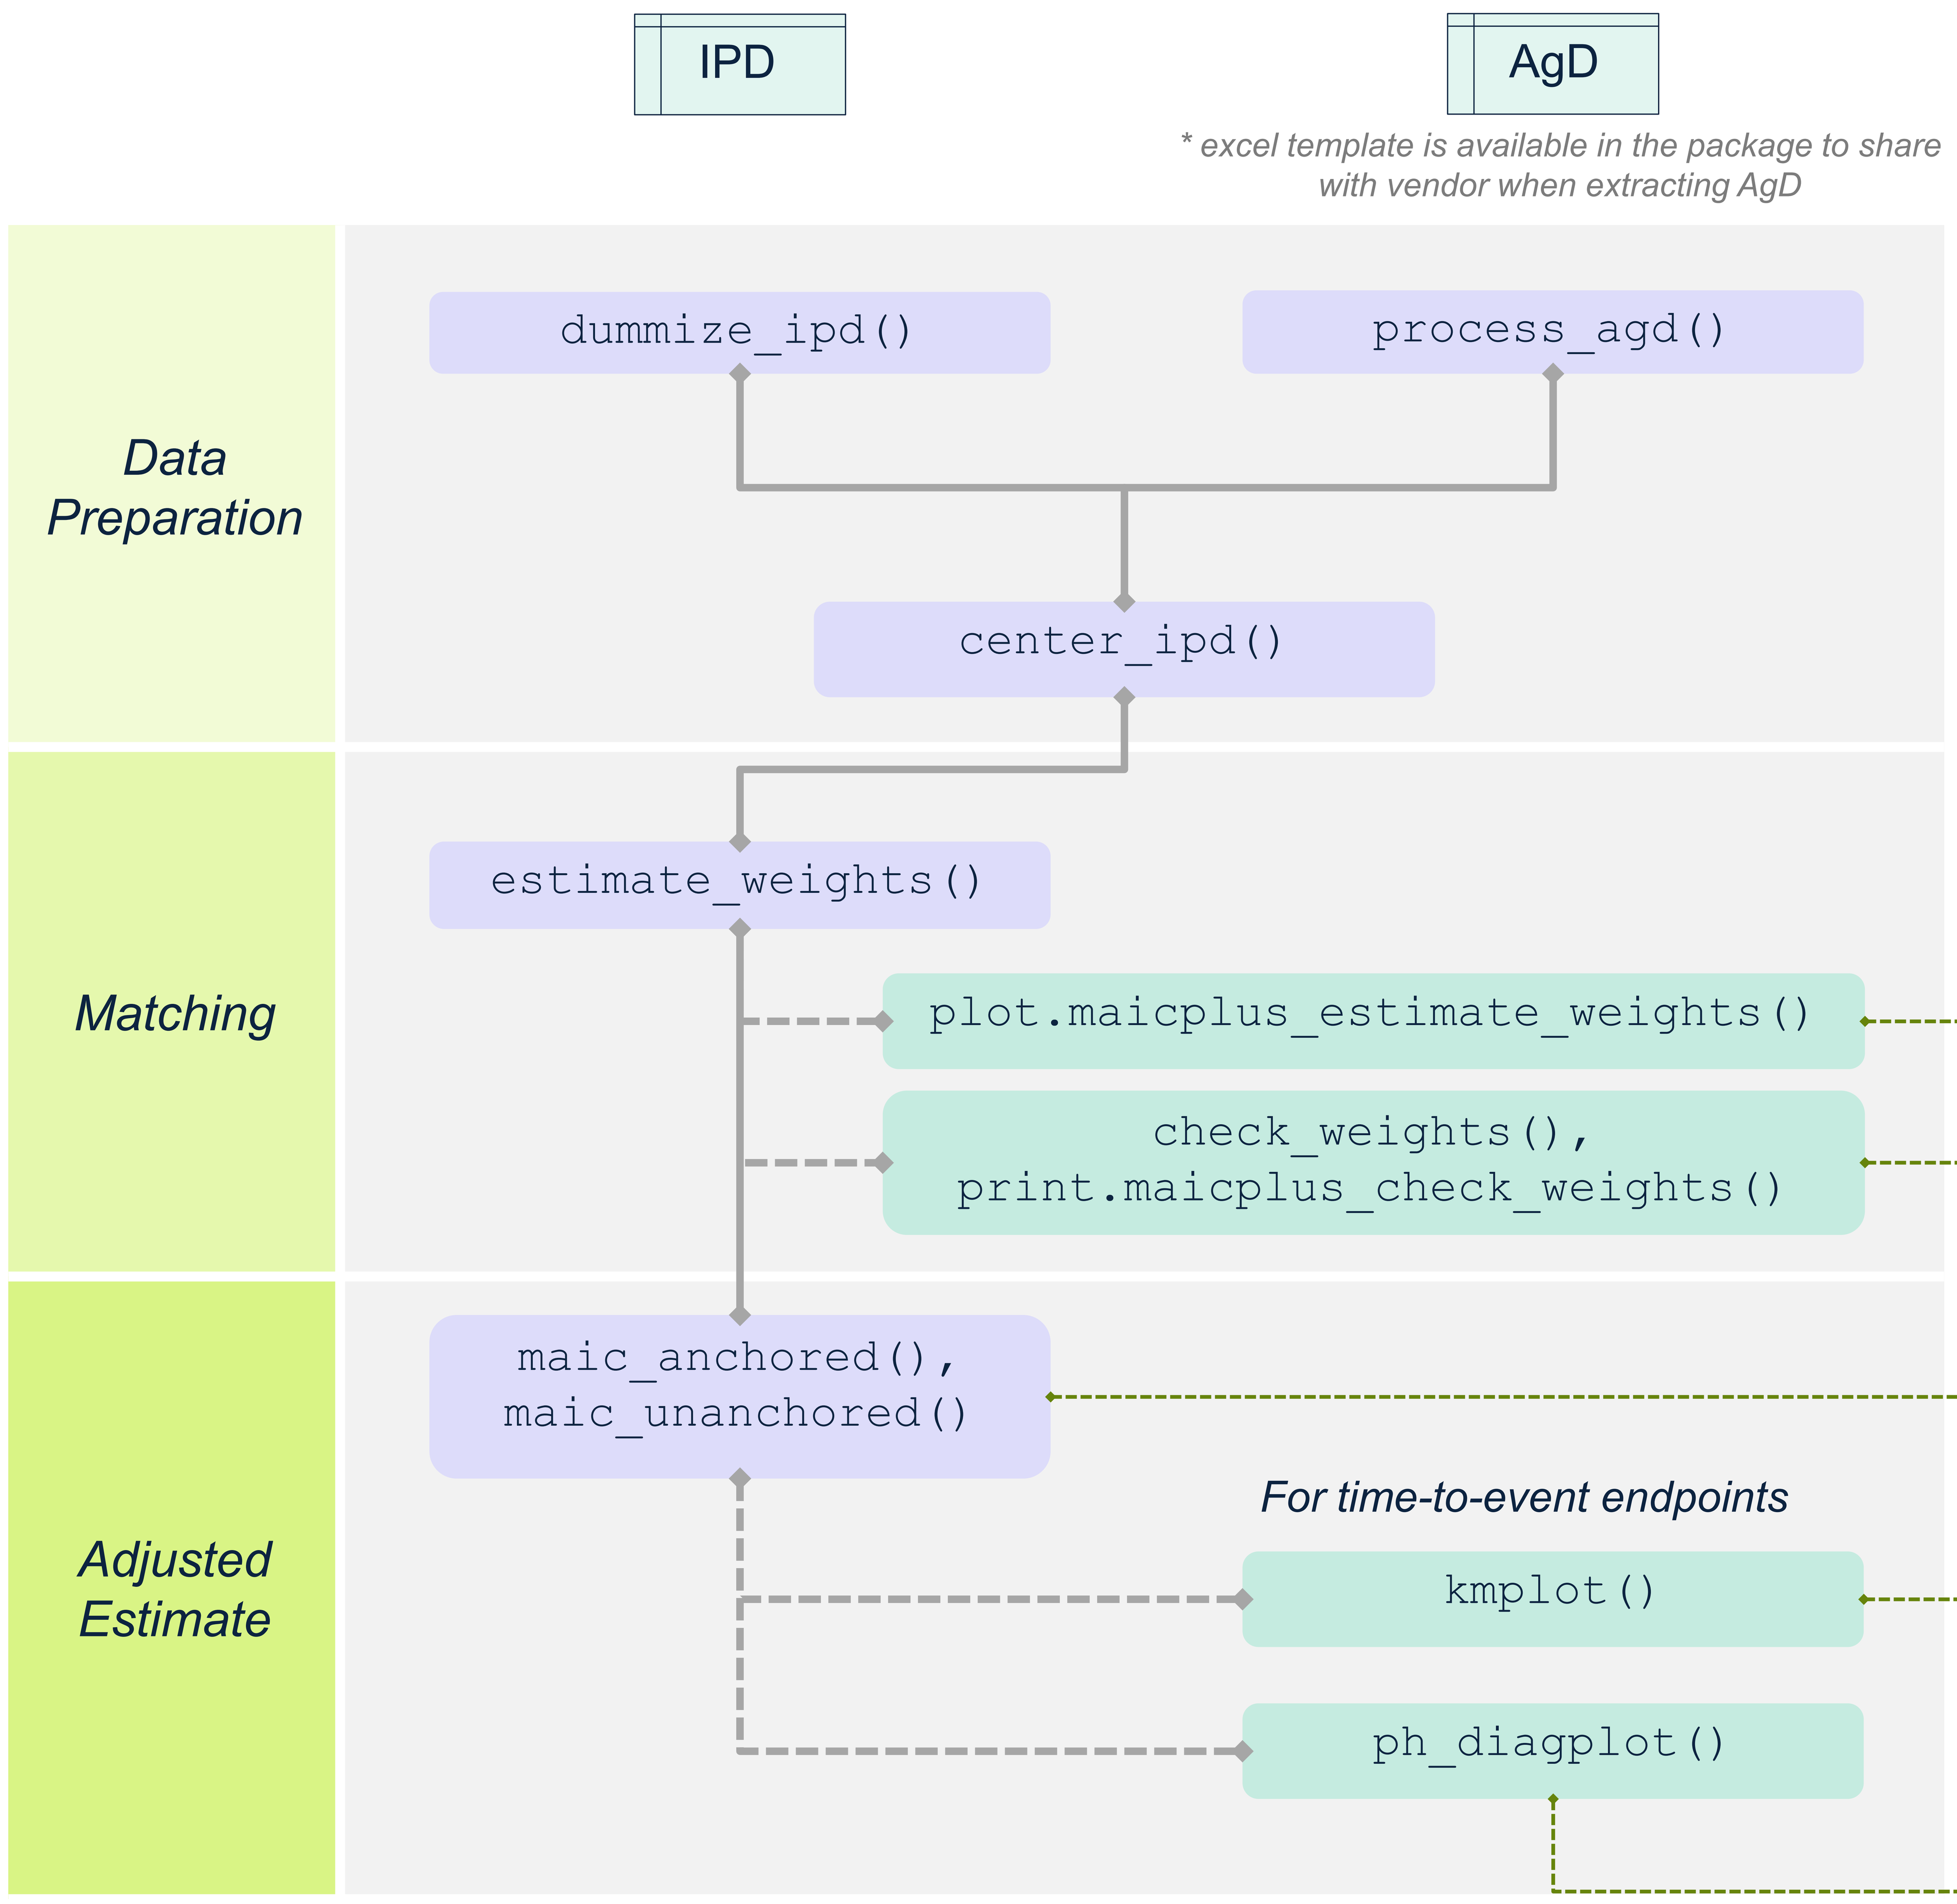
IPD
AgD
* excel template is available in the package to share with vendor when extracting AgD
Data Preparation
dummize_ipd()
process_agd()
center_ipd()
Matching
estimate_weights()
plot.maicplus_estimate_weights()
check_weights(),
print.maicplus_check_weights()
Adjusted Estimate
maic_anchored(),
maic_unanchored()
For time-to-event endpoints
kmplot()
ph_diagplot()

