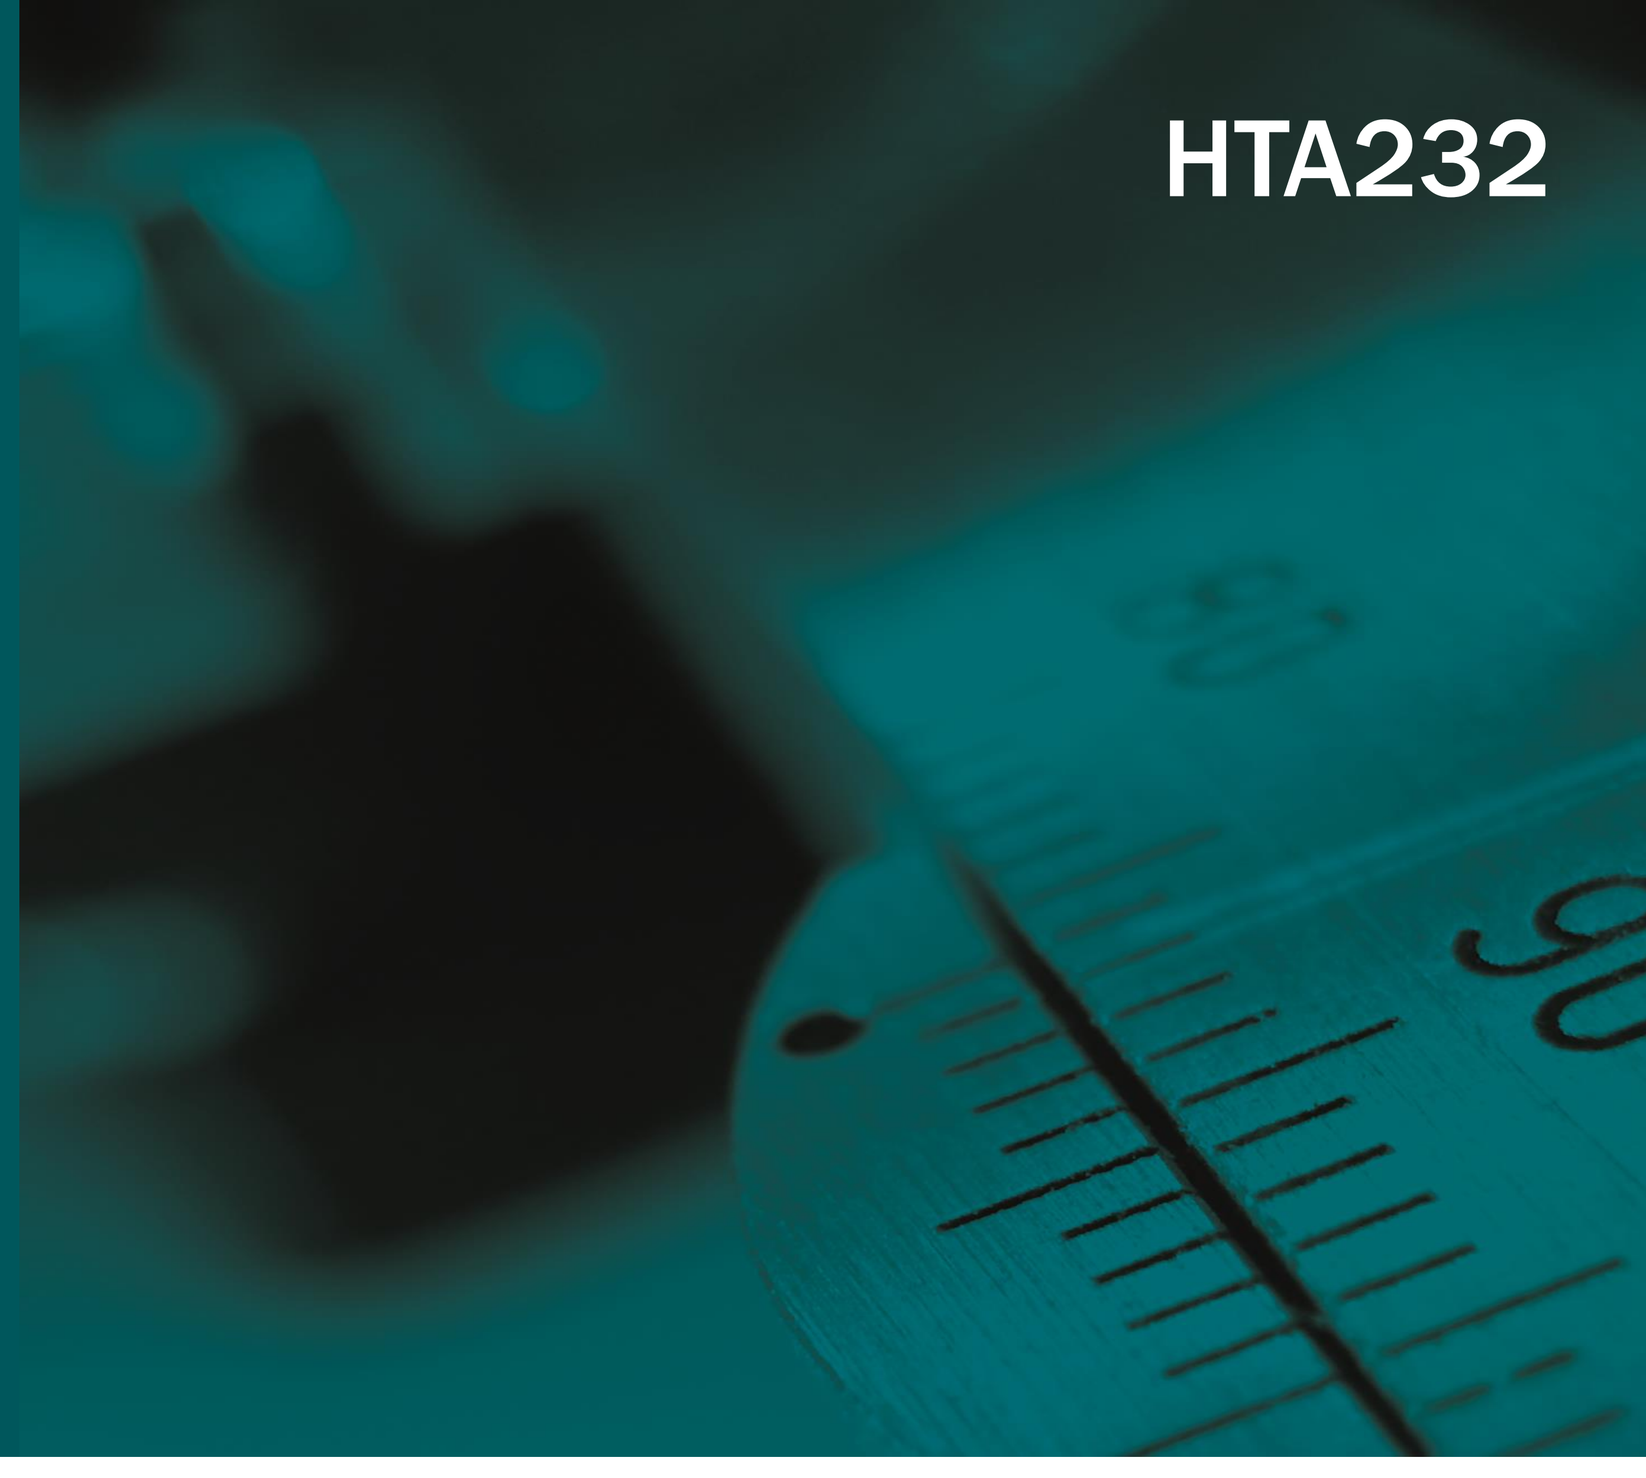

HTA232

A Multi-Criteria Decision Analysis Framework for the Value Assessment of First-Line Treatment of Adult Patients with Advanced Anaplastic Lymphoma Kinase Positive Non-Small Cell Lung Cancer

