Treatment effect waning in immuno-oncology health technology assessments: A review of assumptions and supporting evidence

HTA237

Taylor K,[1] Latimer N,[1,2] Hatswell A,[1] Douglas T,[1] Ho S,[3] Okorogheye G,[3] Borrill J,[3] Chen C,[4] Kim I,[4] Bertwistle D [3]

[1] Delta Hat Ltd, Nottingham, UK; [2] University of Sheffield, Sheffield, UK; [3] Bristol Myers Squibb, Uxbridge, London, UK; [4] Bristol Myers Squibb, Lawrenceville, NJ, USA

What we found

- A variety of methods have been used to apply treatment effect waning (TEW) in health technology assessment (HTA)
- Independent survival models are consistently used, but the extent to which these build waning into the model is not usually considered
- Clinical trials with long-term follow-up seldom provide hazard plots to show how the likelihood of an event may change over time
- The application of TEW in HTA could be improved by following the sequence of steps included in the proposed TEW algorithm

BACKGROUND

- Immuno-oncology therapies (IOs) are a type of biological therapy used to treat cancer by harnessing the body's own immune system to recognize and fight cancer cells.
- Based on their immunological mode of action, it has been suggested that IOs may provide a durable treatment effect for some patients long after treatment has been discontinued.
- Treatment effect waning (TEW) refers to a phenomenon whereby the effects of a treatment attenuate over time.
- Explicit assumptions around the maintenance over time of the treatment effect associated with IOs have been a source of contention in health technology assessment (HTA).
- TEW is commonly incorporated as part of analyses for economic evaluations by HTA agencies such as the National Institute for Health and Care Excellence (NICE), however there is currently no guidance on how TEW should be applied.
- Evidence on the long-term maintenance of IO treatment effects beyond treatment discontinuation has not been comprehensively summarised, leading to greater uncertainty in TEW analyses.

OBJECTIVE

The objective of this study was to provide a clear description of how TEW has been addressed in HTA. Additionally, it aimed to offer an overview of the evidence currently available on the long-term maintenance of IO treatment effects post treatment discontinuation, which could help inform future TEW analyses.

Figure 1. An overview of this multi-faceted review.

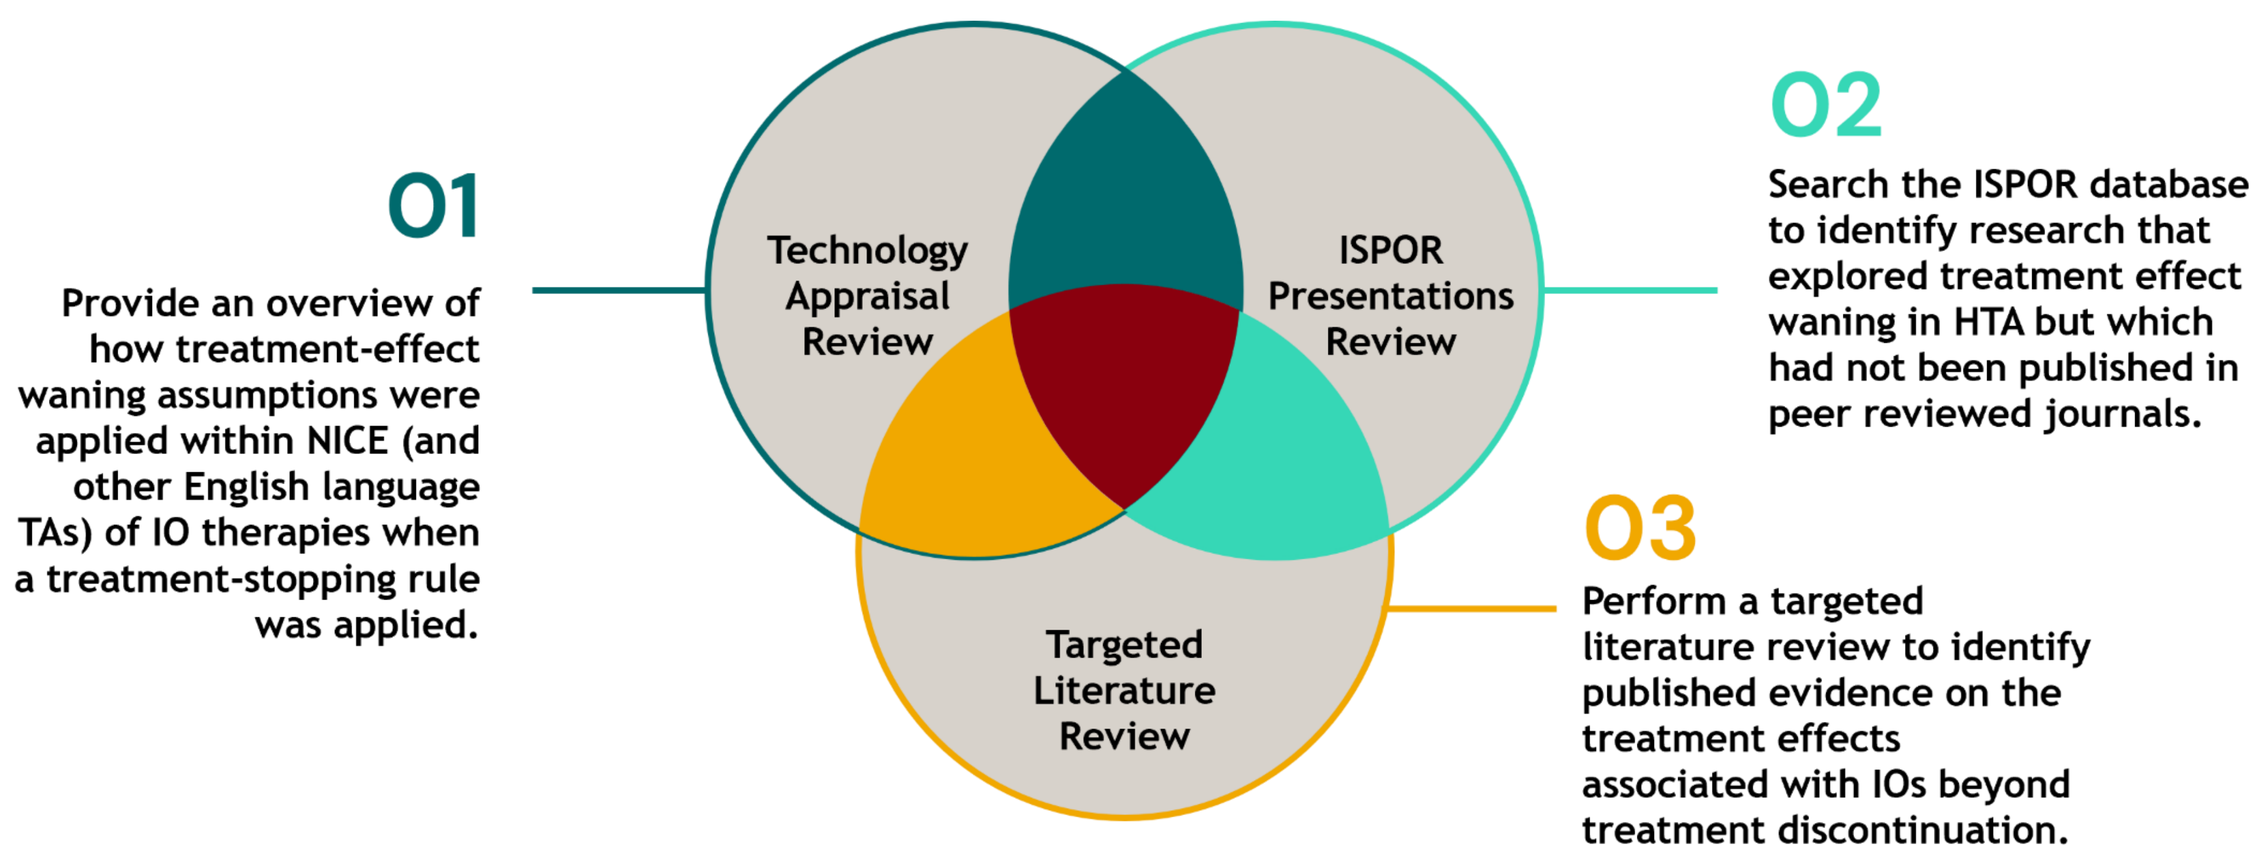
O1
Provide an overview of how treatment-effect waning assumptions were applied within NICE (and other English language TAs) of IO therapies when a treatment-stopping rule was applied.
Technology Appraisal Review
ISPOR Presentations Review
Targeted Literature Review
O2
Search the ISPOR database to identify research that explored treatment effect waning in HTA but which had not been published in peer reviewed journals.
O3
Perform a targeted literature review to identify published evidence on the treatment effects associated with IOs beyond treatment discontinuation.

