EE711

Cost-effectiveness of umeclidinium/vilanterol (UMEC/VI) versus tiotropium/olodaterol (TIO/OLO) in patients with COPD: United Kingdom payer perspective

Digital poster

Kendall R[1], Martin A[2], Shah D[3], Noorduyn SG[4,5], Dasari P[6], Risebrough NA[7], Ismaila AS[8,5]

[1]ICON Health Economics, ICON plc, Vancouver, BC, Canada; [2]Value Evidence and Outcomes, GSK, Brentford, Middlesex, UK; [3]ICON Health Economics, ICON plc, Morriston, NJ, USA; [4]Global Value Evidence and Outcomes, GSK, Mississauga, ON, Canada; [5]Department of Health Research Methods, Evidence and Impact, McMaster University, Hamilton, ON, Canada; [6]ICON Health Economics, ICON plc, Houston, TX, USA; [7]ICON Health Economics, ICON plc, Ontario, Canada; [8]Value Evidence and Outcomes, GSK, Collegeville, PA, USA

Introduction

- Patients with COPD experience substantial morbidity and mortality, resulting in high healthcare resource utilisation and treatment costs.[1]
- Dual long-acting bronchodilators are recommended by the UK NICE guidelines as maintenance therapy for patients with COPD with persistent symptoms or exacerbations.[2]
- Dual therapy with UMEC/VI was compared with TIO/OLO treatment in patients with COPD in a recent NMA, using a frequentist regression-based analysis, with networks stratified by observation time horizon (12 and 24 weeks).[3]
 - The included RCTs had a duration of at least 8 weeks and observed patients with COPD indexed on a dual therapy (LAMA/LABA), testing lung function, health-related quality of life, breathlessness, RMU and exacerbations.[3]
 - The NMA found that at 24 weeks, UMEC/VI provided significantly greater improvements in trough FEV_1 versus TIO/OLO, while TDI focal score showed significantly greater improvement with TIO/OLO than UMEC/VI. At 12 weeks, there were significantly greater improvements in RMU with UMEC/VI versus TIO/OLO. The therapies did not exhibit a statistically significant difference with regard to SGRQ score, time-to-first exacerbation or exacerbation rate.[3]

Objective

Using data from this NMA, this analysis assessed the cost-effectiveness of UMEC/VI versus TIO/OLO for the treatment of COPD, from a UK NHS perspective.

Conclusion

- Based on this analysis, UMEC/VI was the dominant treatment option compared with TIO/OLO in patients with COPD in the UK from an NHS perspective, with lower predicted costs and improved health outcomes.
- These findings suggest that UMEC/VI should be considered as a preferred treatment option for treatment of COPD versus TIO/OLO by physicians in the UK.
- Limitations of this study include:
 - Only 12-week discontinuation rate data were collected, with no data available for treatment discontinuation in the subsequent years, although scenario analyses showed discontinuation rates did not substantially influence the cost-effectiveness of UMEC/VI.
 - While data on baseline fibrinogen and 6MWT at baseline were unavailable, sensitivity analyses for these factors showed that the incremental benefit for QALYs and costs did not change substantially compared with the deterministic results.

Methods

- The GALAXY model[4] is a validated and extensively published economic model employing a linked risk-equation approach to model COPD disease progression and outcomes and evaluate cost-effectiveness.
- For the purposes of this analysis, the GALAXY model was populated with pooled patient baseline characteristics (**Table 1**) and treatment effects (**Table 2**).
 - The population parameters were obtained from an 8-week, multicentre, crossover clinical trial in symptomatic patients with COPD, that compared once-daily UMEC/VI 62.5/25 mcg with once-daily TIO/OLO 5/5 mcg (Study 204990; NCT02799784).[5]
 - The analysis was conducted in accordance with NICE health technology evaluations guidelines.[6]

