A systematic literature review of economic evaluations in primary biliary cholangitis

EE698

Alex Pashley,[1] Elaine A Böing,[2] Peter Serafini,[2] Emma Worthington[1]

[1]*Costello Medical, Cambridge, UK;* [2]*Ipsen, Cambridge, MA, USA*

Presenting author: Elaine A Böing (elaine.boing@ipsen.com)

Background

- Primary biliary cholangitis (PBC) is a rare autoimmune liver disease characterised by progressive cholestasis and biliary fibrosis.[1]
- Up to 40% of patients with PBC do not respond to first-line therapy with ursodeoxycholic acid (UDCA), leading to disease progression and, for some patients, the eventual need for liver transplantation.[2]
- PBC-related healthcare resource utilisation and expenditure are increasing, due, in part, to limited cost-effective second-line therapies for the treatment of PBC.[3,4]

Objective

A systematic literature review (SLR) was conducted to identify economic evaluations relevant for formulary decisions in PBC.

Methods

- The SLR was conducted in accordance with guidance outlined by the Cochrane Collaboration,[5] Preferred Reporting Items for Systematic Reviews and Meta-Analyses (PRISMA)[6] and National Institute of Health and Care Excellence (NICE).[7]
- Relevant articles were identified through searches of MEDLINE, EMBASE and the Health Technology Assessment (HTA) Database in November 2022.
- In addition, congress proceedings from 2021–2022 (n=8), HTA/health economic websites (n=19) and SLR and HTA bibliographies were hand searched.
- Articles were eligible for inclusion if they met the criteria presented in **Figure 1**.
 - Non-English language studies, studies in children/adolescents, in vitro/animal studies and non-original research studies were excluded.
 - Review of abstracts and full texts against the pre-defined eligibility criteria was performed by two independent reviewers; a third independent reviewer was consulted where necessary.
 - Data from included studies were extracted into a pre-specified extraction table.
- The quality of all included economic evaluations was assessed using the Drummond checklist.[8]

Results

- Of 1,480 and 1,124 records identified from database and supplementary searches, respectively, nine articles reporting on eight unique studies met the inclusion criteria (**Figure 2**; **Table 1**).
 - Included studies were from five countries: the United Kingdom (UK), the United States of America (USA), Ireland, Canada and Norway.
 - Critique of the included studies using the Drummond checklist found that most studies were well reported, with the notable exception of the accuracy of cost and outcomes measurements, as resource use was not reported separately from costs in most studies.
- Two types of economic evaluations were reported: cost-utility and cost-effectiveness (**Figure 3**).
 - Markov models were the most commonly used model design, and three quarters of the models used a lifetime horizon (**Figure 3**).
 - Most models included PBC-specific and liver disease-related health states (**Table 1**).
- The majority of studies evaluated obeticholic acid (OCA; **Figure 3**), of which four were submissions to HTA authorities.[10,12,13,15]
 - Model drivers for these studies are summarised in **Table 2**.
- Notably, no studies evaluated fibrates in PBC, and no economic evaluations were published since 2017 which may limit their applicability to current costs.

CONCLUSIONS

- There are limited cost-effectiveness analyses in PBC.
- Findings from this SLR highlight the need for additional, up-to-date economic evaluations and alternative therapies in PBC, which would drive choice for patients and healthcare systems, and a competitive treatment landscape.

Figure 1. Eligibility criteria for inclusion

P	Population	Unselected[a] adult patients (aged ≥18 years) with PBC
I	Intervention	Elafibranor; UDCA; OCA; fibrates; other future treatments (e.g. seladelpar)
C	Comparators	Best supportive care; placebo; any other comparators
O	Outcomes	Incremental cost-effectiveness ratios; cost per clinical outcome; total quality-adjusted life years (QALYs); total life years gained; total costs; incremental costs and QALYs
S	Study design	Cost-utility; cost-effectiveness; cost-consequence; cost-benefit; cost-minimisation

[a]A selected population was defined as a population of patients with PBC who were selected for fulfilling the same additional criterion (e.g. pregnancy, having a specific comorbidity), reducing applicability to the general PBC population.

