CROSS-SECTIONAL RESEARCH ON THE DEVELOPMENT OF CASE MANAGEMENT APPROACH FOR DISABILITY PATIENTS BASED ON THE WORK ABILITY INDEX AND QUALITY OF LIFE MEASURES

Arie Arizandi Kurnianto[1], Nemeskéri Zsolt[2], István Ágoston[1]

[1]Doctoral School of Health Sciences, University of Pécs, Pécs, Hungary, 7621
[4]Department of Cultural Theory and Applied Communication Sciences, Faculty of Cultural Studies, Teacher Training and Rural Development, University of Pécs, Pécs, Hungary, 7633

ISPOR Europe 2022 #ISPOREurope

UNIVERSITY OF PÉCS Faculty of Health Sciences

INTRODUCTION

The Return To Work (RTW) program, an in-kind benefit that provides employees with occupational accident insurance, is one of the novel approaches to case management now being developed for patients with disabilities. Within that concept, a complete rehabilitation program, including medical, vocational, and psychological rehabilitation, has potentially increased the workability index and improved the quality of life for patients with disabilities.

OBJECTIVE

Analyzing the factors associated with improving the work ability index and quality of life in patients with disabilities participating in a return-to-work (RTW) program utilizing a case management system.

METHODS

A total of 154 patients, who are disabled workers administered in the Centre of Return To Work in Indonesia, participated in this study during the outbreaks of COVID-19. Utilizing descriptive statistics, the profile of RTW participants has been outlined. Utilizing specific sociodemographic and occupational factors, the association between RTW outcomes was examined. In this study, quality of life has been measured by a validated questionnaire of WHOQoL-BREF and Work Ability Index by WAI questionnaire from The Finnish Institute of Occupational Health. Meanwhile, correlation and logistic regression have been used to study the association between identified factors of quality of life and work ability index.

RESULTS

Domain Quality of life	Minimum	Maximum	Mean	Std. Deviation
Physical health	39.29	100.00	72.93	11.98
Psychological	41.67	100.00	74.35	10.16
Social Bound	33.33	100.00	76.14	14.48
Environment	43.75	87.50	69.89	8.67

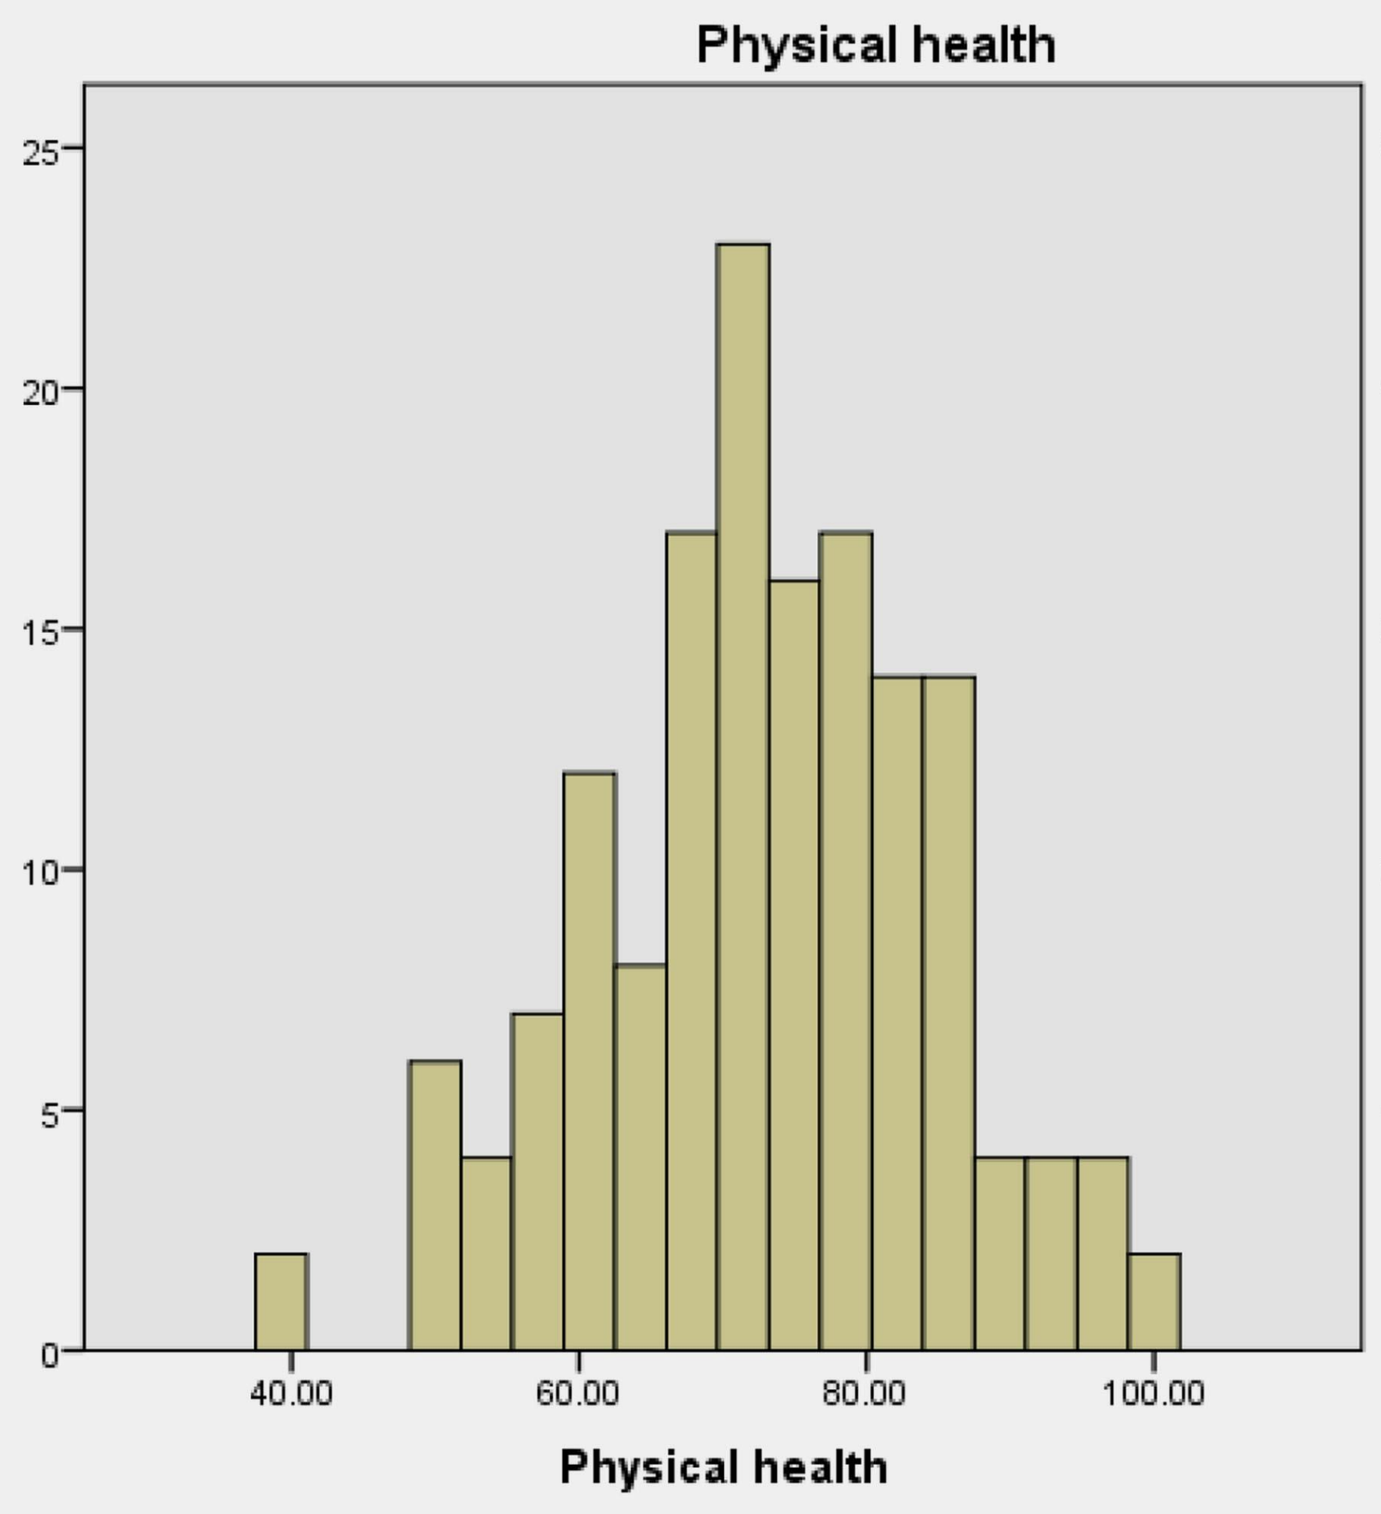
Physical health
40.00
60.00
80.00
100.00
Physical health

