PCR251

ECONOMIC AND HUMANISTIC BURDEN OF VISUAL IMPAIRMENT AND BLINDNESS IN PATIENS AND THEIR CAREGIVERS WITH RETINITIS PIGMENTOSA OR LEBER CONGENITAL AMAUROSIS (e-BIRDs study)

Carles Blanch[1] Jose Borges[1], Núria Perulero[2], Neus Casals[2], David Sánchez[3], Andrés Mayor[4], J. Joaquín Gil[5].

1 Novartis, 2 IQVIA, 3 ONERO , 4 Retina Murcia, 5 Visión España.

INTRODUCTION

Inherited Retinal Dystrophies (IRDs) comprise a heterogeneous group of genetic disorders affecting the retina, caused by mutations in more than 300 genes. **Retinitis pigmentosa (RP)** accounts for approximately half of IRD, with a prevalence ranging from 1 in 3,454 to 1 in 8,357 [1]. This is a group of related eye disorders that cause progressive loss of vision. Although **Leber congenital amaurosis (LCA)** does not have as high a prevalence as RP, with the latter ranging from 1 in 33,000 to 1 in 81,000, LCA is characterized by its acute severity as well as by the early age at which it debuts in child patients. People with this disorder usually have impaired vision throughout their lives from childhood.

OBJECTIVE

- To estimate the direct and indirect costs during the patient's/caregiver's life of visual impairment or blindness due to patients' RP and LCA
- To characterize the humanistic burden of RP or LCA in patients and caregivers in Spain

METHODS

- Cross-sectional study of patients with RP or LCA residing in Spain
- Primary data was collected through telephone surveys completed in Spanish, which took no more than 60 minutes to complete.
- Information about pediatric patients was collected from their parents or legal guardians.
- Parents of patients <18 years of age also answered the questionnaire to assess the impact of the disease on caregivers.
- Participants were identified through a relevant Patient Advocacy Groups (PA) in Spain, among those who registered and using the support of the relevant PA.
- PAs were provided with clear inclusion/exclusion criteria to select relevant participants and all information collected was self-reported.
- The sample was as close as possible to a "freely found" naturalistic sample of those willing and qualified to participate, however, encouragement was given to ensure there was a reasonable representation of RP and LCA patients.
- At the time of recruitment, the PAs indicated the diagnosis of the respondent invited to participate, while the diagnosis of each patient was also self-reported within the survey.
- The patient's informed consent to participate in the survey was made after spontaneous and voluntary telephone contact with IQVIA so that it was registered in the survey.

RESULTS

Sociodemographic characteristics of patients with RP or LCA, as reported by patients (n=41), according to patient age groups and disease

Patient Characteristics		Total RP	Total LCA	TOTAL
Age	Mean (SD)	53.2 (10.5)	53.7 (4.5)	53.2 (10.7)
Gender	Female	18 (47,4%)	2 (66.7%)	20 (48.8%)
	Male	20 (52.6%)	1 (33.3%)	21 (51.2%)

Mean spent (€/year) of patients with RP or LCA, as reported by patients (n=41),

Item	Mean cost
Low vision aids/adaptation	399
Electronic devices/lamps/magnifiers	196
Drugs/supplements	267
Healthcare services	507
Medical visits	185
Paid caregivers	318
Home adaptations	246

Use of low-vision aids/adaptations, as reported by patients, in the total sample (n=41)

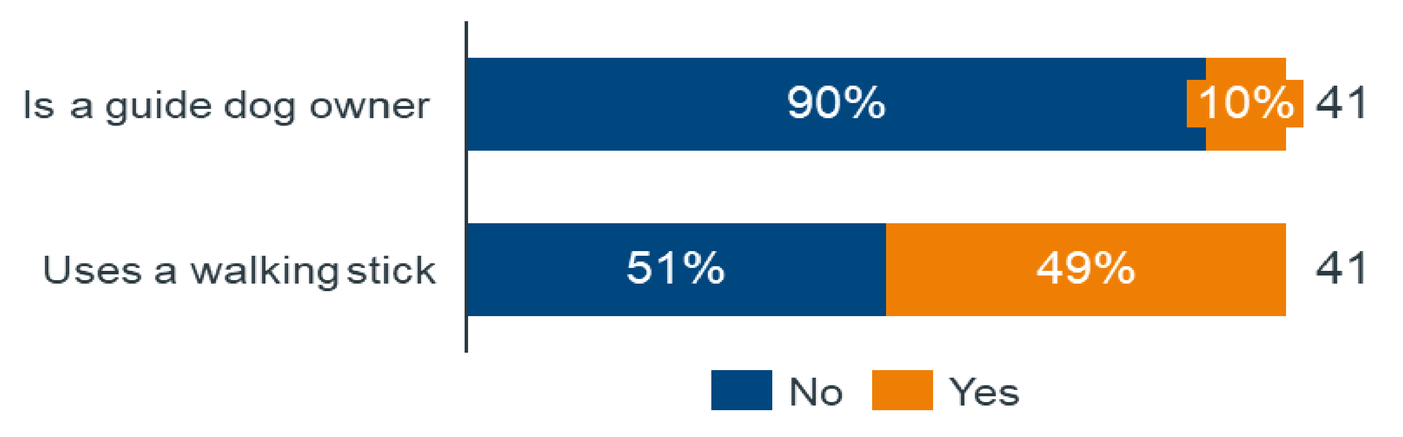

Is a guide dog owner
90%
10%
41
Uses a walking stick
51%
49%
41
No
Yes

Low-vision aids/adaptations related with visual functions (n=41)