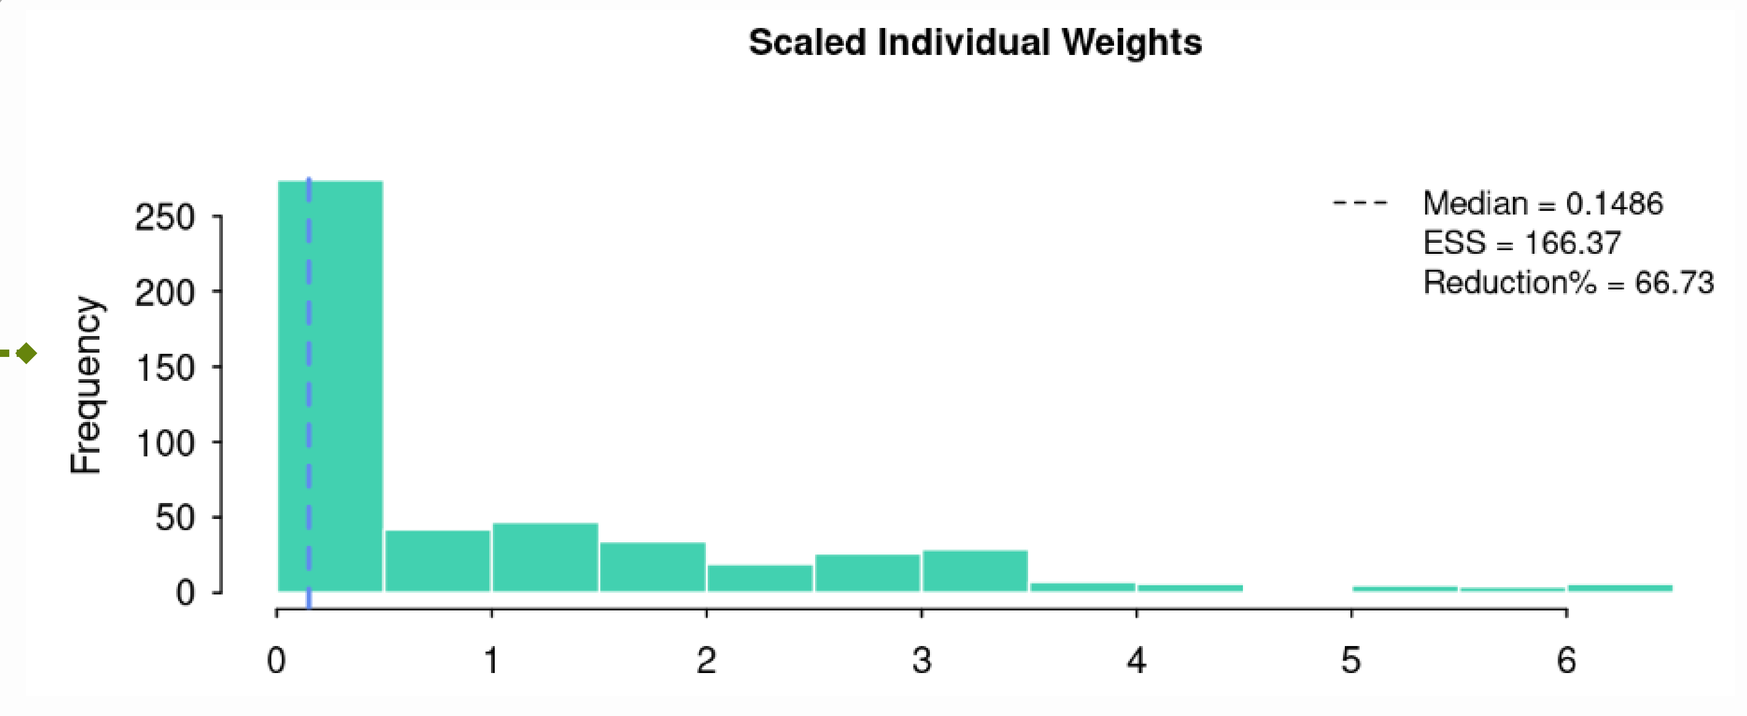
Scaled Individual Weights
Median = 0.1486
ESS = 166.37
Reduction% = 66.73
Frequency

