EE157

The Societal Impact of Inclisiran

Authors: Dennis A. Ostwald, Prof. Dr.* , Malina Müller, Dr.* , Maike Schmitt, Dipl. Kffr.* , Platon Peristeris, PhD* , Adnan Atitallah, Msc* , Lena Steinbeck, Ma* , Jonas Müller** , Katharina Büsch**

*. WifOR Institute, Darmstadt, Germany **. Novartis Pharma AG, Basel, Switzerland

WifOR INSTITUTE

Motivation / Context

Around the globe public budgets on health are overstretched. As governments are under pressure, investment in medical innovations is merely regarded as a cost factor. The traditional healthcare perspective neglects that increasing country health expenditures are necessary to achieve sustainability and development goals.[1,2] Strategic investments in health initiate better population health which leads to economic growth and promotes wealth to society. With the framework of the Societal Impact, we offer a transformative perspective on understanding how health investments affect the economy, society, and quality of every individuals' live.

Health investments have a highly positive return on investment and create a positive feedback loop

Sustainable Development Goal 3 (SDG)[2]

3 GOOD HEALTH AND WELL-BEING

Ensure healthy lives and promote well-being for all at all ages

Sustainable Development Goal 9 (SDG)[2]

9 INDUSTRY, INNOVATION AND INFRASTRUCTURE

Build resilient infrastruture, promote sustainable industrialization and foster innovation

Sustainable Development Goal 8 (SDG)[2]

8 DECENT WORK AND ECONOMIC GROWTH

Promote sustained, inclusive and sustainable economic growth, full and productive employment and decent work for all

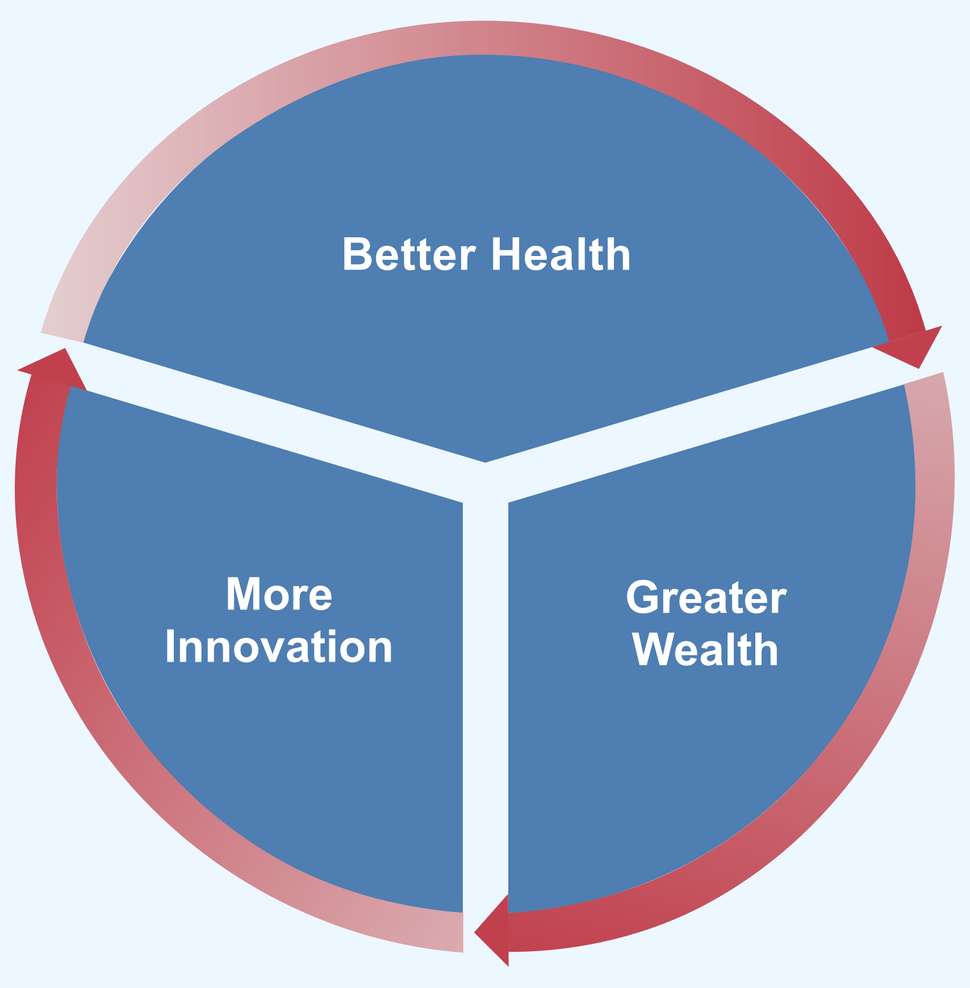
Better Health
More Innovation
Greater Wealth

Cardiovascular disease (CVD) is the leading cause of death worldwide.[3]

CVD-related yearly global costs - including direct healthcare costs as well as productivity losses - are estimated at US$ 1,044 bn until 2030.[4] Atherosclerotic Cardiovascular Disease (ASCVD) is the leading cause of CVD morbidity and mortality.[5] High levels of Low-Density Lipoprotein-Cholesterol (LDL-C), including the cumulative exposure to them, lead to the development of ASCVD. inclisiran is a first in class small interfering RNA targeting hepatic PCSK9 and lowering LDLc as an add-on to statins.

