CO170

Treatment Exposure-Adjusted Event Rates (EAERs) for Grade 3/4 AEs Associated With Emerging and Existing Systemic Therapies for mCRC With at Least 2 Prior Lines of Therapy: Informing Payer and Pathway Formulary Decision Making

Howe A[1], Hernandez LG[1], Paly VF[1], Eng C[2], Dasari A[3], Samuel L[4], Kasper S[5], Tougeron D[6]

[1]Takeda Pharmaceuticals America, Inc., Lexington, MA, USA; [2]Division Hematology and Oncology, Vanderbilt-Ingram Cancer Center, Nashville, TN, USA; [3]Department of Gastrointestinal Medical Oncology, The University of Texas MD Anderson Cancer Center, Houston, TX, USA; [4]Aberdeen Royal Infirmary, Aberdeen, UK; [5]West German Cancer Center, Department of Medical Oncology, University Hospital Essen, Essen, Germany; [6]Department of Hepato-Gastroenterology, Poitiers University Hospital and University of Poitiers, Poitiers, France

Question

Which grade 3/4 AEs are associated with fruquintinib, regorafenib, T/T, and T/T + bev treatment for mCRC and how might this inform payer formulary and pathway decisions?

Study design

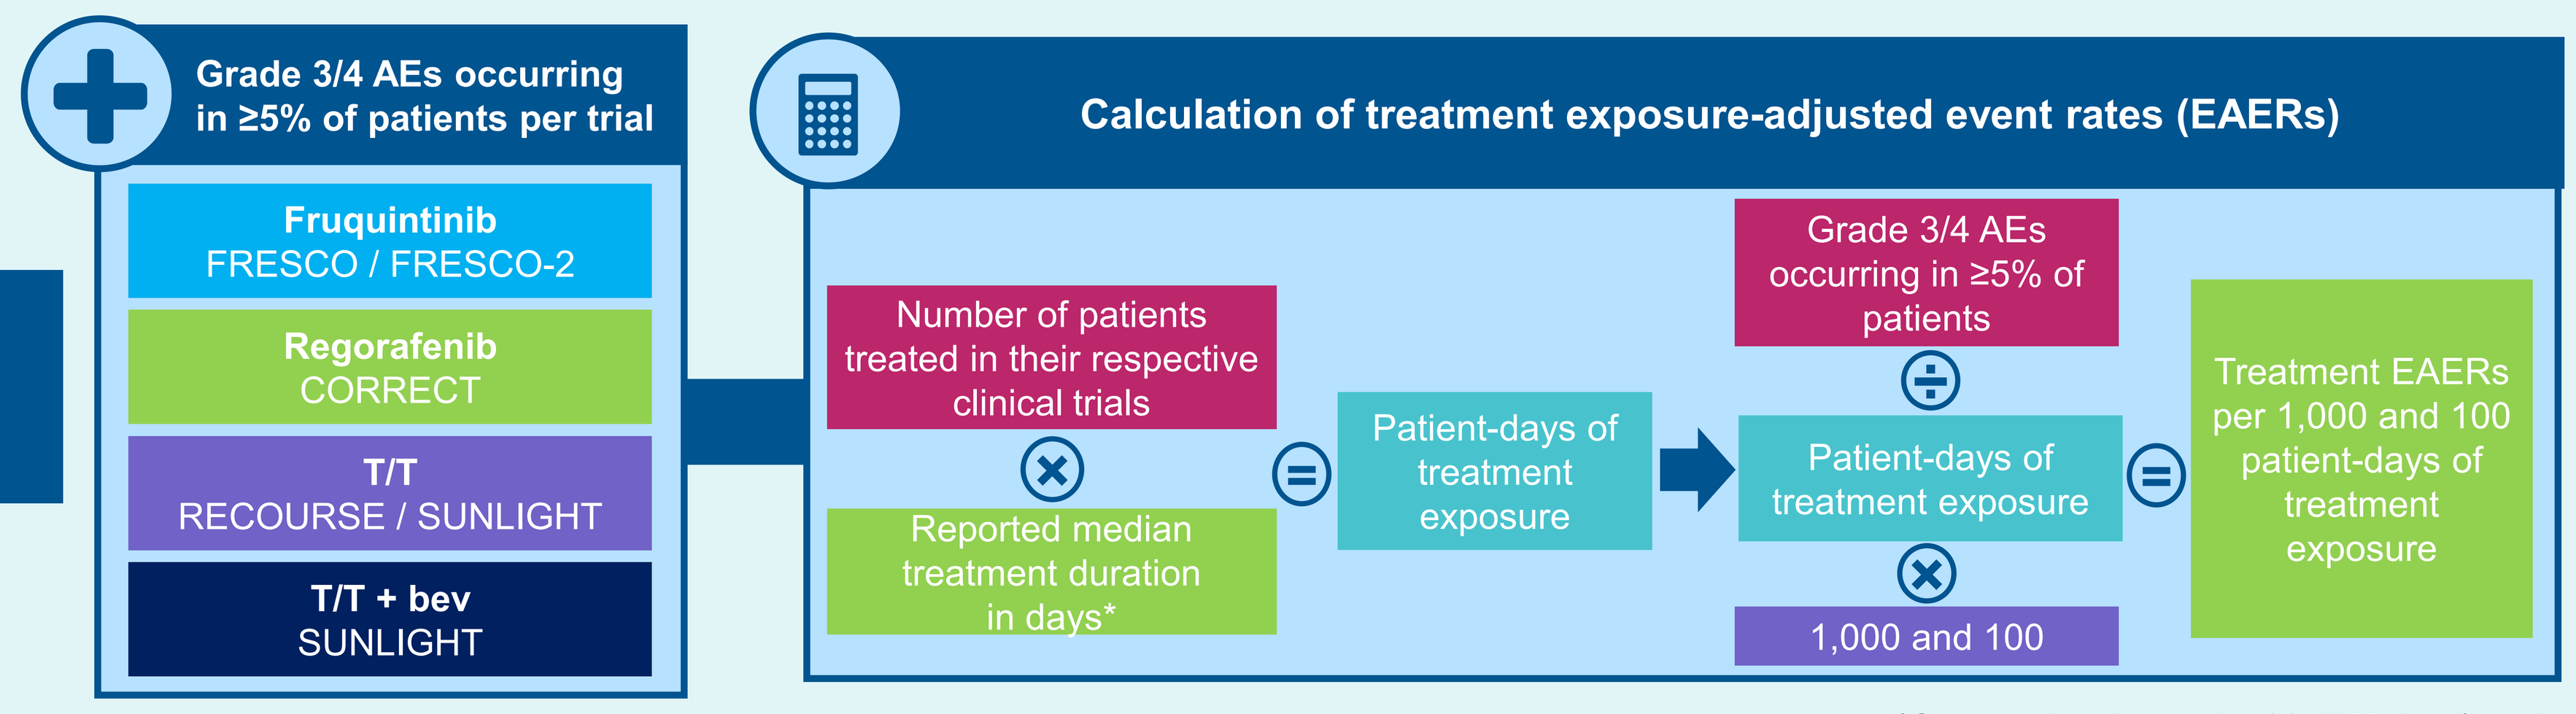

Grade 3/4 AEs occurring in ≥5% of patients per trial
Fruquintinib FRESCO / FRESCO-2
Regorafenib CORRECT
T/T RECOURSE / SUNLIGHT
T/T + bev SUNLIGHT
Calculation of treatment exposure-adjusted event rates (EAERs)
Number of patients treated in their respective clinical trials
Reported median treatment duration in days*
Patient-days of treatment exposure
Grade 3/4 AEs occurring in ≥5% of patients
Patient-days of treatment exposure
1,000 and 100
Treatment EAERs per 1,000 and 100 patient-days of treatment exposure

*Converted to days using 30.4375 days/month.

Key take aways

- **Fruquintinib demonstrates the lowest EAERS of grade 3/4 AEs occurring in ≥5% of patients treated with fruquintinib, regorafenib, T/T, or T/T + bev**
- **This outcome may be considered by population health and healthcare plan decision makers as a value measure when reviewing formulary and pathway choices of emerging and existing systemic therapies for patients with mCRC**

Results

Grade 3/4 treatment EAERs per 1,000 patient-days occurring in ≥5% of patients treated with fruquintinib, regorafenib, T/T, or T/T + bev