** standard footer will be available for the plot and the table in a v0.1.1*

```
> check_weights(weighted_data, agd)
  covariate match_stat internal_trial internal_trial_after_weighted external_trial sum_centered_IPD_with_weights
1       AGE       Mean         59.850                         51.00          51.00                        0.0001
2       AGE         SD          9.011                          3.25           3.25                        0.0125
3  SEX_MALE       Prop          0.380                          0.49           0.49                        0.0000
4     ECOG0       Prop          0.410                          0.40           0.40                        0.0000
5     SMOKE       Prop          0.320                          0.20           0.20                        0.0000
6 N_PR_THER     Median          3.000                          2.00           2.00                        0.0000
```

```
result$inferential$report_median_surv
treatment                type records    n.max  n.start    events     rmean se(rmean)    median   0.95LCL   0.95UCL
    ARM=C IPD, before matching     500 500.0000 500.0000 498.00000  2.633016 0.1211224  1.809502  1.620663  2.018826
    ARM=A IPD, before matching     500 500.0000 500.0000 190.00000  8.709919 0.3551570  7.587826  6.278856 10.288808
    ARM=C  IPD, after matching     500 173.4208 173.4208 173.36540  2.538260 0.1930851  1.773046  1.449432  2.123657
    ARM=A  IPD, after matching     500 173.4208 173.4208  55.54181 10.575576 0.5732742 12.166750 10.244562        NA
    ARM=C        AgD, external     500 500.0000 500.0000 500.00000  2.511139 0.1041033  1.824917  1.645884  1.982455
    ARM=B        AgD, external     300 300.0000 300.0000 178.00000  4.303664 0.3367349  2.746203  2.261184  3.320944
```

```
result$inferential$report_overall
                   Matching treatment     N  n.events(%) median[95% CI]      HR[95% CI] p-Value
          IPD/Overall Survival    ARM=A   500    190(38.0)  7.6[6.3;10.3] 0.23[0.20;0.28]  <0.001
          IPD/Overall Survival    ARM=C   500    498(99.6)   1.8[1.6; 2.0]
 weighted IPD/Overall Survival    ARM=A 173.4   55.5( 32.0) 12.2[10.2; NA] 0.15[0.10;0.22]  <0.001
 weighted IPD/Overall Survival    ARM=C 173.4 173.4(100.0)   1.8[ 1.4;2.1]
          Agd/Overall Survival    ARM=B   300    178( 59.3)   2.7[2.3;3.3] 0.59[0.49;0.70]  <0.001
          Agd/Overall Survival    ARM=C   500    500(100.0)    1.8[1.6;2.0]
                ** adj.A vs B         -     -            -               - 0.26[0.17;0.39]  <0.001
```

