HSD70

ECONOMIC BENEFIT OF EMPOWERING COMMUNITY PHARMACISTS IN THE DIAGNOSIS AND TREATMENT OF ACUTE SORE THROAT IN ENGLAND

Patricia Aluko[1], Kay Edokpayi[1], Flora Cheng[1], Darush Attar-Zadeh[2], Liam Kennedy[1],Yetunde Kolade[1]

1. Reckitt, Slough, United Kingdom, 2. Barnet Clinical Commissioning Group, London, North West, UK.

INTRODUCTION

- About 80% of antibiotics are prescribed in England community settings with only minority due to bacterial infection(often Strep A)[1]
- Rapid diagnostic test can enhance antimicrobial stewardship and guide treatment decisions[1]
- Recent Cochrane review concluded that rapid diagnostic test for sore throat could cut antibiotic prescription by 25%[2]
- Use of Strep A rapid diagnostic test is a routine clinical practice in countries such as Sweden and Denmark[3,4]
- However, NICE guidance concluded that the rapid test for Strep A is not cost-effectiveness in GP settings, while acknowledging further research in Pharmacy settings[5]
- In contrast, a clinical and economic analysis carried out by Health Technology Wales found that rapid test for sore throat carried out in pharmacy setting has 100% probability of being cost-effectiveness at a threshold of £20,000/QALY[6,7]

*QALY: Quality Adjusted Life Year

METHODOLOGY

- Participants aged ≥16 years with sore throat symptoms and FeverPAIN scores 4-5 were tested by pharmacists using OSOM® Strep A rapid diagnostic kit.
- Participants were requested to complete a questionnaire on potential treatment choice (A&E, GP visit, pharmacist, self-care). This was used to determine avoided GP visits if diagnostic service was not available
- Potential cost saving was calculated by estimating the monetary value of avoided GP visits for sore throat and extrapolating findings to UK population