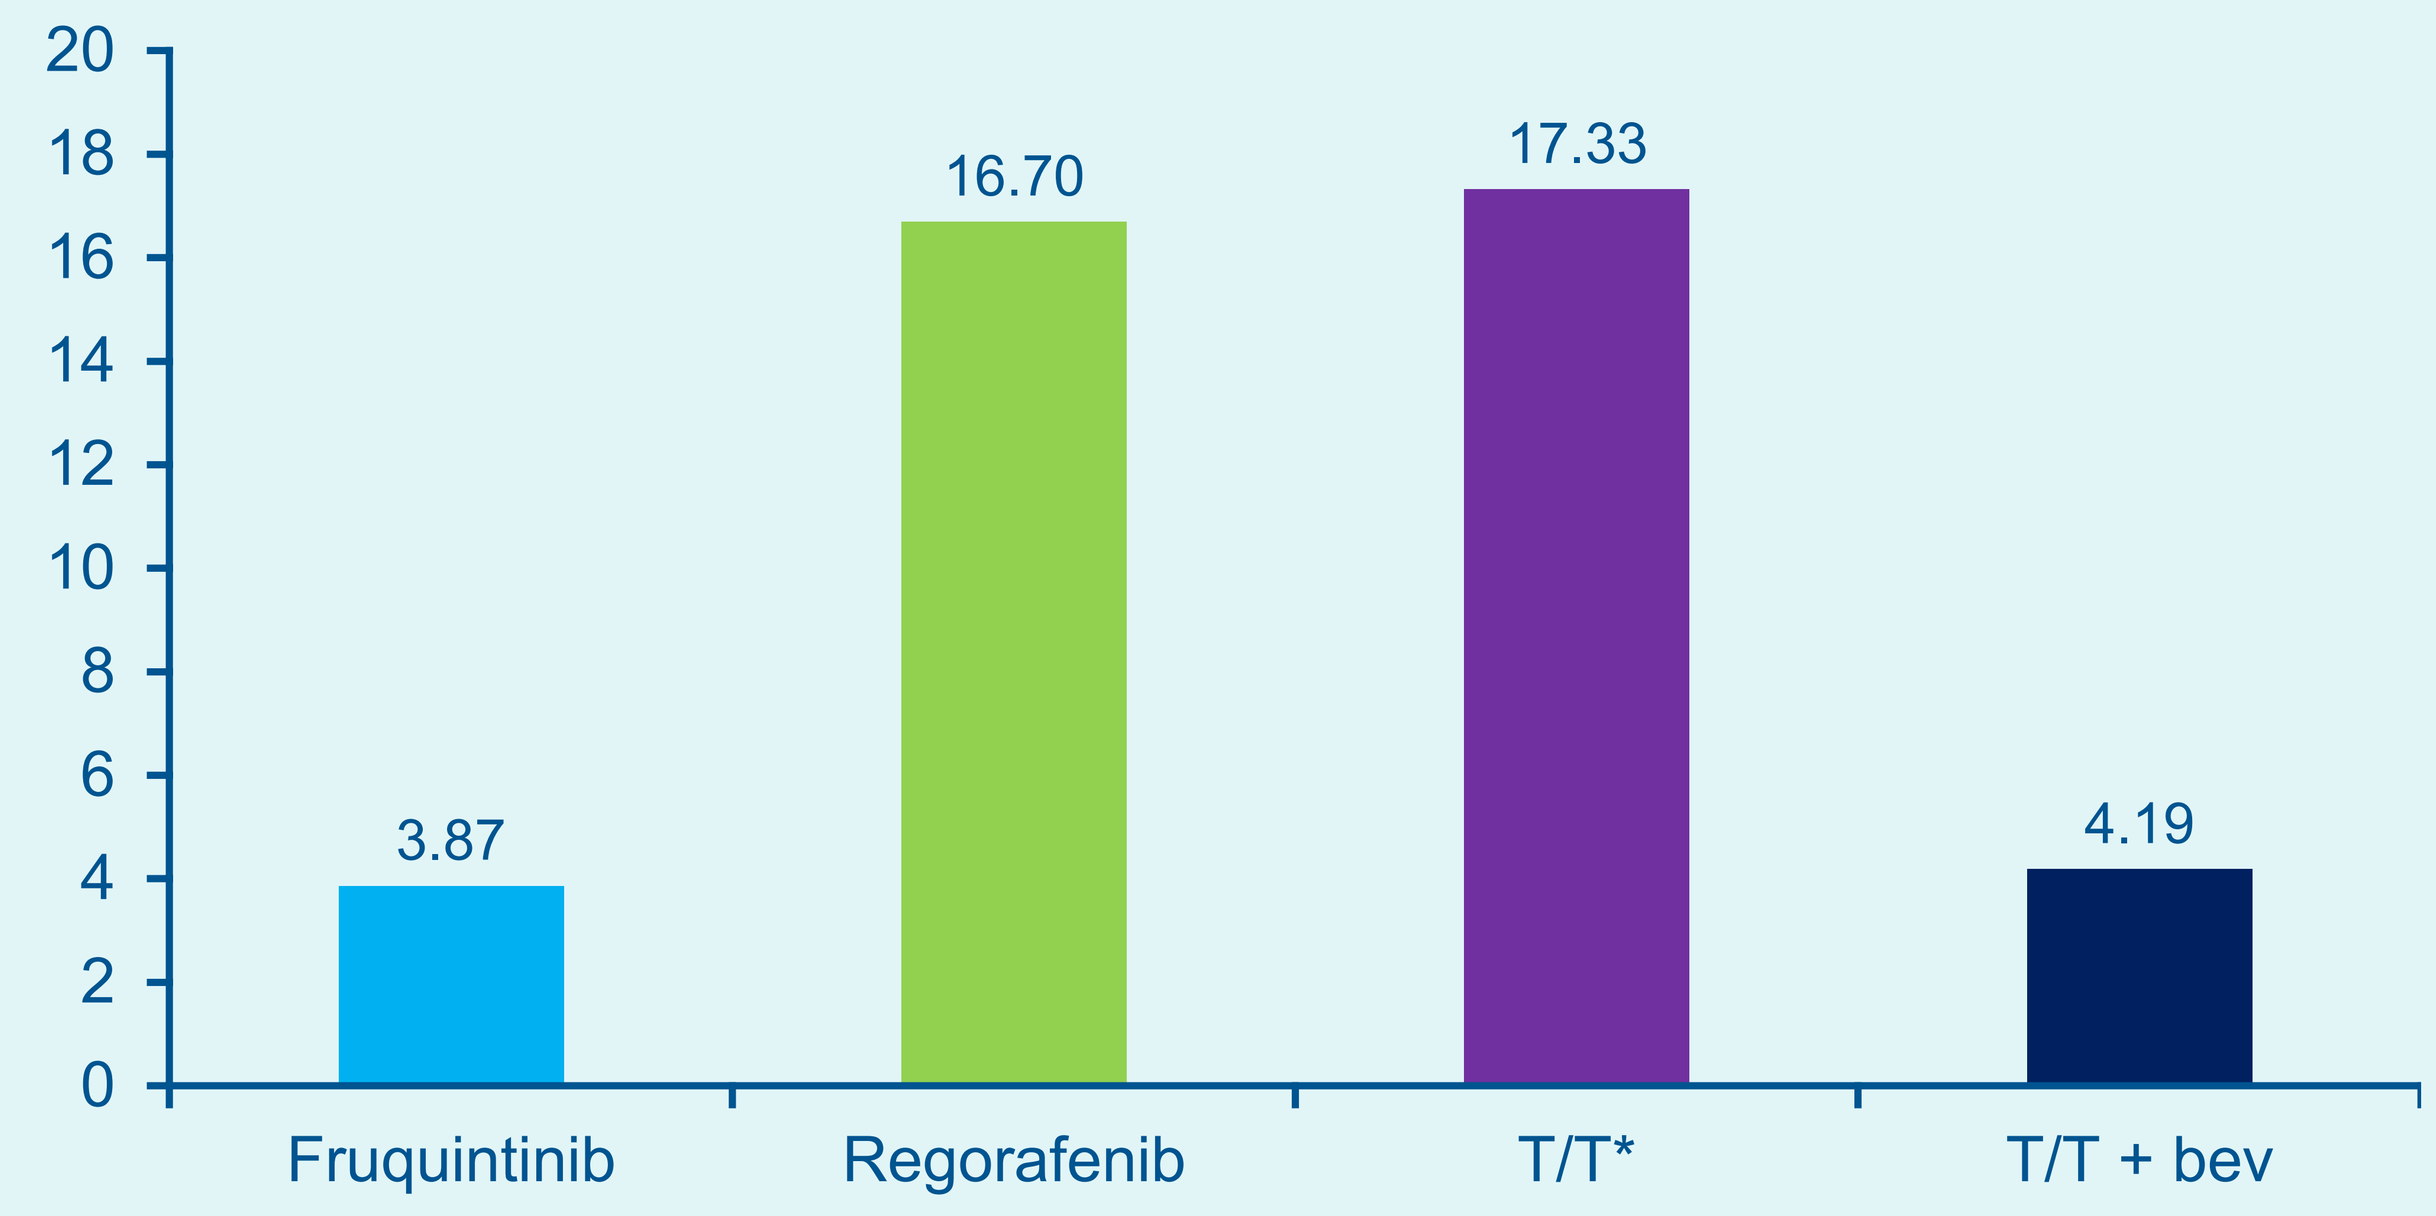

20
18
16
14
12
10
8
6
4
2
0
3.87
16.70
17.33
4.19
Fruquintinib
Regorafenib
T/T*
T/T + bev

*Treatment EAERs per 1,000 patient-days were 24.44 based on the RECOURSE trial alone and 8.71 based on the SUNLIGHT trial alone.

INTRODUCTION

- Colorectal cancer (CRC) is the third-most common cancer type globally and has the second-highest mortality rate of all cancer types[1]
- CRC often presents at an advanced stage, with ~20% to ~30% of patients having metastatic disease (mCRC) at diagnosis, and up to half of patients with localized CRC eventually develop metastases[2–8]
- Systemic therapies such as fruquintinib, regorafenib, and trifluridine/tipiracil (T/T) have demonstrated improvements in survival versus best supportive care (BSC), as well as T/T + bevacizumab (T/T + bev) versus T/T, in the treatment of patients with mCRC who have been previously treated with oxaliplatin- and irinotecan-based chemotherapy[9–13]
- While some relative efficacy data are available for these therapies, there is limited evidence on their relative safety profiles

OBJECTIVE