Objective

Evidence of the impact of CVD beyond the burden on healthcare systems is scarce. The aim of this research is to estimate the Societal Impact of introducing inclisiran according to localized population health approaches around the globe over a ten-year horizon. Thereby, we consider the impact on health, productivity (paid and unpaid work activities), and value creation and translate it into monetary terms.

Methodology

Our methodology consists of two major steps. First, we calculate the health footprint. Therefore, a Markov model was developed based on the inclisiran CEM and adapted to each country to simulate the incremental health gains due to introducing add-on inclisiran treatment.[7] In general, the considered patient population covers adults with pre-existing ASCVD aged 50 years and older. Assumptions regarding specific age-, gender-, and diabetes distribution at baseline as well as uptake values were adapted for each use case. In the second step we evaluate the Societal Impact of the treatment. Therefore, estimated health gains are translated into potential gains in productive time in both paid and unpaid work activities, based on country-specific productivity parameters and socioeconomic data from official statistics. A detailed depiction of the methodology for the Societal Impact evaluation can be found in Ostwald et al. 2023.[8]

	Direct Paid Work Productivity	Indirect / Induced (value chain) Paid Work Productivity	Unpaid Work Productivity
Input parameters	Absenteeism: Work-time loss due to hospitalization + sick leave in first year after event.[9] Presenteeism: Work-time loss while being present at work, due to restricted functioning caused by uncontrolled disease or health risks in the first year after CV event.[10] Return to Work (RTW): The percentage of employees that return to work in the first year after a CV event.[9]	Multipliers: Country-specific spillover factors based on input-output tables to calculate the indirect and induced effects in the overall economy.[11]	Activity restriction: The percentage of restriction in everyday activities in the first year after event, such as cooking, cleaning, childcare, etc.[12]
Outputs: Gain in productive time	• Non-fatal events: Absenteeism Presenteeism Reduced labor force participation (no RTW) • Fatal events: Years of life lost	• Non-fatal events: Absenteeism Presenteeism Reduced labor force participation (no RTW) • Fatal events: Years of life lost	• Unpaid work activities
Monetisation	Using the country-specific economic sector-specific hourly gross value added (GVA) contribution.[13]		