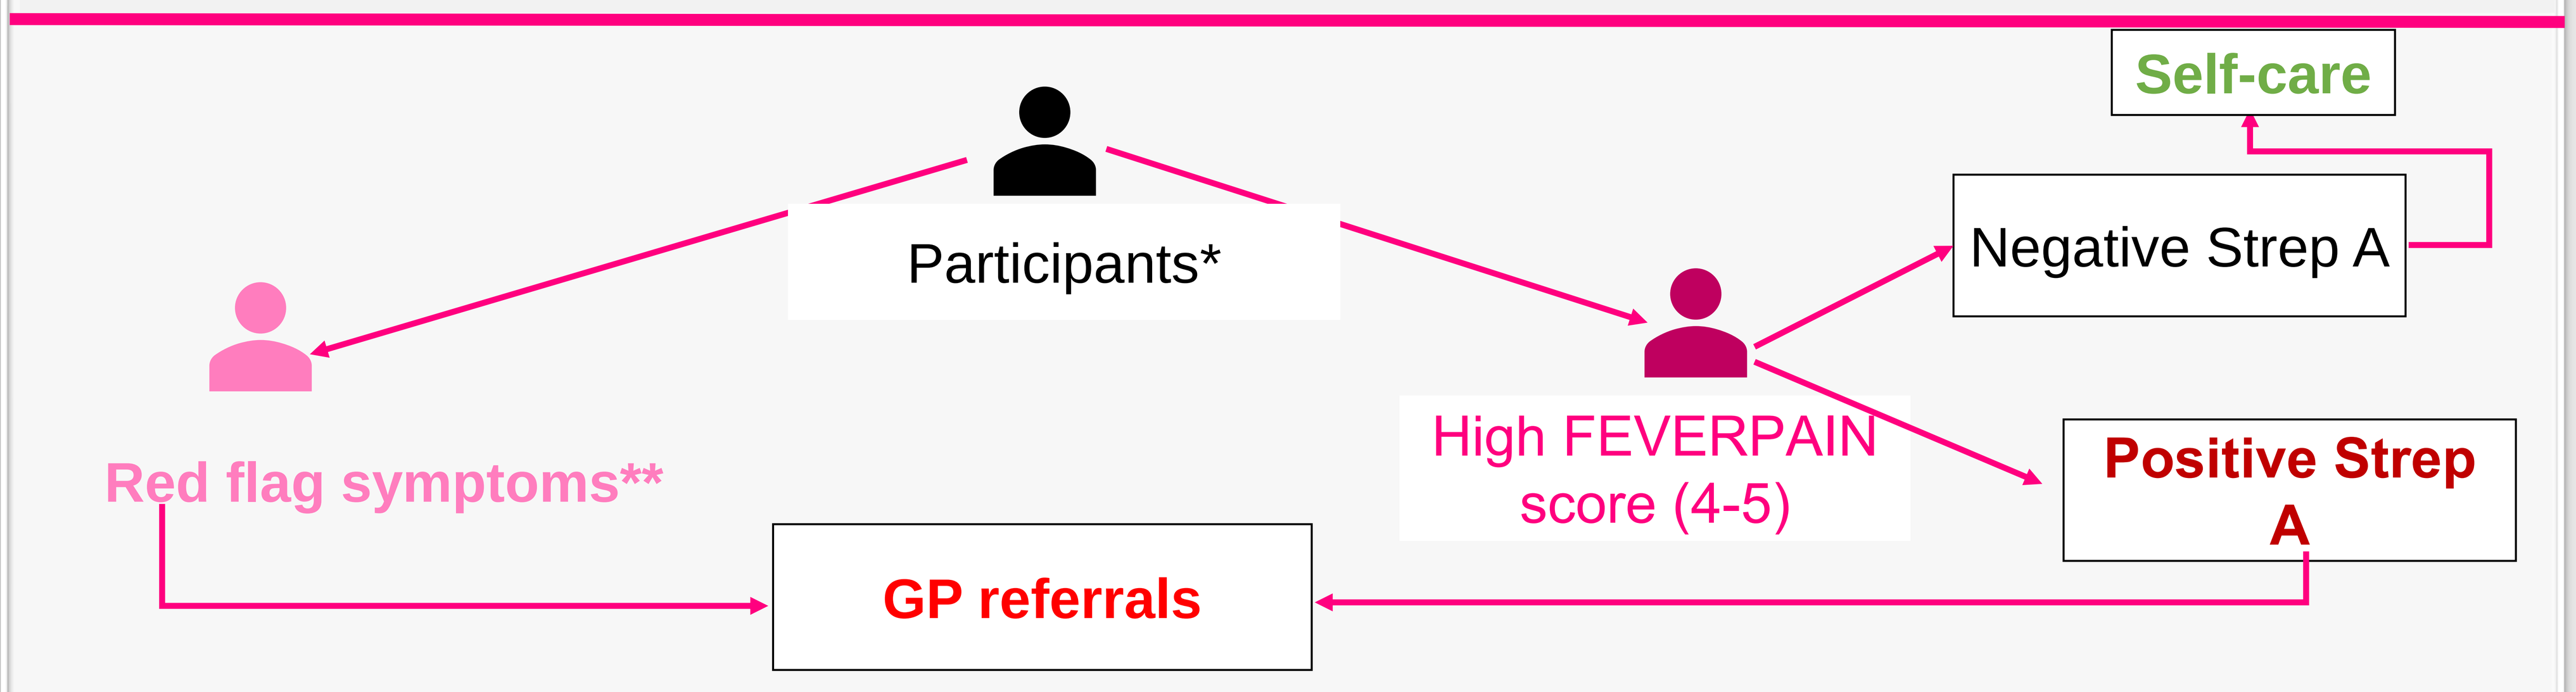
Self-care
Participants*
Negative Strep A
Red flag symptoms**
High FEVERPAIN score (4-5)
Positive Strep A
GP referrals

RESOURCES AND COSTS ESTIMATE

Cost difference = GP consultation cost – (Cost of Advanced pharmacist practitioner + OSOM® Strep A test kit)

Number of GP appointments attended per year between Feb 2018 to Jan 2019 for sore throat symptoms (8-13% of GP consultation)[8,9]

*Patients without signs of viral infection were excluded
**Red flag symptoms refers to trismus, difficulty breathing, neck swelling, torticollis

RESULTS

- Based on FEVERPAIN score, 36 of the 59 participants were eligible for Strep A rapid swab test and only 12 participants tested positive
- The total cost of the swab test service was **£.9.49** which included cost of Strep A swab test kit (£2.16) and 10 mins advanced pharmacist practitioner consultation time (estimated as £7.3) while the cost of GP service was **£31** (with average consultation time of 9.22 mins)

Selected treatment choice pre-study
34 participants selected GP visit, while 19 opted for the pharmacy and 9 participants for OTC self-care.

