Systematic literature review for costs data in haemophilia

Bartlett C[1], Miller P[1], Bracewell J[1], McCool R[1], Jasso-Mosqueda JG[2], Bozzi S[2]

[1]York Health Economics Consortium, UK
[2]Sanofi, France

EE719

BACKGROUND AND OBJECTIVES

Haemophilia A and B are hereditary bleeding disorders resulting in deficiencies of FVIII and FIX clotting factors, respectively. Haemophilia can be severe, moderate, or mild according to the level of deficiency of these clotting factors. The hallmark of haemophilia is recurrent joint bleeding, a primary driver of its morbidity resulting in arthropathy.

Their management includes prevention and treatment of bleeding events. The therapeutic scheme is either episodic, also known as "on demand", or prophylactic. Episodic treatment is used to stop a patient's bleeding event while prophylactic is used to prevent bleeding from occurring [1].

Current guidelines (Medical and Scientific Advisory Council and World Federation of Haemophilia) recommend prophylaxis for patients with severe hemophilia and those with moderate hemophilia and a severe bleeding phenotype [2,3]. Current treatments available are factor replacement therapies (FRTs), non-FRTs, and gene therapy.

- FRTs can be either plasma-derived products or produced through recombinant methods [4]. They include standard half-life therapies, extended half-life therapies and bypassing agents
- Non-FRTs include substitution therapy (monoclonal antibody mimicking the action of FVIII) and haemostatic rebalancing agents [2]

While prophylaxis with FRTs has been the mainstay treatment of many years in patients without inhibitors [4-6], treatment can be complicated by inhibitors development due to the patient's immune response to infused factors. Recombinant or plasma derived by-pass agents are used to treat patients with inhibitors.

This systematic review (SR) was undertaken to identify costs data associated with adolescents and adults with haemophilia A and B, for those with or without inhibitors. The SR focuses on the US and the EU5 (France, Germany, Italy, Spain, UK) to help inform economic evaluations of haemophilia in these populations.

METHODS

Nine bibliographic databases and three conferences were searched to identify studies reporting costs data for adolescents and adults with haemophilia A or B. Targeted searches of three key technology assessment and regulatory agency websites (National Institute for Health and Care Excellence, Canadian Agency for Drugs and Technologies in Health, Institute for Clinical and Economic Review) and two non-database conference searches (European Hematology Association 2022 and World Federation of Hemophilia 2022) were also conducted. Studies published between 2011 and June 2022, conducted in US and EU5 and published in the English language were eligible.

Two reviewers independently assessed records for relevance, with disagreements resolved by discussion. One reviewer extracted data from each study, with a second reviewer checking the extraction. The study findings were summarised.

RESULTS

The searches identified 2,052 unique records. After abstract screening and full-text review, 31 studies (reported in 40 publications) were included for data extraction **(Figure 1)**. Of the 31 studies included, 14 were primary costing studies, 11 were cost-effectiveness analyses and six were cost-modelling studies, with varying haemophilia type and inhibitor status **(Table 1)**.

Figure 1. The simplified PRISMA flowchart (adapted from Moher et al., 2009 [7]