Evaluation around the globe in 17 countries

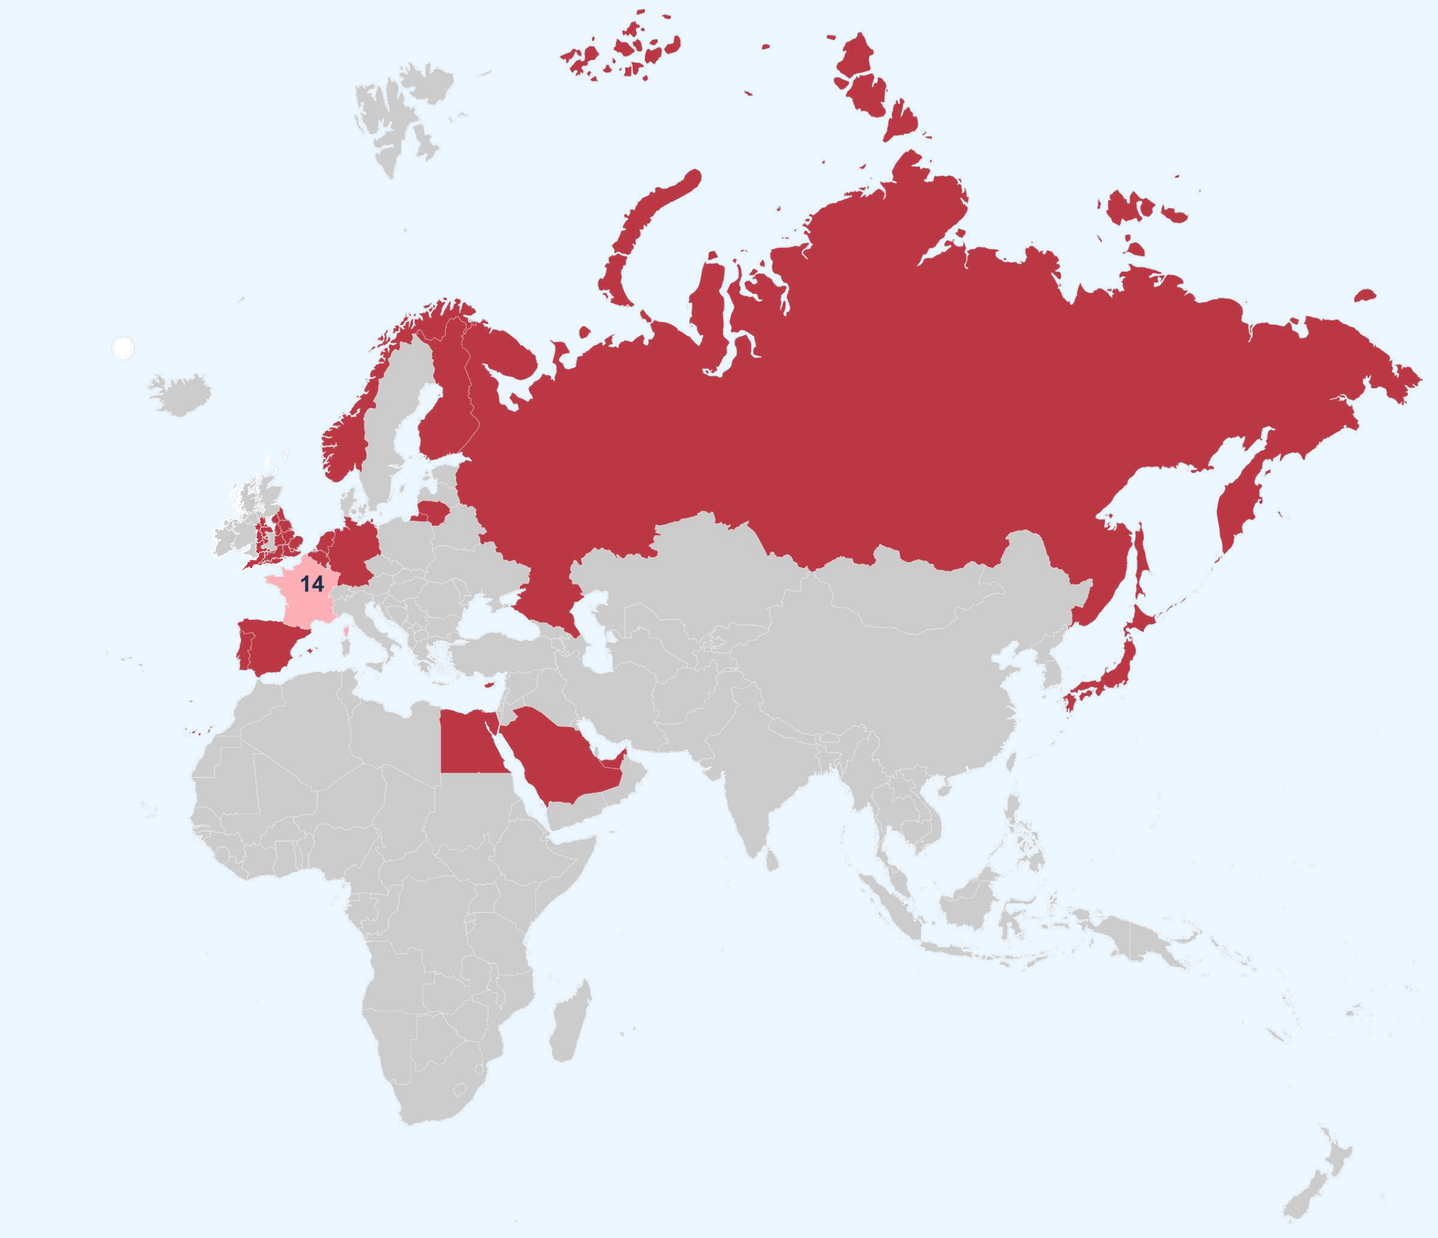
14

Canada

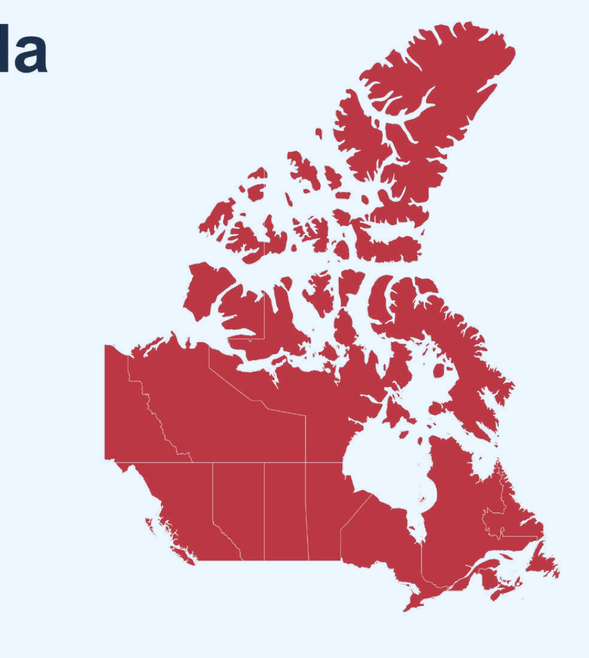

Australia

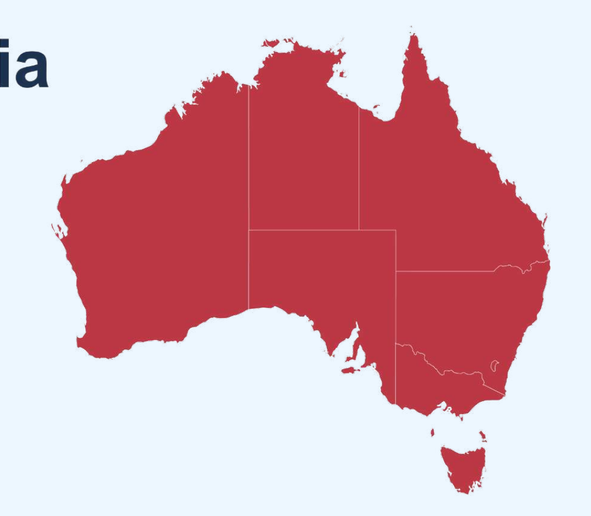

Results

Aggregated results

2,649,086 patients

858,953 CV events avoided

262,493 premature death cases avoided

€45.05b Societal Impact

262,493 avoided premature death cases is equivalent to about 9.8% of all CV death cases in the 17 implemented countries in 2019.[15]