Table 1: **Pooled baseline characteristics**

Characteristic	Study 204990 (pooled data)	SPARK (pooled data)	Notes
Female, %	39.8	23.7	
Age, years, mean (SE)	64.4 (0.55)	63.1 (8.10)	
BMI, %			
Low (<21)	10.0	10.0	Data unavailable in SPARK, obtained from Study 204990
Medium (21–30)	50.0	50.0	Data unavailable in SPARK, obtained from Study 204990
High (>30)	40.0	40.0	Data unavailable in SPARK, obtained from Study 204990
Any CV comorbidity, %	27.0	27.0	Data unavailable in SPARK, obtained from Study 204990
Any other comorbidity, %	78.4	78.4	Data unavailable in SPARK, obtained from Study 204990
History of ≥1 exacerbation, %	19.0	98.0	
mMRC score ≥2, %	100.0	100.0	Data unavailable in SPARK, obtained from Study 204990
Current smokers, %	53.0	38.0	
Height, cm, mean (SE)	169.9 (0.60)	169.9 (0.60)	Data unavailable in SPARK, obtained from Study 204990
Fibrinogen, μg/dL, mean (SE)	453.2 (2.4)	488.5 (2.4)	Derived from a risk equation
6MWT, m, mean (SE)	349.9 (2.7)	308.2 (2.7)	Derived from a risk equation
Number of exacerbations in previous year, mean (SE)	0.18 (0.03)	0.18 (0.03)	Data unavailable in SPARK, obtained from Study 204990
Moderate exacerbations	0.16	0.16	Data unavailable in SPARK, obtained from Study 204990
Severe exacerbations	0.02	0.02	Data unavailable in SPARK, obtained from Study 204990
Starting FEV_1 % predicted, mean (SE)	59.6 (5.6)	37.0 (0.3)	
Starting SGRQ total score, mean (SE)	43.1 (1.00)	53.0 (0.67)	

Table 2: **Treatment effects***

Parameter	UMEC/VI vs TIO/OLO mean difference (95% CI)
FEV_1, change from baseline, mL	41.81 (20.21, 63.40)
SGRQ score, change from baseline	0.79 (–1.09, 2.67)
Moderate/severe exacerbations, relative risk	NA (assumed parity)
RMU, change from baseline in puffs/day	–0.25 (–0.37, –0.13)

**Data obtained from the NMA. Only 12 weeks analysis available.[3]*

- UK healthcare resource unit and drug costs were applied as described in **Table 3**, with costs (GBP, 2022) and QALYs discounted at 3.5% annually.
- Base-case settings and assumptions are shown in **Table 4.**

Table 3: **Cost inputs***

Parameter	Base-case input value (N=1,104) UMEC/VI	Base-case input value (N=1,104) TIO/OLO	Source
Drug costs per day, £	1.08	1.08	BNF 2022[8]
Healthcare costs per year or per exacerbation, £			**Source**
Disease management, COPD severity (FEV_1 % predicted)			
Moderate to severe (50–80%)	128		NICE 2018[9]
Severe (30–<50%)	850		NICE 2018[9]
Very severe (<30%)	1,578		NICE 2018[9]
Exacerbation			
Moderate	88		NICE 2018[9]
Severe	2,379		NICE 2018[9]
Moderate exacerbation event outpatient costs, £			**Source**
Oral corticosteroid: prednisolone	1.04		NICE 2019[2]
Antibiotics			
Amoxicillin	0.06		NICE 2018[9]
Doxycycline	0.14		NICE 2018[9]
Clarithromycin	0.28		NICE 2018[9]

**Unit costs were inflated to 2022 values using the Consumer Price Index data obtained from the Office of National Statistics.[10]*

Table 4: **Base-case model settings and assumptions used in this study**

Base-case model settings	Assumptions
• UK NHS perspective	• Treatment effect was assumed to be persistent at a constant rate for all patients
• Study 204990 population	• Treatment discontinuation was assumed to be zero in the first and subsequent years
• Lifetime horizon (probabilistic analysis)	• UMEC/VI treatment effects for FEV_1 were assumed to begin at the onset of the analysis (zero months)
• 1-year cycle length	
• 3.5% discount rate for costs and benefits	
• Treatment discontinuation and patient productivity costs excluded	

Assessment of impact of uncertainty

- To address the uncertainty in parameter estimation, the base-case analysis was probabilistic (with a deterministic analysis for comparison). Input parameters were assigned distributions which were randomly sampled over 5,000 Monte Carlo simulations.
- One-way sensitivity and scenario analyses were conducted to test robustness of the model results.
 - Scenario analyses used patient baseline demographics from the SPARK study (a 64-week parallel-group study comparing of IND/GLY versus GLY and TIO monotherapies)[7] to evaluate the effects of starting with a population with more severe COPD.
 - Scenario analyses also evaluated the impact of time horizon, discount rates, treatment effects, treatment discontinuation and subsequent treatment, rescue medication cost exclusion and patient productivity cost inclusion.
 - Sensitivity analyses were performed on baseline covariate values not available from Study 204990, and UMEC/VI treatment effects on exacerbation, SGRQ and FEV_1.