**showing anchored case as an example*

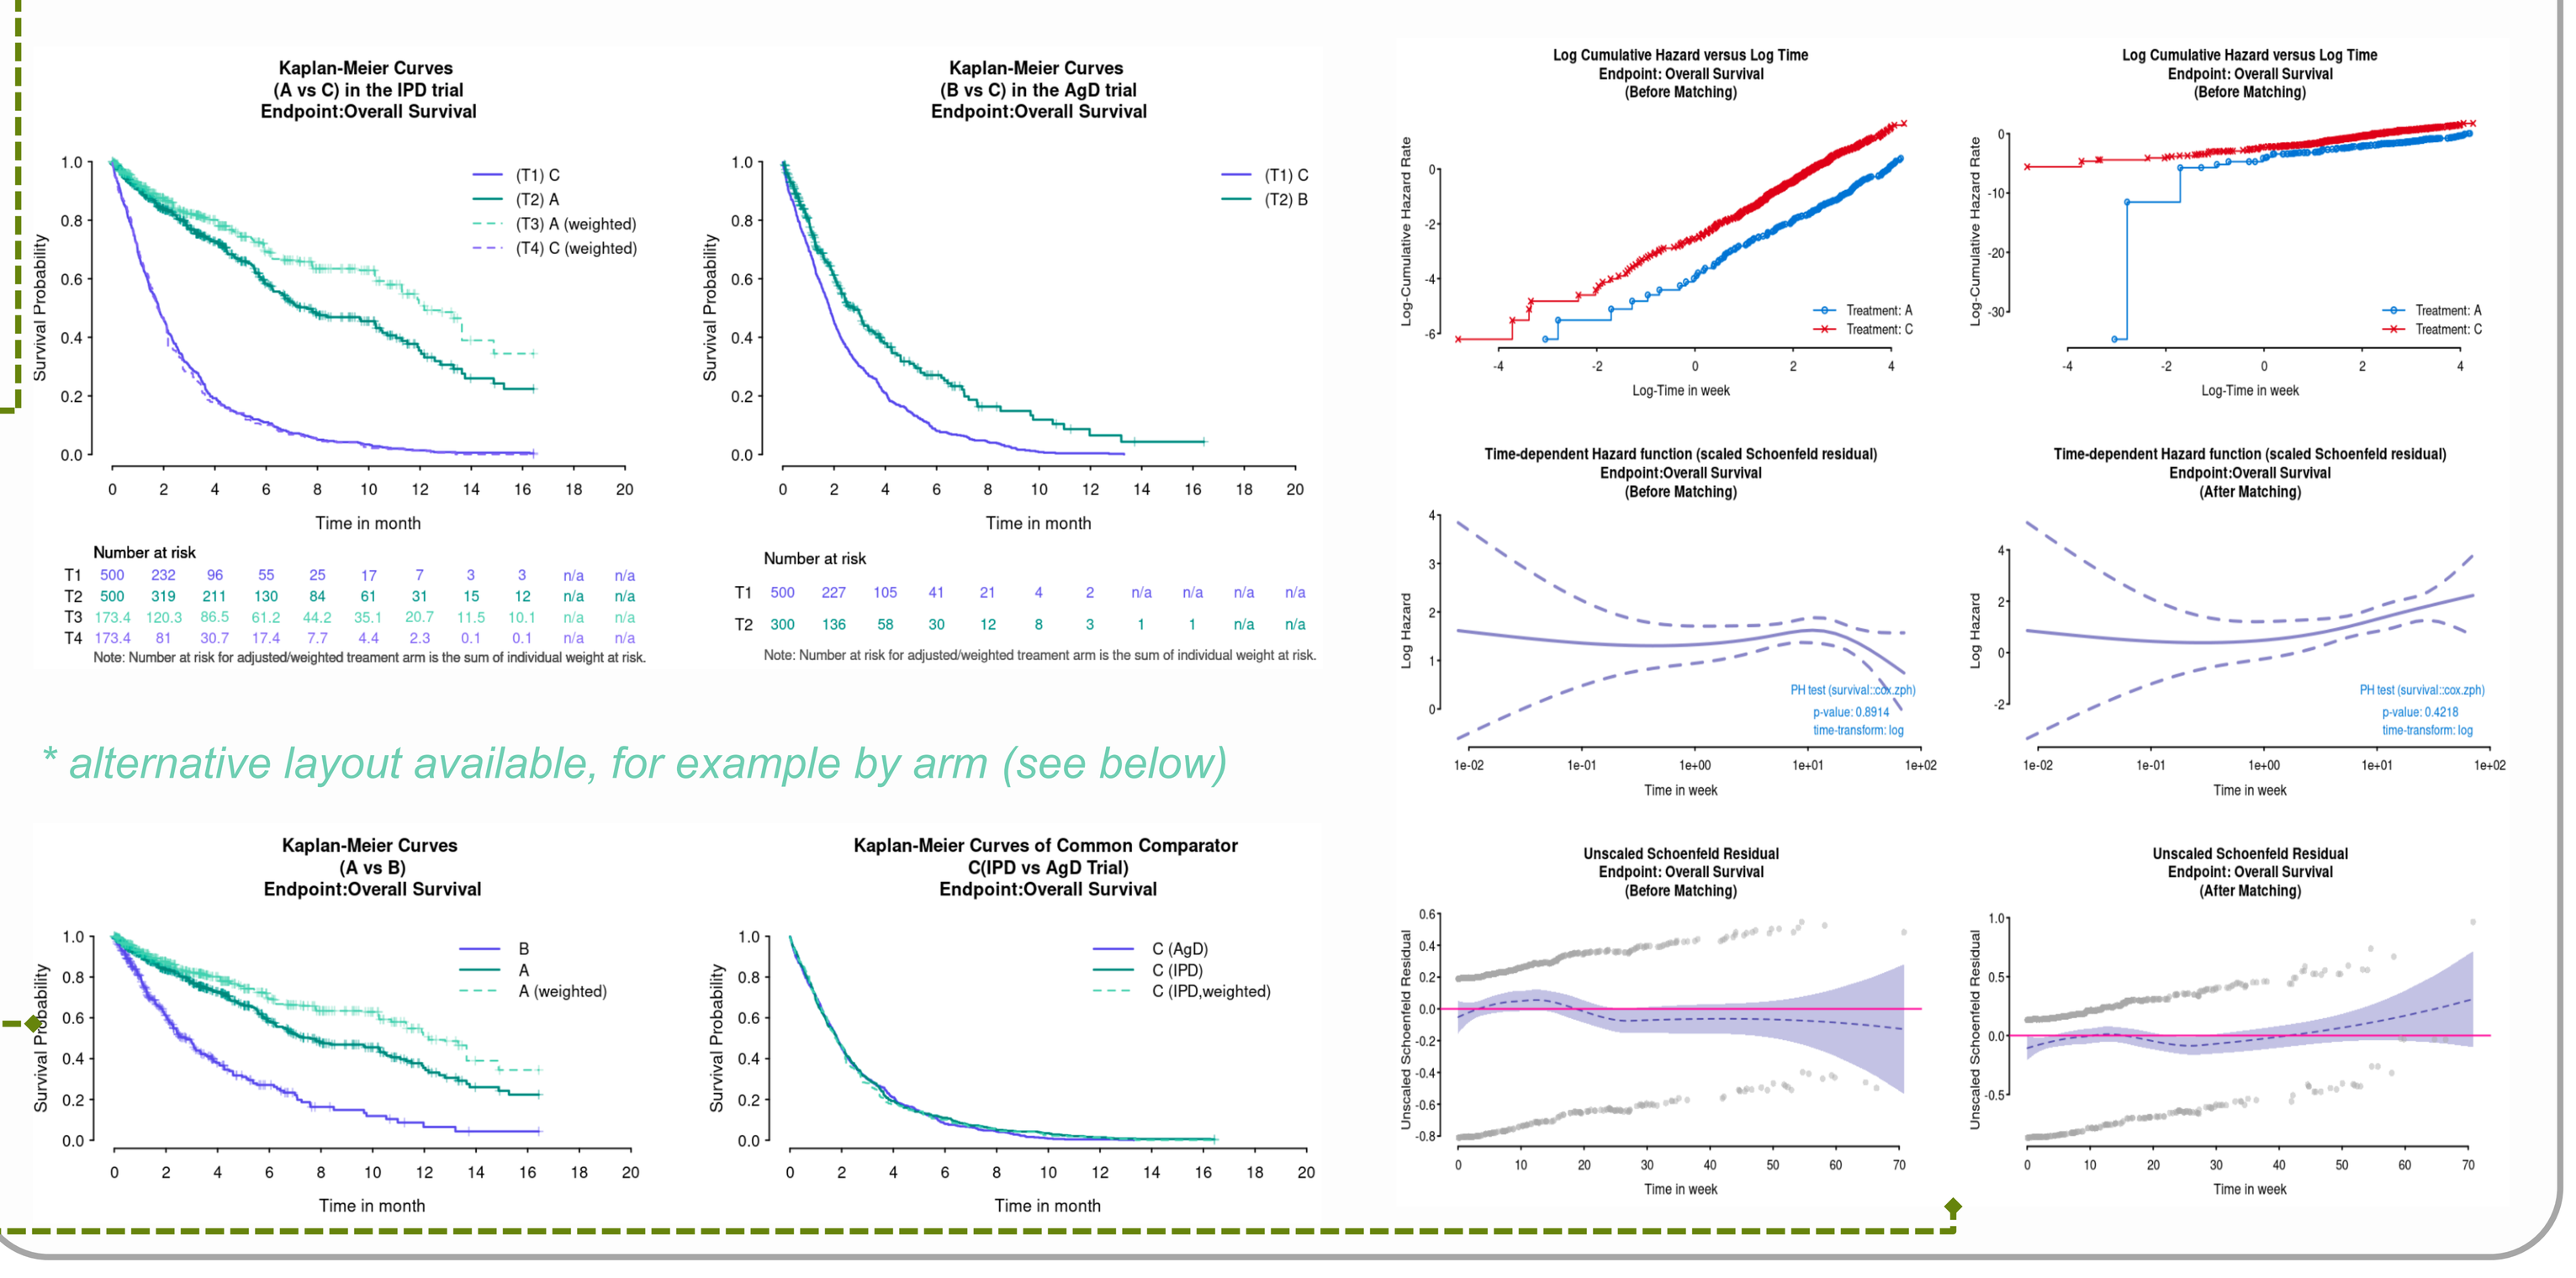
Kaplan-Meier Curves (A vs C) in the IPD trial Endpoint:Overall Survival
Kaplan-Meier Curves (B vs C) in the AgD trial Endpoint:Overall Survival
Log Cumulative Hazard versus Log Time Endpoint: Overall Survival (Before Matching)
Time-dependent Hazard function (scaled Schoenfeld residual) Endpoint:Overall Survival (Before Matching)
Time-dependent Hazard function (scaled Schoenfeld residual) Endpoint:Overall Survival (After Matching)
* alternative layout available, for example by arm (see below)
Kaplan-Meier Curves (A vs B) Endpoint:Overall Survival
Kaplan-Meier Curves of Common Comparator C(IPD vs AgD Trial) Endpoint:Overall Survival
Unscaled Schoenfeld Residual Endpoint: Overall Survival (Before Matching)
Unscaled Schoenfeld Residual Endpoint: Overall Survival (After Matching)
Time in month
Survival Probability
Time in week