Overview of all undiscounted results of the implemented countries

Country	Avoided events			Societal Impact in 2019 Euro
	CV events without premature death	Premature death cases	CV events including premature death cases	
Australia	97,375	31,062	128,437	6,733,416,081 €
Belgium	6,848	2,079	8,927	460,668,118 €
Canada	44,260	14,463	58,723	2,304,437,162 €
Cyprus	2,372	827	3,199	50,826,249 €
Egypt	31,232	8,605	39,837	112,485,654 €
England	104,675	33,972	138,647	9,655,799,936 €
Finland	7,684	2,751	10,435	348,261,524 €
Germany	44,266	15,845	60,111	1,717,125,415 €
Japan [16]	392,995	120,768	513,763	18,883,566,151 €
KSA	18,531	4,885	23,416	598,200,721 €
Lithuania	3,310	923	4,233	73,848,690 €
Netherlands	12,201	4,267	16,468	627,692,309 €
Norway	4,356	1,122	5,478	487,395,164 €
Portugal	43,989	6,946	50,935	1,654,159,171 €
Russia	15,911	4,359	20,270	229,945,987 €
Spain	23,046	7,332	30,378	946,395,771 €
UAE	5,902	2,287	8,189	167,176,699 €

Country-specific results: Due to heterogenous input data, results differ and are hard to compare.

a) Premature CV death and revascularization have highest value of avoided events as illustrated by the results of five representative countries, including Canada, England, Germany, Japan[16] and Spain.

The total Societal Impact of these five representative countries amounts to €12.43b. This is equivalent to about 5% of their total pharmaceutical expenditures in one year.[17]

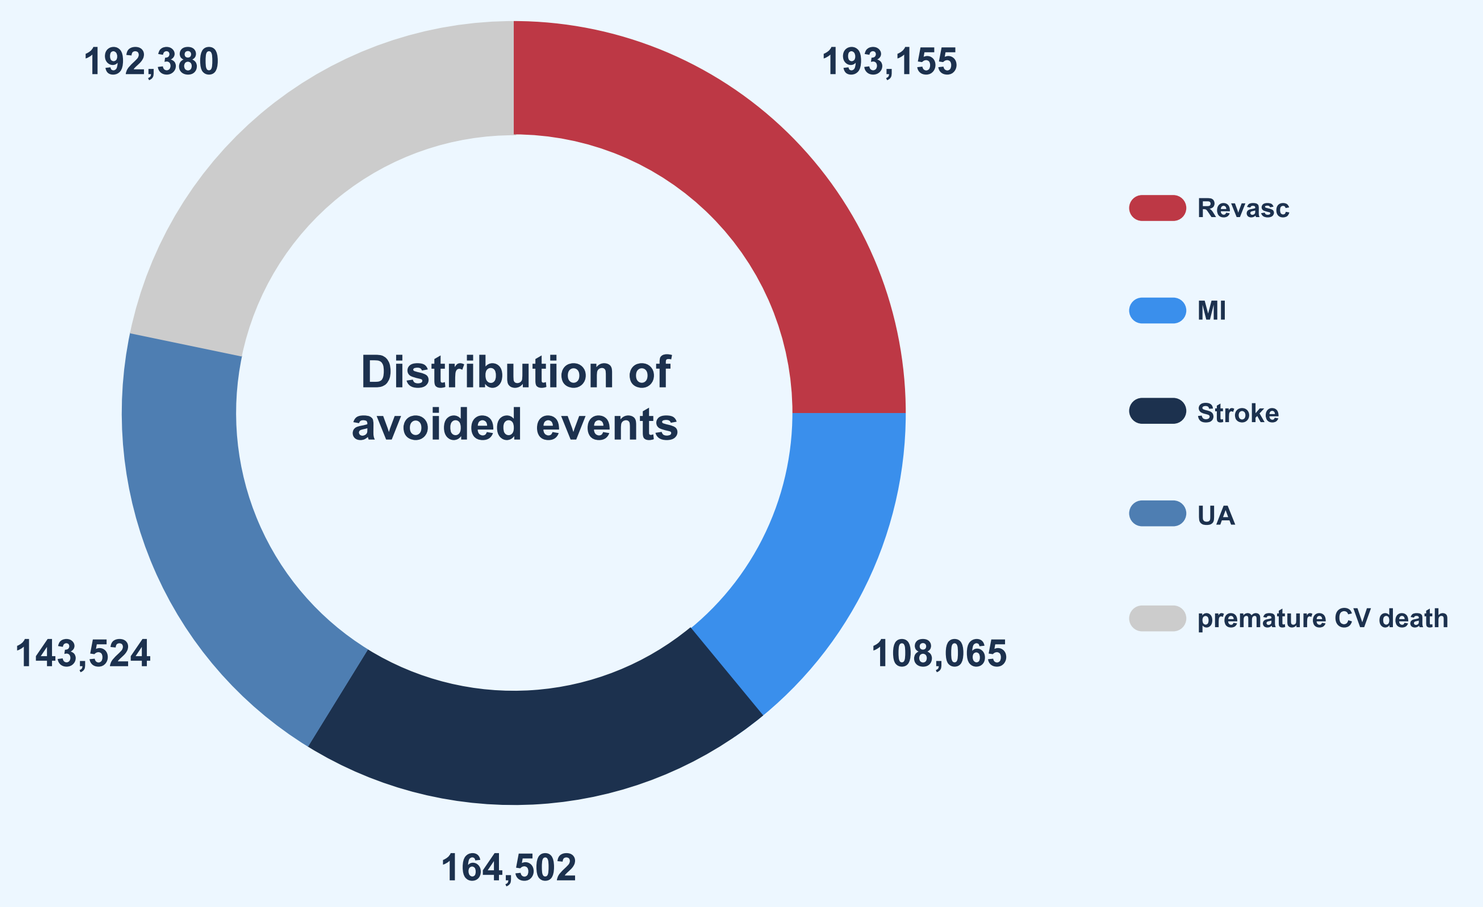
Distribution of avoided events
193,155
108,065
164,502
143,524
192,380
Revasc
MI
Stroke
UA
premature CV death

