RWD130

Healthcare resource utilisation for paediatric and adolescent Duchenne muscular dystrophy patients: analysis of global, real-world data

Jonathan Strober[1], Keiko Ishigaki[2], Valeria Merla[3], Nate Posner[3], Joseph C. Cappelleri[3], Sherry Xiao[3], Anna Talaga[3], Susanne Brunner[3], Halima Iqbal[4], Emma Chatterton[4], Niall Hatchell[4]

[1]UCSF Benioff Children's Hospital, San Francisco, CA; US; [2]Tokyo Women's Medical University, Tokyo, Japan; [3]Pfizer Inc., New York, NY; US; [4]Adelphi Real World, Bollington, UK

Objective

To describe healthcare resource utilisation (HCRU) for male Duchenne muscular dystrophy (DMD) patients aged 18 and under.

Conclusions

A high level of HCRU was reported for adolescents with DMD in both inpatient and outpatient care.

As our data suggests that adolescent patients represent high economic burden on the healthcare system, there is a need for treatment earlier in disease progression to help support cost offsets and benefit patients.

Introduction

- Duchenne muscular dystrophy (DMD) is a childhood-onset genetic disorder, characterised by muscle degeneration, leading to limb weakness, loss of ambulation and progressive cardiac and pulmonary dysfunction[1].
- Previous research has described increasing economic cost of DMD with disease progression[2], with informal care often the main driver of economic burden[3].
- Few studies have utilised real-world data[4] or assessed resource usage with age-based stratification on a global scale.

Methods

- Data were drawn from the Adelphi Real Word DMD Disease Specific Programme (DSP™), a cross-sectional survey, with retrospective data collection of neurologists conducted in France, Germany, Italy, Japan, Spain, the United Kingdom, and the United States from October 2022.
- Interim data was taken in May 2023; data collection is still ongoing.
- The DSP methodology has been previously published and validated[5-7].
- Neurologists reported patient demographics, DMD-related hospitalisations, and use of mobility aids or home adaptations.

Methods (cont.)

- Patients were characterised as child (≤12) or adolescent (≥13).
- Descriptive statistics were reported.

Limitations

- Patients included in the DSP sample may not be truly representative of the overall population of patients, as patients who consult more frequently and require more medical care are more likely to be included.
- Recall bias, a common limitation of surveys, might also have affected responses of both physicians and patients.

Results

Table 1. Patient demographics and clinical characteristics

Characteristics	All patients (n=425)	0–12 years (n=264)	13–18 years (n=161)
Age, mean (SD)	10.4 (4.98)	7.1 (3.12)	15.7 (1.71)
Years since diagnosis, mean (SD)	**n=349** 6.1 (4.72)	**n=218** 3.4 (2.80)	**n=131** 10.5 (3.88)
Hospitalisations	**n=425**	**n=264**	**n=161**
Number of hospitalisations in the last 12 months due to DMD, mean (SD)	0.2 (0.54)	0.2 (0.50)	0.2 (0.59)
Number of patients hospitalised due to DMD in the last 12 months, n (%)	63 (14.8)	34 (12.9)	29 (18.0)
In-patient information	**n=63**	**n=34**	**n=29**
Hospitalizations involving ICU stay in the last 12 months, n (%)	9 (14.3)	3 (8.8)	6 (20.7)
Number of patients who had surgery for DMD-related issue in the last 12 months, n (%)	11 (17.5)	2 (5.9)	9 (31.0)

SD, standard deviation; DMD, Duchenne muscular dystrophy; ICU, intensive care unit

- A cohort of 124 neurologists reported data on 425 male DMD patients, aged 0-18 years. The mean (standard deviation; SD) patient age was 10.4 (SD±4.98, **Table 1**).
- Most patients were classified as early ambulatory (41.8%, **Figure 1**). Overall, 55.7% of adolescent patients were classified as non-ambulatory compared to 13.2% of child patients (**Figure 1**).
- In the 12 months prior to survey, adolescents had a mean (SD) number of consultations 28.5 (47.7), 4.5 (2.8) different physician types involved in their care and 13.7 (13.0) tests/assessments to monitor their condition. For children this was 16.9 (31.0), 3.7 (2.4), and 9.7 (12.6), respectively (**Figure 2**).
- Adolescents (18.0%) had been hospitalised more often due to DMD compared to children (12.9%), with a greater proportion of those hospitalised requiring intensive care (8.8% vs. 20.7%) and having surgery (5.9% vs. 31.0%, **Table 1**).
- Overall, 68.4% of patients (n=425) required mobility aids/supports. Electric wheelchair and orthoses were the most common support required by adolescents and children, respectively (**Figure 3**).
- Adolescents more frequently required mobility aids/supports (61.2% vs. 84.5%; **Figure 3**) and adaptations to their home (52.2% vs. 83.2%, **Figure 4**). The most frequently reported home modification was bathroom adaptation (**Figure 5**).

