Comparison of the Clinical and Economic Impact of Two mRNA COVID-19 Vaccines in High Risk Individuals in Japan

Michele Kohli,[1,*] Yuriko Hagiwara,[2] Ataru Igarashi,[3,4] Keya Joshi,[2] Amy Lee,[1] Michael Maschio,[1] Nicolas Van de Velde,[2] Ekkehard Beck[2]

[1]Quadrant Health Economics Inc., Cambridge, ON, Canada; [2]Moderna, Inc., Cambridge, MA, USA; [3]Department of Health Economics and Outcomes Research, Graduate School of Pharmaceutical Sciences, University of Tokyo, Tokyo, Japan; [4]Department of Health Data Science, Graduate School of Data Science, Yokohama City University, Yokohama, Japan

*Presenting author.

SUPPLEMENTARY MATERIAL

Supplementary Methods

Model Inputs: Population Size and COVID-19 Incidence

- The size of the Japanese adult population aged ≥65 years (n = 37,071,740) was obtained from the World Population Prospects (United Nations)[1]
- The size of the immunocompromised population (≥18 years, n = 4,675,900) was assumed based on the number of patients with autoimmune diseases and oncology patients[2] and was weighted by the distribution by age of the immunocompromised patients[3]
- Individuals who did not receive a Moderna or Pfizer-BioNTech Fall 2023 vaccine were considered to have residual protection against infection and hospitalization from past vaccines
 - The population was stratified into groups based on the highest level of prior vaccination received: 1) no vaccination; 2) primary series; 3) first monovalent booster; 4) second monovalent booster; and 5) bivalent booster[4-6]
 - Residual vaccine effectiveness (VE) for each strata at the start of the model time horizon was calculated from initial VE against omicron,[7-9] monthly waning,[10] and average time since vaccination
- Probability of hospitalization in unvaccinated individuals was calculated by varying the age-specific probabilities to ensure that the number of hospitalizations in individuals who did not receive the Fall 2023 formulas matched the expected number of hospitalizations in Japan[11]

Model Inputs: COVID-19–Associated Outcomes and Vaccine Coverage

Supplementary Table 1. Base-Case Input Probabilities: Adults Aged ≥65 Years and Immunocompromised Population

Age Group	Infection-Related Myocarditis (%)[12]	Infection-Related Hospitalization (%)[11,a]	Hospital Mortality (%)[13,14,b]
Adults ≥65 years			
65-74 years	0.160	23.79	7.07
≥75 years	0.208	39.55	12.91
Immunocompromised adults (≥18 years)			
18-29 years	0.078	2.27	0.22
30-39 years	0.067	3.49	0.22
40-49 years	0.093	3.71	1.58
50-59 years	0.137	7.59	1.58
60-64 years	0.137	20.72	7.02
65-74 years	0.160	23.79	9.87
≥75 years	0.208	39.55	17.59

[a]Hospitalization data from the general population were adjusted to the immunocompromised population based on the data from a previous study.[15]

[b]Age-specific death rates given hospitalization (death after hospitalization regardless of location) for the general population were adjusted to the immunocompromised population using the data from a previous study.[16]