Martins P[1], Vandewalle B[1], Labisa P[2], Cochado S[2]
1. Exigo Consultores, Lisbon, Portugal; 2. Takeda Farmacêuticos Portugal, Lisbon, Portugal

OBJECTIVES

- Establish a multi-criteria decision analysis (MCDA) framework for the value assessment of first-line treatment of adults with advanced, anaplastic lymphoma kinase positive, non-small cell lung cancer (*ALK*+ NSCLC) in Portugal.
- Compare value contributions of the second-generation ALK inhibitors, brigatinib and alectinib – within this novel value assessment framework – from a Portuguese physician decision-making viewpoint.

METHODS

Problem Definition and Stakeholder Involvement

- The objective of the MCDA under consideration was defined ad initium, aiming to develop an assessment tool in a hospital setting regarding the choice of reimbursed treatments for patients with advanced *ALK*+ NSCLC previously not treated with an ALK inhibitor.

Criterion Definition and Structuring

- Relevant criteria (and associated levels) were obtained from a literature review of MCDA frameworks, and initial health technology assessment proposals produced by health authorities, related to the treatment of (lung) neoplasms.
- The initial list was fine-tuned into a final matrix of criteria and levels, contemplating completeness, non-redundancy, non-overlap, and preference independence, during a focus group with three Portuguese physicians experienced in the treatment of adults with advanced *ALK*+ NSCLC.

Weight and Part-Worth Utility Estimation

- Criteria weights and part-worth utilities for the MCDA framework, reflecting preferences towards attributes and their respective levels, were elicited from the same physicians through adaptive conjoint analysis (Lighthouse Studio – Sawtooth Software).

Alternatives Evaluation

- Brigatinib and alectinib were rigorously assessed on all criteria, considering the most recent publicly available evidence.

Global Value Estimation

- An overarching estimate of value, integrating individual alternative scores across criteria, was expressed as share of preference between both alternatives.
- This metric helps to understand how physicians allocate their preferences within a group of different competing products (i.e.: brigatinib and alectinib).

RESULTS

Criterion Definition and Structuring

- The final MCDA criteria and their respective levels, derived from the existing clinical evidence pertaining to the treatments employed in contemporary clinical practice (specifically, brigatinib and alectinib), and refined based on the clinical expertise of the physicians, are presented in Table 1.

Table 1. Final list of criteria and corresponding levels for the MCDA framework.

Criterion	1st level	2nd level	3rd level	4th level
One-year progression-free survival	50%	70%	90%	-
Four-year overall survival	60%	70%	80%	-
Intracranial objective response rate	70%	80%	90%	-
Proportion of discontinuations due to AE	7%	12%	17%	-
Daily dosing frequency (doses per day)	BID	QD		-
Pill burden (tablets/capsules per dose)	4	1		-
Change in health-related quality of life	Clinically significant improvement	Stabilization	Clinically significant deterioration	-
Annual treatment cost	15,000€	30,000€	45,000€	65,000€

AE: adverse events; BID: twice daily; QD: once daily

Weight and Part-Worth Utility Estimation

- Change in health-related quality of life (HRQoL), four-year overall survival (OS) and one-year progression-free survival (PFS) were ranked as the most important criteria, accounting for 62.5% of the total weight in the decision problem, followed by intracranial objective response rate (ORR) and annual treatment cost, representing a further 24.1% (Figure 1).

