Feasibility of using Finnish Real-World Data to build an External Control Arm for the Factor XIa inhibitor asundexian PACIFIC-AF trial

James Potts, Kaisa Laapas, Igor Khorlo, Petri Niemela, Sanna Strom, Liisa Partanen, Samu Kurki, Thomas Viethen, Khaled Abdelgawwad, Jussi Leinonen

MSR 34

Objectives

Bayer has started a collaborative Future Clinical Trials (FCT) project in Finland with several cooperation bodies to develop and test novel approaches for clinical trials. One of the first use cases focused on the capability to use the Finnish healthcare data as a source for creation of an External Control Arm (ECA). Based on the PACIFIC-AF trial[1], a randomized phase II study with asundexian, it was explored whether this ECA could have successfully replaced the Internal Control Arm (ICA).

Methods

PACIFIC-AF trial design

The PACIFIC-AF trial was a randomized active comparator-controlled phase II clinical trial with two investigational treatment arms and one arm (n=250) as the ICA treated with apixaban. At the end of 12 weeks, the primary endpoint of composite ISTH major or clinically relevant non-major bleeding was evaluated in each treatment arm; additionally, different CV events making up the primary composite endpoint were individually assessed.

Finnish Real-World Data (F-RWD)

Finnish Healthcare records fulfilling the RCTs eligibility criteria between January 1, 2013 through September 30, 2019 were used to build an ECA. Additionally, baseline data was collected for one year before the inclusion (baseline for hypertension extended to cover five years before the inclusion and two years for weight/BMI) and followed-up maximally up to December 31, 2020.

Statistical Analysis

Various candidate propensity score (PS) models for the probability of being in the ICA were evaluated using all available known (‘category A’) and possible/probable (‘category B’) confounders. Optimal 1:1 matching with a common support region were used to obtain sufficient overlap. Models with only category A confounders, all category A and B confounders, and category A confounders with forward and backward stepwise selection were compared using the standardized mean difference (SMD) of each confounder; the model with lower SMDs and a substantial number of confounders being well-balanced was chosen as the final model. Sensitivity analyses were explored for improved overlap and SMD[2].

Results

Total of N=8255 from the F-RWD were available; among these, n=3952 were treated with apixaban. Out of these, n=1763 met RCT eligibility criteria, including AF diagnosis, and thus made up the F-RWD subpopulation used for building the ECA. The selected ECA consisted of n=239 participants and balanced across 28 category A confounders, with the largest discrepancy between ICA and the selected ECA being prior use of heparins (22.0% vs 27.6%), respectively (Table 1). Descriptive comparisons between ICA and selected ECA for bleeding and cardiovascular outcomes at 3 months is shown in Table 2, with overlap in all exposure-adjusted incidence rates (EAIR) for all outcomes with 3 or more events. Other weighting/matching approaches proved superior with regards to the SMDs of confounders and the number of subjects matched in the ECA (Figure 1).