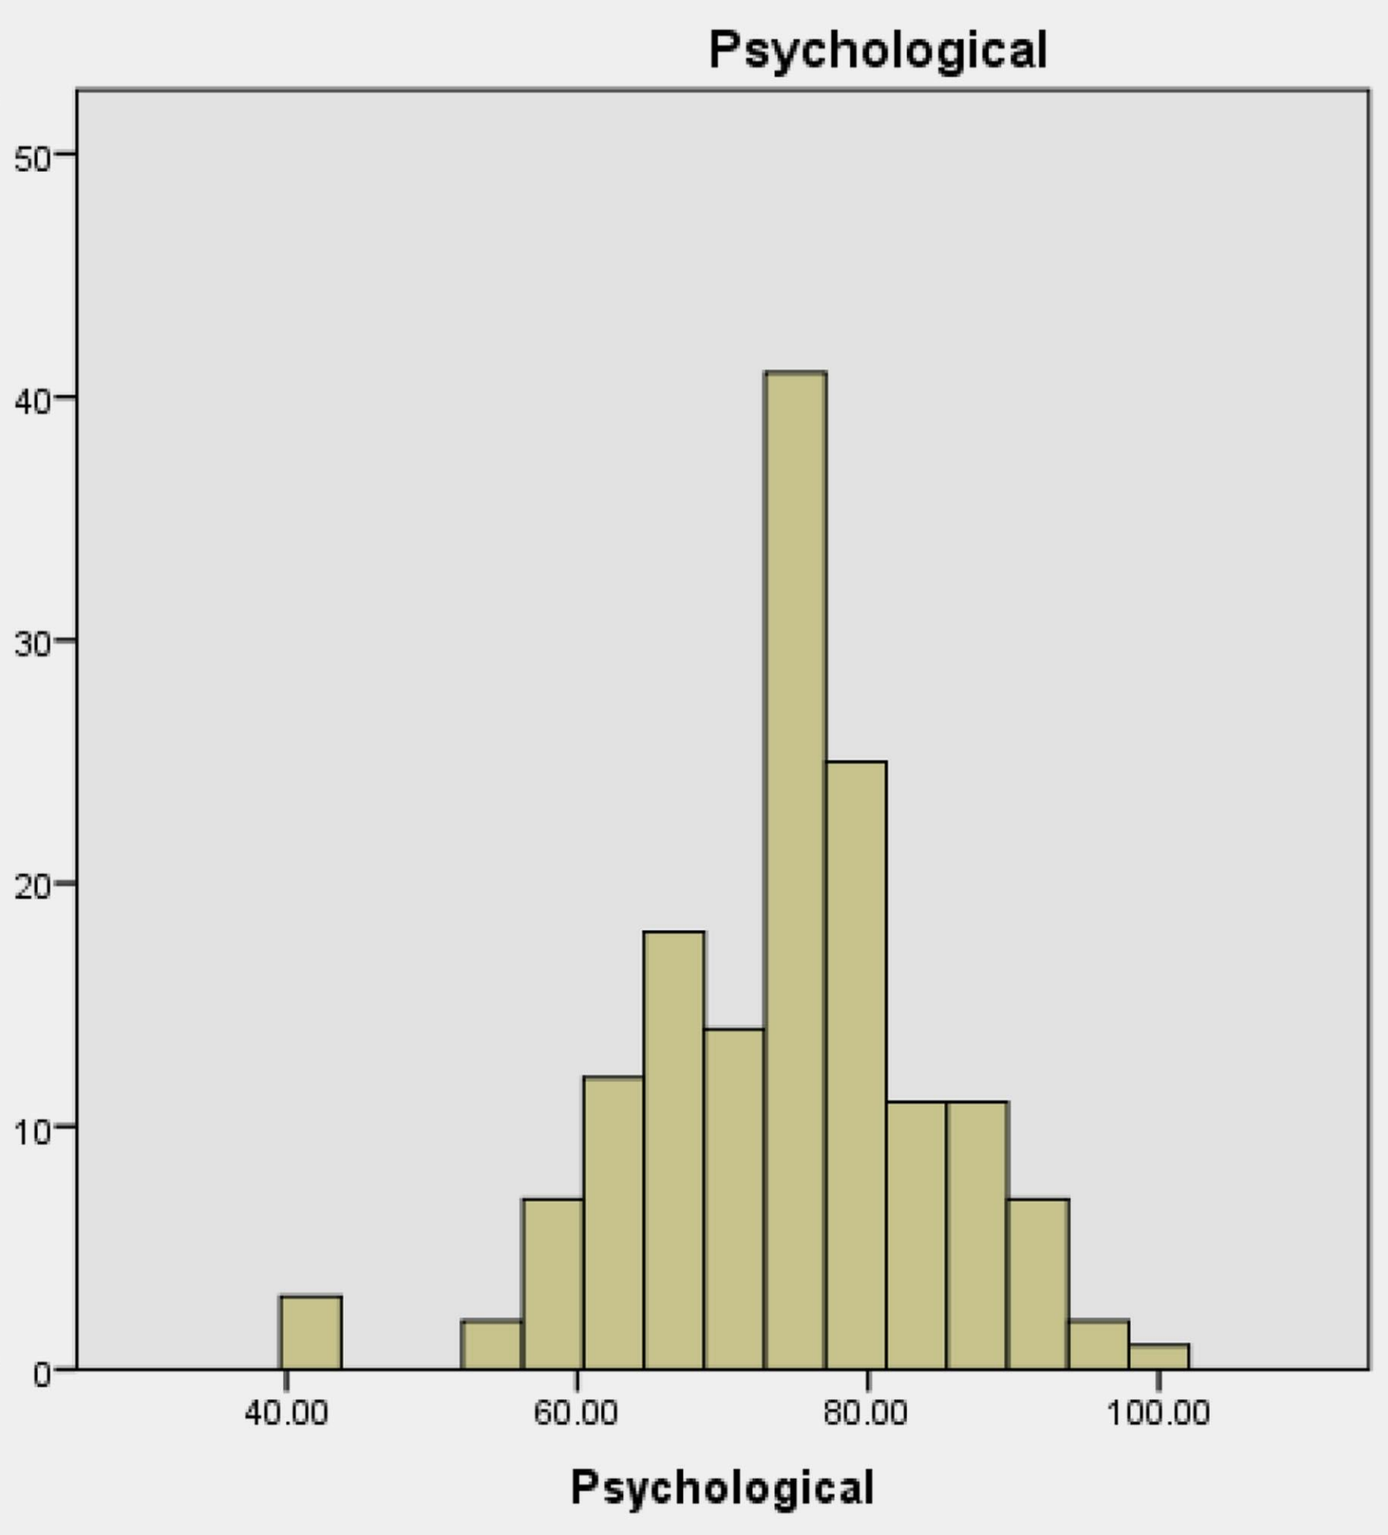
Psychological
40.00
60.00
80.00
100.00
Psychological

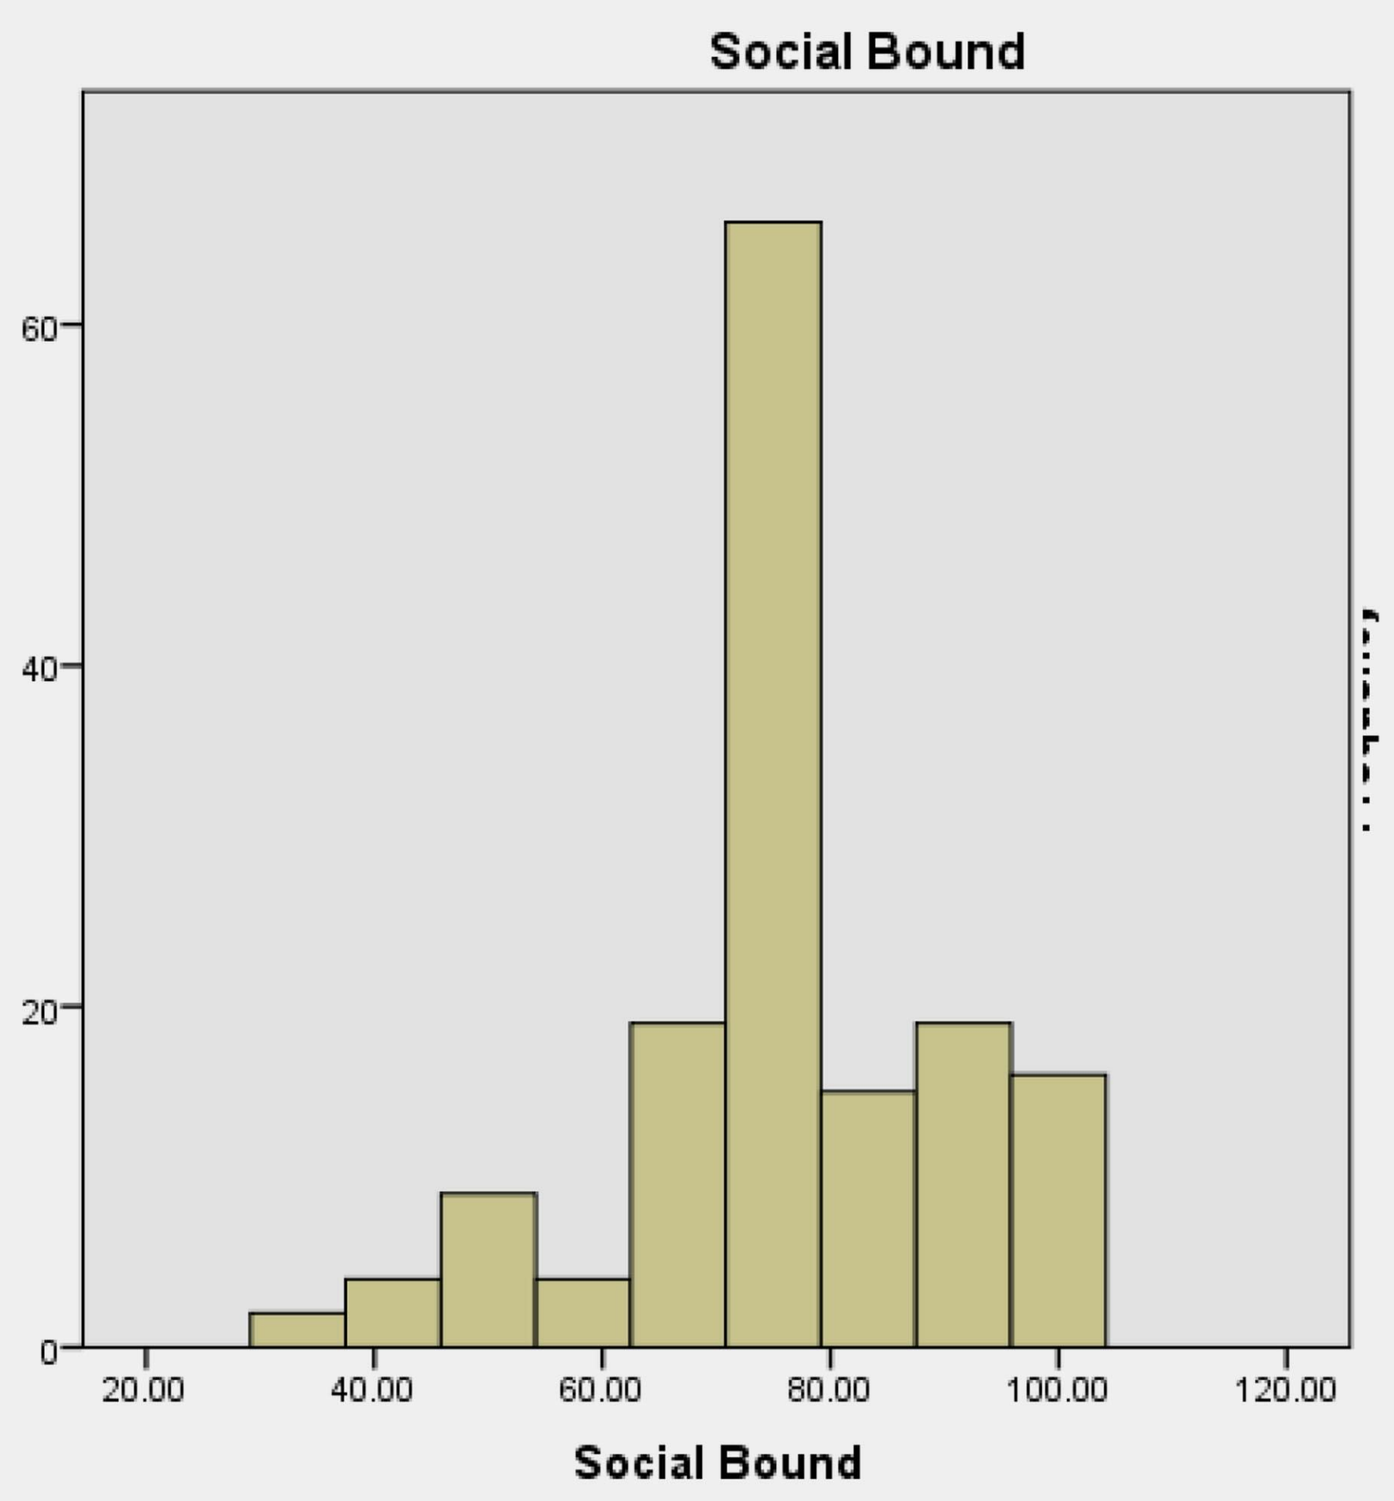
Social Bound
20.00
40.00
60.00
80.00
100.00
120.00
Social Bound