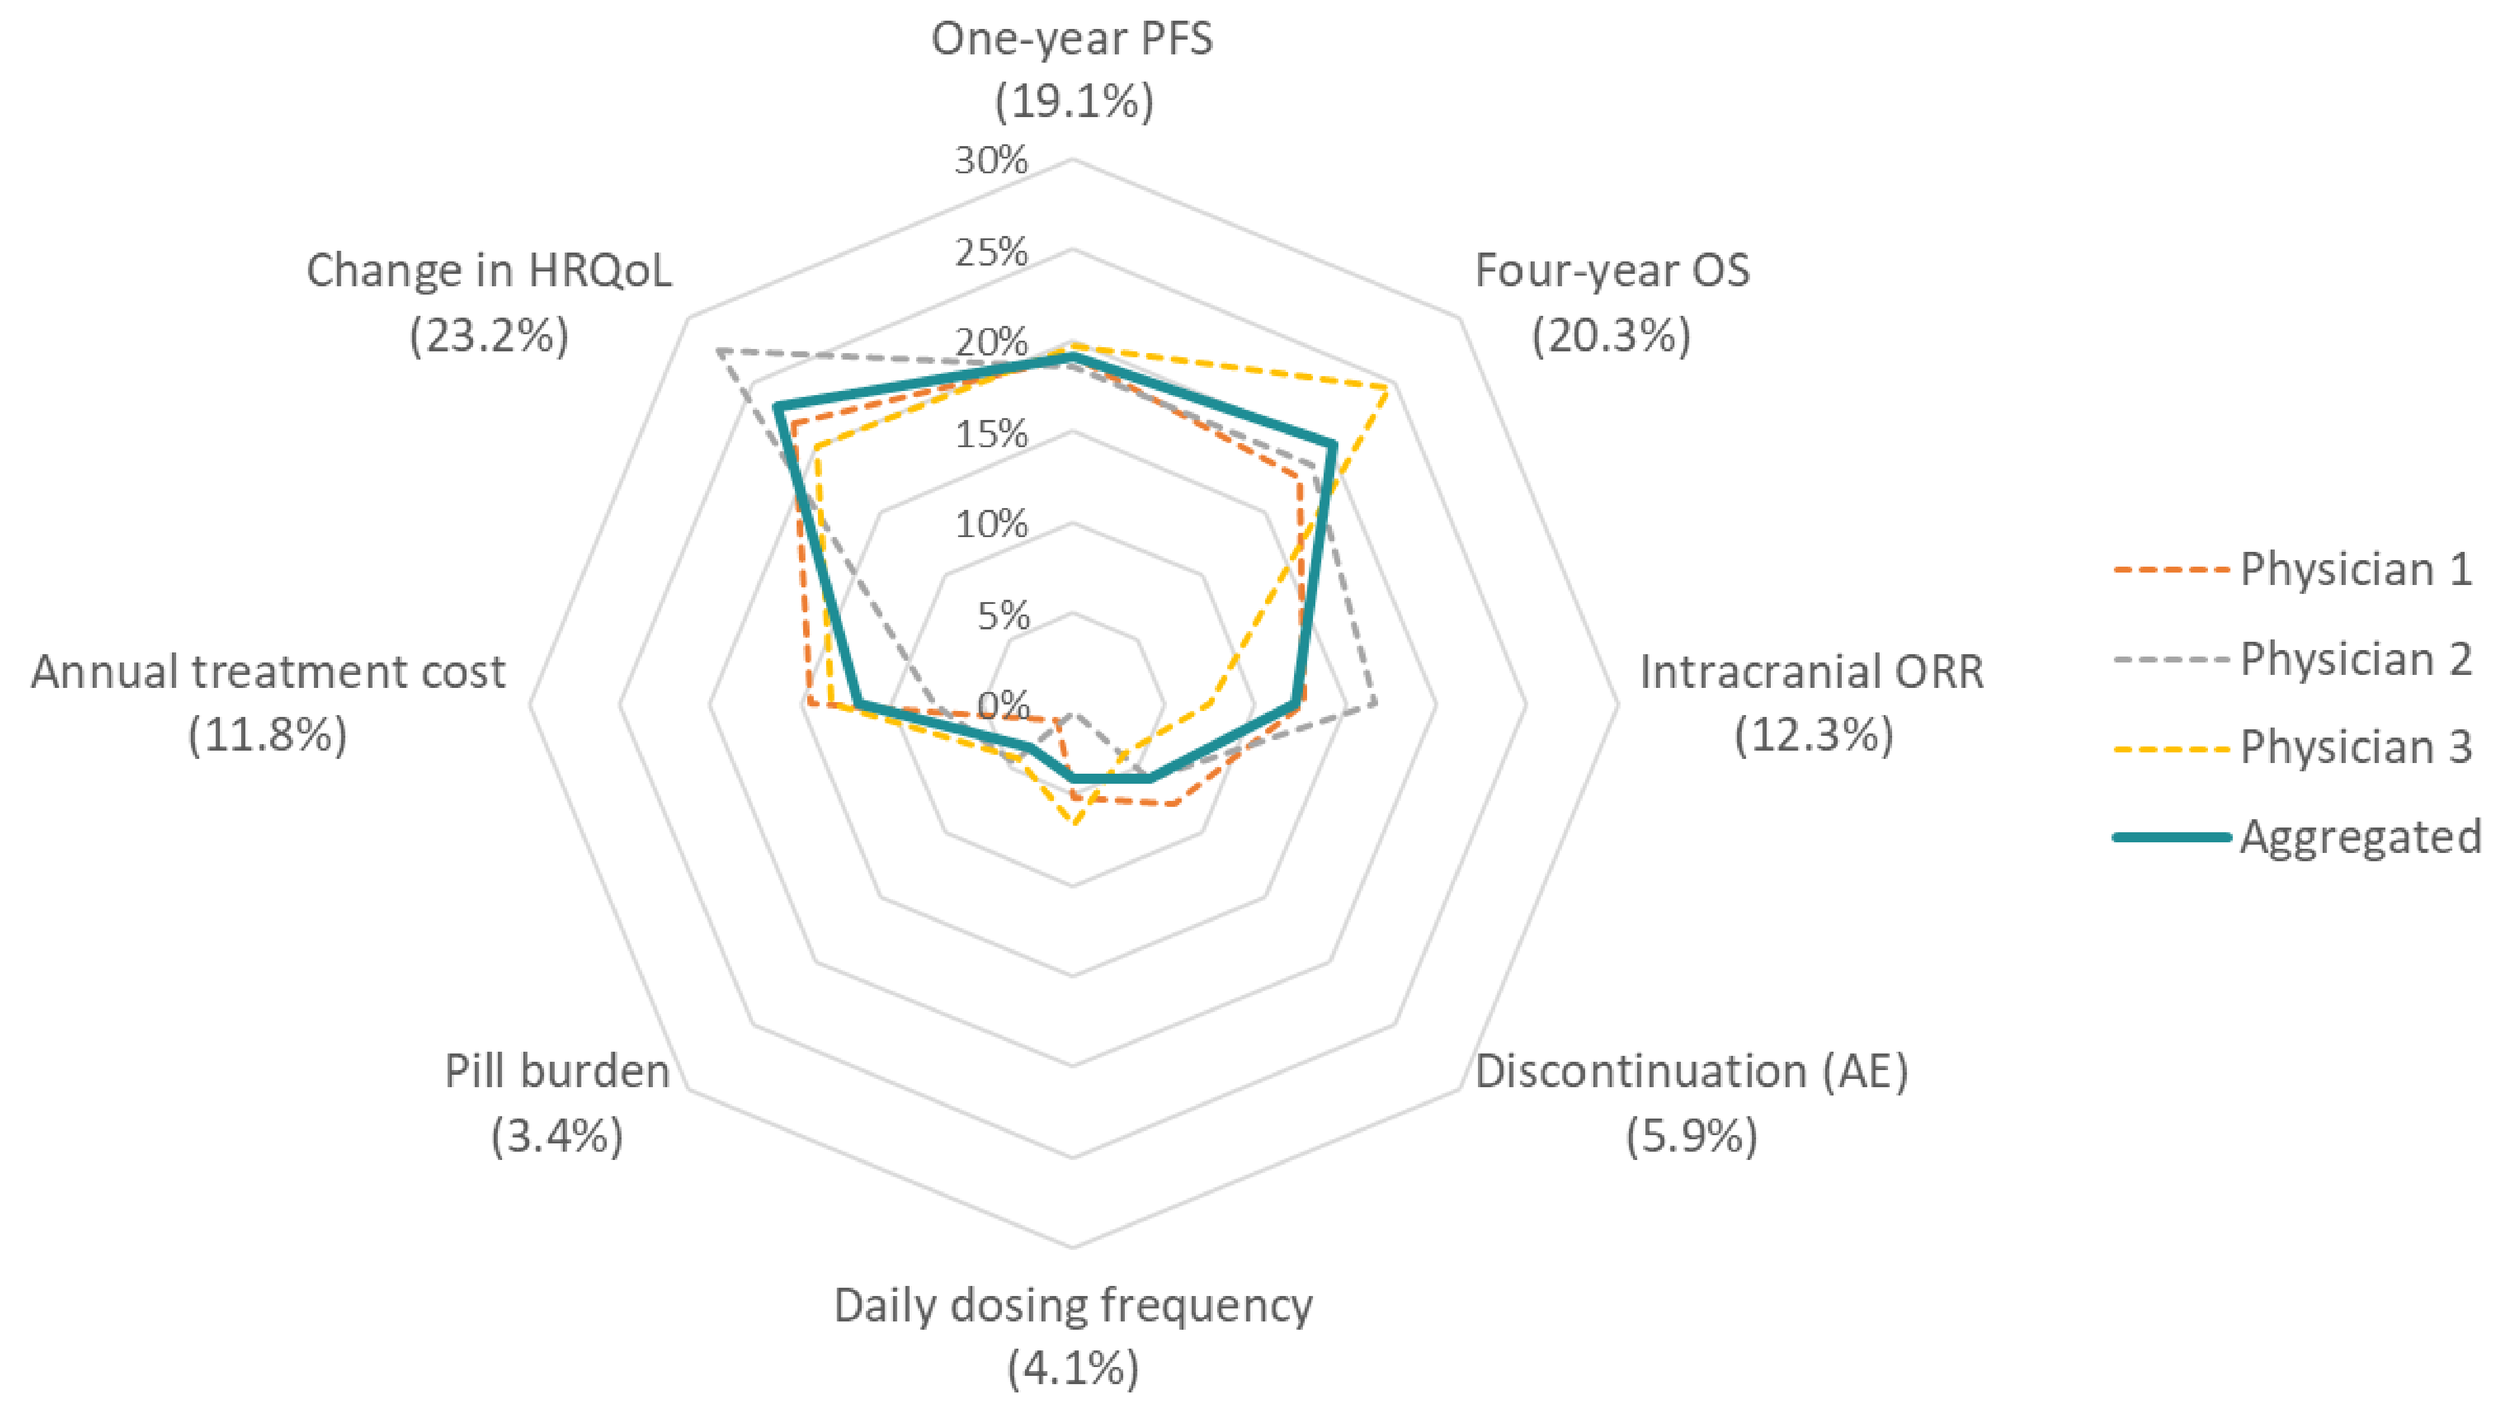
One-year PFS
(19.1%)
30%
25%
20%
15%
10%
5%
0%
Four-year OS
(20.3%)
Intracranial ORR
(12.3%)
Discontinuation (AE)
(5.9%)
Daily dosing frequency
(4.1%)
Pill burden
(3.4%)
Annual treatment cost
(11.8%)
Change in HRQoL
(23.2%)
Physician 1
Physician 2
Physician 3
Aggregated