- This study characterizes the rates of grade 3/4 AEs occurring in ≥5% of patients while receiving treatment with fruquintinib, regorafenib, T/T, or T/T + bev for mCRC previously treated with oxaliplatin- and irinotecan-based chemotherapy, to inform payer formulary and pathway decisions

METHODS

- Treatment exposure-adjusted event rates (EAERs) were used to characterize the rates of grade 3/4 adverse events (AEs) occurring in ≥5% of patients while receiving treatment
- The number of patient-days of treatment exposure was calculated for each treatment by multiplying the number of patients treated in their respective clinical trials by the reported median treatment duration converted to days (using 30.4375 days in a month) (**Table 1**)
 – For fruquintinib, the number of patients treated was calculated as the sum of the number patients from FRESCO (278) and FRESCO-2 (456), and the median treatment duration in months as the average of the median treatment duration from FRESCO (3.68 months) and FRESCO-2 (3.06 months)[11,13]
 – For T/T, the number of patients treated was calculated as the sum of the number patients from RECOURSE (533) and SUNLIGHT (246), and the median treatment duration in months as the average of the median treatment duration from RECOURSE (1.54 months) and SUNLIGHT (2.10 months)[10,12]
- The number of grade 3/4 AEs occurring in ≥5% of patients for each treatment were from their respective clinical trials;[9-13] for fruquintinib and T/T, number of AEs reported is based on the pooled data from FRESCO and FRESCO-2,[11,13] and from RECOURSE and SUNLIGHT[10,12]
- Treatment EAERs were calculated by dividing the number of grade 3/4 AEs occurring in ≥5% of patients by the patient-days of treatment exposure, and multiplied by 1,000 and 100 to calculate treatment EAERs per 1,000 and 100 patient-days of treatment exposure, respectively

Table 1. Number of patients treated, median treatment duration, and treatment exposure with fruquintinib, regorafenib, T/T, and T/T + bev

	Fruquintinib (Pooled FRESCO and FRESCO-2)[11,13]	Regorafenib (CORRECT)[9]	T/T (Pooled RECOURSE and SUNLIGHT)[10,12]	T/T + bev (SUNLIGHT)[12]
Number of treated patients in clinical trial*	734	500	779	246
Median duration of treatment (months)†	3.37	1.70	1.82	5.00
Median duration of treatment (days)‡	102.57	51.74	55.41	152.19
Patient-days of treatment exposure‡	75,290	25,872	43,164	37,438

