COST-EFFECTIVENESS OF DAPAGLIFLOZIN IN THE TREATMENT OF CHRONIC SYMPTOMATIC HEART FAILURE WITH REDUCED EJECTION FRACTION IN THE NETHERLANDS

Ingelin Kvamme[1], Gimon de Graaf[1], Susanne Nekeman[2], Stijn Gijsbers[2], Valérie Wester[1,3], Martine Hoogendoorn[1]

[1]Institute for Medical Technology assessment, Rotterdam; [2]AstraZeneca Netherlands [3]Erasmus School of Health Policy & Management, Rotterdam

✉ Kvamme@imta.eur.nl

This study was financially supported by AstraZeneca

AstraZeneca

Objective

Heart failure is a leading cause of hospitalization and mortality worldwide, and 242,300 cases were reported in 2018 in the Netherlands alone. Dapagliflozin is a SGLT2 inhibitor registered for treatment for chronic symptomatic heart failure with reduced ejection fraction, among other indications. Aim of this study was to estimate the cost-effectiveness of dapagliflozin in Dutch patients with symptomatic chronic heart failure and a reduced ejection fraction (HFrEF) when adding dapagliflozin to standard of care (SoC). Dapagliflozin and SoC were compared to SoC alone. The SoC was based on the background treatment as observed in the DAPA-HF trial, which was in accordance with international and national guidelines for SoC for HF treatment [1-2].