Figure 1. Ambulation status

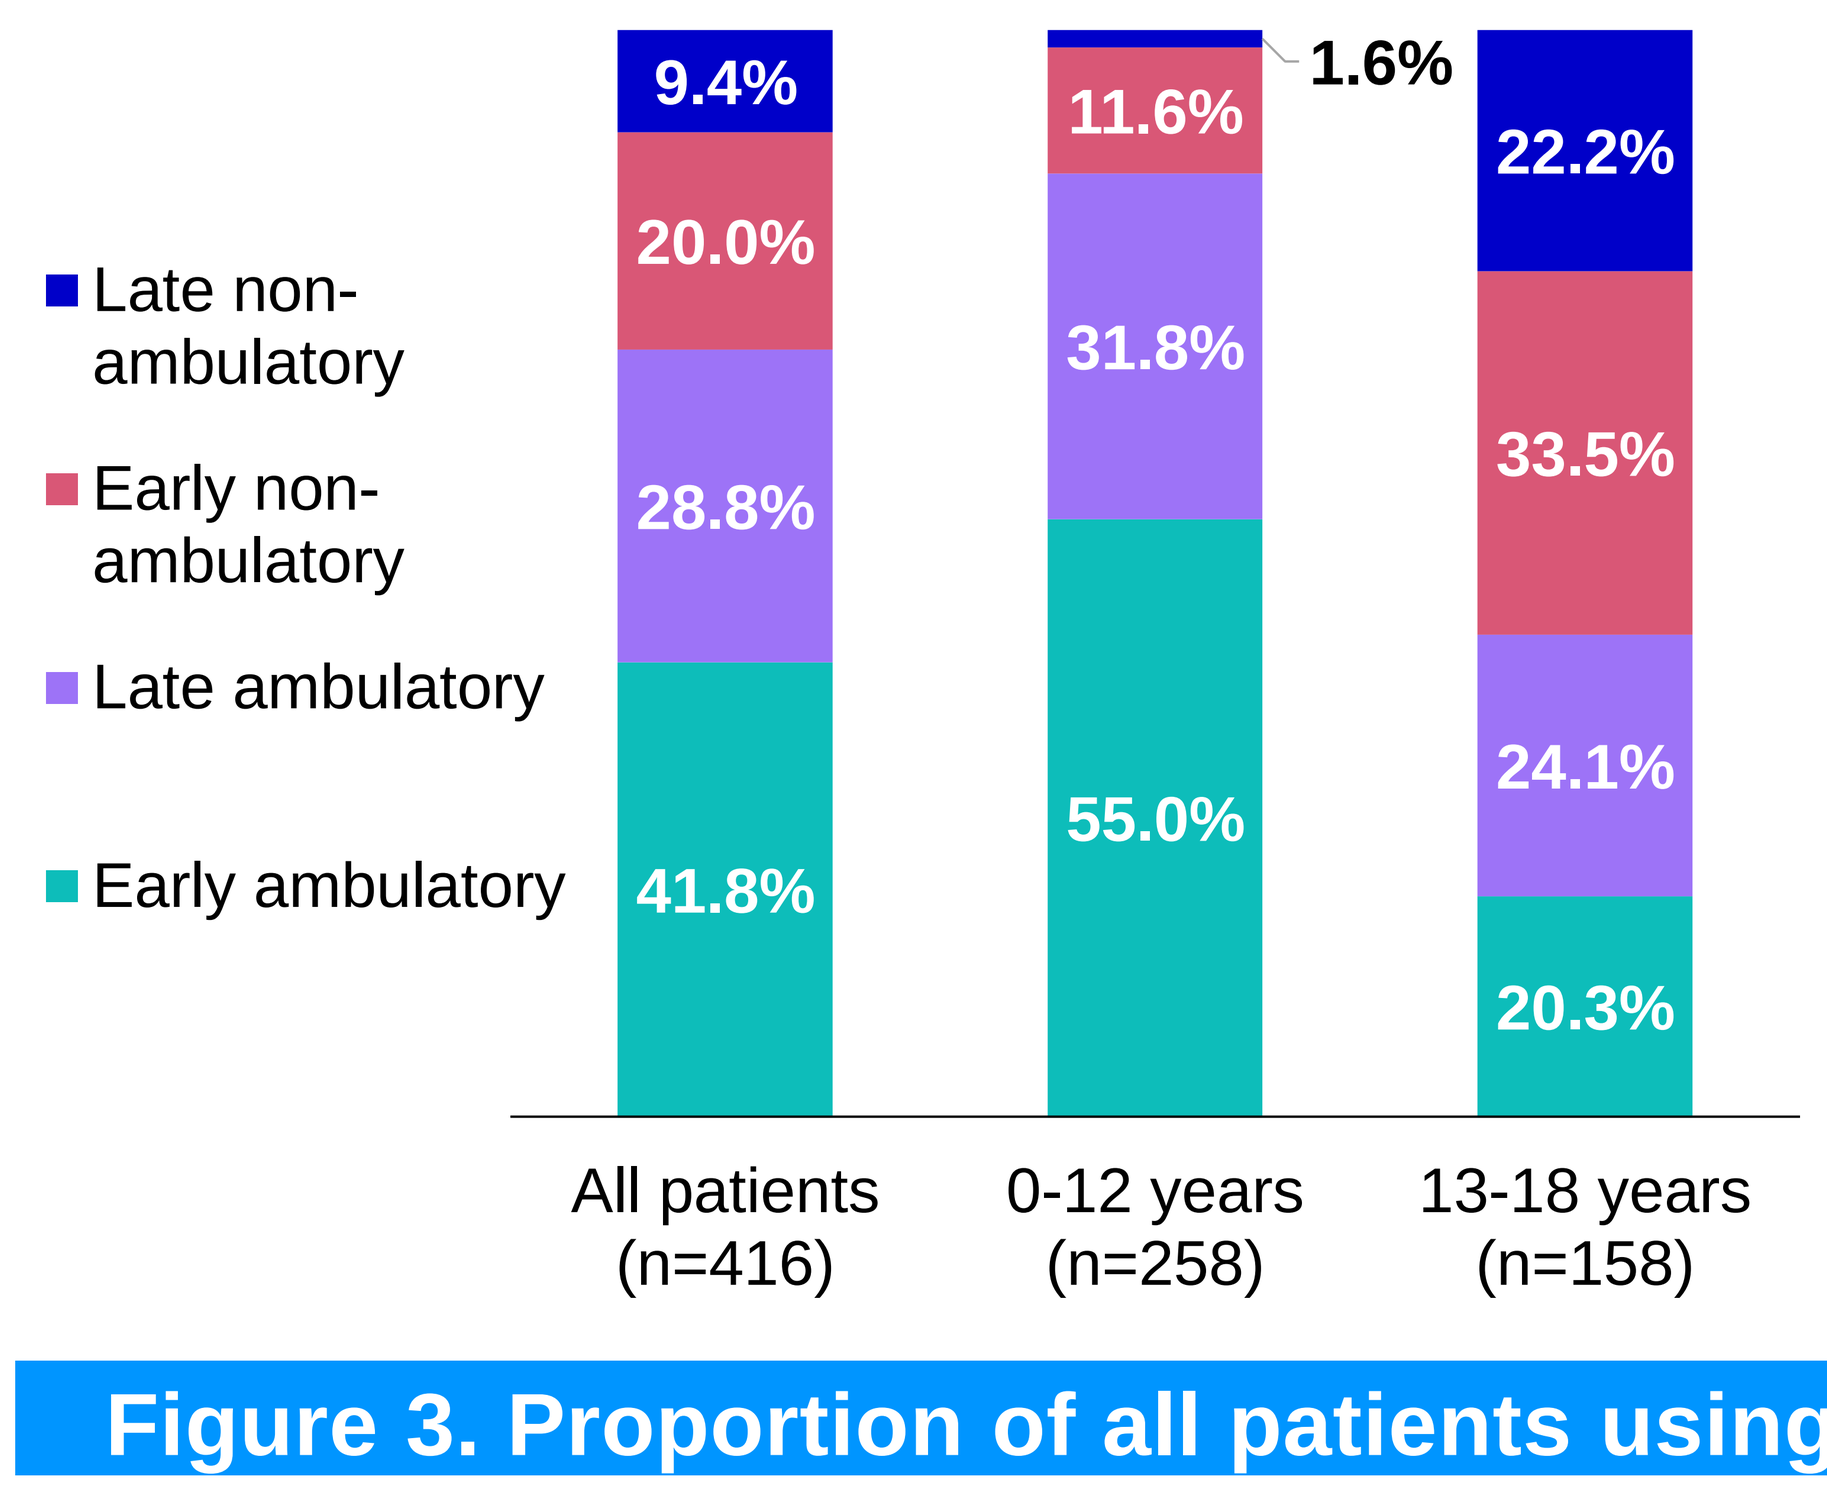
Late non-ambulatory
Early non-ambulatory
Late ambulatory
Early ambulatory
9.4%
20.0%
28.8%
41.8%
1.6%
11.6%
31.8%
55.0%
22.2%
33.5%
24.1%
20.3%
All patients (n=416)
0-12 years (n=258)
13-18 years (n=158)