Results

Deterministic analysis

- Over a lifetime horizon (25 years), UMEC/VI was predicted to result in 0.019 fewer total exacerbations PPPY and provided an additional 0.220 LYs and 0.104 QALYs with cost savings of £1,079 compared with TIO/OLO.
- UMEC/VI was predicted to result in a lower number of both moderate and severe exacerbations, resulting in both lower drug- and non-drug-related healthcare costs.

Probabilistic base-case analysis

- Over a lifetime horizon (25 years), UMEC/VI was predicted to result in 0.019 fewer total exacerbations PPPY and provided an additional 0.216 LYs (95% range: 0.077, 0.391) and 0.102 QALYs (0.038, 0.184), with cost savings of £1,259 (£853, £1,765) compared with TIO/OLO (**Table 5**).

Table 5: **Cost-effectiveness analyses for UMEC/VI versus TIO/OLO over a lifetime horizon**

Probabilistic (25 years)	UMEC/VI	TIO/OLO	Incremental
Cumulative number of exacerbations			
Moderate	5.571	5.580	–0.009
Severe	1.401	1.447	–0.046
Total	6.973	7.028	–0.055
Severe exacerbations PPPY	0.132	0.140	–0.007
Total exacerbations PPPY	0.659	0.678	–0.019
Outcomes at end of timeframe			
Survival at end of time horizon, %	1.0	0.8	0.17
Accumulated LYs (undiscounted)	10.581	10.365	0.216
Accumulated QALYs	5.878	5.776	0.102
Costs at end of timeframe, £			
Accumulated costs total	14,465	15,723	–1,259
Drug costs	8,568	9,262	–694
Total non-drug costs	5,896	6,461	–565
Incremental results			
Incremental cost, £ (95% range)			–1,259 (–1,765, –853)
Incremental LYs, undiscounted (95% range)			0.216 (0.077, 0.391)
Incremental QALYs (95% range)			0.102 (0.038, 0.184)
ICER/QALY (vs TIO/OLO)			Dominant (Dominant, Dominant)

- UMEC/VI was less costly and showed higher QALYs compared with TIO/OLO across almost all simulations and remained the more favourable treatment option compared with TIO/OLO for 100% of 5,000 simulations (**Figure 1**).

