RWD121

MOMENT registry (Met nOn sMall cEll caNcer regisTry) for advanced non-small cell lung cancer harboring *MET* exon 14 (*MET*ex14) skipping

Michael Thomas[1], Petros Christopoulos[1], Wade T. Iams[2], Julien Mazières[3], Alexis B. Cortot[4], Nir Peled[5], Gabriele Minuti[6], Egbert F. Smit[7], Sebastian Ahrens[8], Karin Berghoff[9], S Peter Eggleton[10], Frank Fries[11], Peter Liu[12], **Seyed Hamidreza Mahmoudpour**[13], Christoph Menzel[14], Aurora O'Brate[15], Christopher Stroh[14], Dina Oksen[13]

[1]Department of Thoracic Oncology, Thoraxklinik, Heidelberg University Hospital and National Center for Tumor Diseases (NCT), NCT Heidelberg, a partnership between DKFZ and Heidelberg University Hospital, Germany; Translational Lung Research Center Heidelberg (TLRC-H), Member of the German Center for Lung Research (DZL); [2]Division of Hematology/Oncology, Department of Medicine, Vanderbilt University Medical Center, Nashville, TN, USA; [3]CHU de Toulouse, Université Paul Sabatier, Toulouse, France; [4]Université de Lille, CHU Lille, CNRS, Inserm, Institut Pasteur de Lille, UMR9020 – UMR-S 1277 – Canther, F-59000 Lille, France; [5]The Institute of Oncology, Shaare Zedek Medical Center, Jerusalem, Israel; [6]Clinical Trial Center: Phase 1 and Precision Medicine, IRCCS, Regina Elena National Cancer Institute, 00144, Rome, Italy; [7]Department of Pulmonary Diseases, Leiden University Medical Centre, Leiden, The Netherlands; [8]Biosample and Strategy Operations, Merck Healthcare KGaA, Darmstadt, Germany; [9]Global Patient Safety, Merck Healthcare KGaA, Darmstadt, Germany; [10]Global Clinical Development, Merck Serono Ltd., Feltham, UK, an affiliate of Merck KGaA; [11]Data Monitoring Management & Innovation, Merck Healthcare KGaA, Darmstadt, Germany; [12]Global Development Operations, Merck Serono Pharmaceutical R&D Co., Ltd., Beijing, China, an affiliate of Merck KGaA; [13]Department of Epidemiology, Merck Healthcare KGaA, Darmstadt, Germany; [14]Companion Diagnostics & Biomarker Strategy, Merck Healthcare KGaA, Darmstadt, Germany; [15]Global Medical Affairs, Merck Healthcare KGaA, Darmstadt, Germany

CONCLUSIONS

- **The prospective design and the innovative elements of the MOMENT registry enable the collection of comprehensive, high-quality real-world clinical data from patients with advanced NSCLC harboring *MET*ex14 skipping undergoing systemic anticancer treatment in a routine clinical setting**
- **High-quality data from the MOMENT registry will enable future research and studies to inform the optimal care for this rare patient population**

INTRODUCTION

- *MET*ex14 skipping is a *MET* alteration that activates oncogenic MET signaling in ~3% of patients with NSCLC[1,2]
- Targeted treatment with MET inhibitors such as tepotinib, capmatinib, and savolitinib is now available to these patients in routine clinical practice around the world
- Due to the low frequency of this alteration and the limited number of patients, these agents are typically investigated in open-label, single-arm trials[3–5]
- Since MET biomarker testing has only recently been introduced in most countries, with treatment guidelines (NCCN Guidelines®, ESMO)[6,7] only recently updated, historic RWD on *MET*ex14 skipping NSCLC are limited
- High-quality RWD are important to enable further analyses on these rare patients and their outcomes in real-world care settings

