EE 128

Budget impact of an intrauterine vacuum-induced device for the treatment of patients with abnormal postpartum bleeding or hemorrhage in the United States

Candice Yong[1], Aimee Fox[2], Alex Hirst[2], Robert Hughes[2], Grace Mountain[2], Brian Seal[1], Leon Dupclay[1], Kara M. Rood[3]

[1]Organon & Co., Jersey City, NJ, USA, [2]Adelphi Values, UK, [3]The Ohio State University-Wexner Medical Center, Columbus, OH

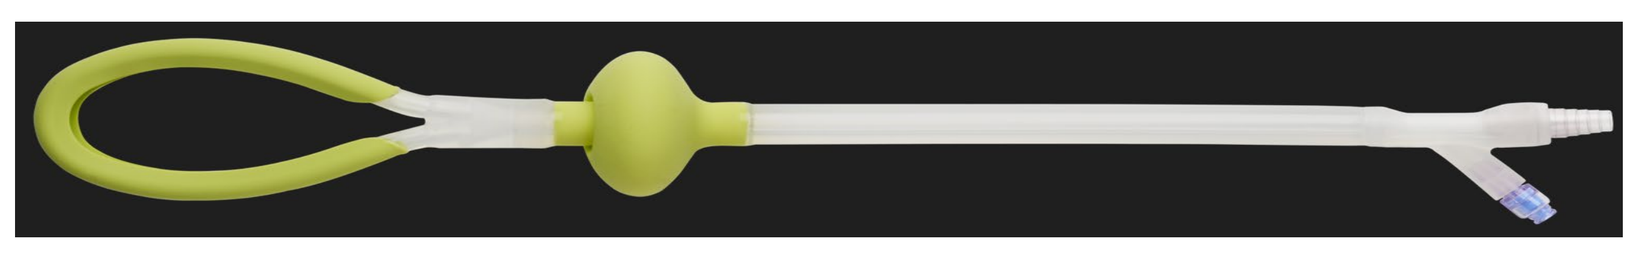

Key Takeaways

- **Introducing a novel intrauterine postpartum hemorrhage (PPH) control device for a hypothetical cohort of 10,000 births, where 1,470 patients experience abnormal postpartum bleeding or PPH, results in hospital cost-savings of $10,426,373 over a 1-year time horizon**
- **The device is associated with a lower overall budget impact due to savings in blood transfusion costs, hospital stay costs, and intensive care admission costs**

1. Background & Objective

- Postpartum hemorrhage (PPH) is one of the most common and dangerous complications associated with severe vaginal bleeding occurring after childbirth.
- It is a leading cause of mortality in childbirth, accounting for about 25% of maternal deaths worldwide.[1]
- With increasing rates of PPH, a novel intrauterine vacuum-induced hemorrhage control device (the Jada® System) has been developed for treatment.
- The budget impact of adding this device (herein referred to as intervention) as a treatment for PPH, compared to current practice was estimated.

3. Results

- The model estimated that 1,470 patients were eligible for treatment.
- Introducing a novel intrauterine vacuum-induced hemorrhage control (VIHC) device for treatment of PPH results in an overall budget impact of -$10,426,373, which is a 7.70% reduction in the budget over the 1-year time horizon.
- This budget impact was -$1,043 per birth and -$7,093 per eligible birth.

Budget impact per scenario	Year 1 total	Per Birth	Per eligible patient
Scenario without the intrauterine device available	$135,329,948	$13,533	$92,061
Scenario with the intrauterine device available	$124,903,575	$12,490	$84,968
Total budget impact	-$10,426,373	-$1,043	-$7,093

- The treatment strategy with the intrauterine VIHC device produced cost savings when considering blood transfusion costs, hospital stay costs, and intensive care admission cost, compared to the balloon tamponade treatment strategy.
- The scenario with the intrauterine VIHC device had lower health resource use compared to the balloon tamponade when considering intensive care admission, rate of hysterectomy, and major blood transfusions with a reduction of 5.6%, 1.8%, and 7.5%, respectively.