Figure 1: **Base-case incremental cost-effectiveness plane**

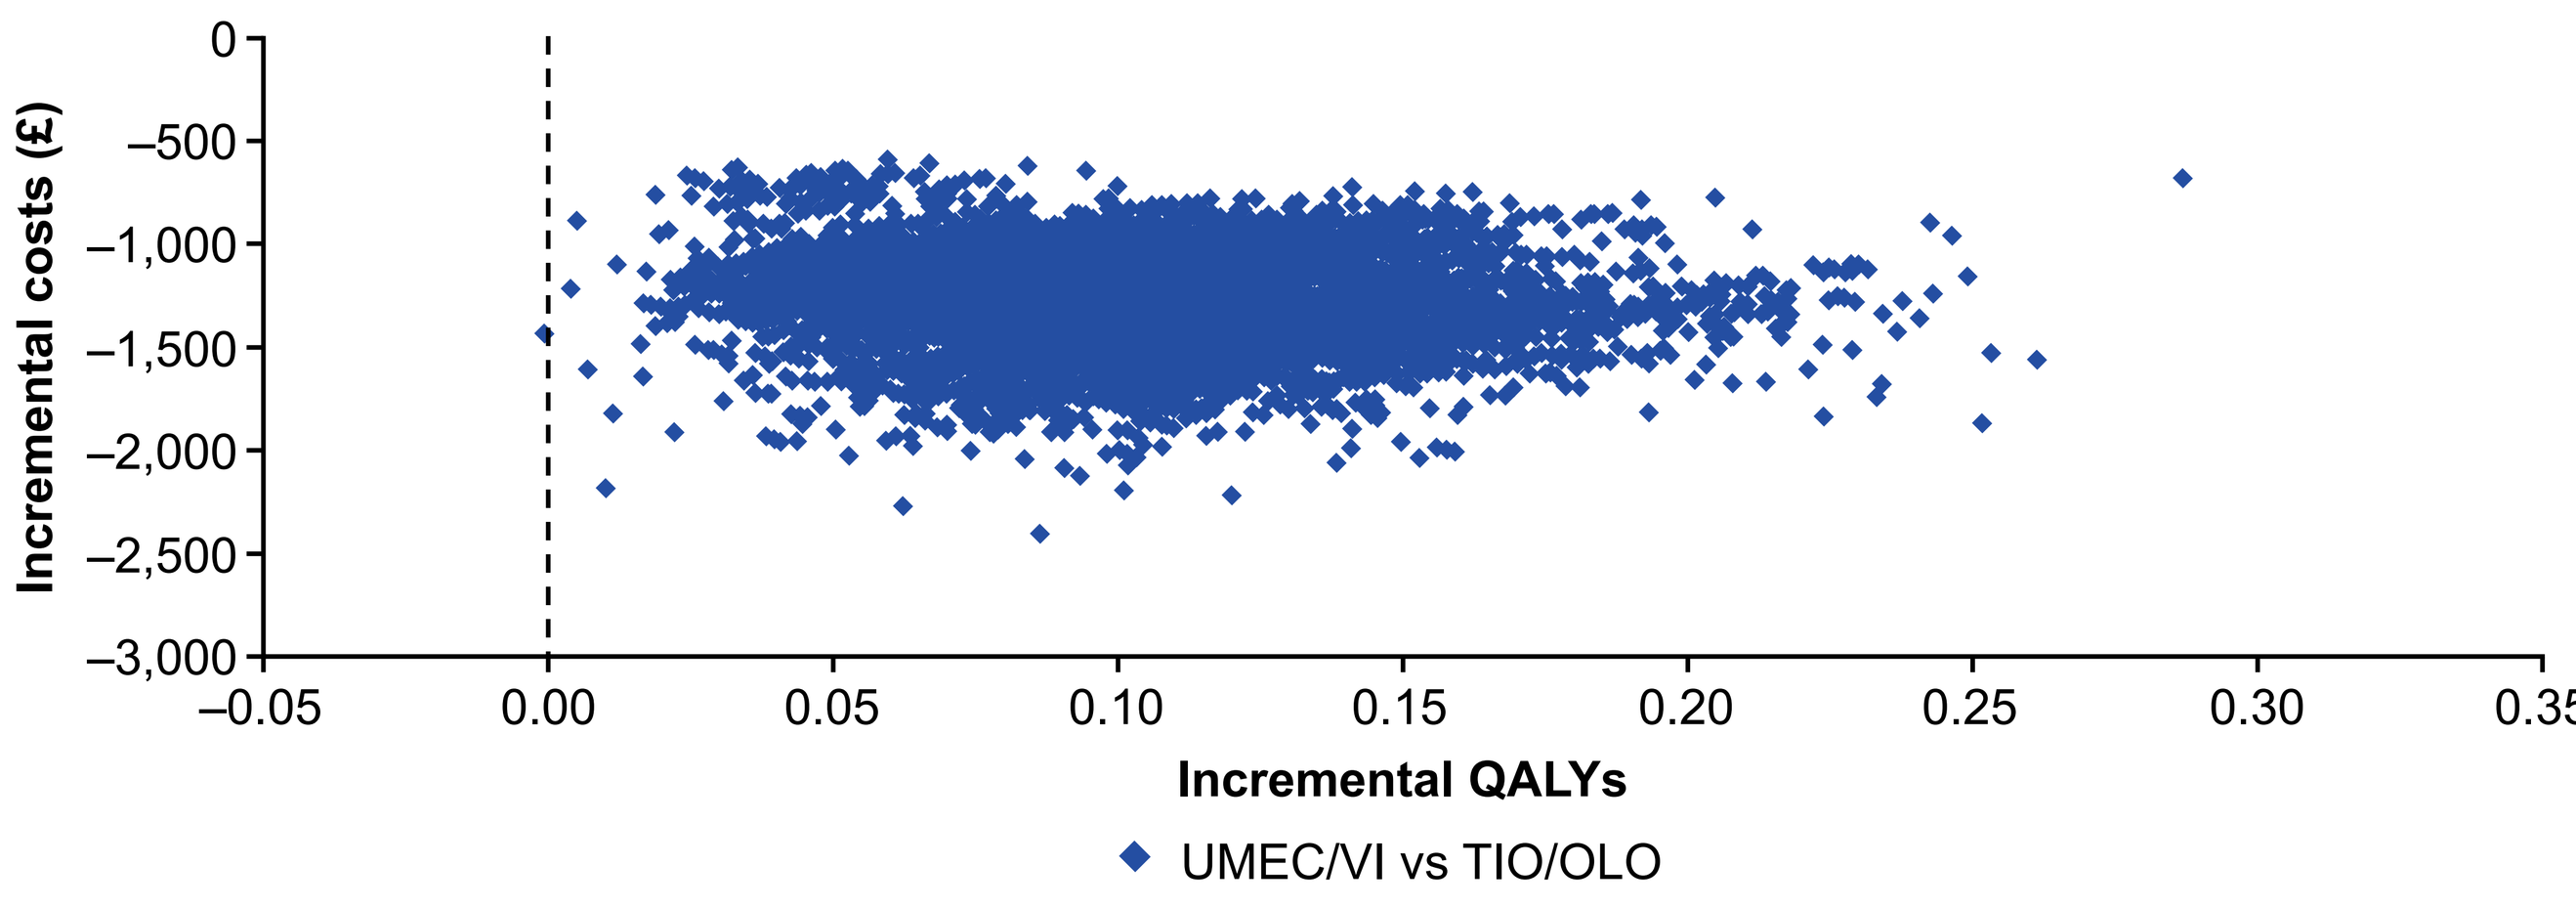
Incremental costs (£)
0
–500
–1,000
–1,500
–2,000
–2,500
–3,000
–0.05
0.00
0.05
0.10
0.15
0.20
0.25
0.30
0.35
Incremental QALYs
UMEC/VI vs TIO/OLO