b) In the majority of countries we observed that the Societal Impact can be achieved in all age groups, as shown in the example of Germany with a relatively old patient population.

Baseline distribution

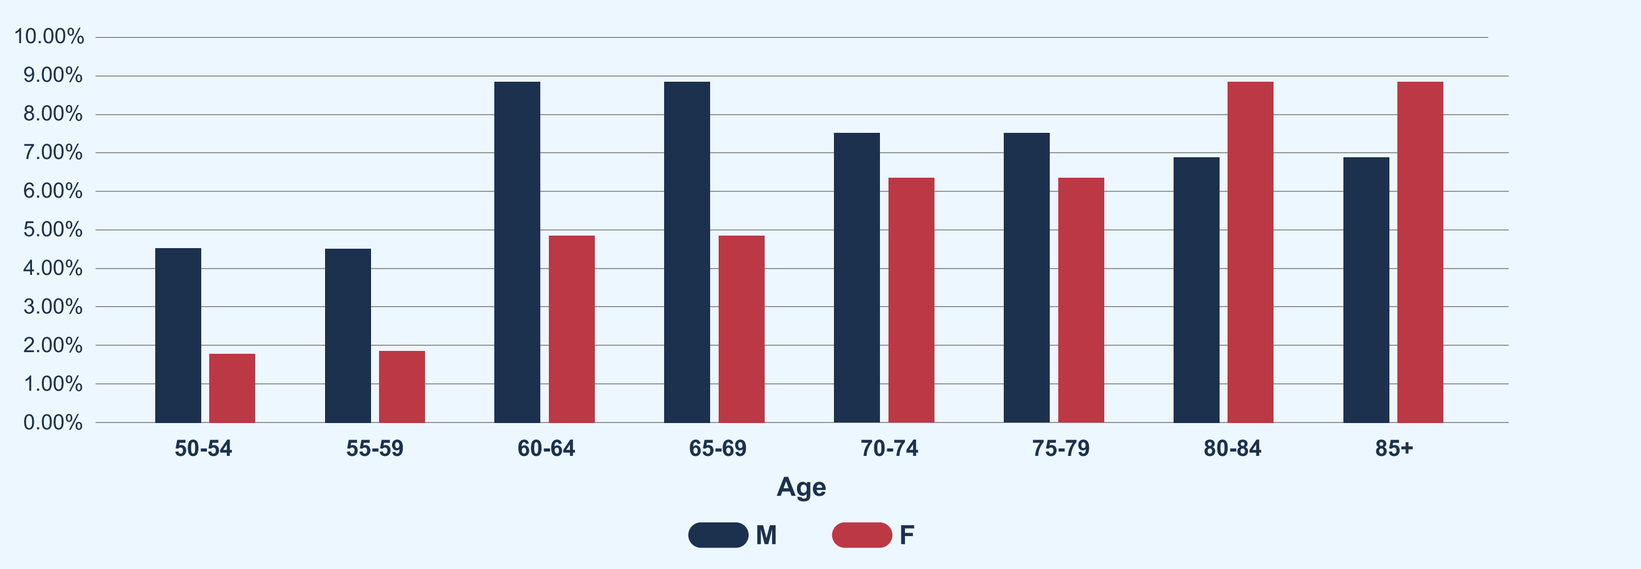
10.00%
0.00%
50-54
55-59
60-64
65-69
70-74
75-79
80-84
85+
Age
M
F