METHODS

Information was reviewed from a range of different, complementary data sources (summarised in **Figure 1**).

Technology appraisal review

- A review of NICE appraisals was conducted, updating a previous review conducted for Bristol-Myers Squibb (BMS) [4].
- NICE technology appraisals (TA)s published up until September 2022 were included if they met the inclusion criteria:
 - The presence of a treatment stopping rule for the IO treatment under investigation
 - Specific assumptions around TEW
- For included TAs, all submission documents were reviewed including final appraisal determinations, committee papers and committee paper slides with relevant information extracted
- As well as NICE, the following HTA agencies were also searched:
 - Scottish Medicines Consortium (SMC)
 - All Wales Medicines Strategy Group (AWMSG)
 - Canadian Agency for Drugs and Technologies in Health (CADTH)
 - The Pharmaceutical Benefits Advisory Committee (PBAC, Australia)
 - Pharmac (New Zealand)
 - The Institute for Clinical and Economic Review (Inst-CER, USA)

Searching the ISPOR scientific presentations database

- A search strategy was generated and presentations that discussed, reviewed, or analysed the relationship between treatment discontinuation and continued survival benefit and/or treatment effect were included, as well as any presentations that discussed, reviewed, or analysed methods used to address TEW in HTA appraisals.
- Relevant presentations published up until ISPOR Europe 2022 were included.

Targeted Literature Review (TLR)

- A search strategy was devised and PubMed was searched on 19 April 2023 to identify published papers from 2010 onwards that specifically analysed or reviewed treatment effects post-treatment discontinuation for IO treatments.