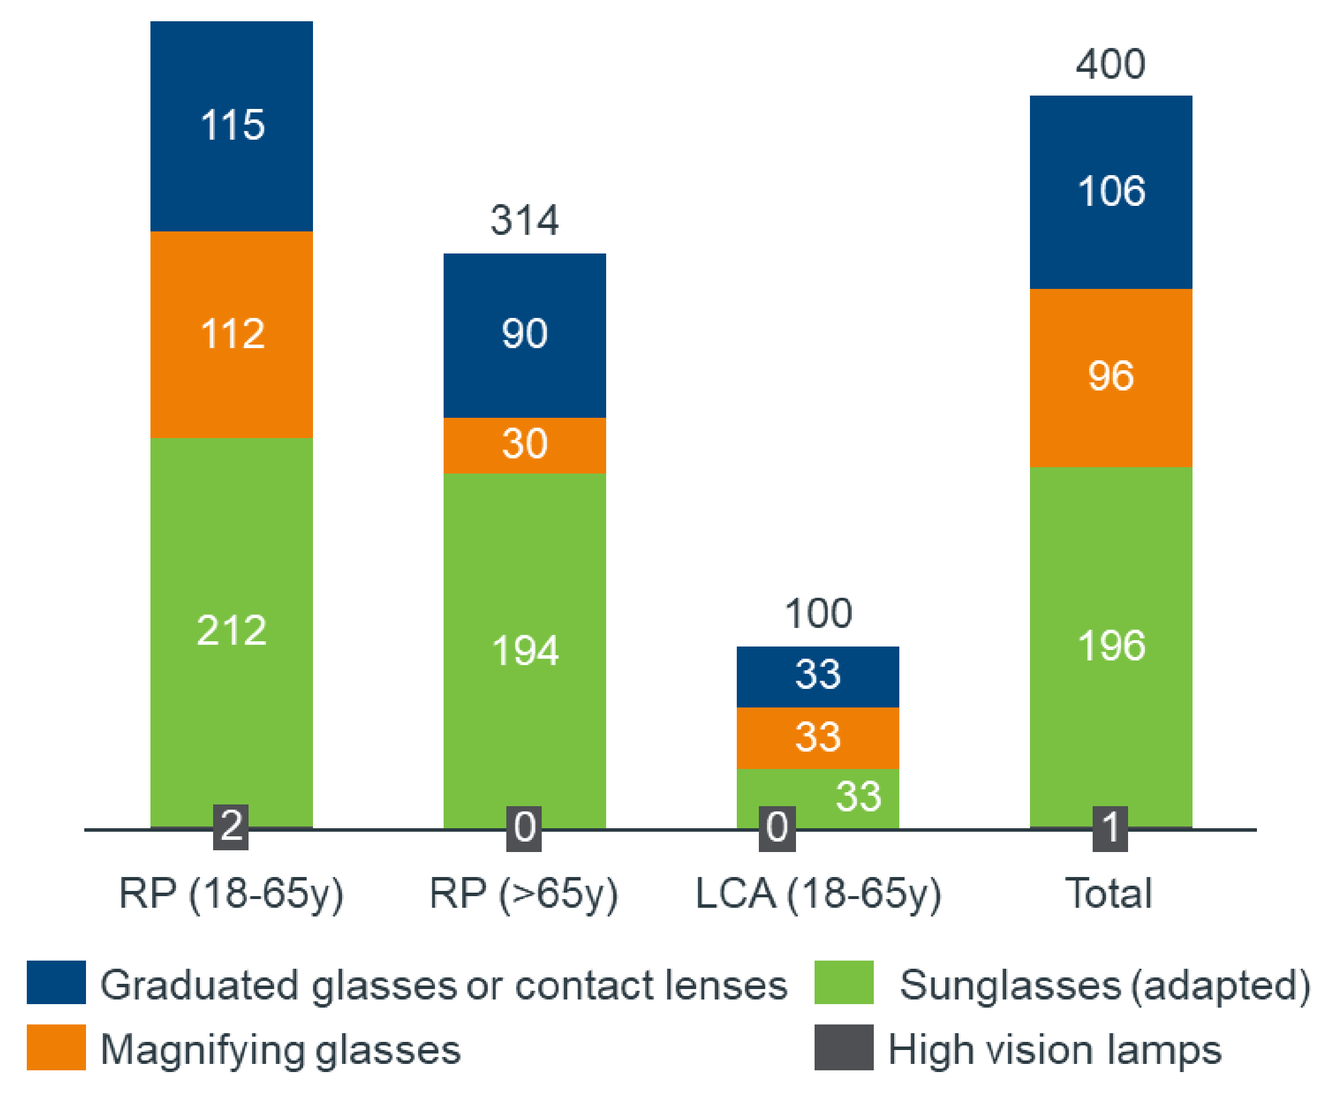

115
112
212
2
314
90
30
194
0
100
33
33
33
0
400
106
96
196
1
RP (18-65y)
RP (>65y)
LCA (18-65y)
Total
Graduated glasses or contact lenses
Sunglasses (adapted)
Magnifying glasses
High vision lamps

How do you attend to your medical appointments (n=38)