Societal Impact

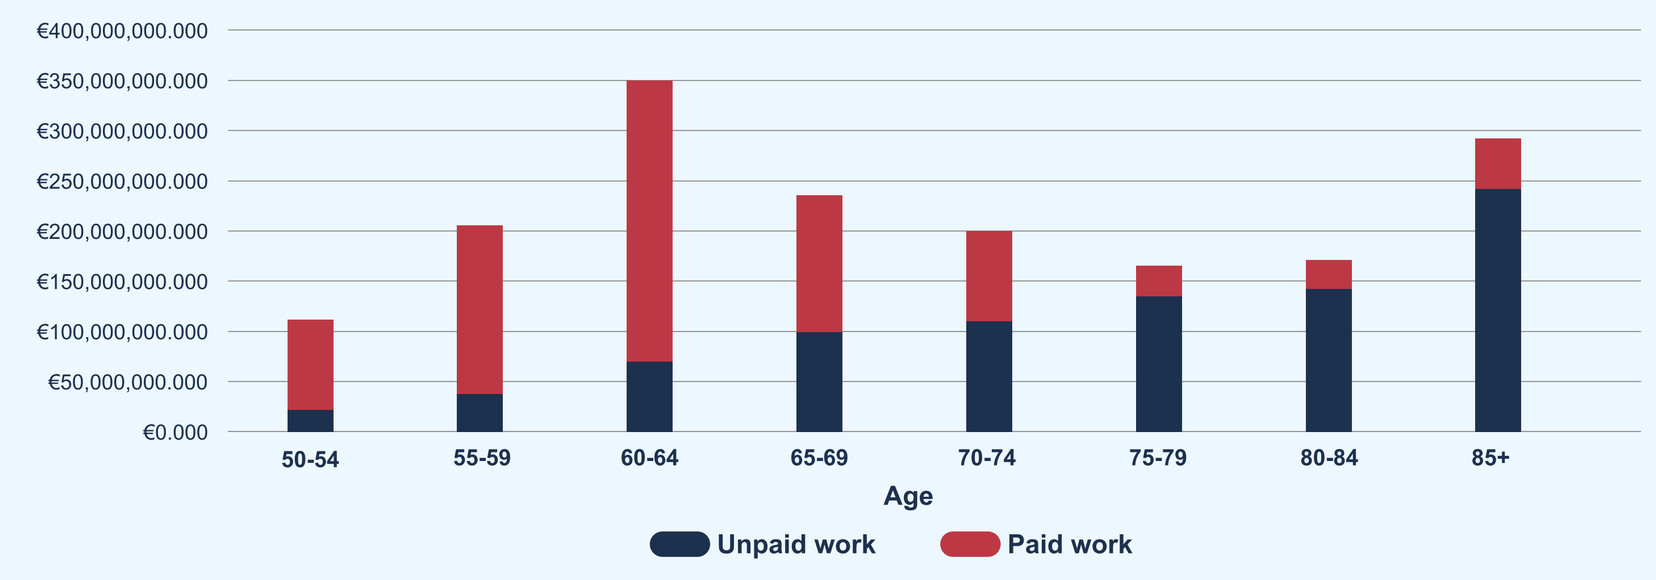
€400,000,000.000
€0.000
50-54
55-59
60-64
65-69
70-74
75-79
80-84
85+
Age
Unpaid work
Paid work

There is positive Return on Investment also for an older population. Even with less paid work productivity, Societal Impact can be achieved due to unpaid work activities.

c) Depending on labour market characteristics and country-specific time use data, potential productivity gains are not homogeneous across countries.

Distribution of potential gain in paid work activities in Japan[16]
Difference in productivity loss (direct)