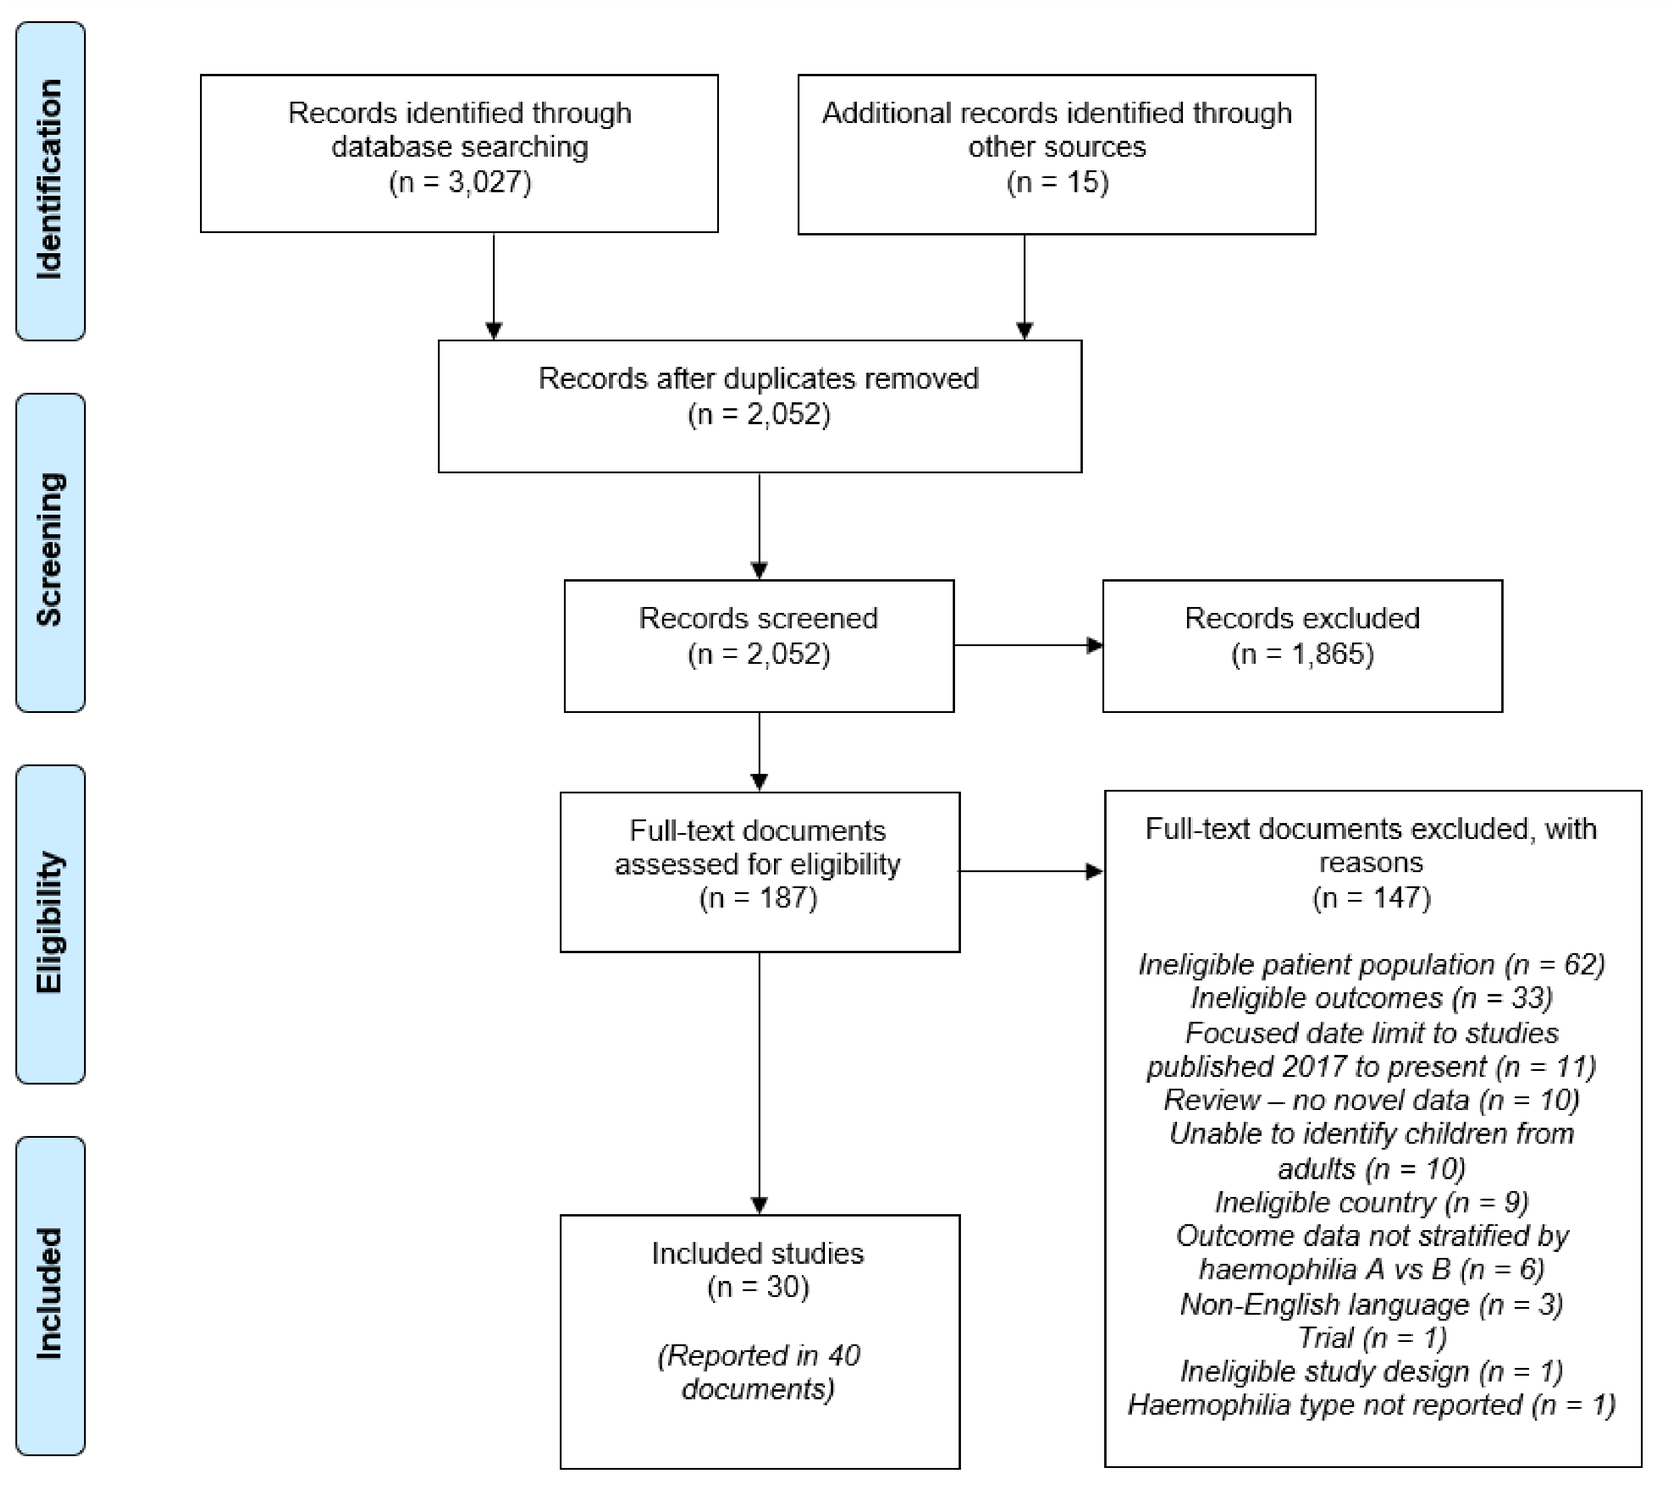
Identification
Screening
Eligibility
Included
Records identified through database searching (n = 3,027)
Additional records identified through other sources (n = 15)
Records after duplicates removed (n = 2,052)
Records screened (n = 2,052)
Records excluded (n = 1,865)
Full-text documents assessed for eligibility (n = 187)
Full-text documents excluded, with reasons (n = 147)
Ineligible patient population (n = 62)
Ineligible outcomes (n = 33)
Focused date limit to studies published 2017 to present (n = 11)
Review – no novel data (n = 10)
Unable to identify children from adults (n = 10)
Ineligible country (n = 9)
Outcome data not stratified by haemophilia A vs B (n = 6)
Non-English language (n = 3)
Trial (n = 1)
Ineligible study design (n = 1)
Haemophilia type not reported (n = 1)
Included studies (n = 30)
(Reported in 40 documents)