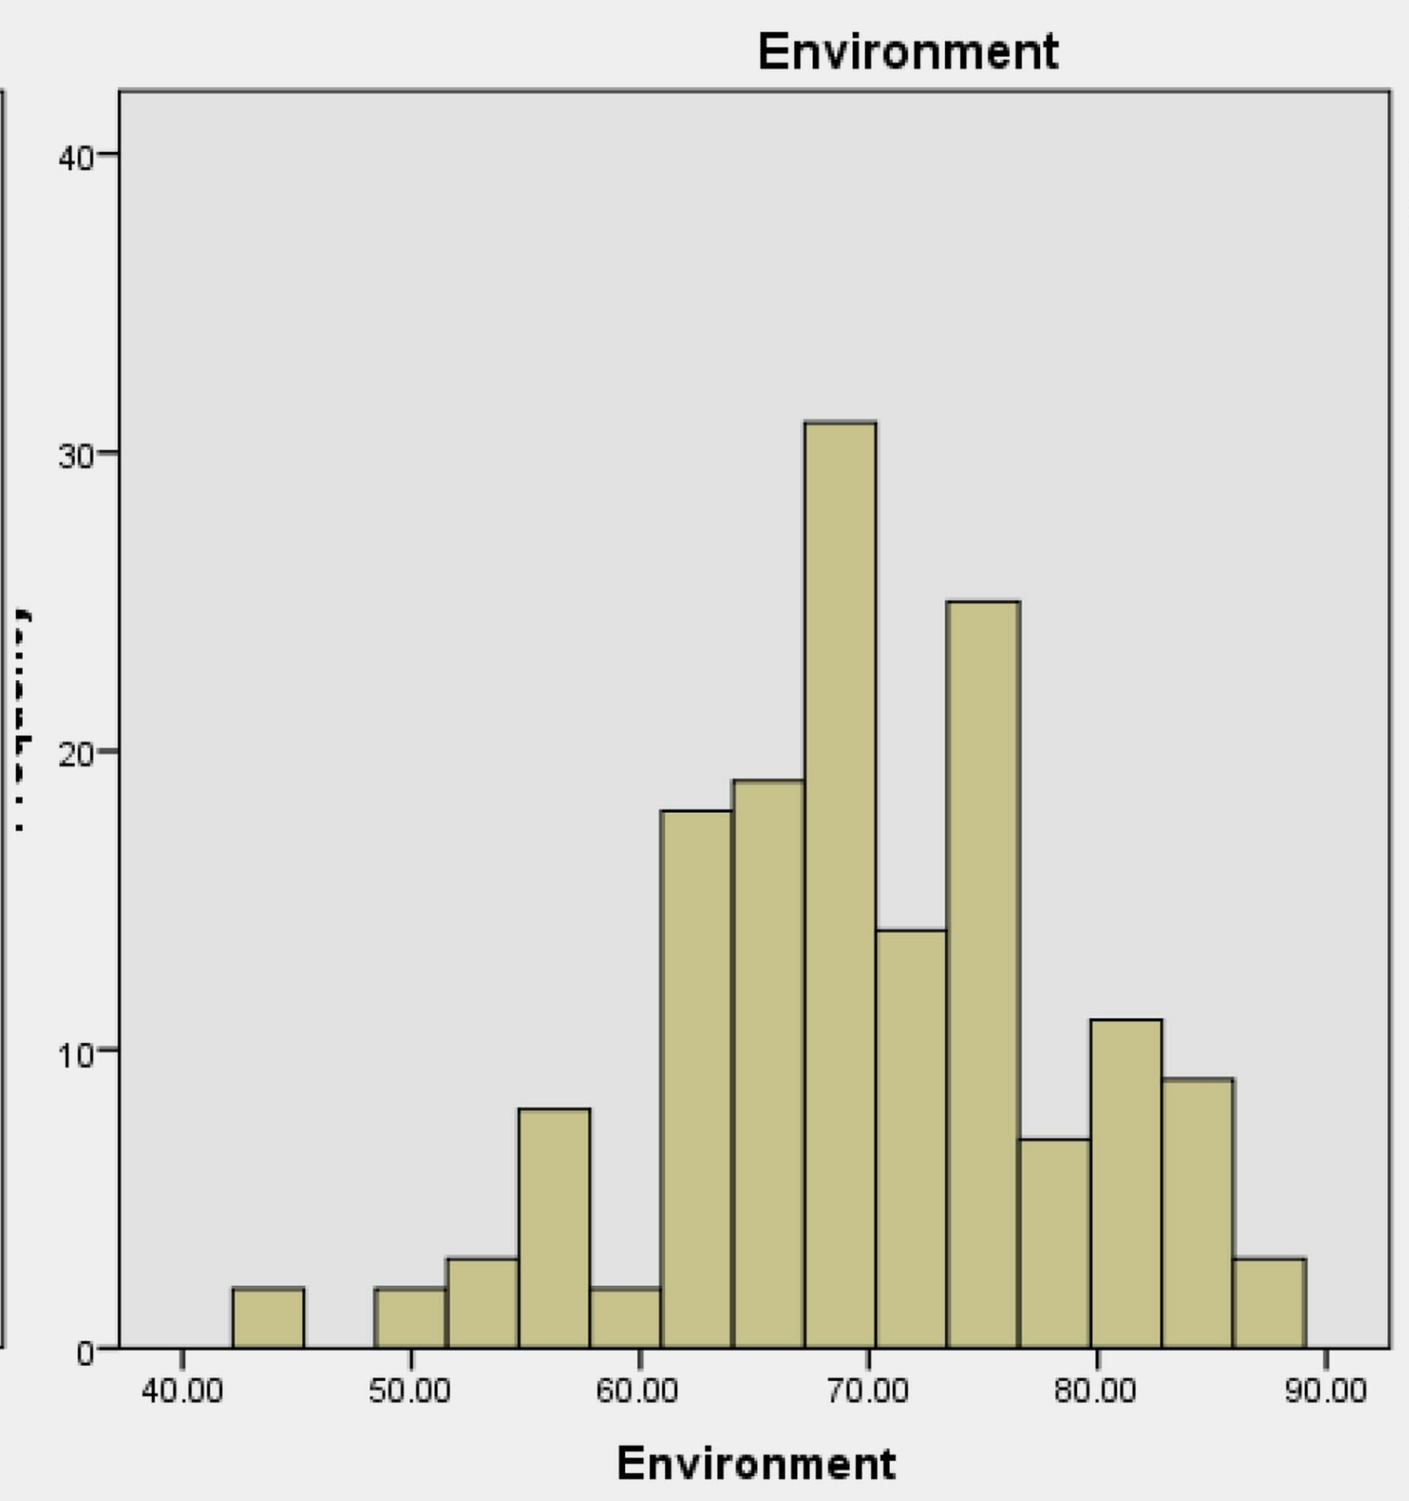
Environment
40.00
50.00
60.00
70.00
80.00
90.00
Environment