OBJECTIVE

- The ongoing MOMENT registry aims to collect data prospectively and enable analyses of uniform, comprehensive, and high-quality data on baseline characteristics, treatment patterns and sequence, and clinical outcomes of patients with advanced NSCLC harboring *MET*ex14 skipping treated in routine clinical practice

MOMENT REGISTRY

Registry design and methods

MOMENT

Registry for advanced NSCLC harboring *MET*ex14 skipping

- MOMENT (NCT05376891) is a non-interventional disease registry collecting data on patients with *MET*ex14 skipping advanced NSCLC receiving any systemic anticancer therapy
- The disease registry design applies available recommendations from the EMA[8] to ensure collection of high-quality harmonized and standardized data with quantifiable quality indicators in terms of completeness, accuracy, validity, consistency, and integrity (e.g. MedDRA, ECOG and NCI-CTCAE) (**Figure 1**; **Table 1**)
- The first patient was enrolled on October 4, 2022
- Approximately 700 patients are expected to be enrolled within 4.5 years

Figure 1. Core data elements of the MOMENT registry

- **Biomarker data*** including *MET*ex14 skipping confirmation, PD-L1 status, and co-mutational profile
- **Patient demographics at diagnosis** including age, sex, smoking status, and country
- **Clinical characteristics over time** including disease stage, histology, ECOG PS, line of therapy, and presence of brain metastasis
- **Exposure to anticancer treatments** including all systemic therapies with reasons for treatment choice and discontinuation
- **Effectiveness outcomes** including longitudinal radiologic tumor assessment†, and mortality data with date and cause of death
- **Safety data** based on a predefined list of all-grade safety events (NCI-CTCAE V5) during anticancer treatment

*Based on tests performed to confirm genomic alterations (laboratory report data including test type, method, results, interpretation). †Including imaging needed to mimic RECIST V1.1 response assessment following clinical practice. Imaging data proposed to be sent to an IRC (if feasible) to independently assess tumor response (impact of any differences could be assessed during data analyses) per RECIST V1.1.

Table 1. Effectiveness and safety assessment

Effectiveness	Safety
• Tumor response per investigator per timepoint for each treatment line (based on RECIST-like criteria*) • Imaging data are collected at baseline and follow-up for central review, and independent assessment of response and progression • Mortality data include date and cause of death	• Data on adverse reactions for each patient include: – Start/stop date – Severity of AEs (Grade 1–5) – Serious AEs – Management, including a need for hospitalization – Dose reductions – Treatment interruptions – Treatment discontinuation – Outcome

*RECIST V1.1 would be possible based on the imaging data and by IRC, if needed in the future.

Eligibility criteria

- The registry enrolls patients with advanced stage (IIIB–IV) NSCLC and confirmed *MET*ex14 skipping, who are initiating or currently treated with at least one systemic therapy (**Figure 2**)

Figure 2. Eligibility criteria

Key inclusion criteria
- Advanced stage (IIIB–IV) NSCLC (all histologies)
- Confirmed *MET*ex14 skipping (by TBx and/or LBx)*
- Initiating or currently treated with systemic anticancer therapy

Exclusion criterion
Patients enrolled in a clinical trial†

Systemic therapies in the MOMENT registry population‡

ICIs§ | Chemotherapy§ | MET inhibitors§ | Other anticancer therapies

*Prior to the initiation of a trial site, local *MET*ex14 skipping detection methods are assessed. †Patients with previous participation in any clinical trial can be included, provided they receive at least one subsequent therapy line in a routine clinical setting. If a patient enters a clinical trial after enrollment into the registry, treatment is blinded for data entry in the eCRF during the time the patient receives any investigational drug. ‡All available anticancer therapies, including those approved, conditionally approved, or available through Early Access. §Given as monotherapy or in combination with other systemic therapies.