Table 1: Known Confounders of Outcome Events by RCT, F-RWD, and selected ECA

Confounder	Parameter	RWD N= 1763	Selected ECA N= 239	RCT N= 250
Age	n	1763	239	250
	Mean (SD)	77.54 (8.921)	75.58 (8.042)	74.27 (8.317)
	Median	78	77	75
Age categorized (years)	≤ 64	105 (5.96%)	24 (10.04%)	34 (13.6%)
	65-69	232 (13.16%)	21 (8.79%)	32 (12.8%)
	70-74	315 (17.87%)	49 (20.5%)	53 (21.2%)
	75-79	344 (19.51%)	68 (28.45%)	61 (24.4%)
	80-84	337 (19.12%)	50 (20.92%)	45 (18.0%)
	85-89	284 (16.11%)	24 (10.04%)	21 (8.4%)
	≥ 90	146 (8.28%)	3 (1.26%)	4 (1.6%)
Sex	Male	804 (45.60%)	128 (53.56%)	141 (56.40%)
	Female	959 (54.40%)	111 (46.44%)	109 (43.60%)
BMI categorized	<= 30	1508 (85.54%)	167 (69.87%)	166 (66.40%)
	> 30	255 (14.46%)	72 (30.13%)	84 (33.60%)
Low body weight	>= 60kg	1593 (90.36%)	210 (87.87%)	221 (88.40%)
	< 60kg	170 (9.64%)	29 (12.13%)	29 (11.60%)
Smoking	No	1632 (92.57%)	225 (94.14%)	240 (96.00%)
	Yes	131 (7.43%)	14 (5.86%)	10 (4.00%)
Time since atrial fibrillation (days)	<= 30	354 (20.08%)	92 (38.49%)	99 (39.60%)
	>30-<90	107 (6.07%)	10 (4.18%)	16 (6.40%)
	>= 90	1302 (73.85%)	137 (57.32%)	135 (54.00%)
Arterial hypertension	No	523 (29.67%)	31 (12.97%)	30 (12.00%)
	Yes	1240 (70.33%)	208 (87.03%)	220 (88.00%)
Hyperlipidemia	No	1458 (82.70%)	158 (66.11%)	158 (63.20%)
	Yes	305 (17.30%)	81 (33.89%)	92 (36.80%)
Chronic heart failure	No	1313 (74.48%)	122 (51.05%)	133 (53.20%)
	Yes	450 (25.52%)	117 (48.95%)	117 (46.80%)
Diabetes mellitus	No	1302 (73.85%)	158 (66.11%)	163 (65.20%)
	Yes	461 (26.15%)	81 (33.89%)	87 (34.80%)
Coronary artery disease	No	1353 (76.74%)	191 (79.92%)	200 (80.00%)
	Yes	410 (23.26%)	48 (20.08%)	50 (20.00%)
Chronic kidney disease	No	1438 (81.57%)	199 (83.26%)	209 (83.60%)
	Yes	325 (18.43%)	40 (16.74%)	41 (16.40%)
Percutaneous coronary intervention	No	1645 (93.31%)	225 (94.14%)	235 (94.00%)
	Yes	118 (6.69%)	14 (5.86%)	15 (6.00%)
Myocardial infarction	No	1655 (93.87%)	217 (90.79%)	214 (85.60%)
	Yes	108 (6.13%)	22 (9.21%)	36 (14.40%)
Anemia	No	1287 (73.00%)	203 (84.94%)	224 (89.60%)
	Yes	476 (27.00%)	36 (15.06%)	26 (10.40%)
History of stroke	No	1571 (89.11%)	214 (89.54%)	230 (92.00%)
	Yes	192 (10.89%)	25 (10.46%)	20 (8.00%)
TIA	No	1674 (94.95%)	223 (93.31%)	237 (94.80%)
	Yes	89 (5.05%)	16 (6.69%)	13 (5.20%)
History of ISTH major bleeding	No	1612 (91.44%)	217 (90.79%)	228 (91.20%)
	Yes	151 (8.56%)	22 (9.21%)	22 (8.80%)
Carotid endarterectomy or stent	No	>1758 (>99.7%)	>234 (>97.9%)	>245 (>98.0%)
	Yes	<5 (<0.28%)	<5 (<2.09%)	<5 (<2.00%)
Peripheral arterial disease	No	1697 (96.26%)	227 (94.98%)	230 (92.00%)
	Yes	66 (3.74%)	12 (5.02%)	20 (8.00%)
Serum creatinine >= 1.5mg/dL	No	1487 (84.34%)	209 (87.45%)	223 (89.20%)
	Yes	276 (15.66%)	30 (12.55%)	27 (10.80%)
Malignancy	No	1515 (85.93%)	196 (82.01%)	205 (82.00%)
	Yes	248 (14.07%)	43 (17.99%)	45 (18.00%)
Prior or concomitant use of SSRIs	No	1653 (93.76%)	233 (97.49%)	244 (97.60%)
	Yes	110 (6.24%)	6 (2.51%)	6 (2.40%)
Use of proton pump inhibitors	No	930 (52.75%)	134 (56.07%)	141 (56.40%)
	Yes	833 (47.25%)	105 (43.93%)	109 (43.60%)
Non-steroidal anti-inflammatory drugs	No	1344 (76.23%)	213 (89.12%)	232 (92.80%)
	Yes	419 (23.77%)	26 (10.88%)	18 (7.20%)
Platelet aggregation inhibitors	No	1599 (90.70%)	221 (92.47%)	234 (93.60%)
	Yes	164 (9.30%)	18 (7.53%)	16 (6.40%)
Prior use of heparins	No	1157 (65.63%)	173 (72.38%)	195 (78.00%)
	Yes	606 (34.37%)	66 (27.62%)	55 (22.00%)
Prior use of NOACS	No	690 (39.14%)	98 (41.00%)	104 (41.60%)
	Yes	1073 (60.86%)	141 (59.00%)	146 (58.40%)