HTA-R Workstream and Workplan of {maicplus}

openstatsware (open-source statistical software) is the Software Engineering (SWE) Working Group in the American Statistical Association (ASA) Biopharmaceutical section (BIOP). Its **HTA-R workstream** aims to develop and maintain a collection of open-source R tools of high quality in the right formats (packages, apps, implementation-focus report/manual) that are useful to both sponsors and payers, to support crucial analytic topics in HTA dossier submission across various countries, particularly the topics with unmet needs in R implementation and/or related to EUnetHTA.

https://rconsortium.github.io/asa-biop-swe-wg/

This workstream is in close collaboration with HTA ESIG in PSI/EFPSI who can help to generate pipeline ideas and ensure relevance of developed tools. See https://www.psiweb.org/sigs-special-interest-groups/hta

Workplan for {maicplus}

- Submit v0.1.0 to CRAN before end of 2023. The package will be available in CRAN in Q1 2024.
- Continue to develop designed/requested functionalities for version 0.1.1 (see issues of our github repo, with label “next version”). Please register your suggestions for the tool there.

Conclusion

It will benefit our HTA community (sponsors and payers) in the long run to have more open-source R tools that are developed and maintained following good software engineering practices. In addition to leveraging each other’s expertise and resource to create a standard tool for a common analysis for HTA submissions, open source collaboration brings in strong transparency of implementation details and real-world testing for high quality. {maicplus} was created as a result of such effort.

Please join us in **HTA-R workstream.** (homepage: *github.com/hta-pharma*)

Reference:

1.Signorovitch, J.E., Wu, E.Q., Yu, A.P., Gerrits, C.M., Kantor, E., Bao, Y., Gupta, S.R. and Mulani, P.M., 2010. Comparative effectiveness without head-to-head trials: a method for matching-adjusted indirect comparisons applied to psoriasis treatment with adalimumab or etanercept. Pharmacoeconomics, 28, pp.935-945.

2.Signorovitch, J.E., Sikirica, V., Erder, M.H., Xie, J., Lu, M., Hodgkins, P.S., Betts, K.A. and Wu, E.Q., 2012. Matching-adjusted indirect comparisons: a new tool for timely comparative effectiveness research. Value in Health, 15(6), pp.940-947.

3.Phillippo, D., Ades, T., Dias, S., Palmer, S., Abrams, K.R. and Welton, N., 2016. NICE DSU technical support document 18: methods for population-adjusted indirect comparisons in submissions to NICE.

4.Glimm, E. and Yau, L., 2022. Geometric approaches to assessing the numerical feasibility for conducting matching-adjusted indirect comparisons. Pharmaceutical Statistics, 21(5), pp.974-987.

ISPOR Europe, 12-15 November 2023, Copenhagen, Denmark