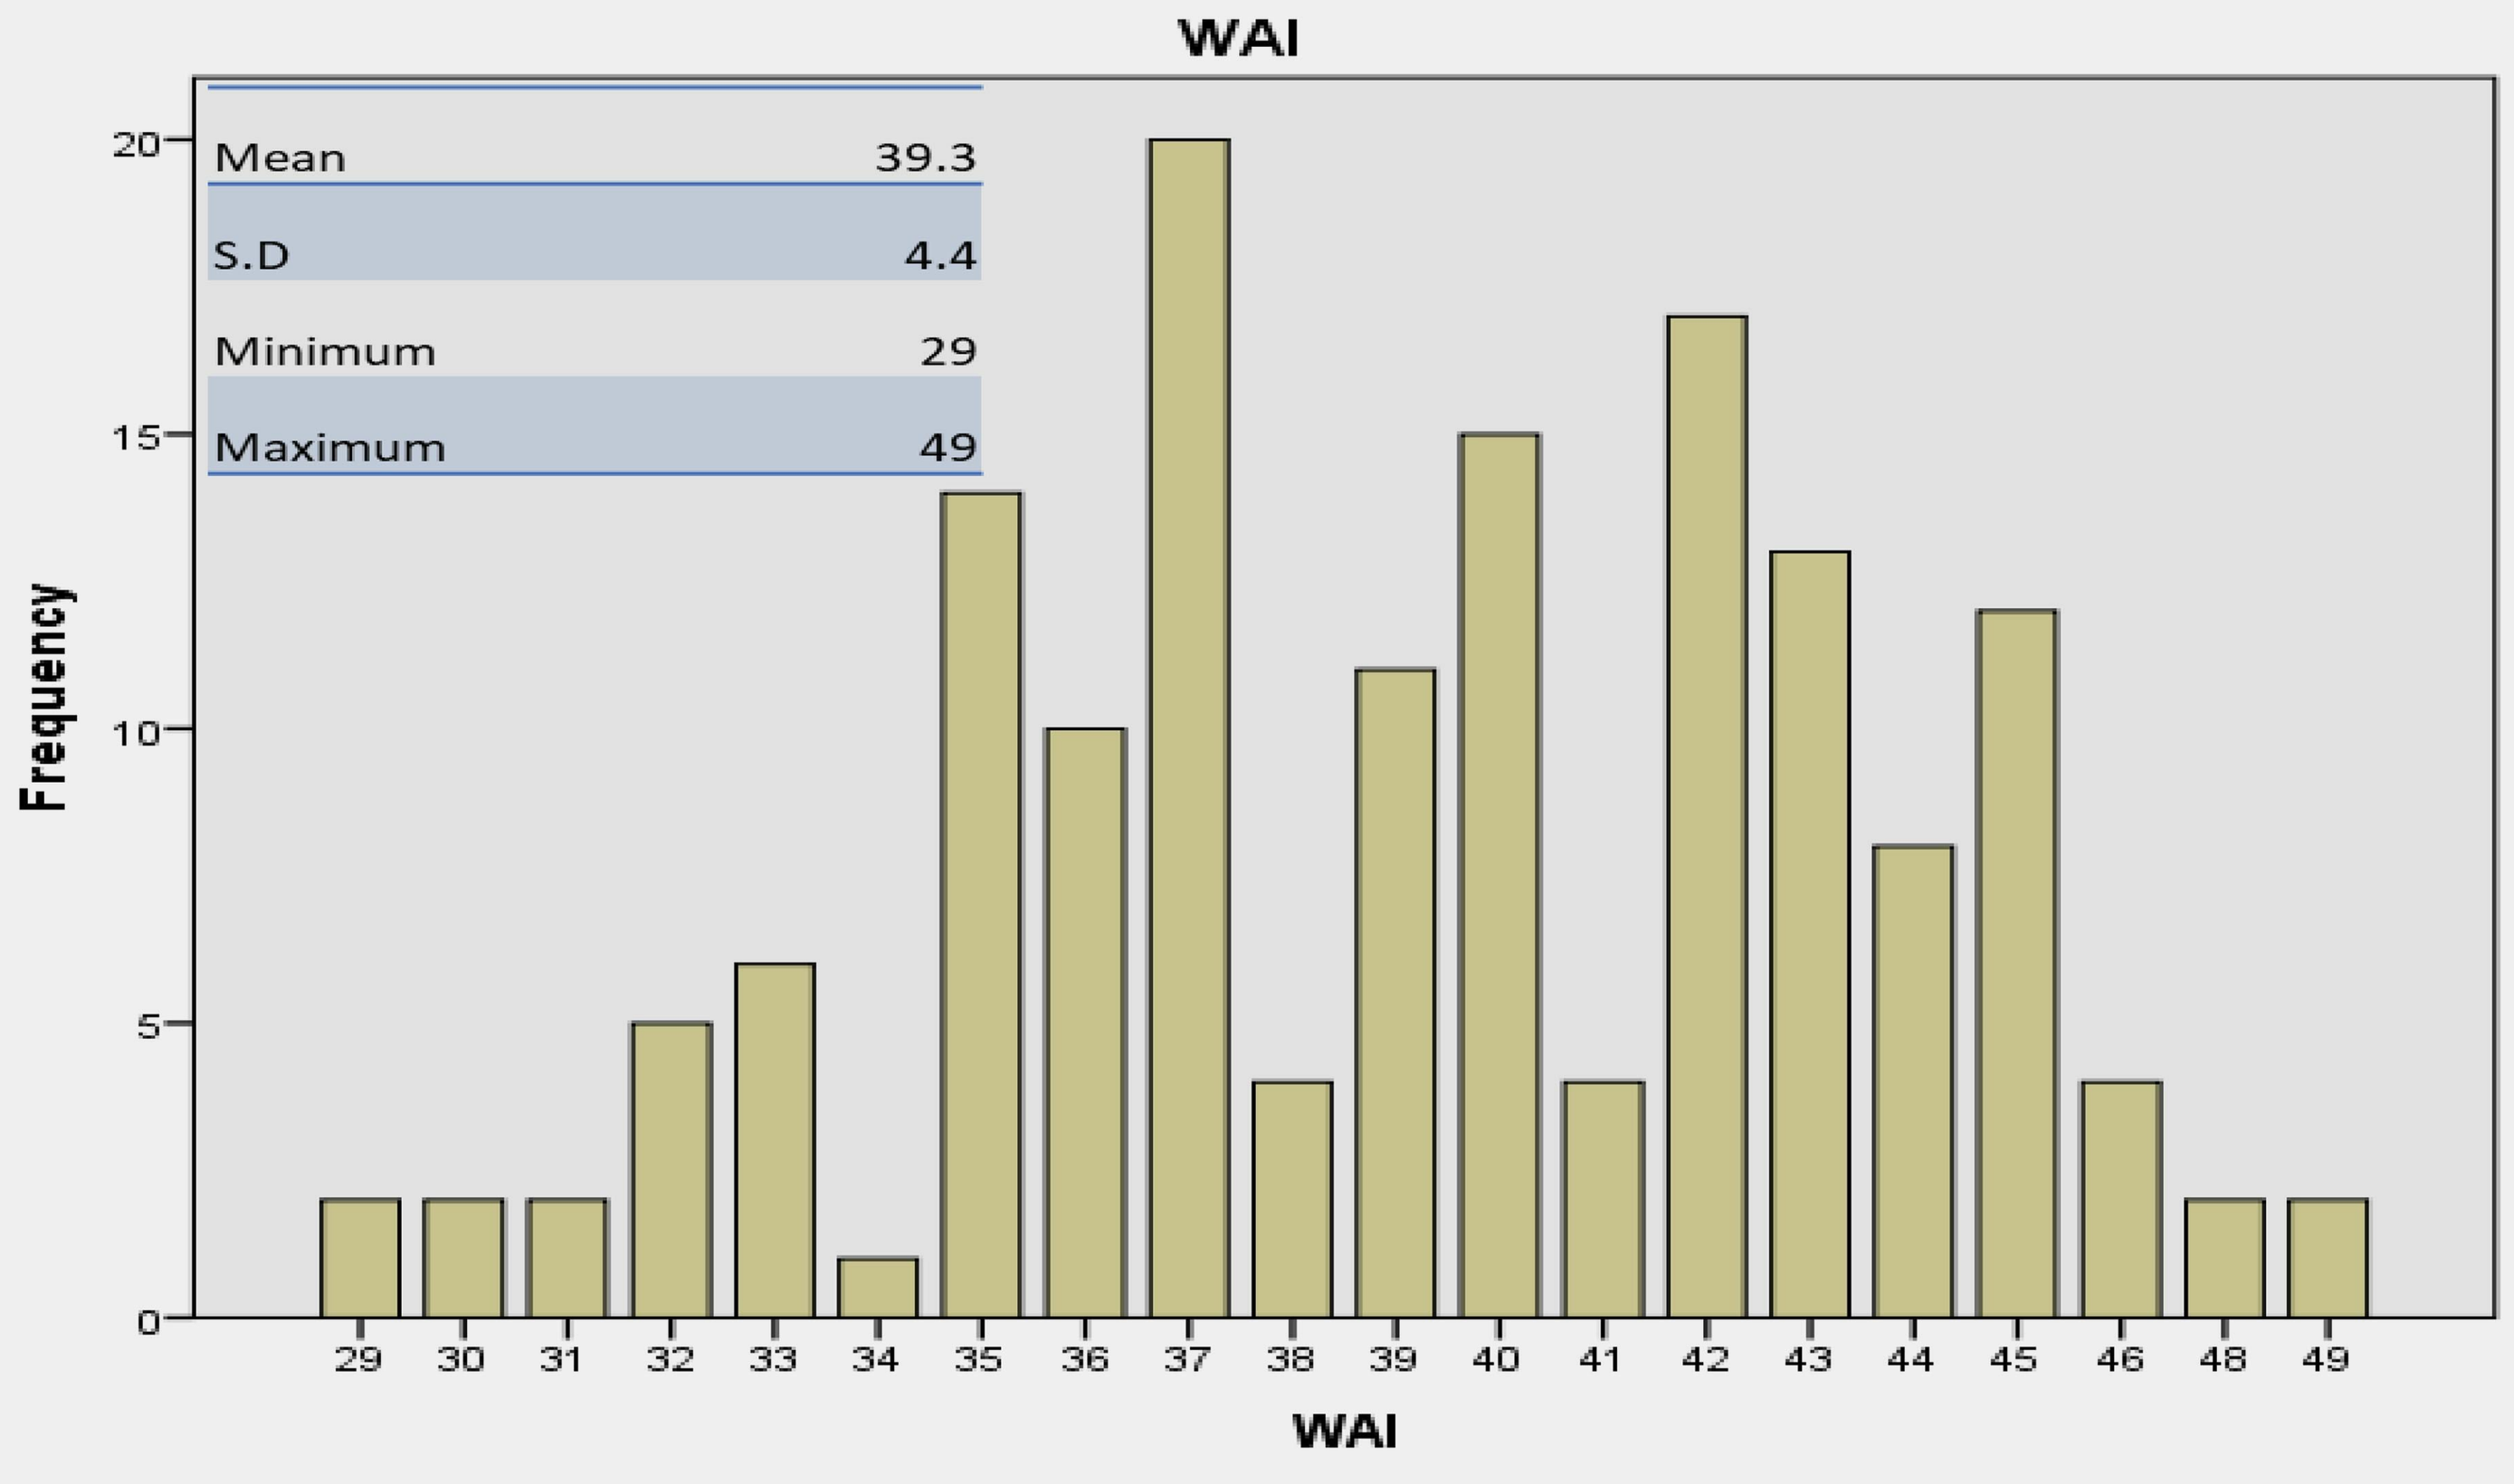
WAI
Mean 39.3
S.D 4.4
Minimum 29
Maximum 49
Frequency
29 30 31 32 33 34 35 36 37 38 39 40 41 42 43 44 45 46 48 49
WAI