Figure 1. Criteria weights for the MCDA framework (aggregated and per physician).

Alternatives Evaluation

- The values assigned to each criterion for each of the alternatives under evaluation (brigatinib and alectinib), in accordance with the available clinical evidence, are presented in Table 2.

Table 2. Values assigned to each criterion according to the alternatives under evaluation, in accordance with the available clinical evidence.

Criterion	Brigatinib	Alectinib
One-year progression-free survival	67.0% [1]	68.4% [5]
Four-year overall survival	66.0% [2]	65.3% [6]
Intracranial objective response rate*	78.0% [1]	81.0% [5]
Proportion of discontinuations due to AE	13.0% [2]	14.5% [6]
Daily dosing frequency (dose per day)	QD [3]	BID [7]
Pill burden (tablets/capsules per dose)	1 [3]	4 [7]
Change in health-related quality of life	Stabilization [2]	Stabilization [8]
Annual treatment cost†	48,618€ [4]	54,969€ [4]

*The value assigned to brigatinib is derived from the indicator "confirmed objective intracranial response in patients with any metastasis (measurable or non-measurable) at the initial assessment"; The value assigned to alectinib is derived from the indicator "response in the central nervous system of patients with measurable or non-measurable metastases at the initial assessment". †The level was defined based on the authors' opinion after evaluation of the available clinical data.

Global Value Estimation

- Based on the treatment profiles of brigatinib and alectinib presented in Table 2, along with the estimated part-worth utilities (not shown), one can estimate the distribution of preference in the choice between these two treatments (in an aggregated manner) for the treatment of advanced *ALK*+ NSCLC. The results are presented in Figure 2.