- At a willingness-to-pay threshold of £20,000 per QALY, the probability that UMEC/VI was cost-effective was 100% versus TIO/OLO (**Figure 2**).

Figure 2: **Base-case net-benefit acceptability curve**

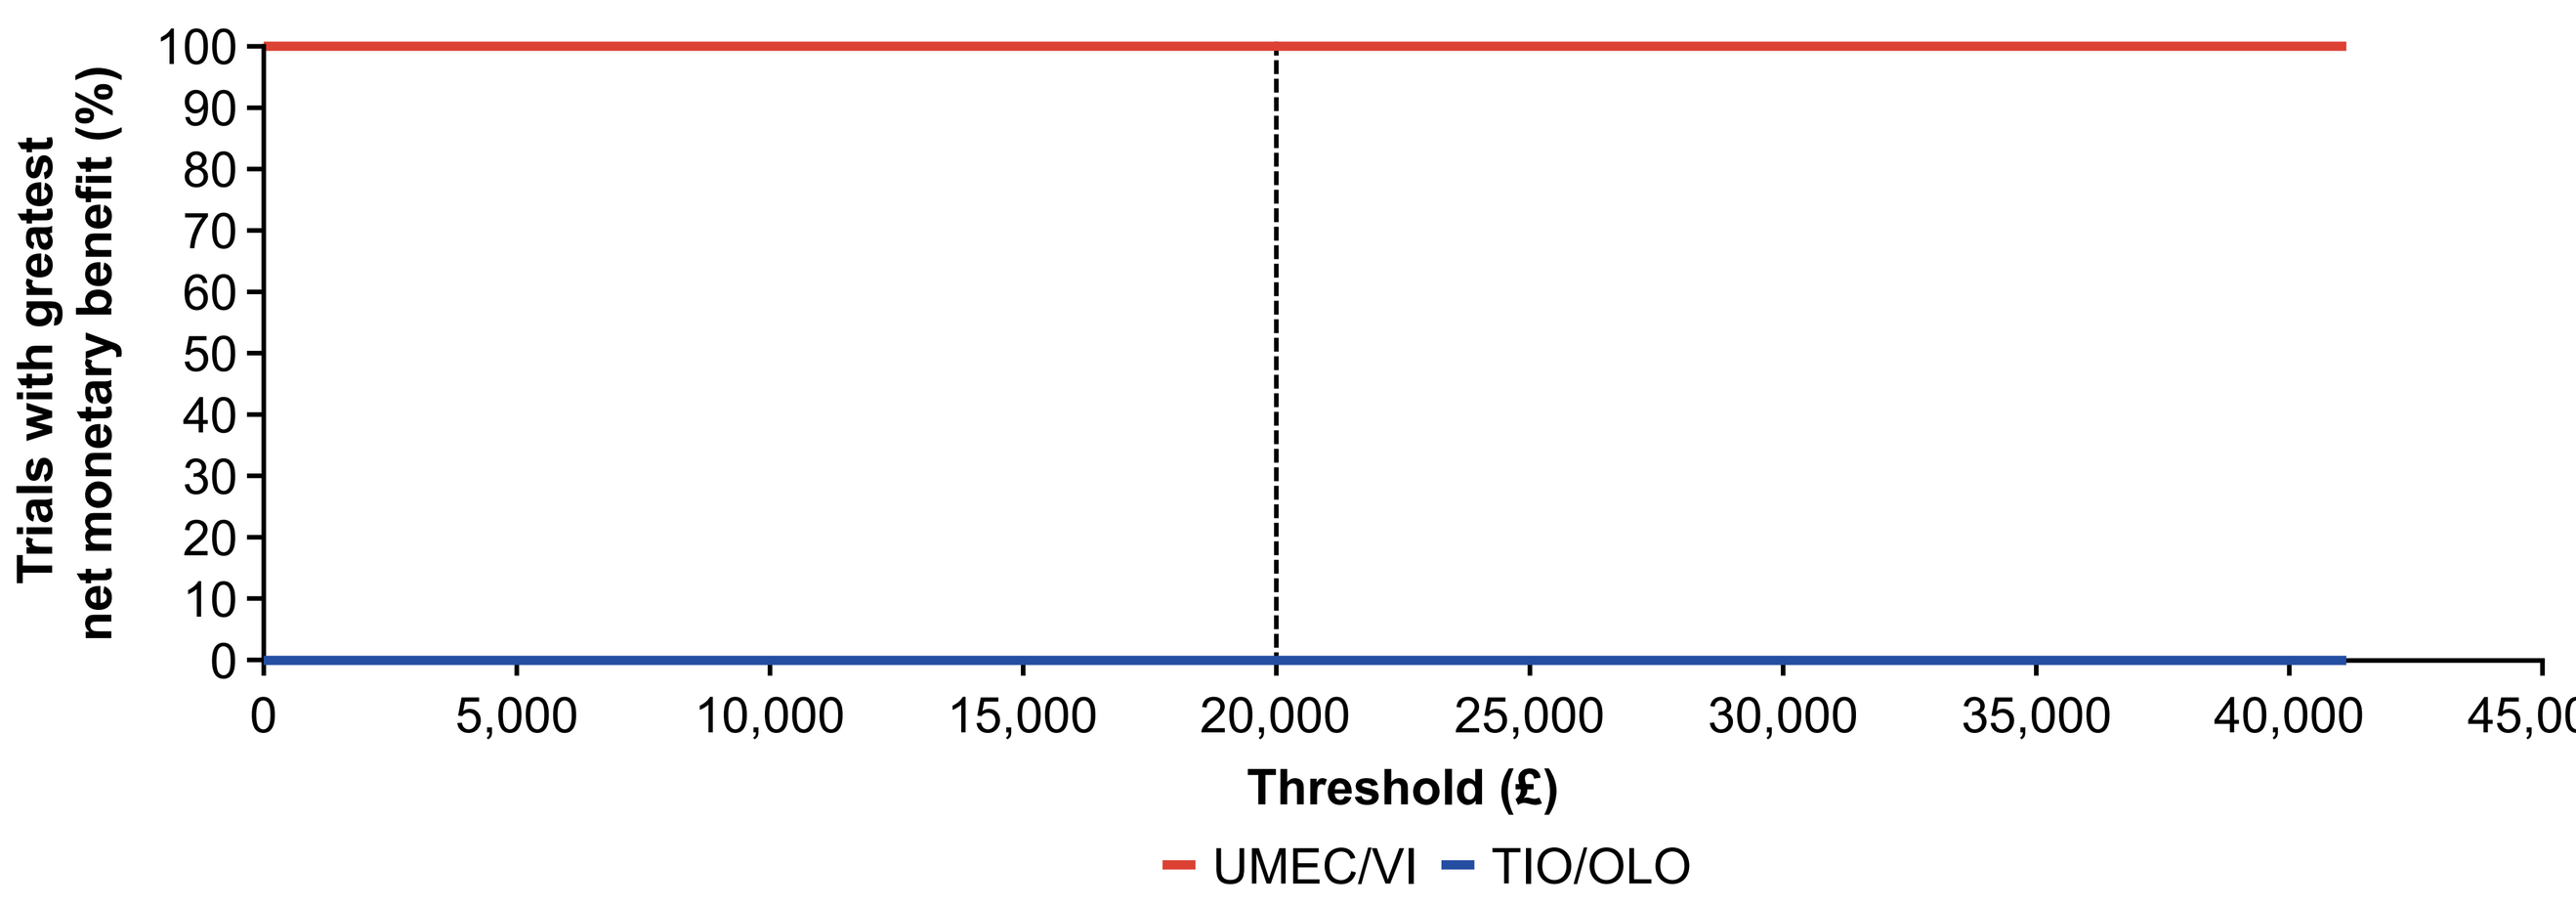
Trials with greatest net monetary benefit (%)
100
90
80
70
60
50
40
30
20
10
0
0
5,000
10,000
15,000
20,000
25,000
30,000
35,000
40,000
45,000
Threshold (£)
UMEC/VI
TIO/OLO