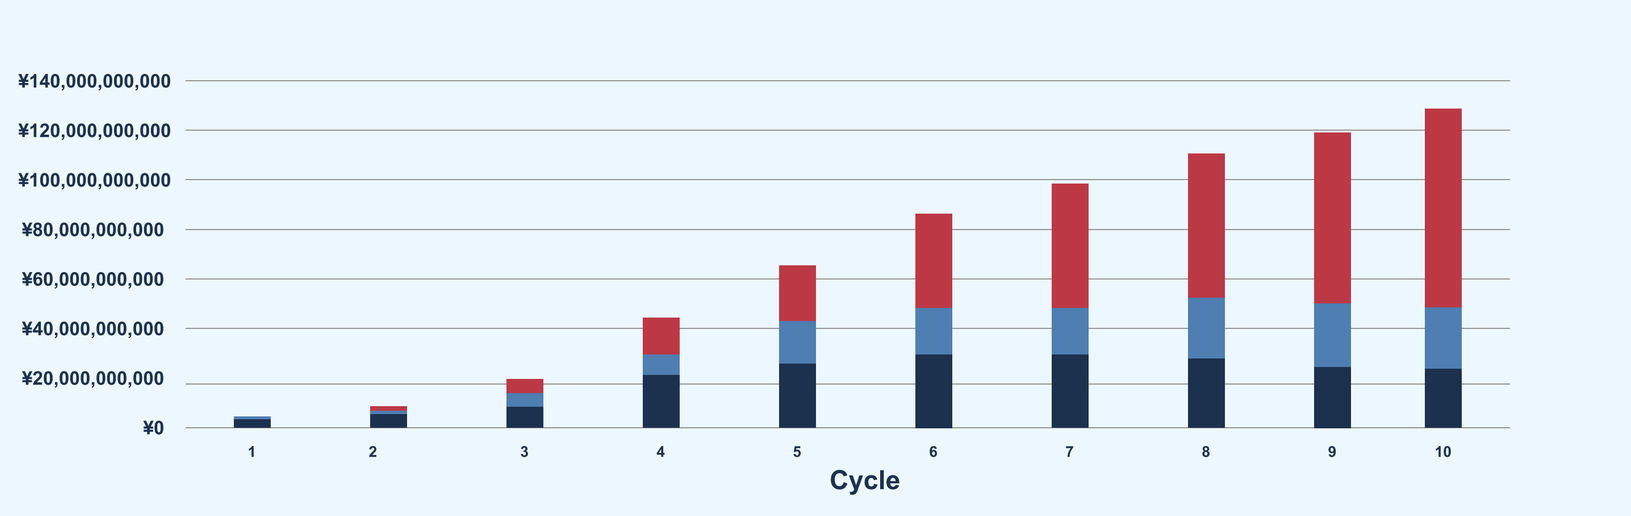
¥140,000,000,000
¥0
Cycle

Distribution of potential gain in paid work activities in Egypt
Difference in productivity loss (direct)

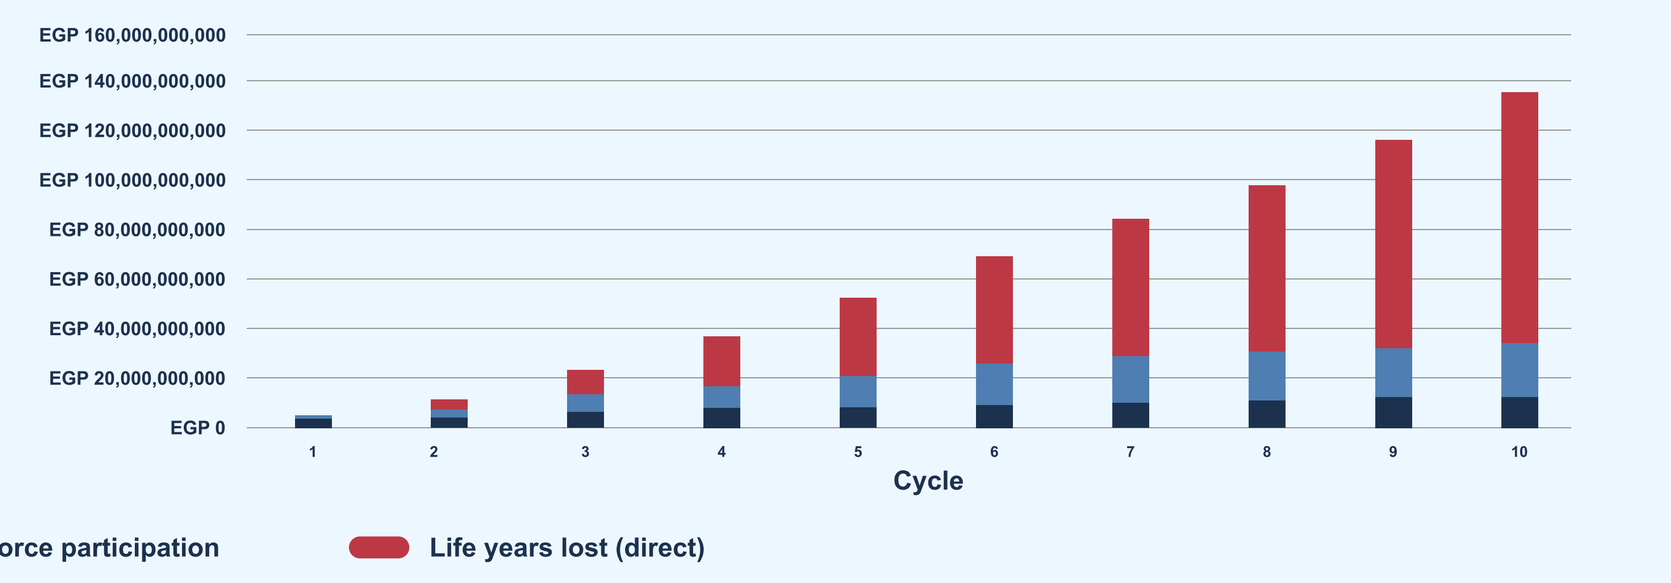
EGP 160,000,000,000
EGP 0
Cycle
Abs. & pres. after CV events (direct)
Reduced labor force participation
Life years lost (direct)

As seen in the examples of Japan[16] and Egypt, avoided productivity losses in paid work can arise of various reasons. In countries with relatively low absenteeism and presenteeism after an event, we observe relatively smaller shares of total avoided productivity losses.

Limitations

As every modelling study, the applied approach has limitations. First, the considered patient population might differ from the general population in terms of employment characteristics and how free time is spent (which affects unpaid work activities). Second, companies could find ways to partially compensate the decrease in productivity given by employees medical conditions. Finally, not all dimensions of value creation could be captured, e.g., the potential variation in caregivers' productivity is not estimated within the present scope of analysis.