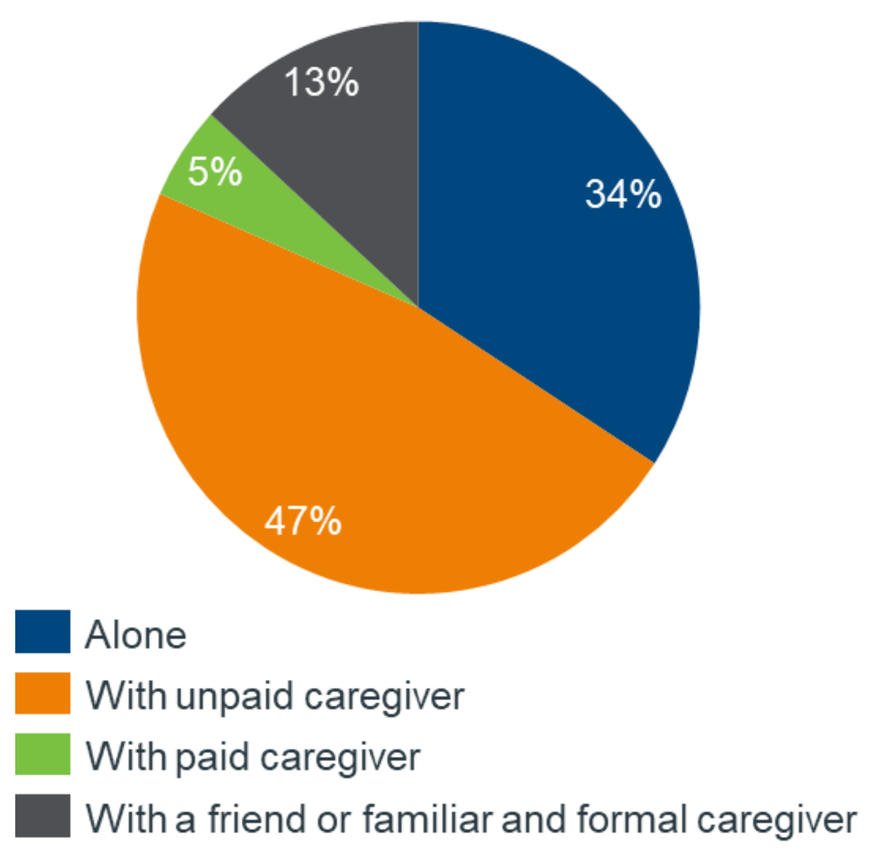

34%
47%
5%
13%
Alone
With unpaid caregiver
With paid caregiver
With a friend or familiar and formal caregiver

- 76.9% could see lights or shadows, and 82.1% experienced night blindness. 46.2% reported good central vision and 7.7% good peripheral vision. When asked about their level of vision and visual acuity, 41.4% responded blind or severely impaired.
- 92.7% had received a genetic test specifically for their inherited retinal disease. 63.1% of these cases reported to be tested after more than 5 years from the symptom's onset. 52.6% were not sure what gene was identified as causing their condition.

Total visits to health care services, in euros/year, as reported by patients, in the total sample (n=41)