Supplementary Figure 1. Fall 2023 Vaccine Coverage by Age Group

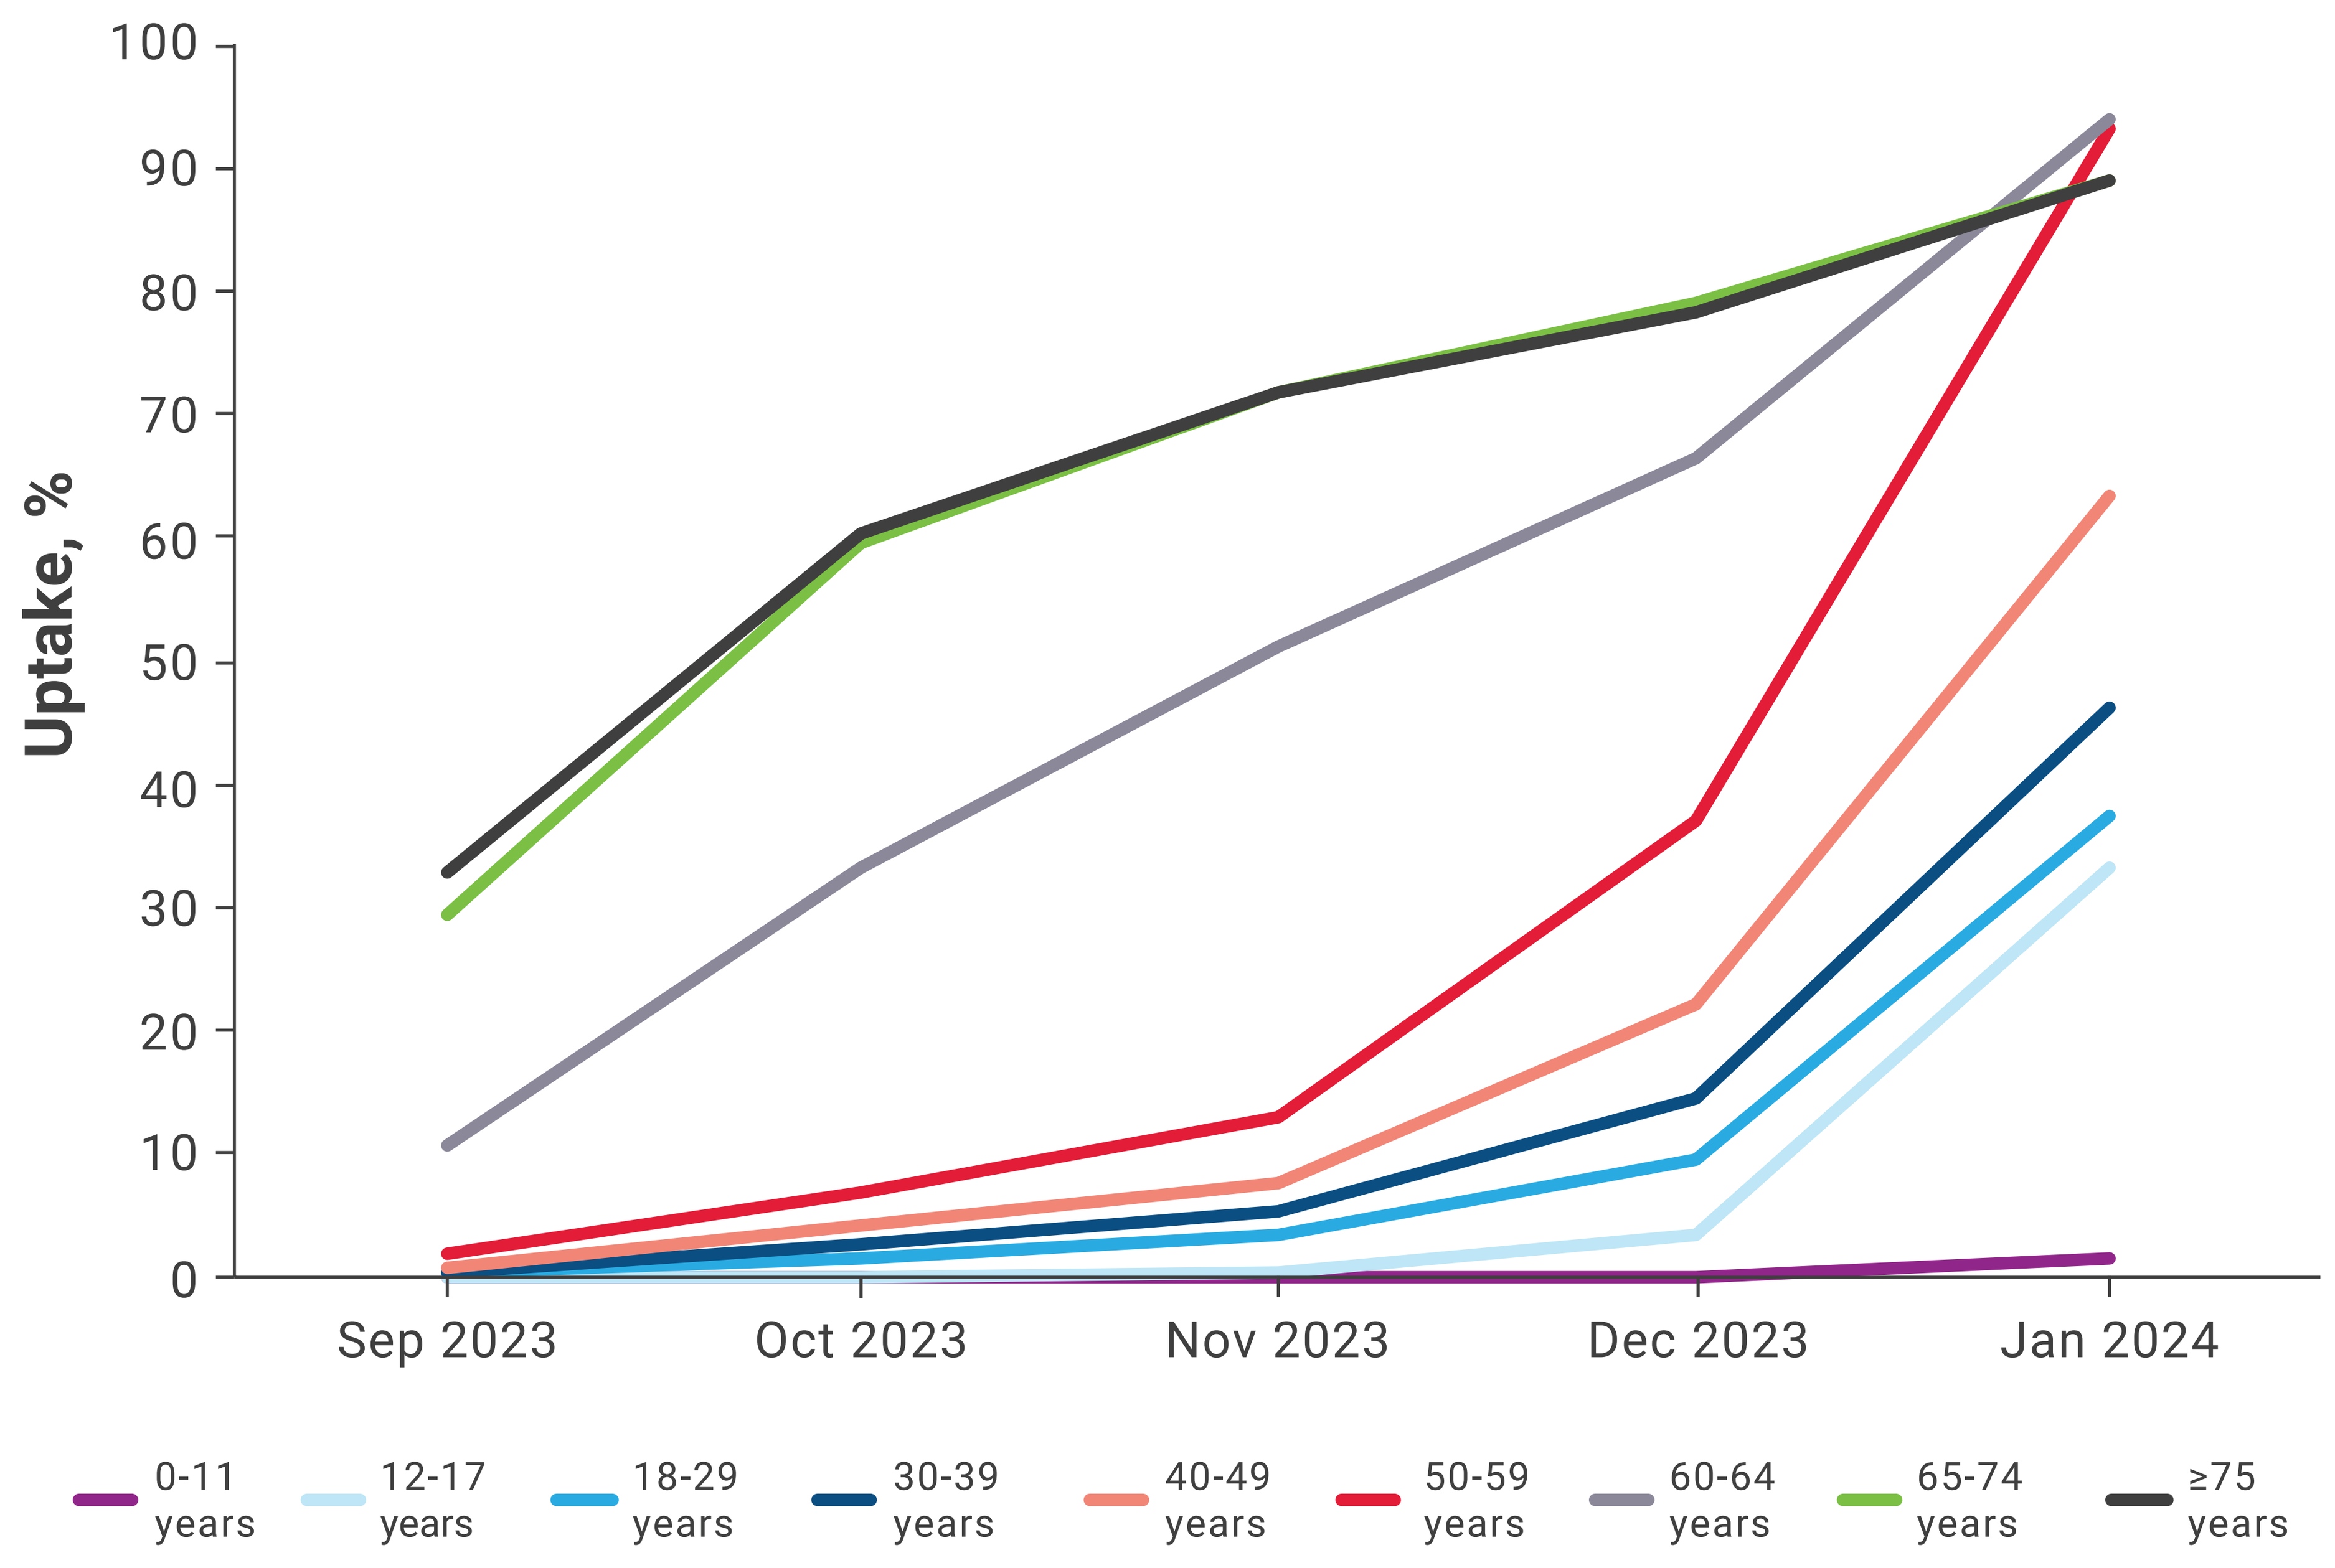
100
90
80
70
60
50
40
30
20
10
0
Uptake, %
Sep 2023
Oct 2023
Nov 2023
Dec 2023
Jan 2024
0-11 years
12-17 years
18-29 years
30-39 years
40-49 years
50-59 years
60-64 years
65-74 years
≥75 years