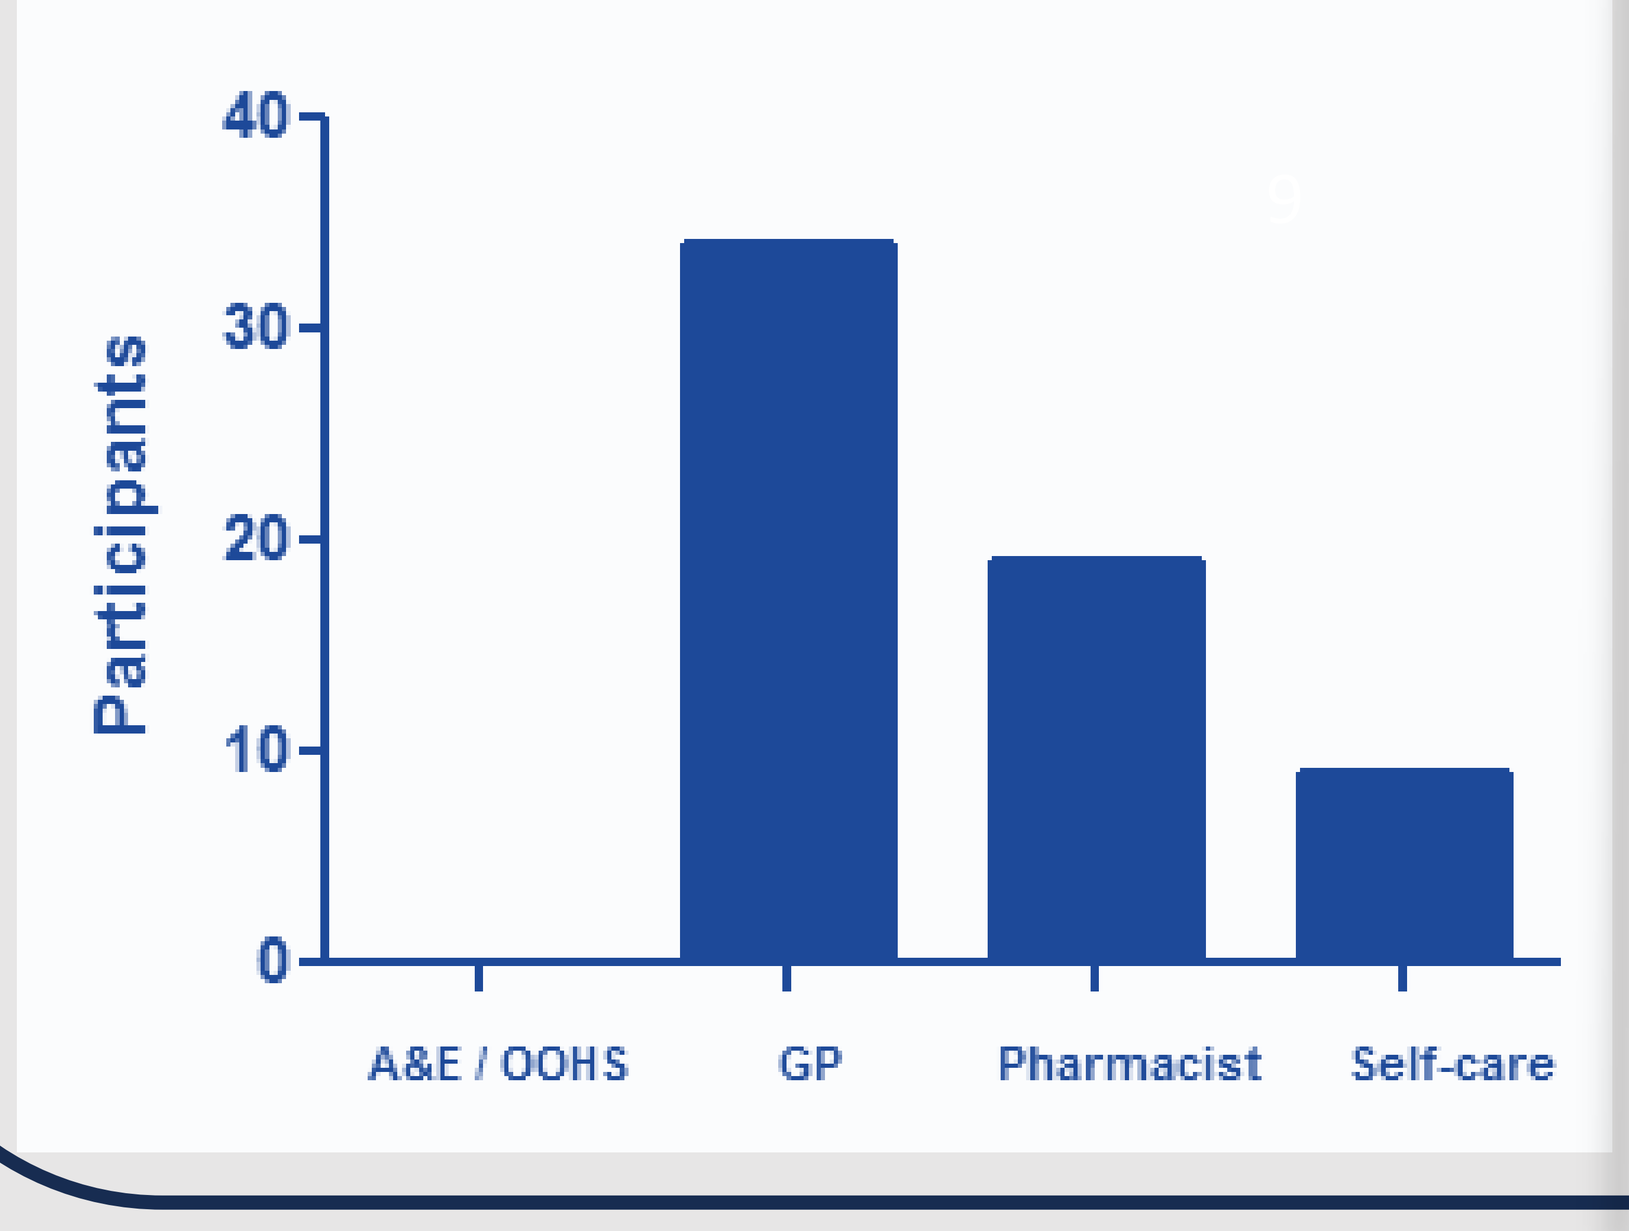
40
30
20
10
0
Participants
A&E / OOHS
GP
Pharmacist
Self-care