Table 1: Number of studies by haemophilia type and inhibitor status

Haemophilia type	Inhibitor status	Number of studies
Haemophilia A	With inhibitors	6
	Without inhibitors	11
	Mixed population of inhibitors	1
	Not reported	3
Haemophilia B	Without inhibitors	4
	Not reported	4
Haemophilia A and B	Without inhibitors	1
	Mixed population of inhibitors	1

Table 2: Bleeding costs by inhibitor status (US)

Type of costs	Inhibitor status	Description	Cost year	Total per patient costs
Direct medical	Patients without inhibitors	Annual costs per bleed	2019	Annual costs per bleed: $2,885 (excluding FIX cost) to $8,413 (excluding FVIII/FIX cost)*
	Patients with inhibitors	aPCC	2017/18	Treating a bleed (up to 6 days): $1,741 to $1,778
		rFVIIa	2017/18	Treating a bleed (up to 6 days): $13,635 to $13,925
		rpFVIII	2018	Treating a bleed (up to 6 days): $6,957
Indirect	Patients without inhibitors	Per treated bleed	2019	$1,163

Key: aPCC – activated prothrombin complex concentrate; FIX – factor IX; FVIII – factor VIII; rFVIIa – recombinant activated factor VII; rpFVIII – recombinant porcine-sequence factor VIII. *Patients with haemophilia A or B.

Table 3: Dental extraction and major surgeries direct costs by inhibitor status (EU5)

Type of costs	Inhibitor status	Description	Cost year	Total per patient costs
Direct medical	Patients without inhibitors	Overall costs	2019/20	Total costs: €235,723*
	Patients with inhibitors	aPCC	2018	€10,101 (dental extraction) to €126,596 (major surgery)
		rFVIIa	2018	Total costs: €14,263 (dental extraction) to €347,731 (major surgery

Key: aPCC – activated prothrombin complex concentrate; rFVIIa – recombinant activated factor VII.
*Patients with haemophilia B.

Table 4: Two-, five- and ten-year direct costs by inhibitor status (US and EU5)

Type of costs	Inhibitor status	Country	Description	Cost year	Total per patient costs
Direct medical at Two years	Patients without inhibitors	US	Episodic treatment	2015	$221,297 (without rFVIIIFc) to $221,658 (with rFVIIIFc)
			Prophylaxis	2015	$598,998 (with rFVIIIFc) to $606,913 (Without rFVIIIFc)
Direct medical at Five years	Patients with inhibitors	EU5	BPA	2017	€2,765,808 (age 12 to 17) to €2,804,532 (age ≥18)
Direct medical at Ten years	Patients without inhibitors	US	Gene therapy	NR	$1,022,249
			Prophylaxis	NR	$1,693,630

Key: BPA – bypassing agent; rFVIIIFc – recombinant factor VIII Fc fusion protein.

This SR identified costs relating to bleeding **(Table 2)**, dental and major surgeries **(Table 3)**, and indirect annual visit costs of €145 to €575 (prophylaxis) and €995 (on demand). Costs were reported for two-, five- and ten-year costs **(Table 4)**, annual costs **(Table 5)**, and lifetime costs **(Table 6)**. This data is largely for haemophilia A, with data for haemophilia B being highlighted by asterisks. This SR shows the cost burden on payers for treating patients with haemophilia: more than $750,000 a year in the US and more than €800,000 in the EU5.