RESULTS

Technology appraisal review

- In total, 59 NICE TAs in IO indications were identified. Of these, 34 included discussion of TEW. After screening for IO agent stopping rules, 19 NICE TAs were included, with publication dates between January 2017 and September 2022 (**Table 1**). 14 were recommended by NICE, 3 were not recommended and 2 were entered into the Cancer Drugs Fund (CDF).
- In 10 of the 19 TAs, TEW was not applied in the company's base-case, implying that a lifetime treatment effect may have been assumed. In these instances, the external assessment group (EAG) and/or the appraisal committee often deemed scenario analyses that included TEW to be more appropriate for decision making.
- Various methods for applying TEW were used across the 19 TAs reviewed, with different assumptions applied. These included:
 1. **Independently fitted survival models -** the ratio of the hazards of the survival models fitted to each treatment arm illustrating a diminishing treatment effect
 2. **Immediate waning -** setting the hazard ratio (HR) to 1 at a specific time point
 3. **Gradual waning -** assumes that the HR for the treatment group relative to the comparator group changes over time, reflecting a gradual decrease in the treatment effect
 4. **Waning incorporating external real-world data (RWD) -** estimating hazard rates using RWD and applying these to treatment arms
 5. **Conditional waning -** assuming waning in only those patients who did not achieve disease control
 6. **Waning involving cure assumptions -** assuming a fixed cure point
- Immediate TEW was used the most throughout the 19 TAs, despite clinical input in 2 appraisals (TA737 and TA683) highlighting that gradual TEW would be more clinically plausible. All the TAs that applied TEW in their base-case used a form of "immediate" TEW. 7 TAs explored gradual TEW within scenario analyses.
- Independent survival models were used in all but one of the TAs and hence constant treatment effects were not assumed in company base case models. However, modelled treatment effects were often not illustrated. Therefore, it was not possible to determine to what extent TEW was already accounted for in the base case model. This is important given that TEW assumptions are generally made when it is thought that treatment effects will attenuate.
- When searching the other English-language HTA agencies, there were several instances where TEW approaches differed between HTA agencies, but this is not surprising given the lack of official guidance around the subject and the different preferences from EAGs and committees. One difference found was the substantially shorter time-horizons used for PBAC (Australia) appraisals.

Searching the ISPOR scientific presentations database

- There were 11 ISPOR presentations included. Two studies investigated the accuracy of different TEW methods used in past NICE appraisals using later data cuts of published data. Conclusions were similar in that gradually equalising hazards of death (gradual waning, described above) demonstrated improved predictive accuracy vs immediate TEW [1,2].
- One review uncovered inconsistencies in assumptions made by companies, ERGs, and NICE ACs around TEW, and highlights the need for further guidance for consistent incorporation of TEW methods in HTA submissions [3].
- Kamgar et al. [4] created smooth HR plots based on pseudo-individual patient level data. The plots illustrate the ratio of hazards between treatment groups, demonstrating whether the treatment effect remains approximately the same over time, or whether the effect appears to increase or decrease. This work demonstrates the plausibility of presenting analyses that are informative for TEW using published data.
- Finally, other work that was included used TEW in analyses, discussed model structures with relevance for TEW or just mentioned TEW in any capacity meaning that the presentations were eligible for inclusion but did not provide information useful for this review.

Table 1. NICE technology appraisals included and assessments of treatment effect waning assumptions.

TA number date	Treatment, disease & treatment line	Stopping rule	TEW - base case [b]	TEW - scenarios	TEW - ERG	TEW - AC
TA428 January 2017	Pembrolizumab Lung, NSCLC, 2L+	2-year	None	HR for OS set to 1 at 3, 5, and 10 years from start of treatment	HR for OS set to 1 at 3 years	Could not agree on a single scenario
TA531 July 2018	Pembrolizumab Lung, NSCLC, 1L	2-year	None	HR for OS set to 1 at 3 and 5 years from start of treatment	No comments	Duration of treatment effect uncertain. Scenarios considered in decision
TA578 / TA798 ^ June 2022	Durvalumab Lung, NSCLC, 2L+	1-year	None	Equal hazard after 7.5 and 10 years from start of treatment	Equal hazard after 3 and 5 years preferred scenarios	Equal hazards after 3 and 5 years appropriate for decision making
TA589 July 2019	Blinatumomab Blood & bone marrow, 2L+	<1-year (up to 2 cycles)	Equal hazard after 11 years	Duration of benefits set to 60 months	No new scenarios explored	AC preferred fixed cure point was 60 months
TA650 September 2020	Pembrolizumab Renal cell carcinoma, 1L	2-year	None	Equal hazard after 10 years from start of treatment & gradual TEW between 5 and 10 years based on response	Equal hazard after 5 and 10 years. Re-ran gradual analyses for all (not just responders)	Not enough evidence for lifetime treatment effect^. TEW after 5 years from start of treatment most plausible
TA661 November 2020	Pembrolizumab Head and neck, 1L	2-year	None	Equal hazard after 3 and 5 years from start of treatment	Equal hazard after 5 years (from starting treatment)	Agreed with the ERG that a 5-year treatment effect was appropriate and consistent with previous Tas
TA683 March 2021	Pembrolizumab Lung, NSCLC, 1L	2-year	Equal hazards after 5 years from start of treatment	Equal hazards after 3 years, 10 years. Lifetime treatment effect^	Gradual waning between 2 and 5 years	Agreed with ERG gradual waning approach
TA692 April 2021	Pembrolizumab Bladder, urothelial carcinoma, 2L+	2-year	Equal hazards after 5 years from start of treatment	Equal hazards after 3 and 10 years. Also, conditional waning on those with/without disease control	Equal hazards after 3 years. Also explored 2, 5 and 10 years in scenarios	3-year treatment effect most plausible
TA705 June 2021	Atezolizumab Lung, NSCLC, 1L	Atezolizumab = none Pembrolizumab = 2 year	Pembrolizumab equal hazards after 5 years from start of treatment. Atezolizumab none	Atezolizumab equal hazards after 5 and 8 years	Maintained company base case and explored several scenarios	AC considered various duration of treatment effect scenarios performed by the ERG
TA724 September 2021	Nivolumab Lung, NSCLC, 1L	2-year	Initially none. Updated base-case equal hazards after 5 years (from stopping treatment)	Equal hazard after 3 years	Equal hazards after 3 and 5 years explored (after starting treatment)	Treatment effect lasting 3 to 5 years most appropriate
TA737 October 2021	Pembrolizumab Oesophageal, 1L	2-year	None. Company claims TEW is reflected in the extrapolation of OS	Gradual waning between 5 and 7 years (equal hazard by 7)	Preferred scenario was waning between 5 and 7 years	All scenarios plausible
TA770 February 2022	Pembrolizumab Lung, NSCLC, 1L	2-year	Equal hazard after 5-years from start of treatment for OS but lifetime treatment effect^ for PFS	Equal hazard after 3 and 10 years from start of treatment	Preference of equal hazard for OS at 5 years. Explored TEW for PFS as scenario	Originally preferred TEW after 3 years but company contested with newer data cut. Settled for TEW after 5 years.
TA772 February 2022	Pembrolizumab Lymphatic, 3L+	2-year	None	Gradual waning between 5 and 7 years from start of treatment (equal hazard by 7)	Explored gradual waning between 3 and 5 years	Did not comment on TEW
TA788 May 2022	Avelumab Bladder, 2L+	2-year in revised base-case	Equal hazard after 5 years from starting treatment	Explored waning at 5,6,7,8 years and gradual waning	Equal hazards at 2,3,4,7,10 years and gradual waning between 2 and 5 years	No stopping rule accepted (to reflect trial) and so waning should not be applied
TA798 June 2022	Durvalumab Lung, NSCLC, 2L+	1-year	None	Equal hazards after 10 years from starting treatment	TEW after 3 years for PFS and 5 years for OS conditional on base case	TEW after 3 and 5 years both appropriate for decision making
TA801 June 2022	Pembrolizumab Breast, 1L	2-year	None	Gradual waning from 4 years using SEER data and equal hazard after 5 years	Preferred equal hazard after 5 years from start of treatment	5-year treatment effect most appropriate
TA802 June 2022	Cemiplimab Skin, 1L	22 months	Equal hazard after 5 years from starting treatment	No waning and gradual waning between 60 and 96 months	Additional TEW scenarios of equal hazards at 42 and 48 months	NR
TA818 August 2022	Nivolumab Lung , mesothelioma, 1L	2-year	None	Gradual waning - decrease linearly between year 5 and 10	Equal hazards after 5 years from start of treatment	Concluded that it was reasonable to assume some TEW, but duration unclear
TA823 September 2022	Atezolizumab Lung, NSCLC, 2L+	1-year	Originally equal hazard after 5 years. Subsequently removed and curves adjusted with 5-year cure assumption	None	ERG noted that the original waning improved CE, therefore recommended removing	None