The findings indicate that scores of physical health and social domain for quality of life among patients reach higher than other domains in this study which remain 72.93 and 76.14, respectively. However, patients' work ability index is generally considered moderate to excellent, with a mean score of 39.3(S.D 4.4). The level of education and all quality of life domains have proven statistically correlated with Work Ability Index (WAI). Moreover, variables of the level of education and physical health as the domain of quality of life significantly can explain factors associated with the work ability index of patients (p<0.05) who have been involved in the framework of case management, The Return To Work Program.

WAI	Frequency	Percent	Std. Error	95% Confidence Interval	
				Lower	Upper
Moderate	42	27.3	3.6	20.1	35.1
Good	84	54.5	4	46.8	63
Excellent	28	18.2	3.3	11.7	24.7

Age	Category of WAI			Total
	Moderate	Good	Excellent	
<40 y.o	34	56	19	109
	22.1%	36.4%	12.3%	70.8%
>40 y.o	8	28	9	45
	5.2%	18.2%	5.8%	29.2%

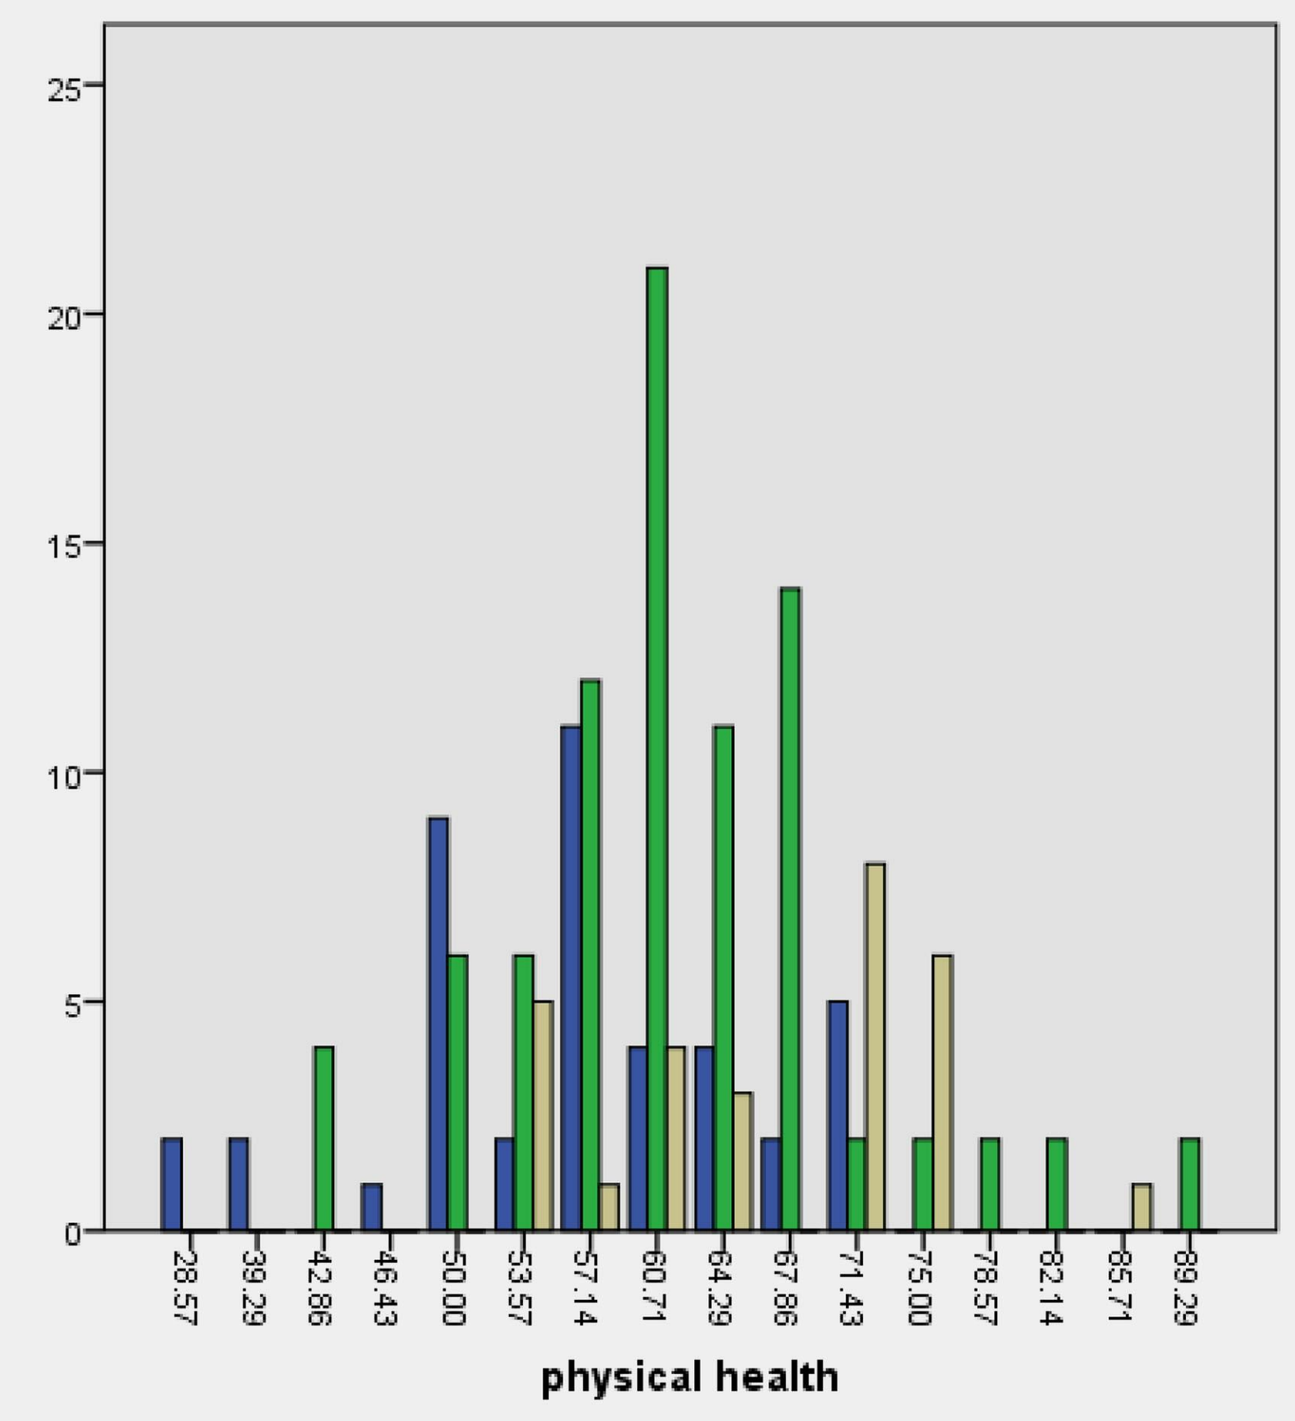
physical health

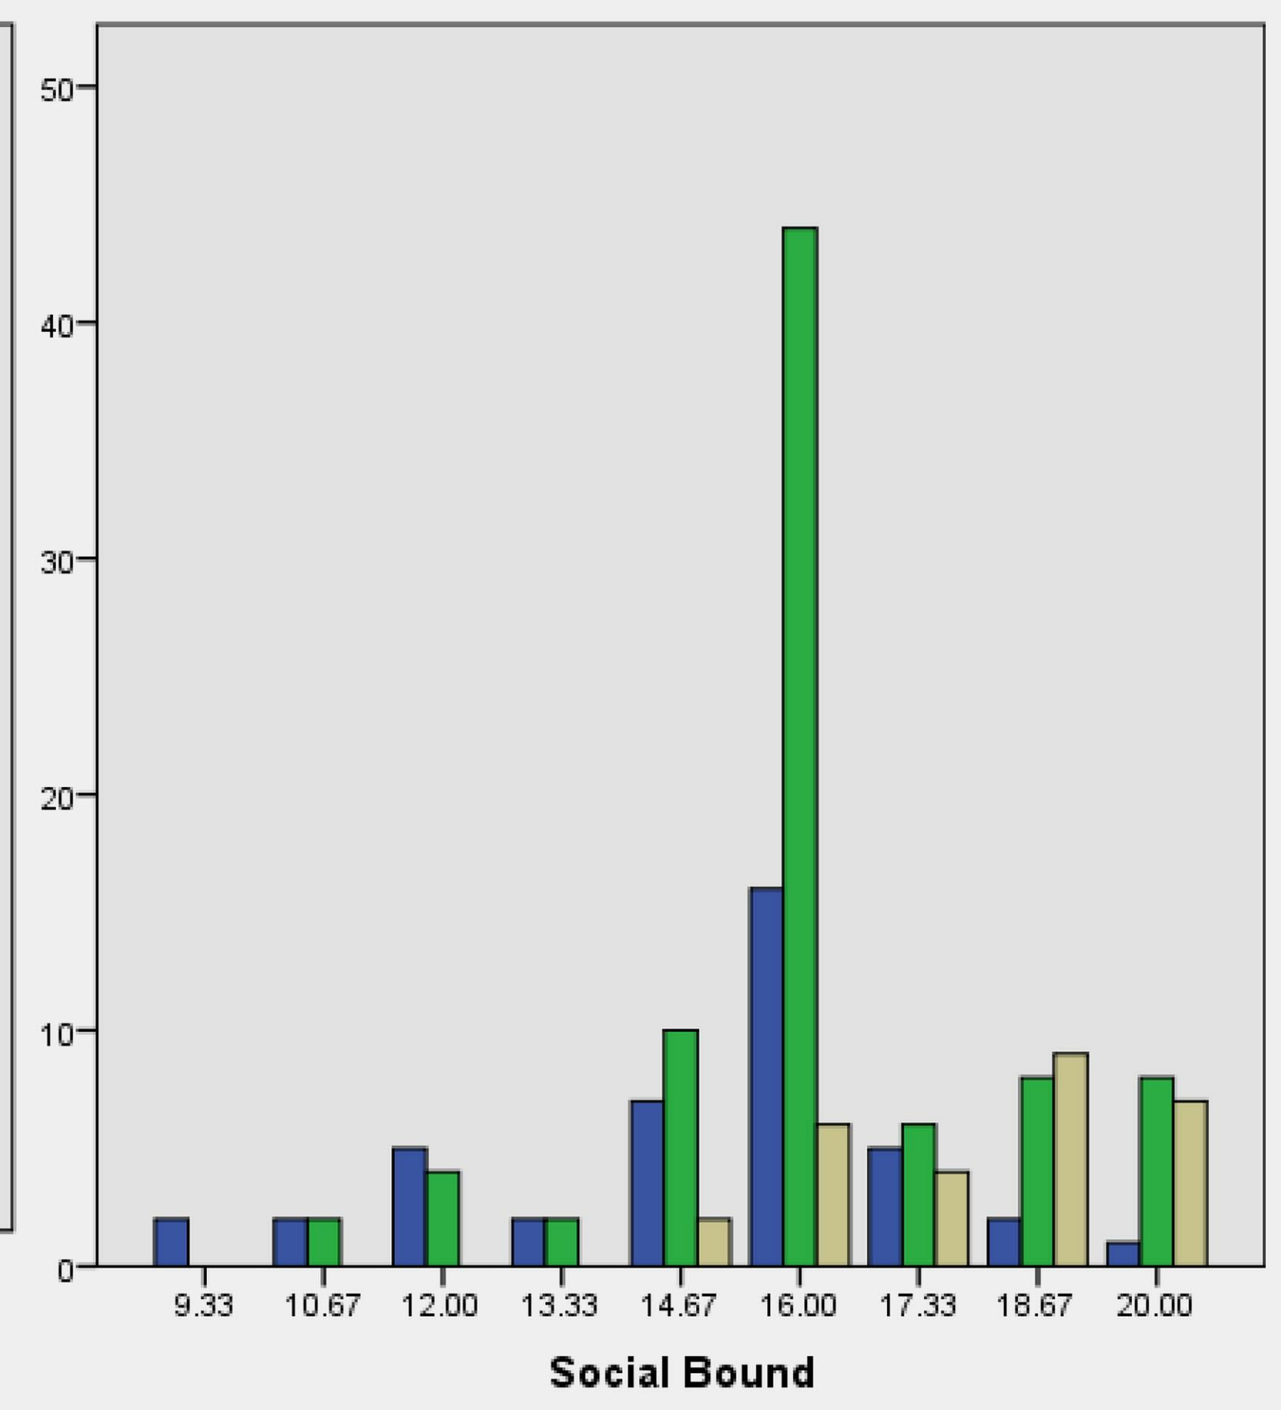
9.33
10.67
12.00
13.33
14.67
16.00
17.33
18.67
20.00
Social Bound