Table 5: Annual direct, direct non-medical and indirect costs by inhibitor status (US and EU5)

Type of costs	Inhibitor status	Country	Description	Cost year	Total per patient costs
Direct medical	Patients without inhibitors	US	FIX	2019	$614,886*
			FVII	2017	Last 12 months of continuous enrollment: $351,065 (≥4 FVII claims) to $650,065 (≥6 FVII claims)
			FVIII	2017	Last 12 months of continuous enrollment: $460,576 (≥6 FVIII claims) to $759,661 (≥273 days of continuous supply of FVIII)
		EU5	Overall costs (excluding FRT)	NR	€2,028*
			Overall costs (excluding costs of therapeutics)	NR (2018 study)	Moderate disease: €1,214 (0 problem joints) to €6,203 (2+ problem joints) Severe disease: €3,749 (0 problem joints) to €9,467 (2+ problem joints)
Direct non-medical	Patients without inhibitors	US	NR	NR	Haemophilia A $3,856, haemophilia B: $2,862**
			Therapies, medications, devices, home alterations, disability entitlement, transit, professional/informal caregivers,	2019	Total (SD) annual costs: $2,371 ($6,184)*
		EU5	Therapies, transport, requirement to aids/equipment, transfer payments	2019	Mean (SD) annual costs: €1,997 (€3,187)*
Indirect	Patients without inhibitors	US	Total indirect costs	NR	Haemophilia A $8,789, haemophilia B $7,692**
			Absenteeism, early retirement/unemployment	2019	$6,931*
		EU5	Work productivity, caregiver burden	2019	€8,973*
Direct medical	Patients with inhibitors	EU5	Prophylaxis	2019	Emicizumab: €540,677
			rFVIIa	2019	On demand €786,106 Prophylaxis: €818,599
Direct non-medical	Patients with inhibitors	EU5	Informal care/daily care	2019	Prophylaxis: €2,240 On-demand: €2,240
Direct medical	Population with mixed inhibitors status /NR	US	Clotting factor and BPA	2011	$218,369
			FIX and BPAs	2019	Mean annual healthcare costs: $80,811 (mild haemophilia) to $632,088 (severe haemophilia)*
			Hospital visits, clotting factor costs, bleed costs	NR	$96,918 (age 45 to 64; non-prophylaxis) to $273,936 (age 45 to 64; prophylaxis)
		EU5	FVIII prophylaxis and for treating bleeds	NR	€260,662 (pharmokinetic driven prophylaxis) to €265,859 (standard prophylaxis)
			Factor consumption costs	NR	Annual factor consumption costs: £142,369 (haemophilia A) to £198,803 (harmophilia B)
			Prophylaxis, bleedings	NR	Mean annual cost: €224,407 (weekly rFIXFc) to €368,587 (weekly rIX-FP)*

Key: BPA – bypassing agent; ED – emergency department; FIX – factor IX; FRT – factor replacement therapy; FVII – factor VII; FVIII – factor VIII; NR – not reported; rFIX-Fc – recombinant coagulation factor IX produced with Fc technology; rFVIIa – recombinant activated factor VII; rIX-FP – recombinant fusion protein containing rFIX fused with recombinant albumin; SD – standard deviation.
*Patients with haemophilia B.
**Patients with haemophilia A or B.

Table 6: Lifetime direct, non-medical and indirect costs by inhibitor status (US and EU5)

Type of costs	Inhibitor status	Country	Description	Cost year	Total per patient costs
Direct medical	Patients without inhibitors	US	FVIII	2019	$18,722,000
			FIX	2019	$20,934,426 (on demand)*
			Prophylaxis	2018	$13,314,045 (rAHF) to $23,406,84 (SoC)
			Valoctocogene roxaparvovec	2019	$13,693,000 to $16,656,470
		EU5	FVIII	NR	70-year costs: £3,458,572 (low dose scenario) to £13,69,229 (high dose scenario)
			rFVIII-FS	2014	€1,452,686 (on demand) to €1,682,380 (late prophylaxis)
	Patients with inhibitors	US	Prophylaxis	2017	$15,144,711 (emicizumab) to $90,182,398 (BPA)
Medical non-direct and indirect	Patients without inhibitors	US	Travel expenses, productivity losses and social benefits	2019	$54,276 (prophylaxis) and $37,400 (on demand)*
Indirect	Patients without inhibitors	EU5	Productivity loss	2014	€19,487 (late prophylaxis) and € 59,050 (on demand)
	Patients with inhibitors	US	Productivity loss	2017	$400,983 (BPA prophylaxis) and $766,602 (no prophylaxis)

Key: BPA – bypassing agent; FIX – factor IX; FVIII – factor VIII; rAHF – recombinant advate antihemophilic factor; rFVIII-FS – Sucrose-formulated recombinant factor VIII.; SoC – standard of care.
*Patients with haemophilia B.

