RWD157

Assessment of Real-World Treatment Patterns and Healthcare Resource Utilization in Patients With Lenalidomide-Refractory Relapsed/Refractory Multiple Myeloma From the US Optum Database

Luciano J Costa[1], Sandhya Nair[2], Xiwu Lin[3], Marguerite O'Hara[4], Mary Slavcev[4], Alexander Marshall[4], Ravi Potluri[5], Rohit Tyagi[5], Hamza Hashmi[6]

[1]University of Alabama at Birmingham, Birmingham, AL, USA; [2]Janssen Pharmaceutica NV, Beerse, Belgium; [3]Janssen Global Services, Horsham, PA, USA; [4]Janssen Global Services, Raritan, NJ, USA; [5]Putnam PHMR, New York, NY, USA; [6]Medical University of South Carolina, Charleston, SC, USA

INTRODUCTION

- Despite recent advancement in treatments, multiple myeloma (MM) remains an incurable disease and patients are at persistent risk of relapsing or becoming refractory to therapies[1-3]
- Patients with relapsed/refractory MM often cycle through multiple treatment regimens, including 3 of the most commonly used classes of therapy (ie, immunomodulatory drugs [IMiDs], proteasome inhibitors [PIs], and anti-CD38 monoclonal antibodies [mAbs])
 - Exposure to 2 or more of these classes of therapy results in increased costs and healthcare resource utilization (HCRU)[4,5]
- With increasing utilization of lenalidomide in earlier lines of therapy (LOTs), including in newly diagnosed MM, treatment options with alternative mechanisms of action (MOA) at relapse are limited

OBJECTIVE

- The objective of our analysis was to characterize real-world treatment patterns and HCRU in patients who had received 1–3 prior LOTs (including lenalidomide) and/or were refractory to lenalidomide

METHODS

Data sources

- Data were extracted from the US Optum Claims database for patients with an index date in or after January 2016
 - Index date was defined as the start of first subsequent LOT post inclusion
 - This analysis considered only the first index event per patient

Inclusion criteria

- Patients with MM had received 1–3 prior LOTs (including a PI and lenalidomide, and were refractory to lenalidomide if only 1 prior LOT)
 - Prior anti-CD38 exposure allowed but patients must not have been anti-CD38 mAb refractory
 - No prior exposure to anti–B–cell maturation antigen therapy allowed

Assessments

- Demographics and baseline characteristics were evaluated for all patients
 - Comorbidities were scored per the Charlson Comorbidity Index (CCI), with an increasing CCI score reflecting increasing mortality risk
 - The presence of the most common Elixhauser comorbidities (based on diagnosis codes) was also assessed
- Treatment patterns were analyzed for the overall cohort
- The following outcomes were stratified by insurance type (commercial or Medicare):
 - Time to next treatment or death (TTNT; estimated using the Kaplan-Meier method starting at index date)
 - HCRU (including per-patient per-month [PPPM] outpatient visits, hospitalizations, emergency room [ER] visits, and laboratory [lab] visits

Statistical analyses

- Descriptive statistics are reported for all analyzed data

MULTIPLE MYELOMA

RESULTS

Patient demographics and baseline characteristics

- Demographics and baseline characteristics were similar across insurance types in the 1646 patients analyzed (**Table 1**)
- Median time from index to end of follow-up was 11 months (interquartile range [IQR], 4–23) overall, 13 months (IQR, 5–25) in patients with commercial insurance, and 10 months (IQR, 4–23) in those with Medicare

TABLE 1: Demographics and baseline characteristics

Variable	Overall (N=1646)	Commercial (n=476)	Medicare (n=1170)
Median age at index date, years (IQR)	73 (66–78)	61 (56–66)	76 (71–80)
Stem cell transplant before index date, n (%)	65 (3.9)	37 (7.8)	28 (2.4)
Median time from MM diagnosis to index date, months (IQR)	15 (7–32)	11 (6–26)	17 (8–33)
Number of prior LOTs, n (%)			
1	724 (44.0)	233 (49.0)	491 (42.0)
2	845 (51.3)	219 (46.0)	626 (53.5)
3	77 (4.7)	24 (5.0)	53 (4.5)
Refractory status, n (%)			
Not refractory to IMiD or PI	203 (12.3)	51 (10.7)	152 (13.0)
Single-refractory (IMiD only)	457 (27.8)	145 (30.5)	312 (26.7)
Single-refractory (PI only)	203 (12.3)	55 (11.6)	148 (12.6)
Double-refractory (IMiD and PI)	783 (47.6)	225 (47.3)	558 (47.7)
Lenalidomide refractory	1220 (74.1)	365 (76.7)	855 (48.3)
Median CCI (IQR)	2 (0–4)	1 (0–4)	2 (0–5)
CCI, n (%)			
0	523 (31.8)	195 (41.0)	328 (28.0)
1	261 (15.9)	61 (12.8)	200 (17.1)
≥2	862 (52.4)	220 (46.2)	642 (54.9)
Elixhauser comorbidities, n (%)[a]			
Hypertension	1042 (63.3)	242 (50.8)	800 (68.4)
Renal failure	581 (35.3)	98 (20.6)	483 (41.3)
Fluid and electrolyte disorders	549 (33.4)	134 (28.2)	415 (35.5)

[a]In ≥30% of patients overall.