Abbreviations: TA, technology appraisal; TEW, treatment effect waning; ERG, evidence review group; AC, appraisal committee; RCT, randomised controlled trial; HR, hazard ratio; OS, overall survival; NR, not reported; CDF, cancer drugs fund; PFS, progression-free survival; CE, cost effectiveness. [a] to determine if the survival models being used assumed proportional hazards / constant treatment effect or were independently fitted to each treatment arm (as this could inform whether waning may be "inbuilt"). [b] in some instances, the base-case changed (e.g., after CDF review); the most recent base-case was included. ^ TA798 guidance replaced TA578.

Targeted Literature Review (TLR)

- In total, 30 articles met the inclusion criteria for the TLR. Included articles were categorised into: articles with clinical trial data, real-world evidence observational analyses and review papers (**Figure 2**). The studies found were IO treatment for advanced/metastatic disease, across a range of solid-tumours.
- Two emerging themes were found from the TLR:
 - There was evidence showing that patients who experienced a complete response are more likely to have favourable survival outcomes [5]. Therefore, there could be merit in applying TEW differently based on patients' responses.
 - Secondly, the optimal duration of treatment for IOs is still unknown, but there was some evidence showing that continuing treatment beyond the initial response might be beneficial for some patients [6].
- An analysis performed by Regan et al. (2019) [7] provided evidence of a sustained treatment effect for nivolumab following discontinuation using treatment free survival as an outcome.
- It would be beneficial if future studies on long-term clinical trial outcomes included smoothed hazard plots compared to background mortality rates, and plots of the observed treatment effect over time to help inform TEW analyses. None such analyses were found within this TLR. Examples can be found in IO survival validation papers that have presented smoothed hazard plots derived from KM curves observed in IO RCTs with long-term follow-up [8,9].

Figure 2. The flow of articles included in the TLR.