Study sites

- The participating sites include oncology and radiotherapy sites, both in the academic and community oncology setting to increase representativeness of the patient population
- The registry is currently operating at more than 60 sites across Europe and North America (**Figure 3**)

Figure 3. MOMENT registry participating countries

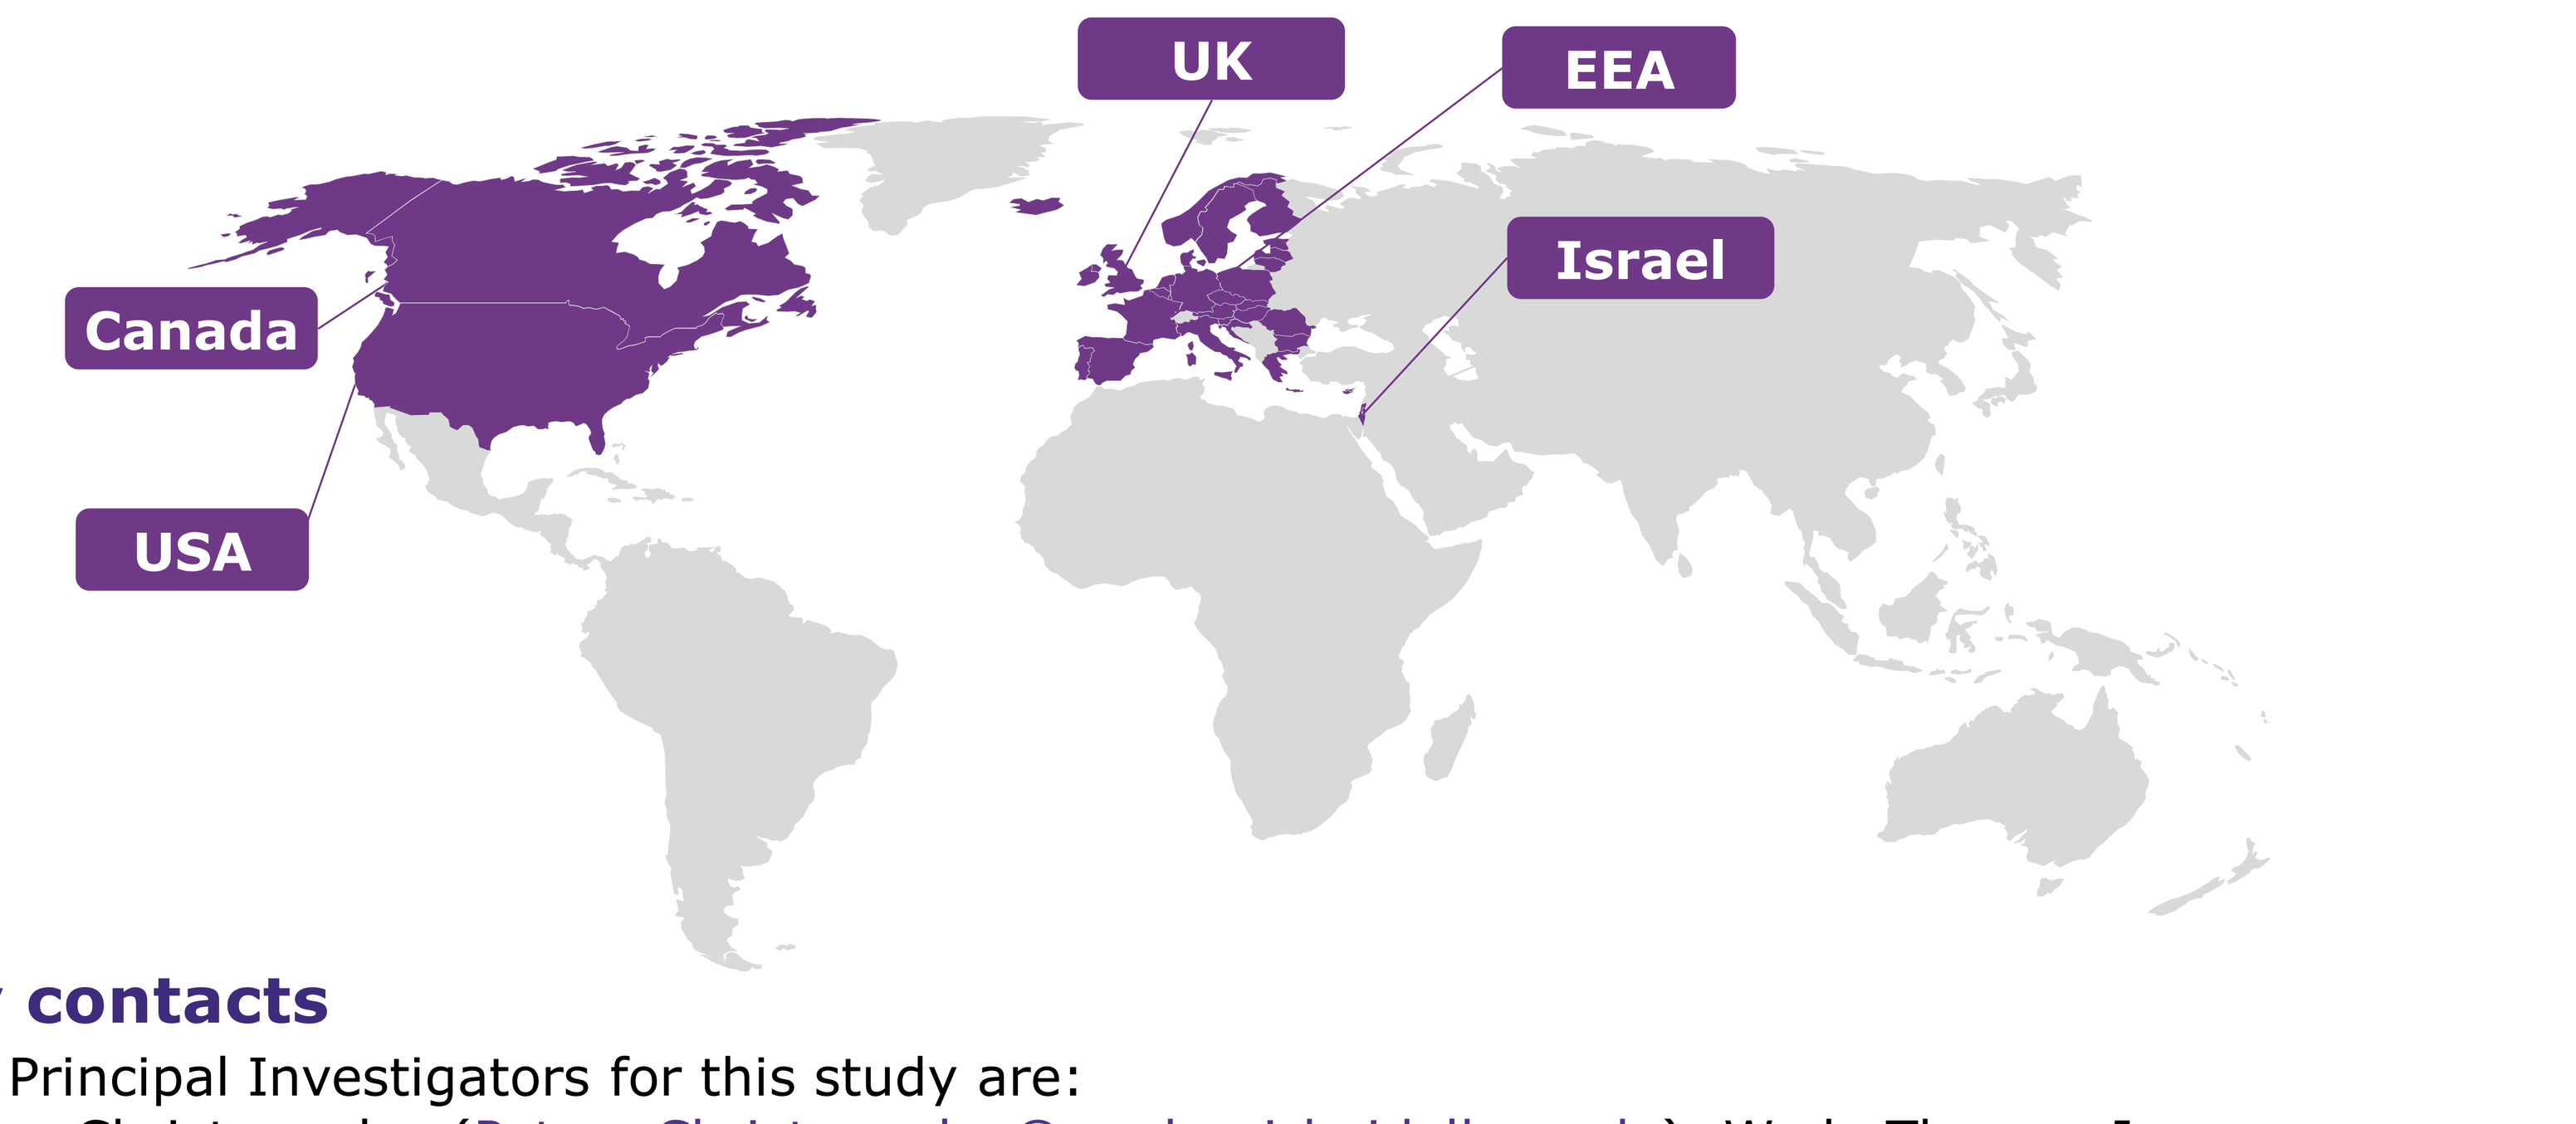

UK
EEA
Israel
Canada
USA

Study contacts

- The Principal Investigators for this study are: Petros Christopoulos (Petros.Christopoulos@med.uni-heidelberg.de), Wade Thomas Iams (wade.t.iams@vumc.org), and Michael Thomas (Michael.Thomas@med.uni-heidelberg.de)
- For further information, please visit https://www.clinicaltrials.gov (ClinicalTrials.gov Identifier: NCT05376891), https://www.encepp.eu/encepp/viewResource.htm?id=47603 (EU