Figure 2. Consultations and testing in the last 12 months

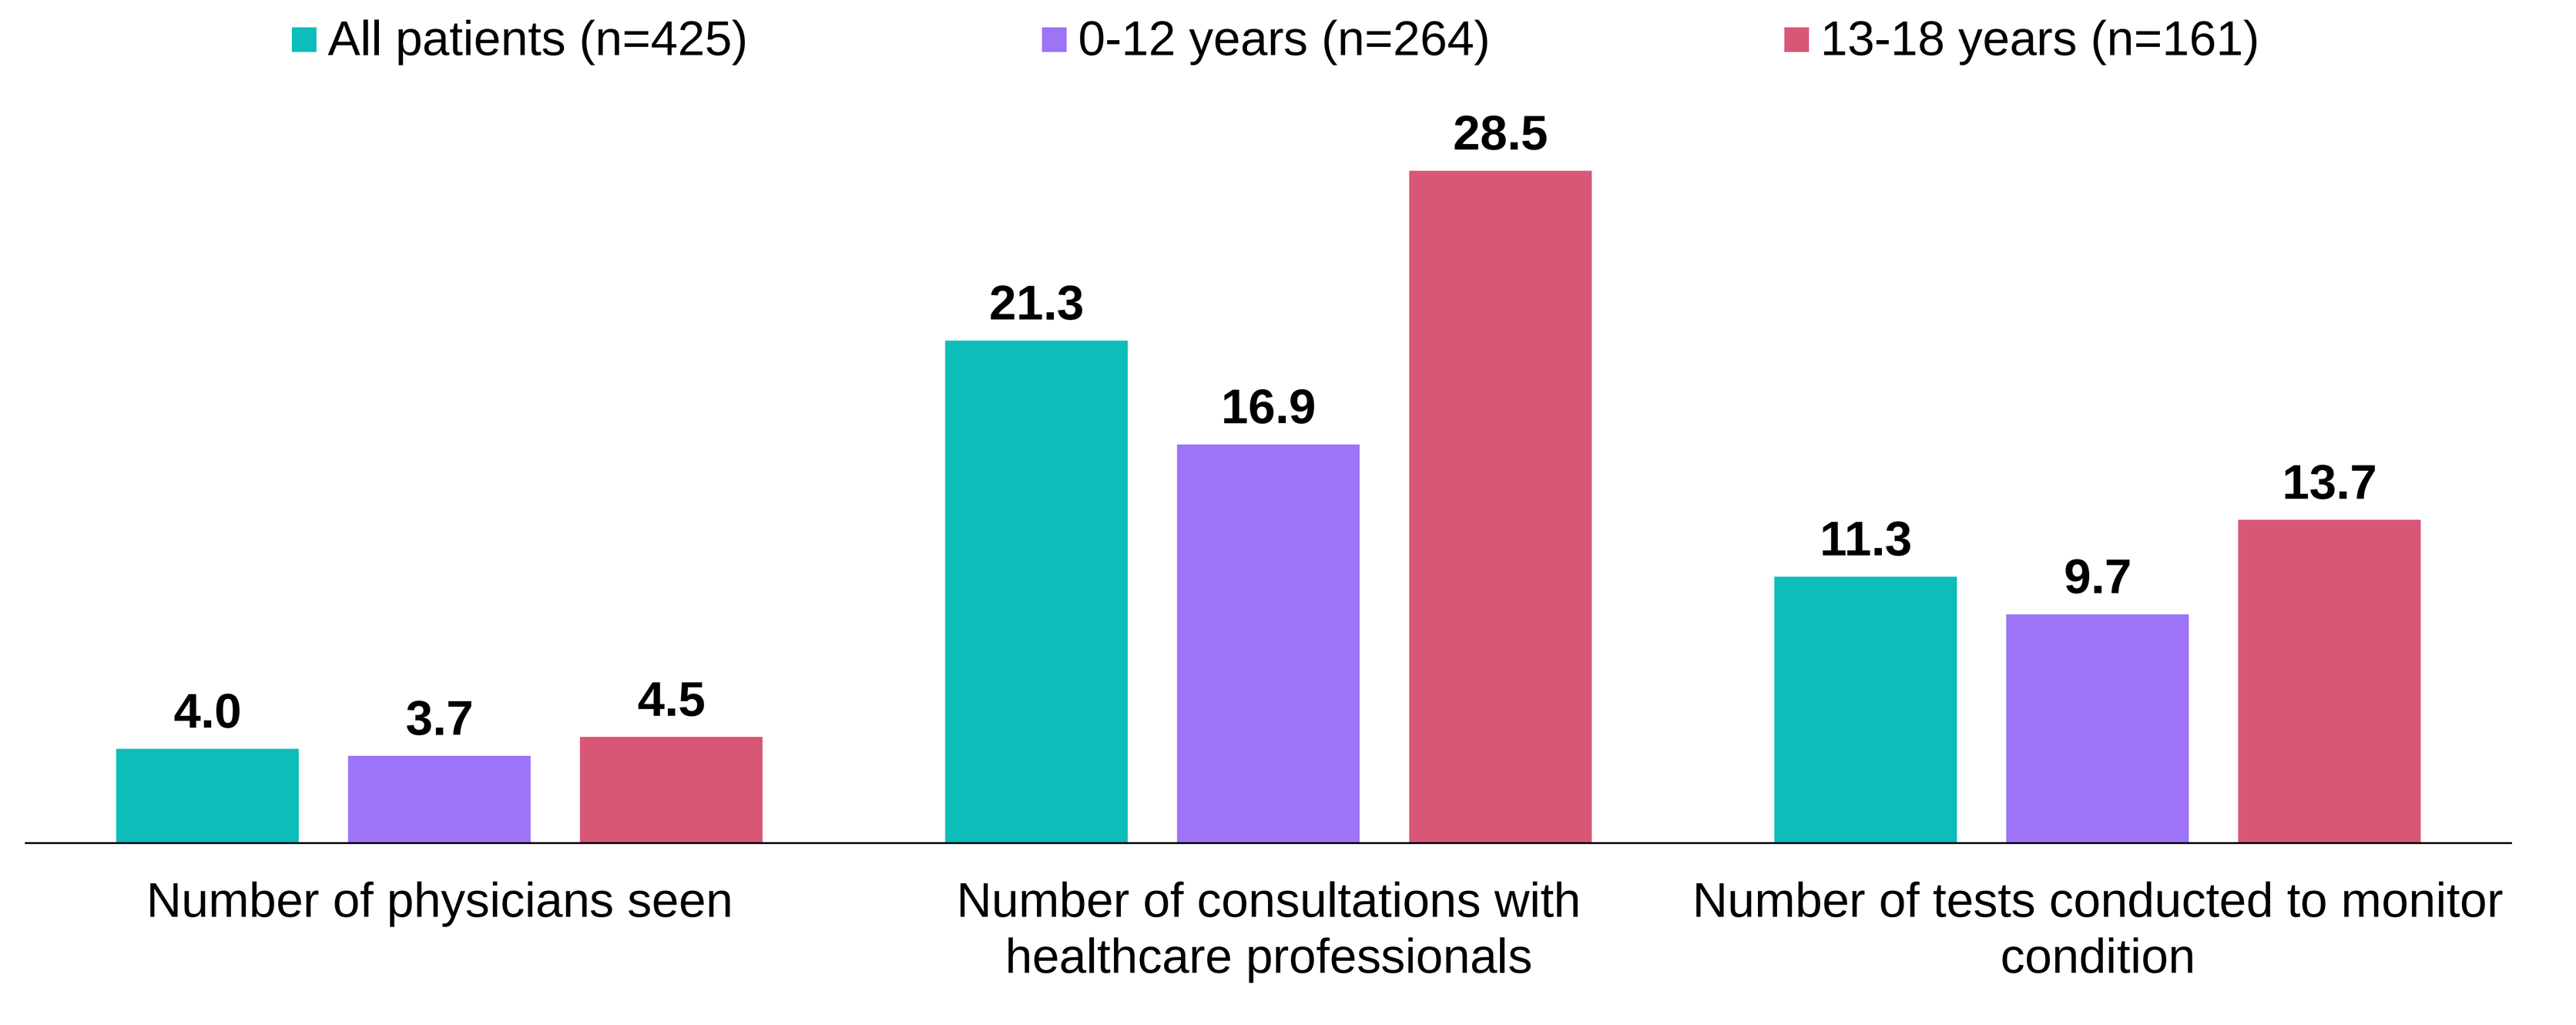
All patients (n=425)
0-12 years (n=264)
13-18 years (n=161)
4.0
3.7
4.5
21.3
16.9
28.5
11.3
9.7
13.7
Number of physicians seen
Number of consultations with healthcare professionals
Number of tests conducted to monitor condition