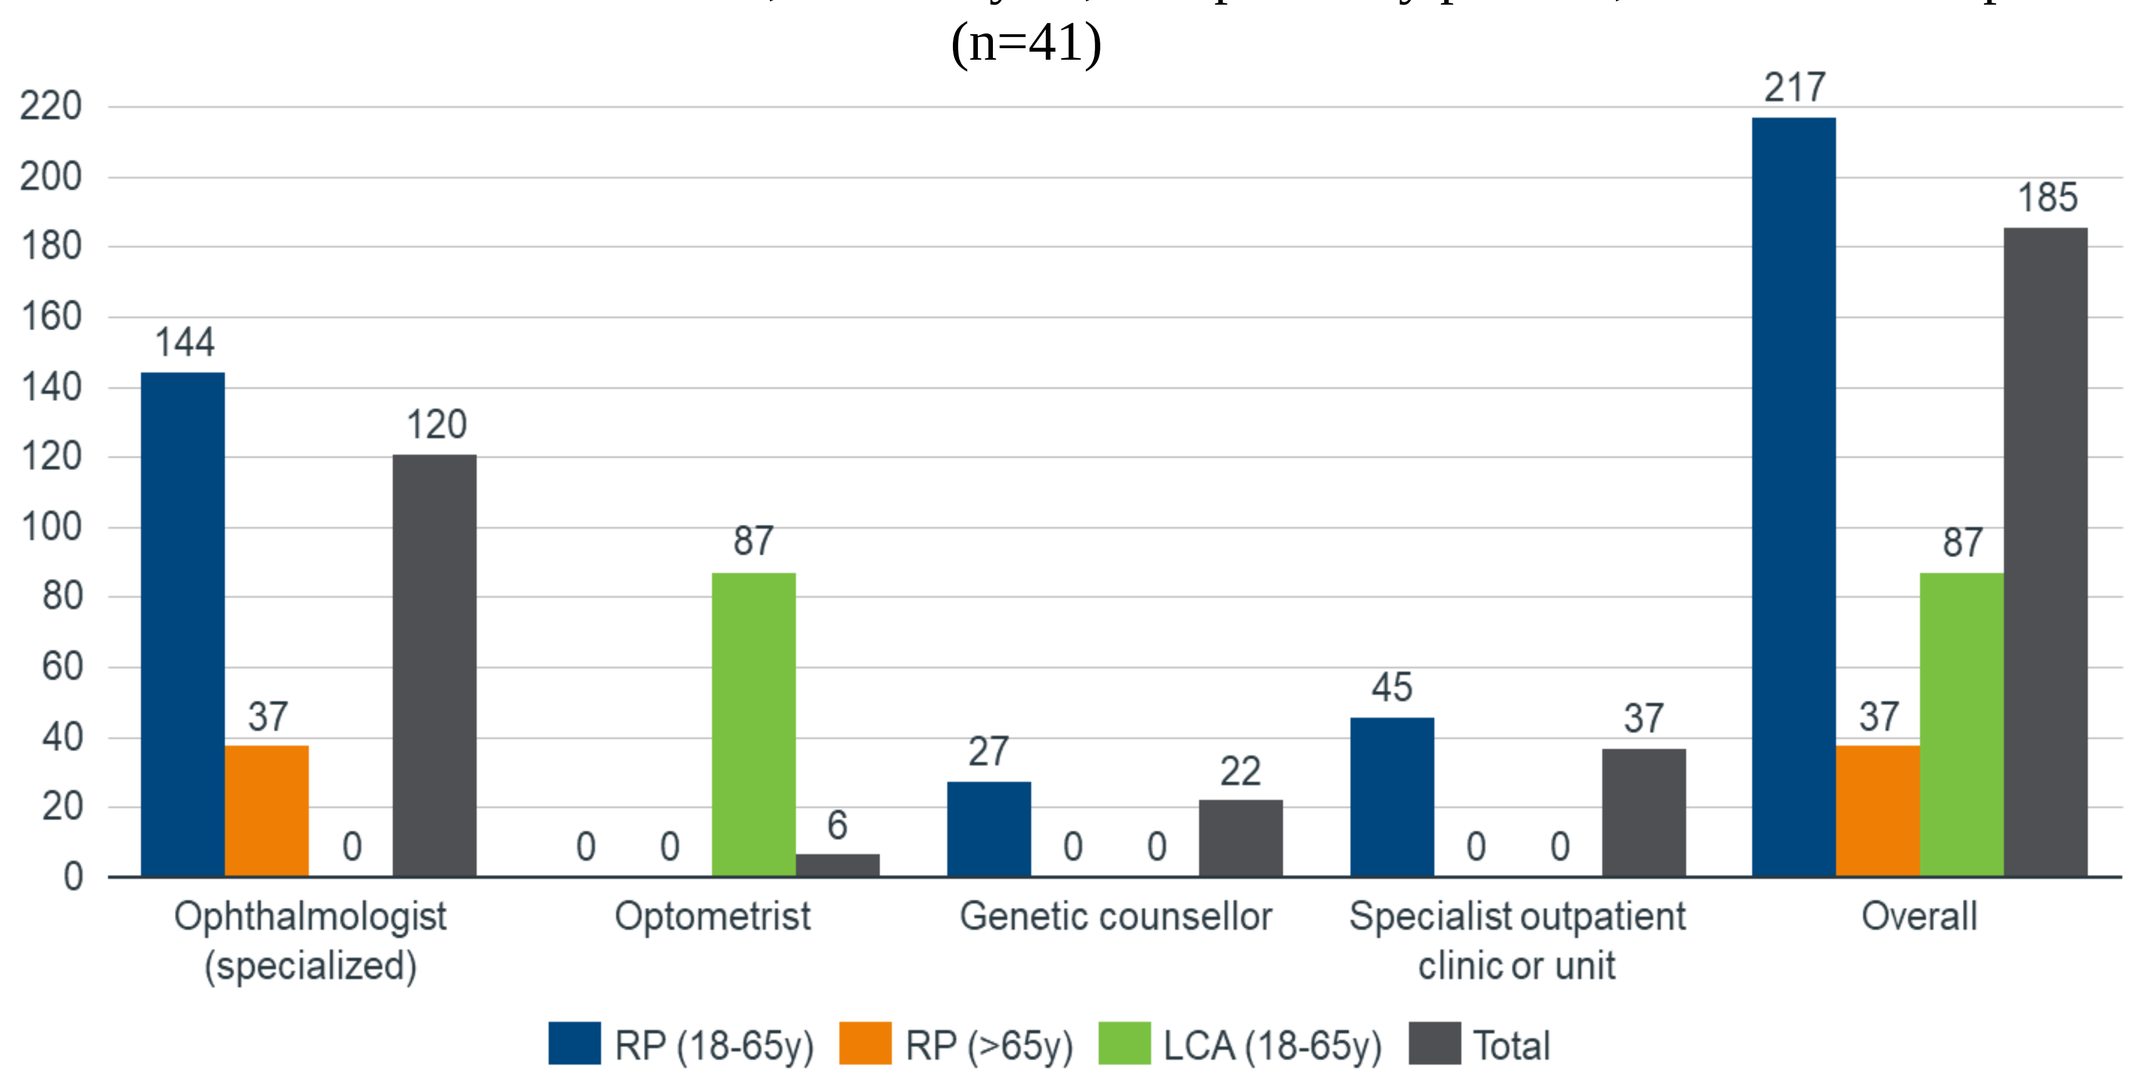

Ophthalmologist (specialized)
144
37
0
120
Optometrist
0
0
87
6
Genetic counsellor
27
0
0
22
Specialist outpatient clinic or unit
45
0
0
37
Overall
217
37
87
185
RP (18-65y)
RP (>65y)
LCA (18-65y)
Total

Uncovered patient's expenses in low-vision medications and supplements, as reported by patients, in the total sample (n=41), by age group and disease, in euros per patient/year

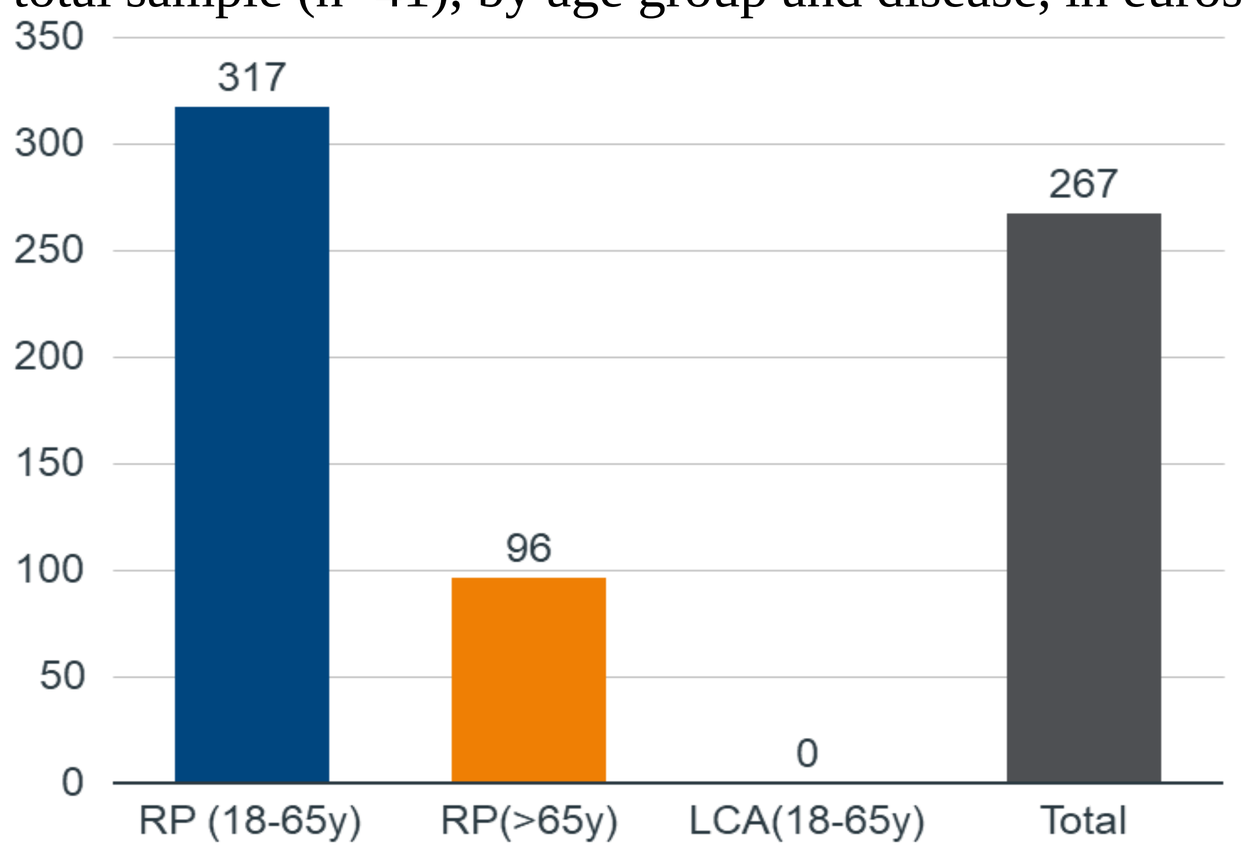

317
96
0
267
RP (18-65y)
RP(>65y)
LCA(18-65y)
Total

Humanistic burden of PR or LCA for caregivers in Spain (n=11)