*For fruquintinib, calculated as the sum of the number of patients from FRESCO (278) and FRESCO-2 (456). For T/T, calculated as the sum of the number of patients from RECOURSE (533) and SUNLIGHT (246).
†For fruquintinib, calculated as the average of the median treatment duration from FRESCO (3.68 months) and FRESCO-2 (3.06 months). For T/T calculated as the average of the median treatment duration from RECOURSE (1.54 months) and SUNLIGHT (2.10 months).
‡Calculated by multiplying the number of patients treated by the median treatment duration in months converted to days (using 30.4375 days in a month).

RESULTS

- Grade 3/4 AEs occurring in ≥5% for any treatment included: anemia, asthenia, diarrhea, fatigue, hand foot syndrome/palmar-plantar erythrodysesthesia, hypertension, leukopenia, neutropenia, rash, thrombocytopenia, and laboratory abnormalities (increased alanine aminotransferase, alkaline phosphatase, aspartate aminotransferase, and total bilirubin) (**Table 2**)
- EAERs per 1,000 patient-days of treatment exposure were estimated as 3.87 for fruquintinib, 17.33 for T/T, 16.70 for regorafenib, and 4.19 for T/T + bev (**Table 3**)
- EAERs per 100 patient-days treatment exposure were estimated as 0.387 for fruquintinib, 1.733 for T/T, 1.670 for regorafenib, and 0.419 for T/T + bev (**Table 3**)
- Treatment EAERs were higher with T/T compared with T/T + bev due a higher rate of TEAEs with T/T in RECOURSE and SUNLIGHT compared with T/T + bev in SUNLIGHT
- However, RECOURSE and SUNLIGHT were conducted and published at least 8 years apart; thus, before the SUNLIGHT trial T/T had a history already in real-world practice with potential better management of T/T and its associated AEs
- A scenario analysis was conducted to characterize the treatment EAERs for T/T based on RECOURSE and SUNLIGHT separately, with the following respective results:
 – Number of grade 3/4 AEs occurring in ≥5% of patients were 611 and 137 in RECOURSE and SUNLIGHT, respectively;
 – Patient-days of treatment exposure were 24,998 and 15,724;
 – Treatment EAERs per 1,000 patient-days were 24.44 and 8.71; and
 – Treatment EAERs per 100 patient-days were 2.444 and 0.871

Table 2. Number of grade 3/4 AEs occurring in ≥5% of patients treated with fruquintinib, regorafenib, T/T, and T/T + bev

	Fruquintinib (Pooled FRESCO and FRESCO-2)[11,13]	Regorafenib (CORRECT)[9]	T/T (Pooled RECOURSE and SUNLIGHT)[10,12]	T/T + bev (SUNLIGHT)[12]
Anemia		29	123	15
Asthenia	35		28	10
Diarrhea	24	41	22	2
Fatigue	21	77	30	3
Hand foot syndrome/palmar-plantar erythrodysesthesia	59	83		
Hypertension	121	38	3	14
Increased alanine aminotransferase	16	27	10	
Increased alkaline phosphatase			42	
Increased aspartate aminotransferase	11	29	23	
Increased total bilirubin	4	60	45	
Leukopenia			113	
Neutropenia			279	106
Rash		29		
Thrombocytopenia		19	30	7
Total	**291**	**432**	**748**	**157**

Table 3. Treatment EAERs for grade 3/4 AEs occurring in ≥5% of patients treated with fruquintinib, regorafenib, T/T, or T/T + bev

	Fruquintinib	Regorafenib	T/T	T/T + bev
Treatment EAERs per 1,000 patient-days*	3.87	16.70	17.33	4.19
Treatment EAERs per 100 patient-days†	0.387	1.670	1.733	0.419

*Calculated by dividing the number of number of grade 3/4 AEs occurring in ≥5% by the patient-days of treatment exposure, multiplied by 1,000.
†Calculated by dividing the number of number of grade 3/4 AEs occurring in ≥5% by the patient-days of treatment exposure, multiplied by 100.