Table 2: Number of Subjects with Events and Exposure-Adjusted Incidence Rates at 3 Months

	F-RWD (N=1763)		Selected ECA (N=239)		RCT (N=250)	
Endpoint	Number of subjects with event n (%)	EAIR (90% CI)	Number of subjects with event n (%)	EAIR (90% CI)	Number of subjects with event n (%)	EAIR (90% CI)
Composite of ISTHMB, CRNMB	47 (2.67)	11.18 (8.64, 13.99)	9 (3.7%)	16.17 (8.44, 25.94)	6 (2.4%)	11.10 (4.86, 19.45)
ISTHMB	16 (0.91)	3.76 (2.36, 5.43)	2		0	
CRNMB	36 (2.04)	8.54 (6.34, 11.01)	8 (3.35%)	14.31 (7.12, 23.52)	6 (2.4%)	11.10 (4.86, 19.45)
Myocardial infarction	35 (1.99)	8.3 (6.13, 10.73)	7 (2.93%)	12.48 (5.86, 21.11)	0	
Ischemic stroke	38 (2.16)	9 (6.74, 11.53)	1		0	
Systemic embolism	1 (0.06)		0		0	
Composite of IS, SE, CV-death	63 (3.57)	14.93 (11.97, 18.15)	4 (1.67%)	7.00 (2.39, 13.57)	3 (1.2)	5.27 (1.44, 11.06)
Composite of IS, SE, MI, CV-death	96 (5.45)	23.05 (19.32, 27.05)	11 (4.6%)	19.67 (11.03, 30.34)	3 (1.2)	5.27 (1.44, 11.06)
CV-death	26 (1.47)	6.08 (4.26, 8.17)	3 (1.26%)	5.23 (1.43, 10.98)	3 (1.2)	5.22 (1.42, 10.95)

ISTHMB = ISTH Major bleeding
CRNMB = Clinically relevant non-major bleeding

Figure 1: Sensitivity of SMD for each known confounder by matching and weighting methods