Figure 1: Markov model for heart failure

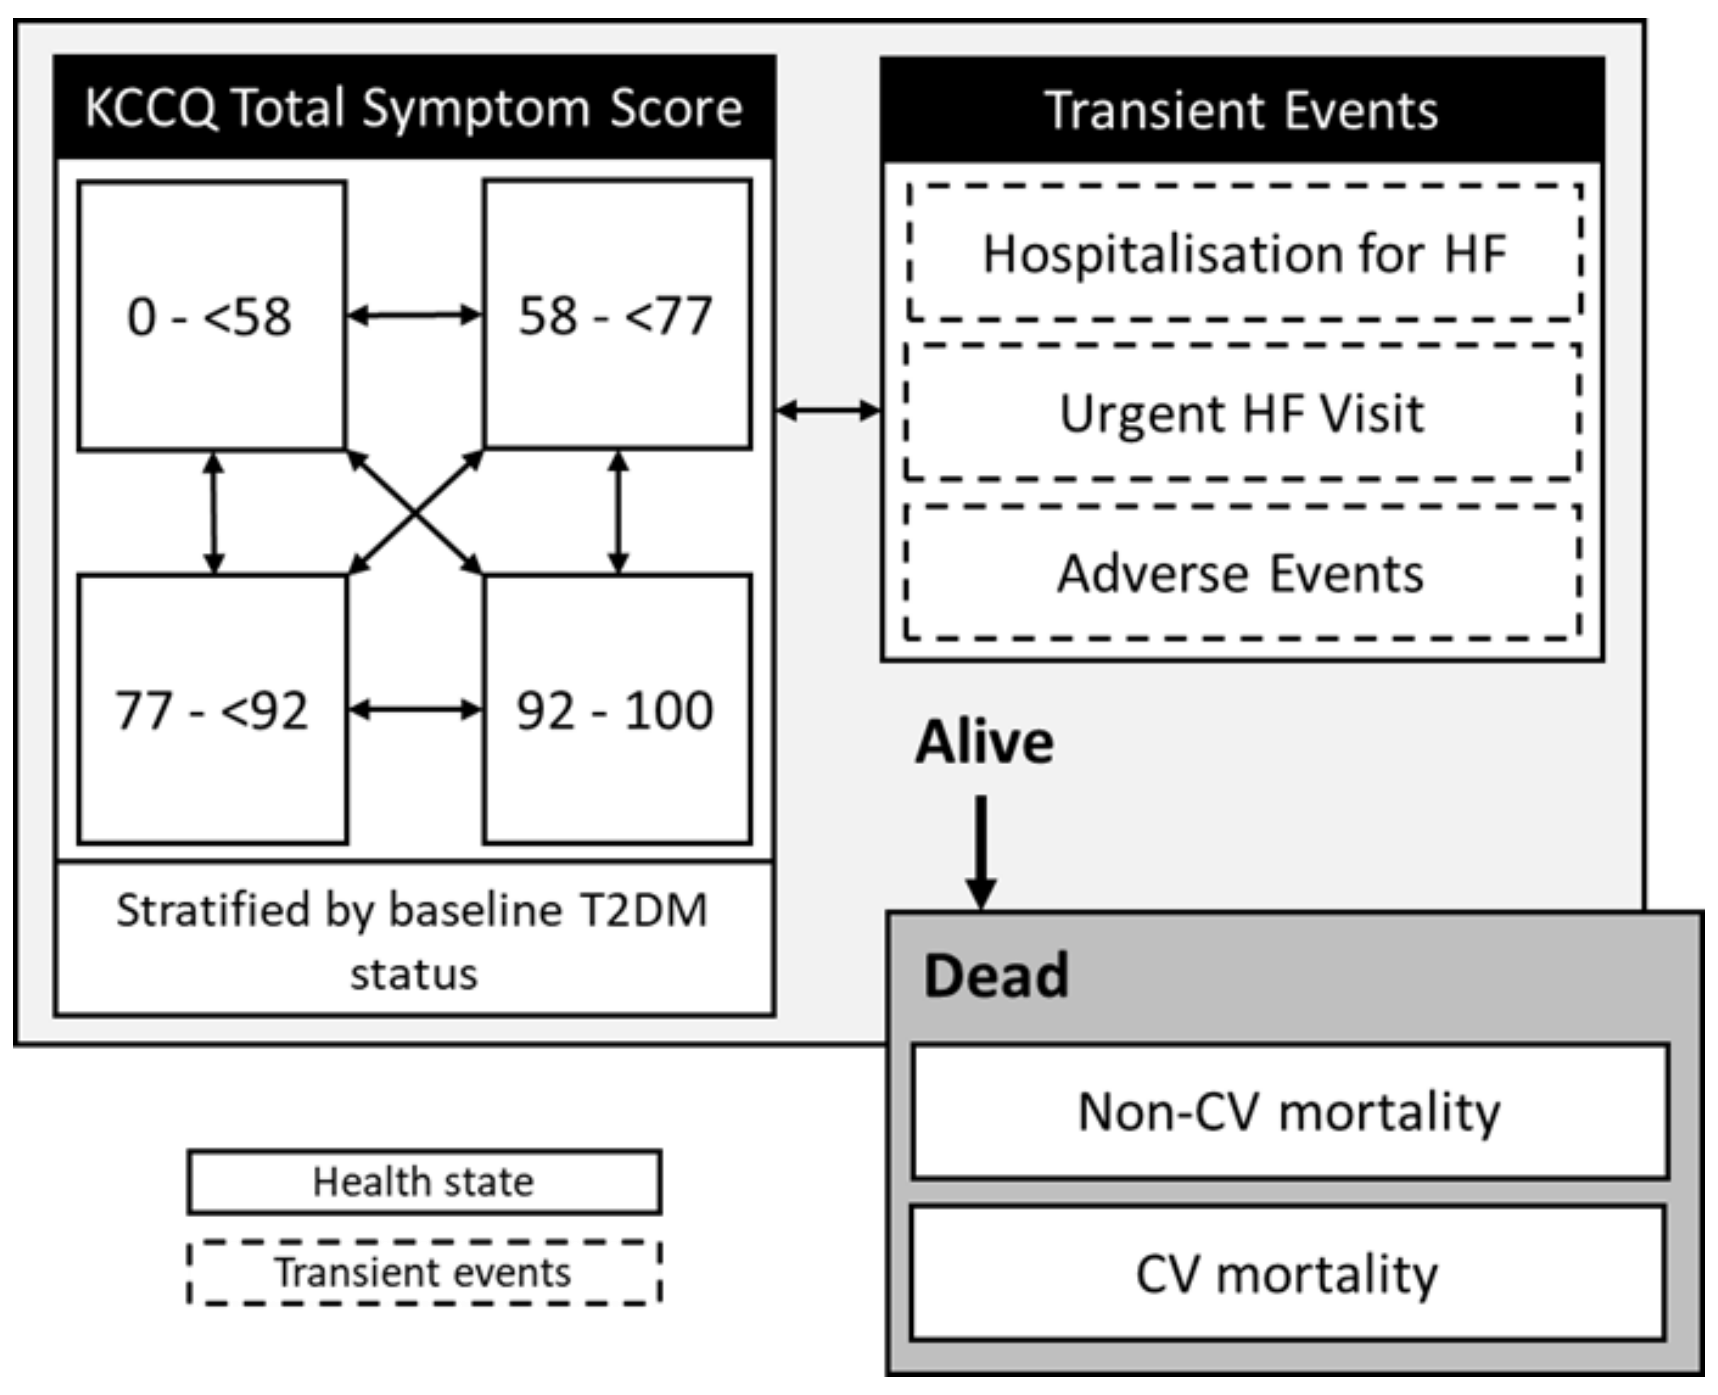
KCCQ Total Symptom Score
0 - <58
58 - <77
77 - <92
92 - 100
Stratified by baseline T2DM status
Transient Events
Hospitalisation for HF
Urgent HF Visit
Adverse Events
Alive
Dead
Non-CV mortality
CV mortality
Health state
Transient events

Methods

A Markov model for heart failure with a one-month cycle length (see figure 1) was developed for assessing dapagliflozin [3]. The analysis was adapted to Dutch guidelines by adopting a lifetime horizon (maximum age limited to 101 years) and a societal perspective, including all costs within the health care sector, patient and family costs like patients' travel costs, informal care costs. Productivity costs were excluded due to the high age of patients. Disease progression was modelled through transitions between discrete health states characterized by Kansas City Cardiomyopathy Questionnaire total symptom score (KCCQ TSS) quartiles. Furthermore, the incidence of hospitalizations for heart failure (HHF) and urgent heart failure visits (UHFV) was modelled. The health states and incidence of events were associated with costs and utility decrements.