Budget impact per scenario per cost category	Scenario with the intrauterine device available	Scenario without the intrauterine device available	Budget impact
Direct treatment costs	$654,534	$295,444	$359,090
Blood transfusion costs	$1,303,486	$1,664,883	-$361,398
Hospital stay costs	$120,337,819	$129,493,084	-$9,155,266
Intensive care admission costs	$2,607,736	$3,876,537	-$1,268,801
Total costs	$124,903,575	$135,329,948	-$10,426,373

- The treatment strategy with the intrauterine VIHC device produced cost savings when considering device in-dwelling time with the time spent in either a delivery suite or a high-intensity suite, compared to the balloon tamponade treatment strategy.
- Cost savings were highest for the high-intensity suite.
- Results were comparable for vaginal delivery vs. cesarean delivery.

Cost impact of device in-dwelling time* (hours) per eligible patient	Scenario with the intrauterine device available	Scenario without the intrauterine device available	Cost difference
Vaginal delivery			
Delivery suite	$187	$1,083	-$896
High-intensity suite	$336	$1,953	-$1,617
Cesarean delivery			
Delivery suite	$277	$1,083	-$806
High-intensity suite	$499	$1,953	-$1,454

*In-dwelling time based on RUBY study for the intrauterine device (3.1 hours for vaginal, 4.6 hours for cesarean), and 18 hours for balloon tamponade.

2. Study design

Model overview:
- A budget impact model (BIM) with a 1-year time horizon was developed to compare the costs of treating PPH patients from the hospital perspective, based on a hypothetical cohort of 10,000 births.
- Two alternate treatment strategies were defined:

1. **Current practice**: without the intervention (treatment with uterotonics and balloon tamponade)
2. **New practice**: with the intervention (treatment with uterotonics and intrauterine VIHC device)

- Patients received uterotonic drugs +/- nonsurgical treatments (balloon tamponade or the intervention), and could progress to subsequent surgical procedures, followed by potential hysterectomy.
- The patient population eligible for treatment with the intervention was estimated using epidemiological data from real-world United States (US) Premier hospital database (2016-2022).
- Probabilities of treatment progression and healthcare resource use (HRU) were sourced from clinical trial data and published literature.
- Cost inputs were estimated from Premier hospital database (2016-2022).
- Model inputs differed by method of delivery; vaginal or caesarean-section.[2,3,4,5]

Model outputs:
- Overall budget impact of introducing the intervention to the market.
- Budget impact per birth and eligible patient.
- Cost difference in device in-dwelling time per birth.
- Budget impact per scenario per cost category.

5. Strengths and Limitations

Strengths:
- The model flexibility alongside the clear and transparent structure allow robust testing of the model results.
- Model development was supported by seeking clinical expert advice to ensure it reflected clinical practice.
- The model captured treatment strategies, which more accurately reflects the potential cost offsets available from the introduction of the novel intrauterine vacuum-induced hemorrhage control device.

Limitations:
- It is assumed that all treatment effects and costs are incurred within the calendar year each patient enters the model.
- All treatment failures result in progression to the next treatment line.
- The market shares for the subsequent treatments were sourced from the Premier analysis but re-weighted to sum to 100%. This was required for the functionality of the model.
- Resource costs associated with each treatment option are cumulative.

6. Conclusion

- The device offers a valuable treatment option for patients with PPH, despite higher acquisition cost compared to the balloon tamponade.
- The device provides cost savings compared to the balloon tamponade due to shorter in-dwelling time and reduced health resource use, including blood transfusions and intensive care admissions.

References

1. Carroli G, Cuesta C, Abalos E, Gulmezoglu AM. Epidemiology of postpartum haemorrhage: a systematic review. Best Practice & Research Clinical Obstetrics & Gynaecology. 2008;22(6):999-1012.; 2. Suarez S C-AA, Borovac-Pinheiro A, et al. Uterine balloon tamponade for the treatment of postpartum hemorrhage: a systematic review and meta-analysis. Am J Obstet Gynecol. 2020;222(4): e1-293.e252.; 3. Organon PREMIER data analysis; 4. Goffman D, et al. Obstet Gynecol, 2023 Nov 1;142(5):1006-16.; 5. Obstetrics & Gynecology. Practice Bulletin No. 183 Summary: Postpartum Hemorrhage. Obstetrics & Gynecology. 2017;130(4):923-925

Disclosures & Funding Statement

- C Yong, B Seal and L Dupclay: Organon employee and stock/shareholder; A Fox, A Hirst, R Hughes and G Mountain are employees of Adelphi Values; K Rood: RUBY Investigator; Advisory Board participation (Cooper Surgical); Advisory Board participation, paid consultant (Organon)
- This study was funded by Organon & Co.

© 2023 Organon group of companies. All rights reserved.