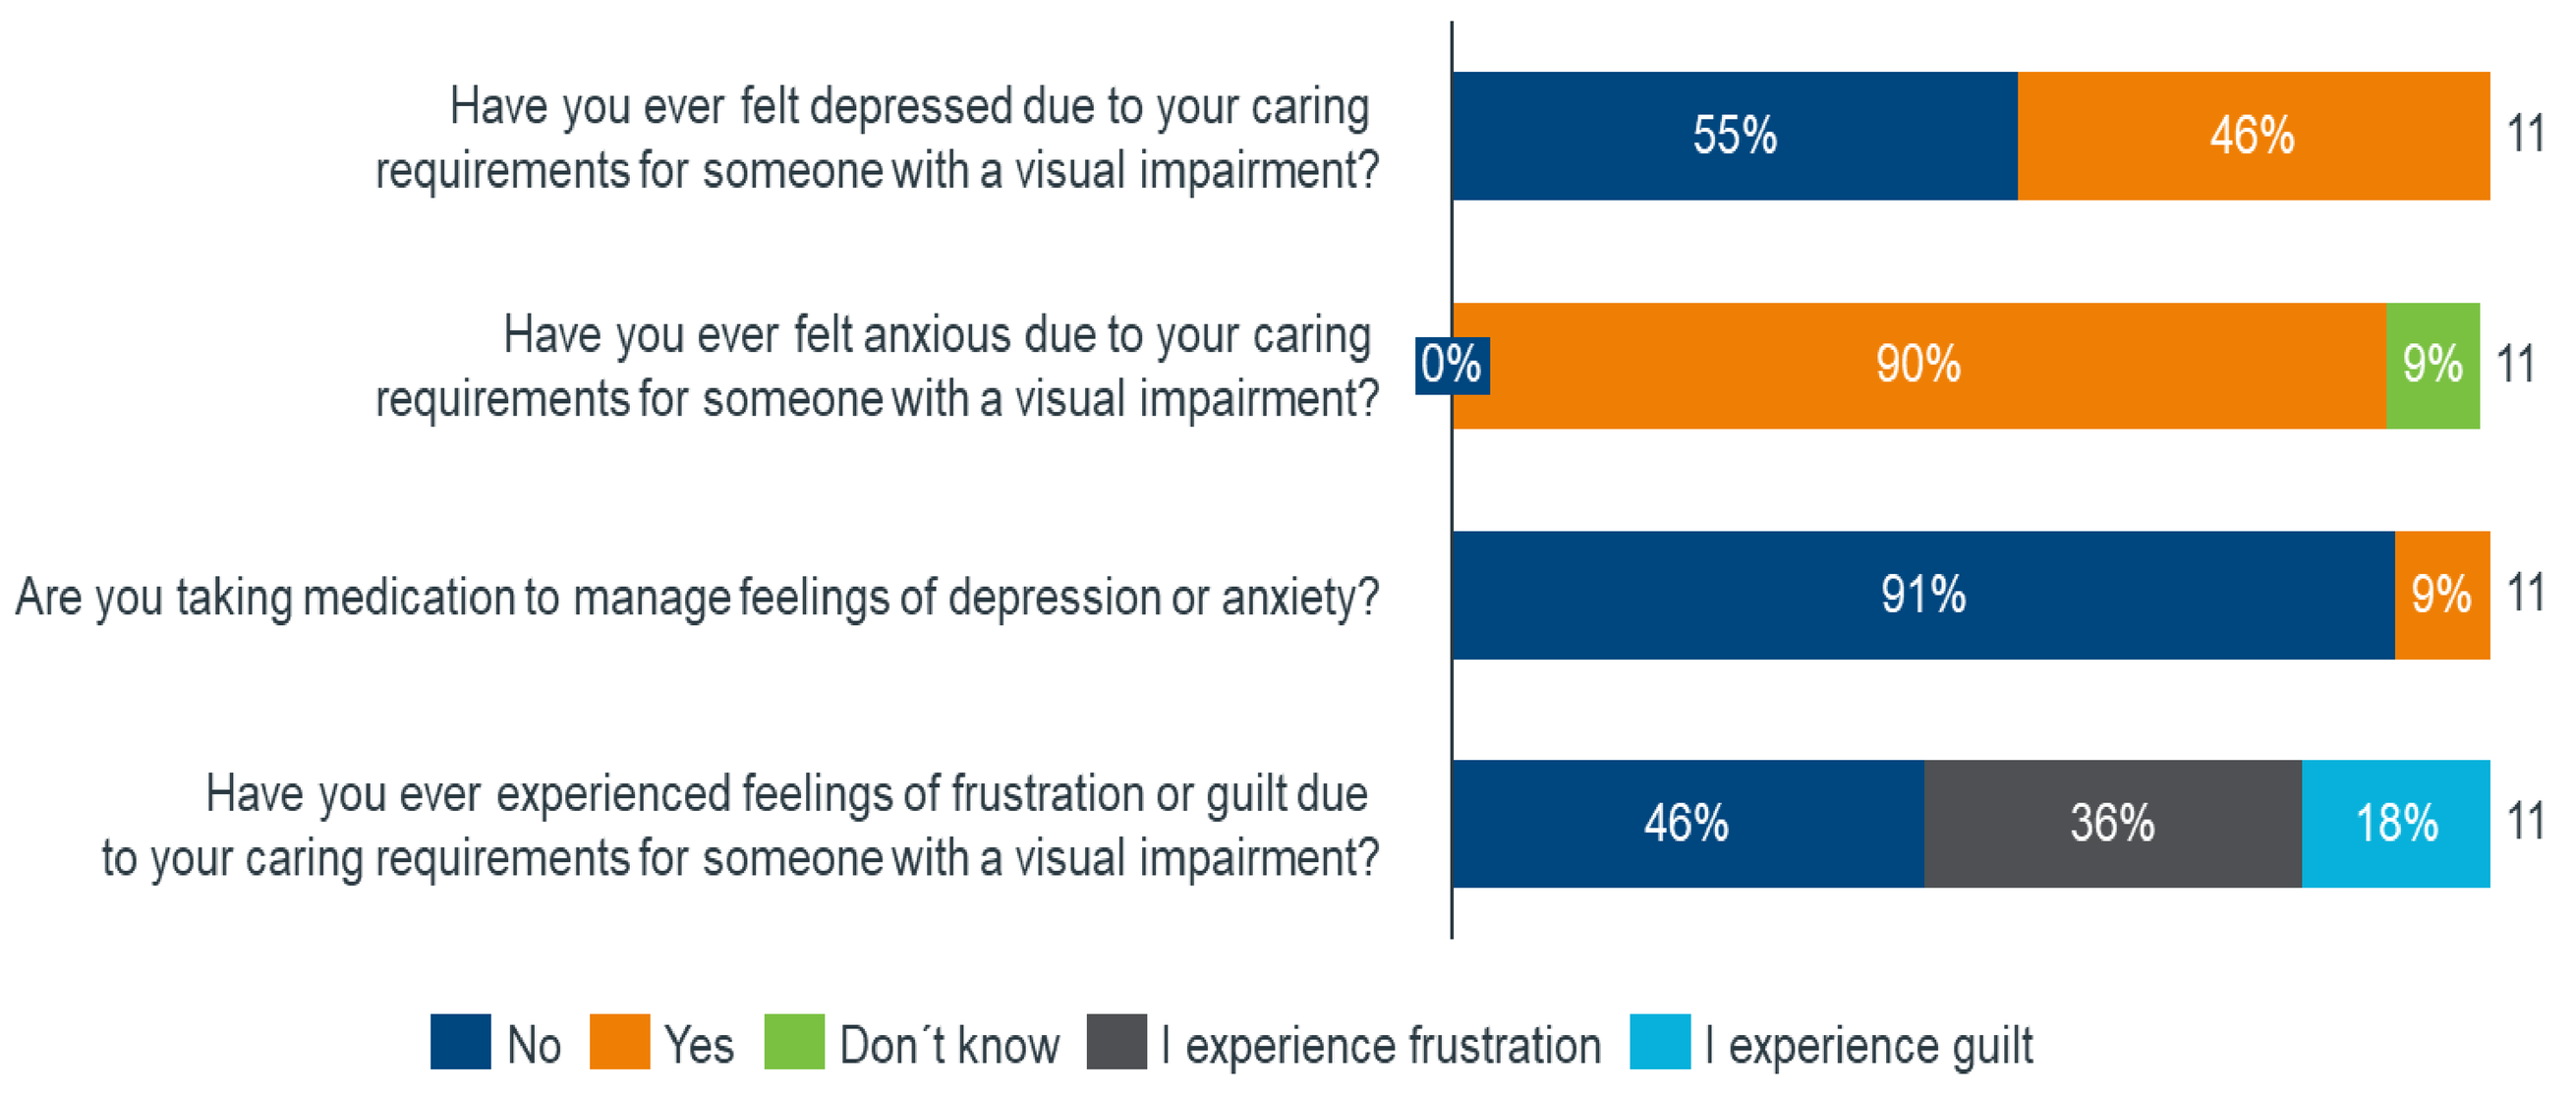

Have you ever felt depressed due to your caring requirements for someone with a visual impairment?
55%
46%
11
Have you ever felt anxious due to your caring requirements for someone with a visual impairment?
0%
90%
9%
11
Are you taking medication to manage feelings of depression or anxiety?
91%
9%
11
Have you ever experienced feelings of frustration or guilt due to your caring requirements for someone with a visual impairment?
46%
36%
18%