Treatment patterns

- The most common treatment regimens were daratumumab, bortezomib, and dexamethasone (6.0%), pomalidomide and dexamethasone (5.4%), and daratumumab, pomalidomide, and dexamethasone (4.7%)
 - The most frequently used regimen type was triplet therapy (33.4%), followed by monotherapy (32.1%), doublets (28.9%), and quadruplets (5.7%)

TTNT

- Median TTNT was 6.1 months (95% CI, 5.7–6.7) overall. Median TTNT was lower for patients with commercial insurance compared with those with Medicare (**Figure 1**)

HCRU

- Mean PPPM visits by insurance type are shown in **Figure 2**, with mean HCRU from the first LOT through loss to follow-up shown in **Table 2** overall and by insurance type
 - Mean (SD) length of inpatient stay was 0.9 (2.0) days overall (0.7 [1.6] days vs 1.0 [2.1] days in patients with commercial insurance and Medicare, respectively)

FIGURE 1: Median TTNT by insurance type

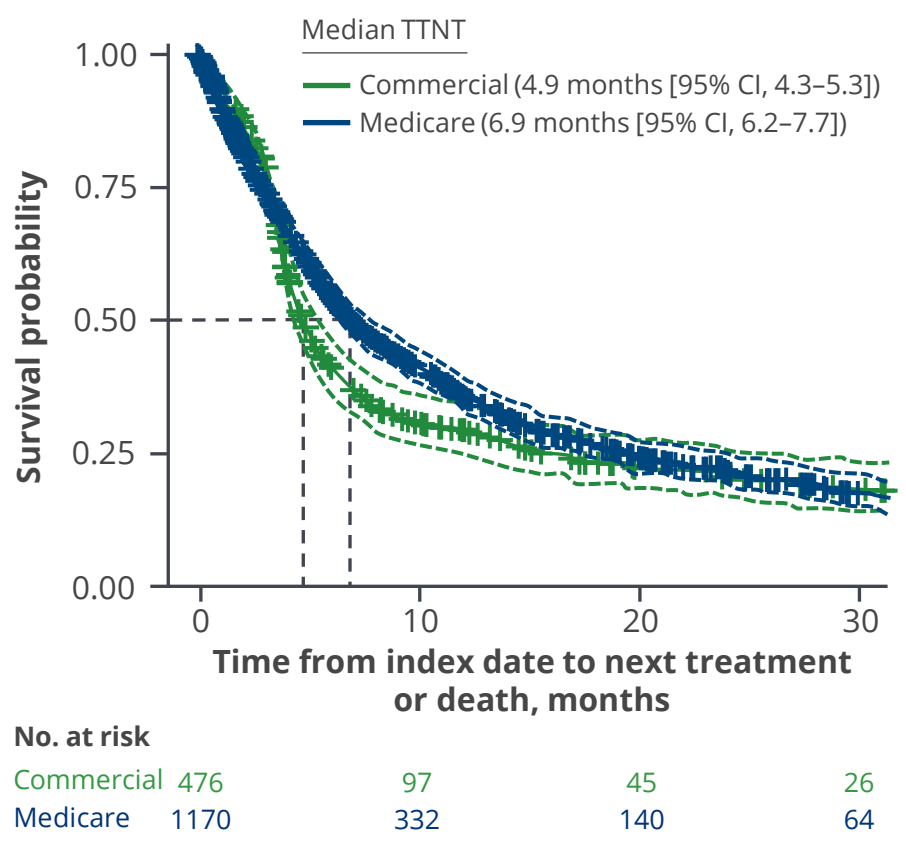
Median TTNT
Commercial (4.9 months [95% CI, 4.3–5.3])
Medicare (6.9 months [95% CI, 6.2–7.7])
Survival probability
1.00
0.75
0.50
0.25
0.00
0
10
20
30
Time from index date to next treatment or death, months
No. at risk
Commercial 476 97 45 26
Medicare 1170 332 140 64

Dotted colored lines represent upper and lower bounds of the 95% CIs.