Scenario and sensitivity analyses

- UMEC/VI was the dominant (lower predicted costs and improved health outcomes) treatment for all scenarios and sensitivity analyses compared with TIO/OLO.
- Cost savings for UMEC/VI compared with TIO were highest in the scenario where treatment discontinuation data were applied in the first year and subsequent years, and lowest when RMU data were excluded.

Abbreviations

6MWT, 6-minute walk test; BMI, body mass index; BNF, British National Formulary; CI, confidence interval; COPD, chronic obstructive pulmonary disease; CV, cardiovascular; FEV_1, forced expiratory volume in one second; GBP, British pound sterling; GLY, glycopyrronium; ICER, incremental cost-effectiveness ratio; IND, indacaterol; LABA, long-acting β_2-agonist; LAMA, long-acting muscarinic antagonist; LY, life-year; mMRC, modified Medical Research Council dyspnoea scale; NHS, National Health Service; NICE, National Institute of Health and Care Excellence; NA, not applicable; NMA, network meta-analysis; OLO, olodaterol; PPPY, per patient per year; QALY, quality-adjusted life-year; RCT, randomised controlled trial; RMU, rescue medication use; SE, standard error; SGRQ, St George's Respiratory Questionnaire; TDI, transition dyspnoea index; TIO, tiotropium; UK, United Kingdom; UMEC, umeclidinium; VI, vilanterol.

References

1. Iheanacho I et al. *Int J Chron Obstruct Pulmon Dis*. 2020;15:439–60
2. National Institute for Health and Care Excellence (NICE). Chronic obstructive pulmonary disease in over 16s: diagnosis and management. 2019
3. Ismaila AS et al. *Adv Ther*. 2022;39(11):4961–5010
4. Briggs AH et al. *Med Decis Making*. 2017;37(4):469–80
5. Feldman GJ et al. *Adv Ther*. 2017;34:2518–33
6. National Institute for Health and Care Excellence (NICE). NICE health technology evaluations: the manual. 2022
7. Wedzicha JA et al. *Lancet Respir Med*. 2013;1(3):199–209
8. National Institute for Health and Care Excellence (NICE). British National Formulary. 2020
9. National Institute for Health and Care Excellence (NICE) Chronic obstructive disease in over 16s: diagnosis and management [H] Economic model report. 2018
10. Office of National Statistics (ONS). Consumer Price Index. 2022

Acknowledgements

Editorial support (in the form of writing assistance, including development of the initial draft based on author direction, assembling tables and figures, collating authors' comments, grammatical editing, and referencing) was provided by Christopher Heath, PhD, at Fishawack Indicia Ltd., UK, part of Avalere Health, and was funded by GSK.

Disclosures

This study was funded by GSK (Study 217635).

RK, **DS, PD** and **NAR** are employees of ICON and hold shares and stocks in ICON. **AM**, **SGN** and **ASI** are employees of GSK and/or hold stocks/shares in GSK. **SGN** is a PhD candidate at McMaster University, Hamilton, ON, Canada. **ASI** is also an unpaid faculty member at McMaster University, Hamilton, ON, Canada.

On behalf of all authors, this poster is presented by **AM**, an employee of GSK.

ISPOR Europe | 12–15 November 2023 | Copenhagen, Denmark

Presenting author: Alan Martin (alan.a.martin@gsk.com)