PAS Register Number: EUPAS47602), or contact Merck Healthcare KGaA, Darmstadt, Germany (email: MomentRegistry@merckgroup.com)

Abbreviations: AE, adverse event; eCRF, electronic case report form; ECOG PS, Eastern Cooperative Oncology Group performance status; EEA, European Economic Area; EMA, European Medicines Agency; ESMO, European Society for Medical Oncology; EU PAS, European post-authorisation study; ICI, immune-checkpoint inhibitors; IRC, independent review committee; LBx, liquid biopsy; MedDRA, Medical Dictionary for Regulatory Activities; MET, mesenchymal–epithelial transition factor; *MET*ex14, *MET* exon 14; NCCN, National Comprehensive Cancer Network; NCI-CTCAE, National Cancer Institute Common Terminology Criteria for Adverse Events; NSCLC, non-small cell lung cancer; PD-L1, programmed death-ligand 1; RECIST, Response Evaluation Criteria in Solid Tumors; RWD, real-world data; TBx, tissue biopsy.

References: 1. Awad M, et al. *J Clin Oncol*. 2016;34:721–730; 2. Reungwetwattana T, et al. *Lung Cancer*. 2017;103:27–37; 3. Wolf J, et al. *N Engl J Med*. 2020;383:944–957; 4. Lu S, et al. *Lancet Respir Med*. 2021;9:1154–1164; 5. Paik P, et al. *N Engl J Med*. 2020;383:931–943; 6. Hendriks LE, et al. *Ann Oncol*. 2023;34(4):339–357; 7. NCCN Clinical Practice Guidelines in Oncology. (NCCN Guidelines®) for Non-Small Cell Lung Cancer V4.2023. © National Comprehensive Cancer Network, Inc. 2023. Accessed October 2023; 8. EMA. Guideline on registry-based studies (EMA/426390/2021). Available at: https://www.ema.europa.eu/en/documents/scientific-guideline/guideline-registry-based-studies_en-0.pdf. Accessed October 2023.