11
No
Yes
Don´t know
I experience frustration
I experience guilt

Percentage of the use of resources (Financial aid), as reported by patients, in the total sample (n=41)

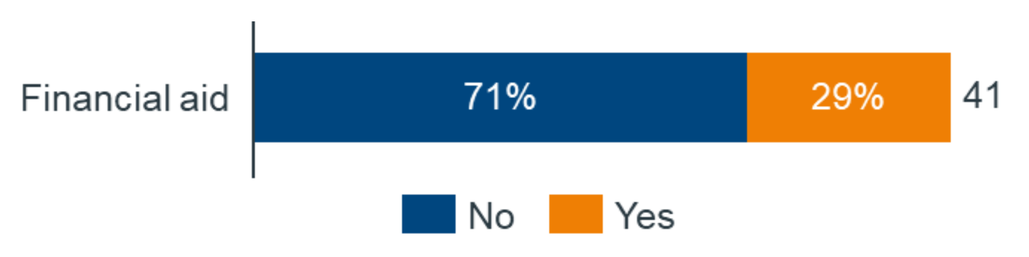

Financial aid
71%
29%
41
No
Yes

Percentage of patients with unpaid caregivers (n=41)

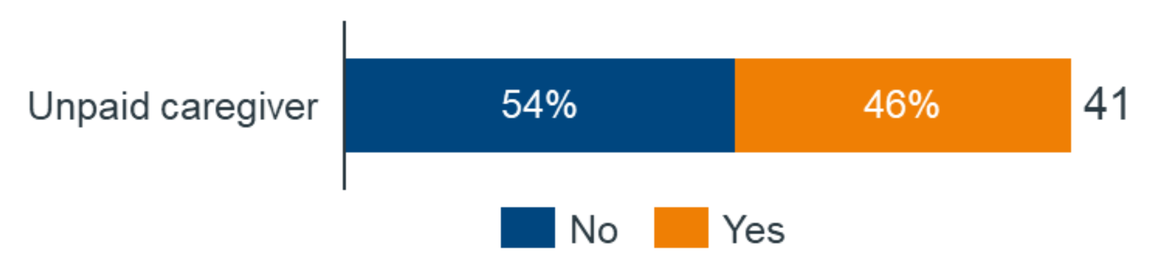

Unpaid caregiver
54%
46%
41
No
Yes

Percentage of the use of resources (Financial aid), as reported by patients, in the total sample (n=41)