Selected treatment choice post-study
After the swab test study, 45 participants opted for self-care

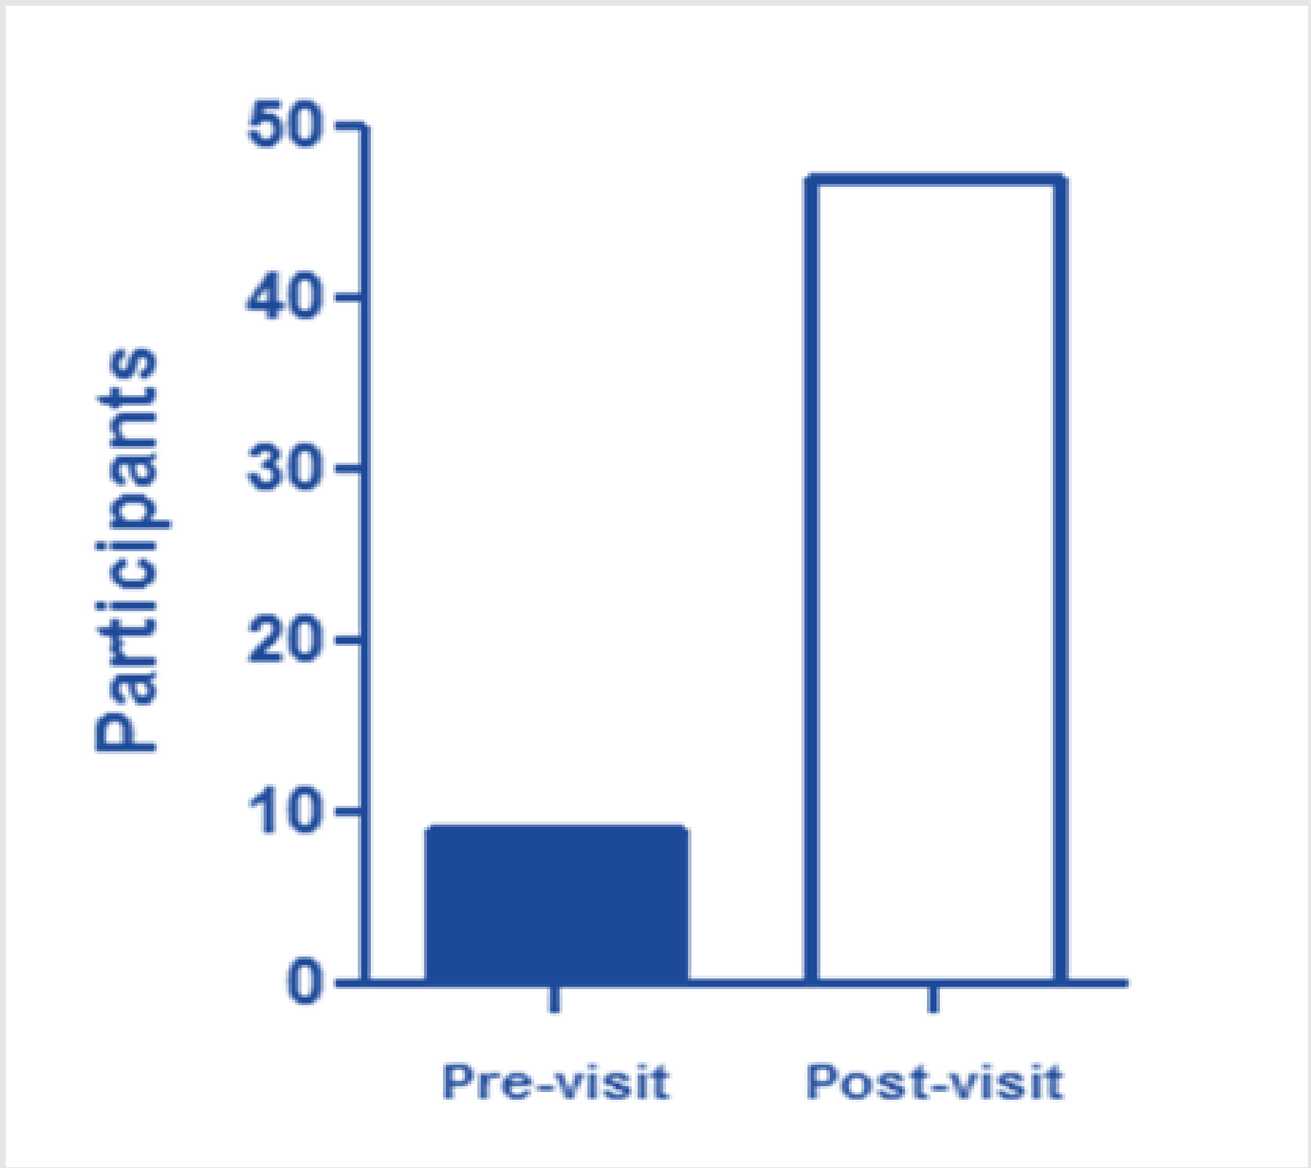
50
40
30
20
10
0
Participants
Pre-visit
Post-visit

GP appointments saved
Only 12 of the 34 participants who opted for GP visit required referral with 22 potential appointments saved