Funding: This study was sponsored by Merck (CrossRef Funder ID: 10.13039/100009945).

Acknowledgments: The authors would like to thank patients, all investigators and co-investigators, and the teams at all participating centers and at Merck. Medical writing support was provided by Ifigeneia Mavragani, PhD, of Syneos Health, UK, and funded by Merck.

Disclosures: MT–Scientific meetings (self) and traveling support (self): BMS, MSD, AstraZeneca, Novartis, Roche, Takeda, Lilly, Chugai, Celgene, Boehringer Ingelheim, Pfizer; advisory board honoraria (self): BMS, MSD, AstraZeneca, Novartis, Roche, Takeda, Lilly, Boehringer Ingelheim, Pfizer; research funding (institution): BMS, AstraZeneca, Roche, Takeda. **PC–**Research funding from Amgen, AstraZeneca, Boehringer Ingelheim, MSD, Novartis, Roche, Takeda; advisory board/lecture fees: AstraZeneca, Boehringer Ingelheim, Chugai, Daiichi Sankyo, Gilead, Novartis, Pfizer, Roche, Takeda. **WTI–**Consulting or advisory role: EMD Serono Research & Development Institute, Inc., Billerica, MA, USA, an affiliate of Merck KGaA, Janssen, Takeda, Bristol Myers Squibb, Mirati, Chardan Consulting, Curio Science, Defined Health, G1 Therapeutics, Genentech, Jazz Pharmaceuticals, Outcomes Insights. **JM–**Advisory boards: Roche, BMS, MSD, AstraZeneca, Pfizer, Novartis, Merck, Amgen, Takeda, Daiichi Sankyo; research funding (institution): Roche/Genentech, AstraZeneca, Bristol Myers Squibb. **ABC–**Research funding: Novartis, Merck; honoraria: AstraZeneca, BMS, Novartis, MSD, Pfizer, Roche, Takeda; consulting or advisory role: AstraZeneca, BMS, Novartis, Pfizer, Roche, Takeda; travel expenses: AstraZeneca, MSD, Novartis, Pfizer, Roche, Takeda. **NP–**Advisor, honoraria, and research funding: Bristol Myers Squibb, Eli Lilly, Foundation Medicine, Guardant Health, Merck Serono Ltd., Herzliya, Israel, an affiliate of Merck KGaA, MSD, Novartis, NovellusDx, Pfizer, Roche, Takeda. **GM–**Consulting or advisory role: AstraZeneca, BMS, Roche. **EFS–**Advisory/consultancy (institution): Lilly, AstraZeneca, Boehringer Ingelheim, Roche/Genentech, Bristol Myers Squibb, Merck, MSD, Takeda, Bayer, Regeneron, Novartis, Daiichi Sankyo, Seattle Genetics; research funding (institution): Boehringer Ingelheim, Bayer, Roche/Genentech, AstraZeneca, Bristol Myers Squibb. **SA**, **KB**, **FF**, **SHM**, **CM**, **AOB**, **CS**, **DO–**Employees of Merck. **SPE–**Employee of Merck Serono Ltd., Feltham, UK, an affiliate of Merck KGaA. **PL–**Employee of Merck Serono Co., Ltd., Beijing, China, an affiliate of Merck KGaA.

Presented at the International Society for Pharmacoeconomics and Outcomes Research (ISPOR) Europe 2023 | November 12–15, 2023 | Copenhagen, Denmark and virtual

POSTER PDF

Copies of this poster obtained through this Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster.

Correspondence:
Hamid.Mahmoudpour@merckgroup.com