Figure 2. PRISMA flowchart of identified economic evaluations

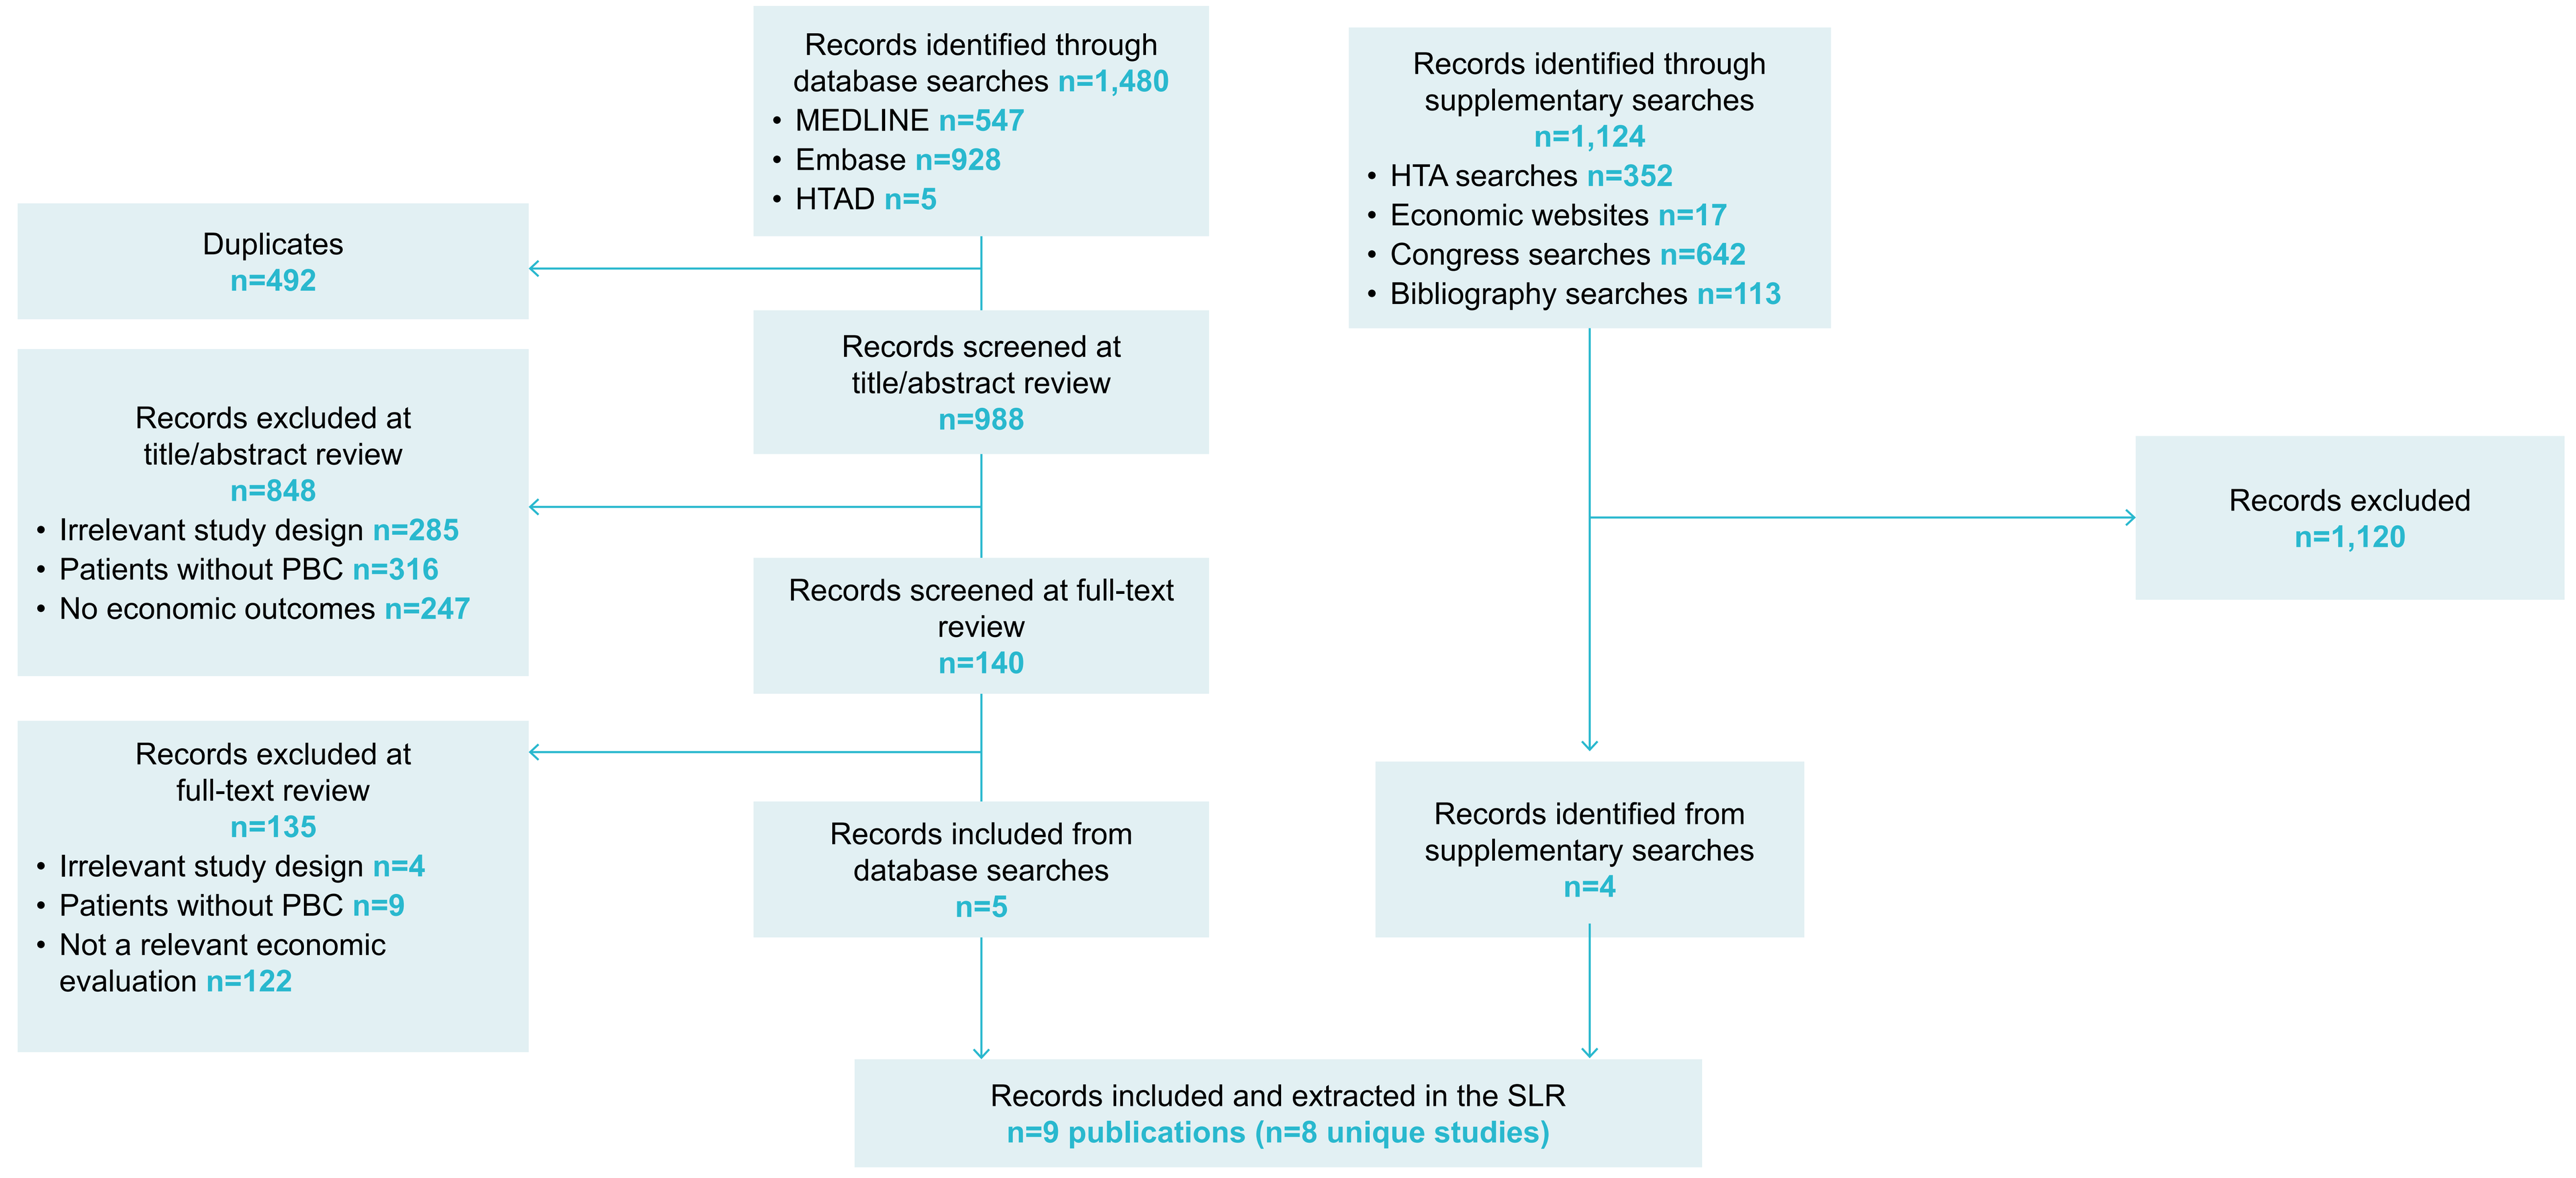
Records identified through database searches n=1,480
• MEDLINE n=547
• Embase n=928
• HTAD n=5
Records identified through supplementary searches n=1,124
• HTA searches n=352
• Economic websites n=17
• Congress searches n=642
• Bibliography searches n=113
Duplicates n=492
Records screened at title/abstract review n=988
Records excluded at title/abstract review n=848
• Irrelevant study design n=285
• Patients without PBC n=316
• No economic outcomes n=247
Records excluded n=1,120
Records screened at full-text review n=140
Records excluded at full-text review n=135
• Irrelevant study design n=4
• Patients without PBC n=9
• Not a relevant economic evaluation n=122
Records included from database searches n=5
Records identified from supplementary searches n=4
Records included and extracted in the SLR
n=9 publications (n=8 unique studies)