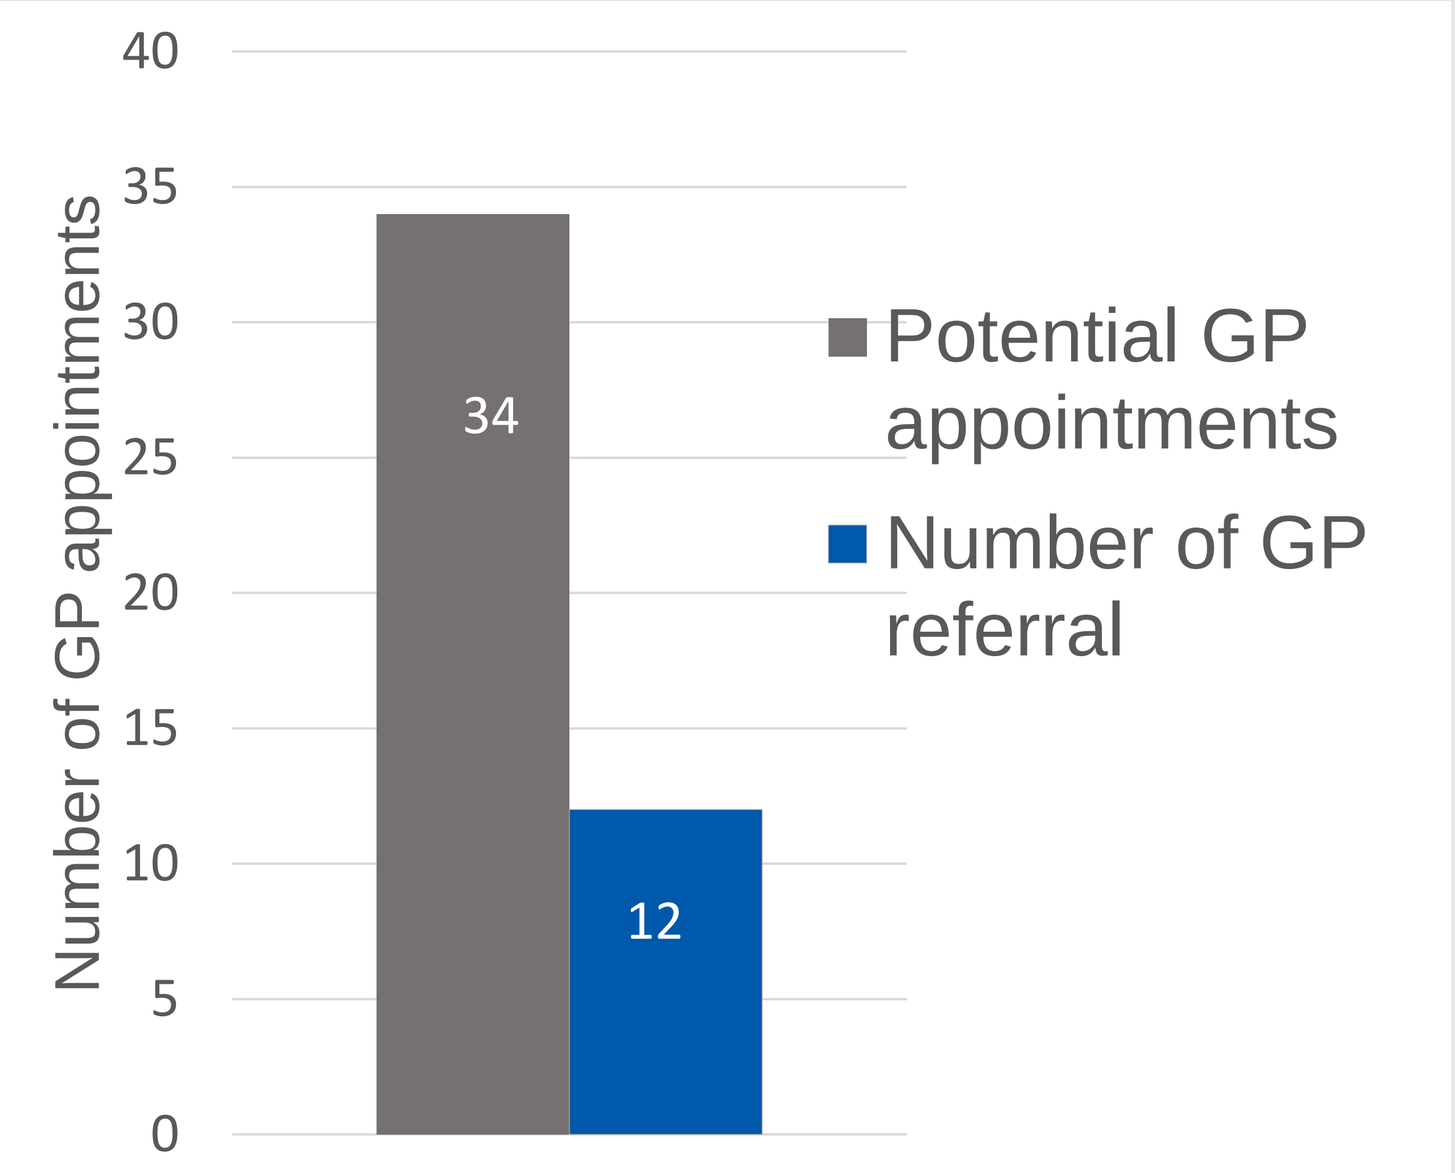
40
35
30
25
20
15
10
5
0
Number of GP appointments
34
12
Potential GP appointments
Number of GP referral

Cost saving per sore throat patient
About £21 could be saved per patient when the swab test is carried out in a pharmacy setting