Model input

In the DAPA-HF trial, dapagliflozin was found to be associated with statistically significant reductions in the primary composite outcome of cardiovascular (CV) death, hospitalization for HF (HHF) or urgent HF visit (UHFV) as a whole (hazard ratio: 0.74, 95% confidence interval: 0.65–0.85), and for each of the individual component endpoints (CV death: 0.82 [0.69–0.98], HHF: 0.70 [0.59–0.83], and UHFV: 0.43 [0.20–0.90]). Dapagliflozin also resulted in a nominal statistically significantly reduction in the incidence of all-cause mortality (ACM) (0.83, 0.71–0.97) [4].

Patient characteristics at initiation were based on the CHECK-HF study [5] (Mean age: 71 years, 34% females, 25% diabetes), which was considered representative for the Dutch HF population. Additional characteristics were sourced from the patient population in the DAPA-HF Trial. Transition rates, risks for events, mortality (all-cause mortality and cardiovascular specific mortality) and utilities were sourced from the DAPA-HF trial data. Additional input parameters for cost and utility decrements came from the Dutch costing manual, literature, and clinical expert opinion. Costs were discounted by 4.0%, effects by 1.5%. Beside the base-case analysis, one-way sensitivity analyses (OWSA), probabilistic sensitivity analysis (PSA) and scenario analyses were performed. A burden of disease analysis using the iMTA iDBC tool showed that the relevant cost-effectiveness threshold for the patient population was €50,000/QALY in the Netherlands.

Results

The base case analysis resulted in a 0.55 gain in QALYs for dapagliflozin + SoC compared to SoC alone (table 1). Cost for dapagliflozin were €3,593 higher, mainly due to the higher acquisition costs of dapagliflozin, which were somewhat compensated by a reduction in hospitalization costs. The resulting ICER was €6,507 per QALY gained. One-way sensitivity analysis varying input parameters by 20% showed that results were most sensitive to the input values used for health state utilities, societal costs and intervention costs (Figure 2). At a threshold of €50,000 per QALY, the cost-effectiveness acceptability curve showed that the probability that dapagliflozin was cost-effective was 100% (Figure 3).

Table 1: Lifetime cost-effectiveness dapagliflozin + SoC vs SoC alone (societal perspective discounted base-case analysis)

Cost-effectiveness	Dapagliflozin + SoC	SoC	Incremental
Total overall cost	€ 31,759	€ 28,167	€ 3,593
Medication costs	€ 4,222	€ 1,299	€ 2,923
HF treatment costs	€ 7,032	€ 7,107	€ -75
Adverse event costs	€ 674	€ 630	€ 45
Societal costs	€ 19,831	€ 19,131	€ 700
Total QALYs	5.197	4.645	0.552
Total life years	6.827	6.156	0.671
Incremental cost/QALY	-	-	**€6,507**
Incremental cost/LYG	-	-	€5,356

Main findings

- **Base case results show that dapagliflozin in addition to standard of care results in an ICER of €6,507 per QALY gained.**
- **The likelihood of dapagliflozin being cost-effective at €50,000/QALY, the threshold relevant for the patient population, is 100%.**
- **The ICER is driven by gains in life-years and QALYs as a result of CV and All-Cause mortality benefits with Dapagliflozin in HFrEF patients.**
- **Dapagliflozin is a cost-effective treatment option for chronic, symptomatic heart failure with reduced ejection fraction in the Netherlands.**

Results (continued)

The most important scenario analyses are shown in Figure 4. When unrelated healthcare costs were included the ICER was €19,139. All scenario analyses resulted in ICERs below the €50,000/QALY threshold.