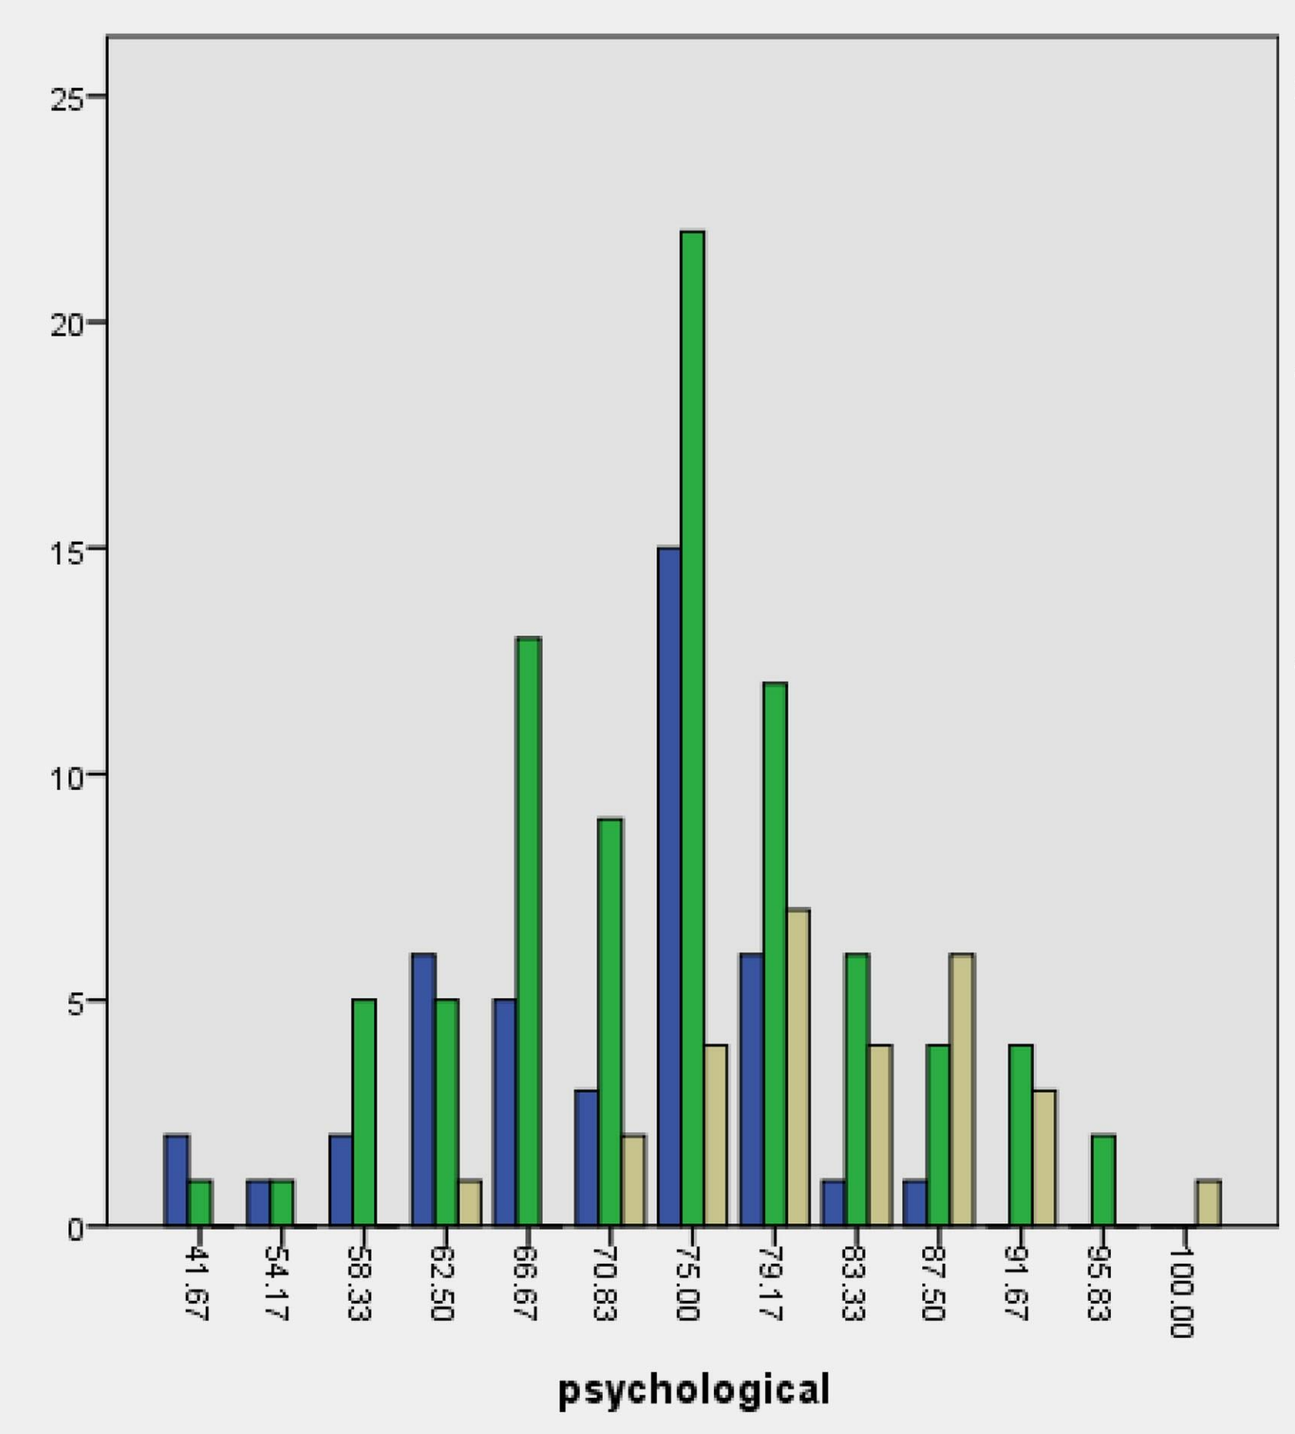
psychological

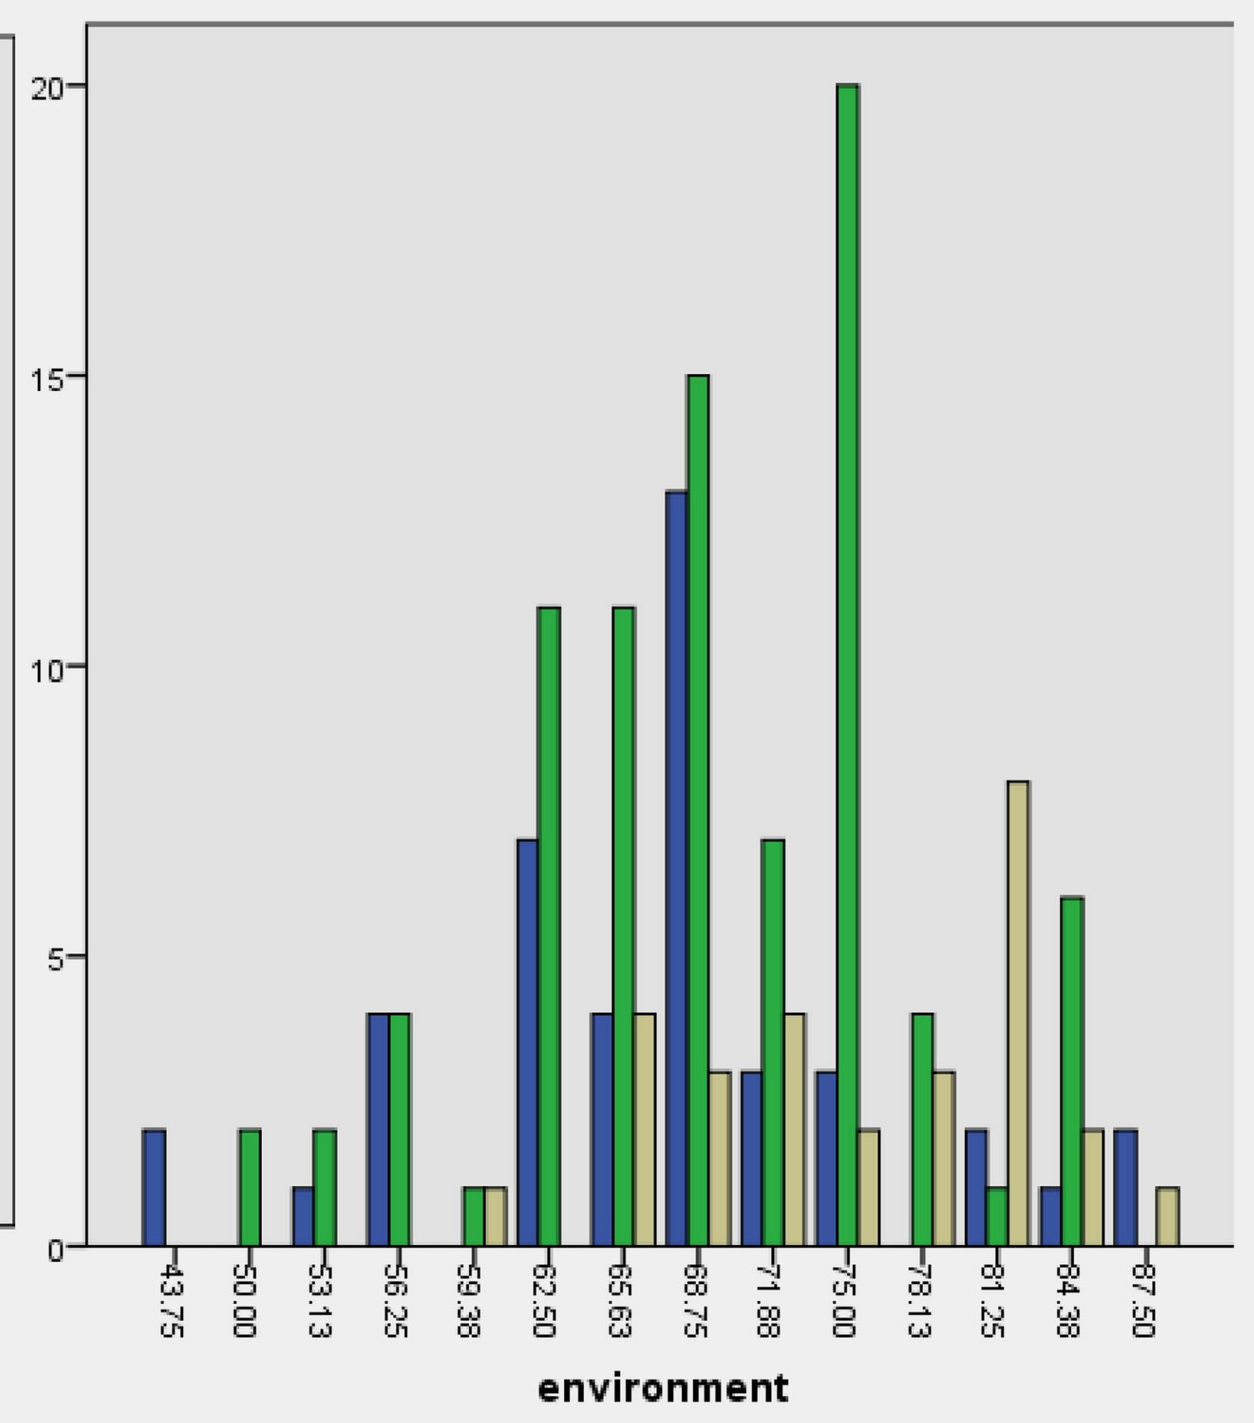
environment

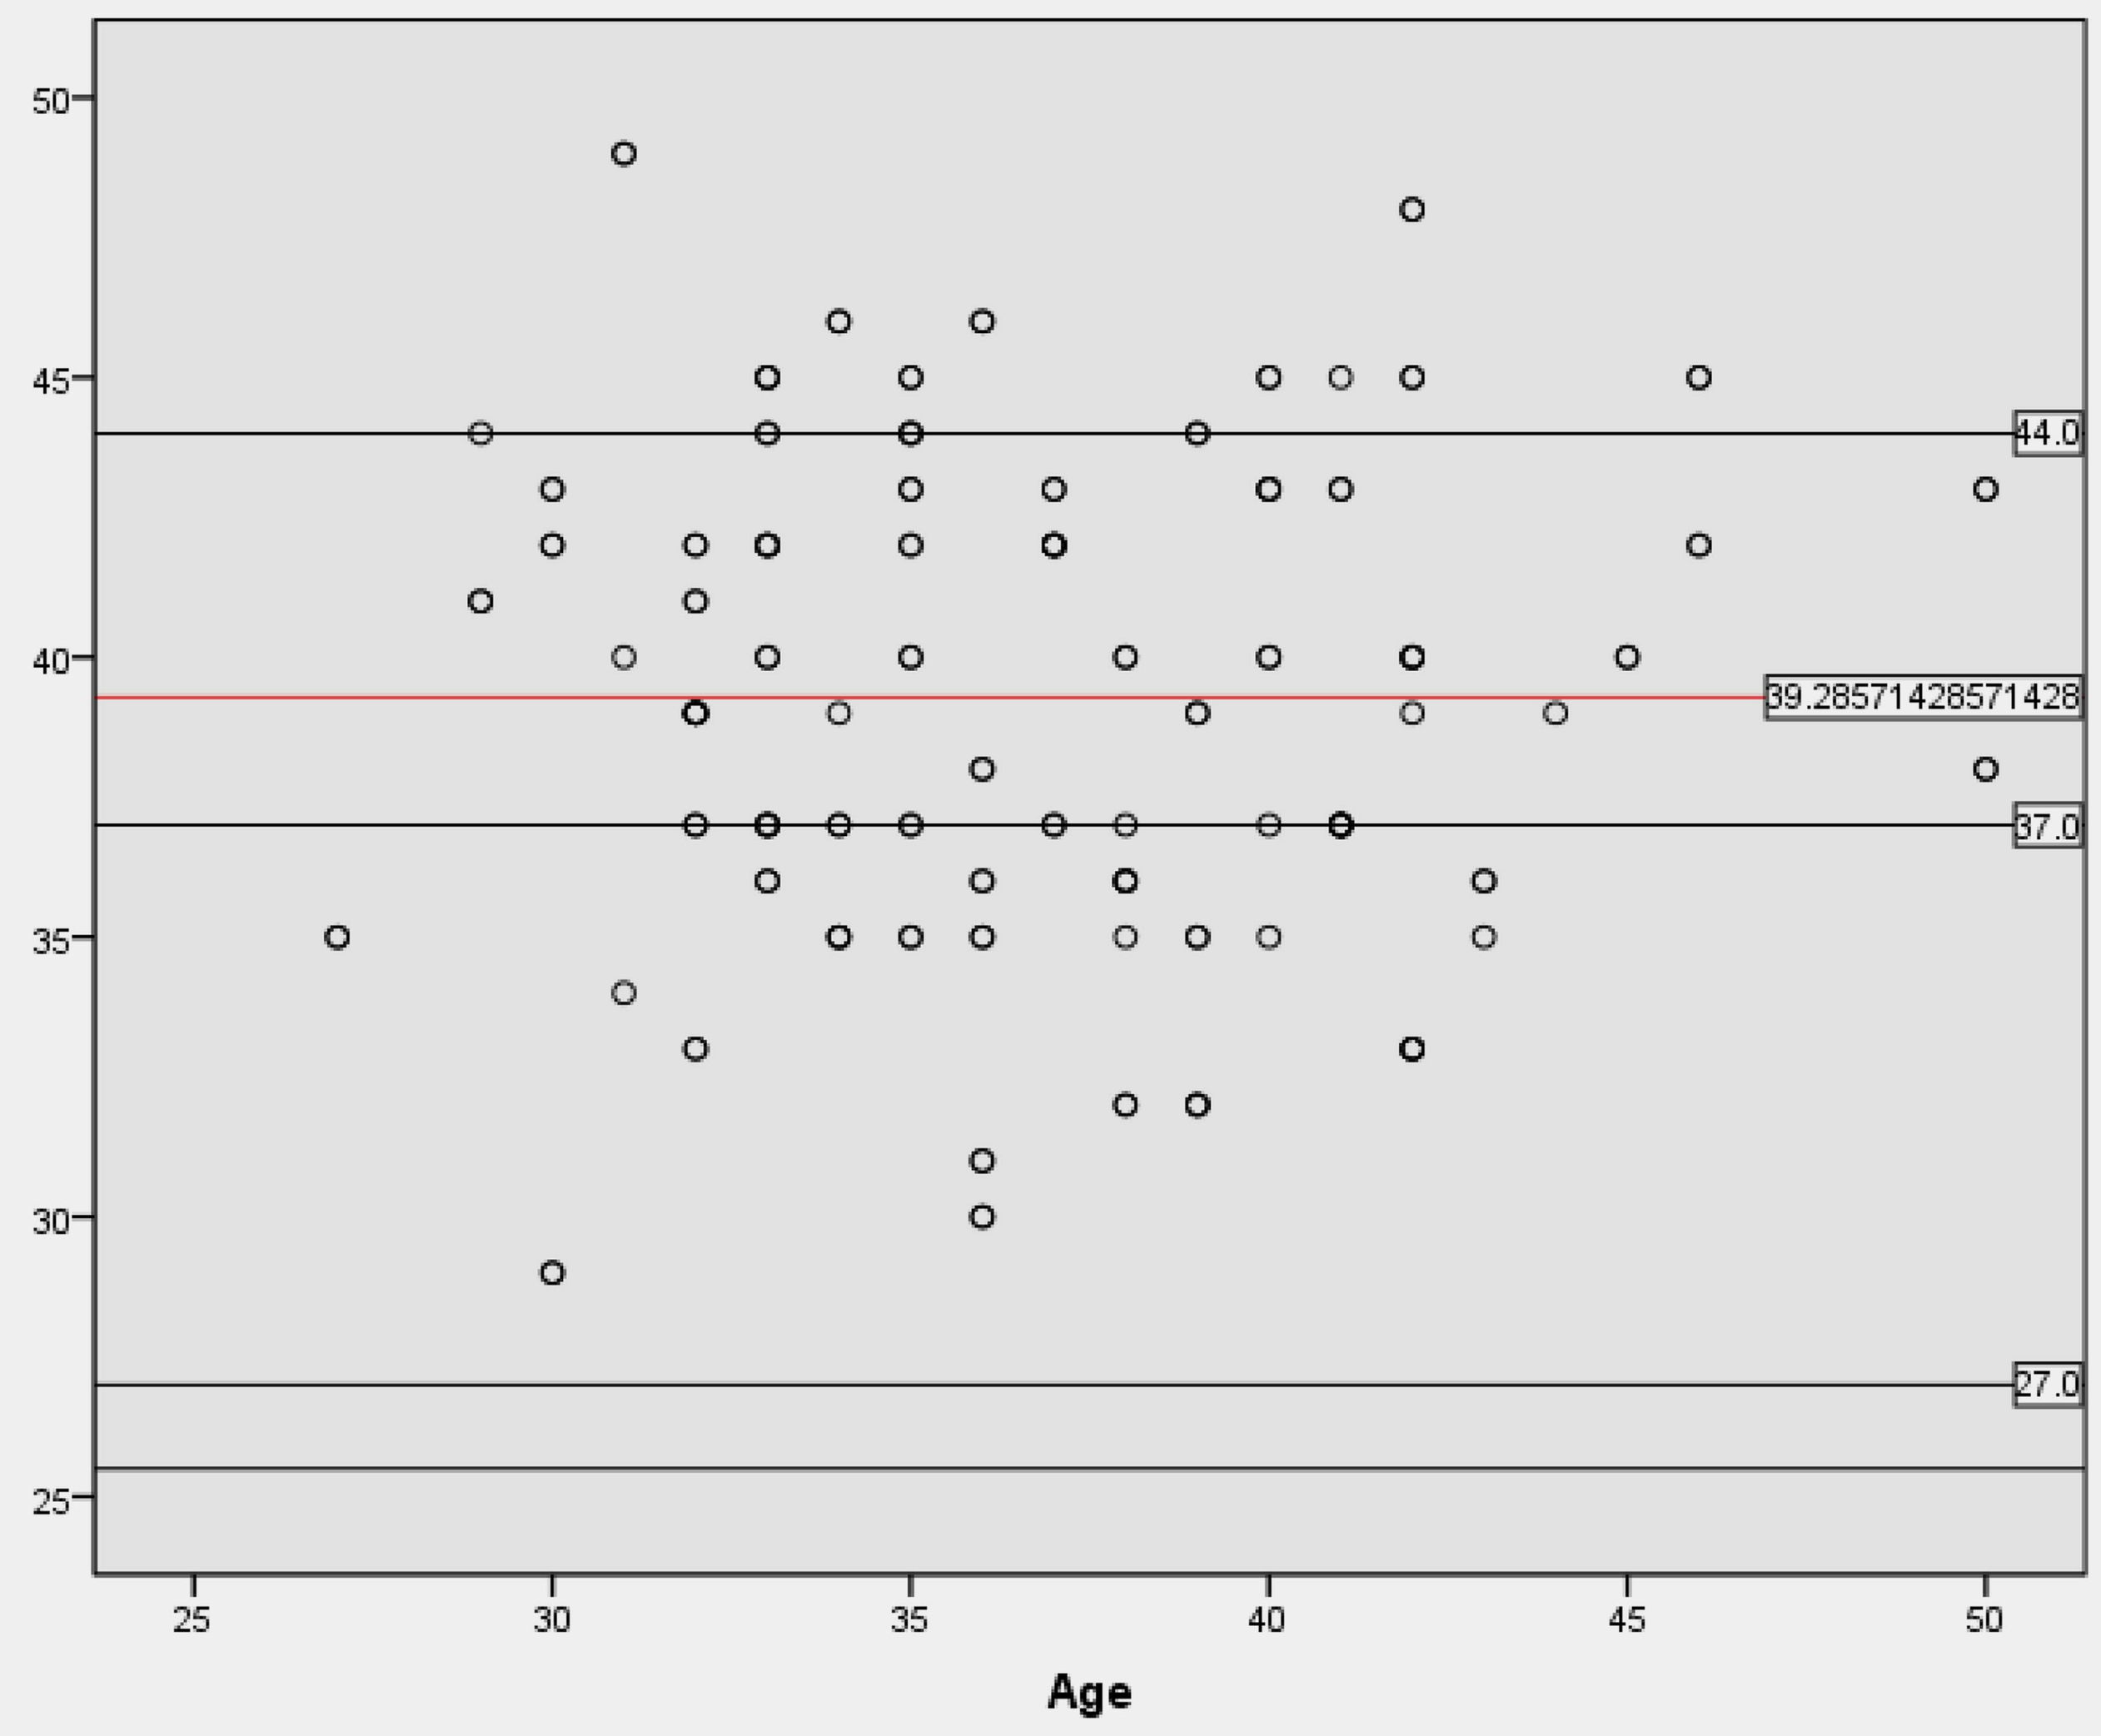
44.0
39.28571428571428
37.0
27.0
Age

CONCLUSION

The research concluded that endeavors to strengthen the workability index and quality of life among disabled workers throughout the RTW program might start by ensuring that the case manager made every effort to meet the worker's and employer's commitments to the RTW program goals. The factors that may contribute to the association of the work ability index are physical health and level of education.

KEYWORDS

Case management, disability patient, Work Ability Index, Quality of Life

CONTACT

Arie Arizandi Kurnianto

Doctoral School of Health Sciences, University of Pécs, Pécs, Hungary, 7621

E-mail: arie.arizandi@bpjsketenagakerjaan.go.id
Phone: +36304553794