LIMITATIONS

- These analyses were based on AE rates reported in randomized clinical trials, which may differ from rates and/or number of events per patient that may occur in real clinical practice

CONCLUSIONS

- Based on the AE rates reported in randomized clinical trials, patients with mCRC previously treated with oxaliplatin- and irinotecan-based chemotherapy experienced lower treatment EAERs associated with grade 3/4 AEs occurring in ≥5% of patients when treated with fruquintinib versus other systemic treatment options
- Population health and healthcare plan decision makers may consider the use of the treatment EAERs of this analysis as a value measure when reviewing formulary and pathway choices of emerging and existing systemic therapies for patients with mCRC
- Future research based on real-world evidence should look into the impact of the sequence of these treatments on efficacy and toxicity

References

1. Shin AE, et al. *Trends Pharmacol Sci*. 2023;44:222–36.
2. Ciardiello F, et al. *CA Cancer J Clin*. 2022;72:372–401.
3. National Cancer Institute. Cancer Stat Facts: Colorectal Cancer. 2022; https://seer.cancer.gov/statfacts/html/colorect.html. Published 2022. Accessed May 5, 2023.
4. Centers for Disease Control and Prevention. United States Cancer Statistics: Data Visualizations Stage at Diagnosis. 2022; https://gis.cdc.gov/Cancer/USCS/#/StageatDiagnosis/. Published 2022. Accessed May 5, 2023.
5. Chen Y, et al. *Support Care Cancer*. 2010;18:217–24.
6. Siegel RL, et al. *CA Cancer J Clin*. 2020;70:145–64.
7. Bouvier AM, et al. *Dig Liver Dis*. 2021;53:639–45.
8. Tamakoshi A, et al. J Epidemiol. 2017;27:S36–42.
9. Grothey A, et al. *Lancet*. 2013;381:303–12.
10. Mayer RJ, et al. *N Engl J Med*. 2015;372:1909–19.
11. Li J, et al. *JAMA*. 2018;319:2486–96.
12. Prager GW, et al. *N Engl J Med*. 2023;388:1657–67.
13. Dasari A, et al. *Lancet*. 2023;402:41–53.

Acknowledgments

This study was funded by Takeda Pharmaceuticals America, Inc. Lexington, MA, USA. Editorial assistance was provided by Leslee Masten of Evidera, a part of Thermo Fisher Scientific.

Disclosures

AH, LGH, VFP: employment and stock ownership (Takeda). **CE**: Consulting: Amgen, Bayer, Ca Institute of Regenerative Medicine, Elevation, GE, GSK, IGM, Merck, Natera, Pfizer, Taiho, Seagen. Financing of Scientific Research To Vanderbilt: Merck, Hutchi-Med, Pfizer, Janssen, Gritstone. **AD**: Institutional, Research Grant: Hutchmed, Guardant Health, Natera Inc, Eisai, Crinetics, AAA / Novartis. Financial Interests, Personal, Advisory Board: Hutchmed, Crinetics, Voluntis, AAA / Novartis, Personalis Inc. **LS**: Institutional: ARCUS, GSK, Mirati, MSD, Novartis. Advisory Board: GSK, SEAGEN & Takeda. **SK**: Personal and institutional grants or contracts: Bristol Myers Squibb, Lilly, and Roche. Payment or honoraria for lectures, presentations, speakers' bureaus, manuscript writing, or educational events from Bristol Myers Squibb, Amgen, Servier, Lilly, Merck Serono, Roche, MSD, and Pierre Fabre. Consulting fees: Novartis and Incyte. Support for attending meetings or travel: Pierre Fabre, Lilly, Amgen, Bristol Myers Squibb, Roche, and Merch Serono. Participation on a data safety monitoring board or advisory board: Novartis. **DT**: Consulting and/or advisory board: Amgen, Roche, Servier, MSD, BMS, Pierre Fabre, Astra Zeneca, Bayer, and Takeda.

To view an electronic version of this poster, scan this Quick Response (QR) code or visit: https://tiny.one/1XL1275Krz

Copies of this poster obtained through QR code are for personal use only and may not be reproduced without permission from the congress and the author of this poster.

Presented at ISPOR Europe 2023, Copenhagen, Denmark, 12–15 November 2023. For questions or comments, please contact Dr Luis Hernandez: luis.hernandez3@takeda.com.