Figure 2: One-way sensitivity analyses for ICER

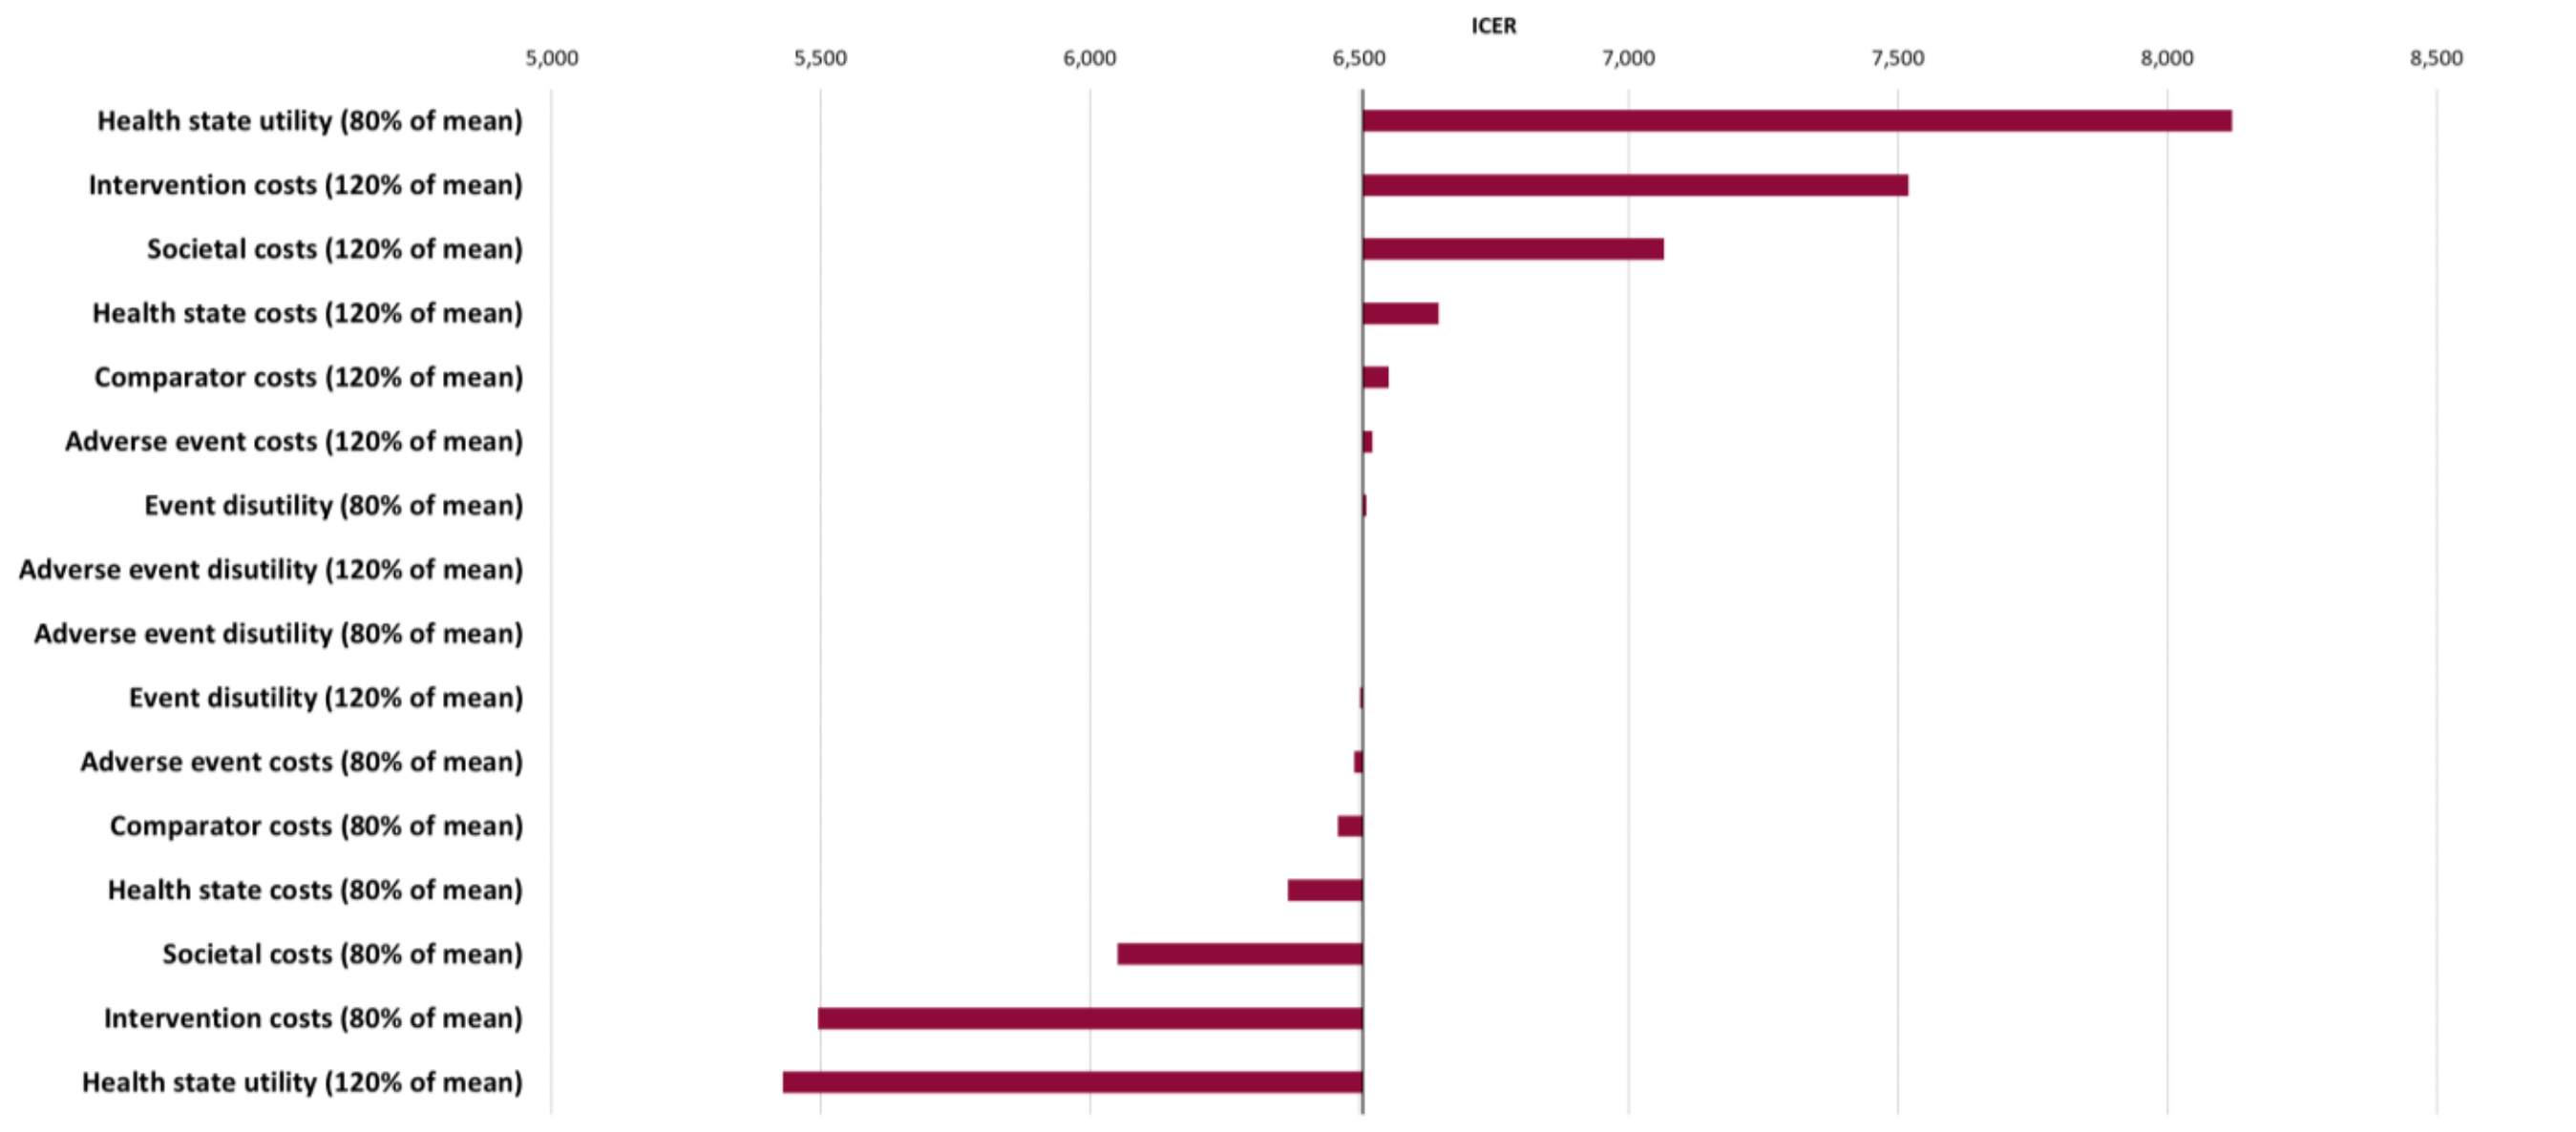
ICER
5,000
5,500
6,000
6,500
7,000
7,500
8,000
8,500
Health state utility (80% of mean)
Intervention costs (120% of mean)
Societal costs (120% of mean)
Health state costs (120% of mean)
Comparator costs (120% of mean)
Adverse event costs (120% of mean)
Event disutility (80% of mean)
Adverse event disutility (120% of mean)
Adverse event disutility (80% of mean)
Event disutility (120% of mean)
Adverse event costs (80% of mean)
Comparator costs (80% of mean)
Health state costs (80% of mean)
Societal costs (80% of mean)
Intervention costs (80% of mean)
Health state utility (120% of mean)