Table 1. Summary of included studies

#	Study name	Country	Perspective	Patient population	Model health states
1	Boberg 2013[9]	Norway	Public healthcare payer	Adult patients with PBC treated with UDCA	3 health states: alive without liver transplantation, alive after liver transplantation and death
2	CADTH 2017[10]	Canada	Public healthcare payer	Adult patients with PBC with inadequate response to or unable to tolerate UDCA	10 health states: including 3 PBC-specific health states (low, moderate and high risk), decompensated cirrhosis, HCC, pre-liver transplant, liver transplant, post liver transplant, PBC re-emergence and excess mortality
3	Longworth 2003[11]	UK	Public healthcare payer	Patients aged ≥16 years with PBC, ALD or PSC listed for an isolated liver transplant	NR
4	NCPE 2017[12]	Ireland	Healthcare payer	Adults with inadequate response to or unable to tolerate UDCA	10 health states: including 3 representing progression of PBC (based on alkaline phosphatase and bilirubin biomarkers) and 7 representing liver disease clinical outcomes (entered once patients progress to decompensated cirrhosis or HCC)
5	NICE 2017[13]	UK	NHS and personal social service	Adult patients with PBC with inadequate response to or unable to tolerate UDCA	9 health states: 3 biomarker-related health states (low, moderate and severe) and 6 liver disease-related health states (decompensated cirrhosis, HCC, pre-transplant state, transplantation, re-emergence of PBC and death)
6	Pasha 1999[14]	Canada	Societal	Patients with PBC from the Mayo and Canadian UDCA trials	Unclear; major events included ascites, varices, variceal bleeds, encephalopathy, liver transplantation and death
7	Samur 2017[4]	USA	NR	Adult patients with PBC with inadequate response to UDCA	PBC health states (stages 1–3; as defined by the Ludwig scoring system) and compensated cirrhosis
8	SMC 2017[15]	UK	Public healthcare payer	Adults patients with PBC with inadequate response or intolerant to UDCA	2 health states on model entry: moderate or high risk of liver disease (high risk also covering compensated cirrhosis)

Figure 3. Characteristics of included economic evaluations (n=8)