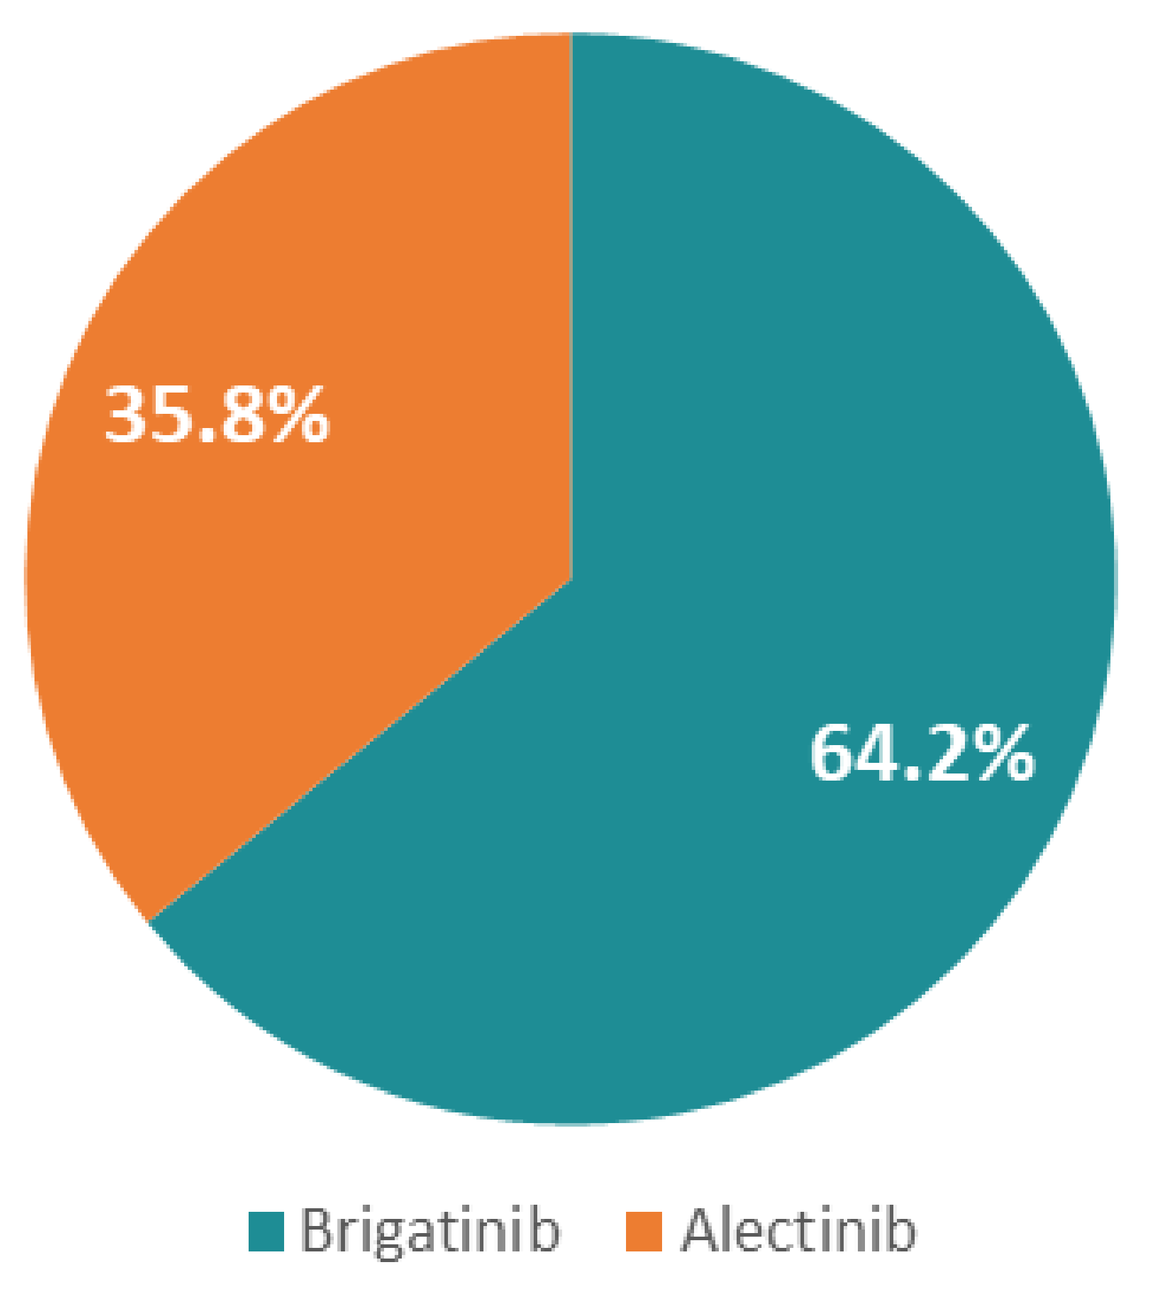
35.8%
64.2%
Brigatinib
Alectinib

Figure 2. Share of preference distribution for the treatment of advanced *ALK*+ NSCLC.

CONCLUSION

The presented value assessment framework provides an objective and transparent method to support medical decision making concerning first-line treatment of adult patients with advanced *ALK*+ NSCLC, easily adaptable to other geographical regions and treatment settings.

REFERENCES: [1] Camidge, D.R., et al., Brigatinib versus Crizotinib in ALK-Positive Non–Small-Cell Lung Cancer. New England Journal of Medicine, 2018. 379(21): p. 2027-2039. [2] Camidge, D.R., et al., Brigatinib Versus Crizotinib in ALK Inhibitor-Naive Advanced ALK-Positive NSCLC: Final Results of Phase 3 ALTA-1L Trial. J Thorac Oncol, 2021. 16(12): p. 2091-2108. [3] European Medicine Agency, Summary of Product Characteristics - Alunbrig (brigatinib). [4] IMPIC Base: Contratos Públicos Online (https://www.base.gov.pt/Base4/pt/detalhe/?type=contratos&id=9275350 & https://www.base.gov.pt/Base4/pt/detalhe/?type=contratos&id=9326304). [5] Peters, S., et al., Alectinib versus Crizotinib in Untreated ALK-Positive Non–Small-Cell Lung Cancer. New England Journal of Medicine, 2017. 377(9): p. 829-838. [6] Mok, T., et al., Updated overall survival and final progression-free survival data for patients with treatment-naive advanced ALK-positive non-small-cell lung cancer in the ALEX study. Ann Oncol, 2020. 31(8): p. 1056-1064. [7] European Medicine Agency, Summary of Product Characteristics - Alecensa (alectinib). [8] Pérol, M., et al., Patient-reported outcomes from the randomized phase III ALEX study of alectinib versus crizotinib in patients with ALK-positive non-small-cell lung cancer. Lung Cancer, 2019. 138: p. 79-87.

ISPOR Europe 2023 | 12-15 November 2023 | Copenhagen, Denmark

Abstract ID: 129930