FIGURE 2: Mean PPPM visits by insurance type

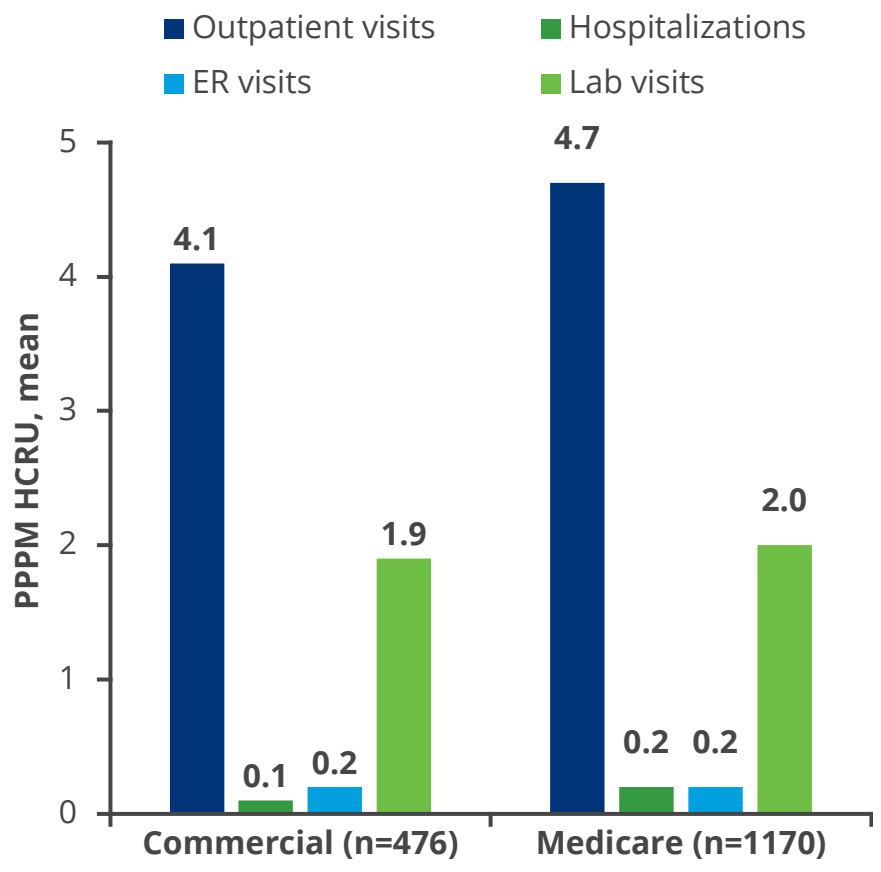
Outpatient visits
Hospitalizations
ER visits
Lab visits
PPPM HCRU, mean
5
4
3
2
1
0
4.1
0.1
0.2
1.9
4.7
0.2
0.2
2.0
Commercial (n=476)
Medicare (n=1170)

TABLE 2: Mean HCRU by insurance type

HCRU	Variable	Overall (N=1646)	Commercial (n=476)	Medicare (n=1170)
Outpatient visits	Patients with ≥1 outpatient visit, n (%)	1556 (94.5)	465 (97.7)	1091 (93.2)
	Outpatient visits PPPM, mean (SD)	4.5 (2.8)	4.1 (2.0)	4.7 (3.0)
Hospital stays	Patients with ≥1 hospitalization, n (%)	960 (58.3)	285 (59.9)	675 (57.7)
	Inpatient stay PPPM, mean (SD)	0.1 (0.4)	0.1 (0.2)	0.2 (0.4)
ER visits	Patients with ≥1 ER visit, n (%)	1022 (62.1)	277 (58.2)	745 (63.7)
	ER visits PPPM, mean (SD)	0.2 (0.3)	0.2 (0.4)	0.2 (0.3)
Lab visits	Patients with ≥1 lab visit, n (%)	1527 (92.8)	459 (96.4)	1068 (91.3)
	Lab visits PPPM, mean (SD)	1.9 (1.2)	1.9 (1.2)	2.0 (1.2)

REFERENCES:

1. Ravi P, et al. *Blood Cancer J* 2018;8:26. 2. Dimopoulos M, et al. *Eur J Haematol* 2010;86:1-15. 3. Touzeau C, et al. *Ann Hematol* 2021;100:1825-36. 4. Madduri D, et al. *Future Oncol* 2021;17:503-15. 5. Yang J, et al. *J Manag Care Spec Pharm* 2023;29:917-26.

Presented at the ISPOR Europe Annual Meeting; November 12–15, 2023; Copenhagen, Denmark

KEY TAKEAWAY

Patients with prior PI and lenalidomide exposure and 1–3 prior LOTs progressed quickly through various regimens and required high HCRU, highlighting an unmet need for newer effective regimens for this population

CONCLUSIONS

The high HCRU observed for patients with 1–3 prior LOTs could be mitigated with earlier use of effective treatments with novel MOAs

The TTNT seen in this population was short, underlining the need for more effective regimens in patients with limited treatment options

This analysis was limited by small patient numbers, consideration of the first index event only, and lack of statistical power to compare patients with different insurance types

ACKNOWLEDGMENTS

This study was funded by Janssen Global Services, LLC. Medical writing support was provided by Emily Bell, BSc, of Eloquent Scientific Solutions, and funded by Janssen Global Services, LLC.

DISCLOSURES

LJC has received grants or contracts from AbbVie, Amgen, and BMS; and has received consulting fees from AbbVie, Amgen, BMS, Pfizer, and Sanofi. **SN**, **XL**, **MO**, **MS**, and **AM** are employees of Janssen. **RP**, **RT**, and **HH** are employees of Putnam PHMR, which was paid by Janssen to conduct these analyses.