Figure 3. Proportion of all patients using mobility aids/supports (n=425)

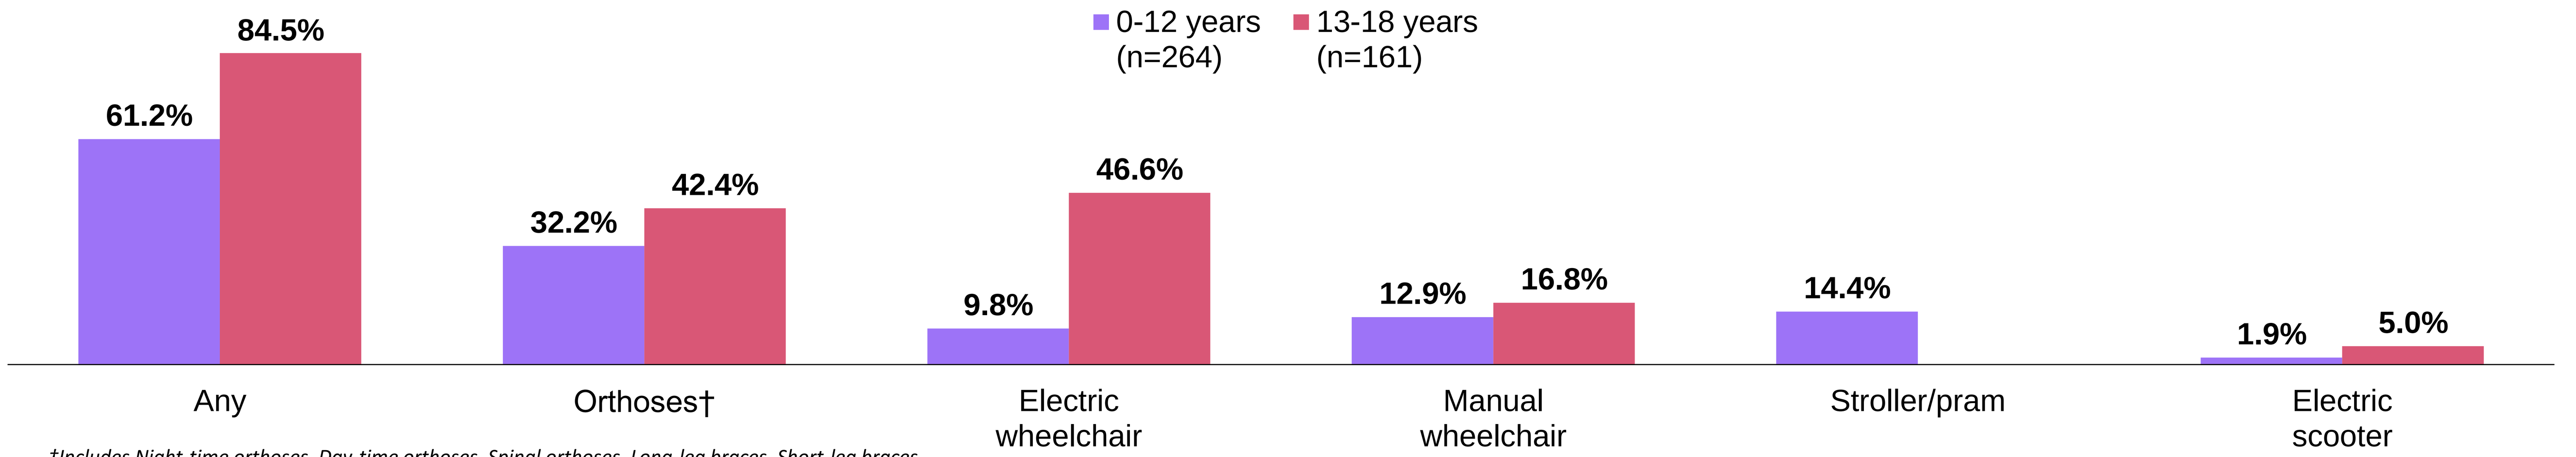
0-12 years (n=264)
13-18 years (n=161)
61.2%
84.5%
32.2%
42.4%
9.8%
46.6%
12.9%
16.8%
14.4%
1.9%
5.0%
Any
Orthoses†
Electric wheelchair
Manual wheelchair
Stroller/pram
Electric scooter

†Includes Night-time orthoses, Day-time orthoses, Spinal orthoses, Long-leg braces, Short-leg braces