DISCUSSION

Most of the costs identified are related to FRTs and non-FRTS, administered as prophylaxis or on demand. Costs increase with disease severity.
Published costs evidence primarily considered haemophilia A without inhibitors **(Table 1)**. Data were not as widely available for haemophilia B and patients with inhibitors. Patients with haemophilia B are a small proportion of the population and there is a general gap in the evidence for these patients. Costs do not appear to vary depending on haemophilia type, but a literature search suggested that costs correlate to disease severity [8].
Whilst comparative data have not been identified in the eligible studies, the literature suggests costs may be higher for patients with inhibitors than without.
Limitations:

- This SR only included studies in which data were reported separately for haemophilia A and B. However, based on the findings of this review, this distinction may not be critical and studies of mixed populations may be useful.
- There was no assessment of the risk of bias of the included studies.

RECOMMENDATIONS

- Treatment associated costs: Country-specific and up-to-date pricing and weight data should be used, and treatments should be established by haemophilia type, severity and inhibitor status so that appropriate comparators can be chosen.
- Bleed costs: Resource use should be costed using country-specific data sources. Clinical opinion can be used to justify applying the same costs regardless of haemophilia type or inhibitor status.
- Other medical costs: Current procedure costs should be sourced from reference costs and tariff sources, unit costs for outpatient appointments and scans should be drawn from country-specific data sources.

REFERENCES:
1. Centers for Disease Control and Prevention. Treatment of Hemophilia. 2023 August 2023]; Available from: https://www.cdc.gov/ncbddd/hemophilia/treatment.html.
2. Srivastava, A., et al., WFH Guidelines for the Management of Hemophilia, 3rd edition. Haemophilia, 2020. 26 Suppl 6: p. 1-158.
3. National Bleeding Disorders Foundation. MASAC Document 267 - MASAC Recommendation Concerning Prophylaxis for Hemophilia A and B with and without Inhibitors. 2023 August 2023]; Available from: https://www.hemophilia.org/healthcare-professionals/guidelines-on-care/masac-documents/masac-document-267-masac-recommendation-concerning-prophylaxis-for-hemophilia-a-and-b-with-and-without-inhibitors.
4. Gale, A.J., Continuing education course #2: current understanding of hemostasis. Toxicol Pathol, 2011. 39(1): p. 273-80.
5. National Bleeding Disorders Foundation. Hemophilia Current Treatments. 2023 August 2023]; Available from: https://www.hemophilia.org/bleeding-disorders-a-z/treatment/current-treatments.
6. Ay, C., et al., Treatment patterns and bleeding outcomes in persons with severe hemophilia A and B in a real-world setting. Ann Hematol, 2020. 99(12): p. 2763-2771.
7.Moher D, Liberati A, Tetzlaff J, Altman DG, Group* P. Preferred reporting items for systematic reviews and meta-analyses: the PRISMA statement. Annals of internal medicine. 2009.151(4):264-69.
8. Coppola A, D'Ausilio A, Aiello A, Amoresano S, Toumi M, Mathew P, et al. Cost-effectiveness analysis of late prophylaxis vs. on-demand treatment for severe haemophilia A in Italy. Haemophilia. 2017.23(3):422-29.

FUNDING DISCLOSURES:
This study was funded by Sanofi.

CONFLICTS OF INTEREST:
S. Bozzi and J.G. Jasso Mosqueda are employees of Sanofi
C. Bartlett , P. Miller, J. Bracewell and R. McCool received consultancy, research grant and expenses from Sanofi