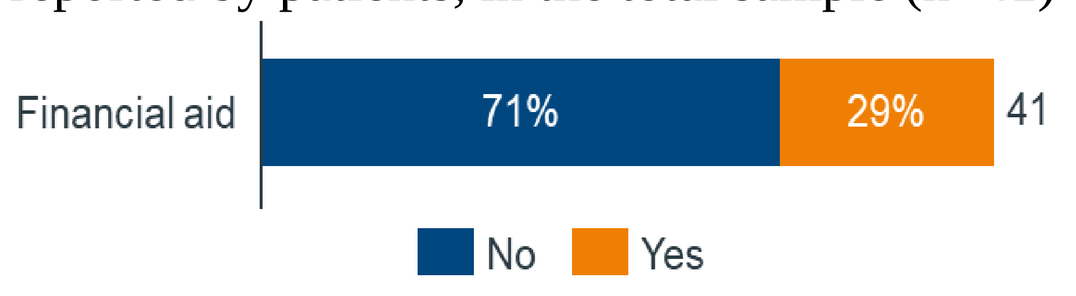

Financial aid
71%
29%
41
No
Yes

Patients' use of paid care. Use of Paid care, as reported by patients, in the total sample (n=41)

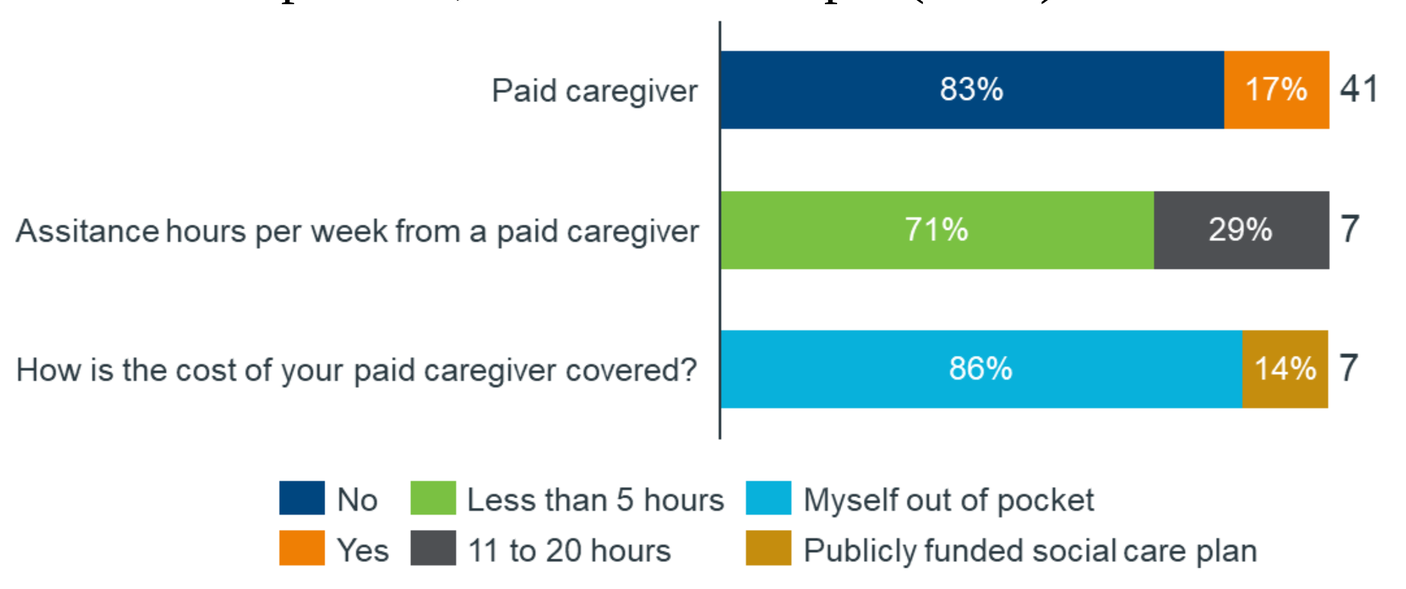

Paid caregiver
83%
17%
41
Assitance hours per week from a paid caregiver
71%
29%
7
How is the cost of your paid caregiver covered?
86%
14%
7
No
Yes
Less than 5 hours
11 to 20 hours
Myself out of pocket
Publicly funded social care plan

Situation of caregivers (n=11)