Figure 4. Proportion of patients with home modifications

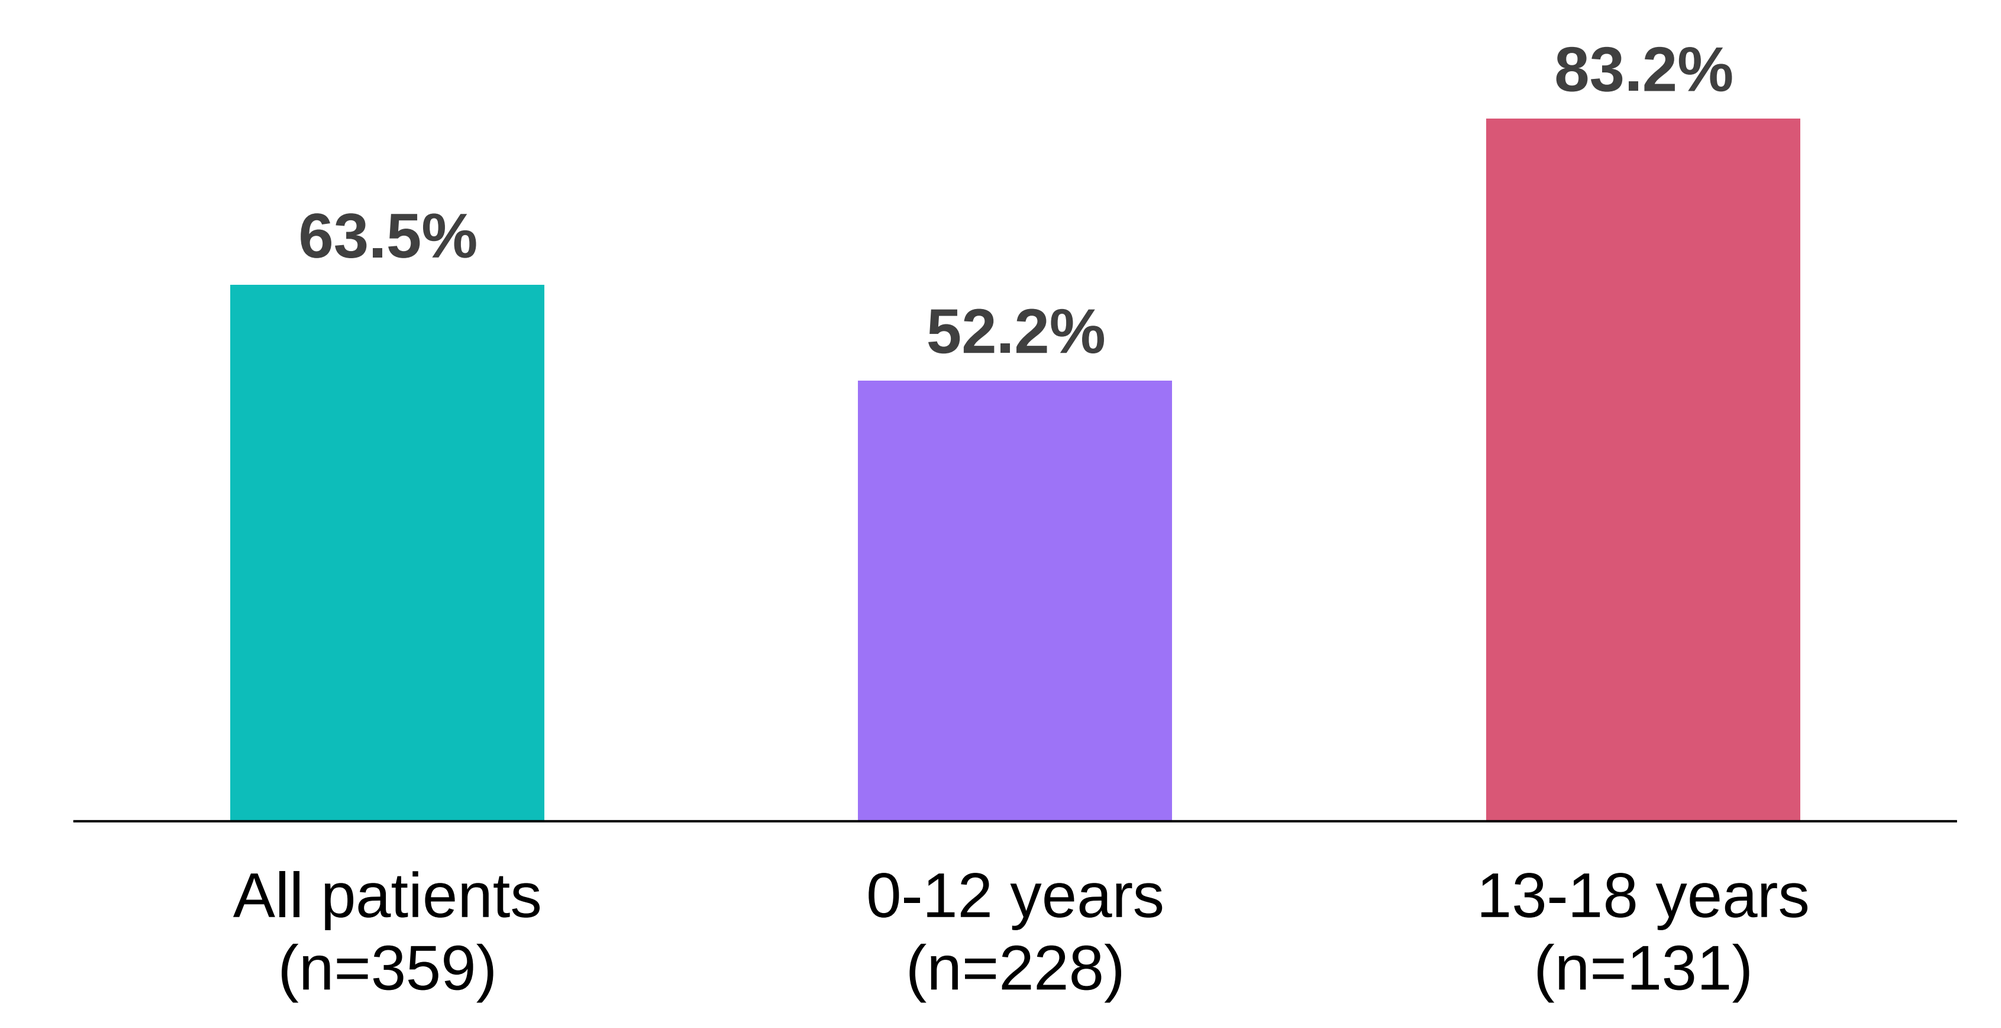
63.5%
52.2%
83.2%
All patients (n=359)
0-12 years (n=228)
13-18 years (n=131)