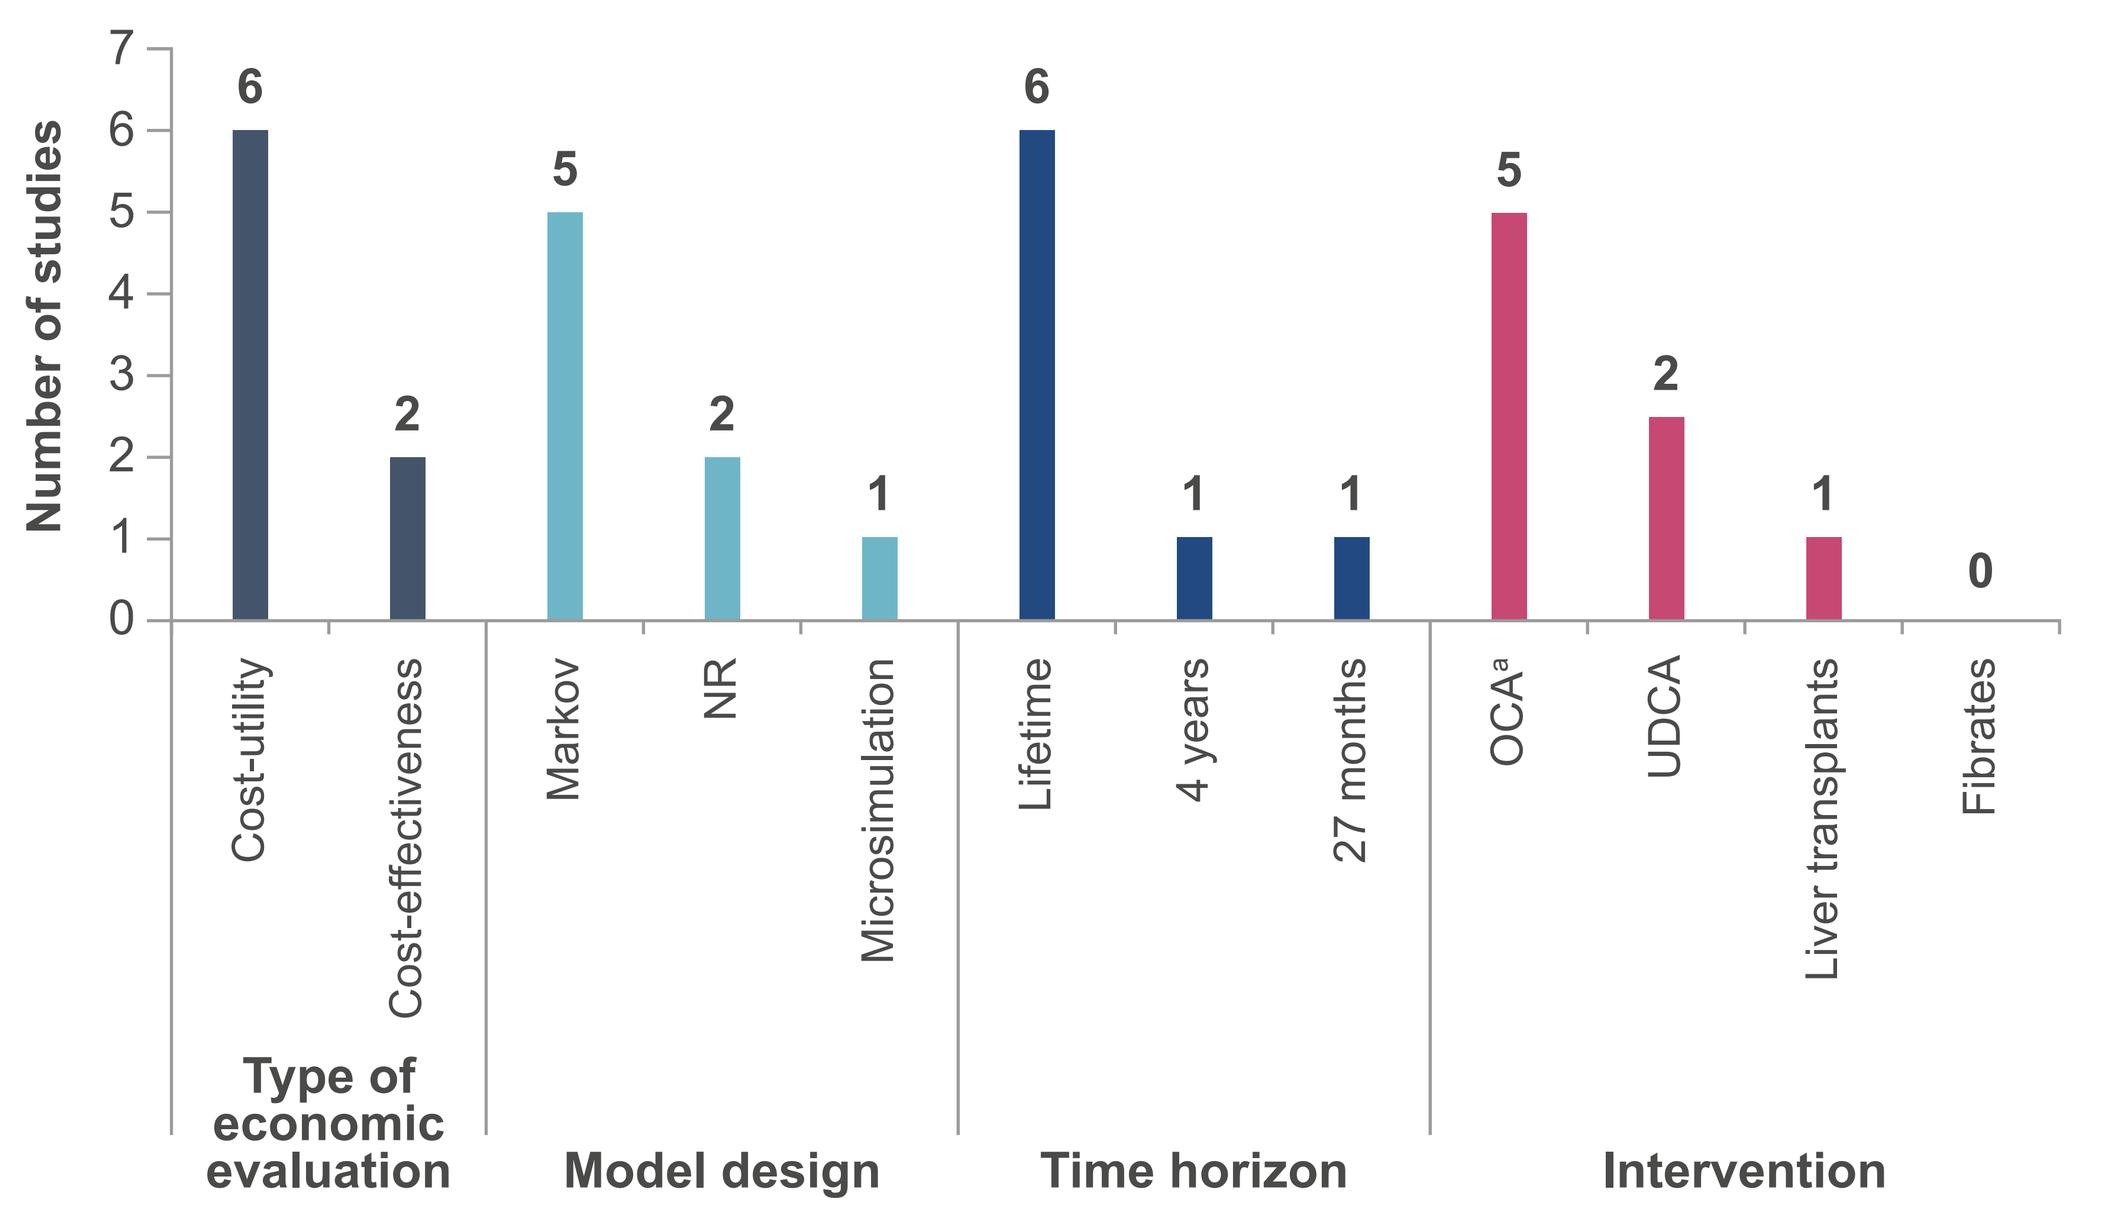
Number of studies
7
6
5
4
3
2
1
0
6
2
5
2
1
6
1
1
5
2
1
0
Cost-utility
Cost-effectiveness
Markov
NR
Microsimulation
Lifetime
4 years
27 months
OCA[a]
UDCA
Liver transplants
Fibrates
Type of economic evaluation
Model design
Time horizon
Intervention