Advantages

The Societal Impact approach provides a framework for a holistic assessment of the broader value of inclisiran from a policy perspective. Each of the country evaluations is based on country-specific input data and characteristics. Potential gains in productivity include both paid and unpaid work activities of the considered patient population. The contribution of individuals to the economy are evaluated using GVA instead of wages which allows to relate the results to the national accounting system. Furthermore, it enables to address besides direct effects also spillover effects along the economic value chains and makes results understandable for multilevel stakeholder decision-making.

Conclusion

The results demonstrate the potential health and socioeconomic value of inclisiran over a large-scale intervention across all analyzed countries. More CV events can be avoided in elderly rather than young patients. However, from an economic perspective, it pays off to intervene early, as the <65 population shows significantly higher productivity gains due to being in employment. This highlights the importance of preventing and treating CV disease, while recognizing health as an asset and a source of social and economic stability that supports policy decision-makers to invest in health.

1. Bundesministerium für wirtschaftliche Zusammenarbeit und Entwicklung (BMZ). 2023. SDG 3: Gesundheit und Wohlergehen. https://www.bmz.de/de/agenda-2030/sdg-3.
2. United Nations (2023). Sustainable Development Goals. 17 Goals to Transform Our World. Available at: https://www.un.org/sustainabledevelopment/.
3. WHO factsheet available at: https://www.who.int/news-room/fact-sheets/detail/cardiovascular-diseases-(cvds).
4. Bloom, D. E., Cafiero, E. T., Jané-Llopis, E., Abrahams-Gessel, S., Bloom, L. R., Fathima, S., Feigl, A. B., Gaziano, T., Mowafi, M., Pandya, A., Prettner, K., Rosenberg, L., Seligman, B., Stein, A. Z., and Weinstein, C. (2011); The Global Economic Burden of Noncommunicable Diseases. Geneva: World Economic Forum.World Heart Federation available at: http://www.championadvocates.org/en/champion-advocates-programme/the-costs-of-cvd.
5. Barquera, S., Pedroza-Tobías, A., Medina, C., Hernández-Barrera, L., Bibbins-Domingo, K., Lozano, R., & Moran, A. E. (2015). Global Overview of the Epidemiology of Atherosclerotic Cardiovascular Disease. Archives of Medical Research, 46(5), 328–338. https://doi.org/10.1016/j.arcmed.2015.06.006.
6. Ray, K. K., Wright, R. S., Kallend, D., Koenig, W., Leiter, L. A., Raal, F. J., Bisch, J. A., Richardson, T., Jaros, M., Wijngaard, P. L. J., and Kastelein, J. J. P. (2020). Two Phase 3 Trials of Inclisiran in Patients with Elevated LDL Cholesterol. New England Journal of Medicine, 382(16), 1507–1519. https://doi.org/10.1056/NEJMoa1912387.
7. NICE. National Institute for Health and Care Excellence. TA733: Inclisiran for treating primary hypercholesterolaemia or mixed dyslipidaemia. Available at: https://www.nice.org.uk/guidance/ta733 (last accessed Nov 2021). National Institute for Health and Care Excellence; 2021.
8. Ostwald DA, Schmitt M, Peristeris P, Gerritzen T, Durand A. The Societal Impact of Inclisiran in England: Evidence From a Population Health Approach. Value Health. 2023 Sep;26(9):1353-1362. doi: 10.1016/j.jval.2023.05.002.
9. Country-specific inputs.
10. Same strategy for all countries based on: Kotseva, K., Gerlier, L., Sidelnikov, E., Kutikova, L., Lamotte, M., Amarenco, P., and Annemans, L. (2019). Patient and caregiver productivity loss and indirect costs associated with cardiovascular events in Europe. European Journal of Preventive Cardiology, 26(11), 1150–1157. https://doi.org/10.1177/2047487319834770.
11. Leontief, W. W. (1937). Interrelation of Prices, Output, Savings, and Investment. The Review of Economics and Statistics, 19(3), 109. https://doi.org/10.2307/1927343; Leontief, W. W. (1944). Output, Employment, Consumption, and Investment. The Quarterly Journal of Economics, 58(2), 290. https://doi.org/10.2307/1883321.
12. Same strategy for all countries based on: Butler, E. N., and Evenson, K. R. (2014). Prevalence of physical activity and sedentary behavior among stroke survivors in the United States. Topics in Stroke Rehabilitation, 21(3), 246–255. https://doi.org/10.1310/tsr2103-246.
13. Fischbach, R., and Wollenberg, K. (2007). Volkswirtschaftslehre 1 Einführung und Grundlagen (13th ed.). Oldenbourg Verlag.
14. Analysis for France is in progress.
15. Institute for Health Metrics and Evaluation (IHME) at the University of Washington. Cardiovascular death cases 2019, accessed on: 9/15/2023. Used with permission. 11 OECD (2023), Pharmaceutical spending (indicator). doi: 10.1787/998febf6-en.
16. For Japan we have preliminary results.
17. OECD (2023), Pharmaceutical spending (indicator). doi: 10.1787/998febf6-en.

Acknowledgements: This study was commissioned and sponsored by Novartis.