Figure 5. Physician-reported home modifications (n=228)

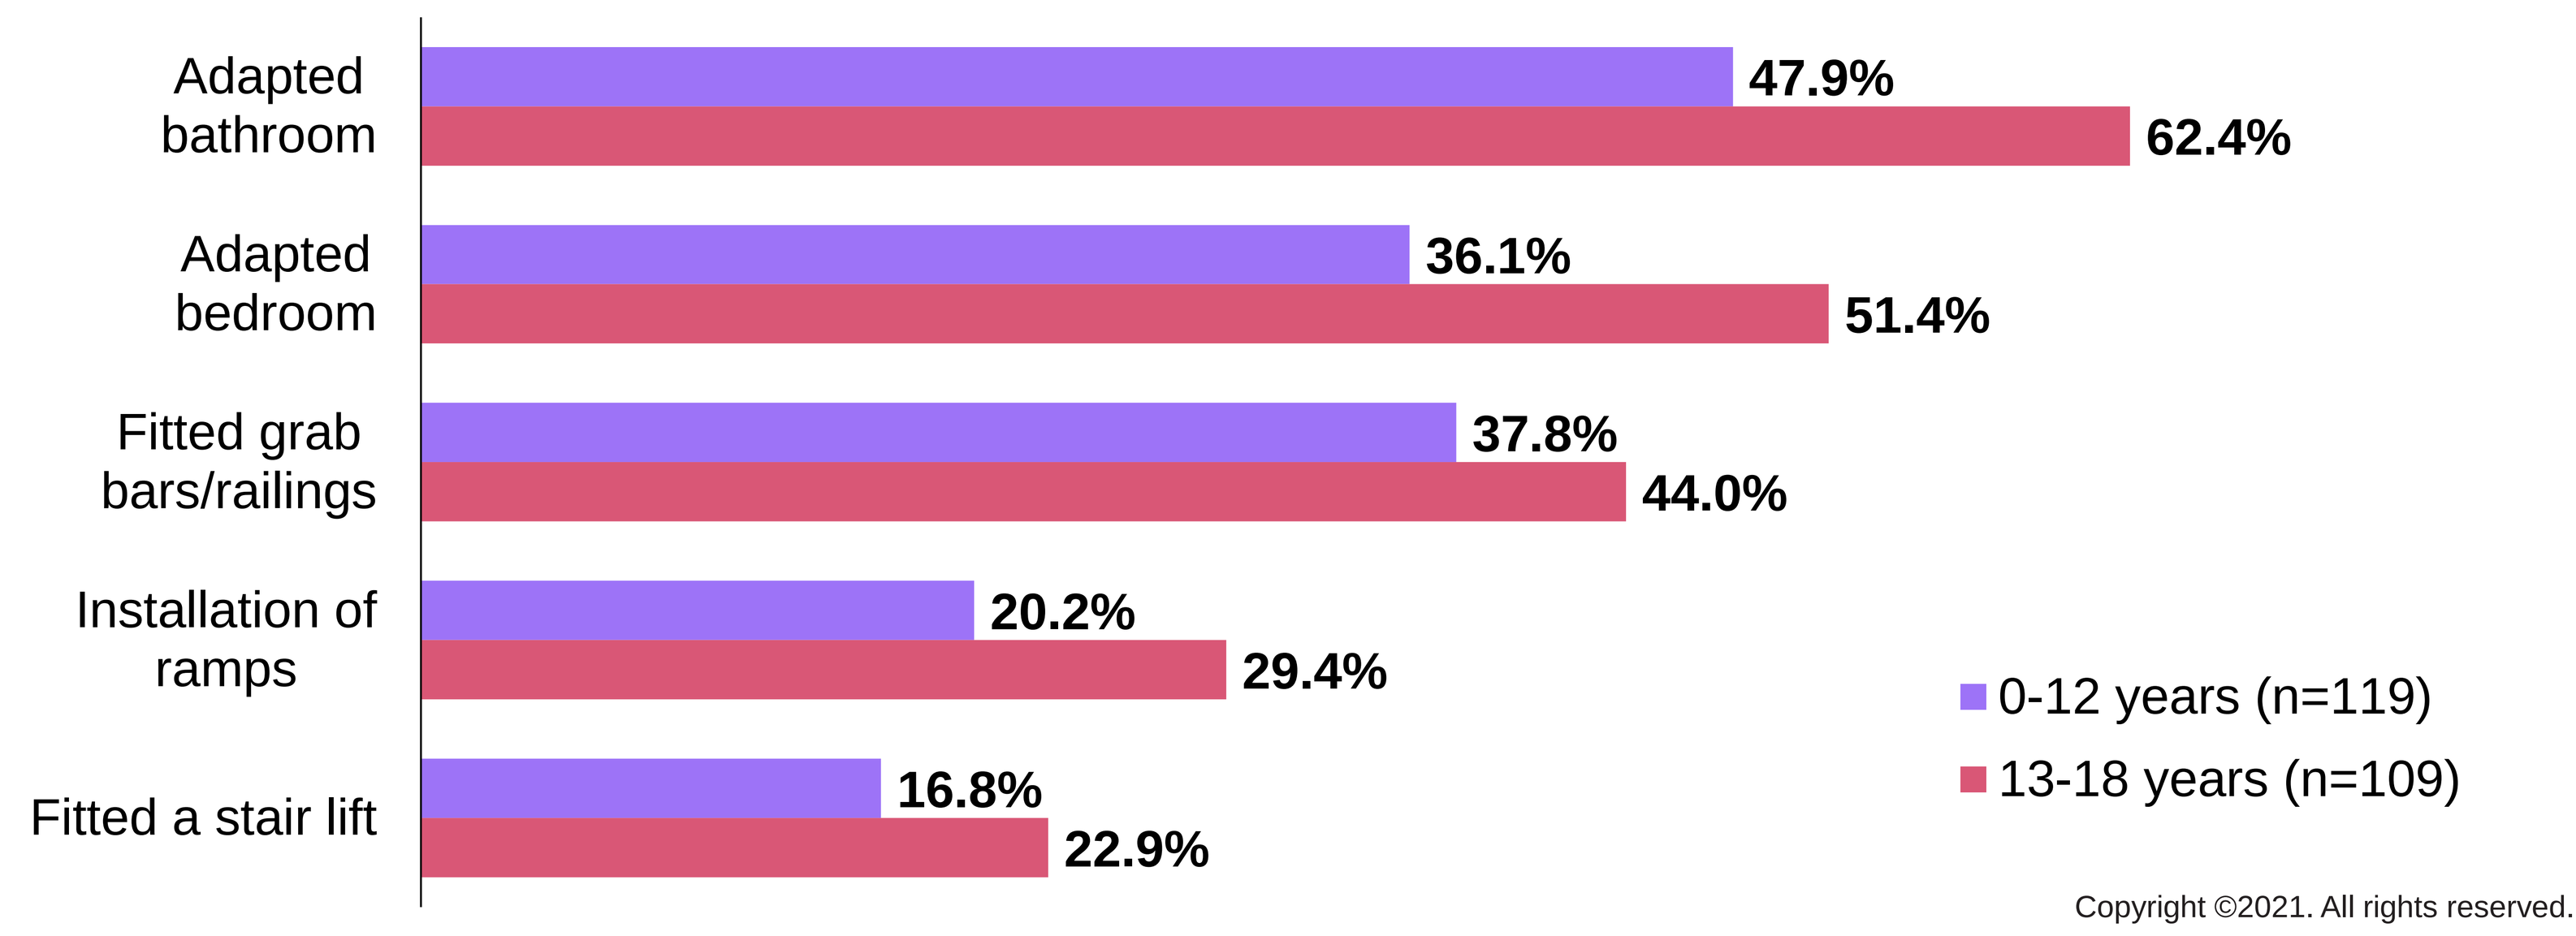
Adapted bathroom
47.9%
62.4%
Adapted bedroom
36.1%
51.4%
Fitted grab bars/railings
37.8%
44.0%
Installation of ramps
20.2%
29.4%
Fitted a stair lift
16.8%
22.9%
0-12 years (n=119)
13-18 years (n=109)