[a]Two studies examined OCA in combination with UDCA.

Table 2. Model drivers reported in studies evaluating OCA

Study name	Model drivers
CADTH 2017[10]	Time horizon; calibration of PBC transition probabilities; discounting; source of utility data
NCPE 2017[12]	Cost of OCA
NICE 2017[13]	Health state utility values for the biomarker component of the model; PBC transition states
Samur 2017[4]	Time horizon; calibration of PBC transition probabilities; discounting; cost of OCA
SMC 2017[15]	Time horizon; calibration of PBC transition probabilities

Abbreviations

ALD: alcohol-related liver disease; HCC: hepatocellular carcinoma; HTA: health technology assessment; HTAD: Health Technology Assessment Database; NHS: National Health Service; NICE: National Institute of Health and Care Excellence; NR: not reported; OCA: obeticholic acid; PBC: primary biliary cholangitis; PRISMA: Preferred Reporting Items for Systematic Reviews and Meta-Analyses; PSC: primary sclerosing cholangitis; QALY: quality-adjusted life year; SLR: systematic literature review; UDCA: ursodeoxycholic acid; UK: United Kingdom; USA: United States of America.

References

1. Lindor KD *et al.* Hepatol 2019;69:394–419; **2.** Corpechot C *et al.* J Hepatol 2011;55:1361–1367; **3.** Hayat M *et al.* Gastroenterol. 2019;156:S–1320; **4.** Samur S *et al.* Hepatol 2017;65:920–928; **5.** Higgins J *et al.* Cochrane Handbook for Systematic Reviews of Interventions. 2019; **6.** Page M *et al.* BMJ 2021;372:n71; **7.** NICE. Process and methods [PMG24]. Last updated: 10 February 2022. Available at: https://www.nice.org.uk/process/pmg24/resources/single-technology-appraisal-and-highly-specialised-technologies-evaluation-user-guide-for-company-evidence-submission-appendices-10956190861/chapter/instructions-for-companies. Last accessed: May 2023; **8.** Drummond M *et al.* Oxford University Press 2015; **9.** Boberg KM *et al.* Aliment Pharmacol Ther 2013;38:794–803; **10.** Anonymous. CADTH Common Drug Reviews 2017;08:08; **11.** Longworth L *et al.* Liver Transpl 2003;9(12):1295–1307; **12.** NCPE. Cost-effectiveness of obeticholic acid (Ocaliva®) for the treatment of primary biliary cholangitis in combination with ursodeoxycholic acid (UDCA) in adults with an inadequate response to UDCA or as monotherapy in adults unable to tolerate UDCA. Last updated: October 2022. Available at: https://www.ncpe.ie/drugs/obeticholic-acid-ocaliva/. Last accessed: May 2023; **13.** NICE. Obeticholic acid for treating primary biliary cholangitis [TA443]. Published: 26 April 2017. Available at: https://www.nice.org.uk/guidance/ta443/. Last accessed: May 2023; **14.** Pasha T. *et al.* Hepatol 1999;29:21–26; **15.** SMC. Obeticholic acid (Ocaliva®). Published: 12 June 2017. Available at: https://www.scottishmedicines.org.uk/medicines-advice/obeticholic-acid-ocaliva-fullsubmission-123217/. Last accessed: May 2023.

Author contributions Substantial contributions to study conception/design, or acquisition/analysis/interpretation of data: **AP, EAB, PS, EW;** Drafting of the publication, or revising it critically for important intellectual content: **AP, EAB, PS, EW;** Final approval of the publication: **AP, EAB, PS, EW.**

Disclosures AP, EW: Employees of Costello Medical; **EAB:** Employee and shareholder of Ipsen; **PS:** Employee of Ipsen.

Medical writing support The authors thank Orla Woodward, PhD, and Beverley Wilson, PhD, of Costello Medical, UK, for providing medical writing support, and the Costello Medical Creative team for design support, which was sponsored by Ipsen in accordance with Good Publication Practice guidelines.

To download the poster, please scan the QR code.

Copies of this ePoster obtained through the QR code are for personal use only and may not be reproduced without written permission from the authors.

Presented at ISPOR EU 2023 | Copenhagen, Denmark | 12–15 November 2023

This study was sponsored by Ipsen