Model Inputs: Cost and Resource Use

Supplementary Table 2. Base-Case Inputs For Cost and Resource Use

Input	Value	Source
Proportion seeking outpatient care	100%	Assumption that all reported cases have sought medical care
Cost of outpatient care	¥44,991	Average of: <60 years[14] ≥60 years[13]
Cost per hospitalization	¥778,075	Same as above
Myocarditis (induced by vaccine or infection)	¥20,262	Claims analysis conducted by Prof. Igarashi[13]
Proportion vaccinated with vaccine-induced myocarditis[a]	0.00182%	COVID-19 vaccine safety updates[17] (males and females weighed equally)

[a]Only applies to individuals aged 18 to 39 years.

References

1. United Nations Department of Economic and Social Affairs PD. World Population Prospects Online Edition. Population by Single Age – Both Sexes. https://population.un.org/wpp/Download/Standard/Population
2. Tokyo Statistical Yearbook. https://www.toukei.metro.tokyo.lg.jp/juukiy/jy-index.htm
3. Ramage-Morin PL, Polsky JY. *Health Rep.* 2020;31(5):3-8.
4. Ministry of Health Labour and Welfare. Government of Japan. The 80th Meeting of the Adverse Reactions Subcommittee of the Immunization and Vaccine Subcommittee of the Health and Welfare Science Council, and the 5th Meeting of the Pharmaceutical Affairs and Food Sanitation Council in FY Reiwa 4 (jointly held). 2023. https://www.mhlw.go.jp/stf/shingi2/0000208910_00043.html
5. Prime Minister's Office of Japan. About the New Coronavirus Vaccine. https://www.kantei.go.jp/jp/headline/kansensho/vaccine.html
6. Open Data. Vaccination. https://data.vrs.digital.go.jp/vaccination/opendata/latest/prefecture.ndjson
7. Vaccine Effectiveness Real-Time Surveillance for SARS-CoV-2 (VERSUS). Vaccine Effectiveness Real-Time Surveillance for SARS-CoV-2 (VERSUS) Study, 5th Report. https://www.tm.nagasaki-u.ac.jp/versus/results/20220608.html
8. Vaccine Effectiveness Real-Time Surveillance for SARS-CoV-2 (VERSUS). Vaccine Effectiveness Real-Time Surveillance for SARS-CoV-2 (VERSUS) Study, 7th Report. https://www.tm.nagasaki-u.ac.jp/versus/results/20221221.html
9. Vaccine Effectiveness Real-Time Surveillance for SARS-CoV-2 (VERSUS). Vaccine Effectiveness Real-Time Surveillance for SARS-CoV-2 (VERSUS) Study, 8th Report. https://www.tm.nagasaki-u.ac.jp/versus/results.html
10. Higdon MM, et al. *Lancet Infect Dis.* 2022;22(8):1114-1116.
11. National Institute of Infectious Diseases. COVID-19 Weekly Surveillance Update: Epidemiologic Situational Awareness. https://www.niid.go.jp/niid/images/epi/PDF/COVID-19_2023w18.pdf
12. Boehmer TK, et al. *MMWR Morb Mortal Wkly Rep.* 2021;70(35):1228-1232.
13. DeSC Healthcare. Insurance Database. https://desc-hc.co.jp/en/company
14. JAST Lab. REZULT Data Overview. https://www.jastlab.jast.jp/rezult_data/
15. Bahremand T, et al. *Lancet Reg Health Am.* 2023;20:100461.
16. Turtle L, et al. *PLoS Med.* 2023;20(1):e1004086.
17. Advisory Committee on Immunization Practices (ACIP). COVID-19 Vaccine Safety Update: Primary Series in Young Children and Booster Doses in Older Children and Adults. https://www.cdc.gov/vaccines/acip/meetings/downloads/slides-2022-09-01/05-COVID-Shimabukuro-508.pdf

Presented at ISPOR Europe; November 12-15, 2023; Copenhagen, Denmark