Presenting author: Valeria Merla

Email for more information
Valeria.Merla@pfizer.com

Click or scan this quick response (QR) to download this poster along with associated material.

Acknowledgements:
We would like to thank the physicians and patients for taking part in this study and providing the information included.
The DSP isa wholly owned Adelphi product, of which Pfizer are one of multiple subscribers.

Disclosures:

- JS is on the advisory board for Sarepta, speaker training for NS Pharma, was PI for Fibrogen pamrevlumab phase 2 trial and currently PI for PTC ataluaren phase 3. JS is also a speaker bureau for Biogen (SMA), consultant for UCB and Janssen/Momenta (MG), and PI for Janssen and Roche (MG)
- KI Keiko Ishigaki received honoraria for speakers bureaus from Nippon Shinyaku Co., Ltd and has an advisory role at Pfizer Japan Inc.
- VM, NP, JCC, SX, AT and SB are employees/stockholders of Pfizer
- NH, EC and HI are employees of Adelphi Real World

References:

1. Szabo SM, et al. Orphanet J Rare Dis. 2021; 16(237)
2. Ryder S, et al. Orphanet J Rare Dis. 12(1):79
3. Cavazza M, et al. Eur J Health Econ. 17(S1):19-29
4. Landfeldt E, et al. AAN. 2014; 83(6): 529-536
5. Anderson P, et al. Curr Med Res Opin. 2008; 24(11):3063-3072
6. Babineaux SM, et al. BMJ Open, 2016; 6(8):e010352
7. Higgins V, et al. Diabetes Metab Syndr Obes. 2016; 9:371-380

Presented at ISPOR EU 12-15 November 2023• Virtual

Copyright ©2021. All rights reserved.