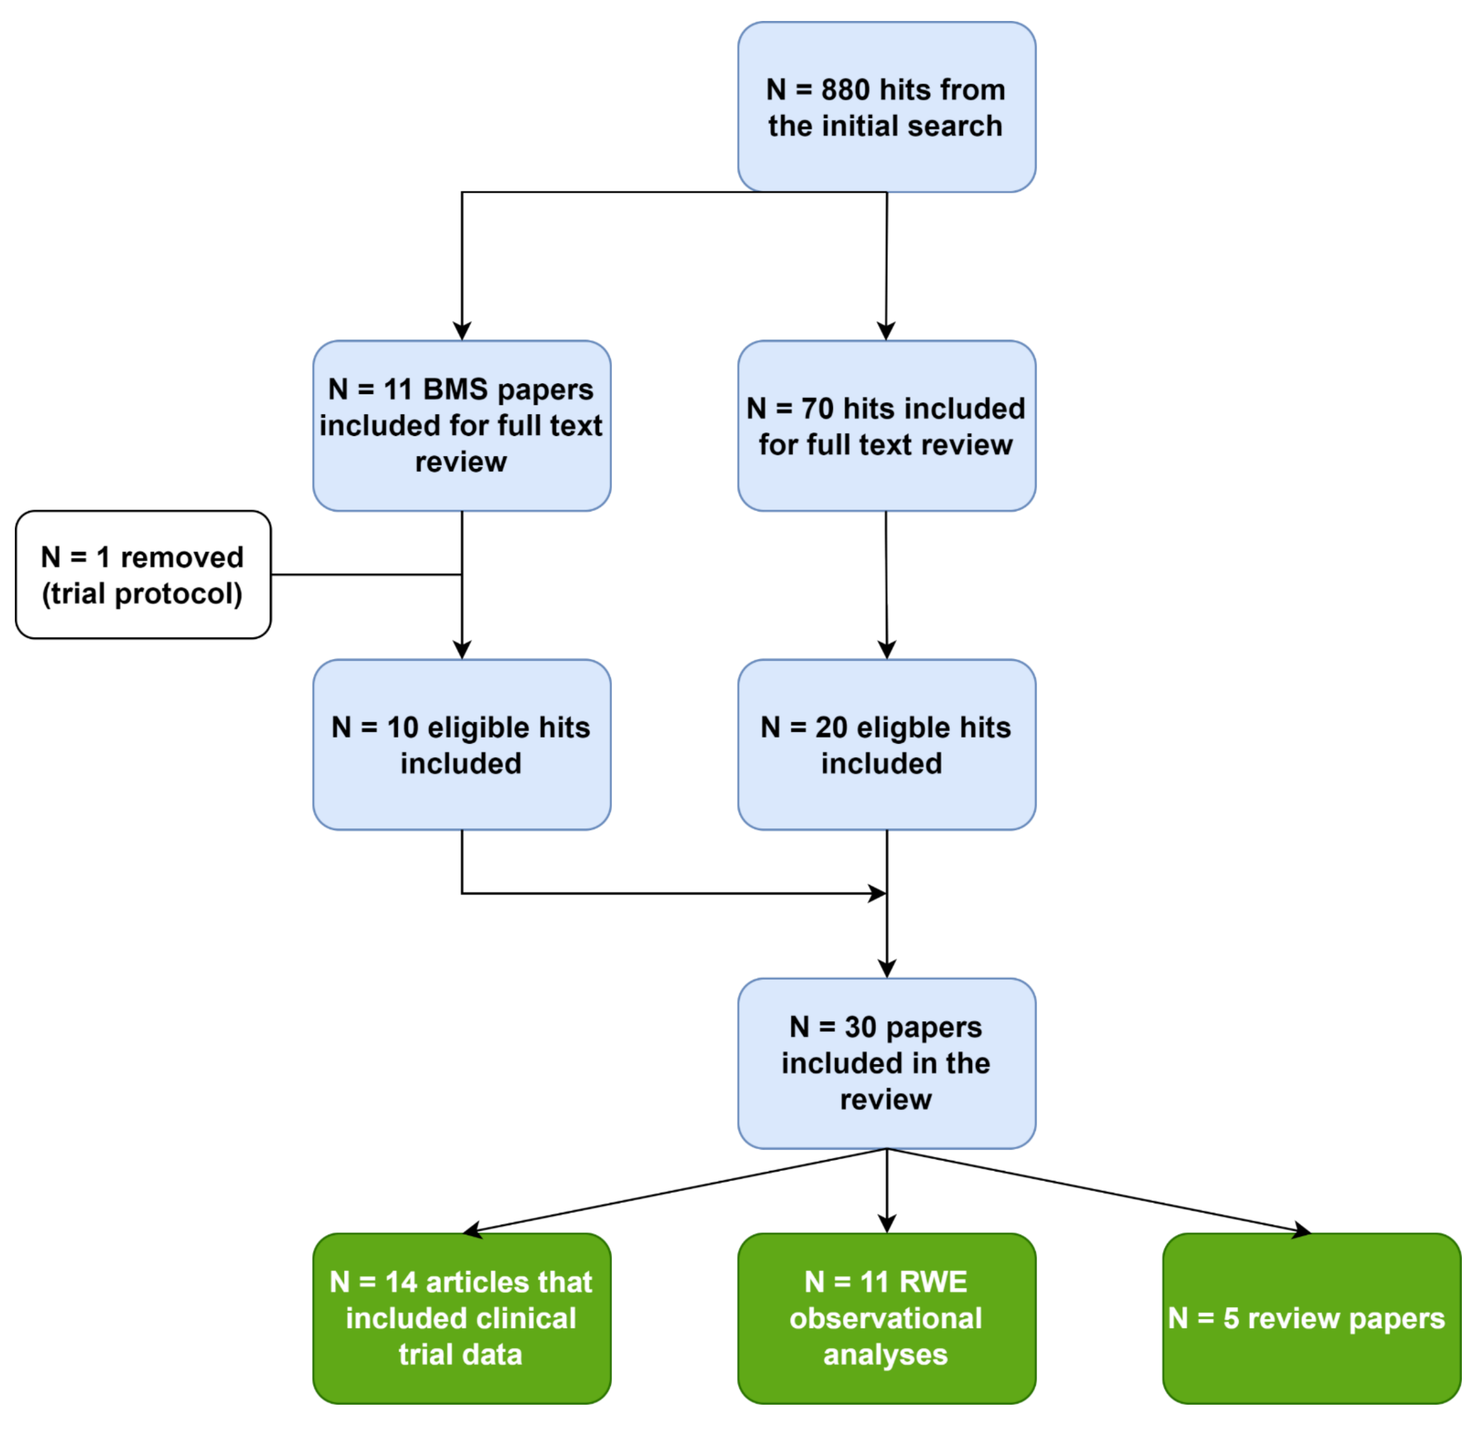
N = 880 hits from the initial search
N = 11 BMS papers included for full text review
N = 70 hits included for full text review
N = 1 removed (trial protocol)
N = 10 eligible hits included
N = 20 eligble hits included
N = 30 papers included in the review
N = 14 articles that included clinical trial data
N = 11 RWE observational analyses
N = 5 review papers

Figure 3. Treatment effect waning algorithm for use in health technology assessment.