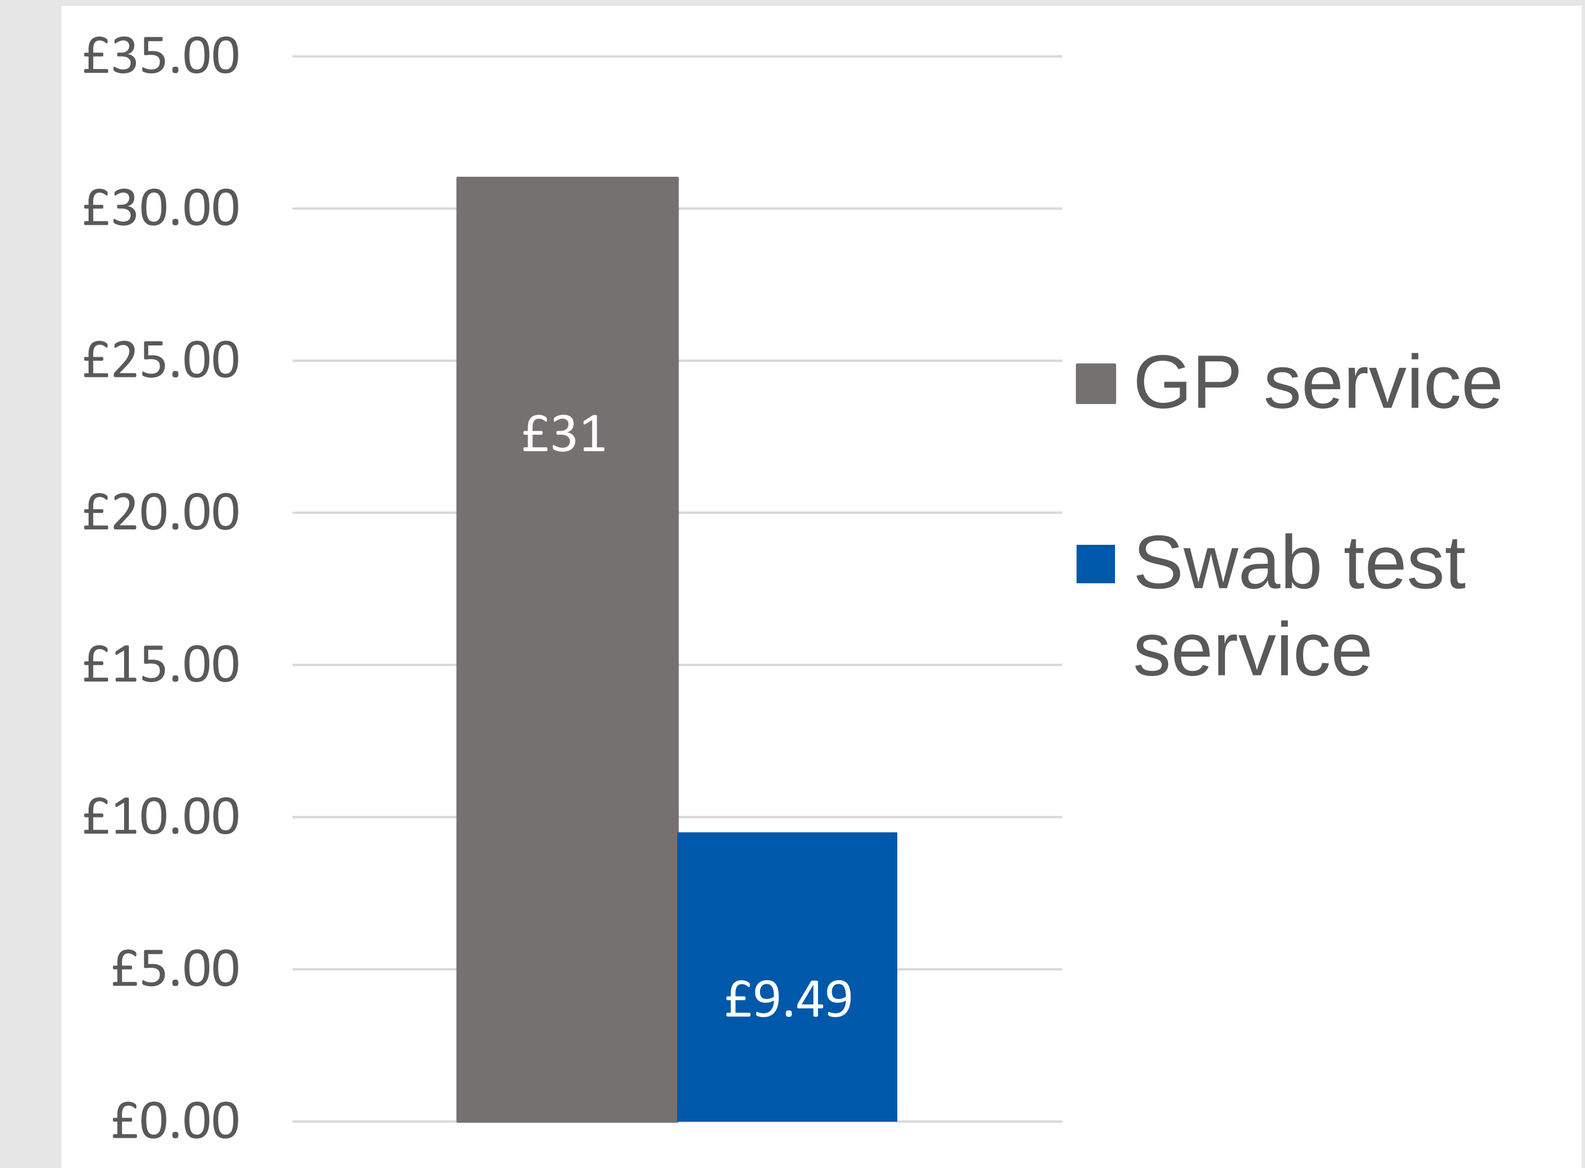
£35.00
£30.00
£25.00
£20.00
£15.00
£10.00
£5.00
£0.00
£31
£9.49
GP service
Swab test service