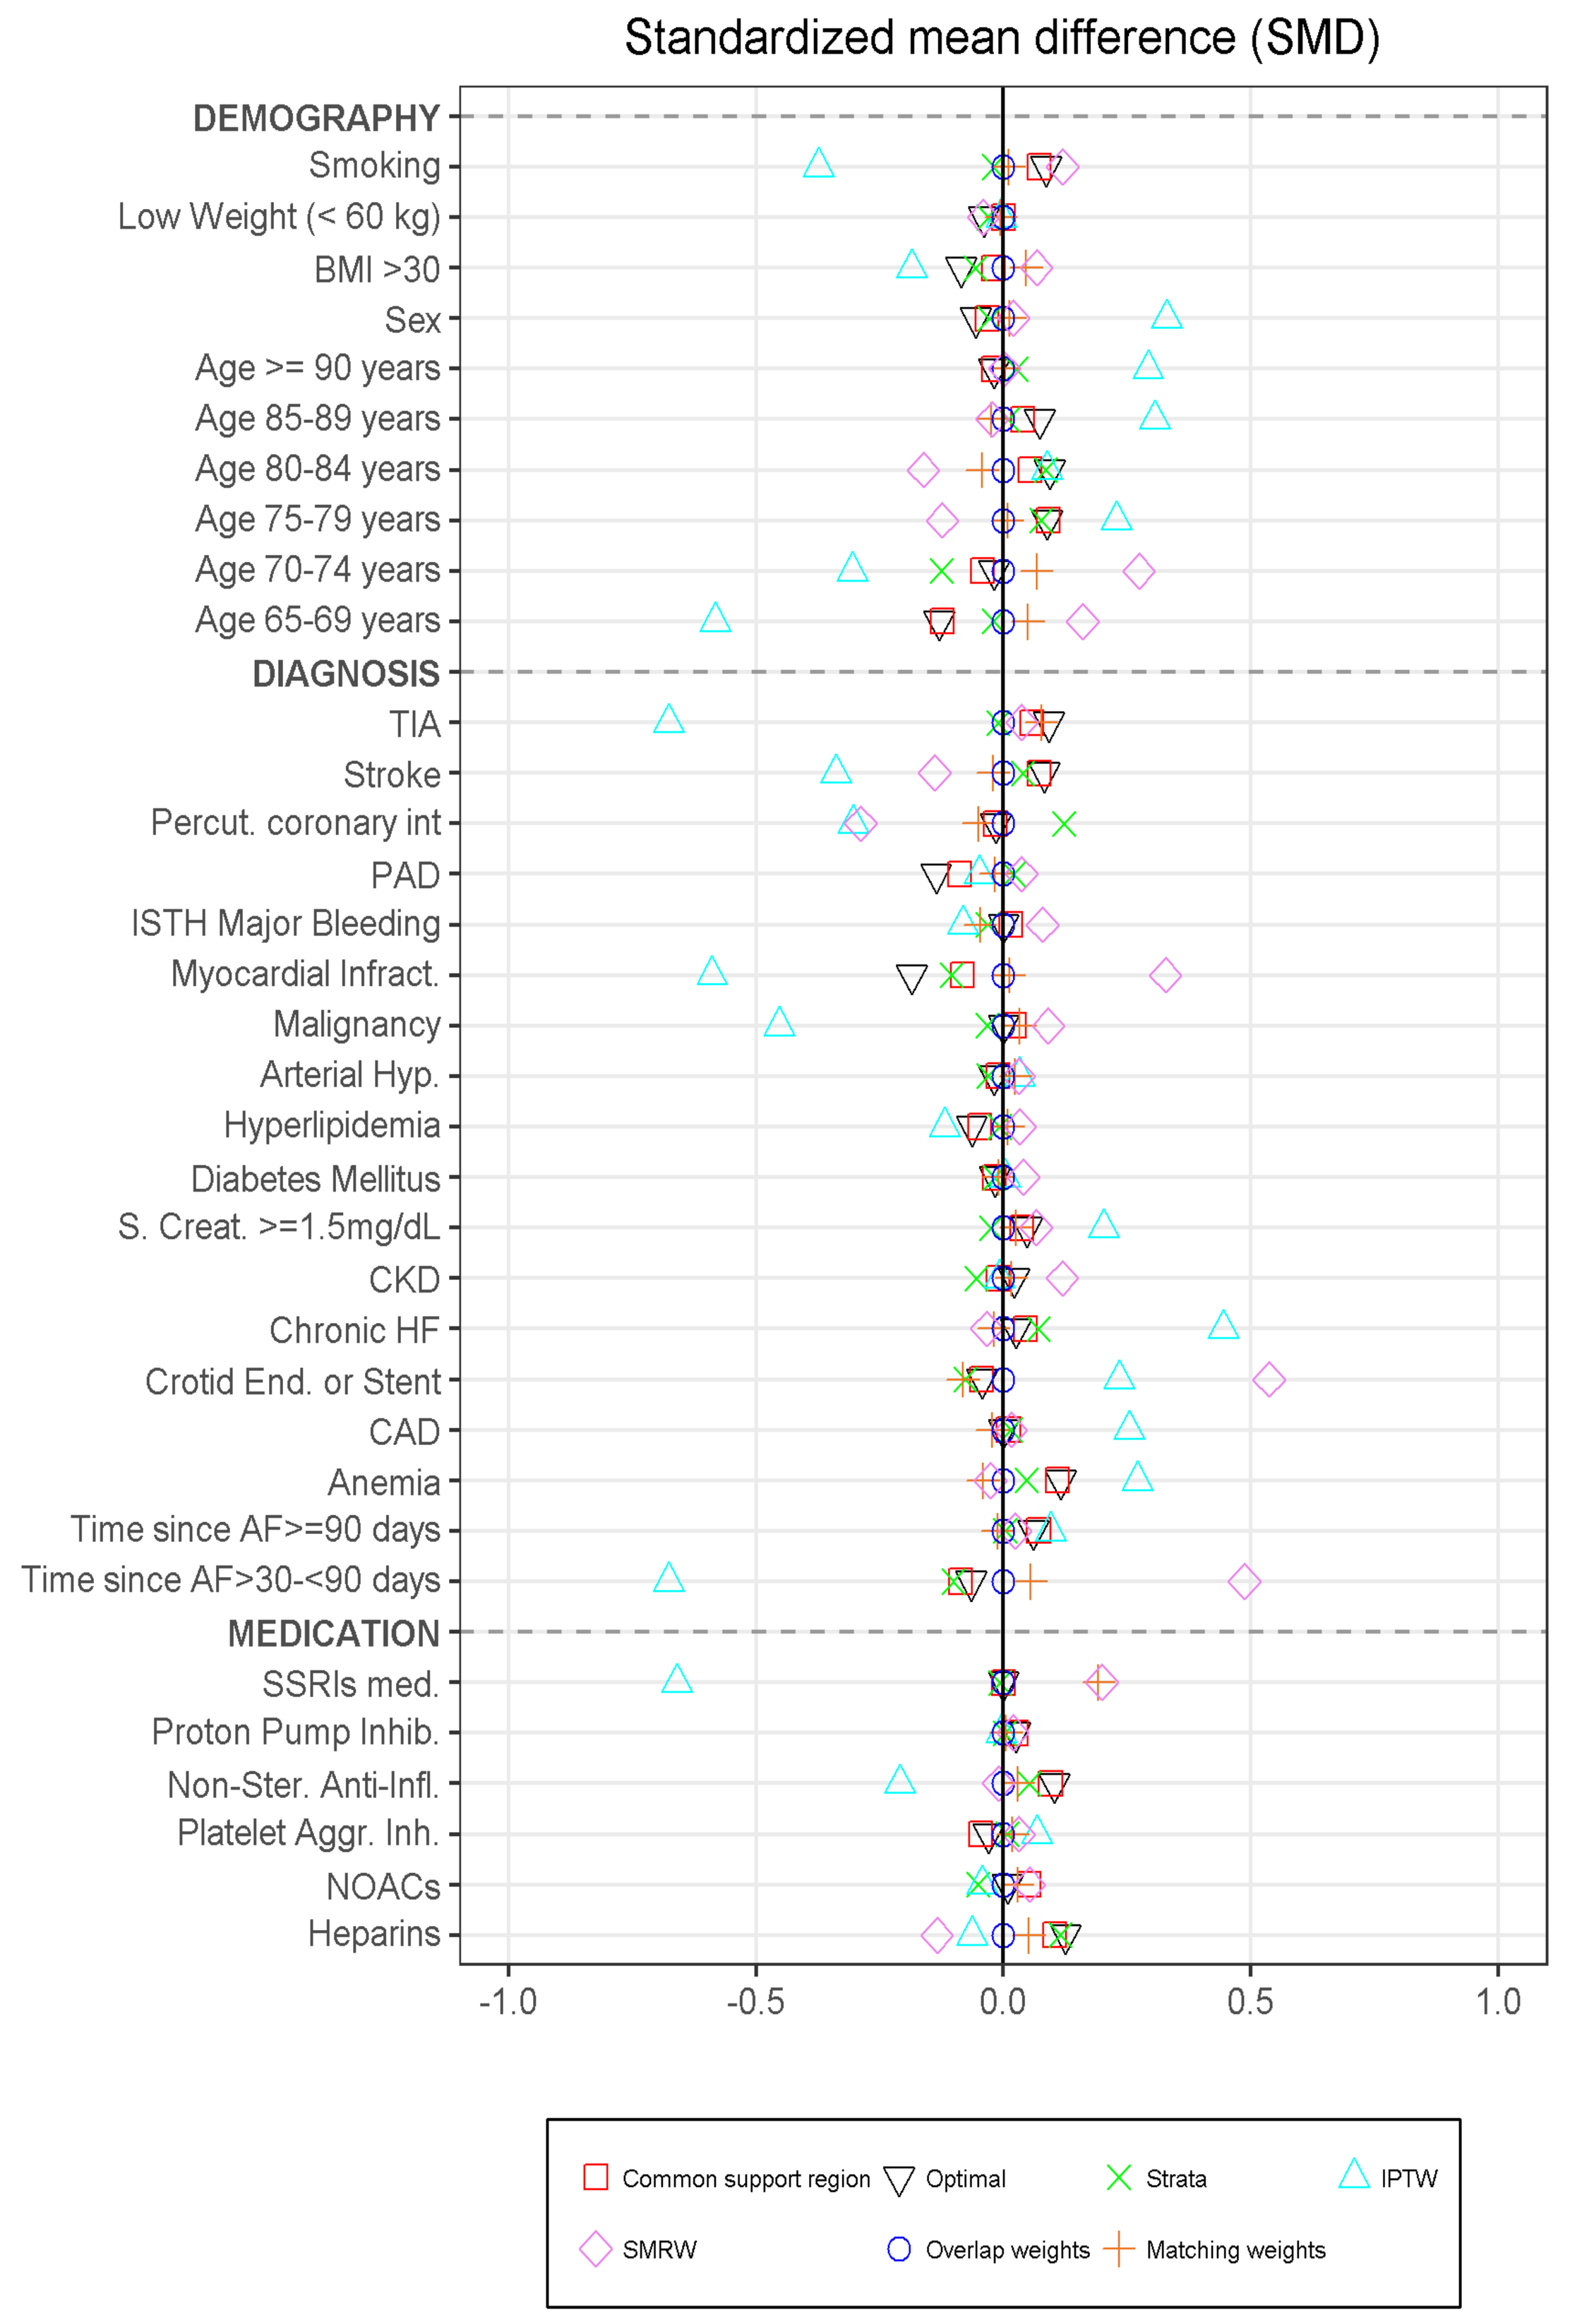
Standardized mean difference (SMD)
DEMOGRAPHY
Smoking
Low Weight (< 60 kg)
BMI >30
Sex
Age >= 90 years
Age 85-89 years
Age 80-84 years
Age 75-79 years
Age 70-74 years
Age 65-69 years
DIAGNOSIS
TIA
Stroke
Percut. coronary int
PAD
ISTH Major Bleeding
Myocardial Infract.
Malignancy
Arterial Hyp.
Hyperlipidemia
Diabetes Mellitus
S. Creat. >=1.5mg/dL
CKD
Chronic HF
Crotid End. or Stent
CAD
Anemia
Time since AF>=90 days
Time since AF>30-<90 days
MEDICATION
SSRIs med.
Proton Pump Inhib.
Non-Ster. Anti-Infl.
Platelet Aggr. Inh.
NOACs
Heparins
-1.0
-0.5
0.0
0.5
1.0
Common support region
Optimal
Strata
IPTW
SMRW
Overlap weights
Matching weights

IPTW = Inverse probability treatment weighting
SMRW = Standardized mortality ratio weighting

Conclusions

- The F-RWD was successfully used to build an ECA that showed baseline and outcome overlap with the PACIFIC-AF ICA.
- Sensitivity analyses showed improvement over the pre-specified primary approach
- A gate-keeping approach to PS-matching and/or weighting methods could be used to establish the most efficient ECA while maintaining requirement of pre-specification of methods in a regulatory setting[3].

References

1. Piccini, JP; Caso, V; Connolly, SJ; et al. Safety of the oral factor Xia inhibitor asundexian compared with apixaban in patients with atrial fibrillation (PACIFIC AF): a multicentre, randomised, double-blind, double-dummy, dose-finding phase 2 study. *The Lancet* April 2022, volume 399(10333).
2. Desai, R; Franklin, JM. Alternative approaches for confounding adjustment in observational studies using weighting based on the propensity score: a primer for practitioners. *BMJ* 2019;367:15657.
3. FDA. Considerations for the design and conduct of externally controlled trials for drug and biological products: Guidance for the industry (*DRAFT GUIDANCE*).