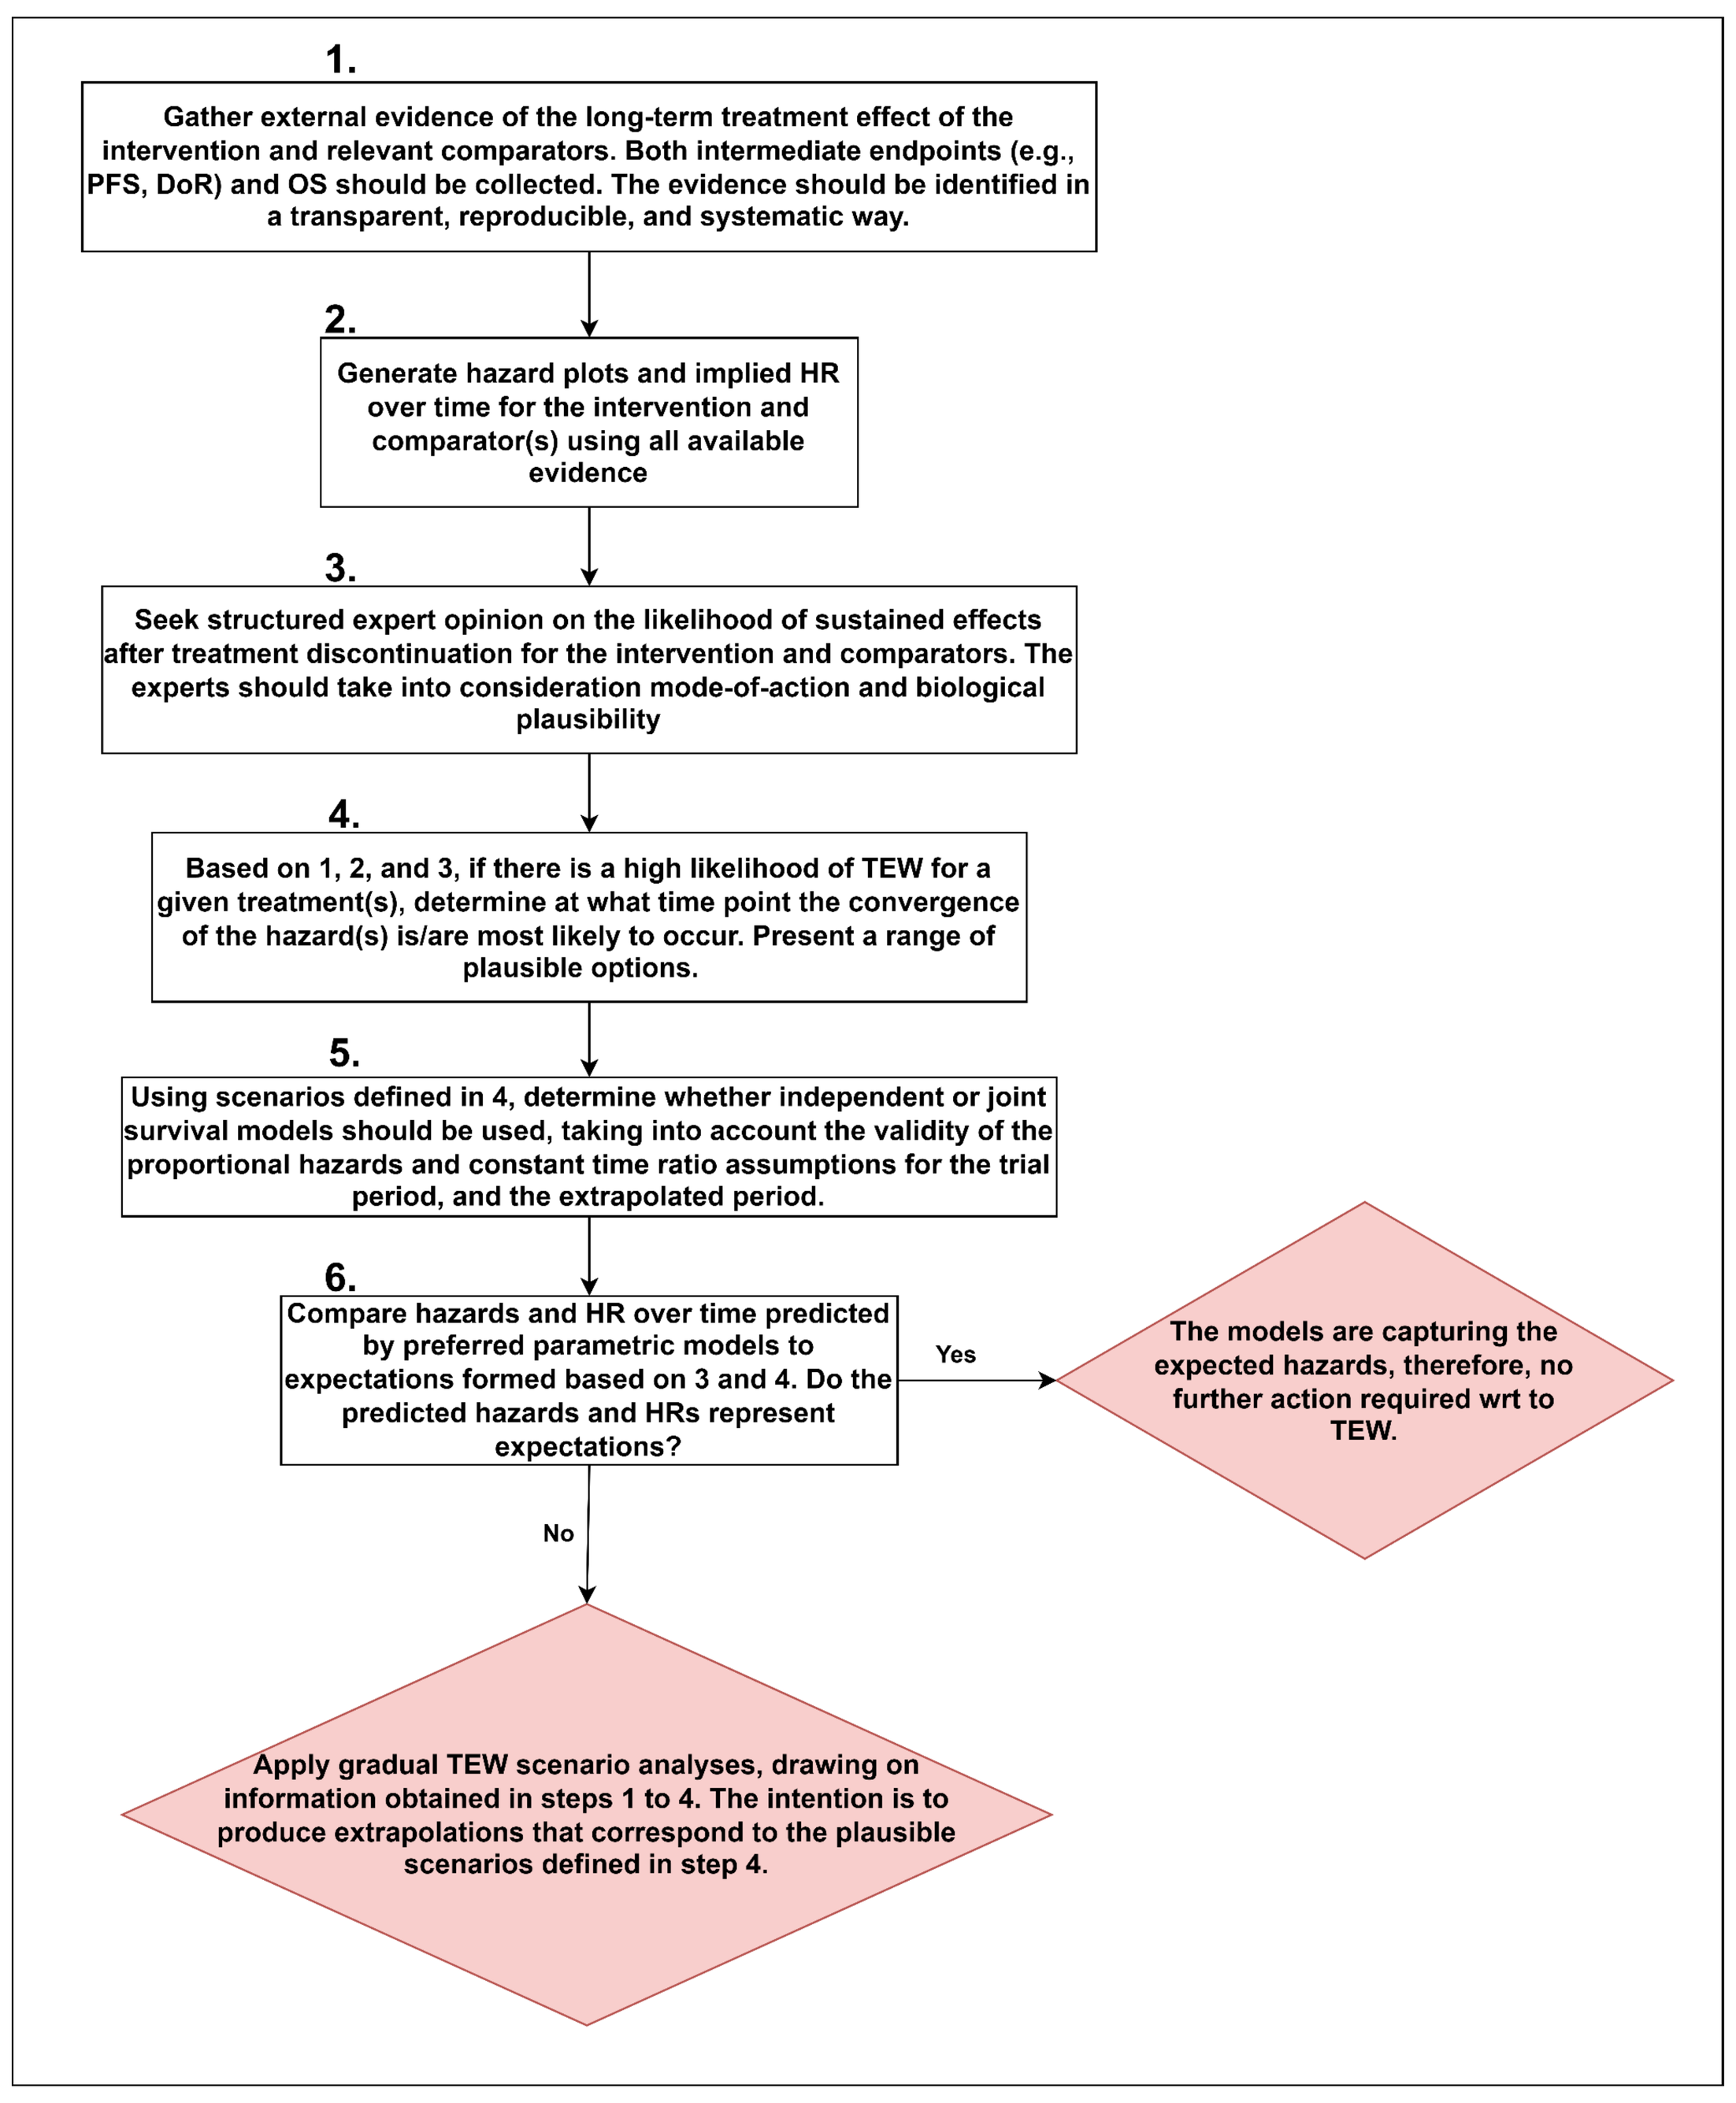
1.
Gather external evidence of the long-term treatment effect of the intervention and relevant comparators. Both intermediate endpoints (e.g., PFS, DoR) and OS should be collected. The evidence should be identified in a transparent, reproducible, and systematic way.
2.
Generate hazard plots and implied HR over time for the intervention and comparator(s) using all available evidence
3.
Seek structured expert opinion on the likelihood of sustained effects after treatment discontinuation for the intervention and comparators. The experts should take into consideration mode-of-action and biological plausibility
4.
Based on 1, 2, and 3, if there is a high likelihood of TEW for a given treatment(s), determine at what time point the convergence of the hazard(s) is/are most likely to occur. Present a range of plausible options.
5.
Using scenarios defined in 4, determine whether independent or joint survival models should be used, taking into account the validity of the proportional hazards and constant time ratio assumptions for the trial period, and the extrapolated period.
6.
Compare hazards and HR over time predicted by preferred parametric models to expectations formed based on 3 and 4. Do the predicted hazards and HRs represent expectations?
Yes
The models are capturing the expected hazards, therefore, no further action required wrt to TEW.
No
Apply gradual TEW scenario analyses, drawing on information obtained in steps 1 to 4. The intention is to produce extrapolations that correspond to the plausible scenarios defined in step 4.