About **14 - 23 million NHS GP appointments** could be saved per year when sore throat patients are treated in community pharmacy setting

Number of GP appointments attended per year	276,516,000
Number of GP appointments for sore throat per year	22,121,280 – 35,947,080
Estimated total number of GP visits that can be prevented per year (Data from potential appointments saved (65%) was applied to estimate GP visits that can be prevented per year	14,378,832 – 23,365,603
Cost of replacing a GP visit with Pharmacist consultation per patient for Strep A swab test	£21.51 (2018/2019 GBP)
Total cost savings from avoided GP visits per year for upper and/or respiratory symptoms	Approx £334 – £543 million (2022/2023 GBP)

CONCLUSION

- Approximately £334 million to £543 million per year could be saved by NHS England when rapid test for Strep A is implemented in pharmacy settings
- Similar finding was observed in a study carried out in Canada with a cost-saving of $1.3 million to $2.6 million per year across five provinces*
- Rapid diagnostic testing for Strep A in community pharmacies in England could alleviate NHS pressure, save costs and empower pharmacists to treat while preventing antimicrobial misuse and promoting self-care treatment.
- Implementation of rapid test for Strep A would also serve as additional safeguard against antimicrobial misuse, considering the upcoming NHS policy that enables pharmacists to prescribe for minor ailments, including sore throats.

References:

1. Firth, I et al. (2023). A Novel Incentive Model for Uptake of Diagnostics to Combat Antimicrobial Resistance. OHE Contract Research Report
2. Cohen JF, et al. (2020). Efficacy and safety of rapid tests to guide antibiotic prescriptions for sore throat. Cochrane Database Syst Rev.
3. Stuhr JK, et al.(2019); Danish GPs' and practice nurses' management of acute sore throat and adherence to guidelines. Fam Pract.
4. Gunnarsson R, et al. (2020); Association between guidelines and medical practitioners' perception of best management for patients attending with an apparently uncomplicated acute sore throat: a cross-sectional survey in five countries. BMJ Open.
5. NICE guidance (2019) .Rapid tests for group A streptococcal infections in people with a sore throat. https://www.nice.org.uk/guidance/dg38.
6. Mantzourani E et al (2020). Impact of a pilot NHS-funded sore throat test and treat service in community pharmacies on provision and quality of patient care. BMJ Open Quality
7. Health Technology Wales (2020) . Evidence Appraisal Report. Rapid antigen detection tests for group A streptococcal infections to treat people with a sore throat in the community pharmacy setting.
8. Mehta N, et al (2017). Antibiotic prescribing in patients with self-reported sore throat. J Antimicrob Chemother.
9. Gulliford, M.C., et al. (2016) Safety of reduced antibiotic prescribing for self limiting respiratory tract infections in primary care: cohort study using electronic health records. BMJ

* Lathia N. et al.(2018). Cost-minimization analysis of community pharmacy-based point-of-care testing for strep throat in 5 Canadian provinces. Can. Pharm. J.

Conflict of Interest:This study was funded by Reckitt. Darush has received honoria from various companies for respiratory related work.

reckitt