Figure 3: Acceptability curve dapagliflozin + SoC vs SoC alone

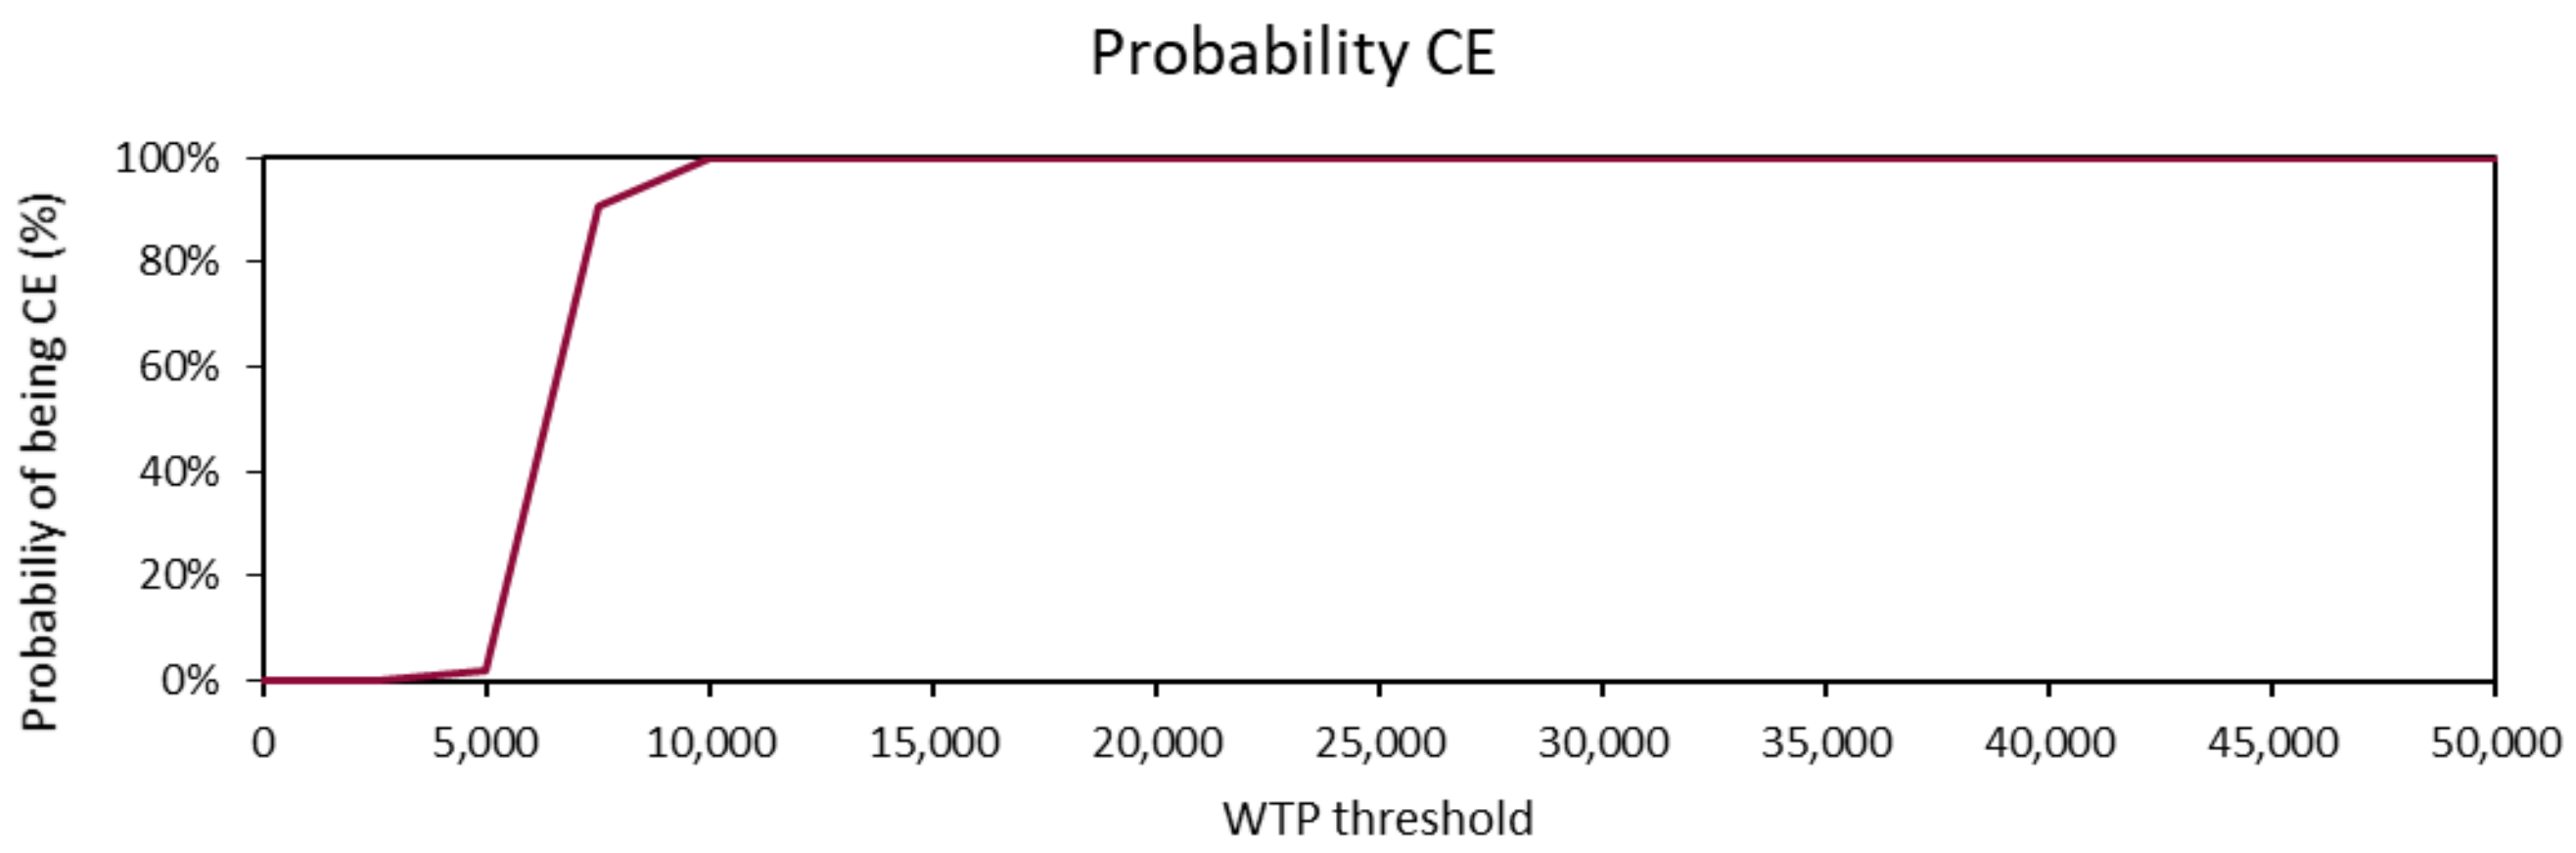
Probability CE
Probabiliy of being CE (%)
100%
80%
60%
40%
20%
0%
0
5,000
10,000
15,000
20,000
25,000
30,000
35,000
40,000
45,000
50,000
WTP threshold