Conclusions

- The HTA agency review revealed that there is no standardised approach adopted by companies in their submissions. EAG / Committee are more conservative and prefer TEW to be incorporated for decision making purposes.
- Independent survival models are commonly used, however modelled treatment effects are often not illustrated meaning that it is difficult to determine whether a waning of treatment effect is already accounted for within the model.
- The ISPOR search uncovered two independent projects that arrived at the same conclusion, suggesting that gradual waning methods based on the equalization of hazards over time may represent a more suitable approach than immediate waning. The preferred NICE method was immediate waning which is a more conservative approach.
- The TLR uncovered promising long-term survival outcomes following discontinuation from IO treatments. Findings consistently indicated that patients who achieved CR tended to experience more favourable survival outcomes and had a higher likelihood of being cured. However, no analyses comparing long-term hazards between IO-treated patients and comparator groups were found.
- Based on the findings from this work, a sequence of steps for assessing the necessity and appropriateness of presenting TEW scenario analyses in HTA is proposed (**Figure 3**).

Acknowledgments

- This study was sponsored by Bristol Myers Squibb.

References

1. Harrington H et al. Forecasting the Long-Term Treatment Effect Duration of Immuno-Oncology Therapies: An Analysis of the Predictive Accuracy of Treatment Waning Methods Applied to Pembrolizumab in Non-Small Cell Lung Cancer. ISPOR EU 2022. 2022 Nov 6.
2. Micallef J et al. When Does a Treatment Effect Really Stop? Exploration of Different Methods for Modelling Treatment Waning. ISPOR US 2022. 2022 May 15.
3. Horscroft J et al. A review of differences in decision-making across NICE health technology assessments of nivolumab. ISPOR EU 2019. 2019 Nov 2.
4. Kamgar F et al. A Review of Treatment-Effect Waning Methods for Immuno-Oncology Therapies in National Institute for Health and Care Excellence Technology Appraisals. ISPOR EU 2022. 2022 Nov 6.
5. Asher N et al. Immunotherapy Discontinuation in Metastatic Melanoma: Lessons from Real-Life Clinical Experience. Cancers. 2021 Jun 20;13(12):3074.
6. Waterhouse DM et al. Continuous Versus 1-Year Fixed-Duration Nivolumab in Previously Treated Advanced Non-Small-Cell Lung Cancer: CheckMate 153. J Clin Oncol. 2020 Nov 20;38(33):3863-73.
7. Regan MM et al. Treatment-Free Survival: A Novel Outcome Measure of the Effects of Immune Checkpoint Inhibition—A Pooled Analysis of Patients With Advanced Melanoma. J Clin Oncol. 2019 Dec 10;37(35):3350-8.
8. Bullement A et al. Survival Extrapolation in Cancer Immunotherapy: A Validation-Based Case Study. Value Health. 2019 Mar;22(3):276-83.
9. Paly V et al. Heterogeneity in Survival with Immune Checkpoint Inhibitors and Its Implications for Survival Extrapolations: A Case Study in Advanced Melanoma. MDM Policy Pract. 2022 Mar 26; 7(1): 1-15

Presented at ISPOR Europe 2023; November 12-15, 2023; Copenhagen, Denmark

Email: ktaylor@deltahat.com

Copies of this poster are for personal use only and may not be reproduced without written permission of the authors.