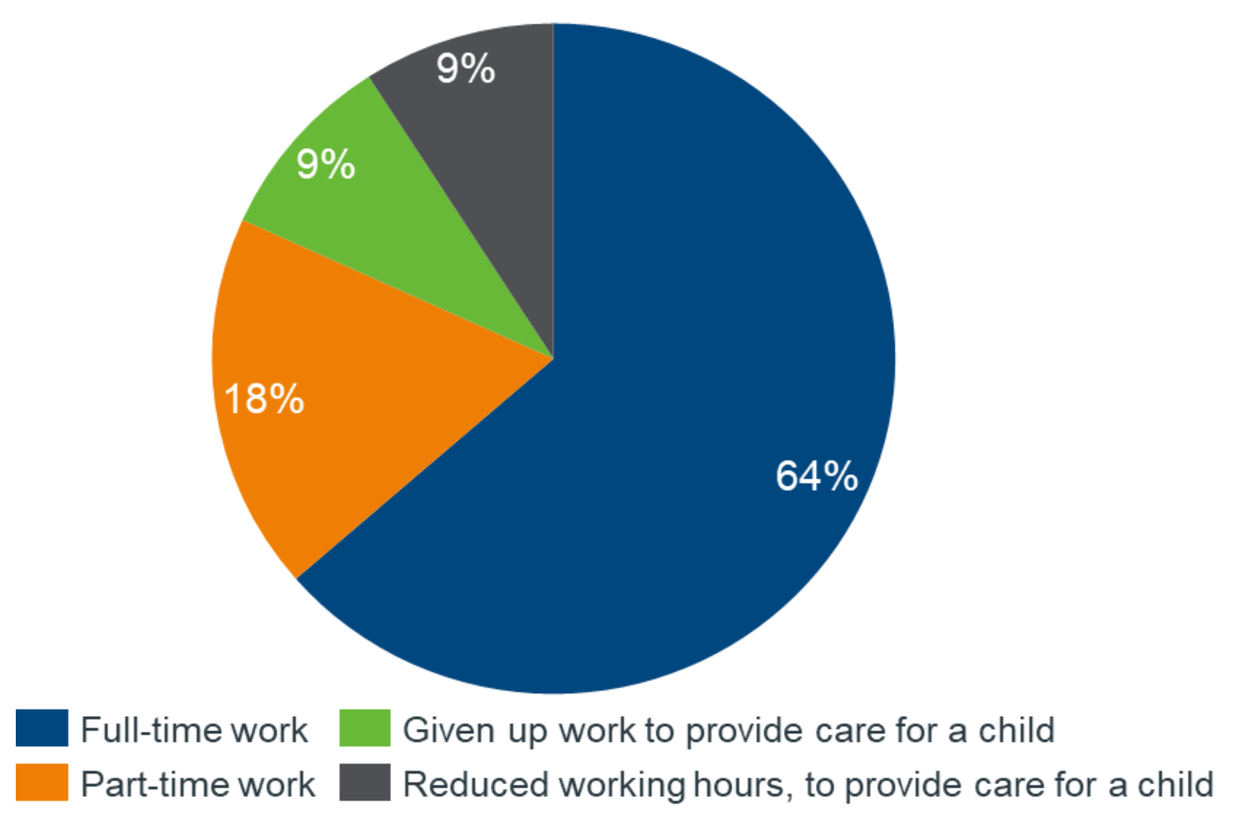

64%
18%
9%
9%
Full-time work
Part-time work
Given up work to provide care for a child
Reduced working hours, to provide care for a child

Number of hours spent as unpaid caregivers (n=17)

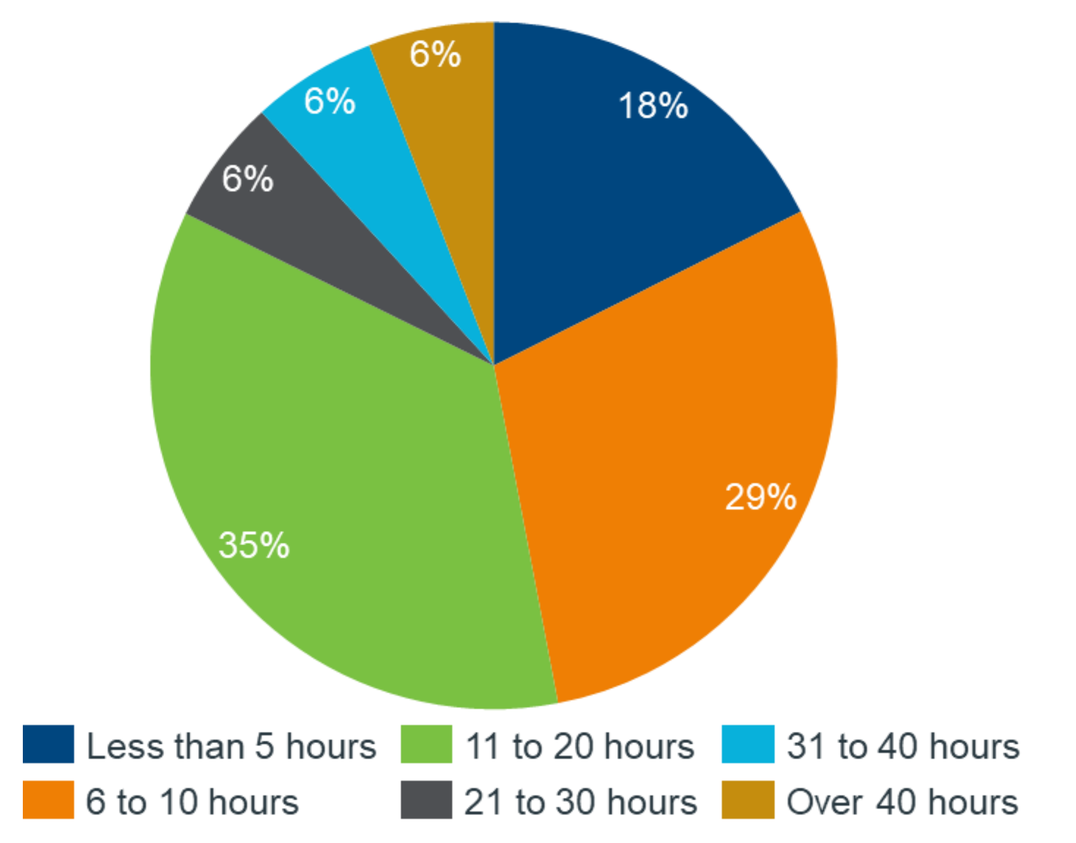

18%
29%
35%
6%
6%
6%
Less than 5 hours
6 to 10 hours
11 to 20 hours
21 to 30 hours
31 to 40 hours
Over 40 hours

Limitations

- ✓ Cross-sectional design (not collect changes over time)
- ✓ Selection bias (registered to support associations)
- ✓ Underrepresentation of LCA patients
- ✓ Self-reported data (recall bias)

CONCLUSIONS

- ➢ A high utilization rates of medical services combined with its low economic impact for patients, suggest that the NHS finances most of all these services.
- ➢ Younger population are the ones who attend more and pay more for they follow-up visits with ophthalmologists and medications/aids.
- ➢ These diseases have a high personal annual cost of low-vision aids/adaptations to cover functional vision and not funded by the NHS.
- ➢ Whilst medical services use to be covered by NHS, the study showed serious humanistic burden mainly due to insufficient financing of care and restrictions in participating within the society.
- ➢ The study also showed the vulnerable position of caregivers in Spain which are mostly unpaid for their tasks and exposed to high psychological burden.