Figure 4: Scenario analysis: ICER dapagliflozin + SoC vs SoC alone

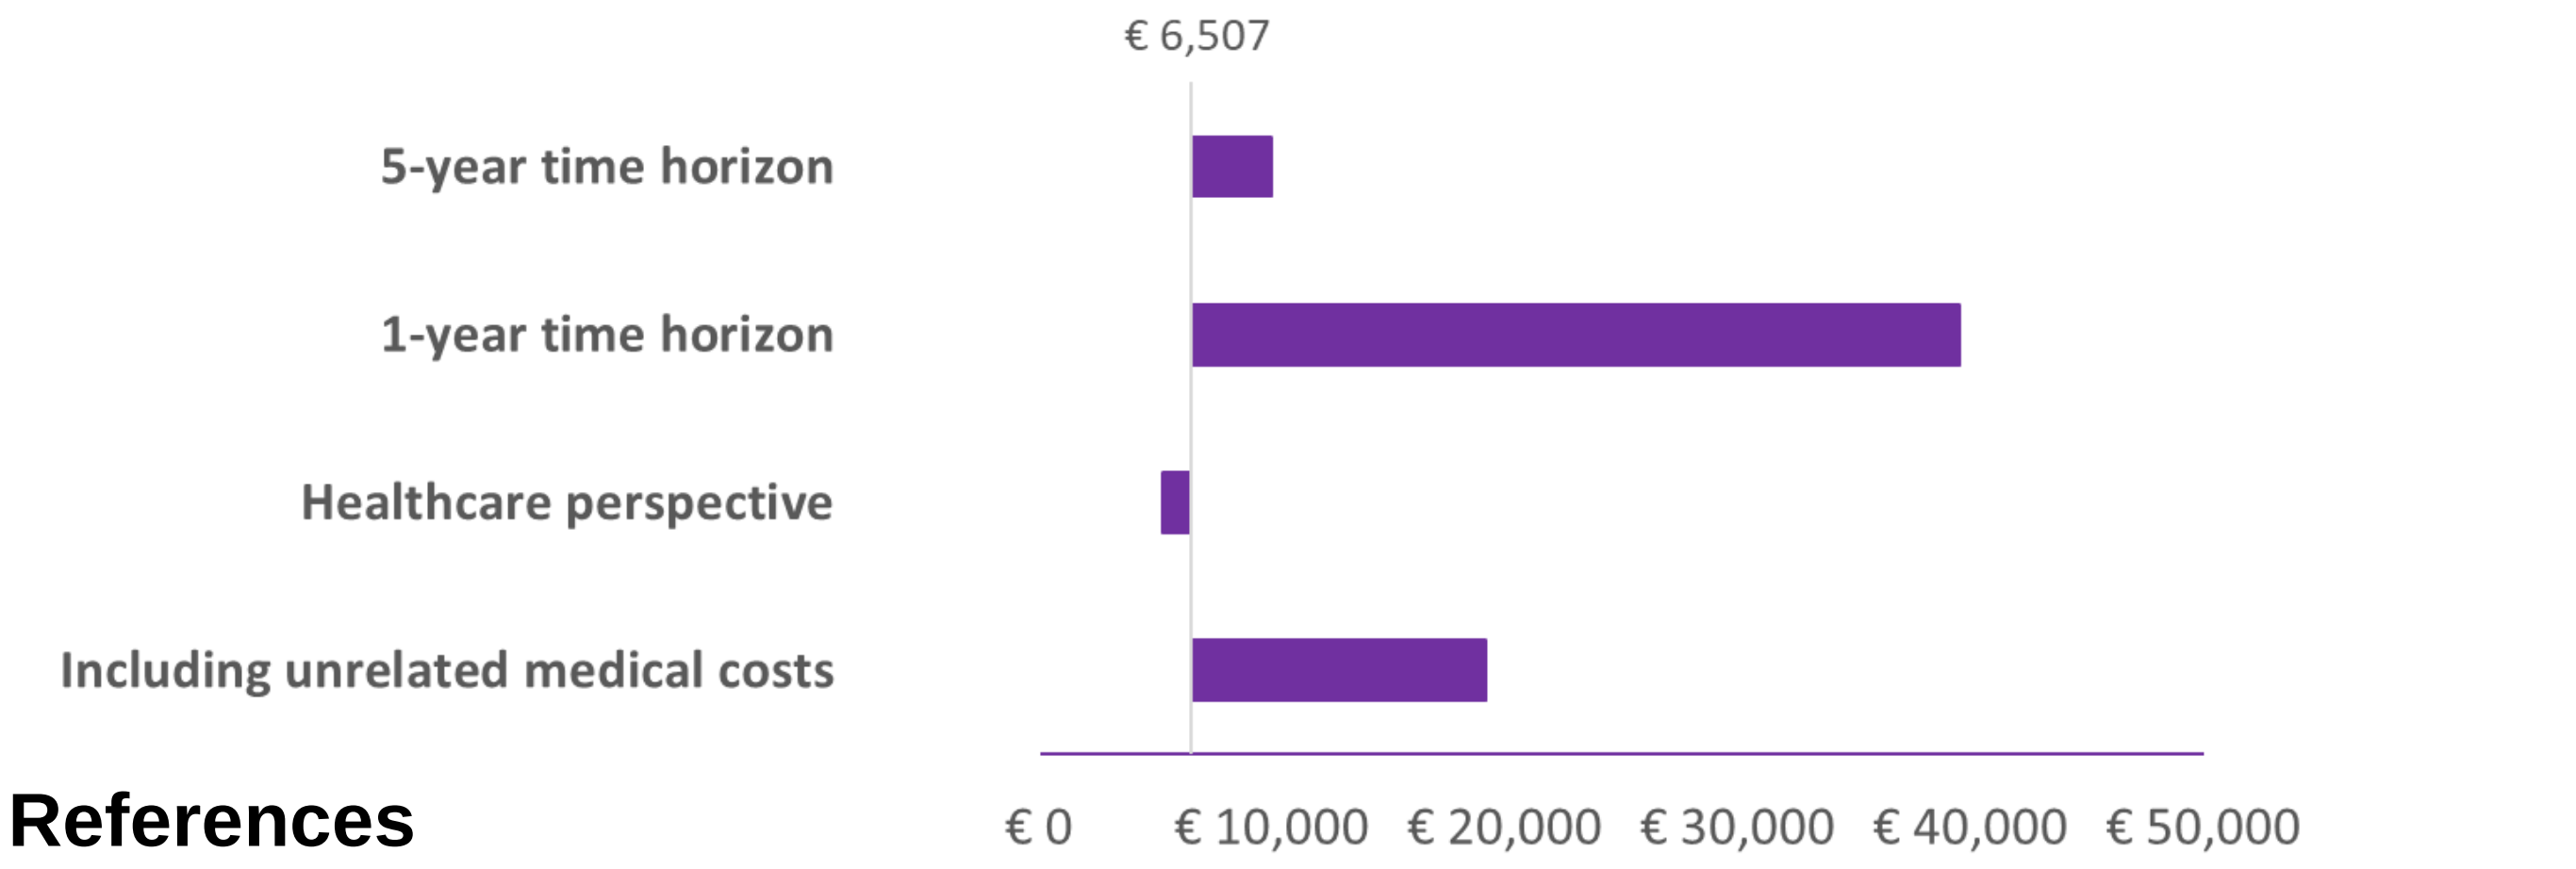
€ 6,507
5-year time horizon
1-year time horizon
Healthcare perspective
Including unrelated medical costs
€ 0
€ 10,000
€ 20,000
€ 30,000
€ 40,000
€ 50,000

References

1. Ponikowski P, et al. 2016 ESC Guidelines for the diagnosis and treatment of acute and chronic heart failure. Eur Heart J 2016;37:2129-2200m. https://doi.org/10.1093/eurheartj/ehw128.

2. Nederlandse Huisartsen Genootschap (NHG). M51 NHG-Standaard Hartfalen 2010;53.

3. McEwan P, et al. 2020 Cost-effectiveness of dapagliflozin as a treatment for heart failure with reduced ejection fraction: a multinational health-economic analysis of DAPA-HF. Eur J Hear Fail.

4. McMurray JJ, et al. 2019 Dapagliflozin in patients with heart failure and reduced ejection fraction. N Engl J Med.;381:1995–2008.

5. Brunner-La Rocca HP, et al. 2019 Contemporary Drug Treatment of Chronic Heart Failure With Reduced Ejection Fraction: The CHECK-HF Registry. JACC Hear Fail.;7(1):13–21.

Erasmus University Rotterdam

Make it happen.

ERASMUS UNIVERSITEIT ROTTERDAM

iMTA